Evaluation of the use of high-efficacy treatments (HETs) in patients with relapsing-remitting multiple sclerosis in Argentina

P1988

Authors: Ricardo Alonso[1,2] , Magdalena Casas[1], Luciana Lazaro[1], Nora Fernandez Liguori[2,3], Cecilia Pita[1], Leila Cohen[1], Juan Ignacio Rojas[4,5], Agustín Pappolla[4], Liliana Patrucco[4], Edgardo Cristiano[4], Marcos Burgos[6], Carlos Vrech[7], Raul Piedrabuena[8], Pablo Lopez[9], Norma Deri[10], Geraldine Luetic[11], Jimena Miguez[12], Mariela Cabrera[13], Alejandra Martinez[14], Gisela Zanga[15], Verónica Tkachuk[16], Santiago Tizio[17], Edgar Carnero Contentti[9] , Eduardo Knorre[18], Felisa Leguizamon[18], Carolina Mainella[19], Pedro Nofal[20], Susana Liwacki[21], Javier Hryb[22], Maria Menichini[11], Claudia Pestchanker[23], Orlando Garcea[1], Berenice Silva[1,12]

Affiliations: 1 Centro Universitario de Esclerosis Múltiple. Hospital Ramos Mejía, Ciudad de Buenos Aires. 2 Servicio de Neurología. Sanatorio Güemes, Ciudad de Buenos Aires. 3 Servicio de Neurología. Hospital Tornú, Ciudad de Buenos Aires. 4 Centro de esclerosis Múltiple Buenos Aires. Ciudad de Buenos Aires. 5 Servicio de Neurología. CEMIC, Ciudad de Buenos Aires. 6 Servicio de Neurología. Hospital San Bernardo, Salta. 7 Servicio de Neurología. Sanatorio Allende, Córdoba 8 Servicio de Neurología. Clínica Universitaria Reina Fabiola, Córdoba 9 Sección de nueroinmunología. Hospital Alemán, Ciudad de Buenos Aires. 10 DIABAI 11 Instituto Neurociencias. Rosario 12 Servicio de neurología. Hospital Italiano. 13 Servicio de neurología. Hospital Militar, Campo de Mayo. 14 Servicio de neurología. Hospital Posadas, Buenos Aires. 15 Servicio de neurología. Hospital Cesar Milstein, Ciudad de Buenos. 16 Servicio de neurología. Hospital de Clínica José de San Martín, Ciudad de Buenos Aires. 17 Servicio de neurología. Hospital Español, La Plata. 18 Servicio de neurología. Hospital Álvarez, Ciudad de Buenos Aires. 19 Servicio de neurología. Hospital Español, Rosario. 20 Servicio de neurología. Hospital Nuestra Señora del Carmen, Tucumán 21 Servicio de neurología. Hospital Córdoba, Córdoba. 22 Servicio de neurología. Hospital Durand, Ciudad de Buenos Aires. 23 Servicio de neurología. Hospital de San Luis, San Luis.

Introduction

Disease-modifying therapies (DMTs) in multiple Sclerosis (MS) can be classified according to their efficacy. DMTs can be divided into two broad classes: drugs of high efficacy, defined as average relapse reduction substantially more than 50% and drugs of moderate efficacy, defined as average relapse reduction between 30 and 50%, compared to placebo or another immunomodulatory treatment, in clinical trials. Different factors tend to lead clinicians or treatment guidelines to recommend high efficacy treatments (HETs) for use in individuals with the most aggressive forms of MS. Despite the evidence showing that HETs are more efficacious in suppressing or delaying relapse activity when initiated early after disease onset to date, there are few data on the characteristics of MS patients treated with HETs in Argentina.

Objective

To analyze the use of HETs in Argentina, focusing on the clinical and sociodemographic characteristics of the patients who use these treatments and the changes in the trend of use over the years.

Methods

Study Population

This retrospective cohort study was done using the Argentina MS patient registry, RelevarEM, the first nationwide MS registry in Argentina and LATAM (Clinical Trials registry number NCTNCT03375177). The inclusion criteria for this study were: Age ≥18 years at disease onset (defined as the first symptom of the inflammatory process), diagnosed with RRMS according to validated diagnosis criteria and under treatment with natalizumab, alemtuzumab, cladribine, rituximab or ocrelizumab Although rituximab is not locally approved for MS treatment, it is used off-label by local neurologists, including some medical experts.

Period definition

Taking into consideration the availability of different DMTs in Argentina, we defined two periods of time (P): from 2015 to 2017 (P1) and from 2018 to 2020 (P2). A comparative analysis between these two periods was performed to assess the tendency of DMTs use over time.

Sample size calculation

The scientific committee responsible for RelevarEM reported that, approximately, 12% of RRMS patients in the registry are currently treated with HETs. Assuming that 12% of the subjects in the population have the factor of interest, the study would require a sample size of 163 for estimating the expected proportion with 5% absolute precision and 95% confidence. In order to improve the robustness of the analysis of the secondary endpoints, as this is a non-interventional and non-prospective study, all patients in the registry that fulfilled the inclusion and exclusion criteria were included in the study.

Statistical analysis

Data analysis was conducted using SPSS Statistics v22. Descriptive analyses of all variables were carried out. Results were presented as frequencies, percentages, ranges, mean and standard deviation values. Comparisons between the two groups were analyzed using Chi-square or Fisher's exact tests for categorical variables. Continuous variables were analyzed using Student t-test or Wilcoxon's tests as appropriate and analysis of variance (ANOVA) with Bonferroni post hoc correction or Kruskal-Wallis test with Dunn post hoc analysis was used to compare three or more groups. Statistical significance was set at p< 0.05.

Results

Sociodemographic and clinical history of patients under HETs/rituximab treatment

315 patients were included in this analysis as they met the inclusion and exclusion criteria. Of them, 67.7% were female, the mean age was 37 ± 10.9 years at study entry, and they had a median disease duration of 6 years (IQR 4 -10). Demographic and clinical characteristics are summarized in Table 1. More than one-third of patients (35%) received HETs or rituximab as their first treatment, while the rest started HETs or rituximab after switching from a prior DMT (Figure 1).

Table 1: Demographic and clinical characteristics of the included patients (n = 315)

Variable	Result
Gender, n (%) - Female - Male	 213 (67.7) 102 (32.4)
Residence, n (%) - Buenos Aires City - Buenos Aires province - Rest of Argentina	 104 (33.2) 100 (31.6) 111 (35.2)
CCI, n (%) - 0 - ≥1	 290 (92) 25 (8)
Mean age at study entry, SD (years)	37 (10.9)
Mean age at onset of symptoms, SD (years)	28,7 ±9.7
Mean age at diagnosis, SD (years)*	30,1 ± 9.7
Median time MS duration, IIQ (years)	6 (4-10)
Patients currently in rehabilitation*, n (%)	58 (18.5)
Mean EDSS score at diagnosis, SD	2,3 ± 1,4
Mean EDSS score at study entry, SD	2,8 ± 2
Positive OCB, n (%)	255 (81.1)
Infratentorial lesions on MRI at the time of diagnosis (%)	241 (76.6)
Spinal cord lesions on MRI at the time of diagnosis (%)	223 (70.9)
Contrast-enhancing lesions on first MRI (%)	203 (64.6)

* Media (DS) **Mediana (IIQ). MS: Multiple sclerosis. EDSS: Expanded Disability Status Scale. OCB: oligoclonal bands. MRI: magnetic resonance imaging. CCI: Charlson comorbidity index

Pharmacological history of patients under HETs or rituximab treatment

The most frequent reason for switching to current HETs was treatment failure to previous DMT (77%). Fingolimod was the most frequent treatment prior to current HET (32%). On the other hand, the time from MS diagnosis to the first HET in treatment-naive patients was shorter than one year (IQR: 0-1 year) and in treatment-experienced patients it was 5 years (IQR: 3-9 years). Almost all patients (97.46%) presented at least one characteristic associated with high-activity MS prior to the initiation of current HETs. More than two thirds of the patients treated with HETs presented clinical or radiological activity in the previous 12 months before starting the current HET.

Figure 1: Pharmacological history prior to current HET/rituximab (n = 315)

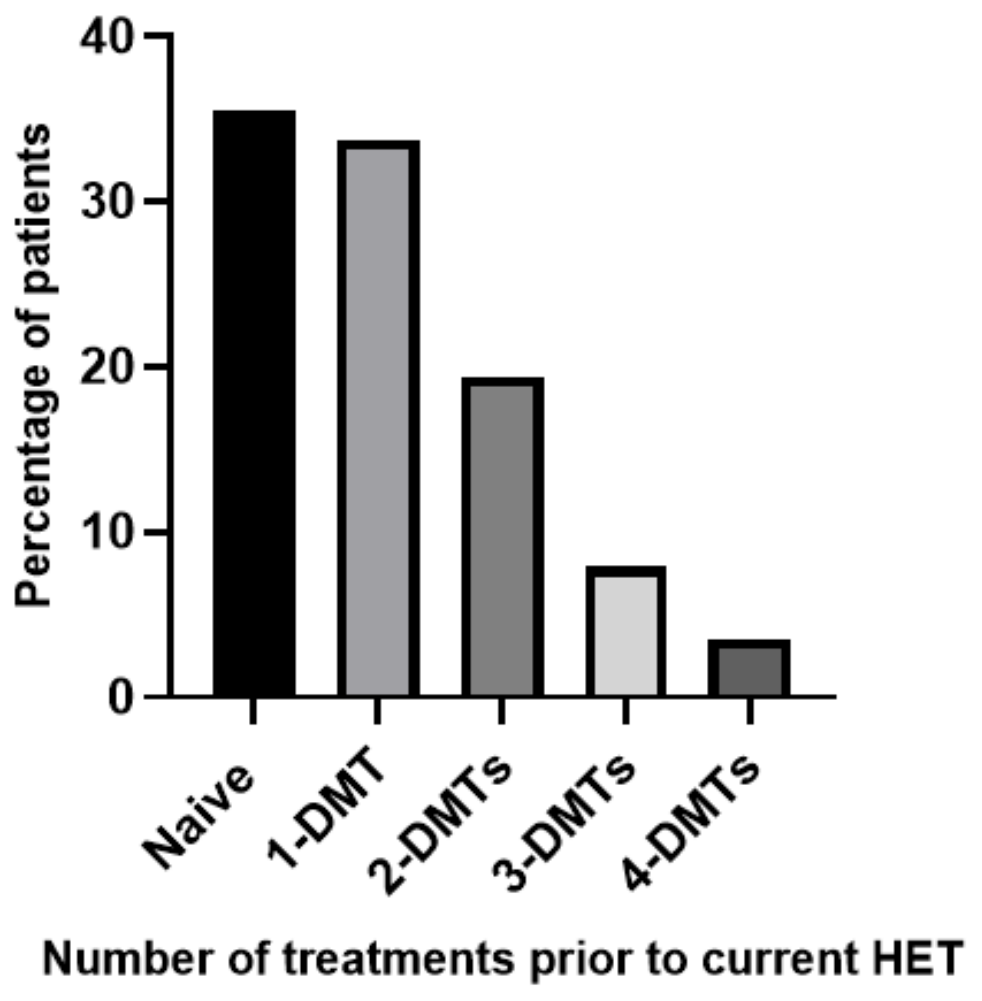
40
30
20
10
0
Percentage of patients
Naive
1-DMT
2-DMTs
3-DMTs
4-DMTs
Number of treatments prior to current HET

DMT: Disease-modifying therapies

Usage trend of HETs and rituximab

A total of 1690 patients included in RelevarEM started some treatment in the 5 years prior data cut off, of which 21.83% were HETs. Between 2015 and 2017 (P1) 729 patients included in RelevarEM started a new typo of treatment, of which 85 (11.65%) were HETs o rituximab. Between 2018 and 2020 (P2) 961 patients included in RelevarEM started a new treatment, of which 284 (29.55%) were HETs or rituximab. When comparing P2 with P1, a significant increase in the use of HETs is observed (p <0.01). The most commonly used HETs were alemtuzumab in 43 patients (50.59%) followed by natalizumab in 39 (45.88%) in P1, while cladribine was prescribed in 129 patients (45.20%) followed by natalizumab in 71 (25%) in P2. Interestingly, a decrease in the prescription of natalizumab and alemtuzumab was found when comparing P2 with P1. The increase in the prescription of HETs was mainly associated with the increase in ocrelizumab in P2 and the approval of cladribine in this period. (Table 2).

Table 2: Evaluation of the use of DMTs in different periods of time (n =1690)

Treatment	P1 (%)	P2 (%)	p-value
HETs + Rituximab	85 (11.65)¥	**284 (29.55)** ¥¥	p < 0.01*
Natalizumab	**39 (45.88)** †	71 (25.0)	p < 0.01*
Alemtuzumab	**43 (50.59)** †	33 (11.62)	p < 0.01*
Ocrelizumab	1 (1.18) †	**36 (12.68)**	p < 0.01*
Cladribina	-	**129 (45.42%)**	-
Rituximab (off label)	2 (2.35) †	15 (5.28)	p 0.37**
No HETs	644 (88.35)¥¥¥	677 (70.45)¥¥¥¥	
Interferon	**78 (12.11)** ††	58 (8.57) ††	p 0.042*
Glatiramer acetate	37 (5.75) ††	30 (4.43) ††	p 0.33*
Fingolimod	**338 (52.48)** ††	270 (39.88) ††	p < 0.01*
Dimethyl fumarate	101 (15.68) ††	**160 (23.63)** ††	p < 0.01*
Teriflunomide	88 (13.66) ††	**125 (18.46)** ††	p 0.021*

*Chi-square ** Fisher test. P1: Period between 2015 and 2017 P2: Period between 2018 and 2020. ¥ Percentage of patients treated with HETs over the total treatments in P1. ¥¥ Percentage of patients treated with no-HETs over the total treatments in P2. ¥¥¥ Percentage of patients treated with no-HETs over the total treatments in P1. ¥¥¥¥ Percentage of patients treated with no-HETs over the total treatments in P1. † Percentage of patients treated with each HET over the total of HETs in P1. †† Percentage of patients treated with each HET over the total of HETs in P1

Conclusions

- More than 35% of patients treated with HETs or rituximab were treatment naïve. Fingolimod was the most frequent treatment (32%) prior to current HET
- We have identified an increasing trend toward the use of HETs in Argentina in relation to the availability of the drugs. Furthermore, we found a statistically significant increase in the use of HETs in the last 5 years. In the first period (from 2015 to 2017), we observed a greater use of natalizumab and alemtuzumab. On the other hand, in the second analyzed period (from 2018 to 2020), we observed a decrease in the prescription of monoclonal antibodies and an increase in the use of cladribine and ocrelizumab.

References

Alonso, R., et. al. , 2021. Usage trend of oral drugs for multiple sclerosis patients in Argentina. Mult Scler Relat Disord 47, 102664.

Rojas, J.I., et. al. The Argentinean multiple sclerosis registry (RelevarEM): Methodological aspects and directions. Mult Scler Relat Disord 32, 133-137.

Rojas, J.I et. al Effectiveness and Safety of Early High-Efficacy Versus Escalation Therapy in Relapsing-Remitting Multiple Sclerosis in Argentina. Clinical neuropharmacology 45(3), 45-51.

Disclosures and funding

This research was supported by Novartis Argentina. The funder of the study had no role in data collection, data analysis nor data interpretation, Novartis collaborated in the writing of the report.

Visit the web at:

https://bit.ly/ectrims2022

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without written permission of the authors

Presenter email address: ricardoalonsohrm@gmail.com

Scan this QR code to download a copy Poster