Longer-term Safety and Efficacy of Ofatumumab in Recently Diagnosed and Treatment Naïve Patients is Consistent with the Overall Population in the ALITHIOS Open-Label Extension Study

P052

Jutta Gärtner[1], Stephen L. Hauser[2], Amit Bar-Or[3], Xavier Montalban[4], Jeffrey A. Cohen[5], Derrick Robertson[6]; Anne H. Cross[7], Carrie M. Hersh[8], Kumaran Deiva[9], Goeril Karlsson[10], Ayan Das Gupta[11], Ronald Zielman[12], Soudeh Ansari[13], Bernd Kieseier[10], Ludwig Kappos[14]

[1]Department of Paediatrics and Adolescent Medicine, Division of Paediatric Neurology, University Medical Centre Göttingen, Georg August University Göttingen, Göttingen, Germany; [2]UCSF Weill Institute for Neurosciences, University of California, San Francisco, San Francisco, CA, USA; [3]Center for Neuroinflammation and Experimental Therapeutics and Department of Neurology, Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA, USA; [4]Department of Neurology Neuroimmunology, Centre d'Esclerosi Múltiple de Catalunya (Cemcat), Hospital Universitari Vall d'Hebron, Barcelona, Spain; [5]Department of Neurology, Mellen MS Center, Neurological Institute, Cleveland Clinic, Cleveland, OH, United States; [6]Multiple Sclerosis Division, Department of Neurology, University of South Florida, Tampa, Florida, USA; [7]Department of Neurology, Section of Neuroimmunology, Washington University School of Medicine, Saint Louis, MO, USA; [8]Cleveland Clinic Lou Ruvo Center for Brain Health, Las Vegas, NV, United States; [9]Department of Pediatric Neurology, University Hospitals Paris Saclay, Hôpital Bicêtre, National Reference Center for Rare Inflammatory Brain and Spinal Diseases, Le Kremlin Bicêtre, France; [10]Novartis Pharma AG, Basel, Switzerland; [11]Novartis Healthcare Pvt. Ltd., Hyderabad, India; [12]Novartis Pharma B.V., Amsterdam, The Netherlands; [13]Novartis Institutes for Biomedical, MA, USA; [14]Research Center for Clinical Neuroimmunology and Neuroscience Basel (RC2NB) and MS Center, Departments of Head, Spine and Neuromedicine, Clinical Research, Biomedicine and Biomedical Engineering, University Hospital and University of Basel, Basel, Switzerland

Introduction

- Ofatumumab, a fully human anti-CD20 monoclonal antibody with a 20 mg subcutaneous monthly dosing regimen, is approved for treating relapsing multiple sclerosis (RMS) in adults[1]
- In Phase 3 ASCLEPIOS I/II trials, ofatumumab demonstrated superior efficacy in reducing the annualised relapse rate (ARR), suppressing MRI lesion activity and delaying disability worsening, while maintaining a favorable safety profile vs teriflunomide in RMS patients[2]
 - In the subgroup of recently diagnosed (≤3 years) and treatment-naïve (RDTN) patients, ofatumumab had a superior benefit–risk profile compared with teriflunomide, with an almost complete abrogation of inflammatory disease activity and no unexpected safety signals, supporting its use as a first-line treatment in early MS[3]
- In the ongoing, open-label, ALITHIOS extension study, ofatumumab has demonstrated well-tolerated safety and sustained longer-term efficacy for up to 4 years in the overall patient population[4,5]

Objective

- To assess the longer-term safety and efficacy of ofatumumab for up to 4 years (data cut-off: 25-Sep-2021) in a subgroup of RDTN (early RMS) patients from the ASCLEPIOS I/II (core) and continued in the ongoing ALITHIOS (extension) trial

Methods

Patient population and outcomes

- The subgroup analysis included data from patients who where recently diagnosed (within 3 years before screening), treatment-naïve (no prior DMT use), prior to enrolment into ASCLEPIOS I/II
- This analysis in early RMS (RDTN) patients comprised of 37.9% (615/1623) of overall patient population in ASCLEPIOS I/II
- Efficacy outcomes (ARR, time-to-3/6-month confirmed disability worsening [3m/6mCDW], number of Gd+T1 lesions, annualised T2 lesion rate) were analysed in two groups:
 - **Continuous group:** Patients randomised to ofatumumab in ASCLEPIOS I/II (core) and continuing ofatumumab in ALITHIOS (extension)
 - **Switch group:** Patients randomised to teriflunomide in ASCLEPIOS I/II, switched to ofatumumab in ALITHIOS
- Safety outcomes were analysed in the below 3 groups:
 - **Overall patient population:** Patients enrolled in ASCLEPIOS I/II and ALITHIOS
 - **Continuous group:** Patients randomised to ofatumumab in ASCLEPIOS I/II and continuing ofatumumab in ALITHIOS
 - **Switch group:** Patients randomised to teriflunomide in ASCLEPIOS I/II, switched to ofatumumab in ALITHIOS

Assessments

- ARR and MRI outcomes were analysed
 - Between-groups (defined as comparison of the cumulative outcomes between the continuous and switch groups) and
 - Within-groups (defined as comparison of the core and extension periods within the continuous and switch groups)
- Safety outcomes were analysed in safety analysis set which includes safety data as of the first dose of ofatumumab for all RDTN patients who received at least one dose of ofatumumab either in the ASCLEPIOS I/II or ALITHIOS trials

Results

Baseline characteristics

- Baseline characteristics of the RDTN subgroup were typical of early RMS patients and were generally balanced between treatment groups (**Table 1**)
- At baseline, mean age of patients was approximately 36 years in the continuous ofatumumab and switch groups; majority of patients were women (>65%)
- The mean EDSS at baseline was approximately 2.2 for both the continuous and switch groups

Table 1. Patient demographics and disease characteristics

Demographics and clinical characterstics[a]	Continuous ofatumumab (N=314)	Switch from teriflunomide to ofatumumab (N=301)	
	Baseline from core study (N=314)	Baseline from core study (N=301)	Baseline from extension study (N=232)
Age, years	36.8±9.40	35.7±9.03	37.7±8.99
BMI, kg/m²	25.93±6.15	26.19±6.05	25.71±5.71
Female, n (%)	69.1	64.8	66.8
Time since diagnosis, years	0.58±0.63	0.53±0.51	2.44±0.59
Time since MS symptom onset, years	3.4±3.96	3.2±4.27	5.16±4.23
EDSS score at baseline	2.30±1.198	2.28±1.203	2.17±1.237
Number of relapses in the last 12 months prior to screening	1.30±0.70	1.4±0.72	0.1±0.41
Number of Gd+ T1 lesions	1.8±4.35	1.4±2.79	0.7±2.01
Total volume of T2 lesions, cm³	10.1±12.22	8.3±8.82	NA[b]
IgG levels at baseline, g/L	10.025±2.01	-	10.27±1.99
IgM levels at baseline, g/L	1.33±0.64	-	1.36±0.691

[a]Values are represented as mean±SD unless specified otherwise; For ofatumumab newly-switched patients, their baseline values from extension study contribute to the overall summary. [b]data is not collected for baseline from extension.
BMI, body mass index; EDSS, Expanded Disability Status Scale; Gd+, gadolinium enhancing; NA, not available; SD, standard deviation.

Efficacy Outcomes

Annualised relapse rate (ARR)

- The between-group analysis over a period of up to 4 years shows that earlier initiation of ofatumumab was associated with a reduction in the cumulative number of relapses by 42%, *P*=0.0013 (**Figure 1A**)
- ARR in the continuous ofatumumab group remained low for up to 4 years after treatment initiation which resulted in an adjusted rate of 1 relapse every 20 years during the extension phase (**Figure 1B**)
- Within-group analysis showed that continuous use of ofatumumab was associated with a significant reduction in ARR by 43.1% with longer-term treatment; while switch from teriflunomide to ofatumumab resulted in a pronounced reduction in ARR (76.6%) (**Figure 1B**)

Figure 1. Between-group comparison of the cumulative number of relapses (A), and within-group comparison of adjusted ARR between the core and extension phase (B)

A Total number of relapses over up to 4 years

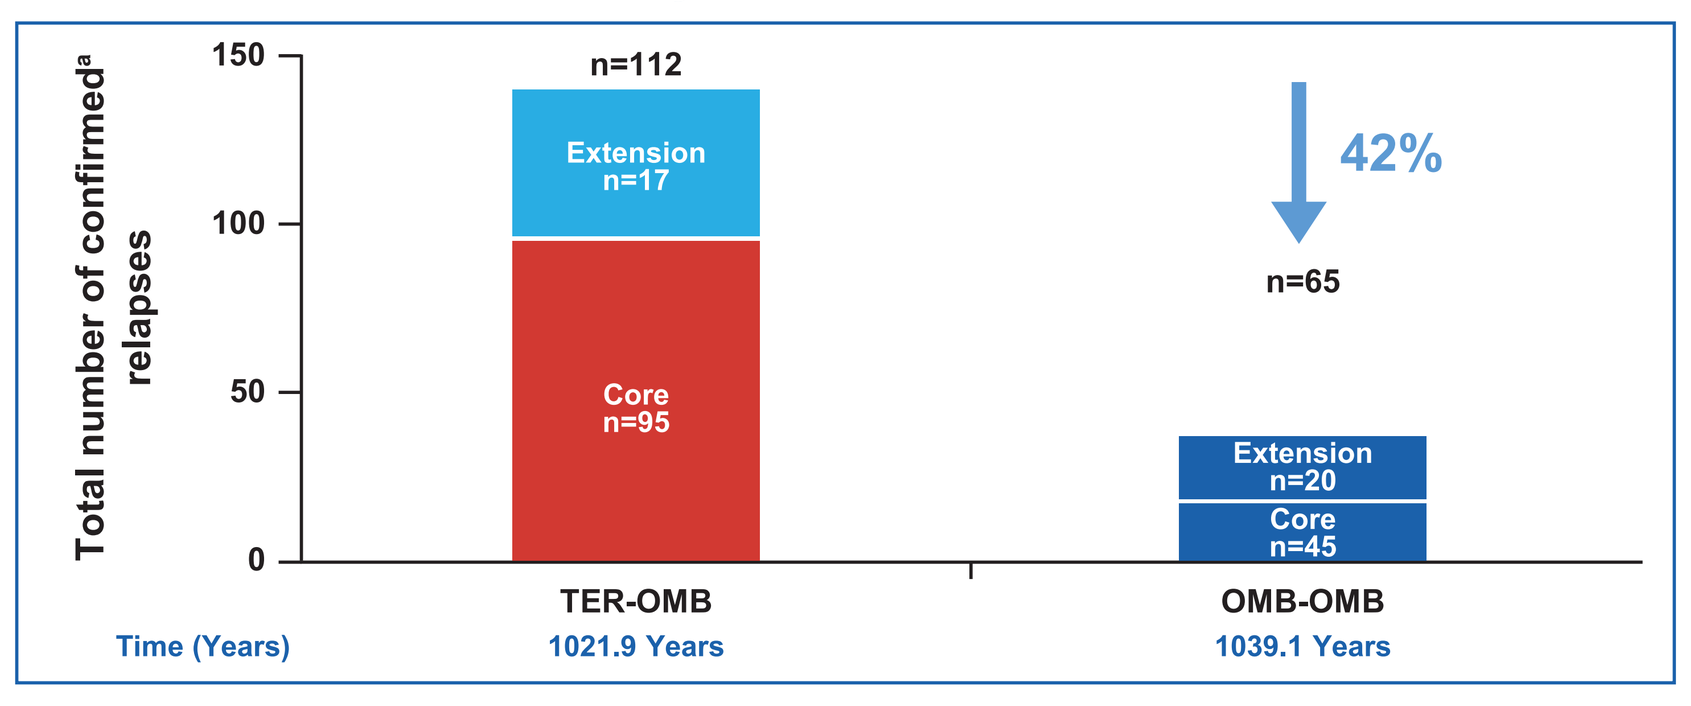

Total number of confirmed[a] relapses
n=112
Extension n=17
Core n=95
42%
n=65
Extension n=20
Core n=45
TER-OMB
OMB-OMB
Time (Years)
1021.9 Years
1039.1 Years

B Within-group comparison[b] between the core and extension phase

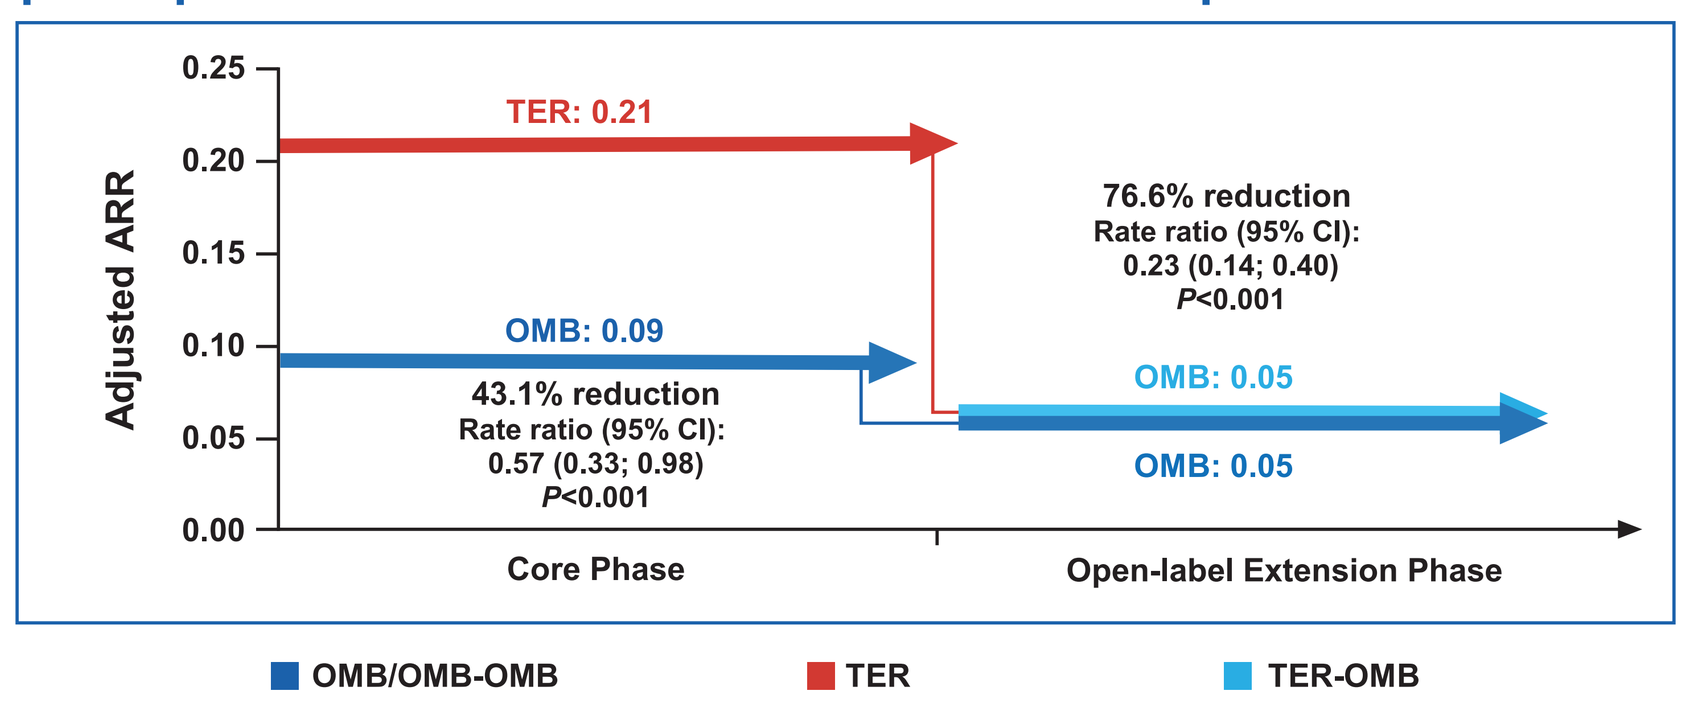

Adjusted ARR
TER: 0.21
76.6% reduction
Rate ratio (95% CI):
0.23 (0.14; 0.40)
P<0.001
OMB: 0.09
43.1% reduction
Rate ratio (95% CI):
0.57 (0.33; 0.98)
P<0.001
OMB: 0.05
OMB: 0.05
Core Phase
Open-label Extension Phase
OMB/OMB-OMB
TER
TER-OMB

[a]Confirmed relapses are those accompanied by a clinically relevant change in the EDSS; [b]Obtained from fitting a piecewise negative binomial model for the time period core phase and extension phase with log-link, adjusted for treatment and region as factors, number of relapses in previous year, baseline EDSS, baseline number of Gd-enhancing lesions and the patient's age at baseline as covariates. The natural log of the time-in-study (in years) by period is used as offset to annualise the relapse rate in each period. Baseline variables are from the core study baseline. All *P* values are nominal *P* values.
ARR, annualised relapse rate, CI, confidence interval; OMB, ofatumumab; OMB-OMB, continuous ofatumumab; TER, teriflunomide; TER-OMB, switch from teriflunomide to ofatumumab.

3- and 6-month CDW

- As shown by the delta at months 36 and 48, and the difference in the cumulative number of events over a period of up to 4 years, earlier treatment with ofatumumab was associated with an efficacy benefit that cannot be recovered in those initially randomised to teriflunomide (**Figure 2** and **Figure 3**)

Figure 2. Between-group comparison of cumulative number of 3mCDW events (A), and time to first 3mCDW (B)

A Cumulative number of first 3mCDW events over up to 4 years

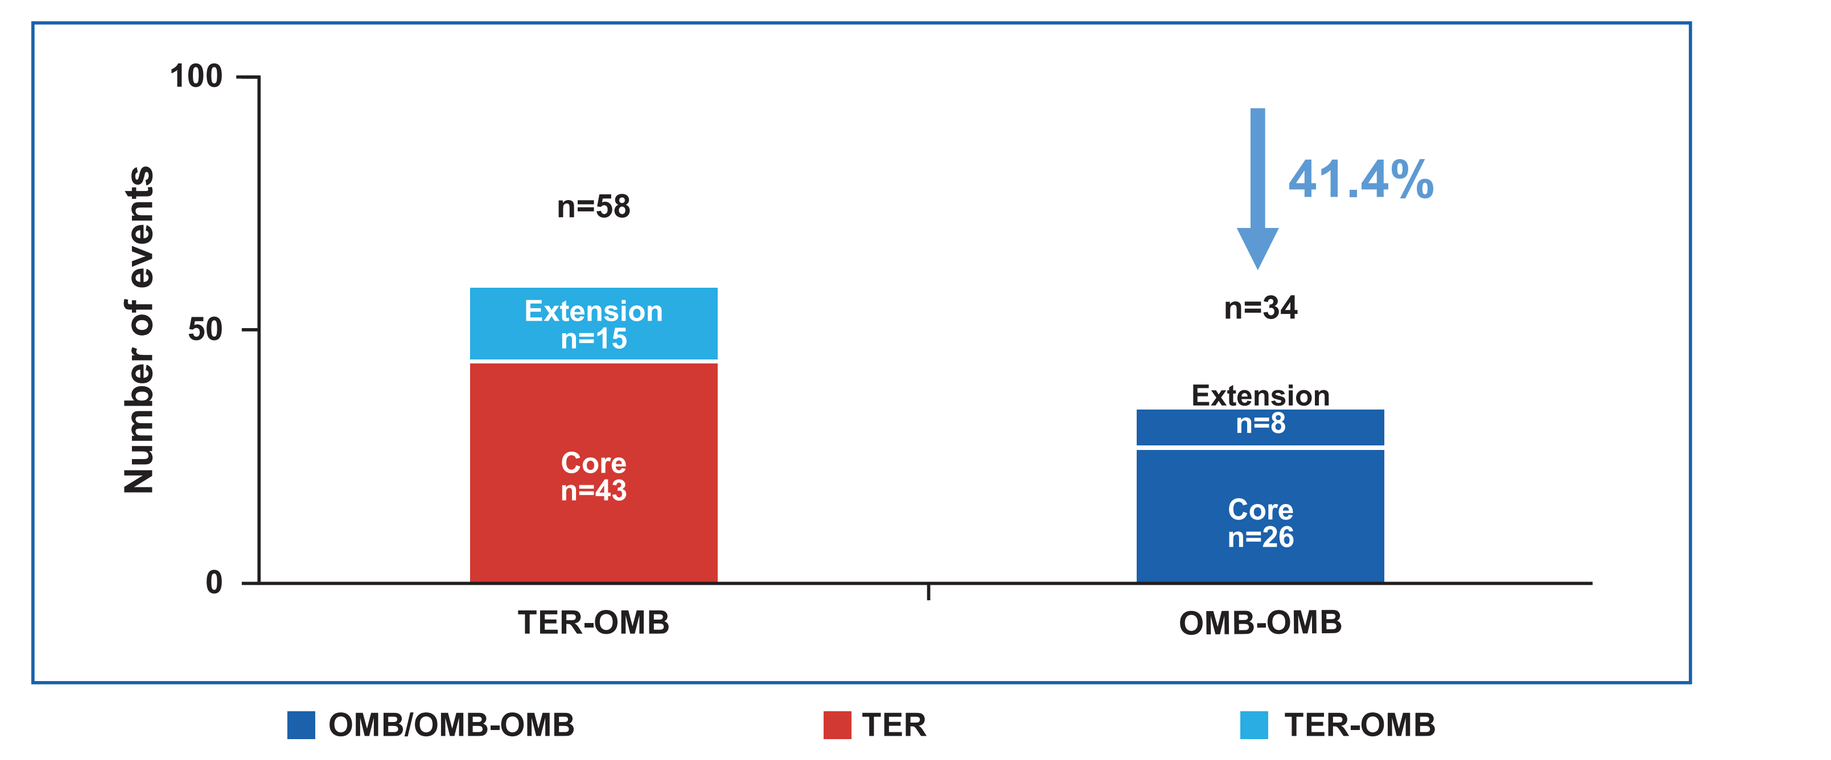

Number of events
n=58
Extension n=15
Core n=43
41.4%
n=34
Extension n=8
Core n=26
TER-OMB
OMB-OMB
OMB/OMB-OMB
TER
TER-OMB

B Time to first 3mCDW – Kaplan-Meier estimates

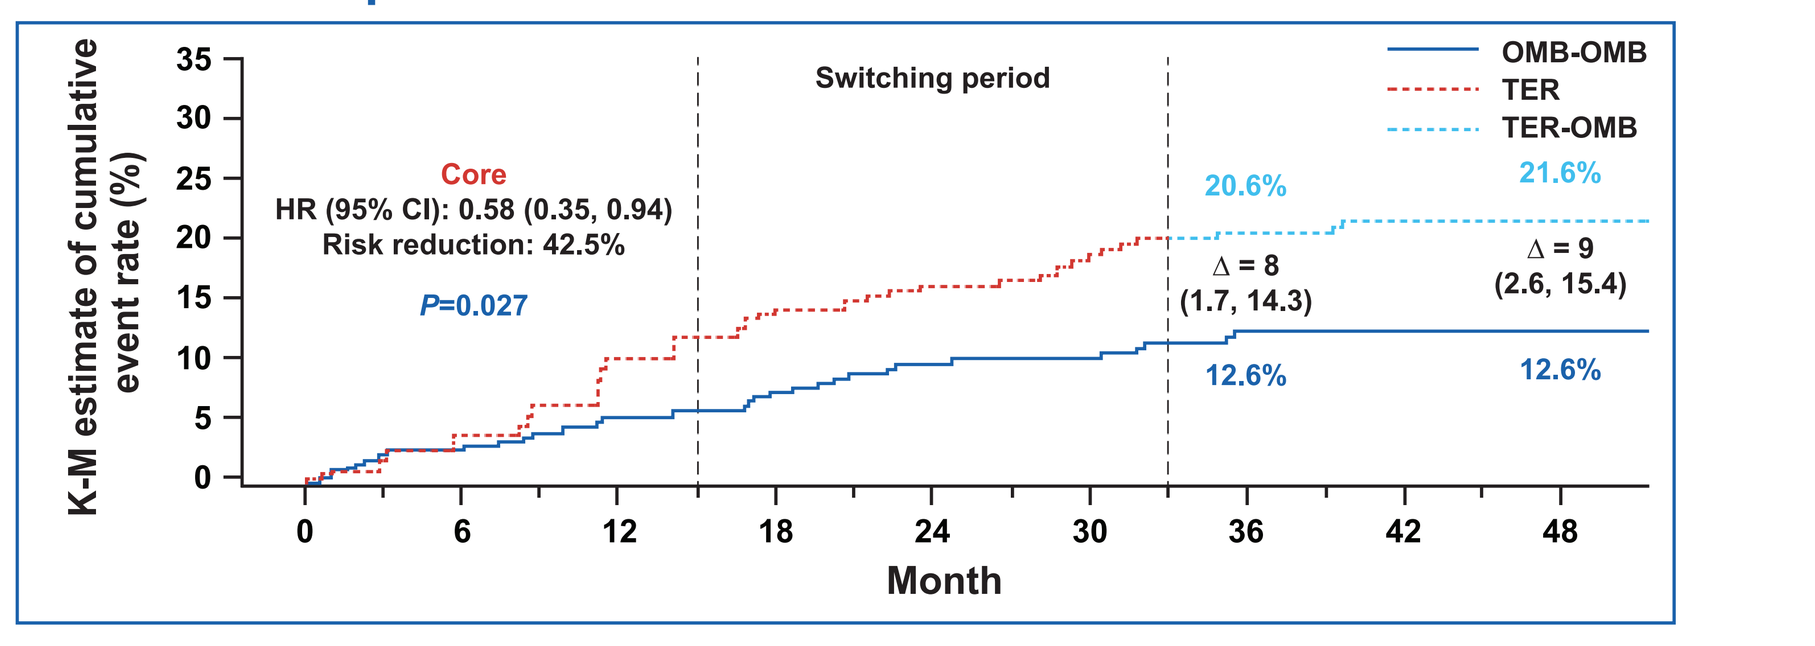

K-M estimate of cumulative event rate (%)
Core
HR (95% CI): 0.58 (0.35, 0.94)
Risk reduction: 42.5%
P=0.027
Switching period
OMB-OMB
TER
TER-OMB
20.6%
21.6%
Δ = 8 (1.7, 14.3)
Δ = 9 (2.6, 15.4)
12.6%
12.6%
Month

Figure 3. Between-group comparison of cumulative number of 6mCDW events (A), and time to first 6mCDW (B)

A Cumulative number of first 6mCDW events over up to 4 years

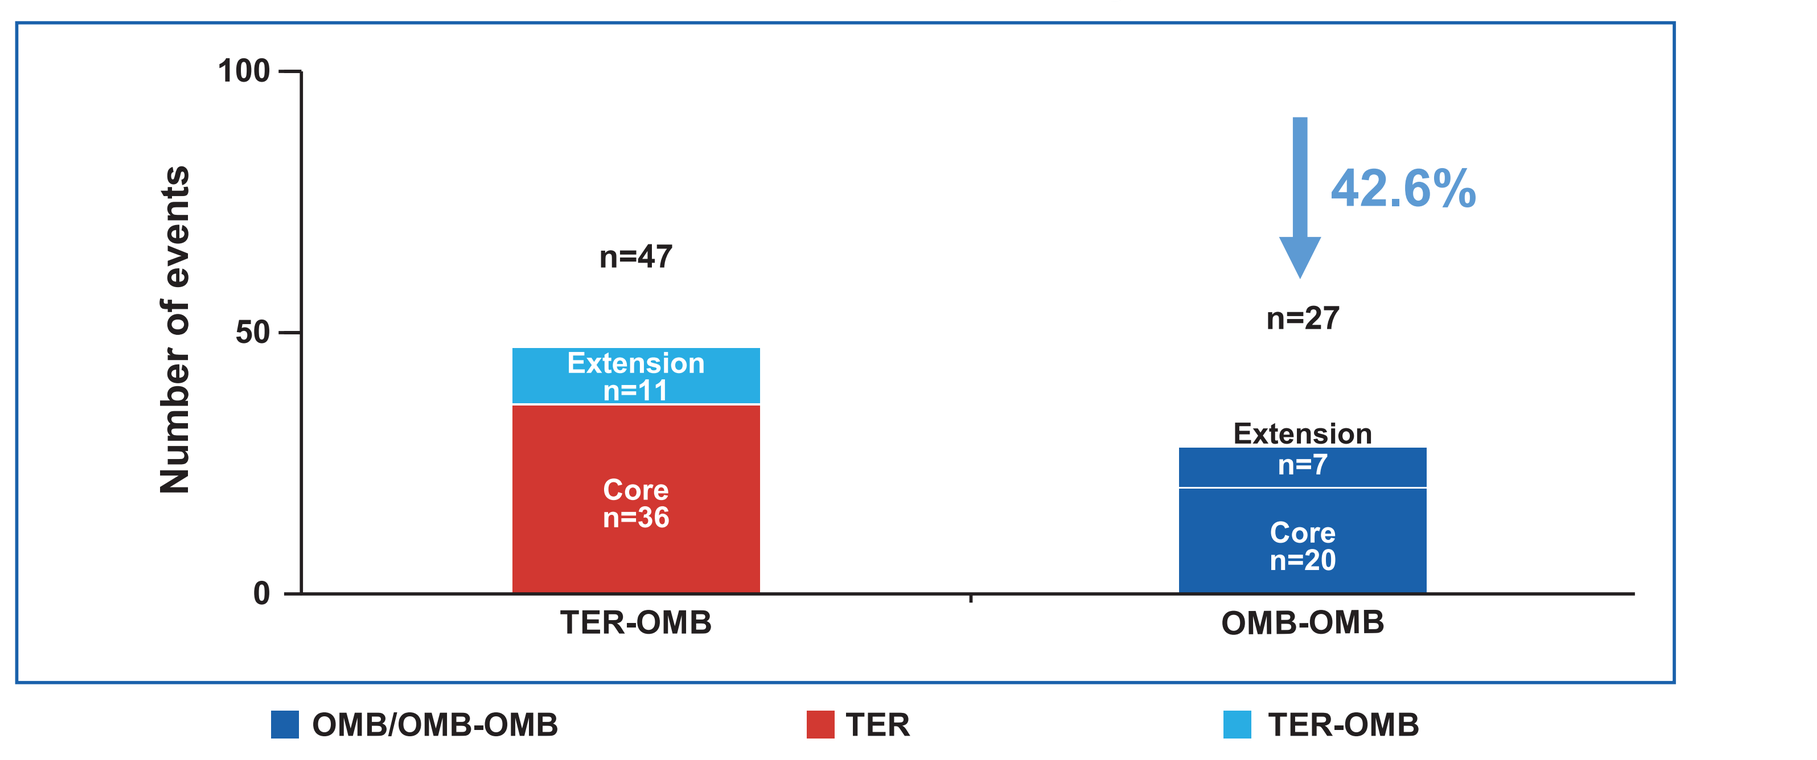

Number of events
n=47
Extension n=11
Core n=36
42.6%
n=27
Extension n=7
Core n=20
TER-OMB
OMB-OMB
OMB/OMB-OMB
TER
TER-OMB

B Time to first 6mCDW – Kaplan-Meier estimates

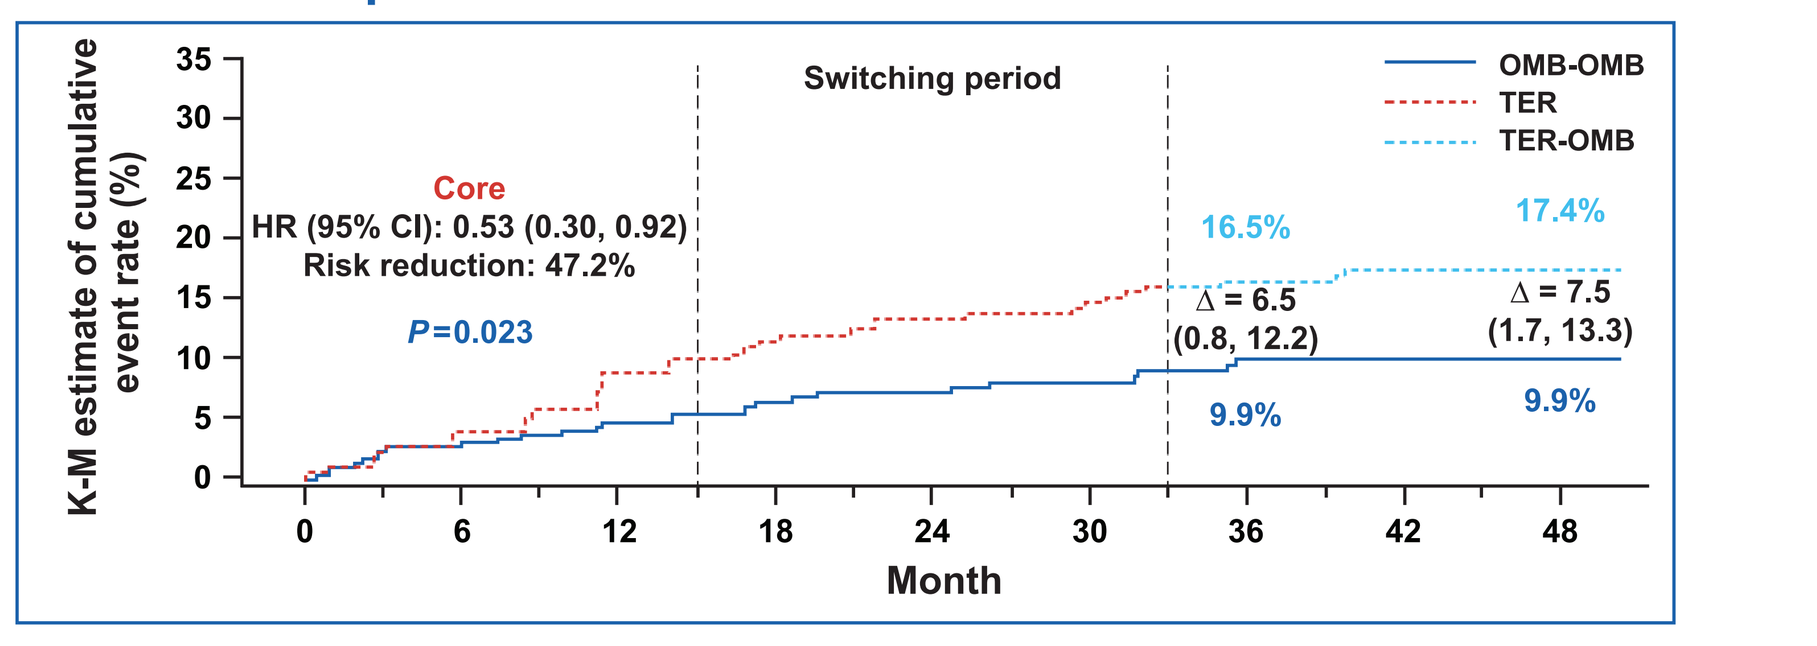

K-M estimate of cumulative event rate (%)
Core
HR (95% CI): 0.53 (0.30, 0.92)
Risk reduction: 47.2%
P=0.023
Switching period
OMB-OMB
TER
TER-OMB
16.5%
17.4%
Δ = 6.5 (0.8, 12.2)
Δ = 7.5 (1.7, 13.3)
9.9%
9.9%
Month

Cut-off for core and extension periods refer to the first dose of ofatumumab in extension. Δ, Difference in KM estimates (TER-OMB minus OMB-OMB). *P* value represented here is *P* value for Log-Rank test.
3mCDW, 3-month confirmed disability worsening; 6mCDW, 6-month confirmed disability worsening; CI, confidence interval; HR, hazard ratio; K-M, Kaplan-Meier, OMB-OMB, continuous ofatumumab; TER-OMB, switch from teriflunomide to ofatumumab.

Mean number of Gd+ T1 lesions

- The between-group analysis over a period of up to 4 years shows that earlier initiation of ofatumumab was associated with a reduction in the cumulative number of Gd+ T1 lesions by 96.6%, *P*<0.0001 (**Figure 4A**)
- The number of Gd+ T1 lesions per scan in the continuous ofatumumab group remained low for up to 4 years after treatment initiation
- Within group analysis showed that continuous use of ofatumumab and switch from teriflunomide was associated with almost complete suppression of Gd+T1 activity (100%, 99.4% respectively) (**Figure 4B**)

Figure 4. Between-group comparison – Cumulative number of Gd+ T1 lesions (A), and within-group comparison of mean number of Gd+T1 lesions between the core and extension phase (B)

A Cumulative number of Gd+T1 lesions over up to 4 years

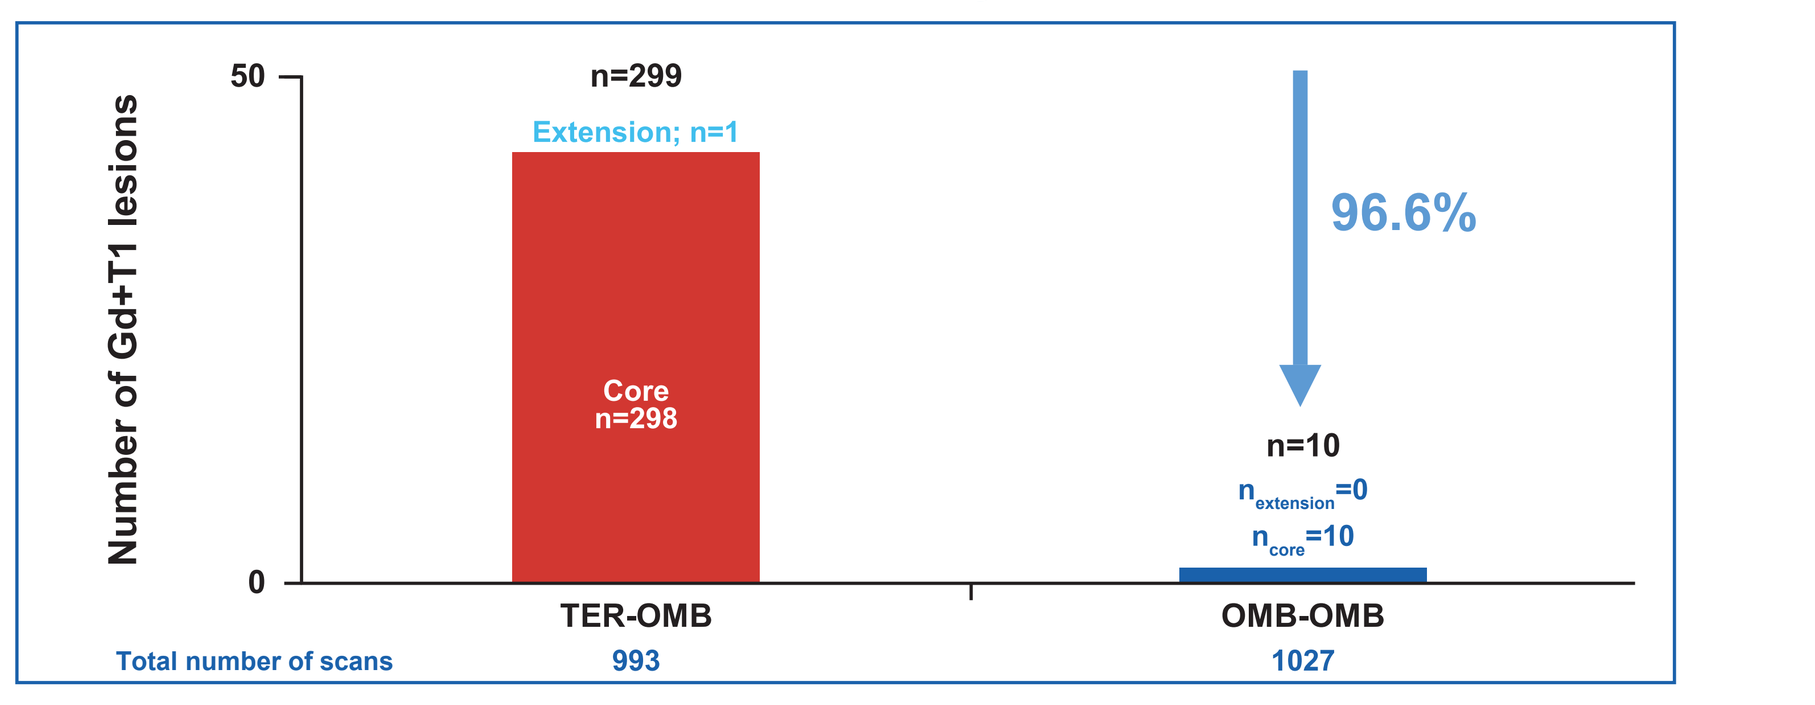

Number of Gd+T1 lesions
n=299
Extension; n=1
Core n=298
96.6%
n=10
n extension=0
n core=10
TER-OMB
OMB-OMB
Total number of scans
993
1027

B Within-group comparison[a] between the core and extension phase

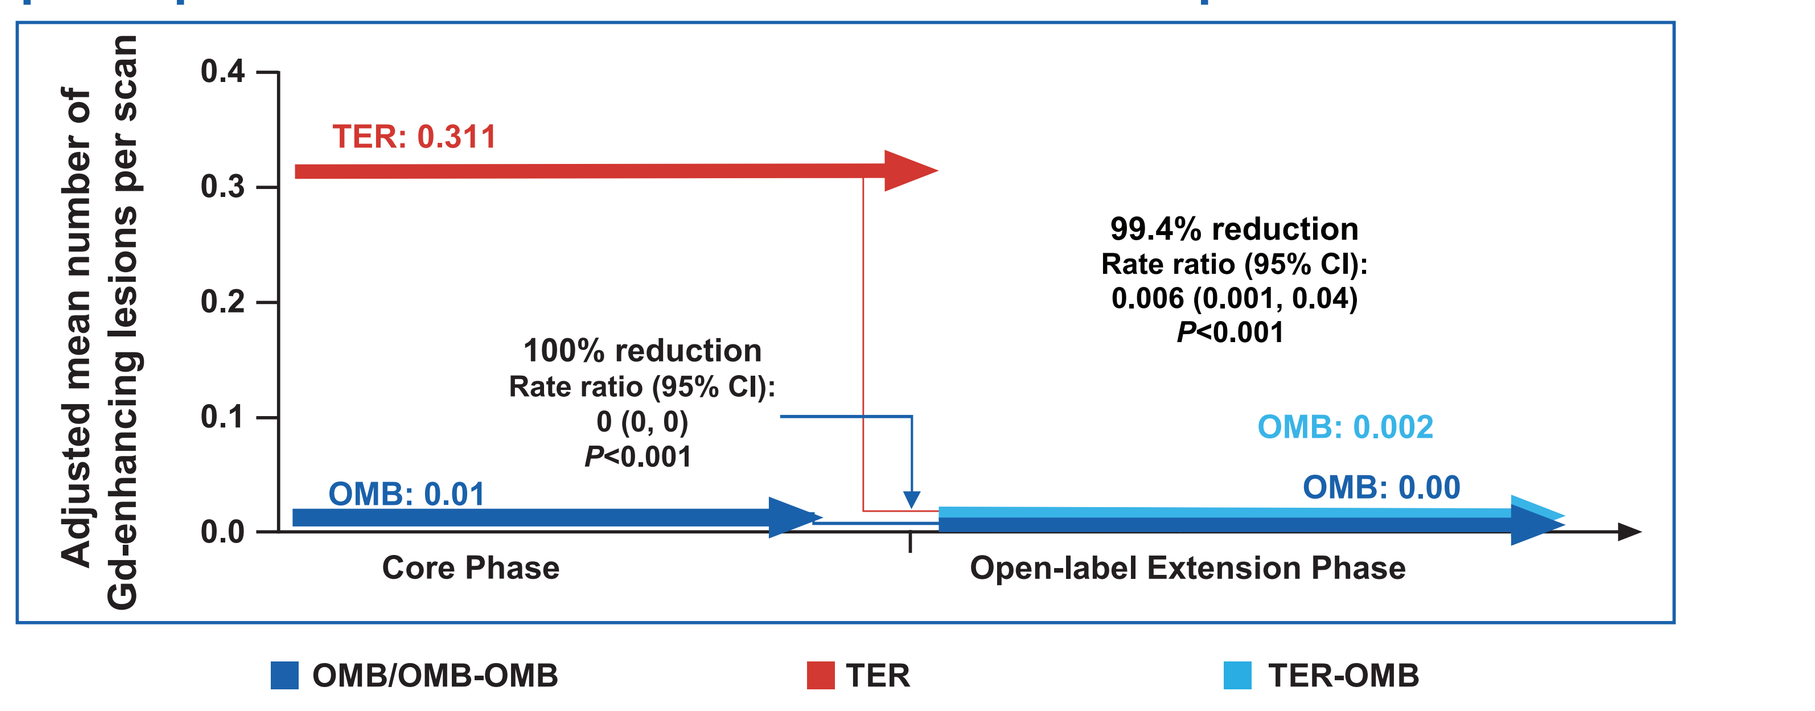

Adjusted mean number of Gd-enhancing lesions per scan
TER: 0.311
99.4% reduction
Rate ratio (95% CI):
0.006 (0.001, 0.04)
P<0.001
100% reduction
Rate ratio (95% CI):
0 (0, 0)
P<0.001
OMB: 0.01
OMB: 0.002
OMB: 0.00
Core Phase
Open-label Extension Phase
OMB/OMB-OMB
TER
TER-OMB

[a]Estimated from fitting a piecewise negative binomial model for the time period core phase and extension phase with log-link, adjusted for treatment and region as factors, baseline number of T1 Gd-enhancing lesions and patient's age at baseline as covariates. The natural log of the number of scans with evaluable Gd-enhancing lesion counts by period is used as offset to obtain the lesion rate per scan in each period. Baseline variables are from the core study baseline. All *P* values are nominal *P* values.
CI, confidence interval; Gd, gadolinium; OMB, ofatumumab; OMB-OMB, continuous ofatumumab; TER, teriflunomide; TER-OMB, switch from teriflunomide to ofatumumab.

Number of neT2 lesions

- Similar to Gd+T1 lesions, the between-group analysis over a period of up to 4 years shows that earlier initiation of ofatumumab was associated with a reduction in the cumulative number of neT2 lesions by 83.4%, *P*<0.0001 (**Figure 5A**)
- The number of neT2 lesions in the continuous ofatumumab group remained low for up to 4 years after treatment initiation; a near complete suppression was observed during the extension phase
- The within-group analysis showed that continuous use of ofatumumab was associated with a reduction in the neT2 lesions by 90.7% with longer-term treatment, while switch from teriflunomide to ofatumumab resulted in a pronounced reduction in the number of neT2 lesions (88.3%) (**Figure 5B**)

Figure 5. Between-group comparison – Cumulative number of neT2 lesions (A), and within-group comparison of adjusted mean annualised rate of neT2 lesions between the core and extension phase

A Cumulative number of neT2 lesions over up to 4 years

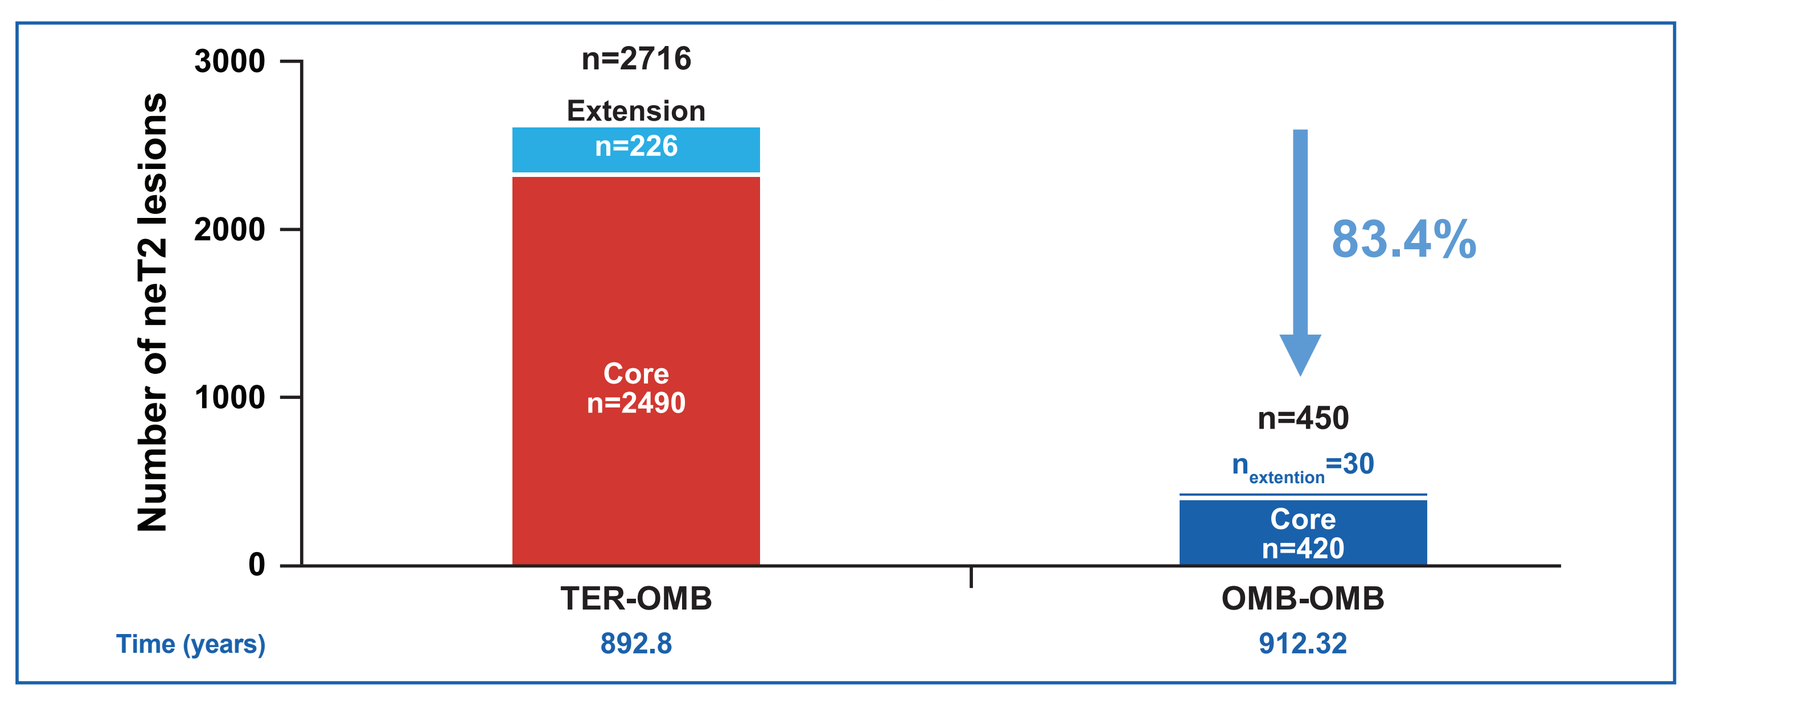

Number of neT2 lesions
n=2716
Extension n=226
Core n=2490
83.4%
n=450
n extention=30
Core n=420
TER-OMB
OMB-OMB
Time (years)
892.8
912.32

B Within-group comparison[a] between the core and extension phase

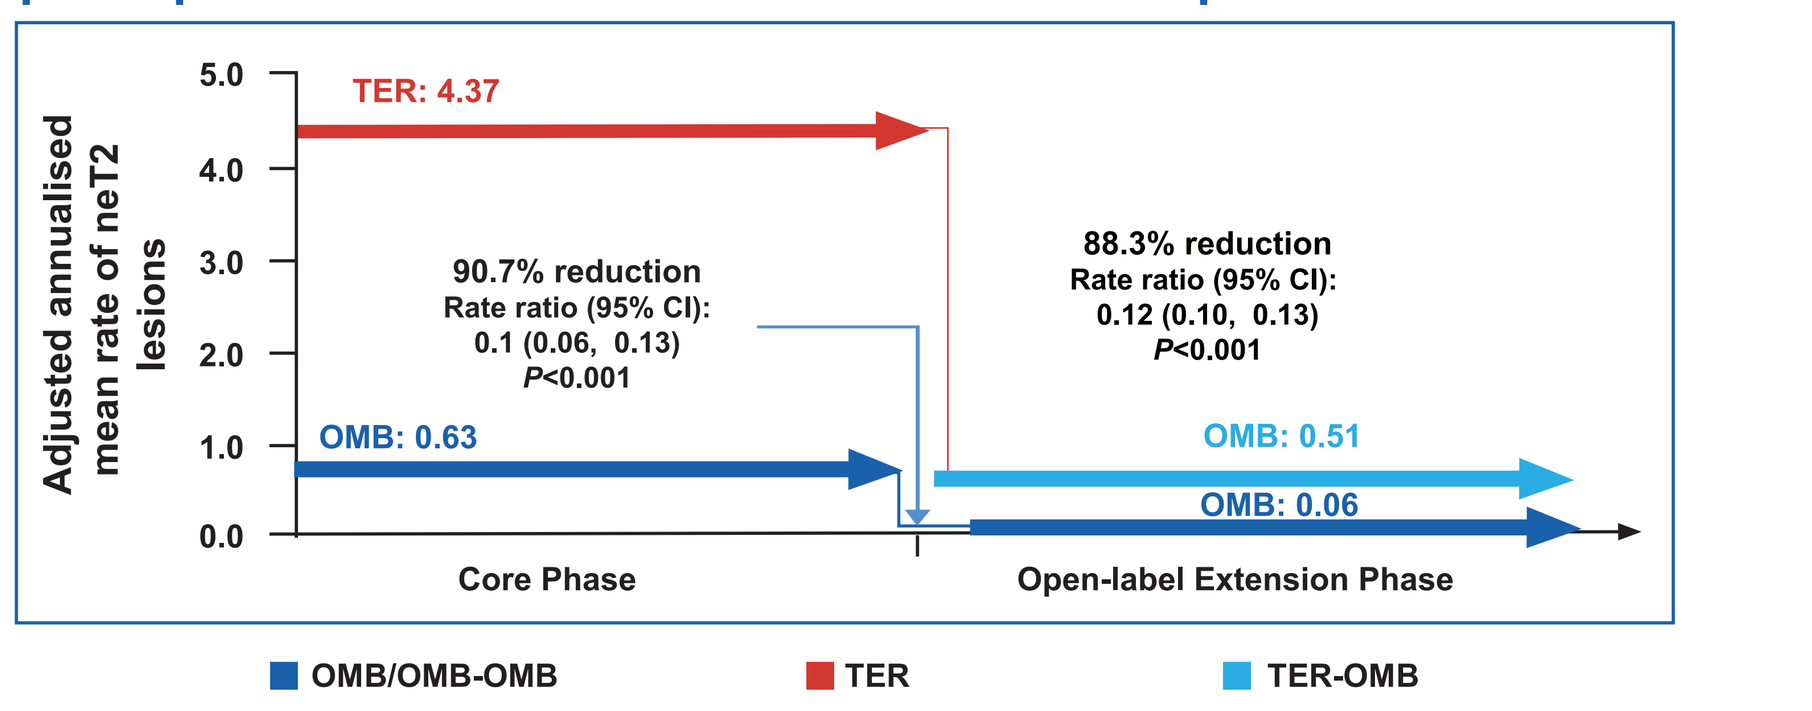

Adjusted annualised mean rate of neT2 lesions
TER: 4.37
88.3% reduction
Rate ratio (95% CI):
0.12 (0.10, 0.13)
P<0.001
90.7% reduction
Rate ratio (95% CI):
0.1 (0.06, 0.13)
P<0.001
OMB: 0.63
OMB: 0.51
OMB: 0.06
Core Phase
Open-label Extension Phase
OMB/OMB-OMB
TER
TER-OMB

[a]Estimated from fitting a piecewise negative binomial model for the time period core phase and extension phase with log-link, adjusted for treatment as factor, baseline volume of T2 lesions and patient's age at baseline as covariates. The natural log of the time-in-study (in years) by period is used as offset to annualise the lesion rate in each period. Baseline variables are from the core study baseline. All *P* values are nominal *P* values.
CI, confidence interval; neT2, new or enlarging T2; OMB, ofatumumab; OMB-OMB, continuous ofatumumab; TER, teriflunomide; TER-OMB, switch from teriflunomide to ofatumumab.

Safety Outcomes

Overall AEs

- The safety and tolerability profile of ofatumumab in RDTN participants was consistent with that in the overall ofatumumab clinical trial safety population[6] (**Table 2**)
- The most common (≥10% of each group) AEs were nasopharyngitis, injection-related systemic reactions, injection-site reactions, headache, upper respiratory tract infections, fatigue, backpain, and COVID-19
- A total of 66 cases of COVID-19 were reported. The majority were mild-to-moderate in severity. The percentage of patients with COVID-19 is consistent with overall ofatumumab clinical trial safety population (14.38%)[6]

Table 2. AEs in RDTN patients as of first dose of ofatumumab (safety analysis set)

Adverse event	OMB-OMB N=314		TER-OMB N=232		OMB overall N=546	
	n (%)	EAIR (95% CI)	n (%)	EAIR (95% CI)	n (%)	EAIR (95% CI)
Patients with at least one AE	294 (93.6)	166.85 (148.83, 187.06)	193 (83.2)	129.42 (112.39, 149.03)	487 (89.2)	149.70 (136.97, 163.60)
AEs leading to OMB discontinuation	33 (10.5)	-	10 (4.3)	-	43 (7.9)	-
Most common AEs (≥10 in any group)						
Nasopharyngitis	90 (28.7)	11.81 (9.60, 14.52)	38 (16.4)	9.24 (6.73, 12.71)	128 (23.4)	10.91 (9.17, 12.97)
Injection-related systemic reaction	80 (25.5)	10.17 (8.17, 12.66)	56 (24.1)	15.61 (12.01, 20.28)	136 (24.9)	11.87 (10.04, 14.05)
Injection-site reactions	54 (17.2)	6.22 (4.76, 8.12)	24 (10.3)	5.65 (3.78, 8.42)	78 (14.3)	6.03 (4.83, 7.53)
Headache	59 (18.8)	6.97 (5.40, 9.00)	19 (8.2)	4.31 (2.75, 6.75)	78 (14.3)	6.06 (4.85, 7.56)
Upper respiratory tract infection	55 (17.5)	6.25 (4.80, 8.14)	22 (9.5)	5.06 (3.33, 7.68)	77 (14.1)	5.85 (4.68, 7.32)
Fatigue	42 (13.4)	4.59 (3.39, 6.21)	9 (3.9)	1.97 (1.03, 3.79)	51 (9.3)	3.72 (2.83, 4.89)
Back pain	36 (11.5)	3.86 (2.79, 5.36)	15 (6.5)	3.34 (2.02, 5.54)	51 (9.3)	3.69 (2.81, 4.86)
COVID-19	35 (11.1)	3.56 (2.56, 4.96)	31 (13.4)	6.90 (4.85, 9.81)	66 (12.1)	4.61 (3.62, 5.87)
Infections	220 (70.1)	51.02 (44.70, 58.22)	123 (53.0)	41.64 (34.90, 49.69)	343 (62.8)	47.21 (42.46, 52.47)
Patients with at least one SAE	51 (16.2)	5.49 (4.17, 7.22)	18 (7.8)	3.94 (2.48, 6.25)	69 (12.6)	4.98 (3.93, 6.30)
Infections[a]	17 (5.4)	1.73 (1.08, 2.79)	7 (3.0)	1.51 (0.72, 3.17)	24 (4.4)	1.66 (1.11, 2.48)
Malignancies	5 (1.6)	0.50 (0.21, 1.20)[b]	1 (0.4)	0.21 (0.03, 1.52)[c]	6 (1.1)	0.41 (0.18, 0.91)
Deaths	2 (0.6)[d]	-	2 (0.9)[e]	-	4 (0.7)	-

Data are shown as the number of participants (%) with at least one event. OMB overall refers to the patients enrolled in ASCLEPIOS I/II and ALITHIOS.
[a]In the continuous ofatumumab group there were three cases of COVID-19/COVID-19 pneumonia, four cases of appendicitis, two cases of pneumonia, and one case each of pneumocystis jiroveci pneumonia, neutropenic sepsis, appendicitis perforated, abscess, herpes zoster, bronchitis, influenza and URTI; in the switch group there were six cases of COVID-19/COVID-19 pneumonia, and one case of appendicitis. [b]In the continuous group there were two cases of basal cell carcinoma, and one case each of ovarian cancer, renal cell carcinoma and uterine leiomyoma; [c]In the switch group there was one case of basal cell carcinoma; [d]In the continuous ofatumumab group, one case of death was due to COVID-19 pneumonia and one case due to completed suicide; [e]in the switch group, one case of death was due to COVID-19 and other was sudden death.
AE, adverse event; CI, confidence interval; EAIR, exposure adjusted incidence rate; OMB, ofatumumab; RDTN, recently diagnosed, treatment-naive; SAE, serious AE;
N: total number of participants included in the analysis.

Injection-related reactions (Systemic and local)

- IRRs were predominantly reported with first injection and the incidence decreased substantially with subsequent injections which is consistent with the overall ofatumumab clinical trial safety population
- Mostly all IRRs (99.3%) were mild-moderate in severity; no Grade 4 IRRs were reported
- No cases of cytokine release syndrome were observed. A total of 2 patients discontinued treatment with injection systemic reaction (one in each group) and none with injection-site reaction

Conclusions

- Long-term, continuous ofatumumab treatment up to 4 years showed sustained benefits on relapses, MRI lesions, and risk of disability worsening in a subgroup of RDTN patients, consistent with that of overall ASCLEPIOS population[3]
 - Sustained differences in efficacy outcomes observed in the continuous versus the switch group highlight the value of earlier initiation of high-efficacy therapy with ofatumumab compared to a lower efficacy therapy
- The safety and tolerability profile of ofatumumab in RDTN participants was consistent with that in the overall ofatumumab clinical trial safety population
- These findings support a favorable benefit–risk profile for ofatumumab in recently diagnosed, treatment naive patients, consistent with that of the overall ASCLEPIOS RMS population, supporting its use in patients at an early stage of their RMS disease course

References

1. KESIMPTA® (ofatumumab) Prescribing Information. https://www.novartis.us/sites/www.novartis.us/files/kesimpta.pdf (accessed February 17, 2022).
2. Hauser SL, et al. *N Engl J Med.* 2020;383:546–57.
3. Gartner, J et al. *Mult Scler.* 2022. 28: 1562–1575.
4. Kappos L, et al. Poster presented at *EAN* 2022. EPR161.
5. Sacca F, et al. Oral presentation at *EAN* 2022. OPR134.

Acknowledgements

The authors acknowledge the following Novartis employees: **Amitha Thakur** and **Saimithra Thammera** for medical writing assistance and coordinating author reviews, and **Moganti Ravi Sankar** for creative design assistance. The final responsibility for the content lies with the authors

Disclosures

This study was funded by Novartis Pharma AG, Switzerland.

Jutta Gartner in the past 3 years, has received fees for lectures and consultancy fees from Bayer, Biogen, Merck, Novartis and Sanofi, as well as funding for a research project from Novartis. **Stephen L. Hauser** has received personal compensation from Annexon, Alector, Accure, and Neurona; he has also received travel reimbursement from F. Hoffmann-La Roche Ltd and Novartis for CD20-related meetings and presentations. **Amit Bar-Or** has participated as a speaker in meetings sponsored by and received consulting fees and/or grant support from: Accure, Atara Biotherapeutics, Biogen, BMS/Celgene/Receptos, GlaxoSmithKline, Gossamer, Janssen/Actelion, Medimmune, Merck/EMD Serono, Novartis, Roche/Genentech, Sanofi-Genzyme. **Xavier Montalban** has received speaking honoraria and travel expenses for participation in scientific meetings, has been a steering committee member of clinical trials or participated in advisory boards of clinical trials in the past years with Actelion, Alexion, Bayer, Biogen, Celgene, EMD Serono, Genzyme, Immunic, Medday, Merck, Mylan, Nervgen, Novartis, Roche, Sanofi-enzyme, Teva Pharmaceutical, TG Therapeutics, Excemed, MSIF and NMSS. **Jeffrey A. Cohen** received personal compensation for consulting for Biogen, Bristol-Myers Squibb, Convelo, Genentech, Janssen, NervGen, Novartis, and PSI; speaking for H3 Communications; and serving as an Editor of Multiple Sclerosis Journal. **Derrick Robertson** has received fees for consulting, contracted research and speaker's bureau from Biogen, Celgene, EMD Serono, Genentech, Sanofi Genzyme, Janssen, TG therapeutics; consulting fees and speakers bureau for Bristol Myers Squibb, Horizon, and Alexion; consulting fees and contracted research for Novartis; consulting fees for Greenwich biosciences; contracted research for GW Pharmaceuticals, PCORI, Atara Biotherapeutics and CorEvitas. **Anne H. Cross** has received consulting fees, support, and honoraria from Biogen, Celgene, Bristol Myers Squibb, EMD Serono, Merck, Genentech, Roche, Greenwich Biosciences (Jazz Pharmaceuticals), Horizon Therapeutics, Janssen (subsidiary of Johnson & Johnson), Novartis, TG Therapeutics, Academic CME, Projects In Knowledge, CME Outfitters, WebMD, Conrad N. Hilton Foundation, Potomac Center for Medical Education, The Consortium of Multiple Sclerosis Centers, and ACTRIMS; has received a grant from the Department of Defense, USA; has been the secretary (elected) of The Consortium of Multiple Sclerosis Centers, member of the scientific advisory board of Race to Erase MS, program committee (chair) of ACTRIMS, member of the COVID-19 advisory committee of the National Multiple Sclerosis Society and National Multiple Sclerosis Society representative on the Progressive MS Alliance; has participated on the data safety monitoring board or advisory board for Race to Erase MS (charity), National Multiple Sclerosis Society, Novartis, EMD Serono, Biogen, Celgene/Bristol Myers Squibb, and TG Therapeutics; has received patent for "Yablonskiy DA, Sukstansky AL, Wen J, Cross AH. Methods for simultaneous multi-angular relaxometry of tissue using magnetic resonance imaging. Patent 15060-630 (015875)." **Carrie M. Hersh** has received speaking and consulting fees from Genentech, Genzyme, Biogen, Novartis, and EMD-Serono. She has received research support paid to her institution by PCORI, Biogen, and Genentech. **Kumaran Deiva** has received personal compensation for speaker activities from Novartis and Sanofi. **Karlsson Goeril, Ayan Das Gupta, Ronald Zielman, Soudeh Ansari, Bernd Kieseier** are employees of Novartis. **Ludwig Kappos'** institution (University Hospital Basel) has received the following exclusively for research support: steering committee, advisory board, and consultancy fees (Actelion, Addex, Bayer HealthCare, Biogen Idec, Biotica, Genzyme, Lilly, Merck, Mitsubishi, Novartis, Ono Pharma, Pfizer, Receptos, Sanofi, Santhera, Siemens, Teva, UCB, and Xenoport); speaker fees (Bayer HealthCare, Biogen Idec, Merck, Novartis, Sanofi, and Teva); and support for educational activities (Bayer HealthCare, Biogen, CSL Behring, 43)

Poster presented at the 38th Congress of the European Committee for Treatment and Research in Multiple Sclerosis (ECTRIMS), 26–28 October, 2022, Amsterdam, the Netherlands

Visit the web at: https://bit.ly/ectrims2022

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Presenter email address: gaertnj@med.uni-goettingen.de

Scan this QR code to download a copy Poster