Poster Presented at the 9th Joint ECTRIMS-ACTRIMS Meeting, 11–13 October 2023, Milan, Italy

P1480

Remibrutinib Ameliorates CNS Autoimmune Disease - Insights From EAE

Bruno Cenni[1], Barbara Nuesslein-Hildesheim[1], Antje Marcantonio[1], Grazyna Wieczorek[1], Frederique Lafossas[1], Meike Lang[1], Pamela Ramseier[1], Sarah Tisserand[1], Giuseppe Locatelli[1], Bernd C. Kieseier[2]

[1]Novartis Institutes for Biomedical Research, Basel, Switzerland; [2]Novartis Pharma AG, Basel, Switzerland

SUMMARY

1. Remibrutinib (LOU064) is a potent, highly selective covalent Bruton's tyrosine kinase inhibitor (BTKi) with a promising profile for multiple sclerosis (MS) and other autoimmune or autoallergic indications. The efficacy and mechanism of action of remibrutinib was assessed in two experimental autoimmune encephalomyelitis (EAE) mouse models for MS

2. Remibrutinib inhibited EAE by a two-pronged mechanism based on the inhibition of pathogenic B-cell autoreactivity, as well as direct anti-inflammatory effects in microglia. Remibrutinib showed efficacy in both models in absence of B-cell depletion, broad T-cell inhibition or reduction of total immunoglobulin (Ig) levels. These findings support the view that remibrutinib may represent a novel treatment option for patients with MS

INTRODUCTION

- BTK regulates the functions of B and myeloid cells, implicated in the pathogenesis of MS[1]. Remibrutinib is a covalent, oral BTKi exhibiting high selectivity and potency, with the potential to minimise off-target toxicity and is currently being investigated in phase 3 trials for the treatment of MS (NCT05147220, NCT05156281)[2]

OBJECTIVE

- To assess the in vivo efficacy of remibrutinib and to better understand its impact on inflammation and tissue destruction in the central nervous system (CNS) in an MS animal model

METHODS

- EAE was induced by immunisation with either human myelin oligodendrocyte glycoprotein (HuMOG), causing a B cell–dependent disease, or rat MOG, causing B cell–independent pathology
- Target engagement was assessed in tissue, and clinical disease activity was determined
- Inflammation, demyelination and the degree of reactive gliosis were studied using immunohistochemistry

RESULTS

REMIBRUTINIB INHIBITS B CELL–DEPENDENT HUMOG EAE

- Remibrutinib dose dependently reduced HuMOG EAE symptoms. Peak spleen BTK occupancy was near-maximal for the 30 mg/kg group and lower for the 3 mg/kg group after the last dose. Interestingly, brain BTK occupancy showed dose-dependent differences with 3 mg/kg reaching only minimal BTK occupancy and 30 mg/kg reaching relatively sustained BTK occupancy[3] (**Figure 1**)

Figure 1. Efficacy and BTK Occupancy in HuMOG EAE

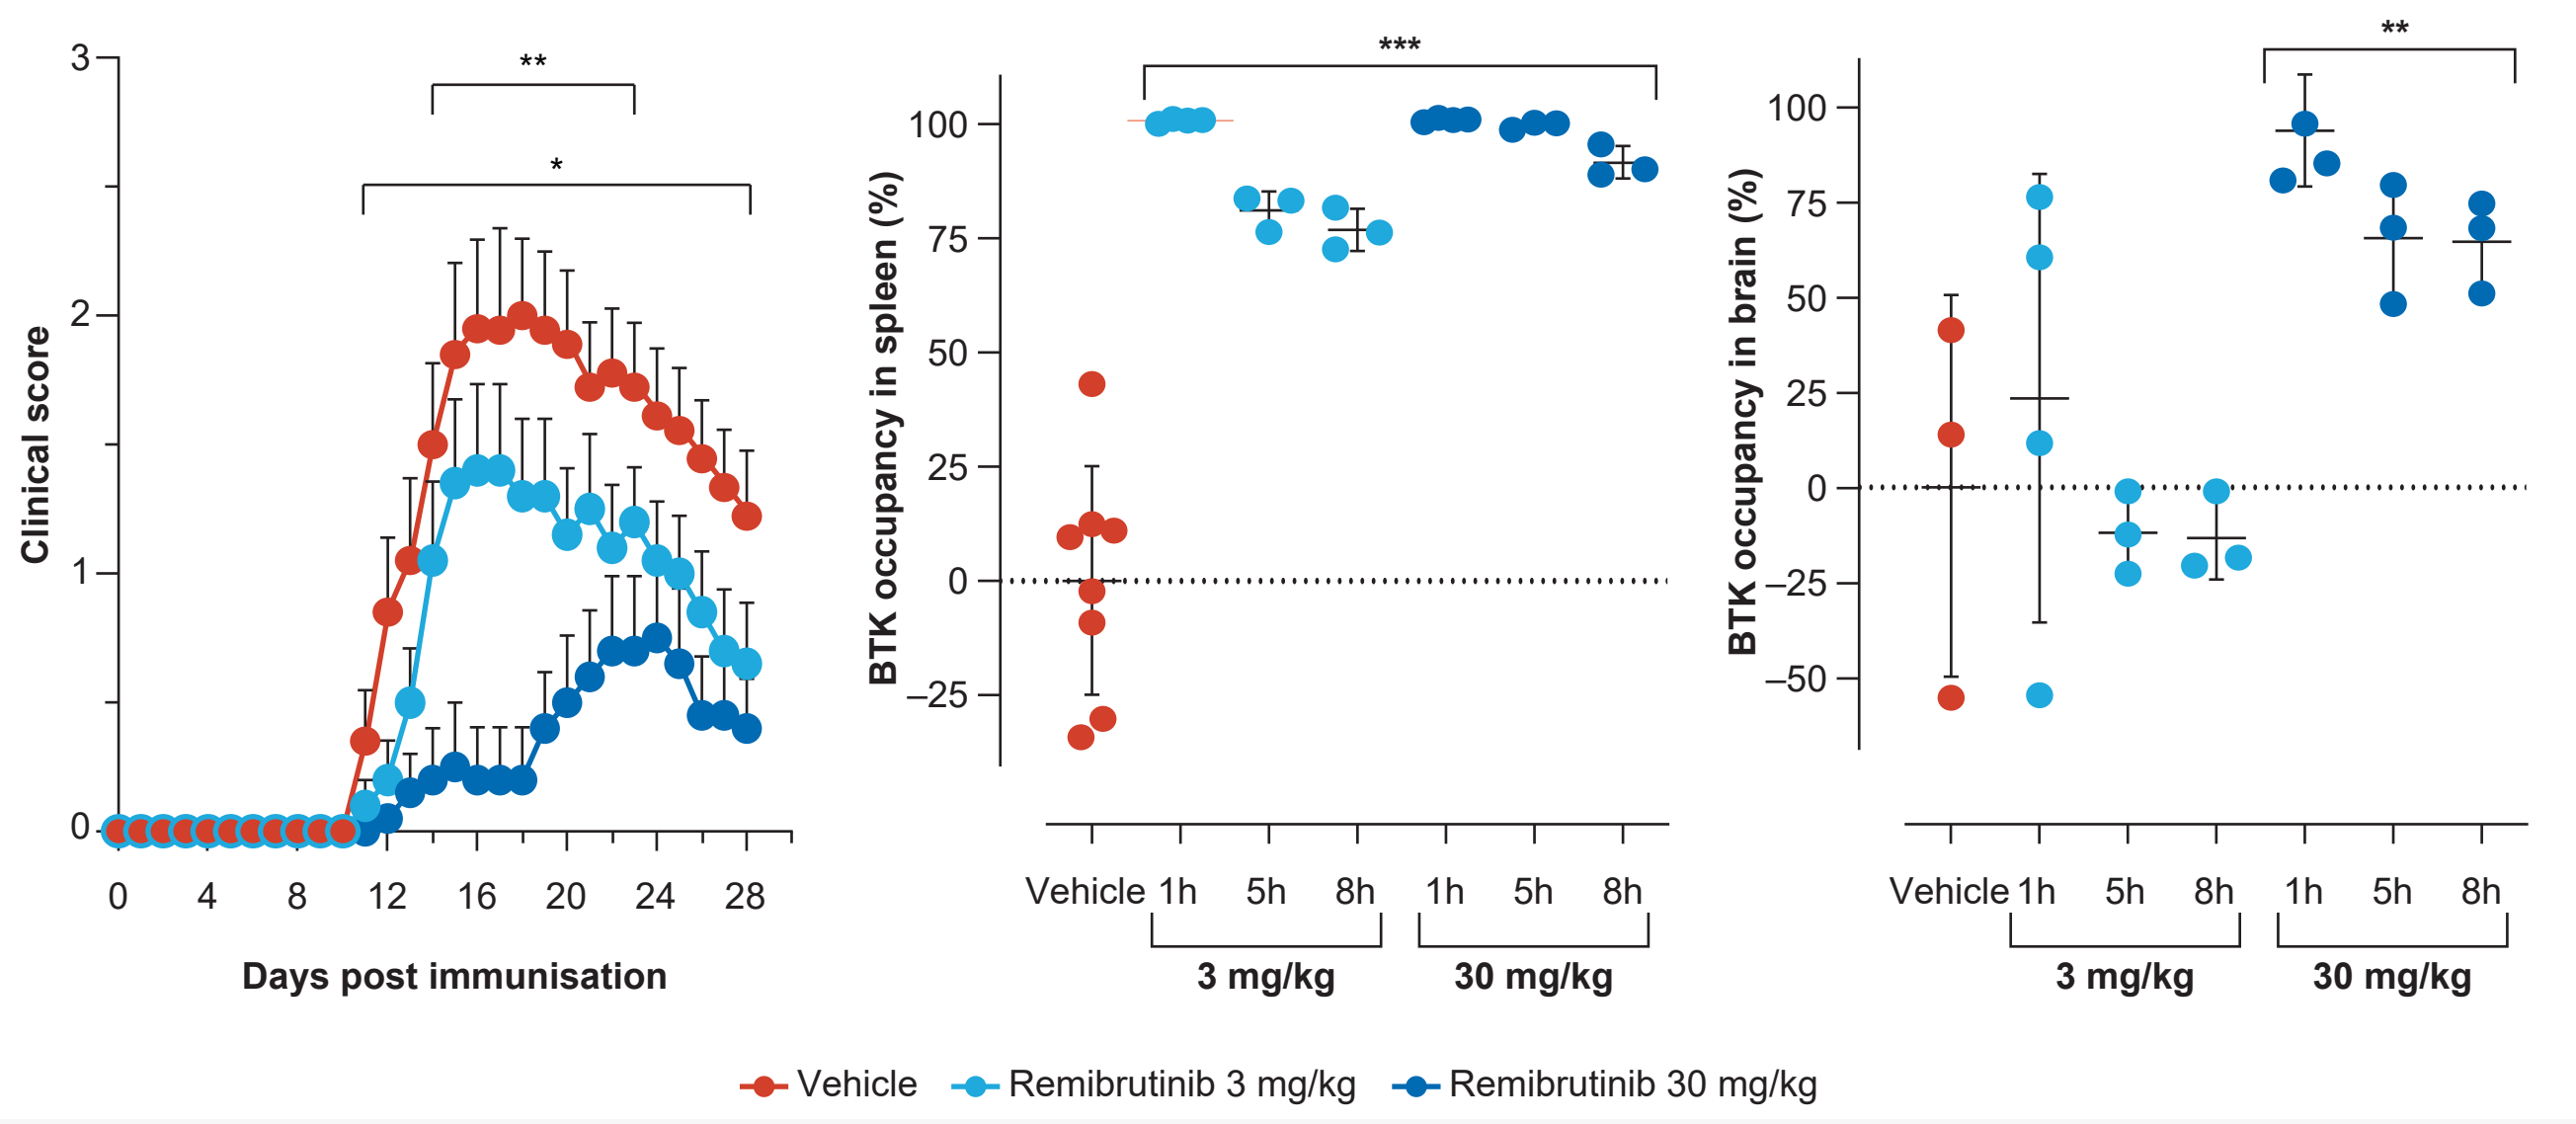

Clinical score
Days post immunisation
BTK occupancy in spleen (%)
BTK occupancy in brain (%)
Vehicle 1h 5h 8h 1h 5h 8h
3 mg/kg
30 mg/kg
Vehicle
Remibrutinib 3 mg/kg
Remibrutinib 30 mg/kg

ANOVA, analysis of variance; BTK, Bruton's tyrosine kinase; EAE, experimental autoimmune encephalomyelitis; h, hour; HuMOG, human myelin oligodendrocyte glycoprotein.
Statistical significance (ANOVA with Dunnett's test) is shown as *p<0.05, **p<0.01, ***p<0.001.

- Remibrutinib inhibited ex vivo HuMOG-specific splenocyte T-cell recall proliferative response, but not polyclonal T-cell proliferation to anti-CD3/CD28, indicating the absence of direct T-cell immune suppression[3] (**Figure 2**)

Figure 2. Ex Vivo Splenocyte HuMOG EAE Recall Proliferative Response

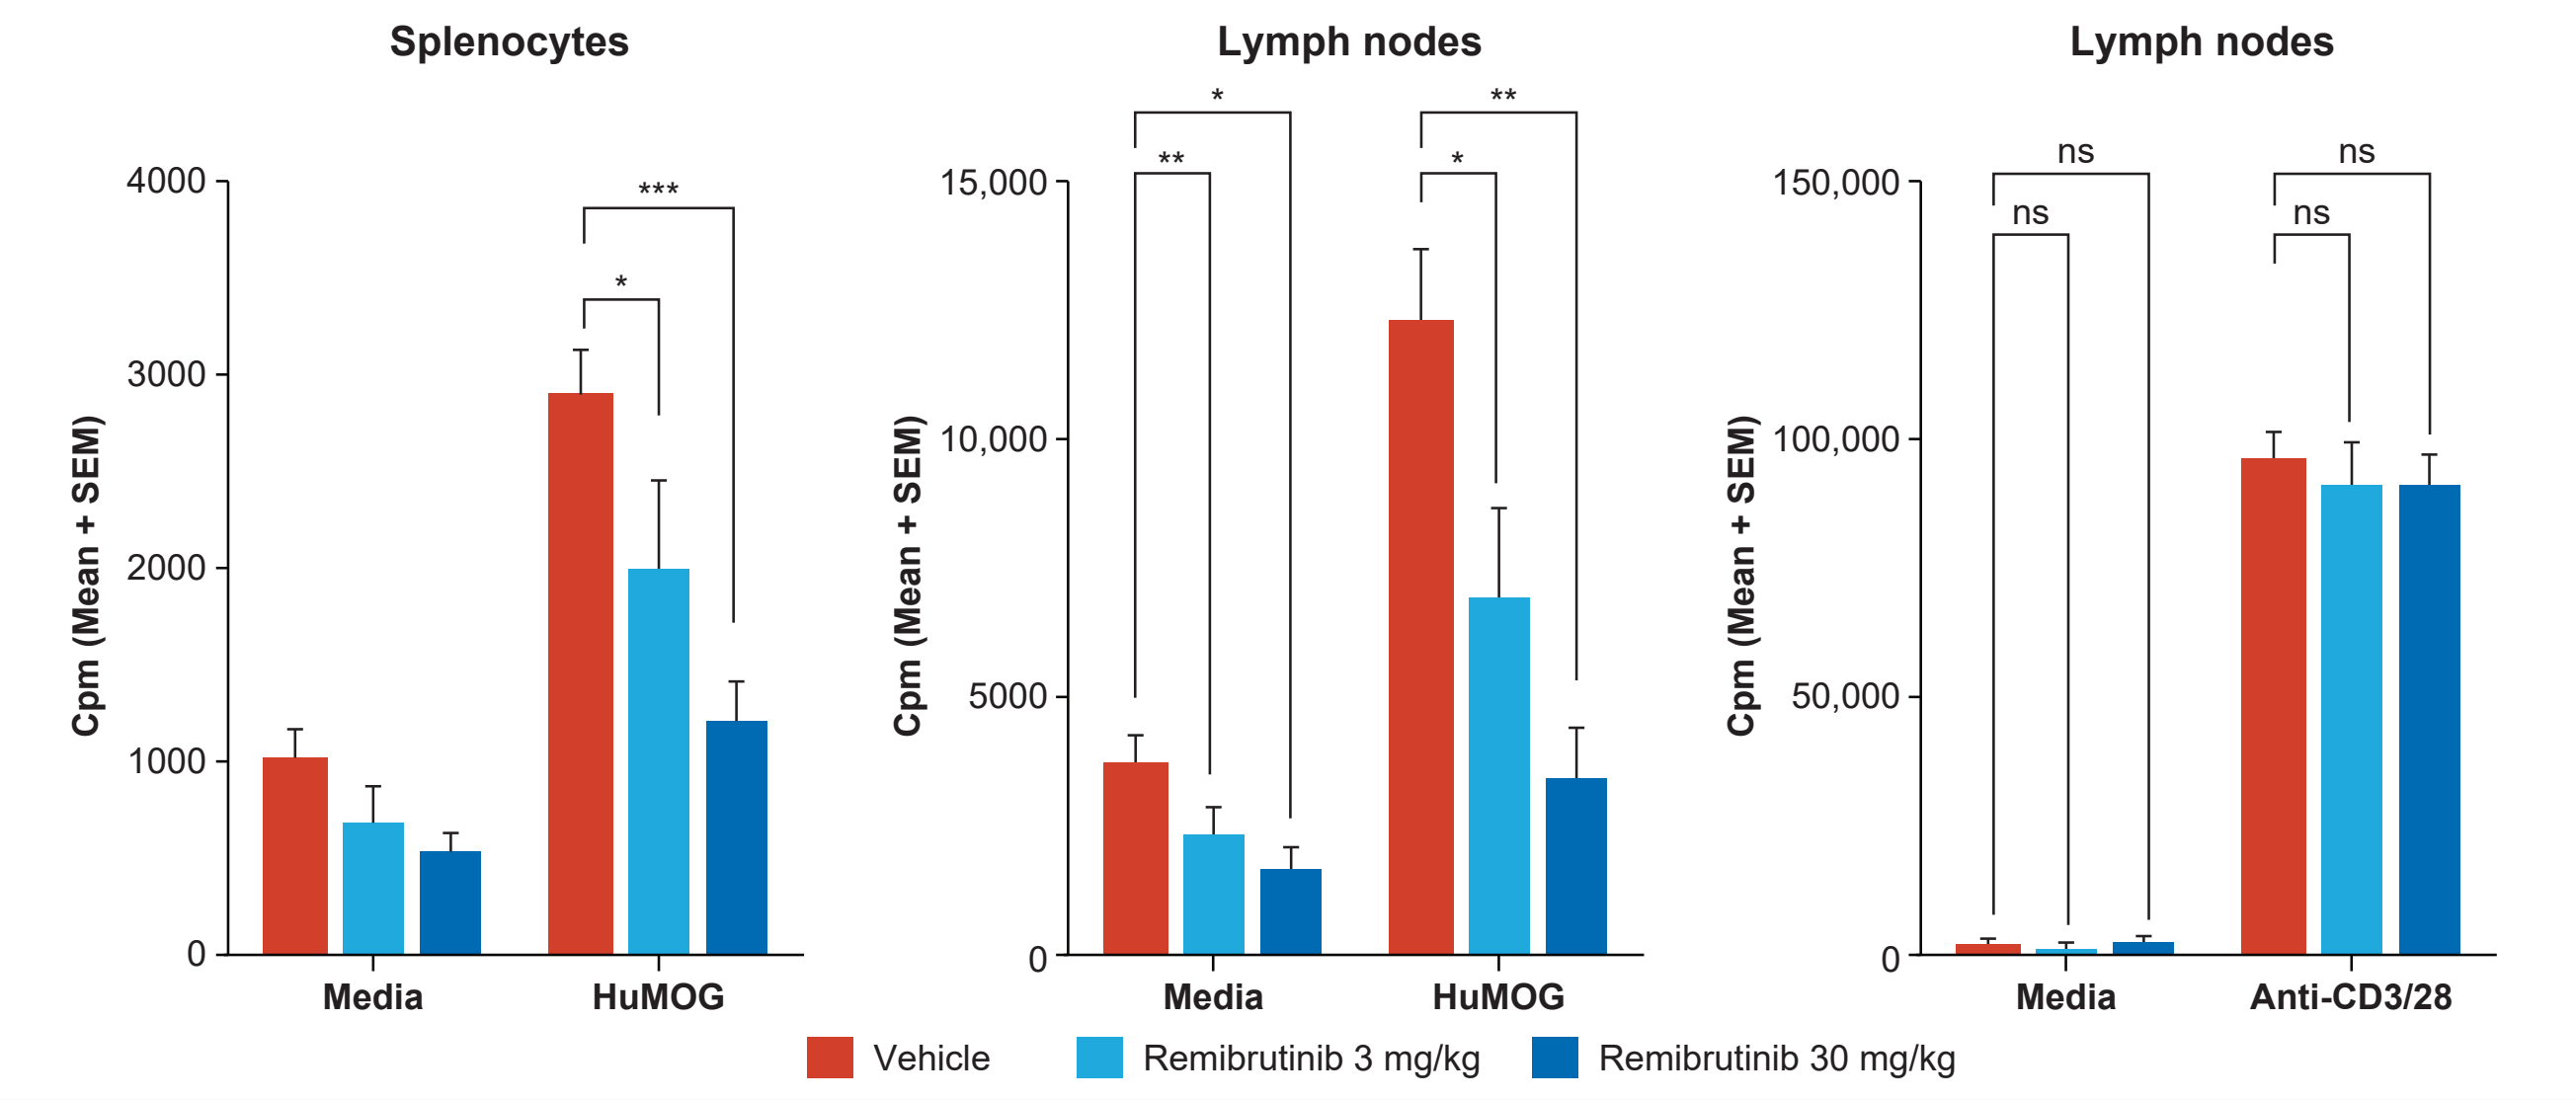

Splenocytes
Lymph nodes
Lymph nodes
Cpm (Mean + SEM)
Media
HuMOG
Anti-CD3/28
ns
Vehicle
Remibrutinib 3 mg/kg
Remibrutinib 30 mg/kg

ANOVA, analysis of variance; CD, cluster of differentiation; Cpm, counts per minute; EAE, experimental autoimmune encephalomyelitis; HuMOG, human myelin oligodendrocyte glycoprotein; ns, not significant; SEM, standard error of the mean.
Statistical significance (ANOVA with Dunnett's test) is shown as *p<0.05, **p<0.01, ***p<0.001.

- Ex vivo analysis of splenocytes, lymph node cells, and blood revealed no changes in B-cell populations or IgG antibody levels, but a clear reduction of CD4+ T-helper 17 (Th17) cells (data not shown)

Figure 3. Histology and Immunohistochemistry of Spinal Cord From HuMOG EAE Mice

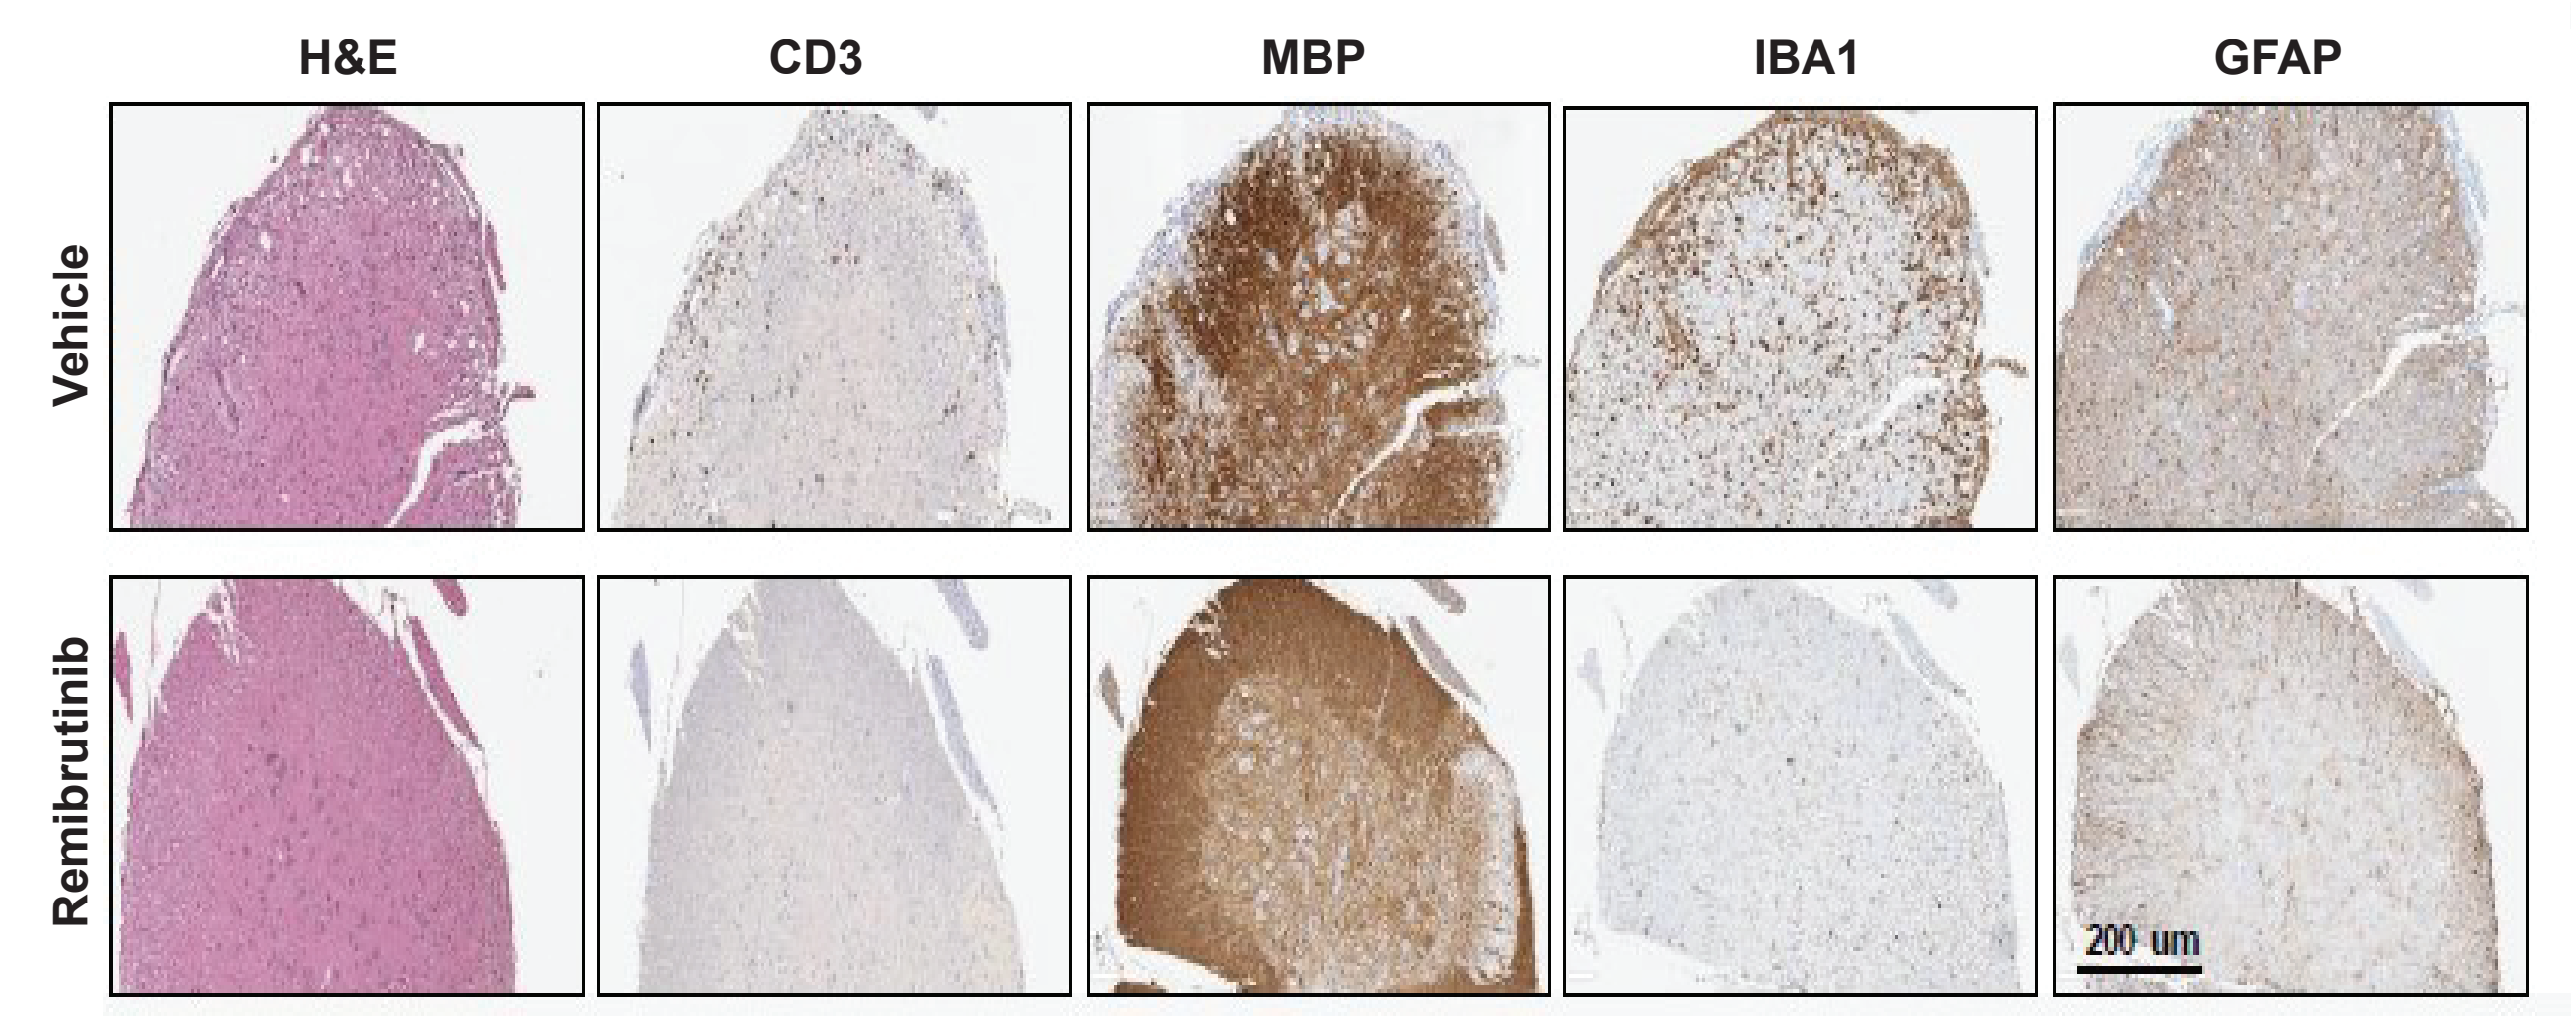

H&E
CD3
MBP
IBA1
GFAP
Vehicle
Remibrutinib
200 um

Spinal cords from HuMOG-induced EAE mice were collected on Day 19 post-immunisation at the peak of the disease. H&E sections were scored on the scale 0-4. Immunohistochemically stained sections were analysed using image analysis platforms HALO® and ASTORIA.

CD3, cluster of differentiation 3 – marker of T cells; EAE, experimental autoimmune encephalomyelitis; GFAP, glial fibrillary acidic protein – marker of astrocytes; H&E, hematoxylin and eosin; HuMOG, human myelin oligodendrocyte glycoprotein; IBA1, ionised calcium-binding adaptor molecule 1 – marker of microglia; MBP, myelin basic protein.

- Histological analysis of the spinal cord at Day 19 post immunisation (**Figure 3–4**) revealed protective effect of remibrutinib at 30 mg/kg b.i.d. by reducing the density and activation status of microglia and astrocytes. Infiltrating T cells were slightly reduced, while demyelination was not changed upon treatment (data not shown)

Figure 4. Semiquantitative (H&E) and Quantitative Results of the Immunohistochemical Stainings

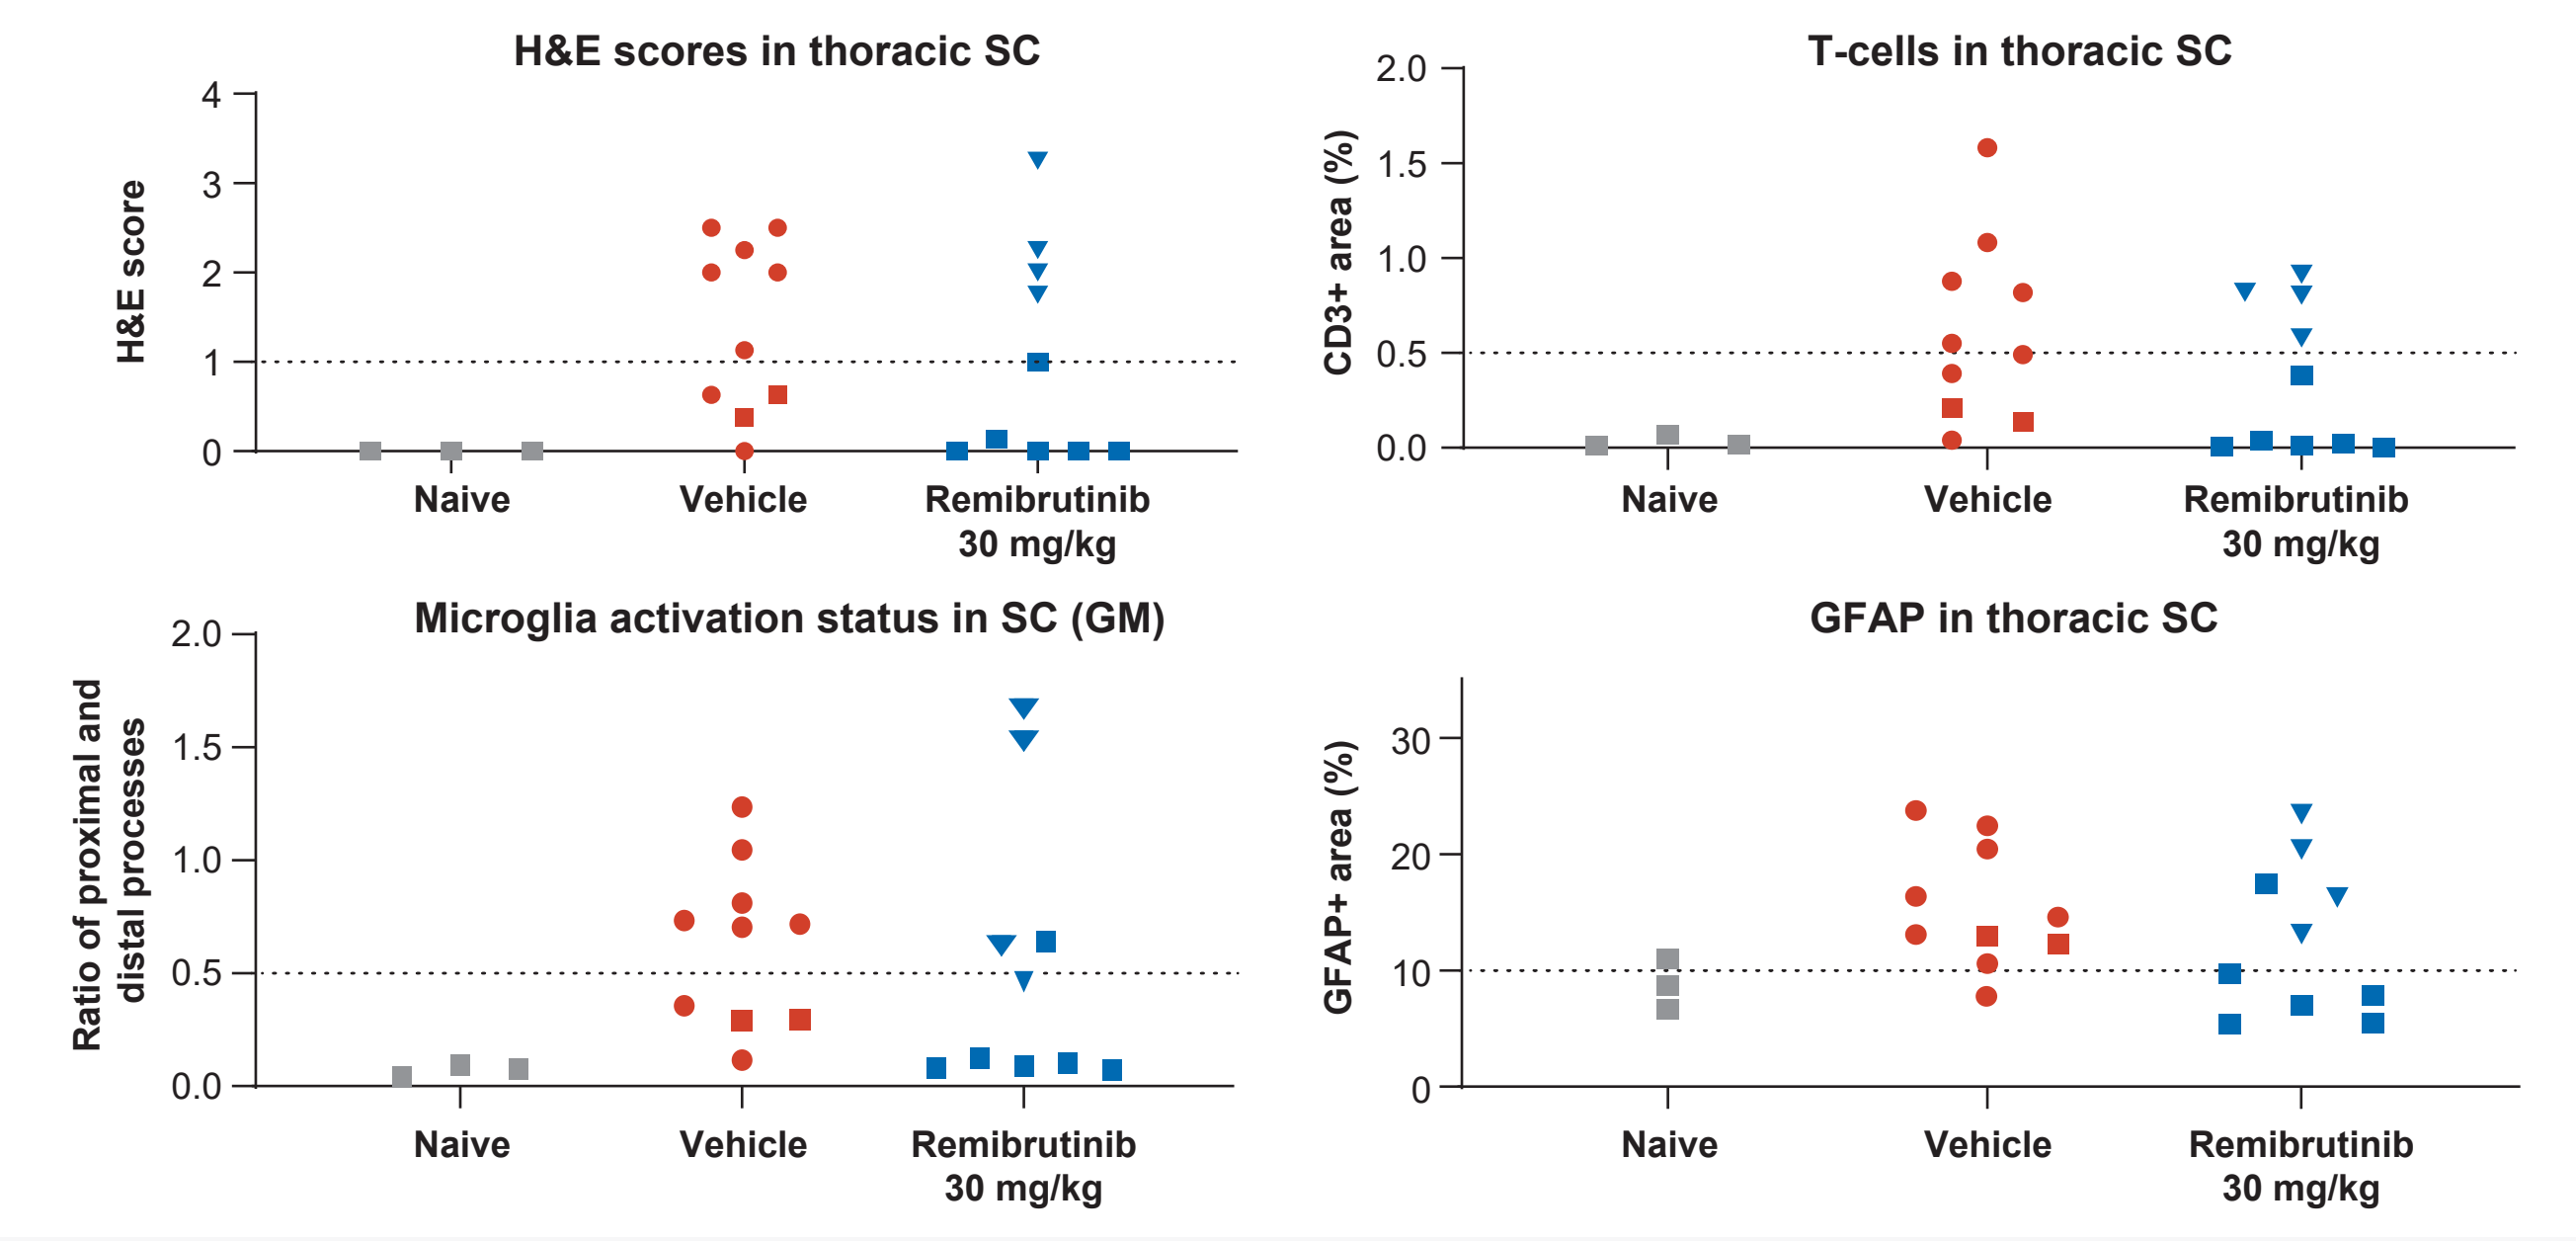

H&E scores in thoracic SC
T-cells in thoracic SC
Microglia activation status in SC (GM)
GFAP in thoracic SC
H&E score
CD3+ area (%)
Ratio of proximal and distal processes
GFAP+ area (%)
Naive
Vehicle
Remibrutinib 30 mg/kg

Squares indicate naive mice with clinical score 0.

CD, cluster of differentiation; GFAP, glial fibrillary acidic protein; GM, gray matter; H&E, hematoxylin and eosin; SC, spinal cord.

REMIBRUTINIB INHIBITS RATMOG EAE

- Remibrutinib orally dosed at 30 mg/kg b.i.d. reduced EAE clinical symptoms (**Figure 5**), suggesting that in the absence of direct T-cell inhibition, the efficacy in this RatMOG EAE model is mediated by myeloid cell and microglia inhibition[3]
- BTK occupancy was measured 16 h after the last dose, showing notable levels of BTK occupancy in the spleen, blood and brain (**Figure 5**)
- Remibrutinib led to a trend in reduction of plasma neurofilament light chain (NfL), a biomarker for neuroinflammation

Figure 5. Efficacy and BTK Occupancy in RatMOG EAE

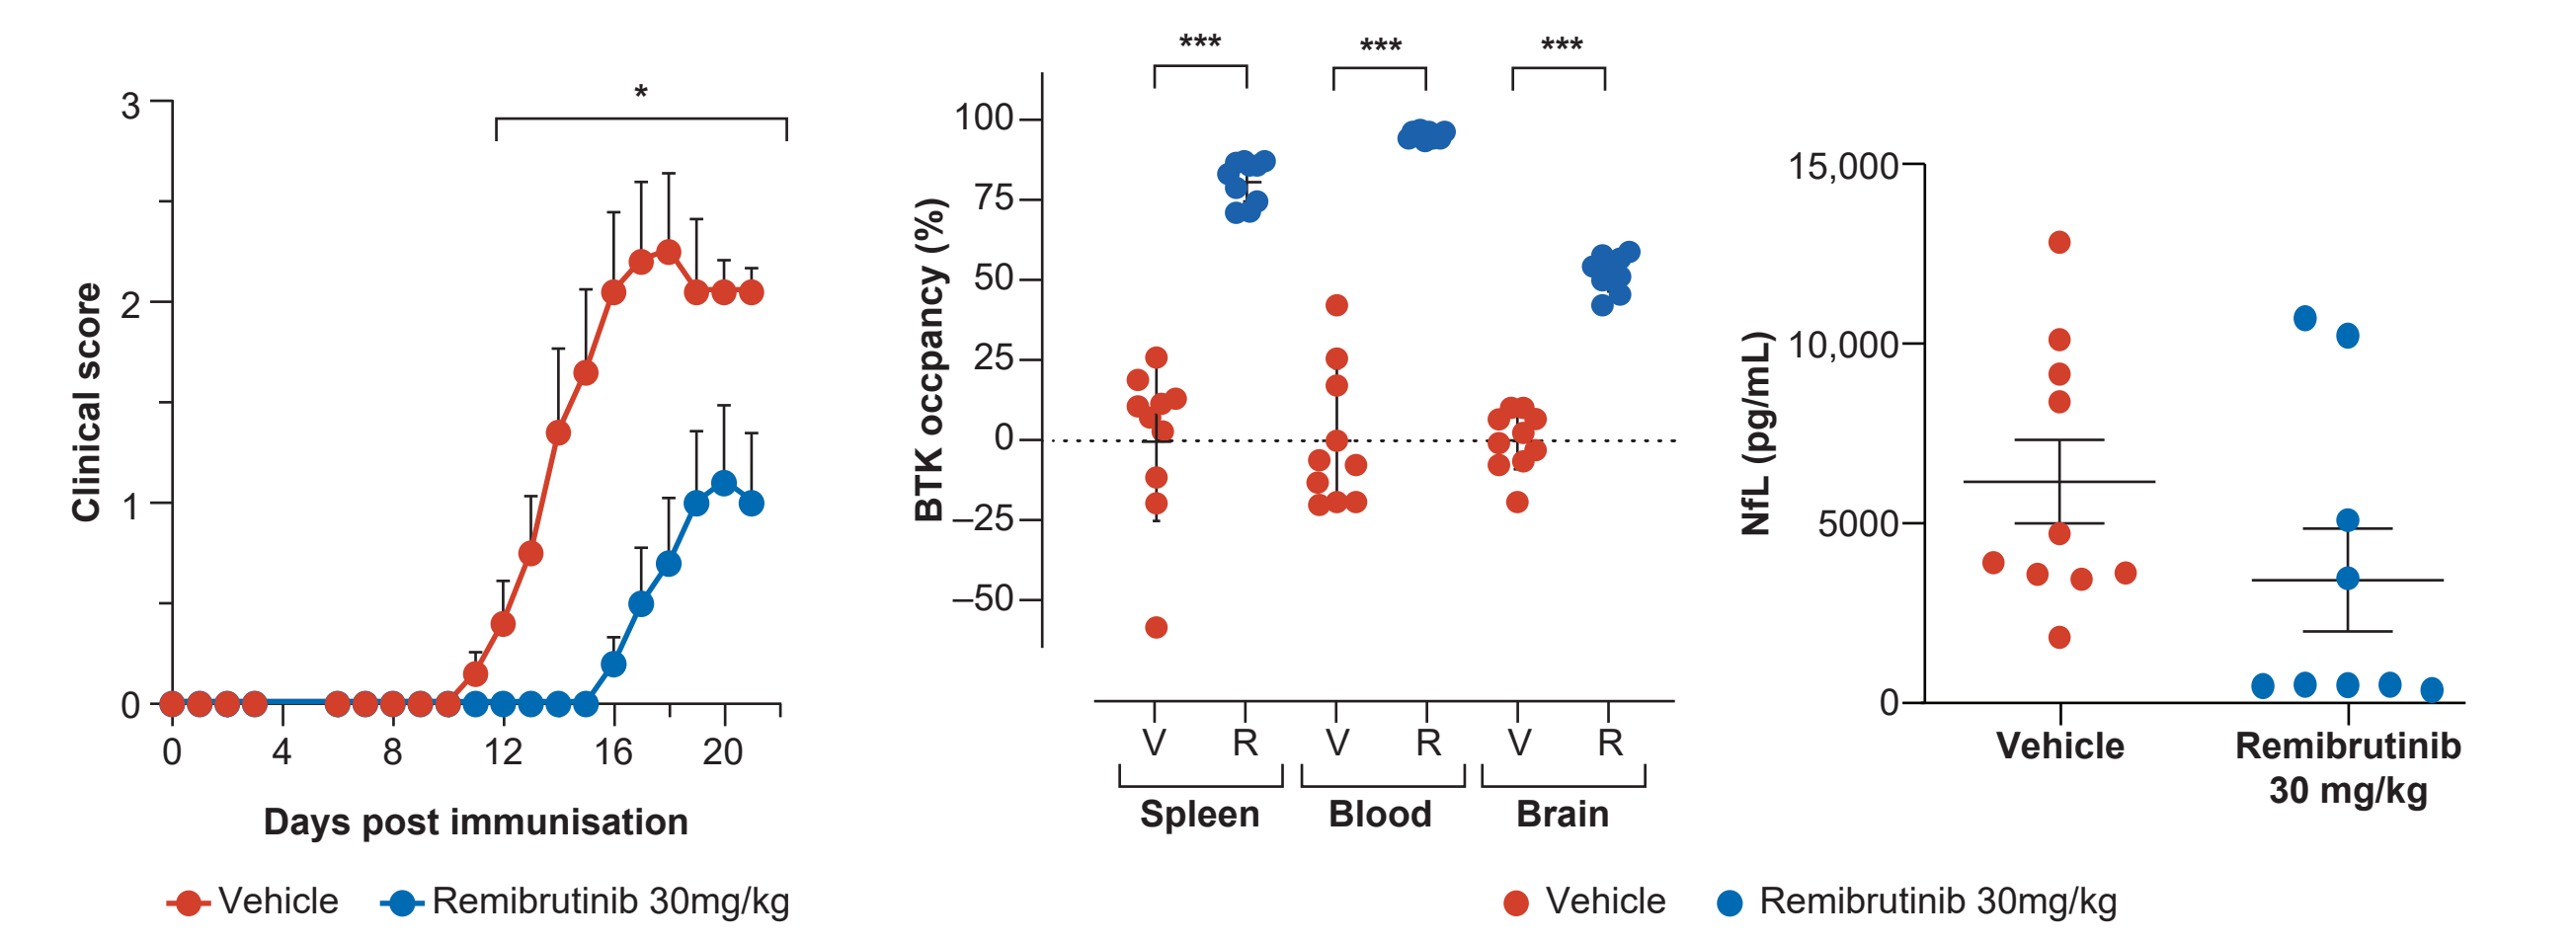

Clinical score
Days post immunisation
BTK occpancy (%)
V R V R V R
Spleen
Blood
Brain
NfL (pg/mL)
Vehicle
Remibrutinib 30 mg/kg
Remibrutinib 30mg/kg

ANOVA, analysis of variance; BTK, Bruton's tyrosine kinase; EAE, experimental autoimmune encephalomyelitis; MOG, myelin oligodendrocyte glycoprotein; NfL, neurofilament light chain; R, remibrutinib; V, vehicle.
Statistical significance (ANOVA with Dunnett's test) is shown as *p<0.05, ***p<0.001.

REDUCTION OF NEUROINFLAMMATION GENE SIGNATURE IN EAE MICROGLIA

- Analysis of scRNA-seq data obtained from brain (**Figure 6**) and spinal cord (data not shown) revealed that remibrutinib significantly downregulated multiple gene sets related to inflammation in microglia, suggesting it has an anti-inflammatory effect specifically in these cells[3]

Figure 6. Microglia RNA Signature in EAE

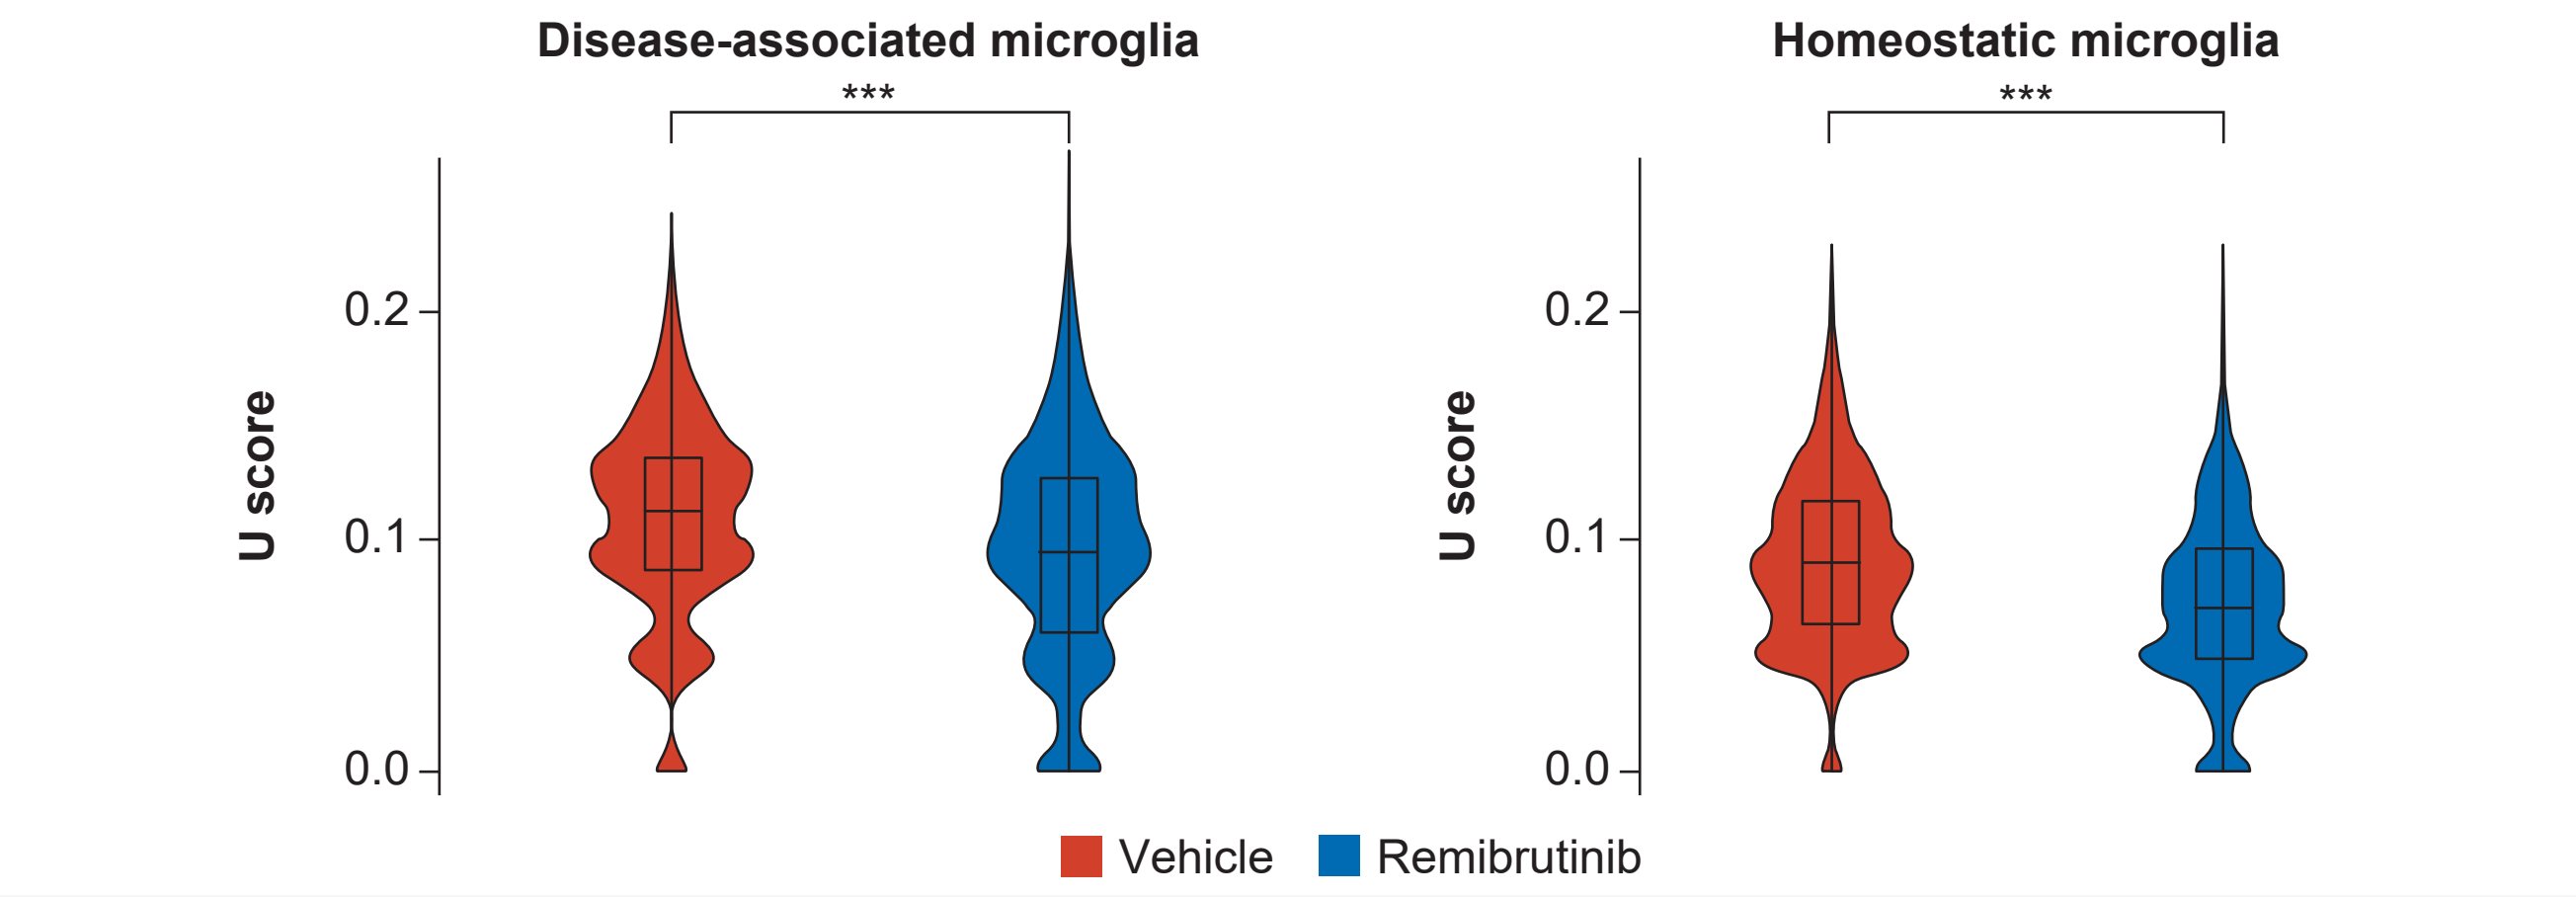

Disease-associated microglia
Homeostatic microglia
U score
Vehicle
Remibrutinib

EAE, experimental autoimmune encephalomyelitis; RNA, ribonucleic acid. ***p<0.001, one-tailed Mann-Whitney U test.

CONCLUSIONS

- Remibrutinib demonstrated clinical efficacy in a BTK-dependent EAE model, which was associated with CNS tissue protection
- Our findings in an experimental MS model support the view that BTK might represent a promising target for treating MS
- The clinical outcomes of the ongoing phase 3 trials studying remibrutinib in MS are warranted to allow translation from model systems to patients

References: 1. Steinmaurer A, et al. *Curr Pharm Des.* 2022;28(6):437-444; **2.** Wiendl H, et al. Poster presented at: American Academy of Neurology; April 2-7, 2022. P7-003; **3.** Nuesslein-Hildesheim B, et al. *J Neuroinflammation*. 2023;20(1):194.

Abbreviations: ANOVA, analysis of variance; **b.i.d.,** twice daily; **BTKi,** Bruton's tyrosine kinase inhibitor; **CD,** cluster of differentiation; **cpm,** counts per minute; **CNS,** central nervous system; **EAE,** experimental autoimmune encephalomyelitis; **GFAP,** glial fibrillary acidic protein; **GM,** gray matter; **h,** hour; **H&E,** hematoxylin and eosin; **HuMOG,** human MOG; **IBA,** ionised calcium-binding adaptor molecule; **IgG,** immunoglobulin G; **MBP,** myelin basic protein; **MOG,** myelin oligodendrocyte glycoprotein; **MS,** multiple sclerosis; **NfL,** neurofilament light chain; **ns,** not significant; **R,** remibrutinib; **RNA,** ribonucleic acid; **SC,** spinal cord; **scRNA,** single-cell RNA; **SEM,** standard error of the mean; **V,** vehicle.

Disclosures: Authors are employees of Novartis, and the studies were funded by Novartis Pharma AG, Basel, Switzerland.

Acknowledgements: The authors thank **Ulrike Naumann, Jochen Singer, Annick Waldt, Rachel Cuttat** for scRNA-seq processing; **Samuel Gaveriaux** for BTK occupancy assays; **Alfred Zimmerlin** for remibrutinib pharmacokinetic analysis; **Sarah Greiner** and **Jelena Filipova** for ELISA and flow cytometry support. Editorial support was provided by Lakshmi Narendra Bodduluru and design support was provided by Sarma Vajhula, both from Novartis Healthcare Pvt. Ltd., Hyderabad, India.

Copyright © 2023 Novartis Pharma AG. All rights reserved.

Scan this QR code to download a copy of the Poster

Visit the web at: https://bit.ly/ectrims2023.

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Presenter email address: bruno.cenni@novartis.com