P-136

Patient-reported Outcomes of Migraine Treatment With Erenumab Versus Topiramate: Results From the HER-MES Study

Uwe Reuter,[1*] Monika Maier-Peuschel,[2] Christian Sieder ,[2] Christian Hentschke,[2] Marc Ehrlich[2]

[1]Charité- Universitätsmedizin Berlin, Germany; [2]Novartis Pharma GmbH, Nuremberg, Germany

INTRODUCTION

- Migraine, the second leading cause of disability, affects 1.04 billion people worldwide[1]
- Existing oral preventive treatments for migraine are associated with low adherence due to suboptimal efficacy and/or poor tolerability[2]
- Use of antibodies targeting the calcitonin gene-related peptide (CGRP) pathway is recommended in patients with migraine in whom ≥2 previous preventive treatment were unsuccessful [3,4]
- Erenumab is the first FDA- and EMA-approved monoclonal antibody targeting the CGRP receptor for migraine prevention
- The efficacy of erenumab compared to placebo has been demonstrated in patients with episodic migraine (EM) and chronic migraine (CM)[5-8]
- HER-MES (Head-to-head study of Erenumab against topiRamate – Migraine study to assess tolerability and efficacy in a patiEnt–centered Setting; NCT03828539) is the first head-to-head, 24-week, randomized, double-blind, double-dummy, Phase 4 study that compared erenumab treatment with a standard-of-care treatment (topiramate) in EM and CM[9]
- HER-MES study demonstrated the superior tolerability and efficacy of erenumab as compared with topiramate in migraine prevention[9]
 - Treatment discontinuation rate due to adverse events (AEs) was lower in erenumab treatment group compared to patients treated with topiramate
 - Significantly more patients achieved ≥50% reduction in monthly migraine days (MMDs) with erenumab compared to topiramate

OBJECTIVE

- To evaluate the effect of 70 mg and 140 mg subcutaneous erenumab compared to the highest tolerated dose of oral topiramate (50–100 mg/day) on patient-reported outcomes (PROs)

METHODS

Figure 1. HER-MES study design

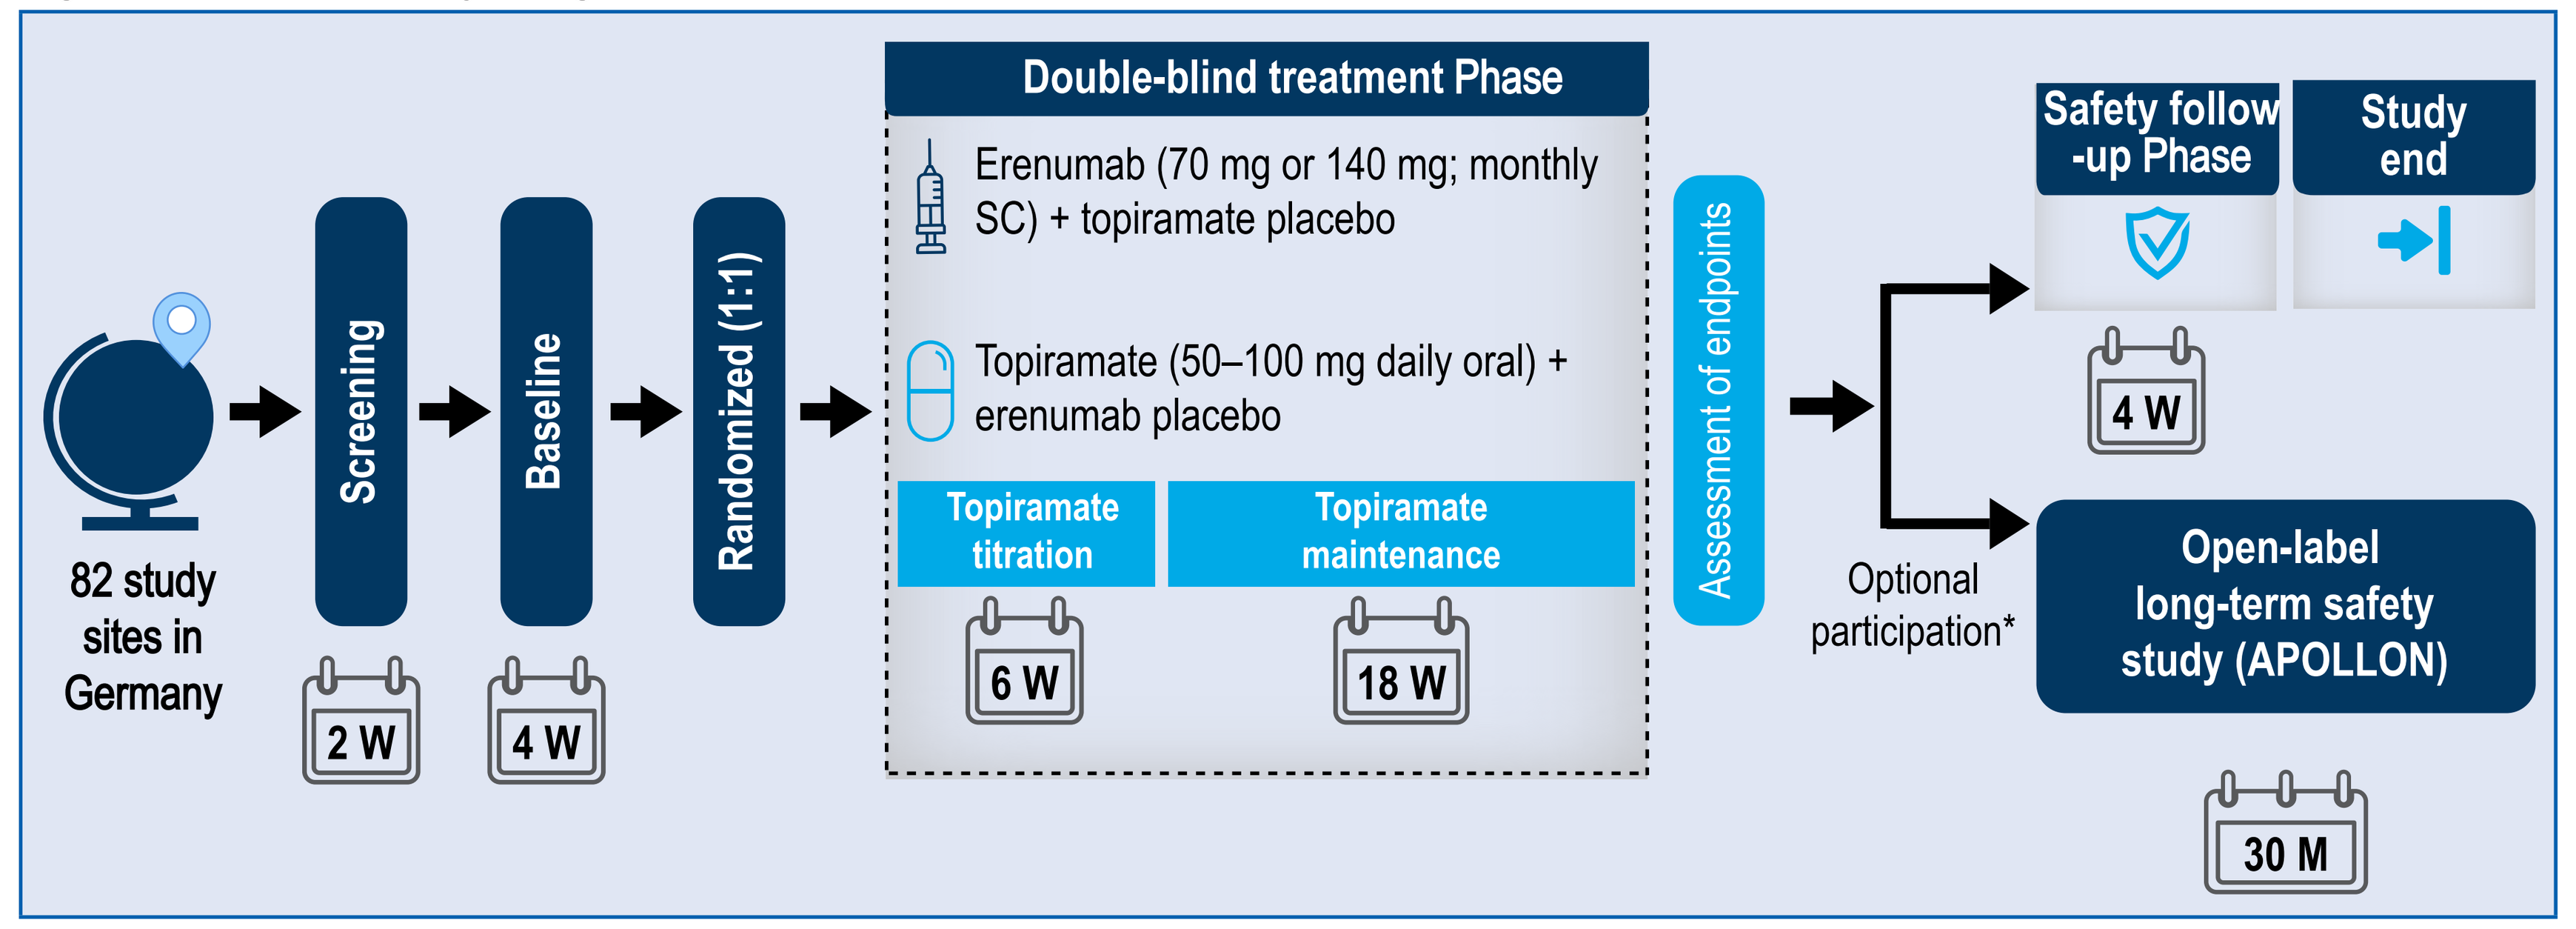
82 study sites in Germany
Screening
2 W
Baseline
4 W
Randomized (1:1)
Double-blind treatment Phase
Erenumab (70 mg or 140 mg; monthly SC) + topiramate placebo
Topiramate (50–100 mg daily oral) + erenumab placebo
Topiramate titration
6 W
Topiramate maintenance
18 W
Assessment of endpoints
Safety follow -up Phase
4 W
Study end
Optional participation*
Open-label long-term safety study (APOLLON)
30 M

*At the end of the double-blind, double-dummy treatment, participants could choose to enter the safety follow-up to end the study (HER-MES) or a long-term study (APOLLON). M, months; SC, subcutaneous; W, weeks

- Patient population included adult patients (aged 18–65 years) with EM and CM
 - All patients were treatment naïve or were not suitable for or had failed up to three previous migraine preventive treatments, and with ≥4 MMDs
- Exploratory endpoints assessed PROs which were
 - Change from baseline to Week 24 in the Headache Impact Test (HIT)-6™ total score was analyzed using a linear mixed model for repeated measures (MMRM)
 - Change from baseline to Week 24 in quality of life (QoL) measured for the physical component summaries (PCS) and mental component summaries (MCS) of the Short Form 36 Health Survey Questionnaire, version 2 (SF-36 v2) was assessed using a linear MMRM
 - The proportion of patients achieving a ≥5-point reduction from baseline in HIT-6™ and SF-36 v2 score was analyzed using a logistic regression model

RESULTS

- Patients were randomized (1:1) to subcutaneous monthly erenumab 70 mg or 140 mg (n=389) or oral topiramate 50 to 100 mg daily (n=388) treatment groups
- Baseline disease characteristics were comparable between both treatment groups (**Table 1**)
 - Majority of patients were treatment naïve (59.4%) or received only one prior preventive migraine treatment (30.7%)
- A greater proportion of patients in the erenumab group as compared to the topiramate group reported improvements in PROs
 - A significantly greater proportion of patients in the erenumab group compared with the topiramate group (72.2% vs 53.9%; p<0.001) achieved a ≥5-point reduction from baseline to Week 24 in HIT-6™ total score (**Figure 2A**)
 - Adjusted mean (SE) decreases from baseline to Week 24 in HIT-6 score were -10.9 (0.44) and −7.72 (0.44) in the erenumab and topiramate groups, respectively (**Figure 2B**)
 - A total of 47.7% patients in the erenumab group and 37.4% patients in the topiramate group had a ≥5-point increase in the physical component (PC) of SF-36 (**Figure 3A**)
 - The adjusted mean change (SE) in the PC of SF-36 was 5.48 (0.36) in the erenumab group and 3.63 (0.36) in the topiramate group (**Figure 3B**)
 - The proportion of patients with a ≥5-point increase was low in the SF-36 mental component (MC) with a total of 25.3% patients in the erenumab group and 16.8% patients in the topiramate group (**Figure 3A**)
 - The adjusted mean change (SE) in the SF-36 MC was 1.00 (0.46) in the erenumab group and −1.18 (0.46) in the topiramate group (**Figure 3B**)

Table 1. Demographics and baseline disease characteristic

Baseline characteristics	Erenumab (N=388)¶	Topiramate (N=388)	All patients (N=776)
Age (years)	40.8 (12.4)	40.7 (12.4)	40.7 (12.4)
Prior prophylactic treatment failure status*, n (%)			
Treatment naïve	232 (59.8)	229 (59.0)	461 (59.4)
Prior treatment failure	156 (40.2)	159 (41.0)	315 (40.6)
1	115 (29.6)	123 (31.7)	238 (30.7)
2	37 (9.5)	31 (8.0)	68 (8.8)
3	4 (1.0)	5 (1.3)	9 (1.2)
Monthly migraine days	10.3 (4.0)	10.5 (3.8)	10.4 (3.9)
HIT-6™ score‡	63.6 (4.2)	63.9 (4.1)	63.8 (4.1)
SF-36 v2 score§			
Physical component	45.3 (7.1)	44.8 (7.2)	45.0 (7.2)
Mental component	51.5 (8.5)	52.1 (8.1)	51.8 (8.3)

All data values are represented as mean (SD) unless specfied
*Prior prophylactic treatments were propranolol/metoprolol, amitriptyline, flunarizine.
¶One patient did not receive study medication and was excluded from the analysis.
‡The HIT-6 is a headache-specific 6-item questionnaire to measure headache impact on daily activities. A higher score indicates a greater adverse impact. n=385 in erenumab group and n=384 in topiramate group.
§The SF-36 v2 an extensive measure of general health-related quality of life (HRQoL) and not related to a specific disease. It uses 36 questions to assess several different health domains and provides a summary of the broad components of health. The physical health component and the mental health component of SF-36 v2 are derived from eight subscales. A higher score indicates more favourable health state.

Figure 2A. Proportion of patients achieving at least a 5-point change from baseline to Week 24 in HIT-6™ total score (full analysis set)

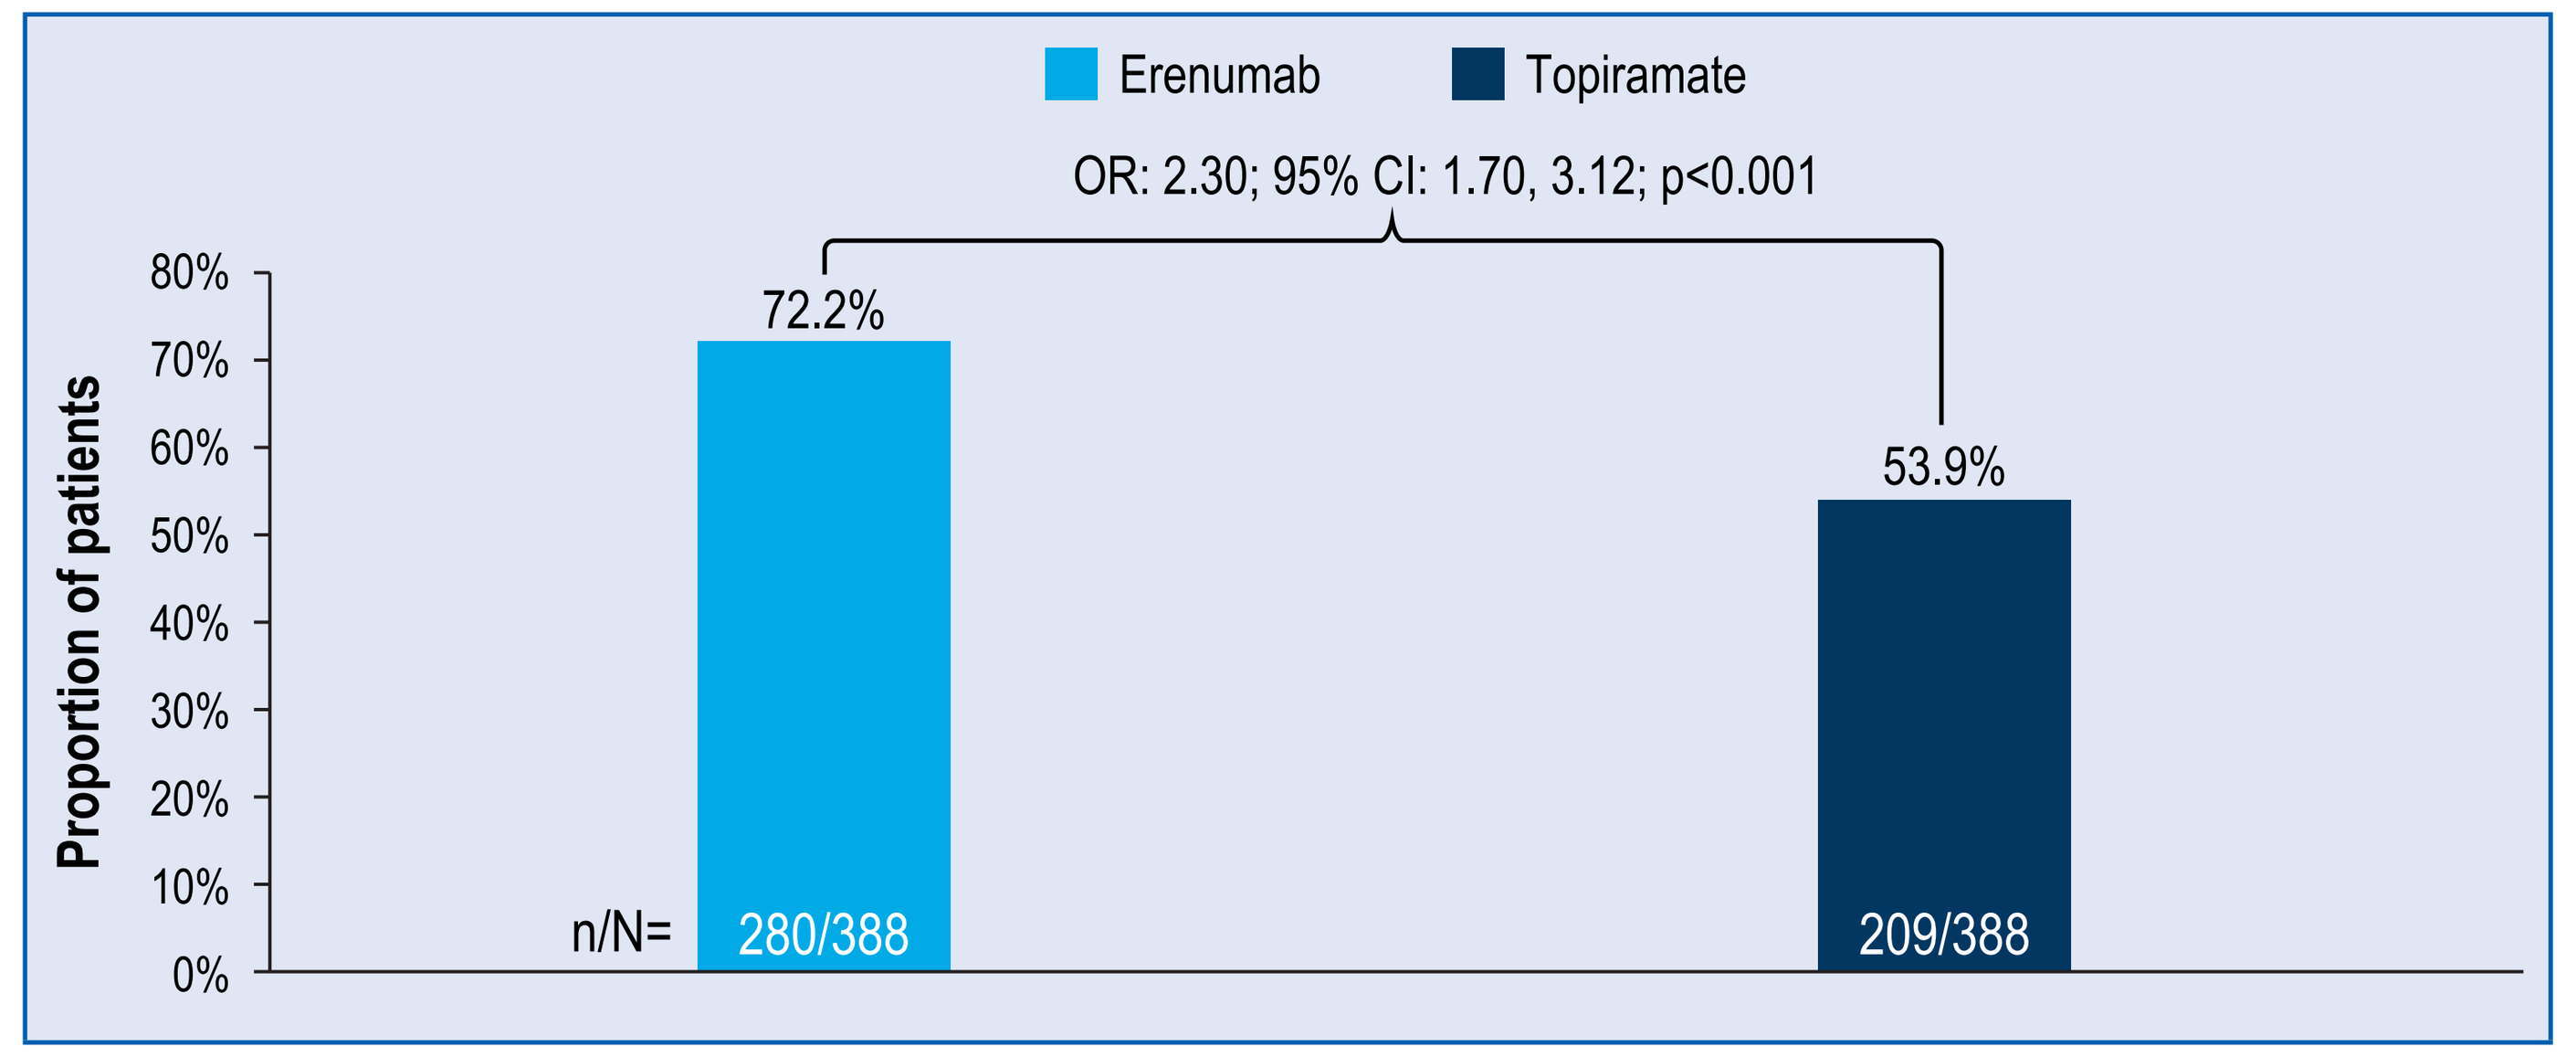
Erenumab
Topiramate
OR: 2.30; 95% CI: 1.70, 3.12; p<0.001
Proportion of patients
80%
70%
60%
50%
40%
30%
20%
10%
0%
72.2%
53.9%
n/N=
280/388
209/388

CI, confidence interval; HIT-6™, Headache Impact Test; n, The number of patients who responded; N, The total number of patients in the treatment group with response variable defined; OR, odds ratio
The OR is obtained from a logistic regression model that includes treatment group, baseline value and stratification factor (MMD at baseline).

Figure 2B. Change from baseline to week 24 in the HIT-6™ score (full analysis set)

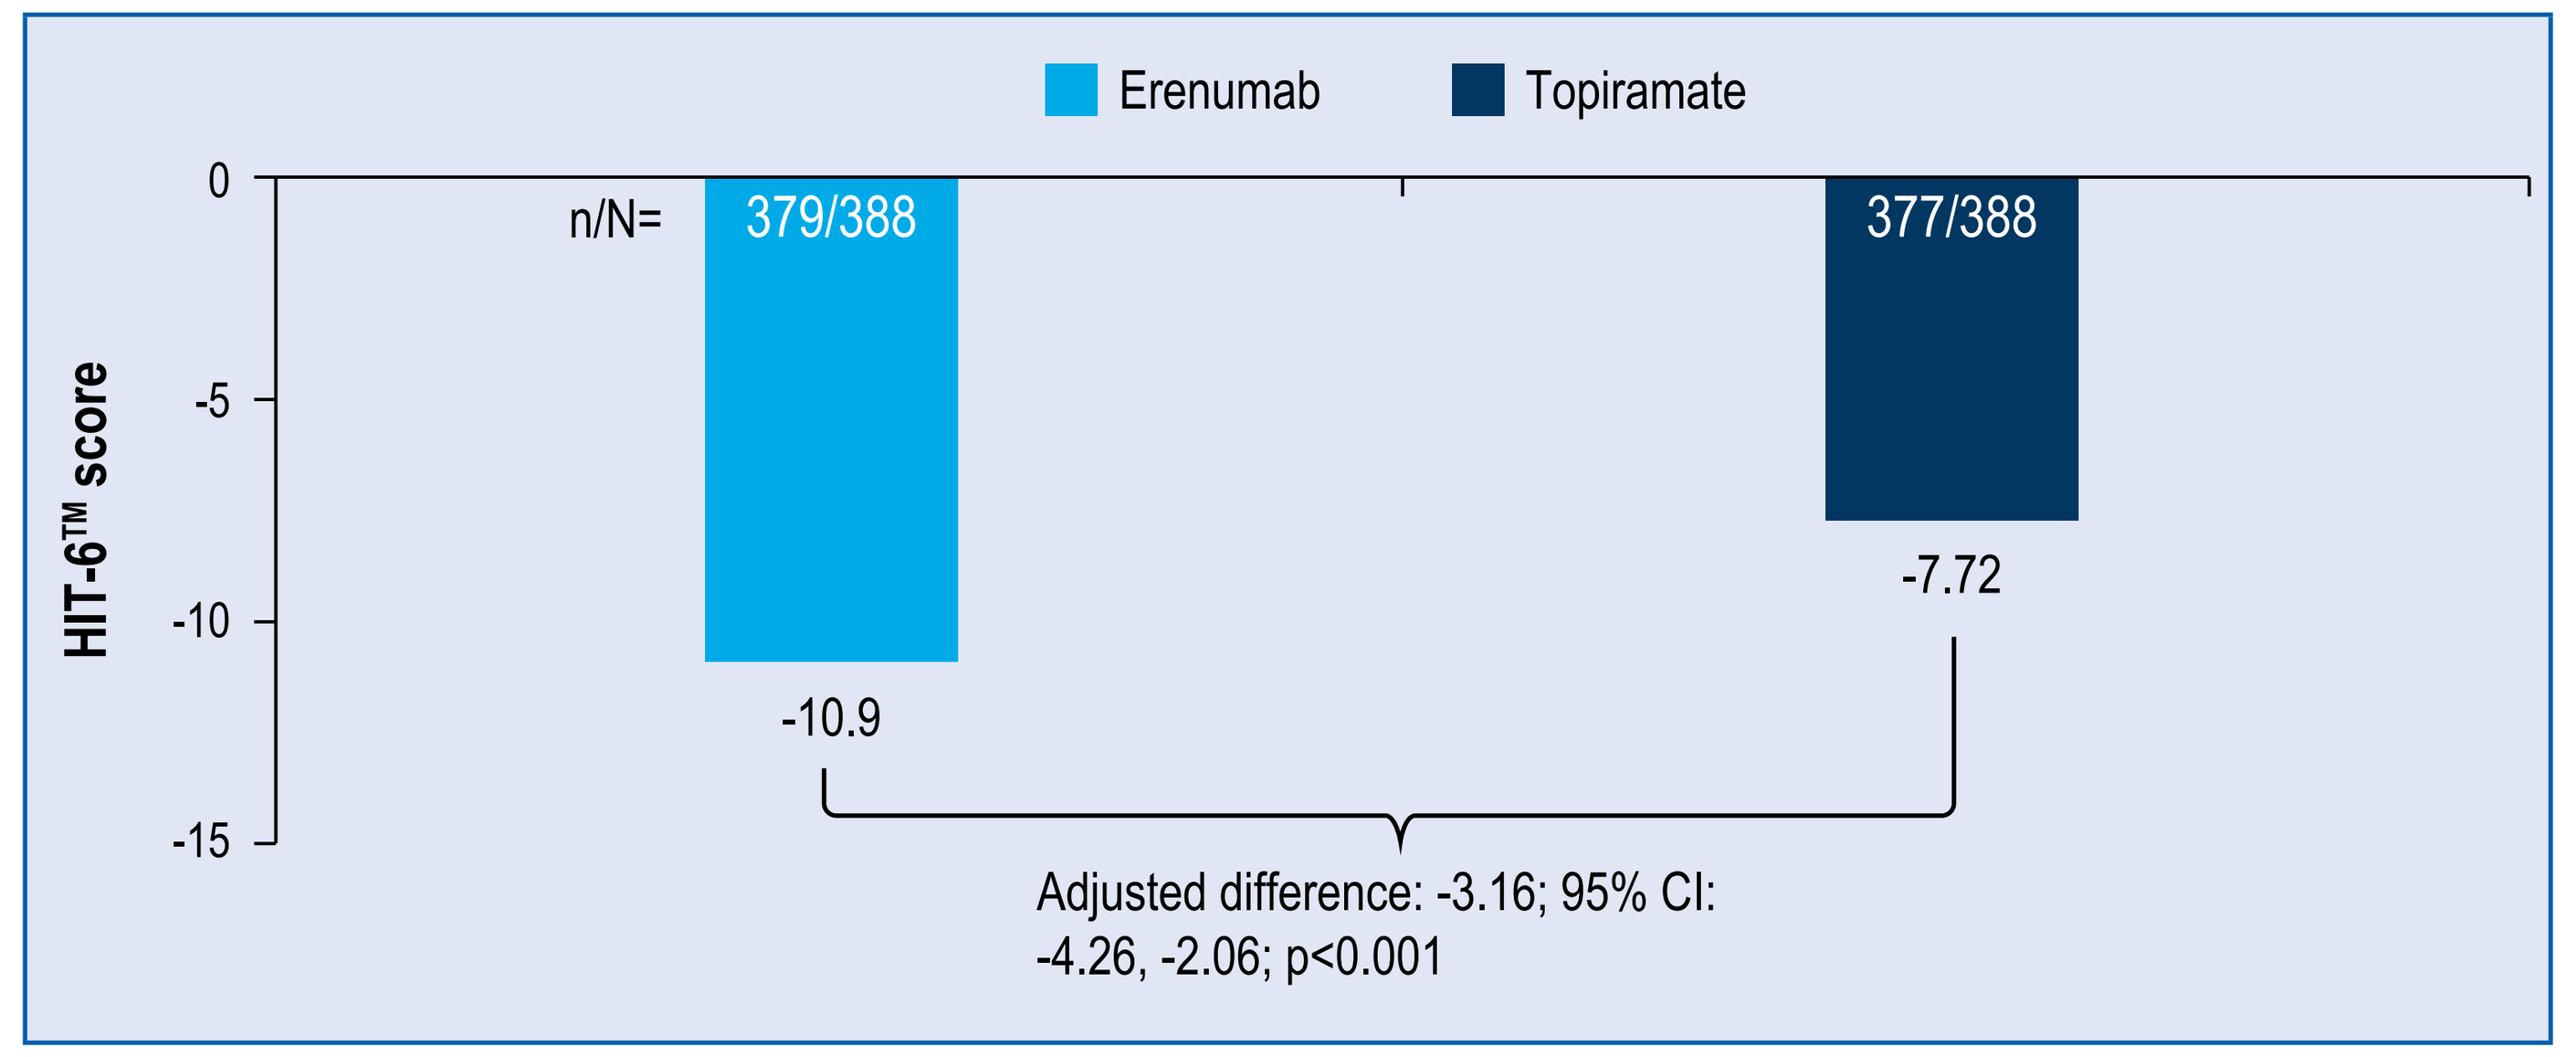
Erenumab
Topiramate
HIT-6™ score
0
-5
-10
-15
n/N=
379/388
377/388
-10.9
-7.72
Adjusted difference: -3.16; 95% CI: -4.26, -2.06; p<0.001

CI, confidence interval; HIT-6™, Headache Impact Test; MMRM, mixed model for repeated measures; n, number of patients in the analysis; N, total number of patients enrolled in the erenumab or topiramate groups
Linear MMRM includes treatment group, baseline value, stratification factor, scheduled visit and interaction between treatment group and scheduled visit.

Figure 3A. Proportion of patients achieving at least a 5-point increase from baseline to Week 24 in QoL measured for the physical and mental component summaries of the SF-36 v2 (full analysis set)

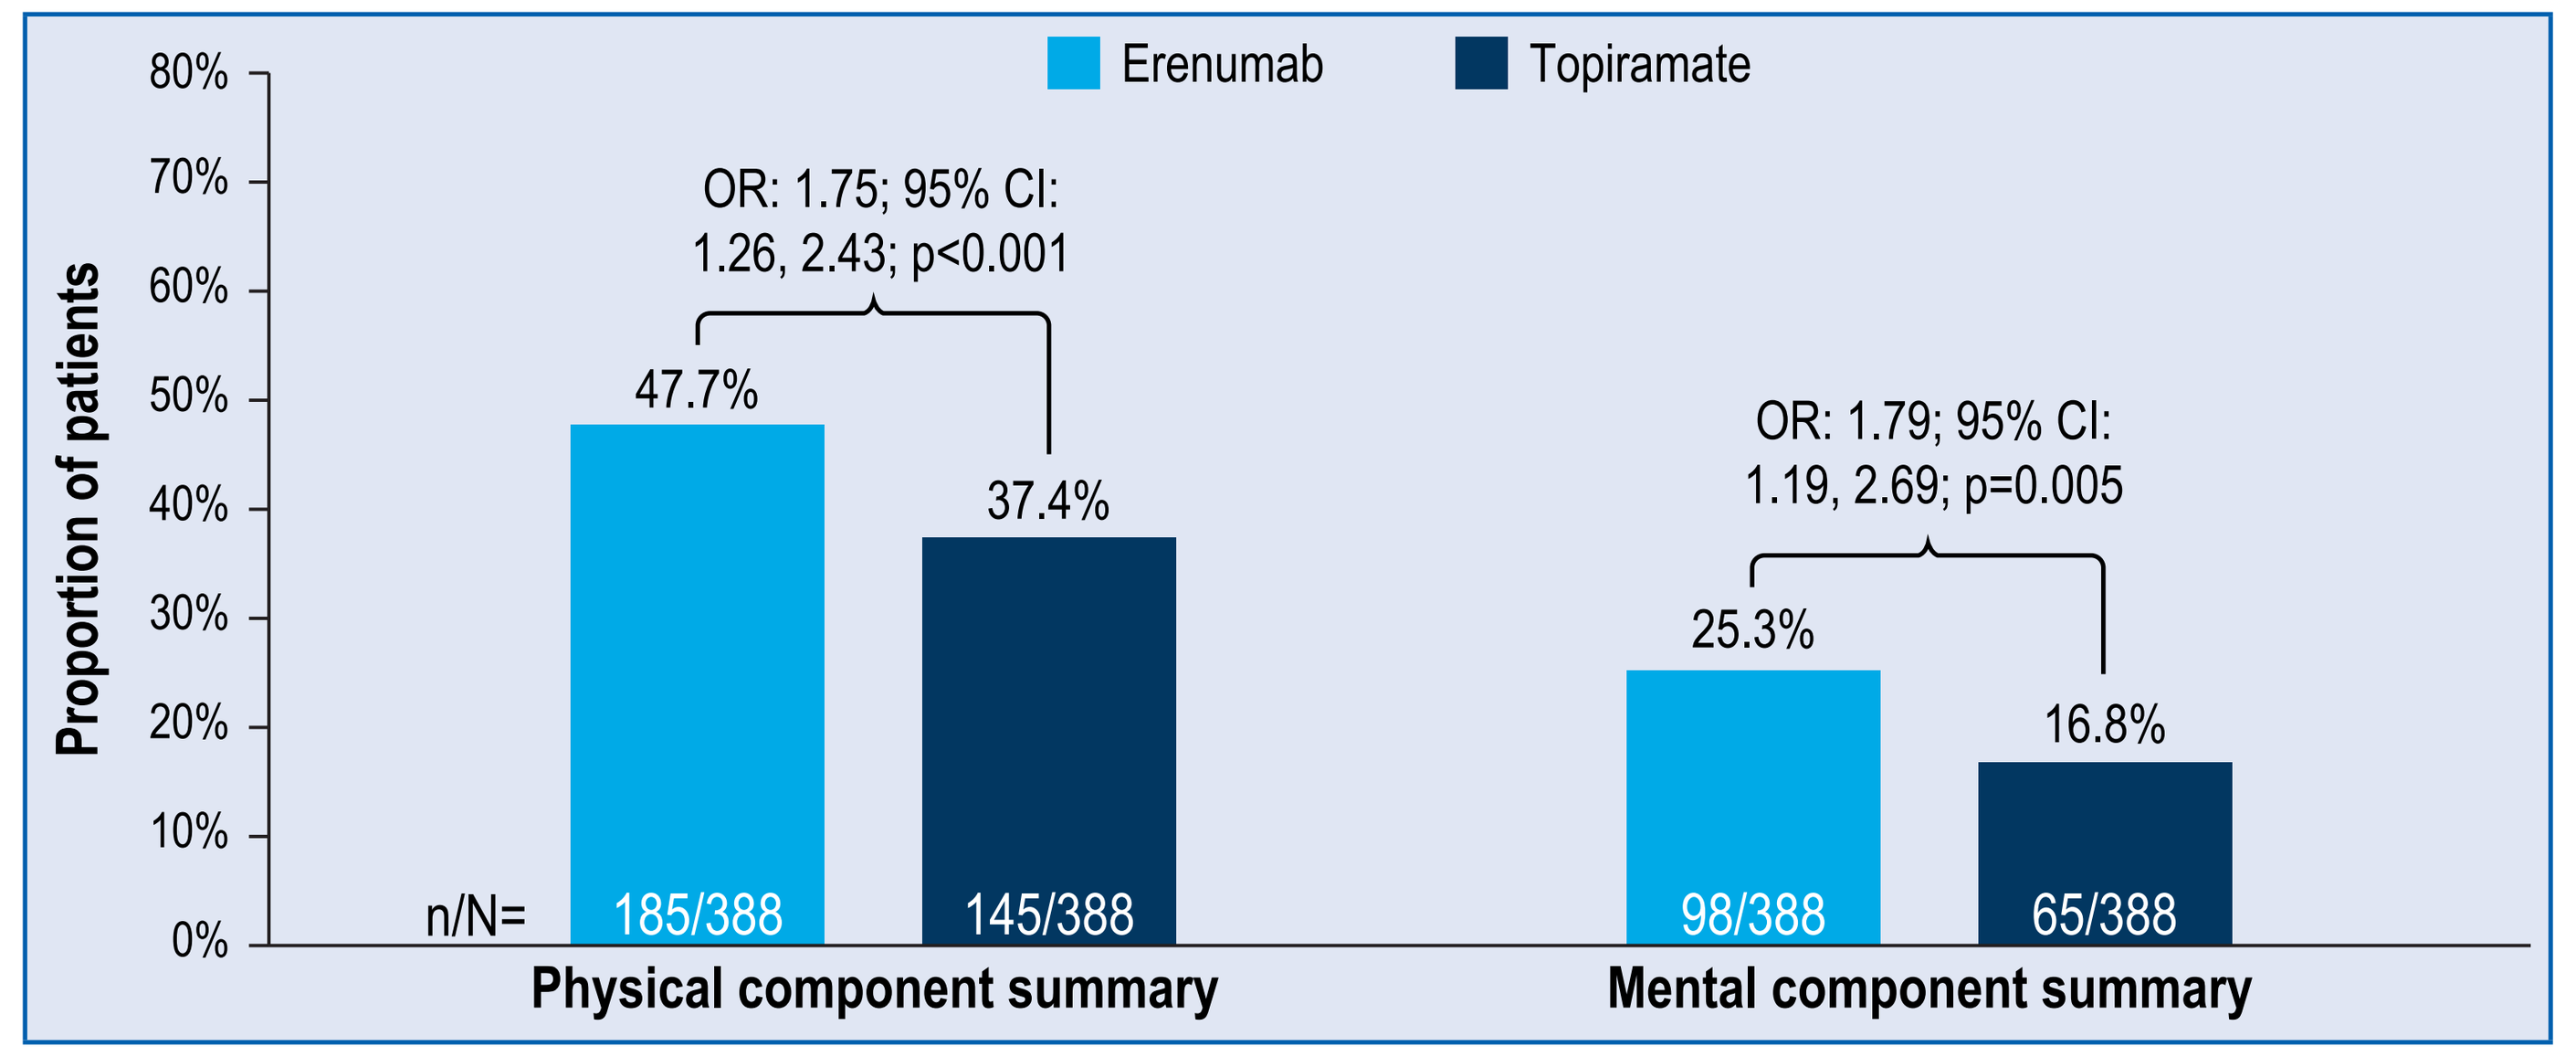
Erenumab
Topiramate
Proportion of patients
80%
70%
60%
50%
40%
30%
20%
10%
0%
OR: 1.75; 95% CI: 1.26, 2.43; p<0.001
47.7%
37.4%
OR: 1.79; 95% CI: 1.19, 2.69; p=0.005
25.3%
16.8%
n/N=
185/388
145/388
98/388
65/388
Physical component summary
Mental component summary

CI, confidence interval; MMD, monthly migraine days; n, The number of patients who responded; N, The total number of patients in the treatment group with response variable defined; OR, odds ration; QoL, quality of life; SF-36 v2, Short Form 36 Health Survey Questionnaire, version 2
The OR is obtained from a logistic regression model that includes treatment group, baseline value and stratification factor (MMD at baseline).

Figure 3B. Change from baseline to Week 24 in QoL measured for the physical and mental component summaries of the SF-36 v2 (full analysis set)

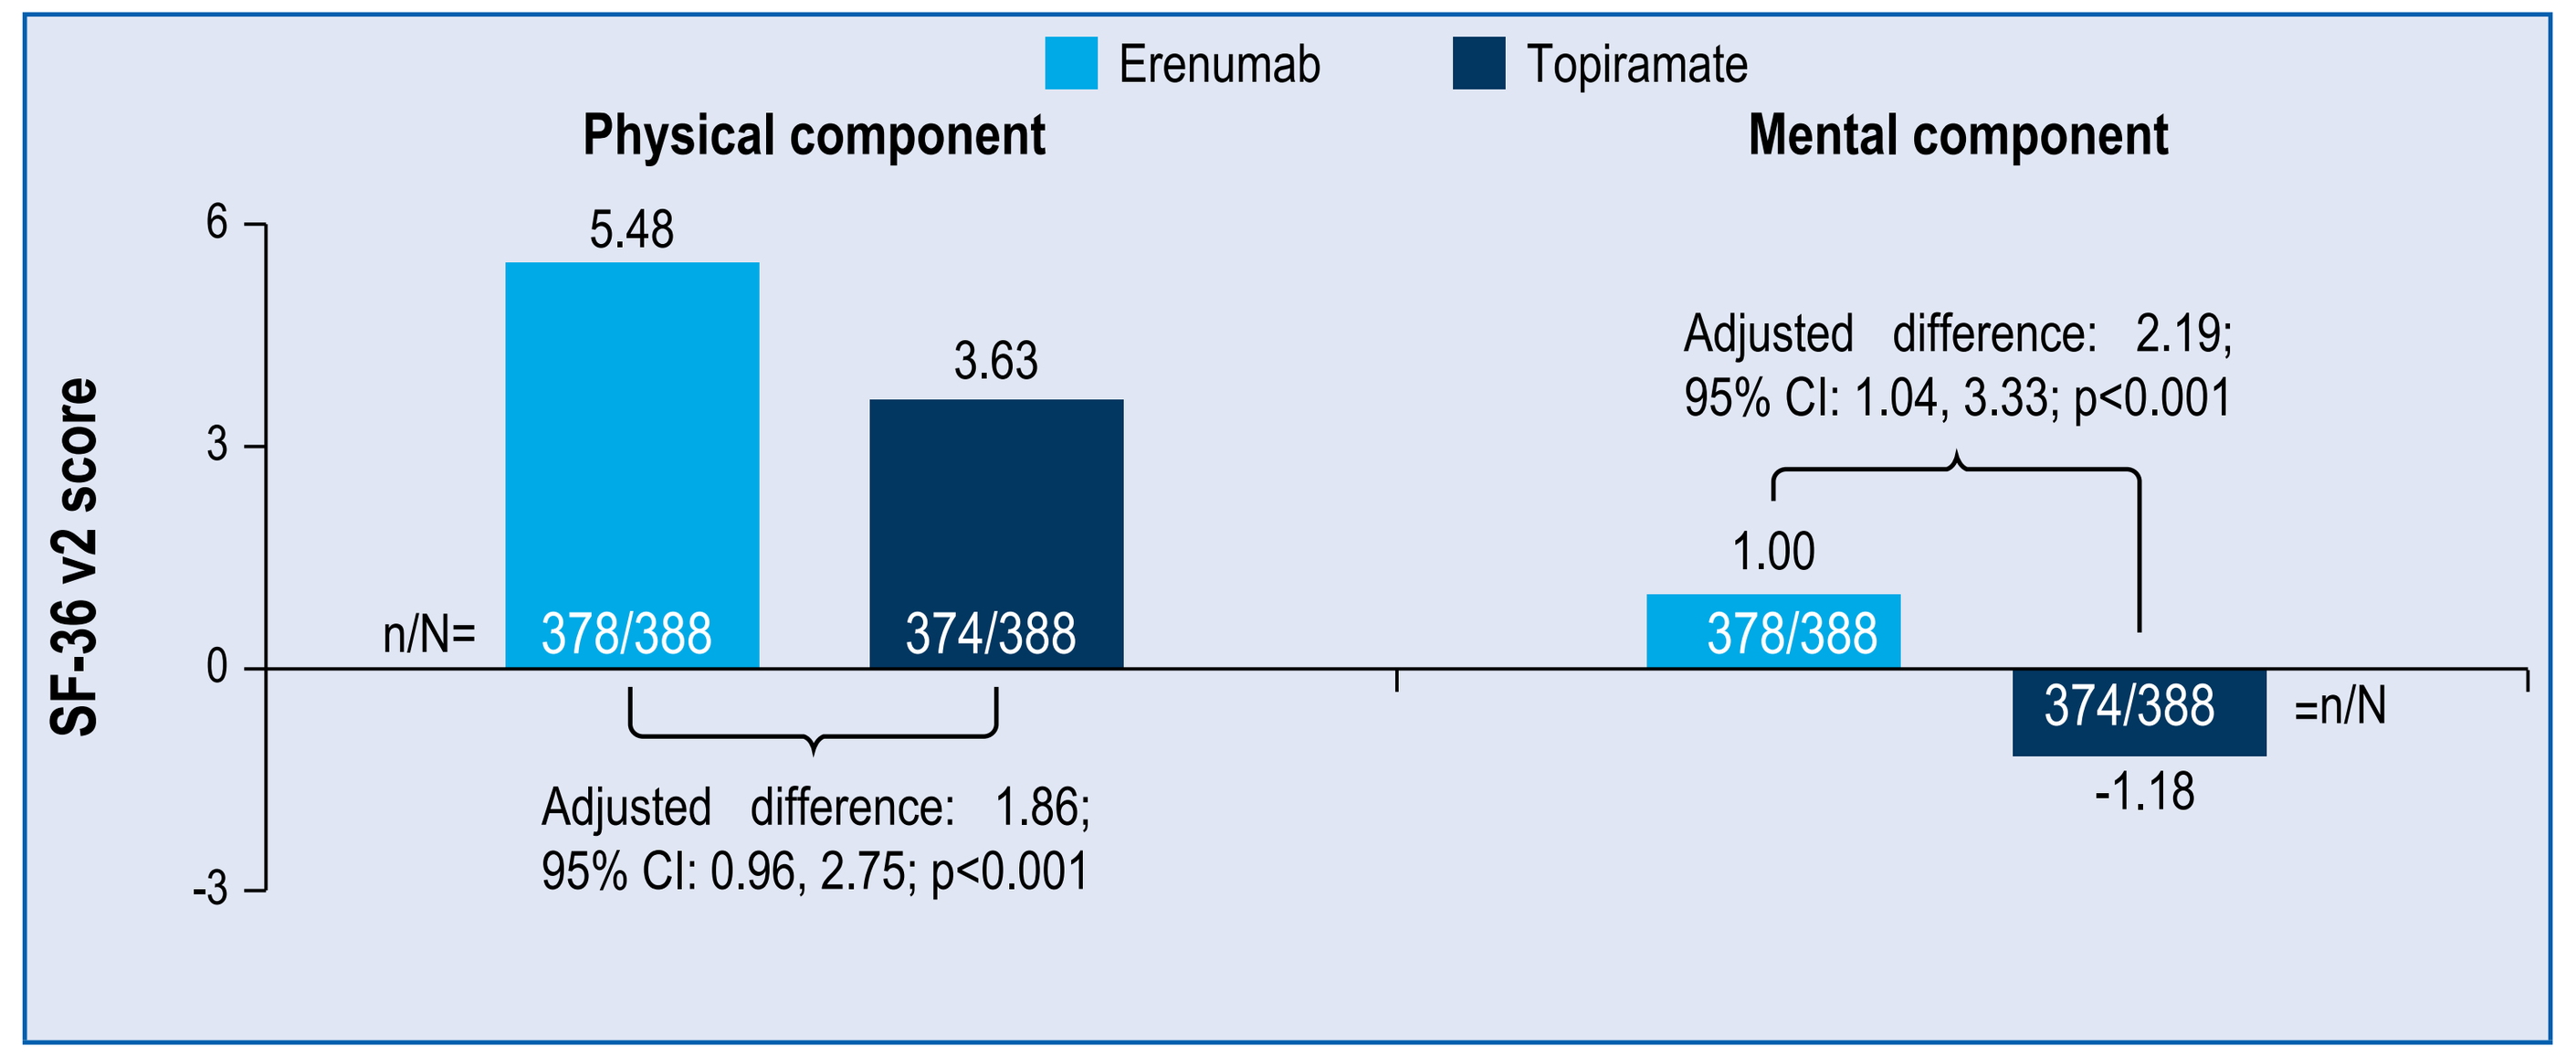
Erenumab
Topiramate
Physical component
Mental component
SF-36 v2 score
6
3
0
-3
5.48
3.63
n/N=
378/388
374/388
Adjusted difference: 1.86; 95% CI: 0.96, 2.75; p<0.001
Adjusted difference: 2.19; 95% CI: 1.04, 3.33; p<0.001
1.00
378/388
374/388
=n/N
-1.18

CI, confidence interval; MMRM, mixed model for repeated measures; n, number of patients in the analysis; N, total number of patients enrolled in the erenumab or topiramate groups; SF-36 v2, Short Form 36 Health Survey Questionnaire, version 2.
Linear MMRM includes treatment group, baseline value, stratification factor, scheduled visit and interaction between treatment group and scheduled visit. The model utilizes unstructured covariance matrix in the repeated statement.

CONCLUSIONS

- HER-MES primary data demonstrated that erenumab had a lower rate of discontinuation than topiramate due to better tolerability. The clinical benefit of erenumb is further supported by the improved QoL in migraine patients as evidenced by relevant PRO measures
- Treatment with erenumab compared with topiramate showed significant improvements in headache-specific functional impairment and general health-related QoL, demonstrating the superiority erenumab over topiramate on PROs
- Majority of patients (59.4 %) included in the study did not receive any previous preventive treatment indicating the higher burden of untreated population

REFERENCES

1. GBD 2016. *Lancet.* 2017; 390(10100): 1211–1259.
2. Blumenfeld AW, et al. *Headache.* 2013; 53(4): 644–55.
3. The American Headache Society. *Headache.* 2019; 59: 1–18.
4. Sacco S, et al. *J Headache Pain.* 2019; 20: 6.
5. Goadsby PJ, et al. *New Eng J Med.* 2017; 377: 2123–2132.
6. Reuter U, et al. *Lancet.* 2018; 392: 2280–2287.
7. Tepper S, et al. *Lancet Neurol.* 2017; 16: 425–434.
8. Ashina M, et al. *Cephalalgia.* 2018; 38(10): 1611–1621.
9. Reuter U, et al. *Cephalalgia*. 2022; 42 (2): 108–118.

ACKNOWLEDGMENTS

Medical writing support was provided by Rachna Shukla and design support was provided by Madhavi Kanithi of Novartis Healthcare Pvt. Ltd., Hyderabad, India. The final responsibility for the content lies with the authors.

FUNDING SOURCE

This study was funded by Novartis Pharma GmbH, Nuremberg, Germany and Novartis Pharma AG, Basel, Switzerland. Erenumab is co-developed by Amgen and Novartis.

DISCLOSURES

Uwe Reuter received honoraria for consulting and lectures within the past 3 years from Abbvie, Amgen, PharmAllergan, Eli Lilly, Lundbeck, Novartis Pharma, Hormosan Pharma, electroCore, Medscape, Novartis, Pfizer, StreaMedUp, Teva; he holds no stocks of pharmaceutical companies or medical device companies.

Marc Ehrlich, **Christian Sieder**, **Christian Hentschke**, and **Monika-Maier Peuschel** are employees at Novartis.

64th American Headache Society (AHS), Annual Scientific Meeting, June 9–12, 2022

© 2022 Novartis Pharma AG. All rights reserved

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the author

Presenter email address: uwe.reuter@charite.de

Scan to download a copy of this poster

NOVARTIS AMGEN

American Headache Society's 64th Annual Scientific Meeting