ePoster at the European Academy of Neurology (EAN) 2023, July 1–4, 2023

EPO-248

Erenumab discontinuation in migraine patients: final results of the APOLLON study

Hartmut Göbel[1], Mirja Koch[2], Cordula Weiss[3]

[1]Kiel Migraine and Headache Center, Kiel, Germany; [2]Novartis Pharma AG, Basel, Switzerland; [3]Novartis Pharma GmbH, Nuremberg, Germany.

CONCLUSIONS

1 **About 15% of patients participating in APOLLON availed of the option of taking a drug holiday and interrupted their treatment for about 89 days (median). After treatment interruption, most patients returned to the same dose of erenumab as before their drug holiday.**

2 **Patients who were on acute headache medication reported a slight increase in use of acute headache medication during treatment interruption. Within three months after returning to erenumab, they were able to reduce the intake of acute headache medication to pre-drug holiday levels.**

3 **The average number of MHDs and MMDs increased from 4.6±3.3 to 7.3±5.3 and 4.0 ±2.8 to 7.0±5.1 days respectively within two months of treatment interruption. After re-uptake of treatment following their drug holiday, patients reported a decrease to 5.2±3.3 MHDs and 4.7±3.2 MMDs in the first month.**

4 **Providing insights into the patients´ response after erenumab treatment discontinuation and subsequently after the re-uptake of treatment, the results can further inform current guidelines on the treatment of migraine with the monoclonal antibody erenumab.**

Presenter email address: carola.wagner@novartis.com

INTRODUCTION

- The monoclonal antibody erenumab is an EMA and FDA approved anti-CGRP pathway treatment developed for the prevention of episodic and chronic migraine.
- Erenumab was found to be effective in German and international studies.
- A recent update to the EHF guideline on the use of monoclonal antibodies suggests pausing the treatment after 12 to 18 months, yet data on the impact of treatment discontinuation is limited. The APOLLON study (Assessment of Prolonged safety and tOLerability of erenumab in migraine patients in a Long-term OpeN-label study) assessed long-term data and the impact of treatment interruption in migraine patients in Germany who previously participated in a 24-week, head-to-head trial comparing the tolerability of erenumab and topiramate (HER-MES, NCT03828539).

OBJECTIVE

- Assessment of long-term safety and tolerability of erenumab in migraine patients.
- Assessment of relevance, characteristics and impact of treatment discontinuation in patients previously treated with erenumab.

METHODS

Study Design

The APOLLON study (Figure 1) was an open-label study with the following phases:

- Screening phase: up to 2 weeks
- Open-label treatment: 128 weeks; during the open-label treatment epoch, it was at the discretion of the treating physician to change the erenumab dose at each planned visit from 70 mg to 140 mg or vice versa..
- Optional drug holiday: up to 24 weeks; a drug holiday could be initiated after at least 12 weeks of treatment. Impact of treatment discontinuation on monthly migraine days was assessed 4 weeks prior to, during and 12 weeks after drug holiday.
- Follow-up phase: 4 weeks; part of routine safety monitoring

Assessments

- Headache diary
- HIT-6 (headache impact test) questionnaire
- TSMQ (treatment satisfaction) questionnaire

Figure 1. Study Design

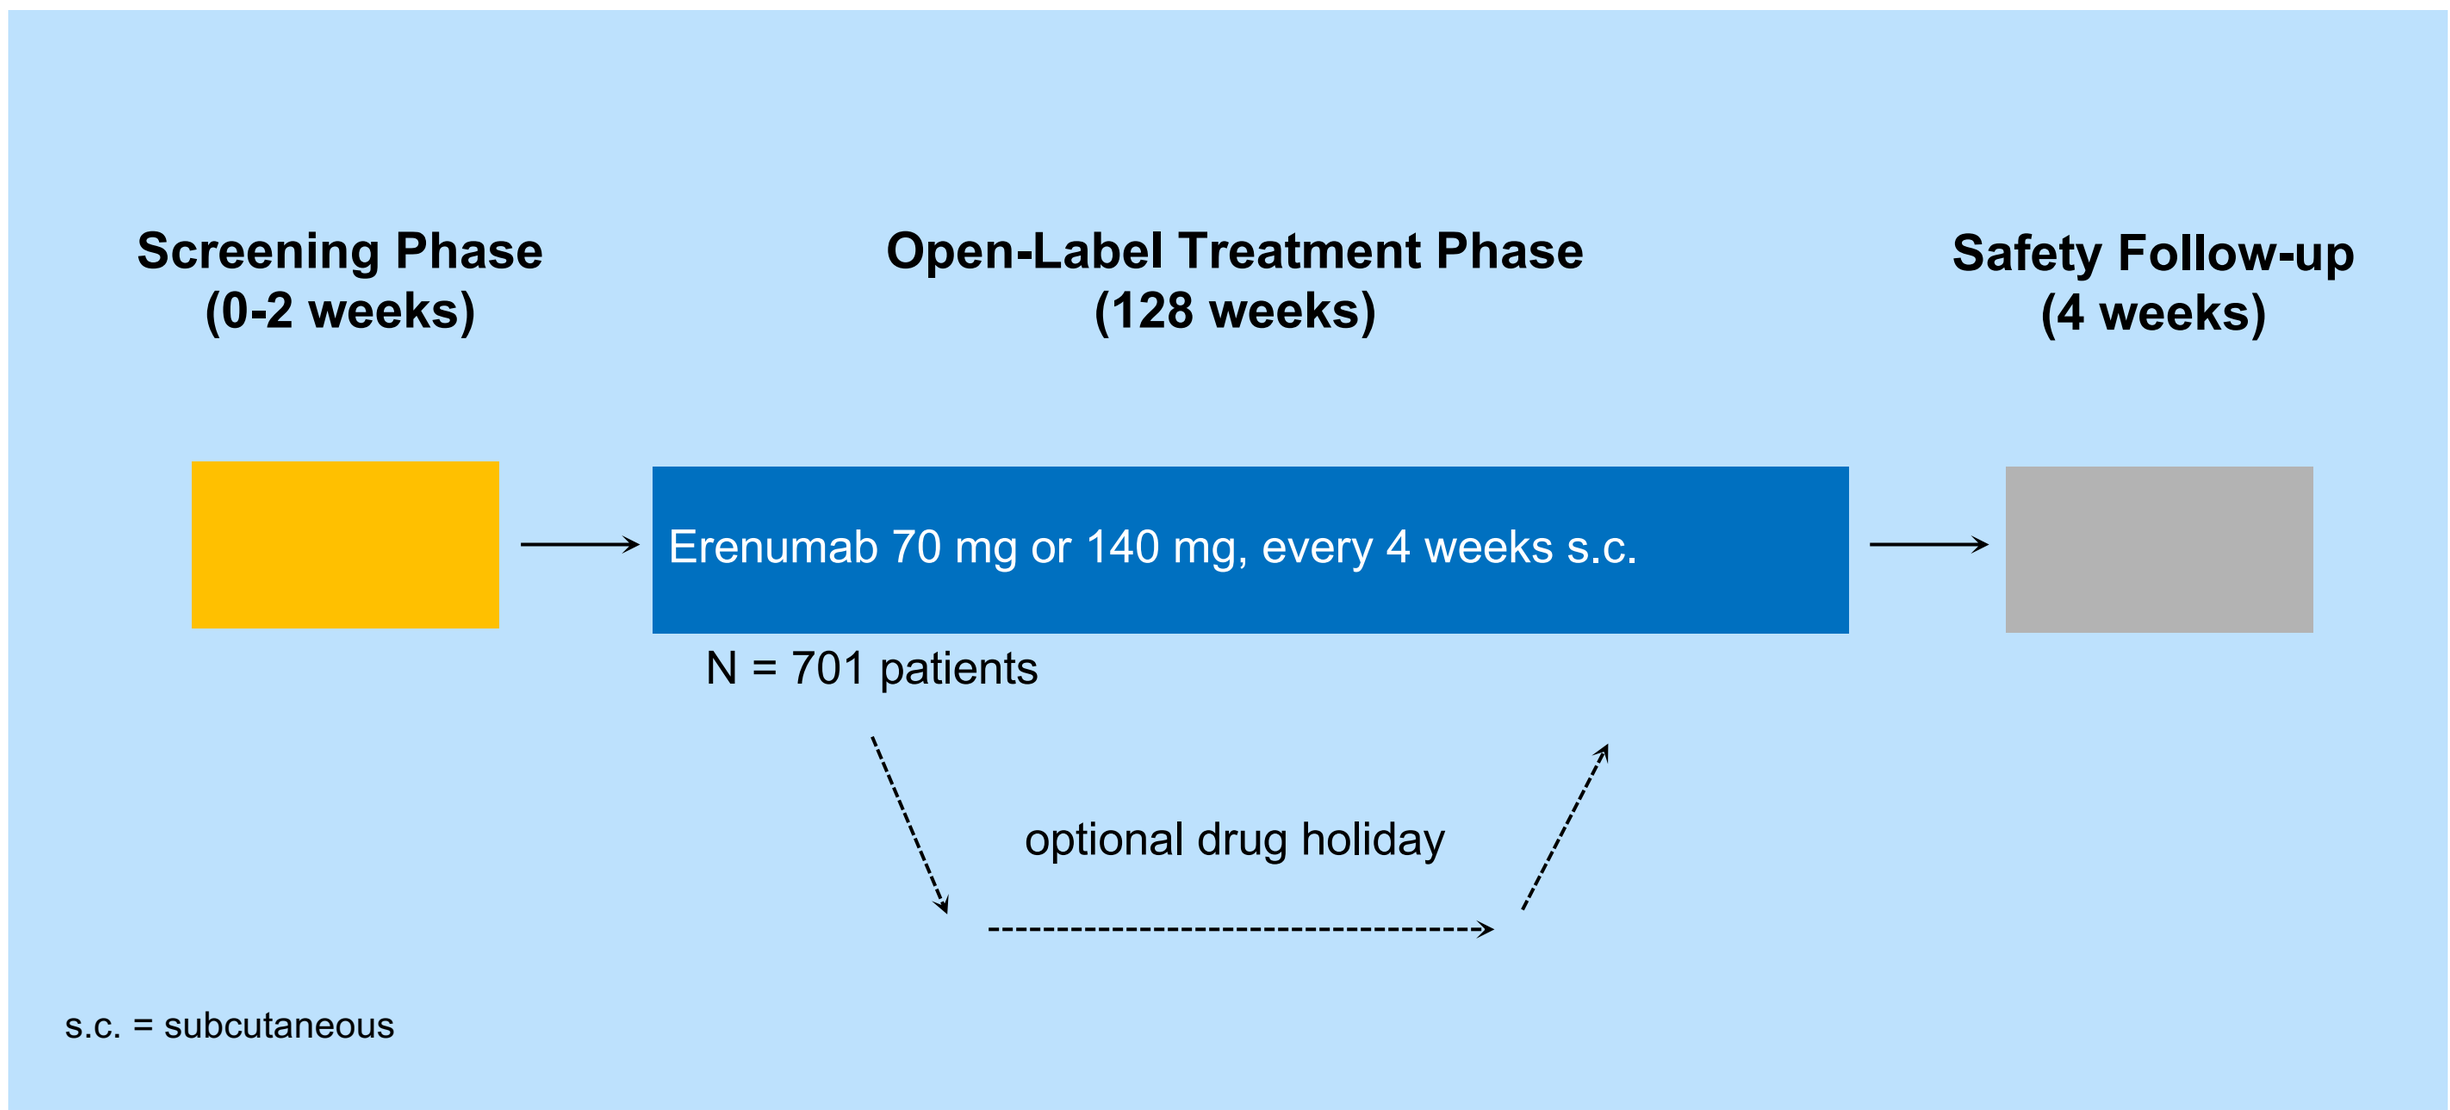
Screening Phase
(0-2 weeks)
Open-Label Treatment Phase
(128 weeks)
Safety Follow-up
(4 weeks)
Erenumab 70 mg or 140 mg, every 4 weeks s.c.
N = 701 patients
optional drug holiday
s.c. = subcutaneous

RESULTS

Patient characteristics and baseline information

- In total, 701 patients at 80 participating sites in Germany were included in the APOLLON study. Baseline characteristics shown in Tab. 1 are based on inclusion in the HER-MES study.
- Patients were on average 41.8±12.3 years old at inclusion in HER-MES.
- On average, patients had a disease duration of 22±7 years and about two thirds of patients reported suffering from migraine with aura (66.0%).
- At baseline, the average number of monthly headache days (MHDs) was 11.5±4.1 days, the average number of monthly migraine days (MMDs) was 10.4±3.8 days.
- 42.0% patients had ≥1 treatment failure prior to inclusion in HER-MES.
- Almost all patients (97.1%) took acute headache medication (Fig. 2).

Table 1. Baseline characteristics.

Baseline characteristic	Patients (N=701)
Age, years±SD	41.8±12.3
Female, n (%)	608 (86.7)
Disease duration, years±SD	22±12
Aura present, n (%)	463 (66.0)
Monthly headache days[a], days±SD	11.5±4.1
Monthly migraine days[a], days±SD	10.4±3.8
Monthly migraine days categories – stratification factor[b]	
4-7 monthly migraine days, n (%)	165 (23.6)
8-14 monthly migraine days, n (%)	470 (67.1)
≥15 monthly migraine days, n (%)	65 (9.3)
Any acute headache medication, n (%)	681 (97.1)
Migraine specific medication, n (%)	571 (81.6)
Non-migraine specific medication, n (%)	109 (15.5)

[a]Normalized to 28 days.
[b]Differs from monthly migraine days categories due to protocol deviations; n=700 due to protocol deviation.

Figure 2. Patients (n, %) with prior prophylactic treatment failures[a].

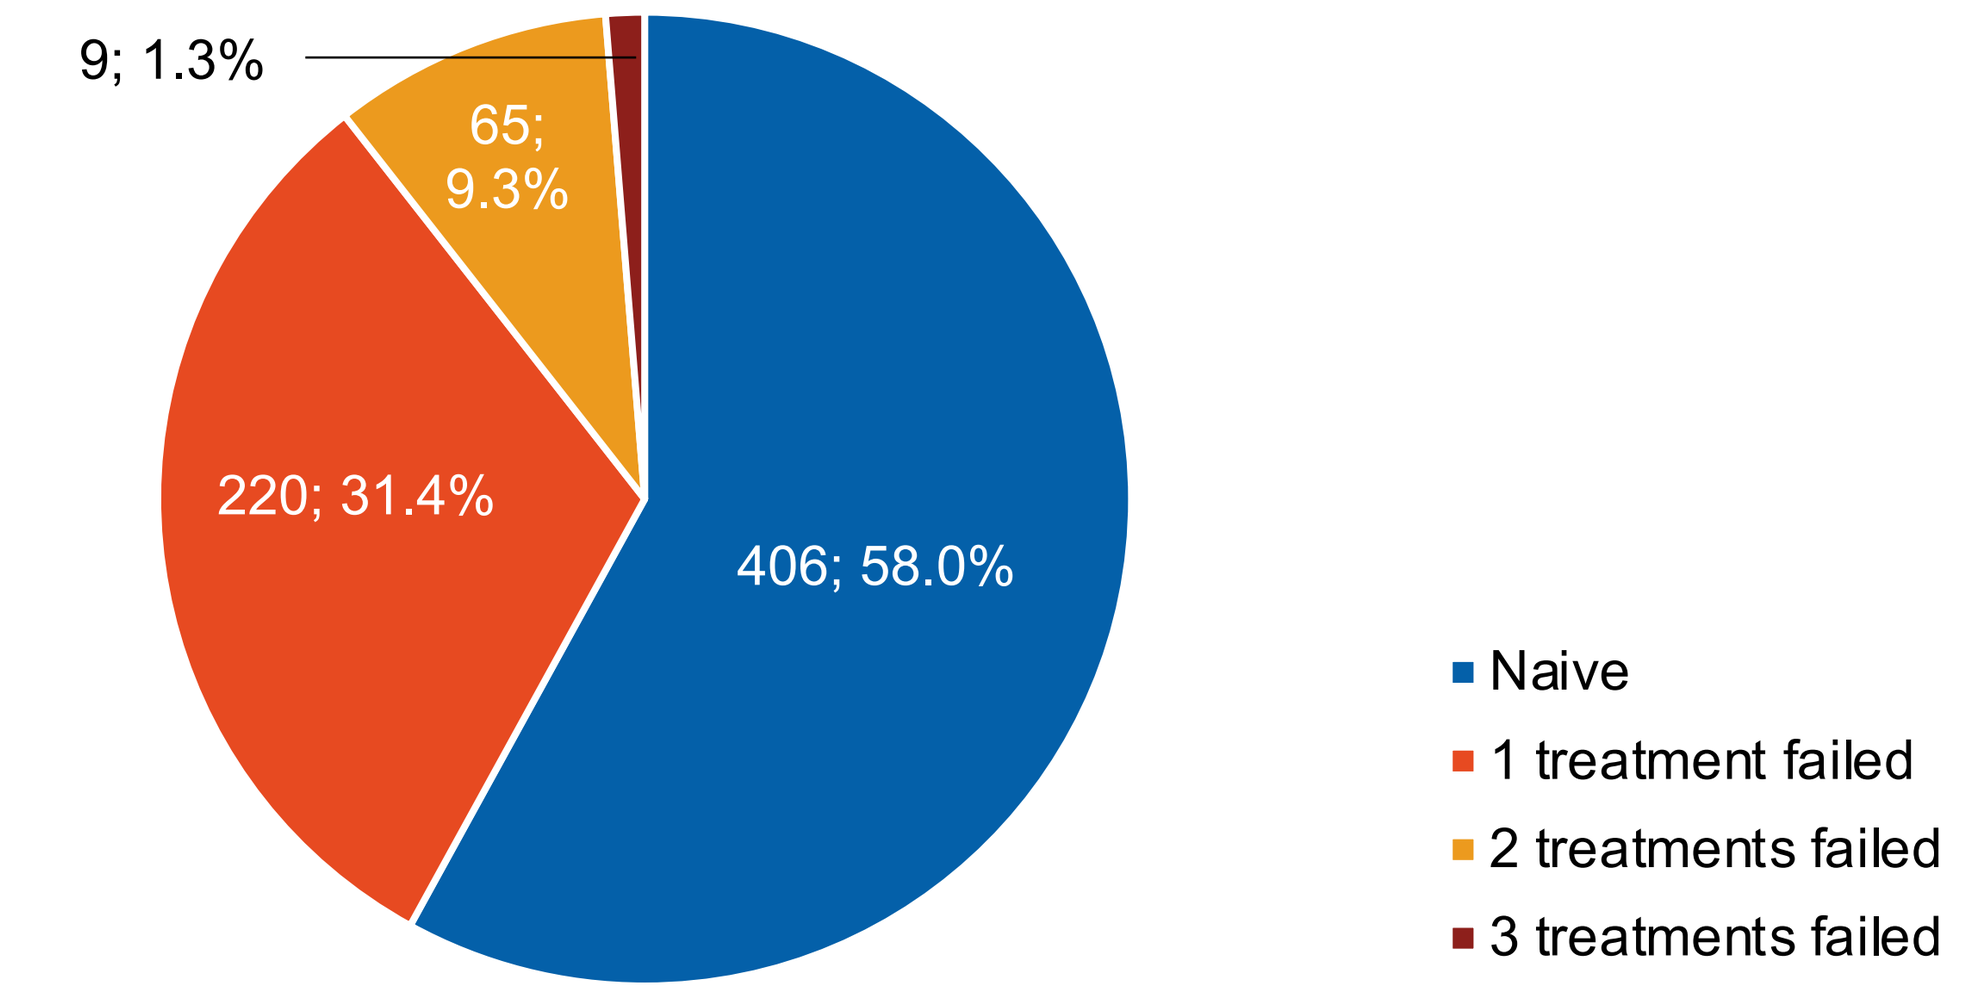
9; 1.3%
65; 9.3%
220; 31.4%
406; 58.0%
Naive
1 treatment failed
2 treatments failed
3 treatments failed

[a]Prior treatment failure of propranolol/metoprolol, amitriptyline, flunarizine.

Drug holidays

- 108 patients (15.4%) planned and took a drug holiday (Tab. 2).
- The majority of patients (64.8% of patients who took a drug holiday) returned to the study.
- Treatment interruption due to drug holiday amounted to about 89 days (median).
- Most patients (88.6% of patients who returned to the study) also returned to the same dose of erenumab as before their drug holiday. Three patients (4.3%) returned to a lower dose.

Table 2. Patients taking drug holiday.

Variable	n (%)
Study population, n	701
Drug holiday planned, n (% of study population)	108 (15.4)
Drug holiday taken, n (% of drug holiday planned)	108 (100.0)
Returned from holiday, n (% of drug holiday taken)	70 (64.8)
Drug holiday duration in days, mean±SD [min; median; max]	138±127 [27; 89; 743]
Dose after returning to study treatment[a]	
Returning to previous dose, n (% of returned from drug holiday)	62 (88.6)
Returning to dose decrease, n (% of returned from drug holiday)	3 (4.3)
Returning to dose increase, n (% of returned from drug holiday)	0 (0.0)

[a]For some patients information about their dose before/after the drug holiday was missing.

Use of acute headache medication during/after drug holiday

- At drug holiday initiation, patients had an average of 3.3±2.6 days with use of acute headache medication per month (Fig. 3).
- Within two months of treatment interruption due to drug holiday, the average number of monthly days with use of acute headache medication increased to 4.9±3.6 days.
- Three months after returning to the study medication, patients reported a reduction to 3.4±2.9 monthly days with use of acute headache medication.

Figure 3. Days with use of acute headache medication drug holiday initiation and first 3 months during/after drug holiday.

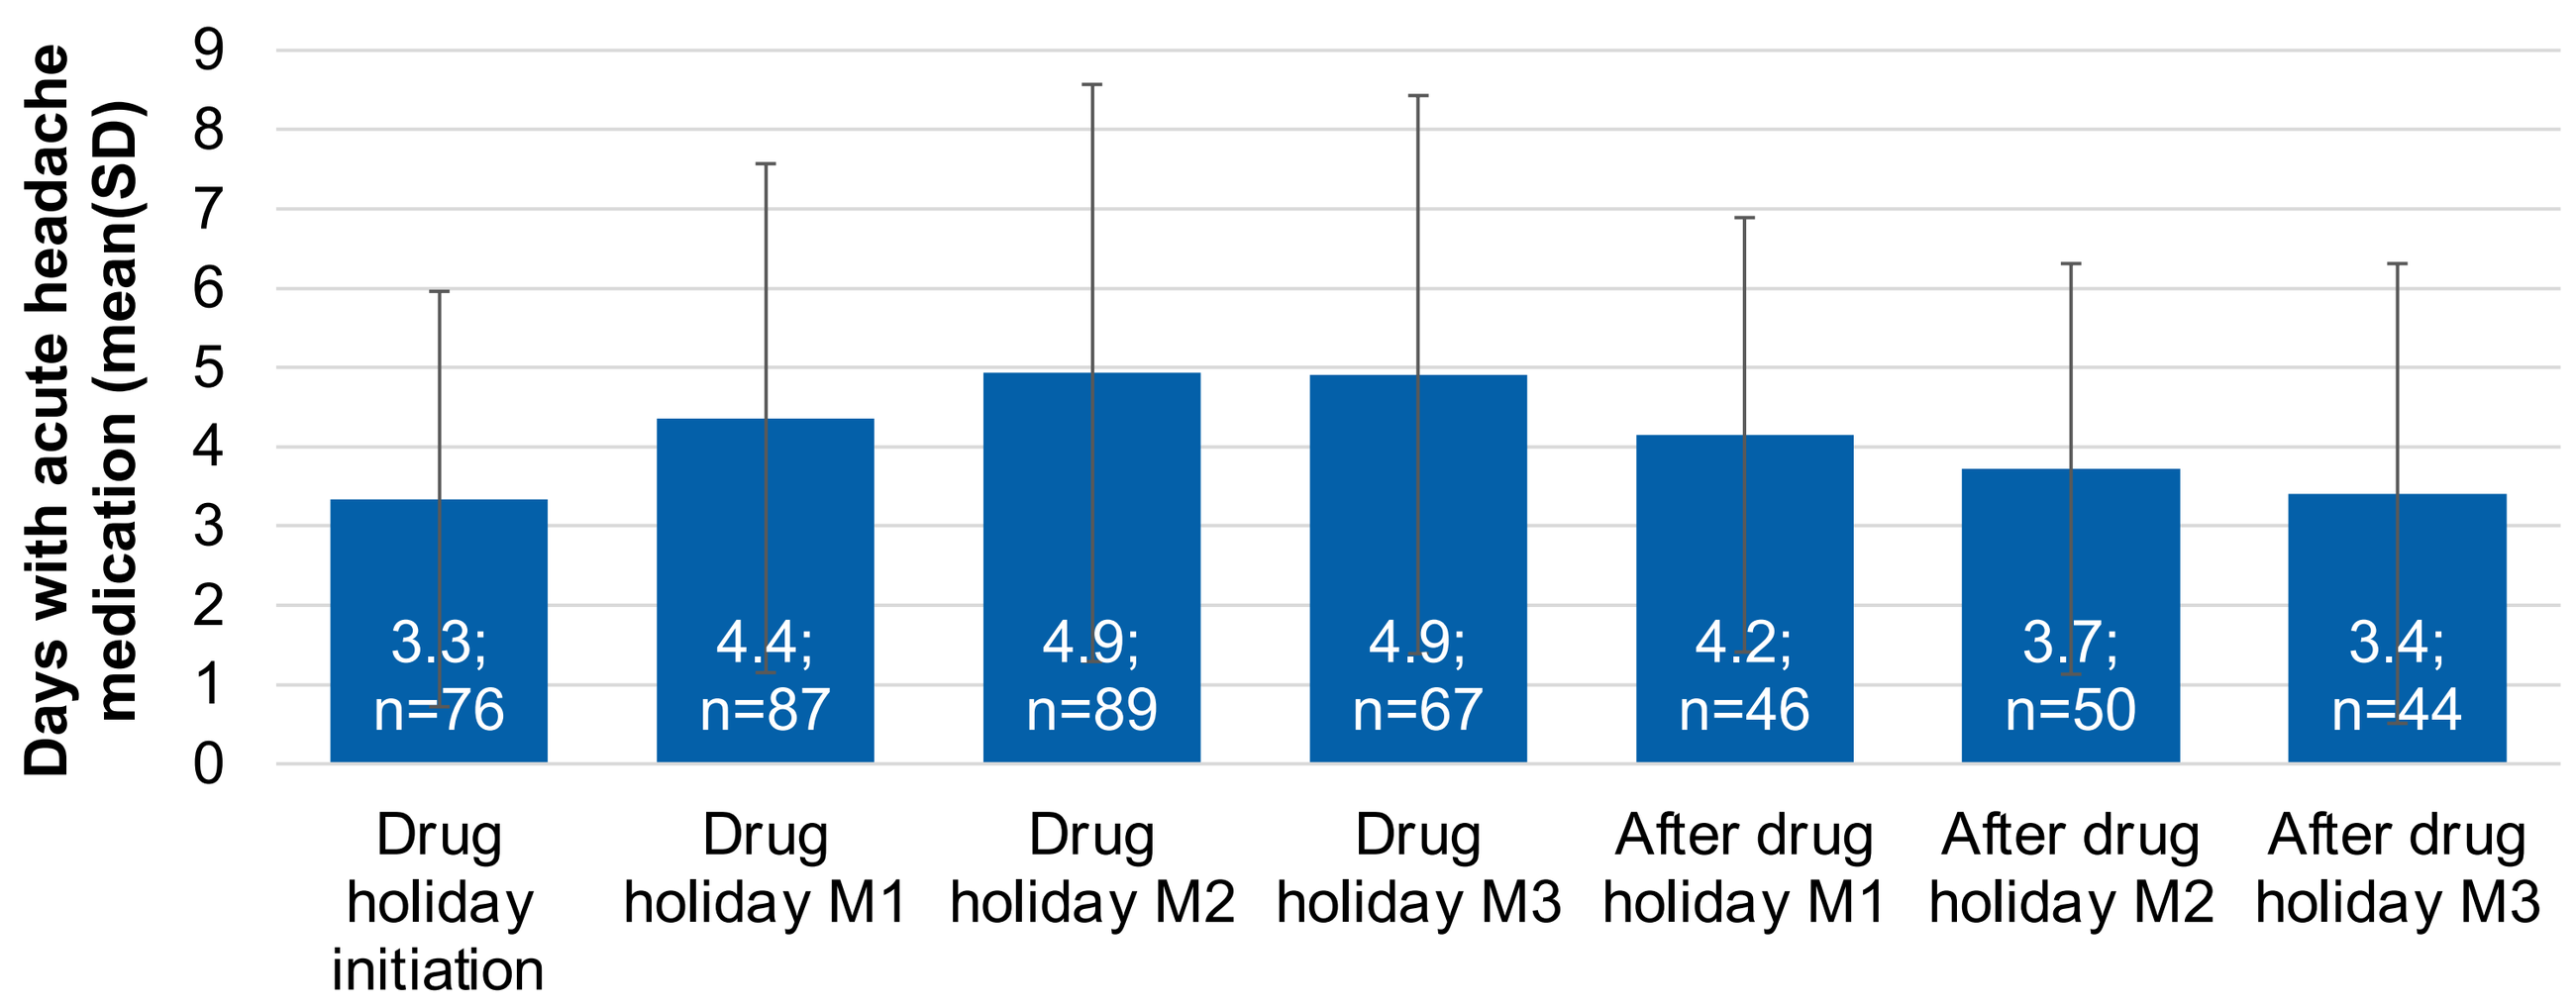
Days with acute headache medication (mean(SD)
9
8
7
6
5
4
3
2
1
0
3.3; n=76
4.4; n=87
4.9; n=89
4.9; n=67
4.2; n=46
3.7; n=50
3.4; n=44
Drug holiday initiation
Drug holiday M1
Drug holiday M2
Drug holiday M3
After drug holiday M1
After drug holiday M2
After drug holiday M3

Monthly headache and migraine days during/after drug holiday

- At drug holiday initiation, patients had an average of 4.6±3.3 MHDs which increased by about two days to 6.8±4.6 MHDs within the first month of drug holiday (Fig. 4).
- After returning to treatment, the averge number of MHDs decreased to about pre-drug holiday level (4.8±3.1 MHDs) within two months.
- The average number of MMDs at drug holiday initiation was 4.0±2.8 MMDs (Fig. 5).
- Within the first month of treatment interruption, the MMDs increased by about 2 days to an average of 6.0±4.3 MMDs and reached a relatively stable level of 7.0±5.1 MMDs on average in the second month of the drug holiday.
- After returning to treatment, patients reported a decrease to 4.7±3.2 MMDs. Overall, this value remained stable during the first three months after re-uptake of treatment.

Figure 4. Monthly headache days (MHDs) at drug holiday initiation and first 3 months during/after drug holiday.

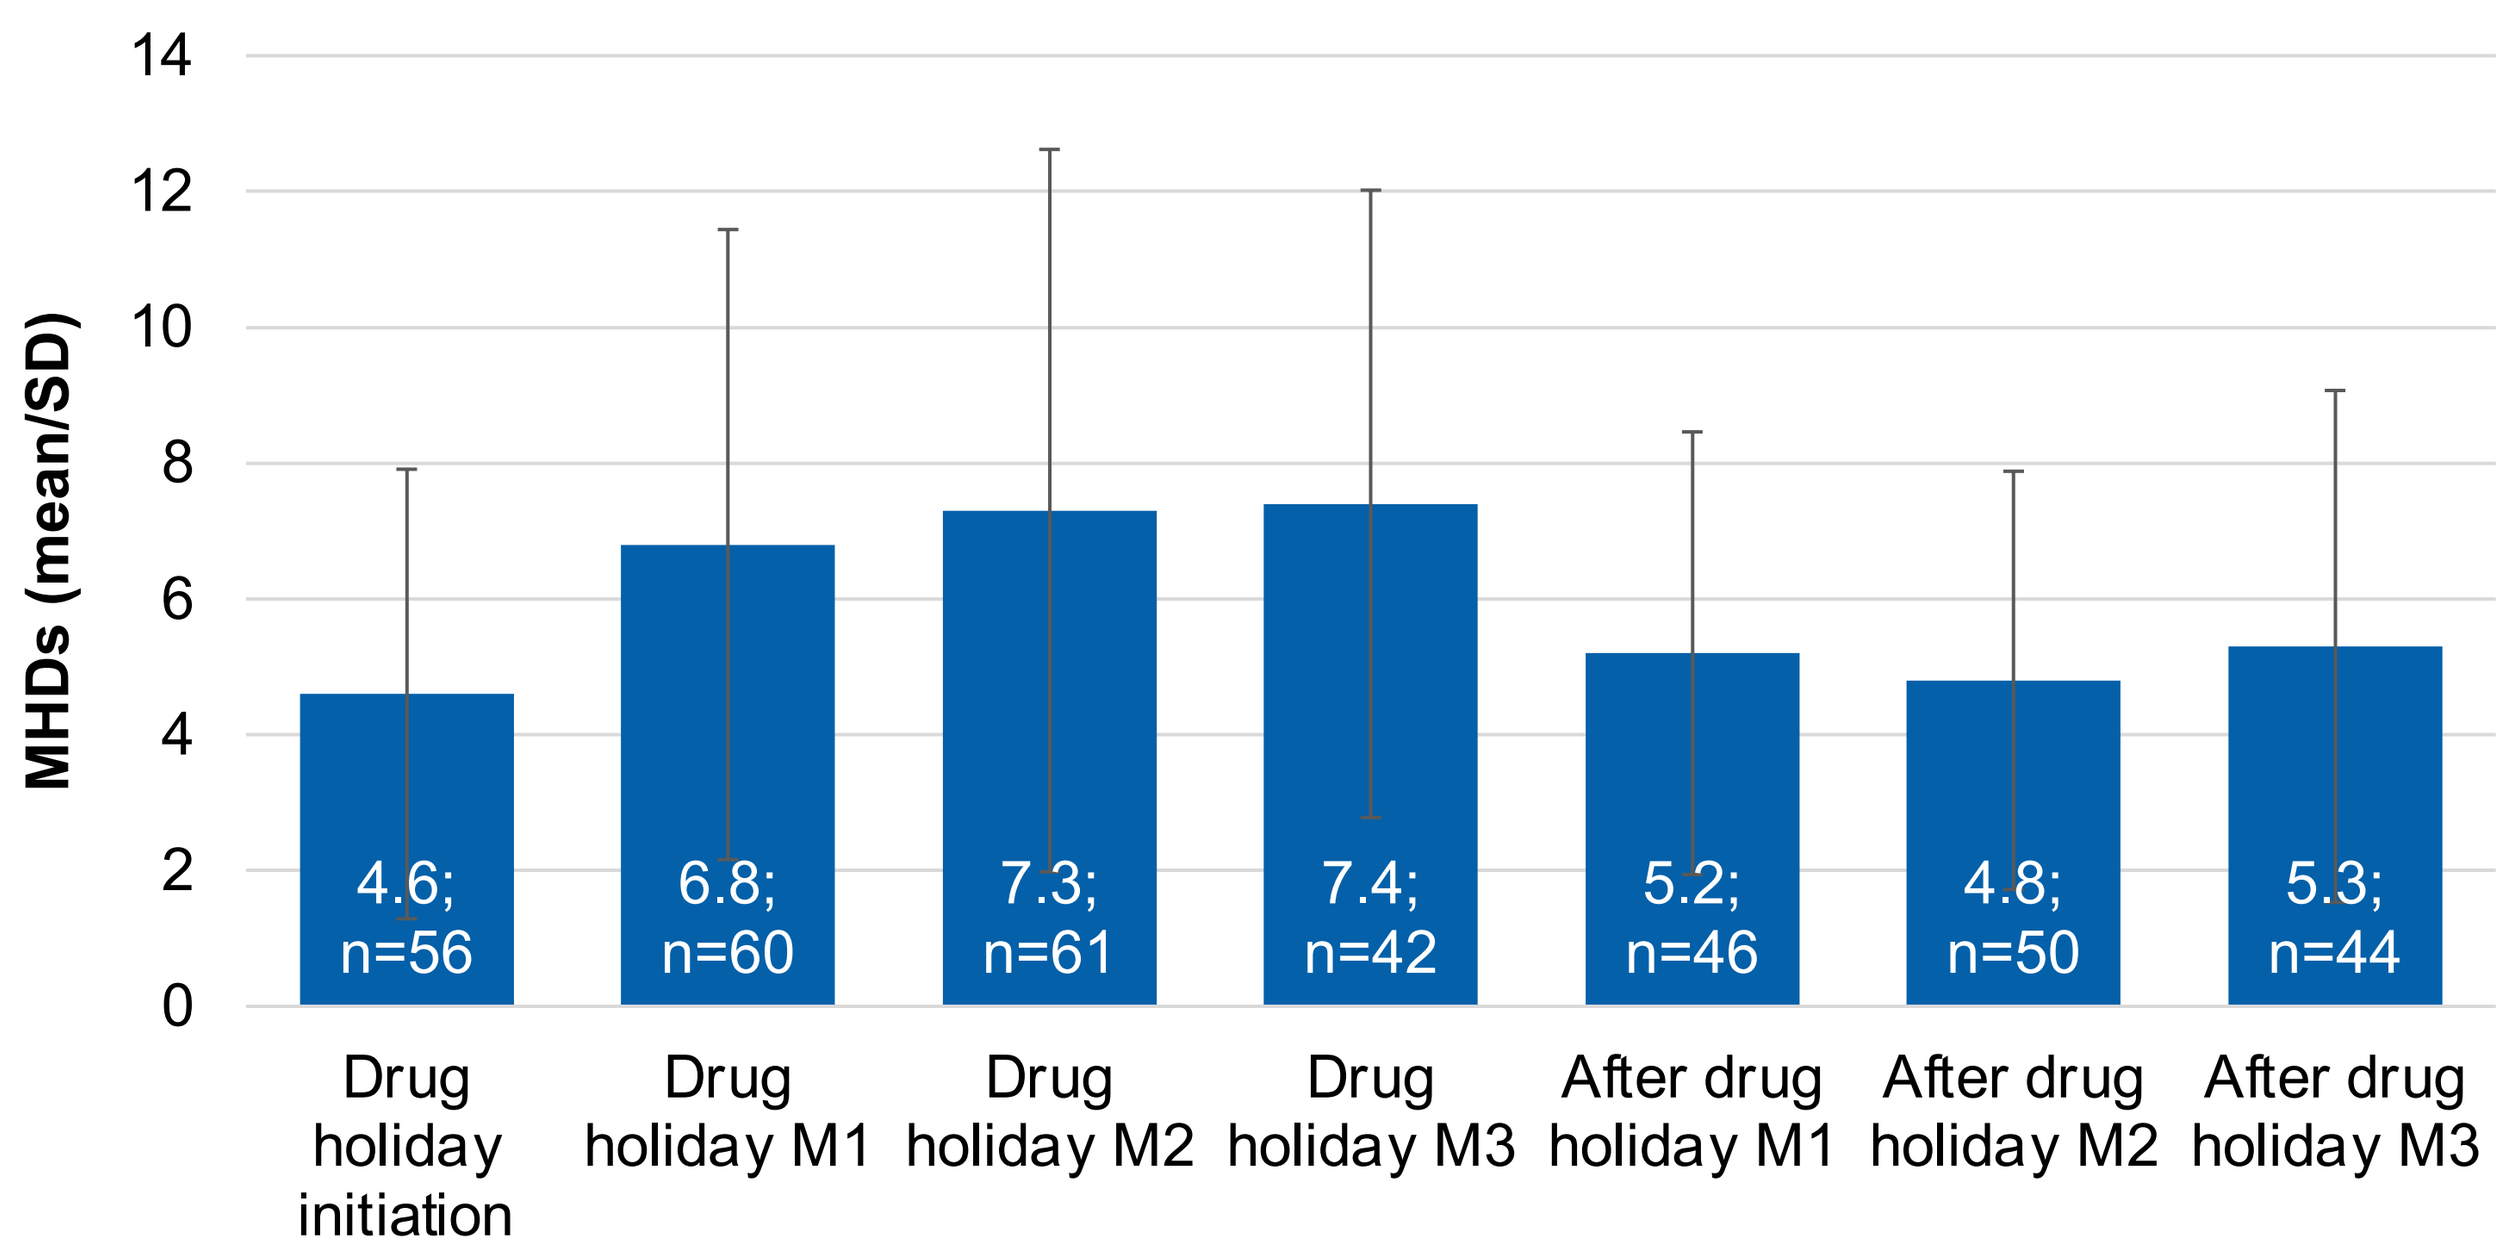
MHDs (mean/SD)
14
12
10
8
6
4
2
0
4.6; n=56
6.8; n=60
7.3; n=61
7.4; n=42
5.2; n=46
4.8; n=50
5.3; n=44
Drug holiday initiation
Drug holiday M1
Drug holiday M2
Drug holiday M3
After drug holiday M1
After drug holiday M2
After drug holiday M3

Figure 5. Monthly migraine days (MMDs) at drug holiday initiation and first 3 months during/after drug holiday.

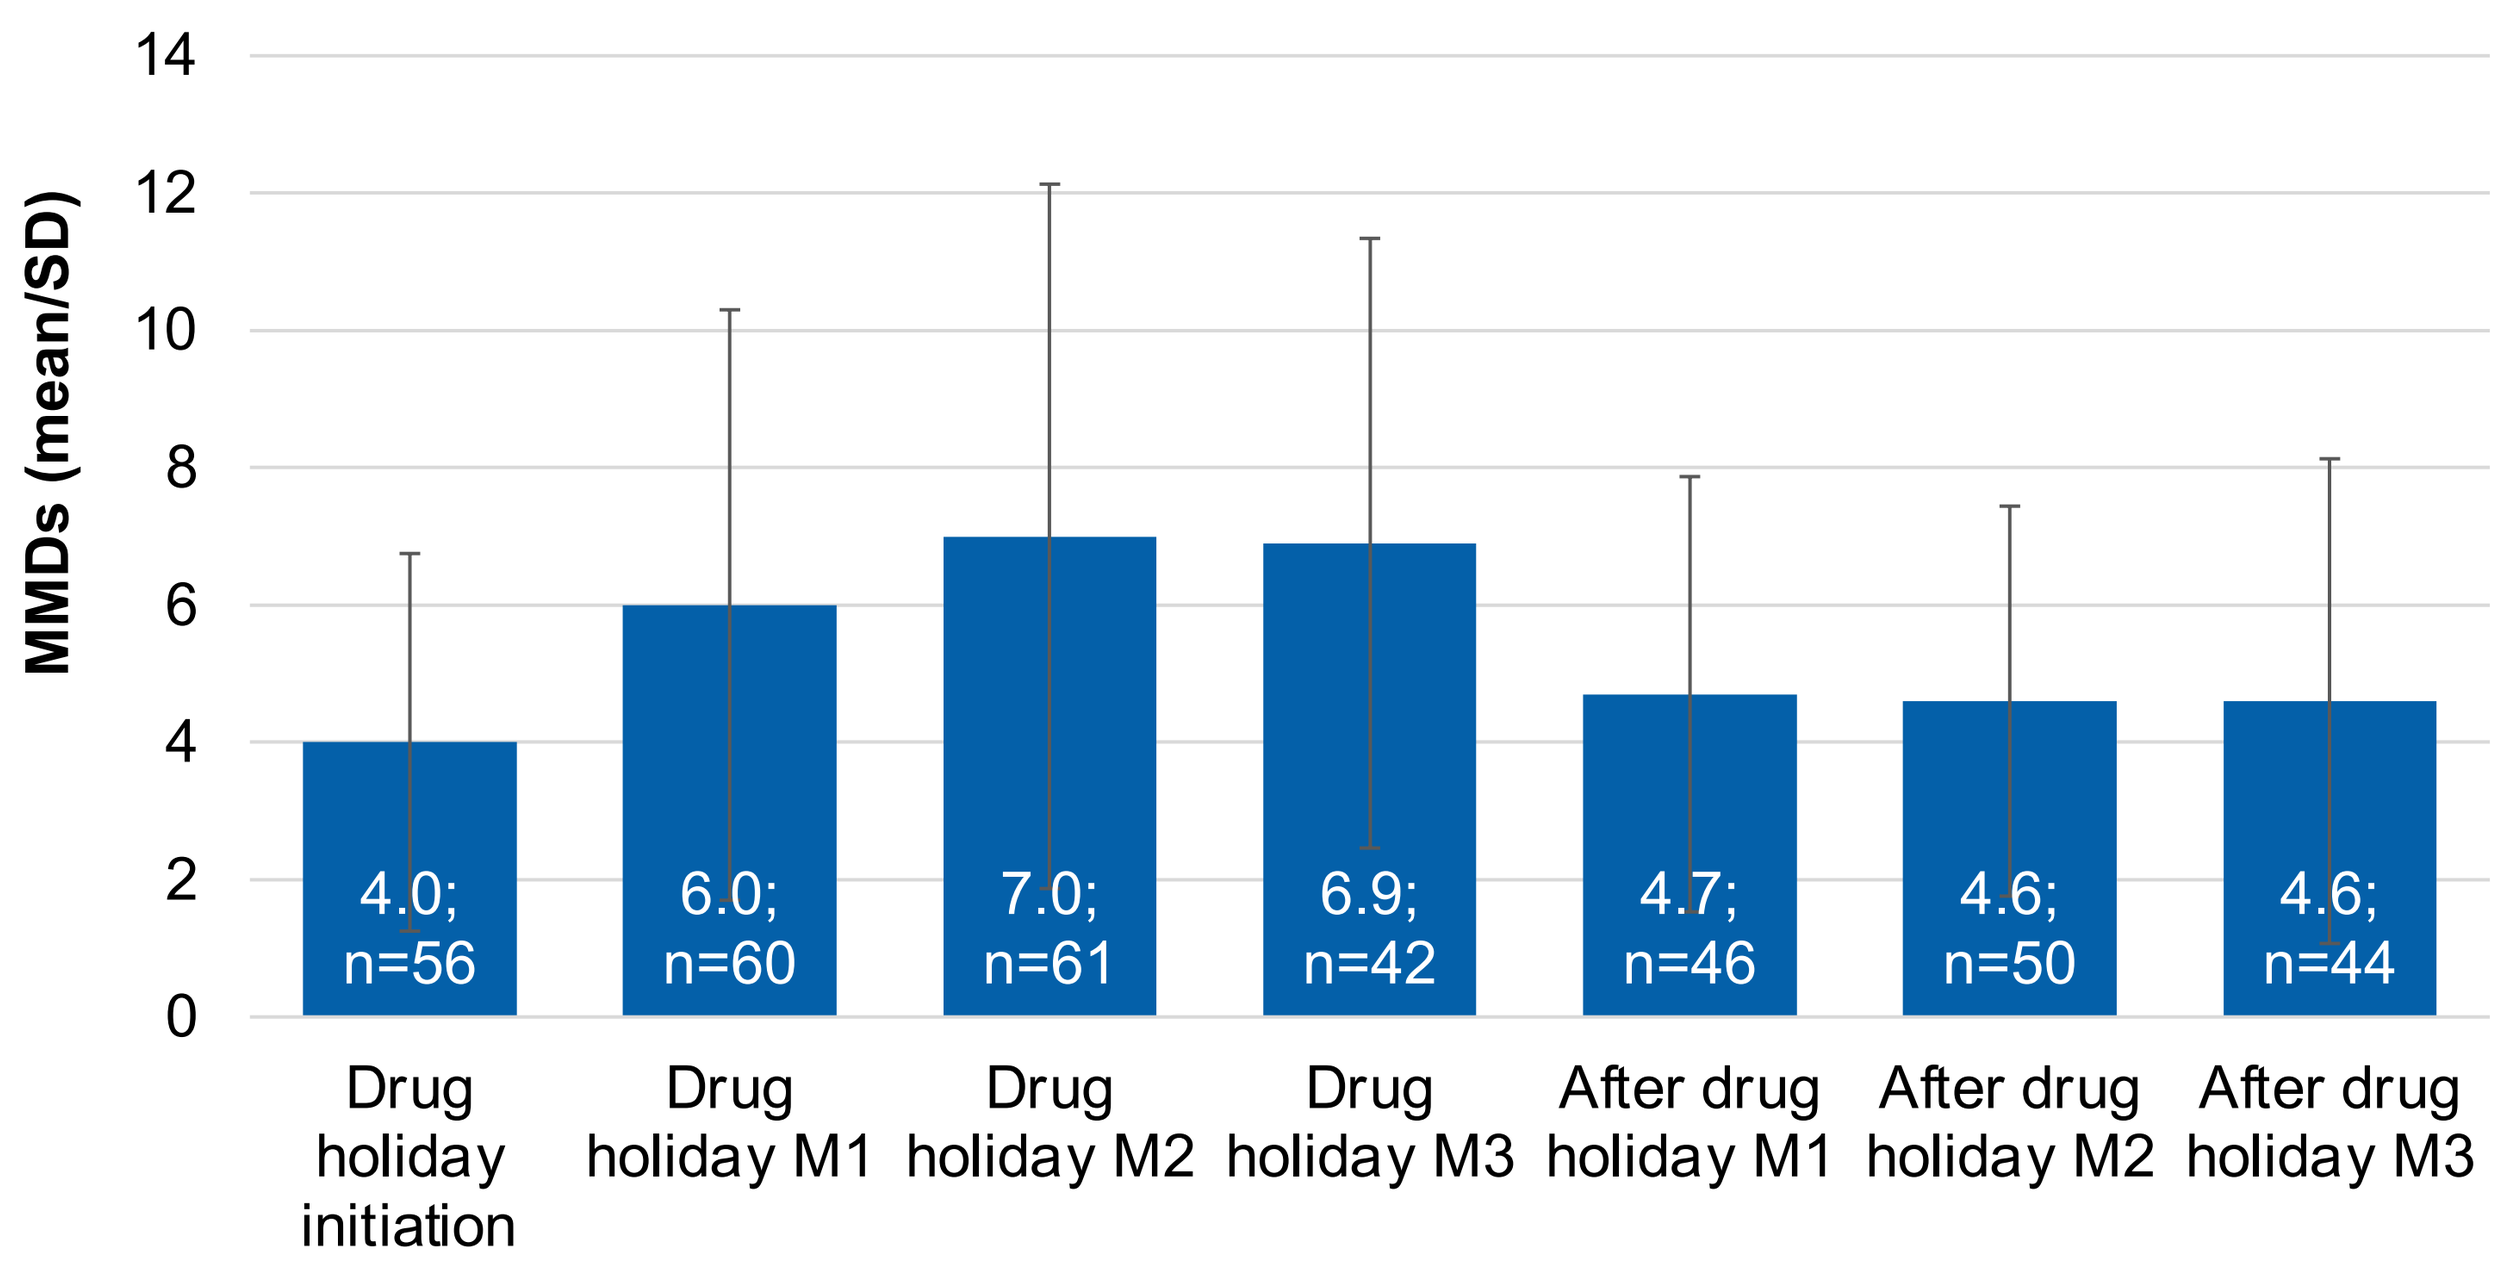
MMDs (mean/SD)
14
12
10
8
6
4
2
0
4.0; n=56
6.0; n=60
7.0; n=61
6.9; n=42
4.7; n=46
4.6; n=50
4.6; n=44
Drug holiday initiation
Drug holiday M1
Drug holiday M2
Drug holiday M3
After drug holiday M1
After drug holiday M2
After drug holiday M3

Disclosures: Hartmut Göbel received honoraria for consulting and lectures from Allergan, Almirall, Astra Zeneca, Bayer Vital, Berlin-Chemie, Bionorica, Bristol-Myers-Squibb, Elli Lilly, Fujisawa, GlaxoSmithKline, Grünenthal, Hermal, Hormosan, Ipsen-Pharma, Janssen-Cilag, Johnson&Johnson, Krewel-Meuselbach, Klosterfrau, Lichtwer, Menarini Pharma, Merz Pharmaceuticals, Minster Pharmaceuticals, MSD, Novartis Pharma, Pfizer, Pharmacia, Sandoz, Schaper und Brümmer, Schwarz-Pharma, Teva, Weber&Weber, Smith Kline Beecham. Mirja Koch is an employee of Novartis AG. Cordula Weiss is an employee of Novartis Pharma GmbH.

This study is supported by Novartis Pharma GmbH, Nuremberg, Germany. Erenumab is co-developed by Amgen and Novartis.