Poster Presented at the 9th Joint ECTRIMS-ACTRIMS Meeting, 11–13 October 2023, Milan, Italy

P179

Efficacy of Ofatumumab on Microglial Activity and Brain Iron in Patients With Relapsing Forms of Multiple Sclerosis: Results From a 9-Month Study

Tarun Singhal[1, 2], Ferdinand Schweser[3], Steven Vaquerano[2], John Hunter Ficke[2], Preksha Kukreja[2], Hong Pan[1, 2], Eero Rissanen[1, 2], Kelsey Carter[2], Gauruv Bose[1, 2], Kristin Galetta[1, 2], Bonnie Glanz[1, 2], Jonathan Zurawski[1, 2], Steven Cicero[2], Maria K. Houtchens[1, 2], Christopher Severson[1, 2], Shipra Dubey[1, 2], Bo Yeun Yang[1, 2], Rohit Bakshi[1, 2], Tanuja Chitnis[1, 2], Brandon Brown[4], Ibolya Boer[5], Howard Weiner[1, 2]

[1]Harvard Medical School, Boston, MA, USA; [2]Department of Neurology, Brigham and Women's Hospital, Boston, MA, USA; [3]Department of Neurology, Jacobs School of Medicine & Biomedical Sciences, University of Buffalo, Buffalo Neuroimaging Analysis Center, Buffalo, NY, USA; [4]Novartis Pharmaceutical Corporation, East Hanover, NJ, USA; [5]Novartis Pharma AG, Basel, Switzerland

SUMMARY

1. An open label, single-centre, observational study was conducted to determine the effect of ofatumumab on microglial activation and brain iron in 10 patients with active relapsing multiple sclerosis (MS)
2. Ofatumumab treatment was associated with decreased microglial activation in cortical grey matter (CoGM), decreased brain iron in CoGM and normal-appearing white matter (NAWM), and reduced serum neurofilament light chain (NfL) levels at 9 months
3. Peripheral CD19+ cell depletion preceding the effect of ofatumumab on microglial activation may suggest an indirect, downstream effect of B-cell depletion on microglial activity in patients with relapsing forms of MS
4. The relationship between changes in cortical microglial activation, brain iron and serum NfL in response to ofatumumab warrants further investigation

INTRODUCTION

- Overactivation of microglia, the innate immune cells of the central nervous system, has been implicated in the pathogenesis of MS[1]
- Upon activation, human microglia cells form clusters in active lesion rims and in normal tissue. These clusters are detectable via targeting of the glial marker 18-kilodalton translocator protein (TSPO)[2,3]
- Iron accumulation has been described within macrophages/microglia at the edges of slowly expanding and some inactive lesions; iron accumulation and its liberation during demyelination are thought to be one of the key factors of neurodegeneration in progressive MS[4]
- Histopathologically, chronic active lesions are also characterised by progressive tissue matrix damage, driven by a rim of iron-laden activated microglia at the lesion edge[4,5]
- In animal models, B-cell depletion using anti-CD20 antibodies led to reduced microglial activation and lesion formation[6]
- The potential impact of ofatumumab on microglial activation and its association with peripheral B-cell depletion and brain iron changes in MS is unknown

OBJECTIVE

- To determine the effect of ofatumumab on microglial activation and brain iron, using [F-18]PBR06 positron emission tomography (PET) and quantitative susceptibility mapping (QSM) in patients with MS, in relation to peripheral B-cell depletion, serum biomarker measurement and changes in clinical impairment, longitudinally over 9 months

METHODS

- The study design is presented in **Figure 1.** A statistical parametric mapping (SPM) analysis was performed to assess the reduction in PET and QSM signals following treatment with ofatumumab
- NAWM, CoGM and the thalamic region of interest (ROI) in standard atlas space were interrogated for peak cluster T-values corresponding to p<0.05
- Serum biomarker assessments included serum NfL and glial fibrillary acidic protein (GFAP) measurements
- Clinical evaluations included the change in Expanded Disability Status Scale (EDSS), and the timed 25-foot walk test (T25FWT) scores. Anxiety and depression scores were evaluated using HADS
- All parameters assessed at 9 months were compared with baseline values
- At the time of the analyses, 9-month PET and QSM data were available for only five patients due to unavailability of the tracer

Figure 1. Study Design

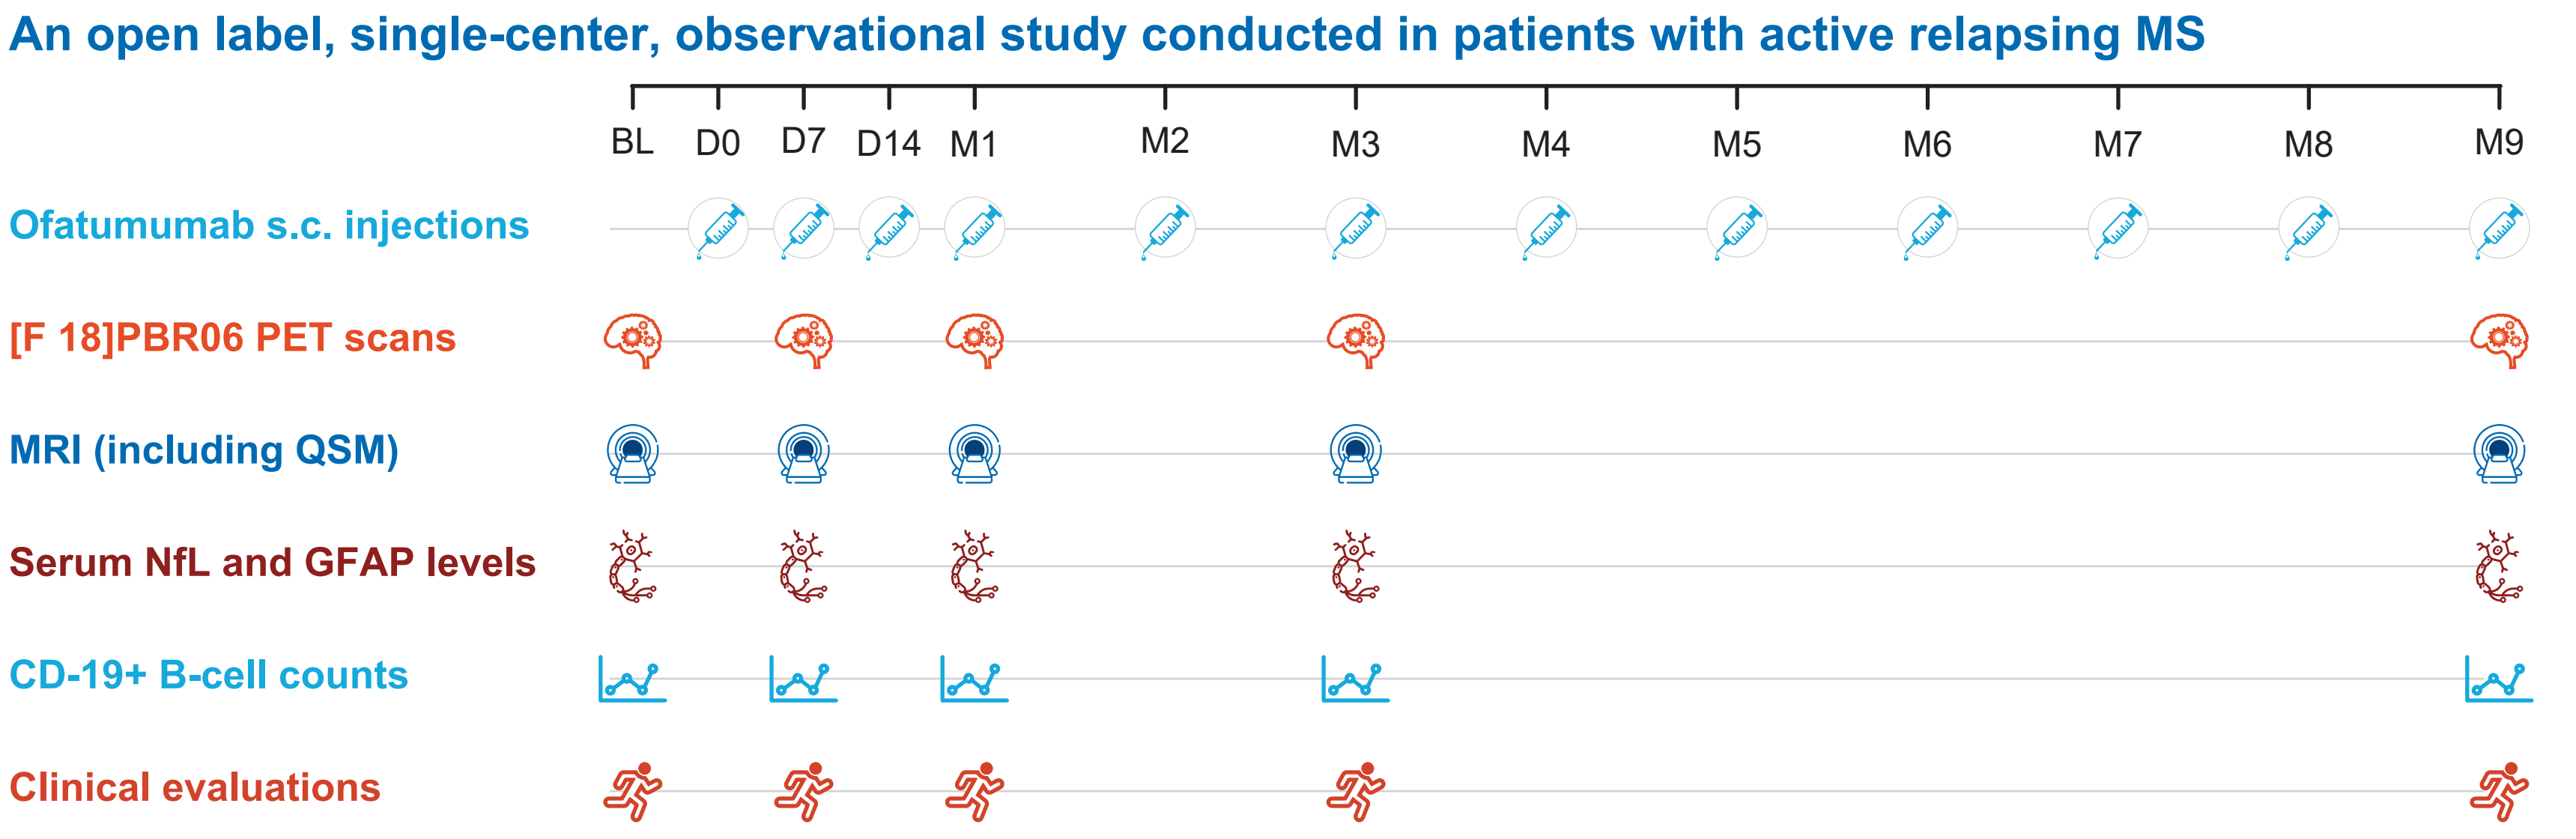
An open label, single-center, observational study conducted in patients with active relapsing MS
BL D0 D7 D14 M1 M2 M3 M4 M5 M6 M7 M8 M9
Ofatumumab s.c. injections
[F 18]PBR06 PET scans
MRI (including QSM)
Serum NfL and GFAP levels
CD-19+ B-cell counts
Clinical evaluations

BL, baseline; D, day; GFAP, glial fibrillary acid protein; M, month; MRI, magnetic resonance imaging; MS, multiple sclerosis; NfL, neurofilament light chain; PET, positron emission tomography; QSM, quantitative susceptibility mapping; s.c., subcutaneous.

RESULTS

PATIENT CHARACTERISTICS

- Ten patients with relapsing MS (mean±SD age: 40.2±12 years; mean EDSS score: 3.0) were enrolled in the study

EFFECT OF OFATUMUMAB ON MICROGLIAL CLUSTERS

- Ofatumumab treatment significantly decreased PET uptake in CoGM (p<0.01) after 9 months compared to baseline (**Figure 2**)
- No clusters of significantly reduced PET uptake were detectable in NAWM and thalamic ROIs

Figure 2. [F-18]PBR06-PET: 9 Months Versus Baseline (N=5)*

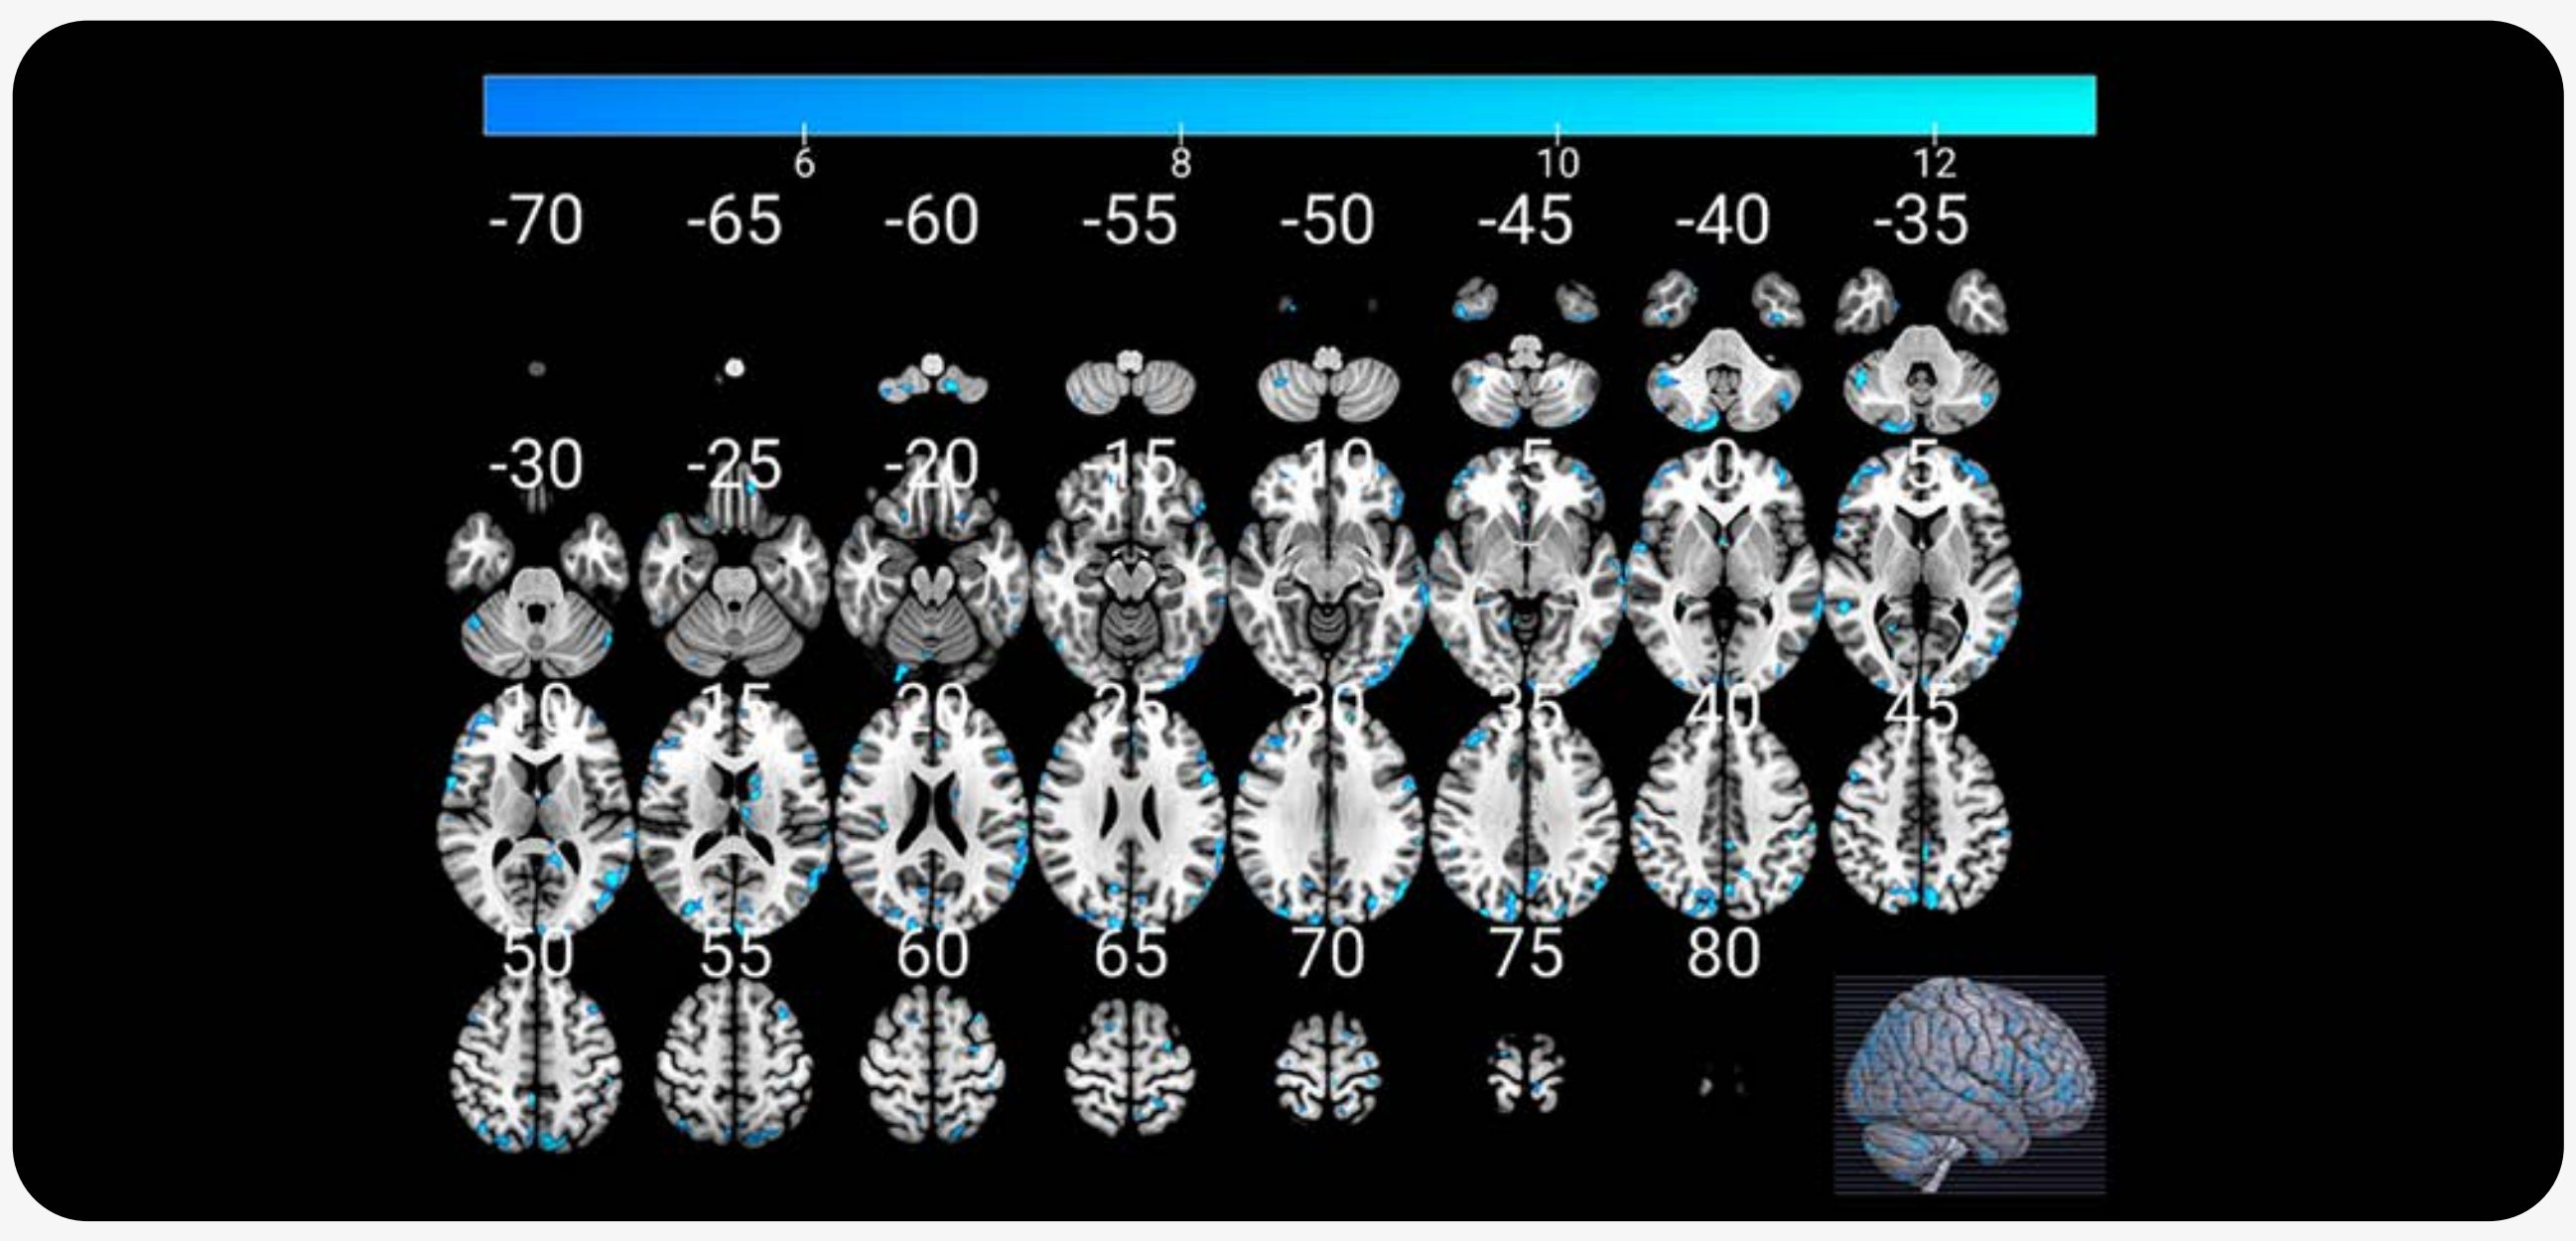
6 8 10 12
-70 -65 -60 -55 -50 -45 -40 -35
-30 -25 -20 -15 -10 -5 0 5
10 15 20 25 30 35 40 45
50 55 60 65 70 75 80

*PET scans could only be conducted in five patients due to the unavailability of the tracer. PET, positron emission tomography.

EFFECT OF OFATUMUMAB ON IRON LEVELS IN THE BRAIN

- After 9 months of ofatumumab treatment, clusters of decreased QSM signal, representing decreased brain iron, were seen in CoGM and NAWM using SPM analysis (p<0.01; **Figure 3**)

Figure 3. Change in QSM Signal: 9 Months Versus Baseline (N=5)

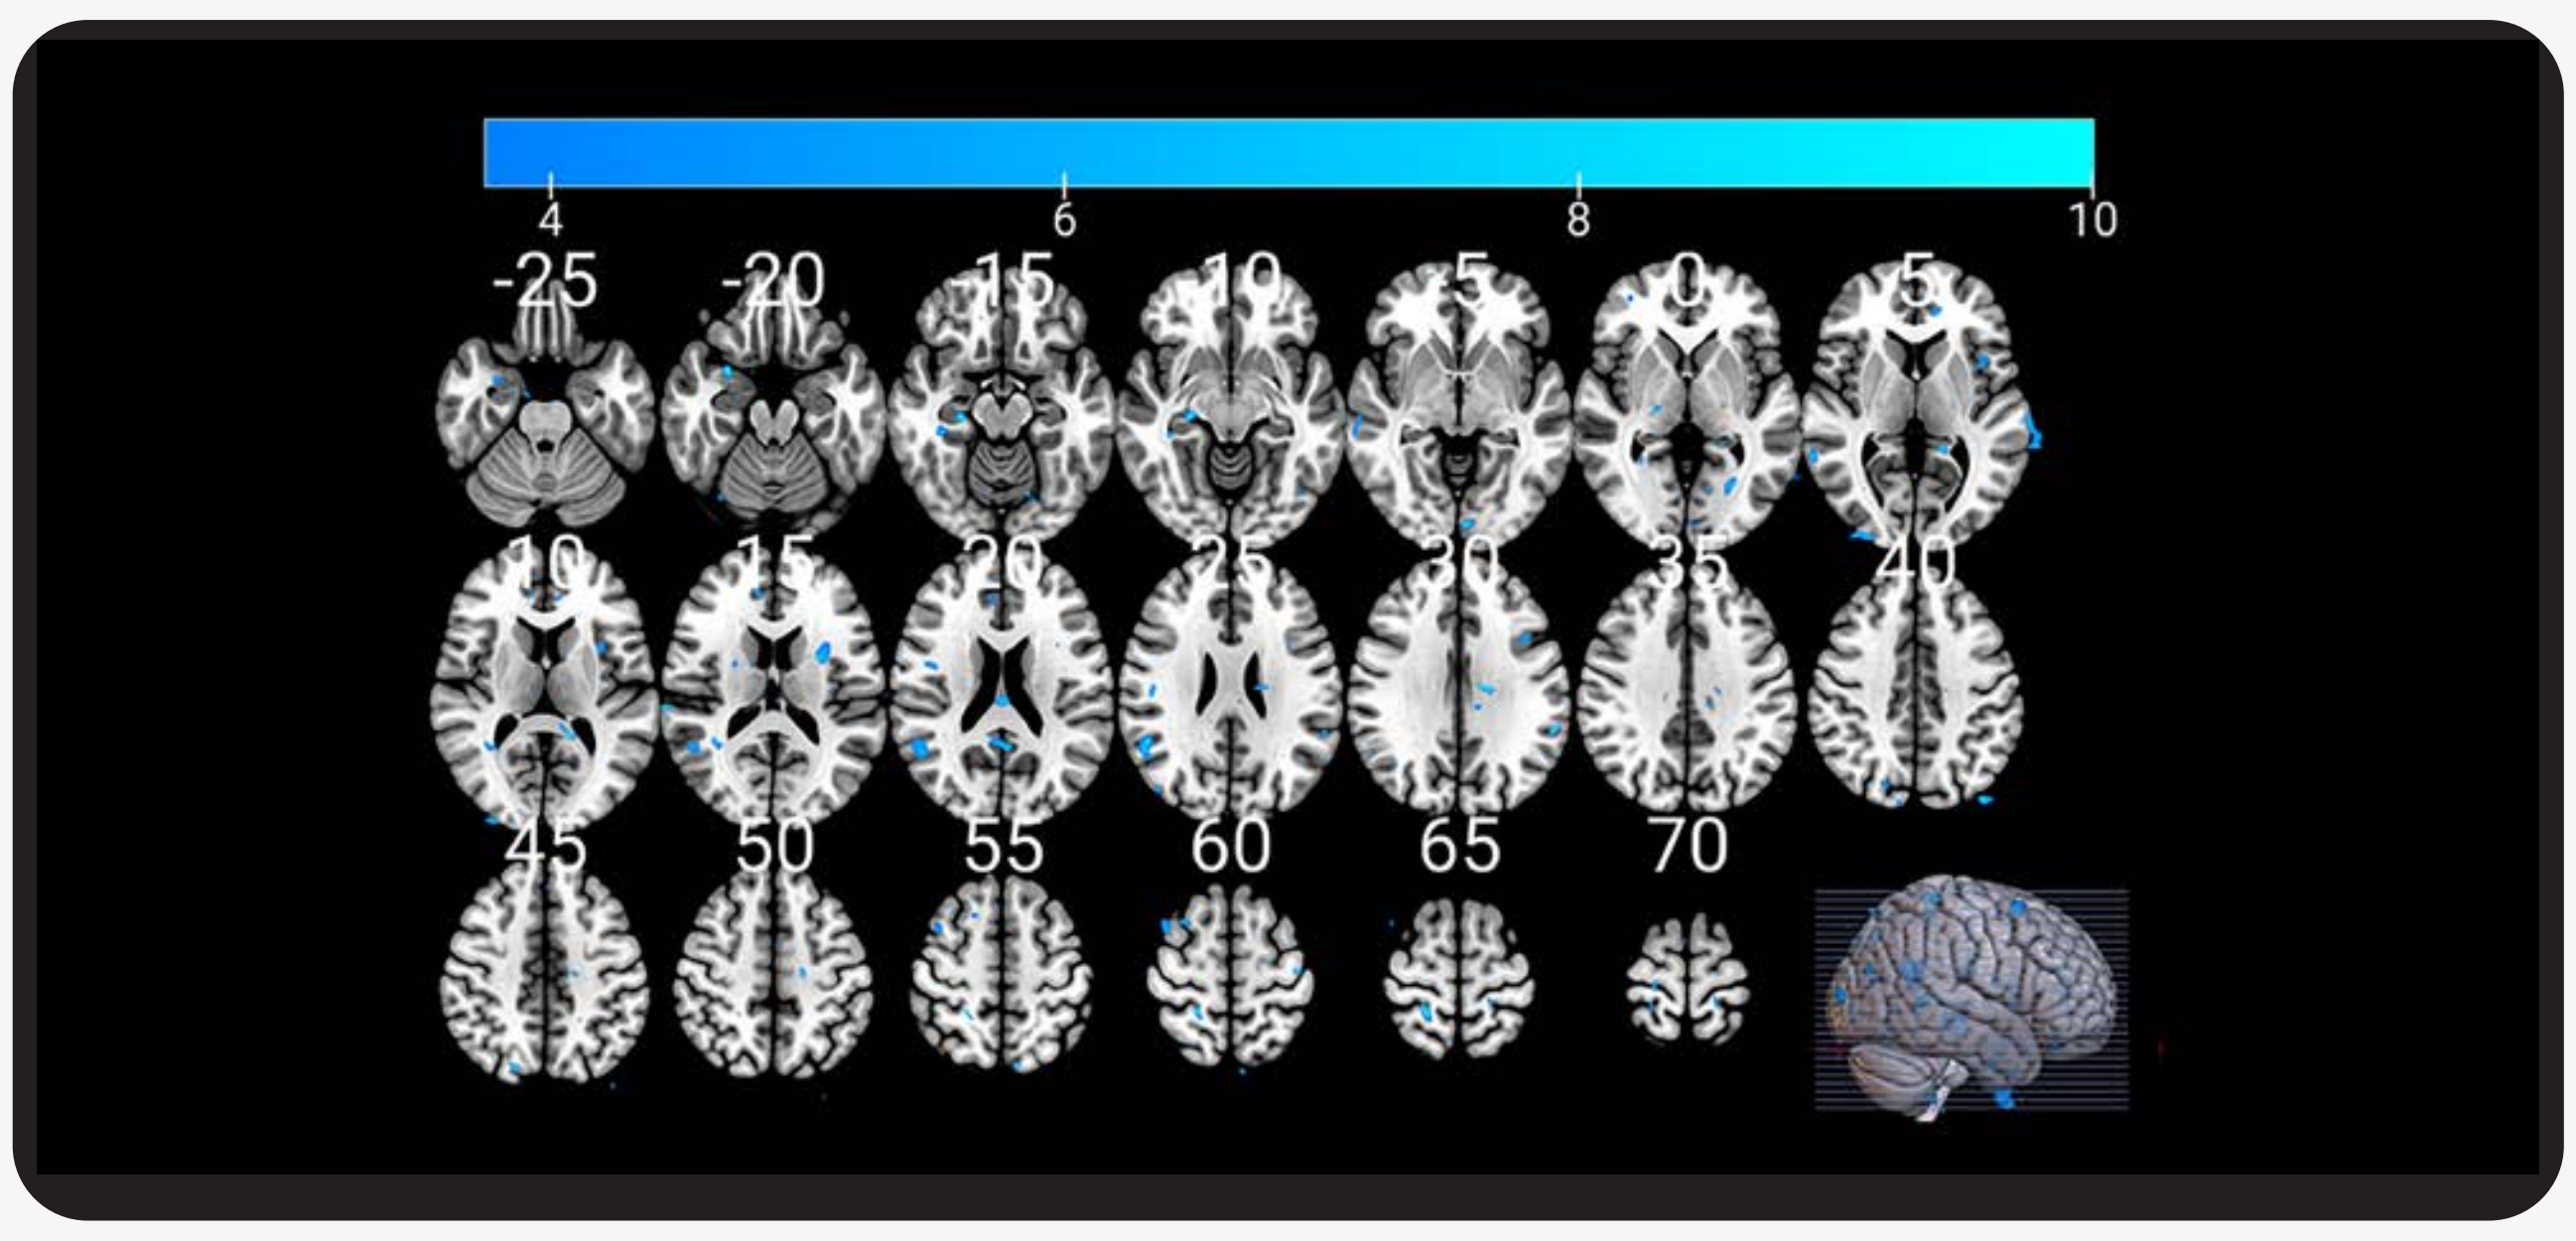
4 6 8 10
-25 -20 -15 -10 -5 0 5
10 15 20 25 30 35 40
45 50 55 60 65 70

QSM, quantitative susceptibility mapping.

EFFECT OF OFATUMUMAB ON B-CELLS

- CD-19+ B-cells decreased significantly after 1 week of ofatumumab treatment

EFFECT OF OFATUMUMAB ON SERUM BIOMARKERS

- At 9 months, ofatumumab treatment significantly reduced serum NfL levels from baseline while no reduction was observed in serum GFAP levels (**Figure 4**)

Figure 4. Effect of Ofatumumab on Serum Biomarkers

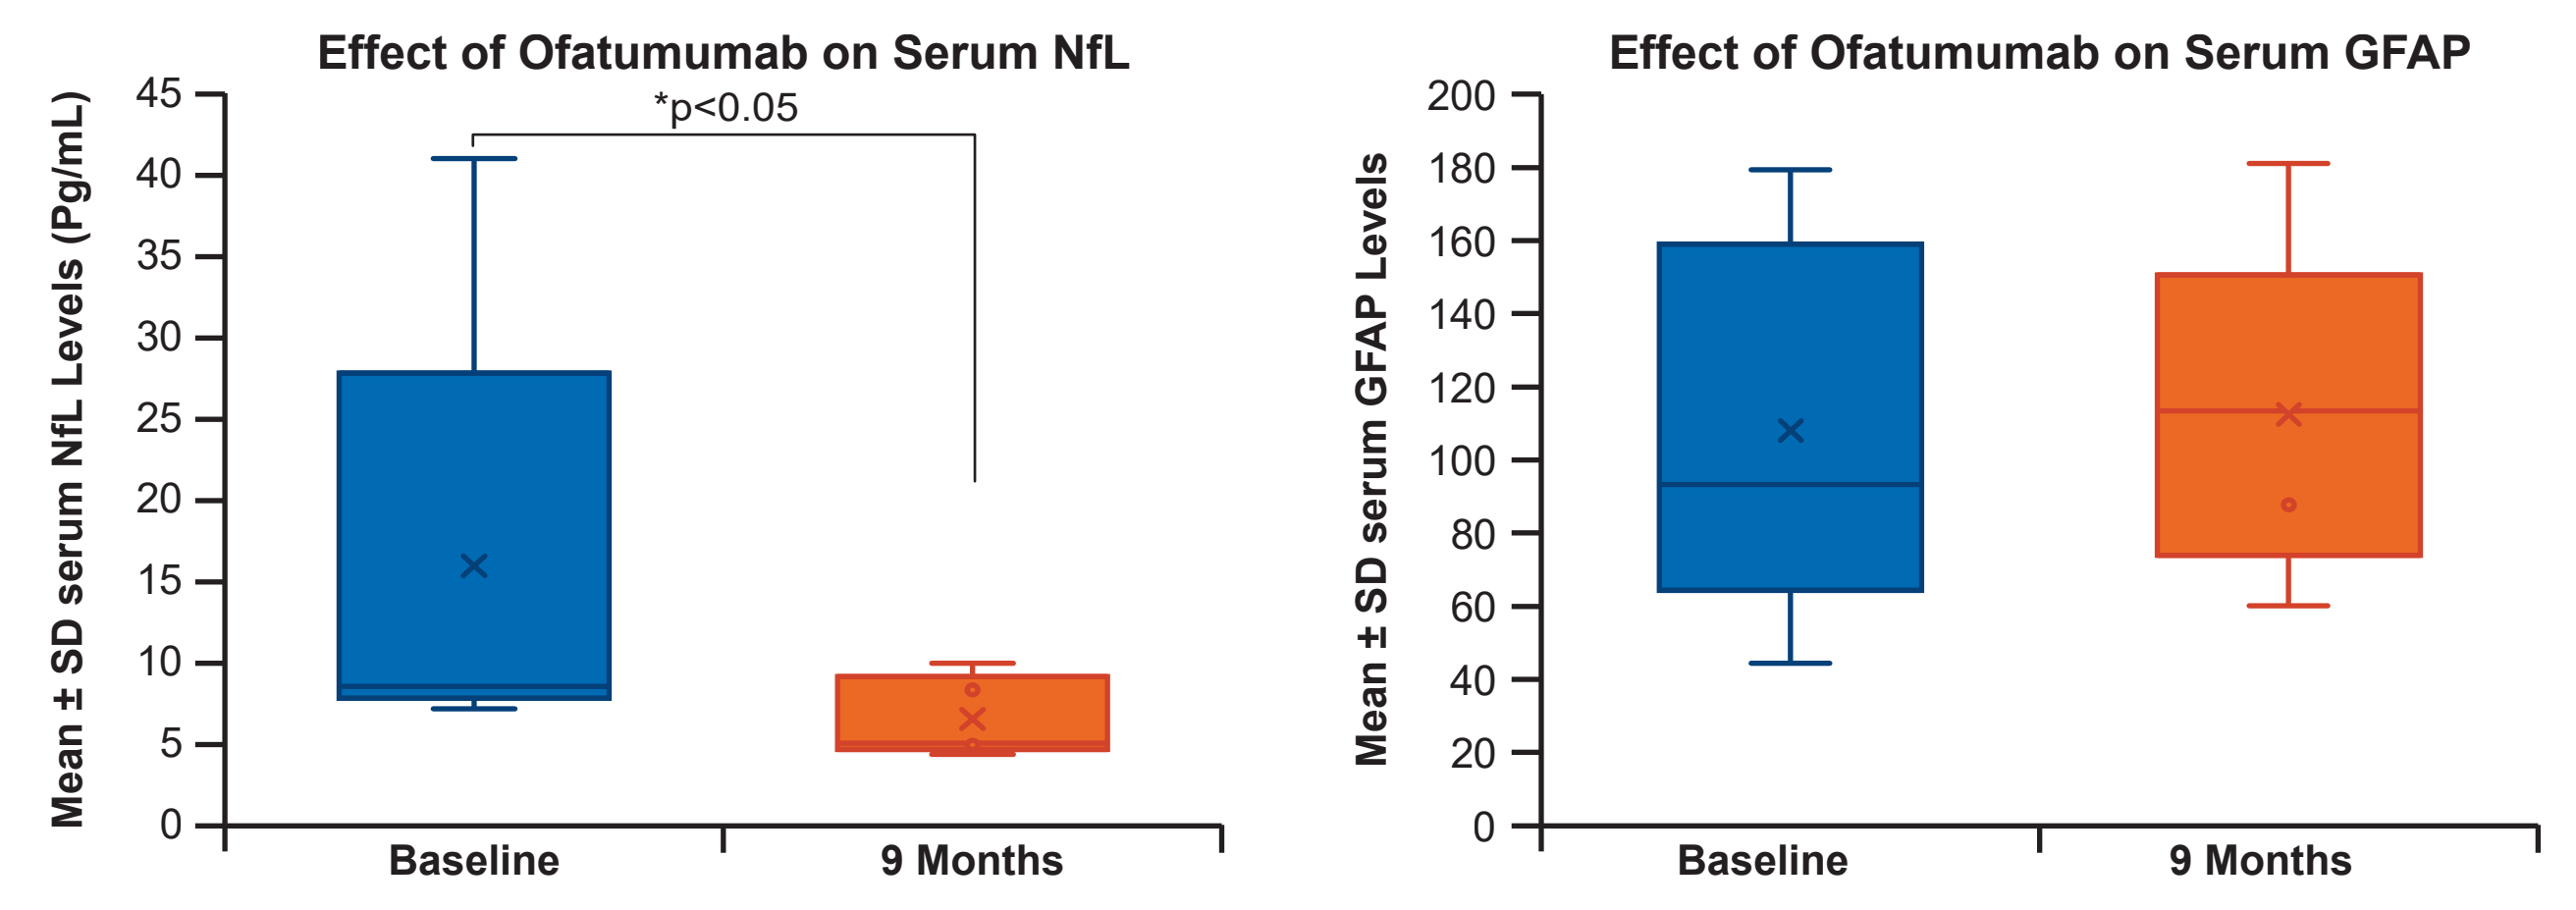
Effect of Ofatumumab on Serum NfL
*p<0.05
Mean ± SD serum NfL Levels (Pg/mL)
0 5 10 15 20 25 30 35 40 45
Baseline
9 Months
Effect of Ofatumumab on Serum GFAP
Mean ± SD serum GFAP Levels
0 20 40 60 80 100 120 140 160 180 200
Baseline
9 Months

*Significant difference from baseline; GFAP, glial fibrillary acidic protein; NfL, neurofilament light chain; SD, standard deviation.

EFFECT OF OFATUMUMAB ON CLINICAL ASSESSMENTS

- At 9 months, EDSS and T25FWT scores remained stable while hospital anxiety and depression scores decreased significantly from baseline following ofatumumab treatment (both p<0.05; **Figure 5**)

Figure 5. Effect of Ofatumumab on Anxiety and Depression Scores

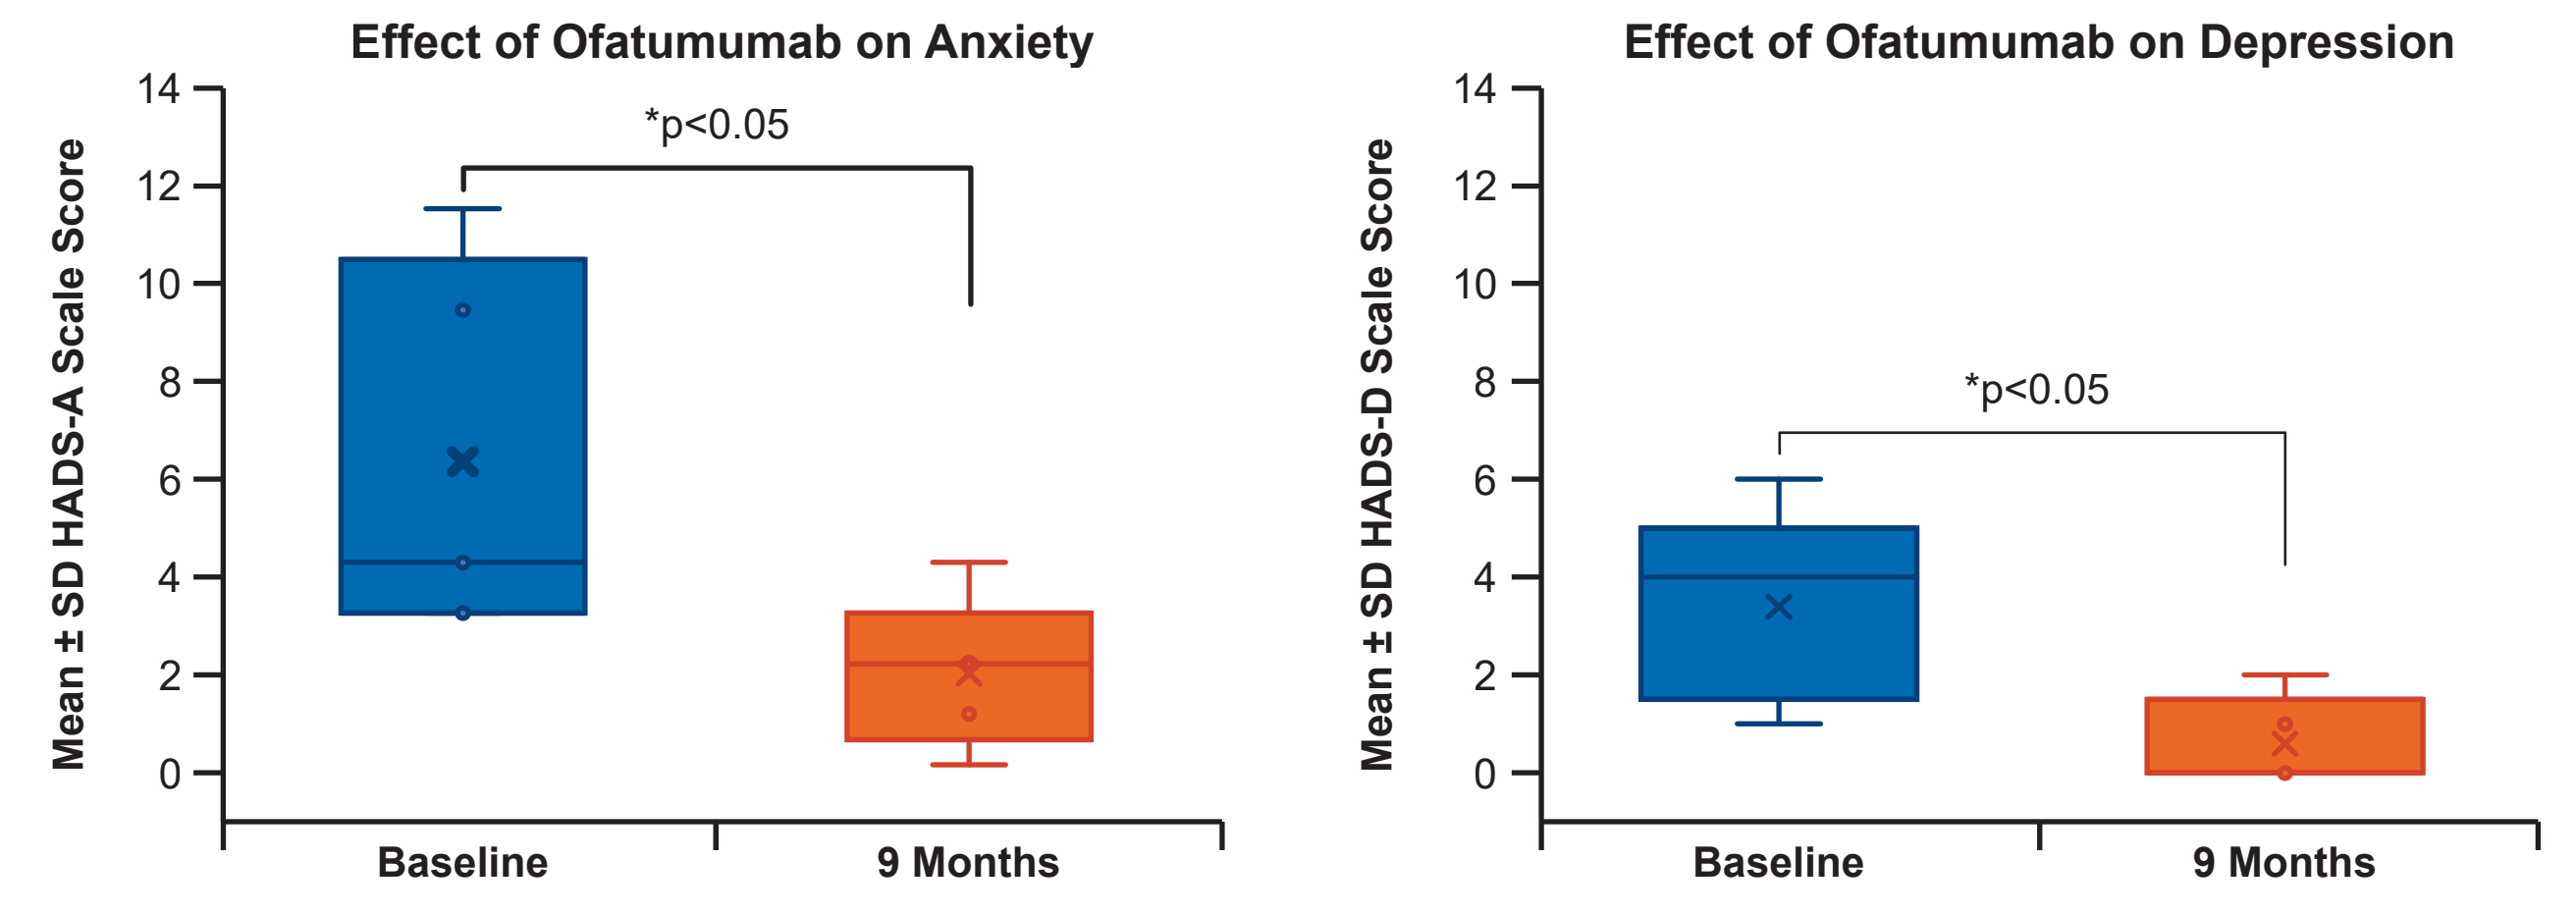
Effect of Ofatumumab on Anxiety
*p<0.05
Mean ± SD HADS-A Scale Score
0 2 4 6 8 10 12 14
Baseline
9 Months
Effect of Ofatumumab on Depression
*p<0.05
Mean ± SD HADS-D Scale Score
0 2 4 6 8 10 12 14
Baseline
9 Months

*Significant difference from baseline; HADS, Hospital Anxiety and Depression Scale.

CONCLUSIONS

- In this study, ofatumumab treatment was associated with decreased microglial activation in CoGM and decreased brain iron and serum NfL levels at 9 months
- Depletion of peripheral CD-19+ B-cells observed prior to microglial activation may suggest an indirect, downstream effect of B-cell depletion on microglial activity in patients with relapsing MS
- Further research is needed to examine the correlation among alterations in cortical microglial activation, brain iron, and serum NfL in response to ofatumumab

References: 1. Luo C, et al. *Neuropsychiatr Dis Treat*. 2017;13:1661-7; **2.** Nutma E, et al. *Glia*. 2021;69(10):2447-2458; **3.** Singhal T, et al. *Neurol Neuroimmunol Neuroinflamm*. 2019;6:e587; **4.** Dal-Bianco A, et al. *Acta Neuropathol*. 2017;133(1):25-42; **5.** Bagnato F, et al. *Brain*. 2011;134(12):3602-15; **6.** Anthony DC, et al. *Ann Clin Transl Neurol*. 2014;1:659–669.

Abbreviations: CoGM, cortical grey matter; **d**, day; **EDSS**, Expanded Disability Status Scale; **GFAP**, glial fibrillary acidic protein; **HADS**, Hospital Anxiety and Depression Scale; **m**, month; **MS**, multiple sclerosis; **NAWM**, normal-appearing white matter; **NfL**, neurofilament light chain; **PET**, positron emission tomography; **QSM**, quantitative susceptibility mapping; **ROI**, region of interest; **s.c.**, subcutaneous; **SPM**, statistical parametric mapping; **T25FWT**, timed 25-foot-walk test

Disclosures: Tarun Singhal has received research support from Novartis Pharmaceuticals and Genzyme-Sanofi, consulting fees from Novartis Pharmaceuticals and Genentech, speaking fees from Tiziana Life Sciences, and research funding from Nancy Davis Foundation's "Race to Erase MS" program, Ann Romney Center for Neurologic Diseases, Harvard Neuro-Discovery Center, National Multiple Sclerosis Society, Department of Defense, and Water Cove Charitable Foundation. **Ferdinand Schweser**, **Steven Vaquerano**, **John Hunter Ficke**, **Preksha Kukreja**, **Hong Pan**, **Kelsey Carter**, **Gauruv Bose**, **Kristin Galetta**, **Bonnie Glanz**, **Jonathan Zurawski**, **Steven Cicero**, **Maria K Houtchens**, **Christopher Severson, Shipra Dubey**, and **Bo Yeun Yang** have nothing to disclose. **Eero Rissanen** has received a fellowship grant from the Sigrid Jusélius Foundation and research grant from the Sakari Alhopuro Foundation. **Rohit Bakshi** has received consulting fees from Bristol-Myers Squibb and EMD Serono and research support from Bristol-Myers Squibb, EMD Serono and Novartis. **Tanuja Chitnis** has received compensation for consulting from Biogen, Novartis Pharmaceuticals, Roche Genentech and Sanofi Genzyme. She has received research support from the National Institutes of Health, National MS Society, US Department of Defense, Sumaira Foundation, Brainstorm Cell Therapeutics, EMD Serono, I-MAB Biopharma, Mallinckrodt ARD, Novartis Pharmaceuticals, Octave Bioscience, Roche Genentech and Tiziana Life Sciences. Disclosures do not conflict with the work being presented. **Brandon Brown** and **Ibolya Boer** are employees of Novartis. **Howard Weiner** has received research support from Cure Alzheimer's Fund, EMD Serono, Inc., Genentech, Inc., National Institutes of Health, National Multiple Sclerosis Society, Sanofi Genzyme, and Verily Life Sciences. He has received payment for consulting from Genentech, Inc, IM Therapeutics, I-MAB Biopharma, MedDay Pharmaceuticals, Tiziana Life Sciences and vTv Therapeutics.

Acknowledgements: This study was funded by Novartis Pharma AG, Basel, Switzerland. Medical writing support was provided by Venkateswarlu Bonala and Sreelatha Komatireddy and design support by V.S.Hari Prasad, all of Novartis Healthcare Pvt. Ltd., Hyderabad, India. The final responsibility for the content lies with the authors.

Copyright © 2023 Novartis Pharma AG. All rights reserved.

Scan this QR code to download a copy of the Poster

Visit the web at:
https://bit.ly/ectrims2023.

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Presenter email address:
tsinghal@bwh.harvard.edu