Presented at 38th Congress of the European Committee for Treatment and Research in Multiple Sclerosis • 26-28 October 2022 • Amsterdam, The Netherlands

P1211

Open-label, Multicentre, Phase 4 Study Assessing Immune Response to Influenza Vaccine in Patients With Relapsing Multiple Sclerosis Treated With Ofatumumab: Interim Results

Brian Steingo,[1] Adnan Subei,[2] Mark J. Tullman,[3] Jeffrey Gitt,[4] Elisabeth Lucassen,[5] James Stankiewicz,[5] Xiangyi Meng,[5] Bianca Weinstock-Guttman[6]

[1]Infinity Clinical Research, Sunrise, FL, USA; [2]Memorial Healthcare System, Hollywood, FL, USA; [3]Mercy Clinic, Washington, MO, USA; [4]Center for Neurology and Spine, Phoenix, AZ, USA; [5]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [6]Jacobs School of Medicine and Biomedical Sciences, University at Buffalo, The State University of New York, Buffalo, NY, USA

SUMMARY

1 **The effect of ofatumumab (OMB) treatment on the humoral immune response to influenza vaccination in patients with RMS is being assessed in an ongoing 3-cohort, open-label, multicentre, prospective, phase 4 study**

2 **The findings from the present interim analysis suggest that OMB-treated patients with RMS are likely able to mount an immune response following inactivated influenza vaccination and will help to inform the coordination of vaccination with OMB treatment in patients with RMS**

3 **These interim observations are consistent with current European and US product labelling with respect to administration of inactivated vaccines[1,2]; additional data available on completion of this study may provide further insights in this area**

INTRODUCTION

- Ofatumumab (OMB; Kesimpta®) is a fully human anti-CD20 monoclonal antibody administered by monthly subcutaneous injection (20 mg in 0.4 mL)[1,2]
- OMB is approved for the treatment of adults with relapsing multiple sclerosis (RMS) in the European Union and United States[1,2]
- Vaccinations comprise an important component of MS management and there is a need for data regarding whether or not treatment with OMB impacts the humoral immune response to vaccines, including the influenza vaccine, in patients with RMS

OBJECTIVE

- To report the interim results of a study assessing whether patients with RMS treated with OMB 20 mg every 4 weeks could mount a humoral immune response to the 2020-2021 or 2021-2022 inactivated influenza vaccines

METHODS

STUDY DESIGN

- An ongoing 3-cohort, open-label, multicentre, prospective, phase 4 study (NCT04667117; **Figure 1**)
 - Cohort 1: vaccinated ≥2 weeks before starting OMB
 - Cohort 2: vaccinated ≥4 weeks after starting OMB
 - Cohort 3: vaccinated ≥4 weeks after starting interferon or glatiramer acetate
- Cohort 1 received OMB 2, 3 and 4 weeks after vaccination, followed by monthly doses from Week 6; Cohorts 2 and 3 continued OMB or other disease-modifying therapy (DMT) per their current dosing schedule
- Patients in all cohorts underwent humoral immunity titre evaluations before vaccination and 4 weeks (Week 4) post vaccination
- Patients in Cohorts 1 and 2 could continue receiving monthly doses of OMB during an optional extension period and take monthly doses of OMB
- Patients with recent infections or who had been or were being treated with certain immunosuppressive or immunomodulatory therapies were excluded from the trial

STUDY ENDPOINTS

- **Primary endpoint:**
 - Patients achieving seroprotection to influenza at Week 4 (defined as a post-vaccination antibody titre ≥40)
- **Secondary endpoints:**
 - Achieving seroconversion (defined as post-vaccination humoral immunity titres ≥4-fold increase or ≥40 in those with pre-vaccination titres ≥10 or <10, respectively)
 - Safety (any adverse events [AEs], serious AEs [SAEs] and AEs leading to discontinuation)

Figure 1. Study Design

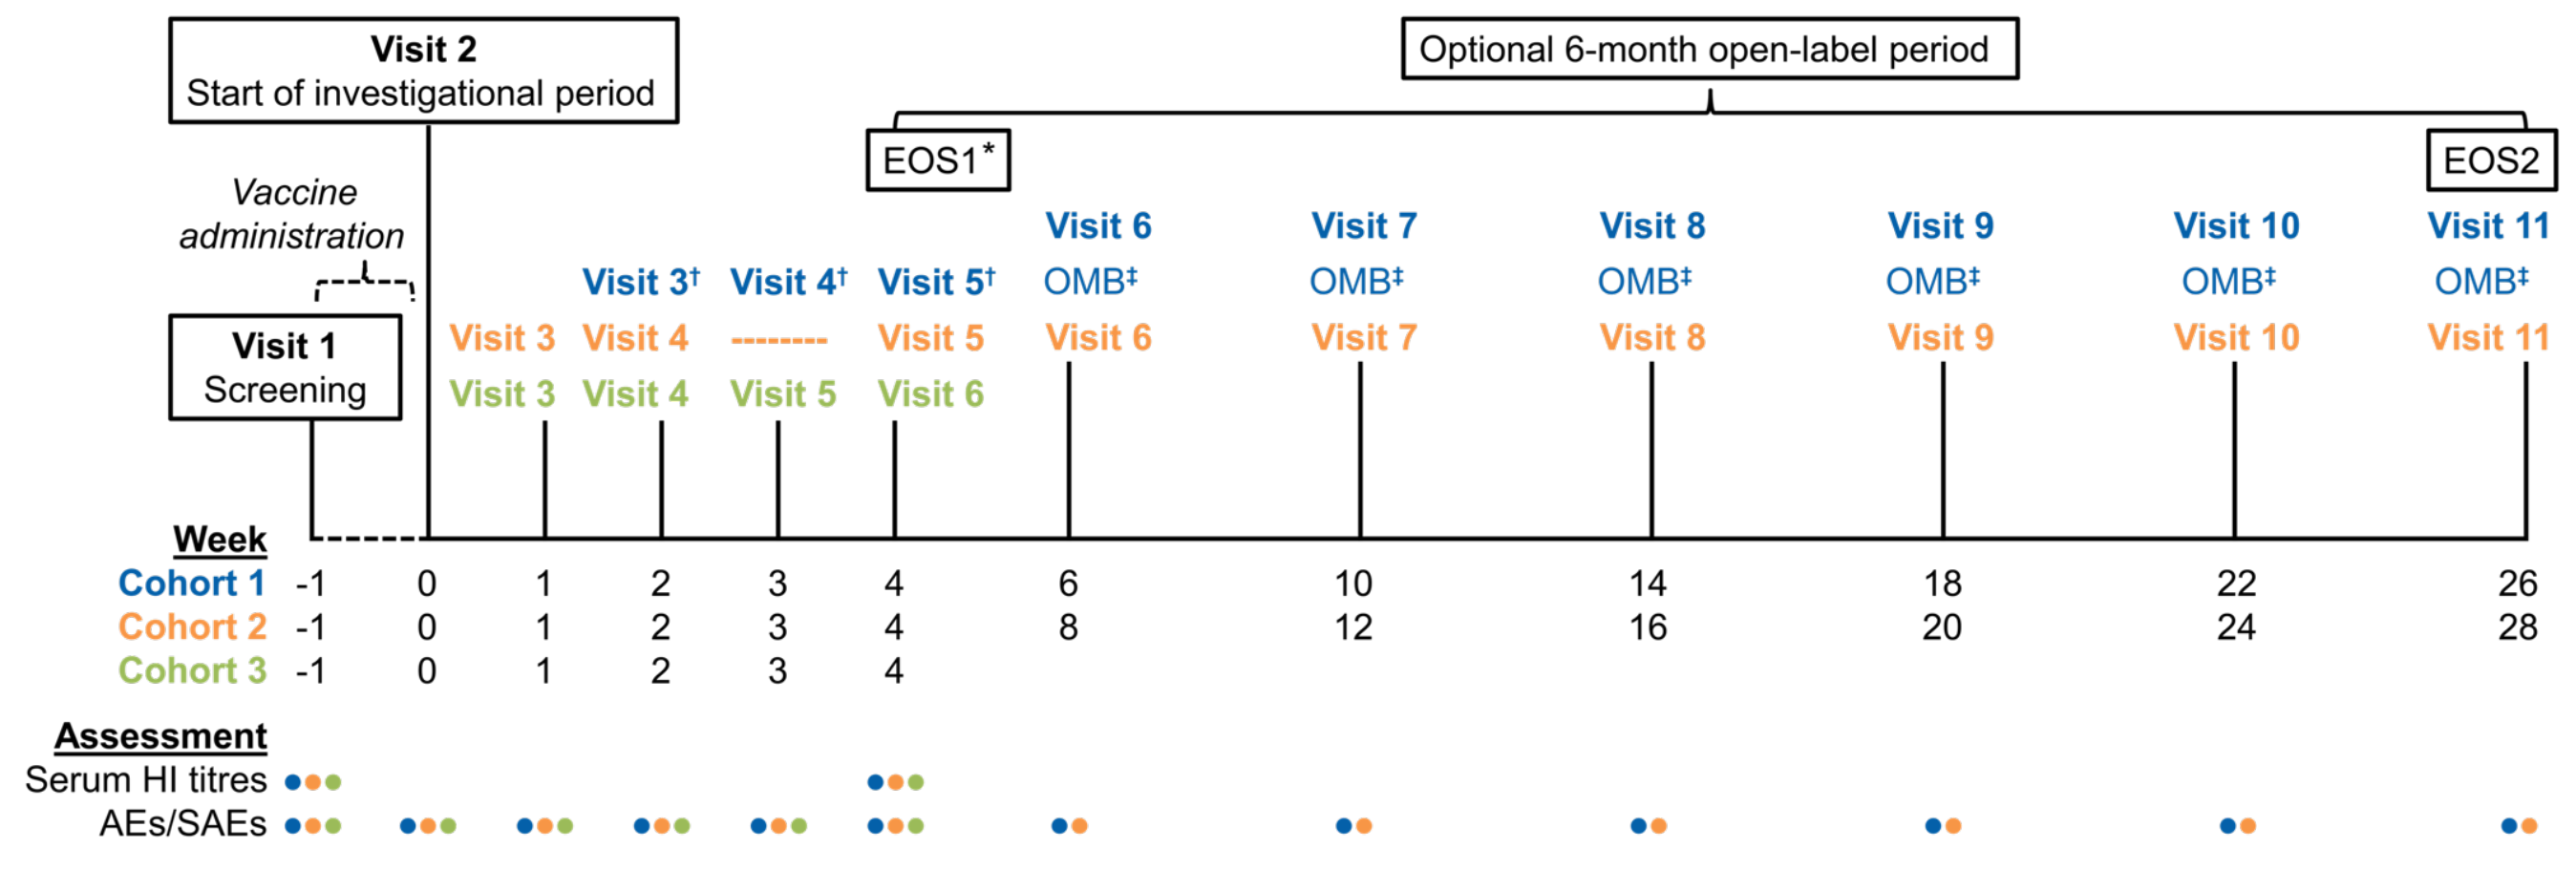
Visit 2
Start of investigational period
Optional 6-month open-label period
EOS1*
EOS2
Vaccine administration
Visit 1
Screening
Visit 3 Visit 3
Visit 3† Visit 4 Visit 4
Visit 4† -------- Visit 5
Visit 5† Visit 5 Visit 6
Visit 6 OMB‡ Visit 6
Visit 7 OMB‡ Visit 7
Visit 8 OMB‡ Visit 8
Visit 9 OMB‡ Visit 9
Visit 10 OMB‡ Visit 10
Visit 11 OMB‡ Visit 11
Week
Cohort 1 -1 0 1 2 3 4 6 10 14 18 22 26
Cohort 2 -1 0 1 2 3 4 8 12 16 20 24 28
Cohort 3 -1 0 1 2 3 4
Assessment
Serum HI titres
AEs/SAEs

AE, adverse event; EOS, end of study; HI, haemagglutination inhibition; OMB, ofatumumab; SAE, serious adverse event
*Participants in Cohort 3 will not enter the extension; †Weekly OMB loading dose; ‡Monthly OMB dose

RESULTS

PATIENT DEMOGRAPHICS AND DISPOSITION

- This interim analysis included 39 patients with a mean age of 41 years (range, 22-53)
- Of these patients, 77% (30/39) were female, 90% (35/39) were White and 44% (17/39) had used a previous DMT for MS treatment (**Table 1**)

Table 1. Patient Demographics and Clinical Characteristics at Screening

Characteristic	Cohort 1 (n=22)	Cohort 2 (n=7)	Cohort 3 (n=10)	Overall (N=39)
Age, years				
Mean (range)	41.4 (24-53)	38.4 (22-53)	41.3 (27-51)	40.8 (22-53)
Sex, n (%)				
Female	19 (86.4)	3 (42.9)	8 (80.0)	30 (76.9)
Male	3 (13.6)	4 (57.1)	2 (20.0)	9 (23.1)
Race, n (%)				
White	21 (95.5)	7 (100.0)	7 (70.0)	35 (89.7)
Black or African American	1 (4.6)	0	3 (30.0)	4 (10.3)
Ethnicity, n (%)				
Hispanic or Latino	6 (27.3)	2 (28.6)	0	8 (20.5)
Not Hispanic or Latino	16 (72.7)	5 (71.4)	10 (100.0)	31 (79.5)
Any previous MS DMT, n (%)	0	7 (100.0)	10 (100.0)	17 (43.6)
Glatiramer acetate	0	0	8 (80.0)	8 (20.5)
Interferon beta-1a	0	0	1 (10.0)	1 (2.6)
Ofatumumab	0	7 (100.0)	0	7 (18.0)
Peginterferon beta-1a	0	0	1 (10.0)	1 (2.6)

DMT, disease-modifying therapy; MS, multiple sclerosis

SEROPROTECTION AT WEEK 4

- Overall influenza seroprotection at Week 4 was achieved in 64%, 67% and 69% of patients in Cohort 1 (n=22), Cohort 2 (n=7) and Cohort 3 (n=10), respectively
- From baseline to Week 4, seroprotection tended to increase after influenza vaccination, although there was variation across influenza strains (**Table 2**). Seroprotection at Week 4 among commonly tested strains was as follows:
 - Influenza A Cambodia: 87% (13/15) in Cohort 1, 80% (4/5) in Cohort 2 and 67% (6/9) in Cohort 3
 - Influenza A Victoria: 87% (13/15), 100% (5/5) and 78% (7/9)
 - Influenza A Wisconsin: 53% (8/15), 60% (3/5) and 67% (6/9)
 - Influenza B Phuket: 64% (14/22), 86% (6/7) and 60% (6/10)

SEROCONVERSION AT WEEK 4

- Overall seroconversion at Week 4 was seen in 45%, 10% and 33% of patients in Cohort 1, Cohort 2 and Cohort 3, respectively
- The rates of seroconversion at Week 4 were variable across vaccine influenza strains (**Table 3**); however, trends for lower rates of seroconversion in Cohort 2 vs Cohorts 1 and 3 were broadly consistent across individual influenza A and B strains. Seroconversion among commonly tested strains was as follows:
 - Influenza A Cambodia: 73% (11/15) in Cohort 1, 20% (1/5) in Cohort 2 and 33% (3/9) in Cohort 3
 - Influenza A Victoria: 80% (12/15), 20% (1/5) and 33% (3/9)
 - Influenza A Wisconsin: 40% (6/15), 20% (1/5) and 44% (4/9)
 - Influenza B Phuket: 41% (9/22), 14% (1/7) and 20% (2/10)

Table 2. Percentages of Patients With Seroprotection at Baseline and Week 4 by Influenza Strain

Strain	% of Patients With Seroprotection					
	Cohort 1 (n=22)		Cohort 2 (n=7)		Cohort 3 (n=10)	
	Baseline	Week 4	Baseline	Week 4	Baseline	Week 4
Influenza A Brisbane	88.9 (8/9)	55.6 (5/9)	100.0 (3/3)	66.7 (2/3)	100.0 (1/1)	100.0 (1/1)
Influenza A Cambodia	20.0 (3/15)	86.7 (13/15)	60.0 (3/5)	80.0 (4/5)	55.6 (5/9)	66.7 (6/9)
Influenza A Kansas	100.0 (9/9)	55.6 (5/9)	100.0 (3/3)	66.7 (2/3)	100.0 (1/1)	100.0 (1/1)
Influenza A Michigan	88.9 (8/9)	55.6 (5/9)	100.0 (3/3)	66.7 (2/3)	100.0 (1/1)	100.0 (1/1)
Influenza A Singapore	88.9 (8/9)	55.6 (5/9)	100.0 (3/3)	66.7 (2/3)	100.0 (1/1)	100.0 (1/1)
Influenza A Victoria	60.0 (9/15)	86.7 (13/15)	100.0 (5/5)	100.0 (5/5)	88.9 (8/9)	77.8 (7/9)
Influenza A Wisconsin	26.7 (4/15)	53.3 (8/15)	40.0 (2/5)	60.0 (3/5)	55.6 (5/9)	66.7 (6/9)
Influenza B Colorado	11.1 (1/9)	33.3 (3/9)	66.7 (2/3)	33.3 (1/3)	0 (0/1)	100.0 (1/1)
Influenza B Phuket	13.6 (3/22)	63.6 (14/22)	85.7 (6/7)	85.7 (6/7)	70.0 (7/10)	60.0 (6/10)
Influenza B Washington	33.3 (5/15)	66.7 (10/15)	20.0 (1/5)	20.0 (1/5)	44.4 (4/9)	55.6 (5/9)

Table 3. Percentages of Patients Achieving Seroconversion at Week 4 by Influenza Strain

Strain	% of Patients Achieving Seroconversion		
	Cohort 1 (n=22)	Cohort 2 (n=7)	Cohort 3 (n=10)
Influenza A Brisbane	44.4 (4/9)	0 (0/3)	0 (0/1)
Influenza A Cambodia	73.3 (11/15)	20.0 (1/5)	33.3 (3/9)
Influenza A Kansas	22.2 (2/9)	0 (0/3)	0 (0/1)
Influenza A Michigan	33.3 (3/9)	0 (0/3)	100.0 (1/1)
Influenza A Singapore	22.2 (2/9)	0 (0/3)	0 (0/1)
Influenza A Victoria	80.0 (12/15)	20.0 (1/5)	33.3 (3/9)
Influenza A Wisconsin	40.0 (6/15)	20.0 (1/5)	44.4 (4/9)
Influenza B Colorado	33.3 (3/9)	0 (0/3)	100.0 (1/1)
Influenza B Phuket	40.9 (9/22)	14.3 (1/7)	20.0 (2/10)
Influenza B Washington	33.3 (5/15)	0 (0/5)	33.3 (3/9)

SAFETY

- Overall, 36% (14/39) of patients experienced ≥1 AE (**Table 4**)
- AEs were most frequent in Cohort 1, where 55% (12/22) patients experienced an AE vs 1 each in Cohorts 2 and 3 (14% [1/7] and 10% [1/10], respectively)
- The higher frequency of AEs in Cohort 1, which included injection-related reactions (IRRs) not reported in Cohorts 2 and 3, is likely related to OMB initiation
 - IRRs were reported in 4 patients (Grade 2, n=3; Grade 1, n=1), beginning with the first OMB injection
- No SAEs or AEs resulting in discontinuation were reported

Table 4. AEs That Occurred in ≥5% of Patients

AE, n (%)	Cohort 1 (n=22)	Cohort 2 (n=7)	Cohort 3 (n=10)	Overall (N=39)
Any AE	12 (55)	1 (14)	1 (10)	14 (36)
Injection-related reaction	4 (18)	0	0	4 (10)
Headache	3 (14)	0	0	3 (8)
Nausea	3 (14)	0	0	3 (8)
Chills	2 (9)	0	0	2 (5)
Fatigue	2 (9)	0	0	2 (5)
Pain	2 (9)	0	0	2 (5)
Hypoaesthesia	1 (4.6)	0	1 (10)	2 (5)
COVID-19	0	1 (14)	1 (10)	2 (5)

AE, adverse event; COVID-19, coronavirus disease 2019

ABBREVIATIONS: AE, adverse event; **COVID-19**, coronavirus disease 2019; **DMT**, disease-modifying therapy; **EOS**, end of study; **HI**, haemagglutination inhibition; **IRR**, injection-related reaction; **MS**, multiple sclerosis; **OMB**, ofatumumab; **RMS**, relapsing multiple sclerosis; **SAE,** serious adverse event. **ACKNOWLEDGEMENTS:** The study was supported by Novartis Pharmaceuticals Corporation. Medical writing support was provided by Nancy Nguyen, PharmD, of Envision Pharma Group and was funded by Novartis Pharmaceuticals Corporation. This poster was developed in accordance with Good Publication Practice (GPP3) guidelines. Authors had full control of the content and made the final decision on all aspects of this poster. **DISCLOSURES: Brian Steingo** has received honoraria and payments for research, speaking engagements and advisory boards from Biogen, Bristol Myers Squibb, EMD Serono, Genentech, Janssen, Novartis and Sanofi. **Adnan Subei** has received consulting fees from Biogen; and has received research support from Novartis. **Mark J. Tullman** has received consulting fees, research support and/or speaking honoraria from Banner Life Sciences, Biogen, Bristol Myers Squibb, EMD Serono, Genentech, Genzyme, Horizon Therapeutics, Novartis and TG Therapeutics. **Jeffrey Gitt** has received honoraria for promotional programmes from Alexion, Allergan, Bristol Myers Squibb and EMD Serono. **Elisabeth Lucassen**, **James Stankiewicz** and **Xiangyi Meng** are employees of and stockholders in Novartis Pharmaceuticals Corporation. **Bianca Weinstock-Guttman** has received consulting fees from Biogen, Celgene, EMD Serono, Genentech and Janssen; and has received research support from Biogen, Celgene, EMD Serono, Genentech and Novartis

REFERENCES: 1. Novartis Pharmaceuticals Corporation. Prescribing information. Kesimpta® 2020. Accessed August 25, 2022. www.novartis.us/sites/www.novartis.us/files/kesimpta.pdf; **2.** European Medicines Agency. Summary of product characteristics. Kesimpta 2021. Accessed August 25, 2022. www.ema.europa.eu/en/documents/product-information/kesimpta-epar-product-information_en.pdf

Scan to download a copy of this poster
Copies of this poster and its content, obtained through this QR code, are for personal use only and may not be reproduced without written permission from the authors