Poster Presented at the 9th Joint ECTRIMS-ACTRIMS Meeting, 11–13 October 2023, Milan, Italy

Meta-analysis for Neurofilament Light Chain (NfL) as Biomarker in Mouse Experimental Autoimmune Encephalomyelitis (EAE) Studies

P118

Marc Bigaud, Sarah Tisserand, Pamela Ramseier, Uffelmann Tatjana, Catherine Huck, Giuseppe Locatelli, Bruno Cenni, Barbara Nuesslein-Hildesheim

Novartis Pharma AG, Basel, Switzerland

SUMMARY

1 The neurofilament light chain (NfL) level in cerebrospinal fluid (CSF; cNfL) and/or plasma (pNfL) is considered as a promising prognostic biomarker of disease activity in multiple sclerosis (MS)

- – To assess the preclinical value of cNfL and pNfL, data from 25 mouse experimental autoimmune encephalomyelitis (EAE) studies performed over the last 2 years were subjected to meta-analysis

2 In EAE-induced mice, cNfL and pNfL levels markedly increased over time with maximal values observed at peak of disease (Day 19 post-disease induction [pDI])

- – At 19 days and 28 days pDI, significant correlations were shown between cNfL and pNfL levels and between cNfL or pNfL versus EAE clinical scores, consistent with observations in humans

3 This meta-analysis supports the translational value of cNfL and/or pNfL in mouse EAE studies

INTRODUCTION

- NfL, a neuron-specific cytoskeletal protein released upon neuro-axonal damage, is considered as a promising prognostic biomarker of disease activity in MS[1,2]
- In humans, NfL measurements obtained from CSF and plasma are highly correlated[3]
 - – As pNfL can be easily measured, it has the potential to be used as an additional tool to evaluate disease prognosis in people living with MS
- However, the value of NfL as a biomarker in preclinical models such as mouse EAE is still debated

OBJECTIVE

- To assess the correlation between cNfL and pNfL, and between cNfL or pNfL and clinical scores in an EAE mouse model

METHODS

- Twenty-five independent EAE studies were conducted over 2 years in C57BL/6J mice
 - – EAE was induced via ratMOG$_{28–152}$ in complete Freund's adjuvant
 - – Studies assessed the effects of pharmacological interventions in EAE-induced mice by analysing clinical scores, cNfL and pNfL levels (NfL ELISA kit #10-7001, Uman Diagnostics)
- All available data were pooled and subjected to meta-analysis

RESULTS

NfL Levels in Healthy (Naive) and EAE-Induced (Control) Mice

- **Naive mice:** Mean±SEM cNfL and pNfL levels were 10.4±0.1 and 0.42±0.03 ng/mL, respectively, indicating a CSF/plasma ratio of ~25
- **EAE-induced mice:** cNfL and pNfL levels significantly increased pDI, although with high inter-individual variability (**Figure 1**)
 - – At peak of disease, which occurred typically around Day 19 pDI, cNfL and pNfL reached maximal mean values of 287±27 (n=114) and 8.0±0.8 (n=126) ng/mL, respectively, indicating a CSF/plasma ratio of ~35
 - – These cNfL and pNfL levels remained elevated up to 1-month pDI (n=61 and 50, respectively) and showed a ~10-fold reduction at 2 months pDI (n=10 and 9, respectively)

Figure 1. Longitudinal Changes in NfL Levels in the CSF (A) and Plasma (B) of Mice Upon EAE Induction

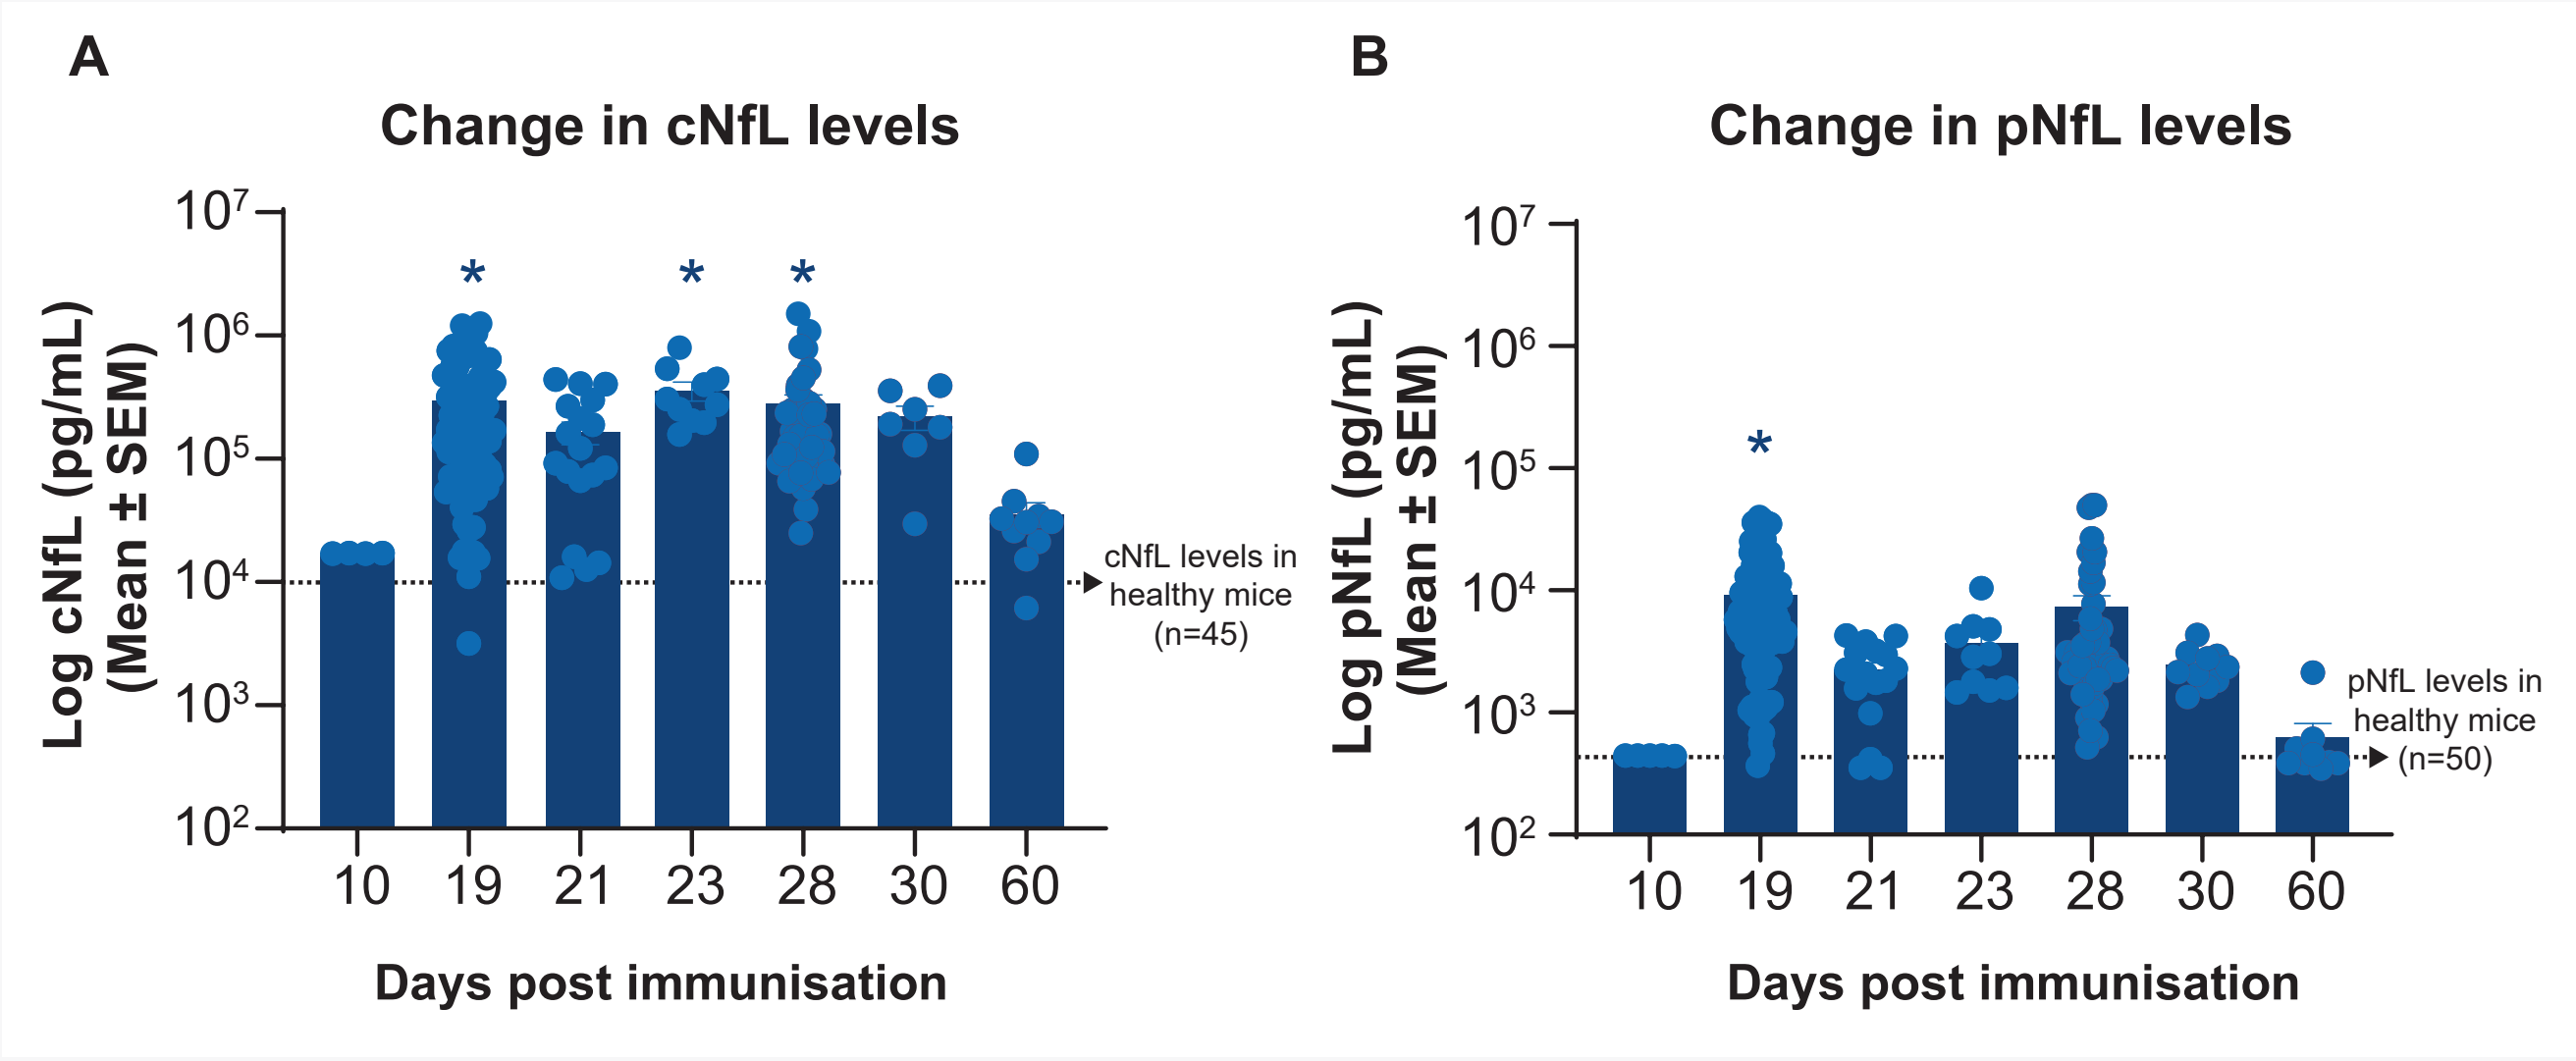
A
Change in cNfL levels
Log cNfL (pg/mL) (Mean ± SEM)
10^7
10^6
10^5
10^4
10^3
10^2
*
*
*
cNfL levels in healthy mice (n=45)
10 19 21 23 28 30 60
Days post immunisation
B
Change in pNfL levels
Log pNfL (pg/mL) (Mean ± SEM)
10^7
10^6
10^5
10^4
10^3
10^2
*
pNfL levels in healthy mice (n=50)
10 19 21 23 28 30 60
Days post immunisation

*p<0.0001 vs healthy (naive) mice (analysed using two-way ANOVA). Dotted line indicates the mean NfL value in healthy (naive) mice. A total of 45 and 50 mice were used for assessing the mean value of cNfL and pNfL, respectively.

ANOVA, analysis of variance; cNfL, cerebrospinal fluid NfL; CSF, cerebrospinal fluid; EAE, experimental autoimmune encephalomyelitis; NfL, neurofilament light chain; pNfL, plasma NfL; SEM, standard error of mean.

Correlation Between cNfL or pNfL Versus Clinical Scores, and Between cNfL and pNfL Levels

- Overall, the meta-analysis demonstrated that correlations between cNfL or pNfL vs EAE clinical scores (**Figure 2**) and between pNfL vs cNfL levels (**Figure 3**) were highly significant ($p<0.0001$ for all), supporting pNfL as a robust predictor of cNfL in EAE mouse models

Figure 2. Correlations Between cNfL (A) or pNfL (B) Levels vs the EAE Clinical Scores Observed at 19 Days or 28 Days After EAE Induction

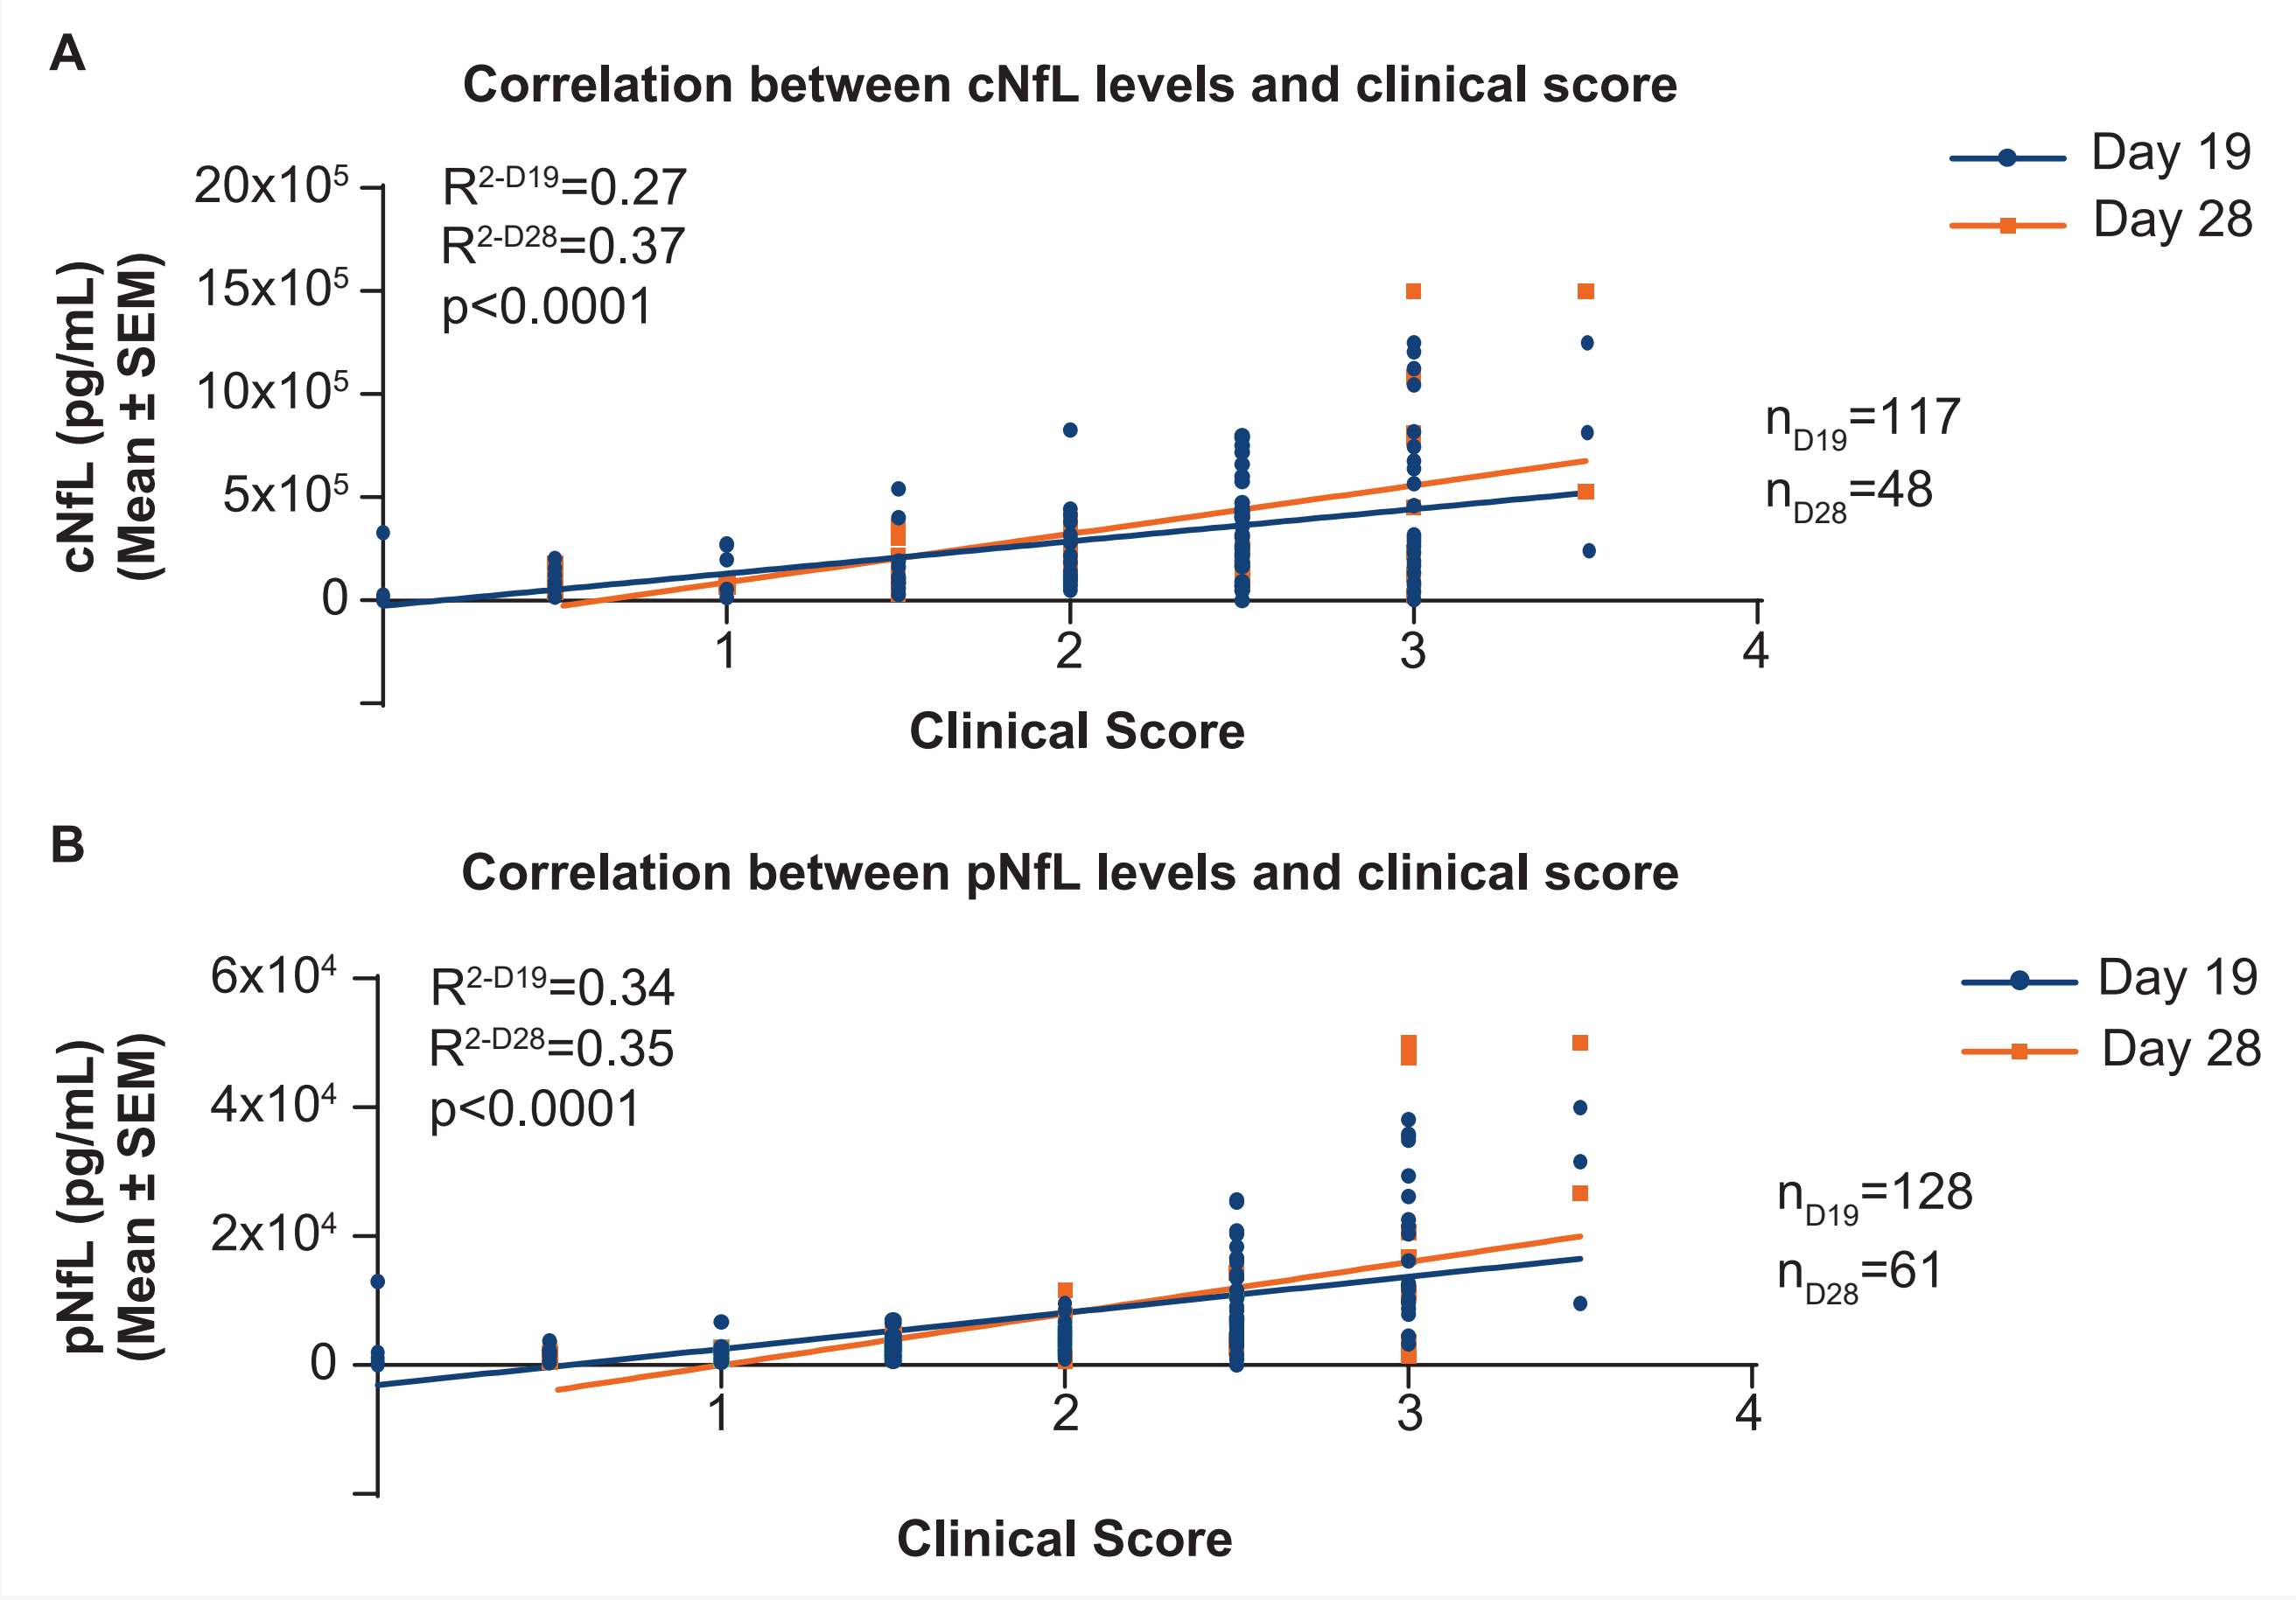
A
Correlation between cNfL levels and clinical score
cNfL (pg/mL) (Mean ± SEM)
20x10^5
15x10^5
10x10^5
5x10^5
0
$R^{2\text{-}D19}$=0.27
$R^{2\text{-}D28}$=0.37
p<0.0001
Day 19
Day 28
n_{D19}=117
n_{D28}=48
1 2 3 4
Clinical Score
B
Correlation between pNfL levels and clinical score
pNfL (pg/mL) (Mean ± SEM)
6x10^4
4x10^4
2x10^4
0
$R^{2\text{-}D19}$=0.34
$R^{2\text{-}D28}$=0.35
p<0.0001
Day 19
Day 28
n_{D19}=128
n_{D28}=61
1 2 3 4
Clinical Score

"n" refers to the number of mice with available data at that timepoint.
cNfL, cerebrospinal fluid NfL; EAE, experimental autoimmune encephalomyelitis; NfL, neurofilament light chain; pNfL, plasma NfL; SEM, standard error of mean.

Figure 3. Correlations Between cNfL and pNfL Levels Measured in EAE Mice at 19 Days (A) and 28 Days (B) After EAE Induction

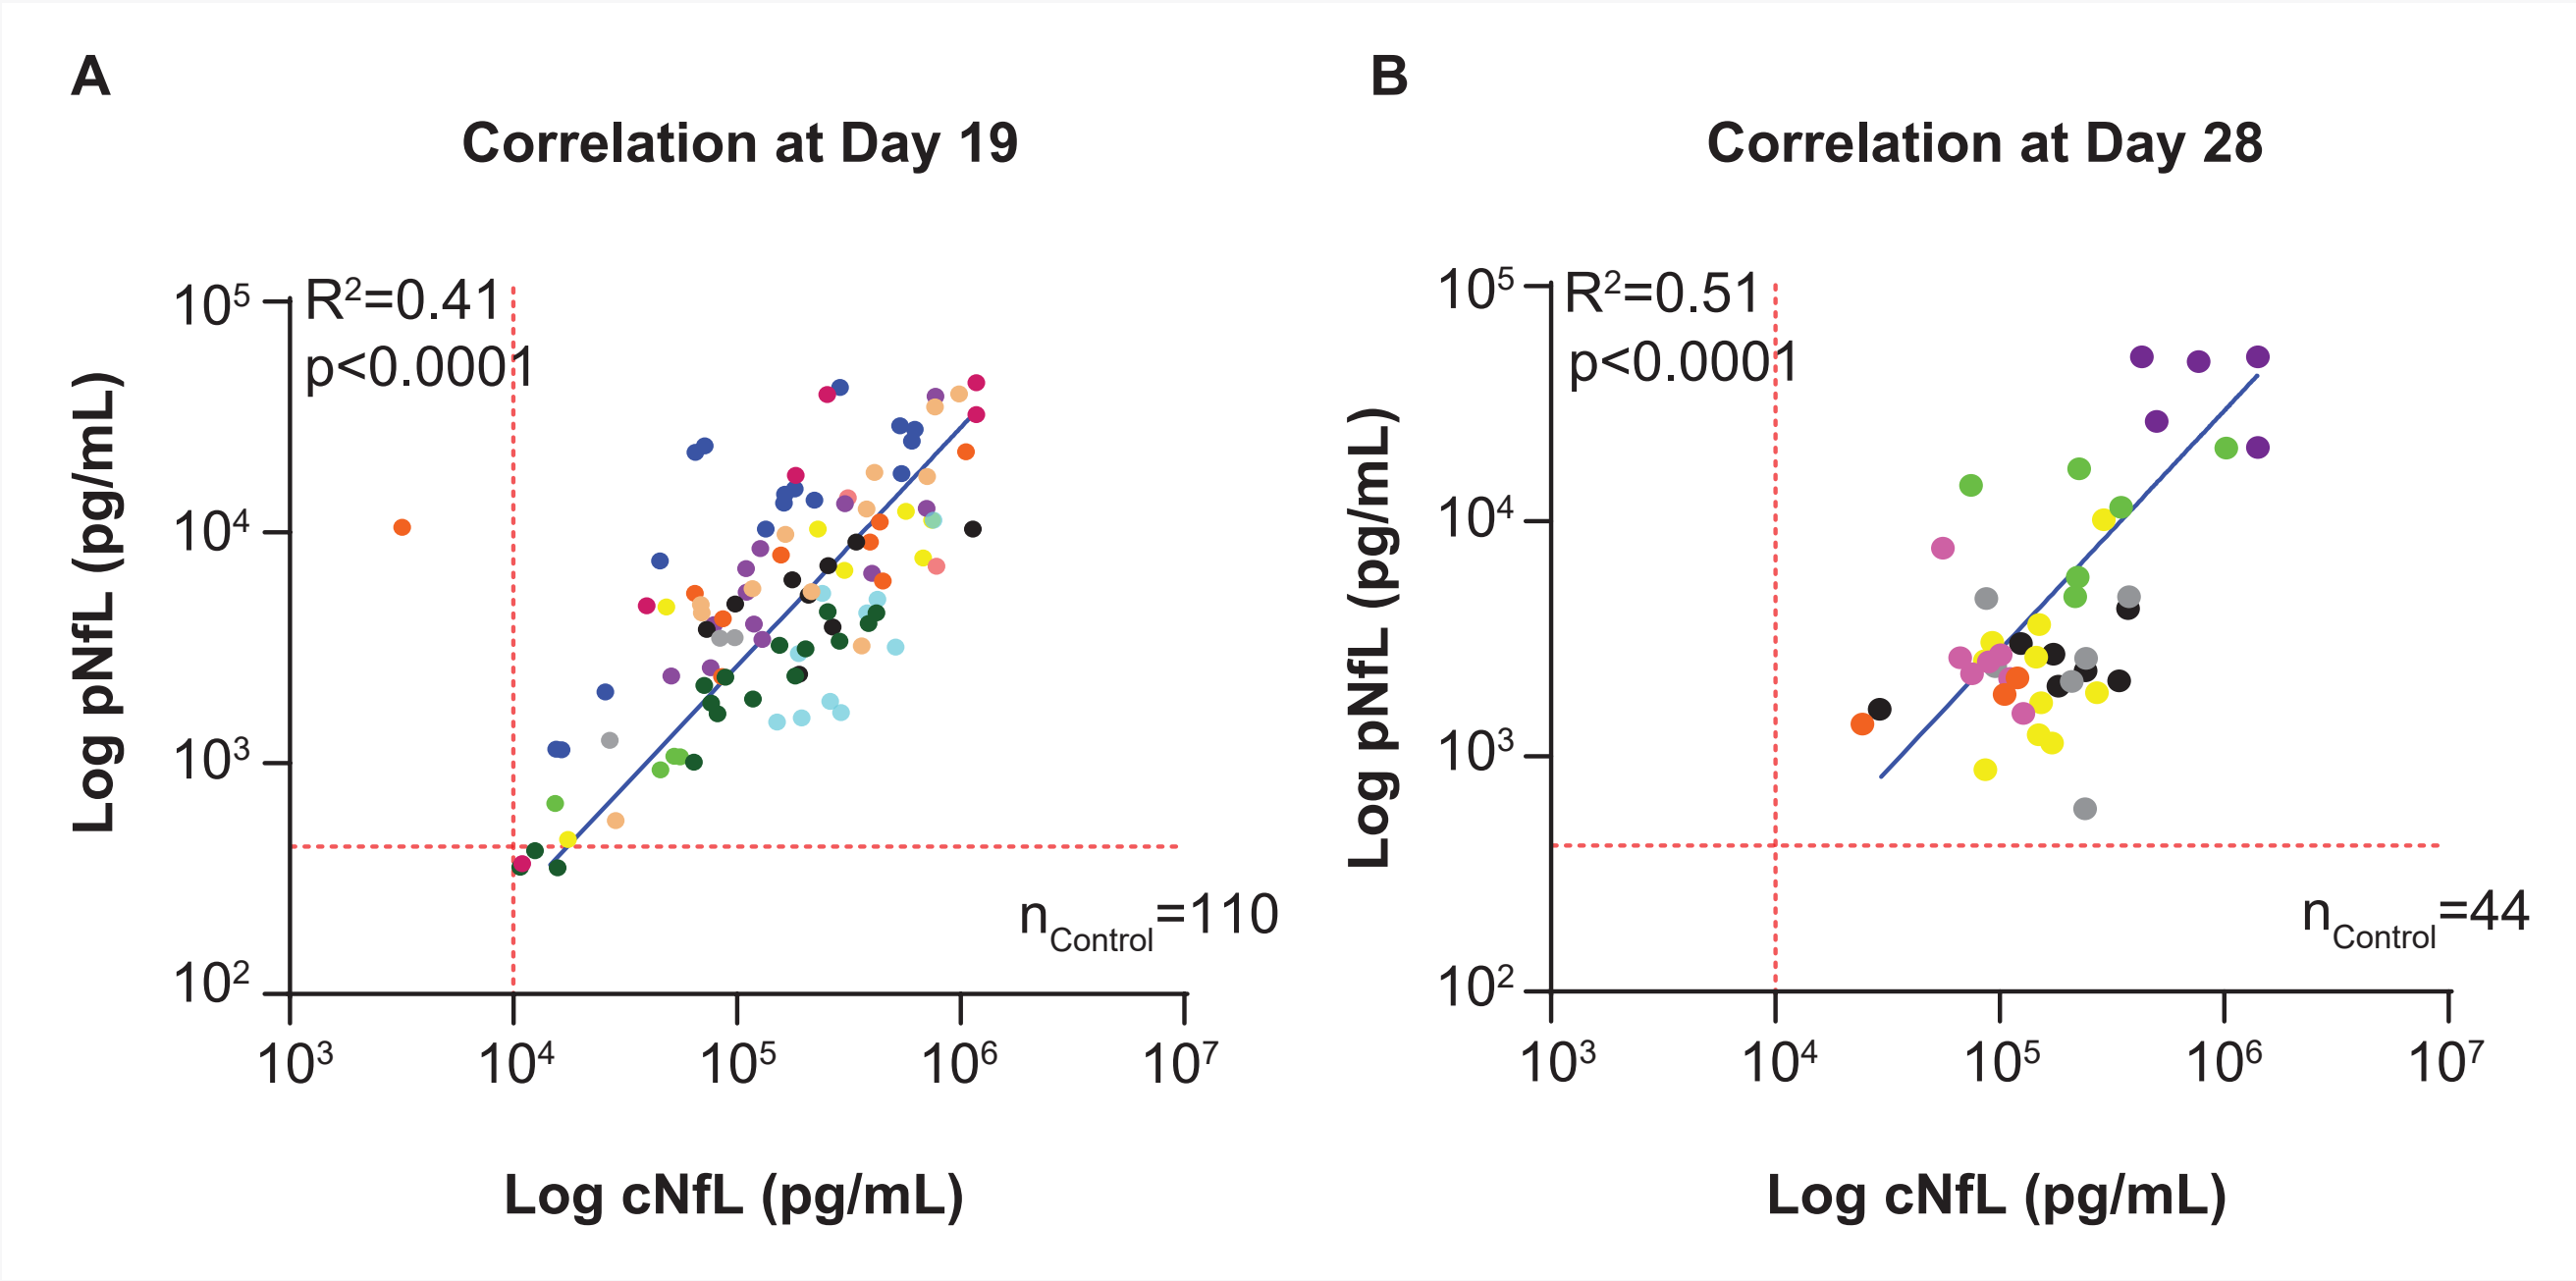
A
Correlation at Day 19
Log pNfL (pg/mL)
R^2=0.41
p<0.0001
$n_{Control}$=110
10^5
10^4
10^3
10^2
10^3 10^4 10^5 10^6 10^7
Log cNfL (pg/mL)
B
Correlation at Day 28
Log pNfL (pg/mL)
R^2=0.51
p<0.0001
$n_{Control}$=44
10^5
10^4
10^3
10^2
10^3 10^4 10^5 10^6 10^7
Log cNfL (pg/mL)

"n" refers to the number of mice with available data at that timepoint. Control indicates EAE-induced mice. Each colour corresponds to an individual study. cNfL, cerebrospinal fluid NfL; EAE, experimental autoimmune encephalomyelitis; NfL, neurofilament light chain; pNfL, plasma NfL.

Effect of Remibrutinib on cNfL and pNfL Levels

- Remibrutinib, a novel, potent, and highly selective Bruton's tyrosine kinase inhibitor currently in development for MS, achieved significant reductions in both cNfL and pNfL as well as clinical scores in EAE mice (**Figure 4**)

Figure 4. Effect of Remibrutinib Treatment (in Diet, 30 mg/kg of Food) on the cNfL and pNfL Levels Measured in EAE Mice at 19 Days Post Induction

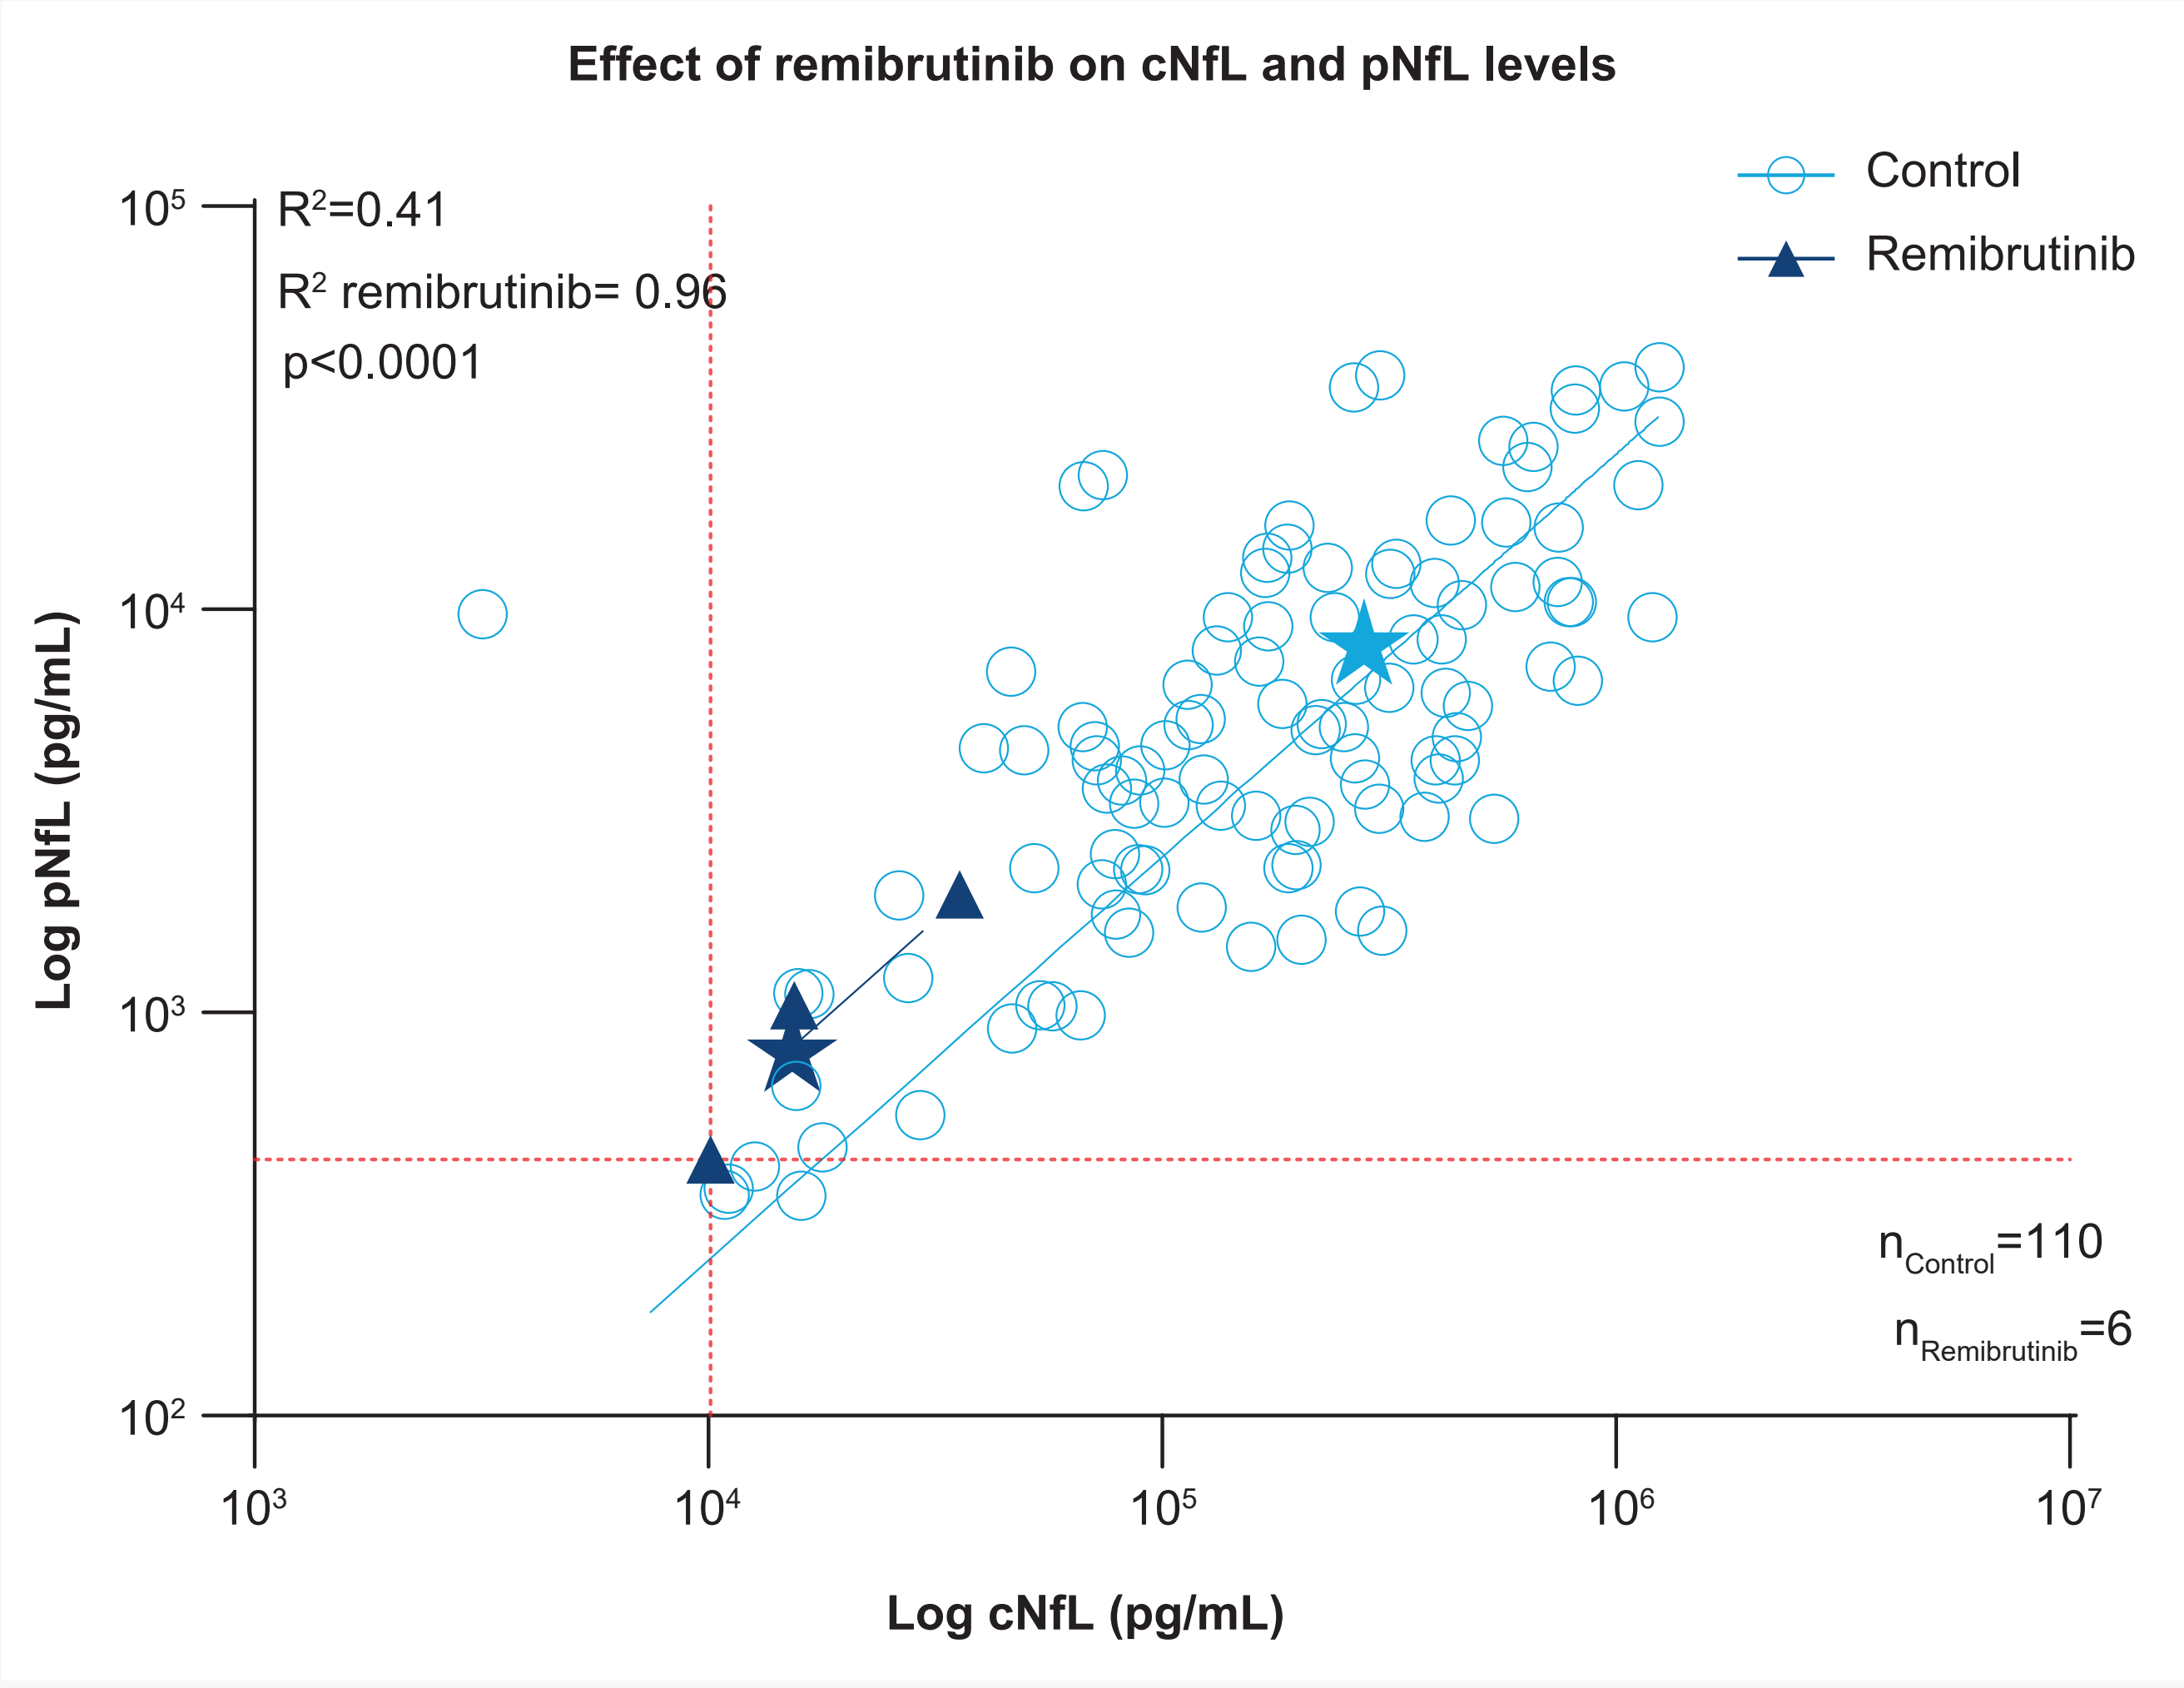
Effect of remibrutinib on cNfL and pNfL levels
Control
Remibrutinib
R^2=0.41
R^2 remibrutinib= 0.96
p<0.0001
Log pNfL (pg/mL)
10^5
10^4
10^3
10^2
$n_{Control}$=110
$n_{Remibrutinib}$=6
10^3 10^4 10^5 10^6 10^7
Log cNfL (pg/mL)

"n" refers to the number of mice with available data at that timepoint. Control refers to EAE-induced mice. Data from a single study are presented for remibrutinib.

cNfL, cerebrospinal fluid NfL; EAE, experimental autoimmune encephalomyelitis; NfL, neurofilament light chain; pNfL, plasma NfL.

CONCLUSIONS

- In mice, pNfL was highly correlated with cNfL, consistent with observations in humans
- The present meta-analysis supports the translational value of cNfL and/or pNfL in mouse EAE studies to assess neuroaxonal damage, disease activity, and possibly the therapeutic potential of new disease-modifying therapies in development

References: **1**. Ning L, et al. *PLoS One*. 2022;17(9):e0274565; **2**. Ziemssen T, et al. *Front Immunol*. 2022;13:852563; **3**. Piehl F, et al. *Mult Scler*. 2018;24(8):1046–1054.

Abbreviations: ANOVA, analysis of variance; **cNfL**, cerebrospinal fluid NfL; **CSF**, cerebrospinal fluid; **EAE**, experimental autoimmune encephalomyelitis; **MOG**, myelin oligodendrocyte glycoprotein; **MS**, multiple sclerosis; **NfL**, neurofilament light chain; **pDI**, post disease induction; **pNfL**, plasma NfL; **SEM**, standard error of mean.

Disclosures: Authors are employees of Novartis Pharma AG.

Acknowledgements: This study was funded by Novartis Pharma AG, Basel, Switzerland. Editorial support was provided by Lakshmi Narendra Bodduluru and design support was provided by Ravikishor Babu, both from Novartis Healthcare Pvt. Ltd., Hyderabad, India. The final responsibility for the content lies with the authors.

Copyright © 2023 Novartis Pharma AG. All rights reserved.

Scan this QR code to download a copy of the Poster

Visit the web at:
https://bit.ly/ectrims2023.

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Presenter email address:
marc.bigaud@novartis.com