P847

High Prevalence of Intolerability With Interferon Beta and Glatiramer Acetate in Patients With MS

Jong-Mi Lee[1], Jacqueline Nicholas[2], Carrie Hersh[3], Eddie Jones[4], James Pike[4], Patricia Dominguez-Castro[5], Vladimir Bezlyak[6], Carol Lines[6], Marina Ziehn[6], Gavin Giovannoni[7]

[1]Stanford Healthcare, Stanford, Palo Alto, CA, USA; [2]OhioHealth Multiple Sclerosis Center, Columbus, OH, USA; [3]Cleveland Clinic Lou Ruvo Center for Brain Health, Las Vegas, NV, USA. [4]Adelphi Real World, Manchester, UK; [5]Novartis Global Service Center, Dublin, Ireland; [6]Novartis Healthcare Private Limited, Hyderabad, India; [6]Novartis Pharma AG, Basel, Switzerland; [7]Queen Mary University of London, Barts and The London School of Medicines and Dentistry, London, UK

Introduction

- Interferon beta (IFNβ) and glatiramer acetate (GA) are injectable disease-modifying therapies (DMTs) commonly used as first-line treatment in multiple sclerosis (MS)[1]
- However, the administration of IFNβ and GA can often be associated with injection-site reactions (ISRs), that can continue throughout treatment[2]
- These ISRs may lead to poor adherence and affect disease outcomes[2]

Objective

- To describe the characteristics of patients with MS currently treated with first-line injectable IFNβ and GA including
 - The occurrence of intolerability, including ISRs, in patients who were either currently or previously treated with IFNβ and GA
 - Understand the treatment patterns of these patients

Methods

Study design

- This non-interventional, retrospective, real-world analysis included data from adult MS patients (aged, ≥18 years) in the Adelphi MS Disease-Specific Programme (MS-DSP) in the United States (US), United Kingdom (UK) and European Union (France, Germany, Italy and Spain) between 2016 and 2019
- The Adelphi DSP database is a disease specific, cross-sectional, multinational ongoing study (database) that captures real-world clinical practice and perceptions of MS patients, caregivers and physicians[3]

Results

- A total of 3964 patients with relapsing-remitting MS (RRMS; France: 247; Germany: 921; Italy: 429; Spain: 298; UK: 309; US: 1760) were on injectable DMTs
- The mean age of the total sample was 39.3 years, the majority of the sample were females 69% **(Table 1)**

Table 1. Patient demographics and baseline characteristics

Characteristics	All patients (N=3964)	France (N=247)	Germany (N=921)	Italy (N=429)	Spain (N=298)	UK (N=309)	US (N=1760)
Mean age (SD)	39.30 (11.76)	36.90 (11.33)	36.30 (10.92)	38.00 (10.66)	35.60 (9.82)	35.90 (10.03)	42.90 (12.10)
Sex, Female, n (%)	2716 (69)	181 (73)	612 (66)	288 (67)	204 (68)	224 (72)	1207 (69)
Age at RRMS diagnosis, Mean (SD)	33.80 (9.68)	32.30 (9.02)	31.60 (8.89)	33.30 (9.18)	31.90 (7.56)	30.40 (8.25)	36.40 (10.26)
Time since initial MS diagnosis (days), Mean (SD)	1799.40 (1995.42)	1795.00 (1968.00)	1445.60 (1711.40)	1599.70 (1625.33)	1535.50 (1488.89)	1863.30 (1951.73)	2089.90 (2265.27)
Most recent EDSS score, Mean (SD)	2.00 (1.53)	1.20 (1.40)	1.80 (1.20)	2.30 (1.44)	1.70 (1.17)	2.40 (0.00)	2.10 (1.74)
Number of relapses in the past 12 months, n (%)							
0	1933 (49)	137 (55)	448 (49)	180 (42)	156 (52)	166 (54)	846 (48)
1	961 (24)	46 (19)	261 (28)	131 (31)	107 (36)	80 (26)	336 (19)
2	321 (8)	28 (11)	101 (11)	41 (10)	17 (6)	26 (8)	108 (6)
3	63 (2)	3 (1)	13 (1)	2 (0)	3 (1)	1 (0)	41 (2)
≥4	40 (1)	0 (0)	3 (0)	3 (1)	0 (0)	0 (0)	34 (2)
Missing	646 (16)	33 (13)	95 (10)	72 (17)	15 (5)	36 (12)	395 (22)

EDSS, Expanded Disability Status Scale; RRMS, relapsing-remitting MS; SD, standard deviation

Most common injectables by drug class and country

- The most common injectables were IFNβ (56.4%), followed by GA (38.2%) and PEGylated IFNβ (5.5%)
- When segregated by country, IFNβ was the most common injectable therapy in every country except in the US, where GA had a higher prevalence **(Figure 1)**

Figure 1. Most common injectable per drug class for the total sample and segregated by country

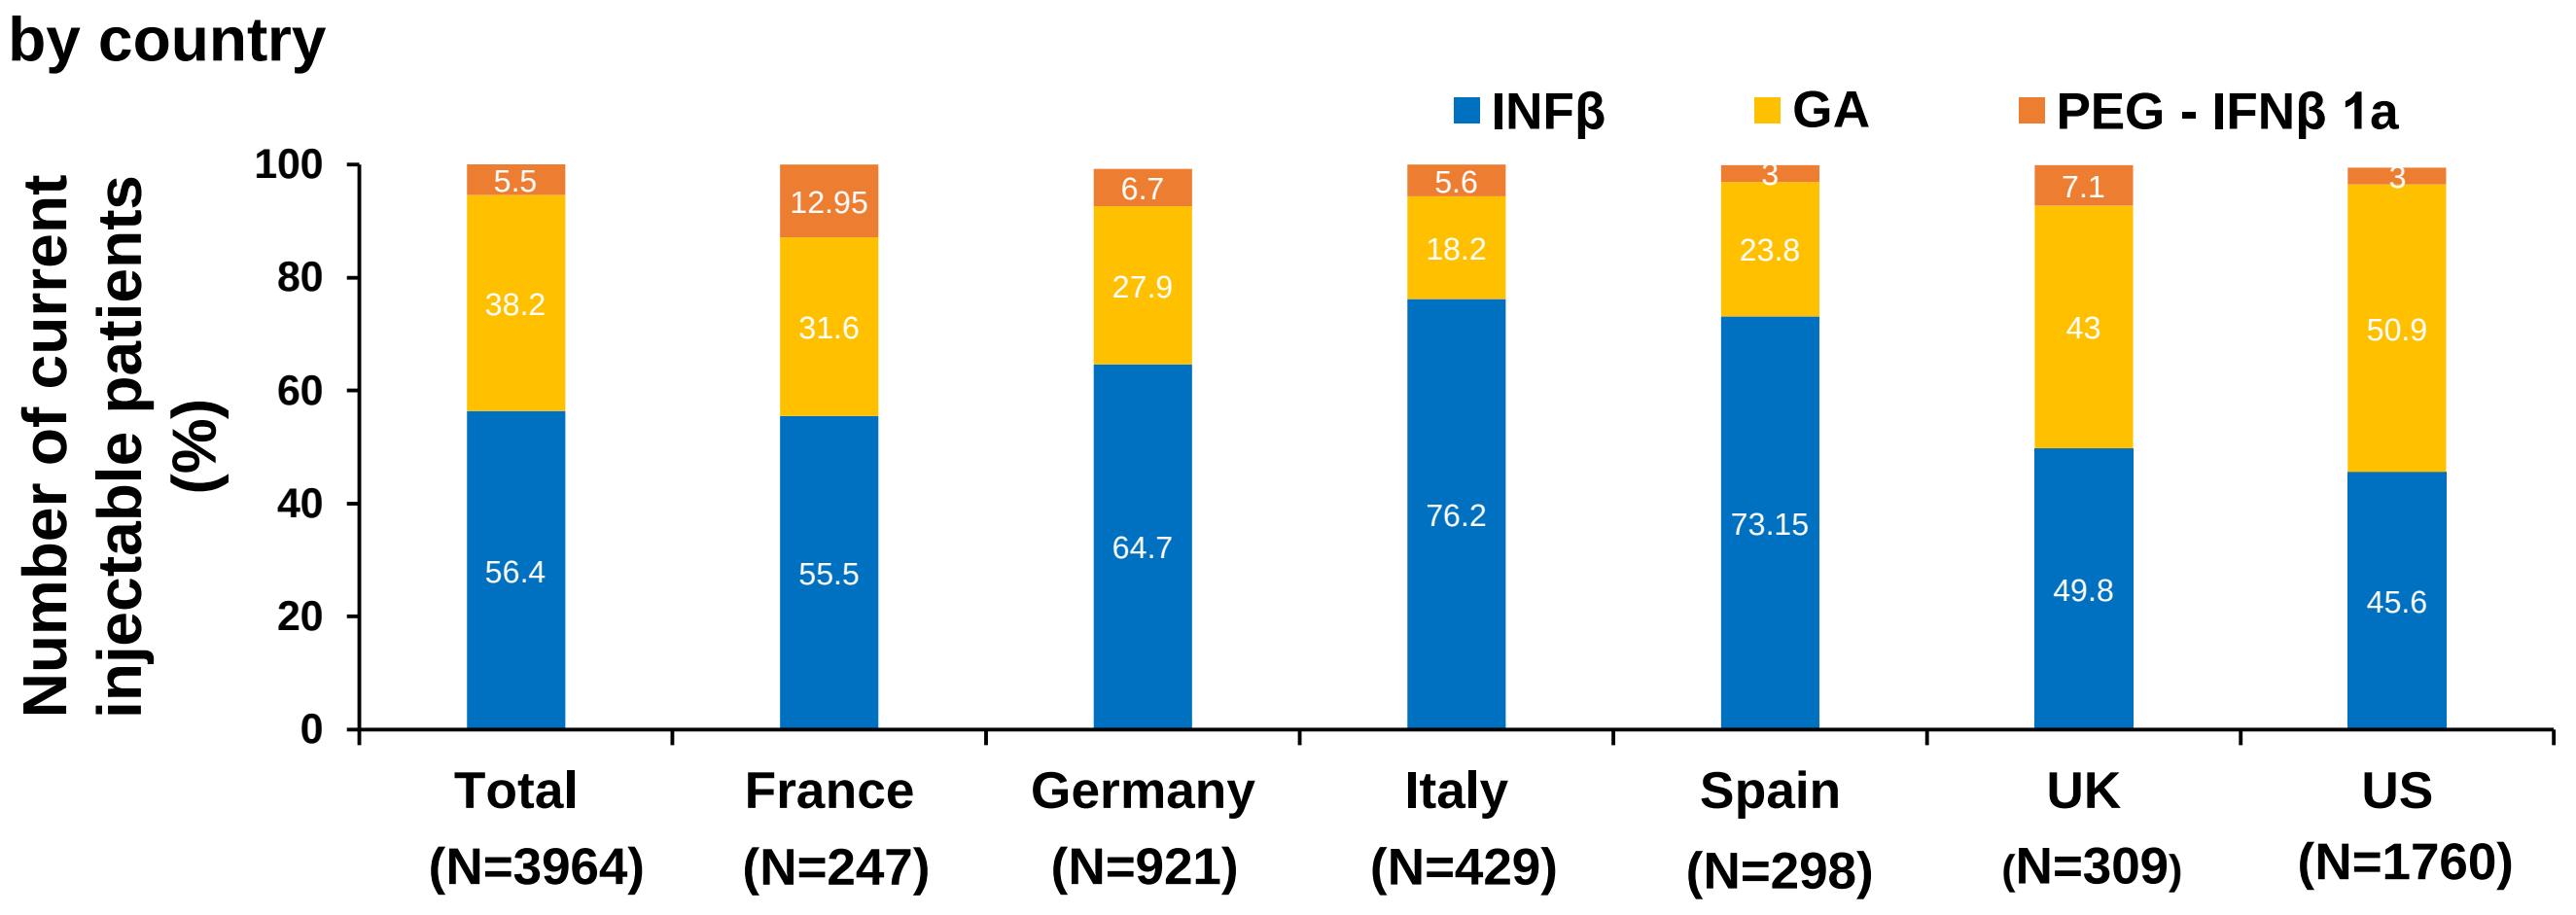
INFβ
GA
PEG - IFNβ 1a
Number of current injectable patients (%)
100
80
60
40
20
0
5.5
38.2
56.4
12.95
31.6
55.5
6.7
27.9
64.7
5.6
18.2
76.2
3
23.8
73.15
7.1
43
49.8
3
50.9
45.6
Total (N=3964)
France (N=247)
Germany (N=921)
Italy (N=429)
Spain (N=298)
UK (N=309)
US (N=1760)

Occurrence of adverse events (AEs)

- Of the patients who were on injectables, 29.1% reported AEs, with 13.2% experiencing ISRs which was the most common side effect reported followed by flu-like symptoms **(Figure 2)**

Figure 2. Occurrence of ISRs and other tolerability issues* (≥1%) in RRMS patients currently being treated with injectable DMTs (N=3964, %)

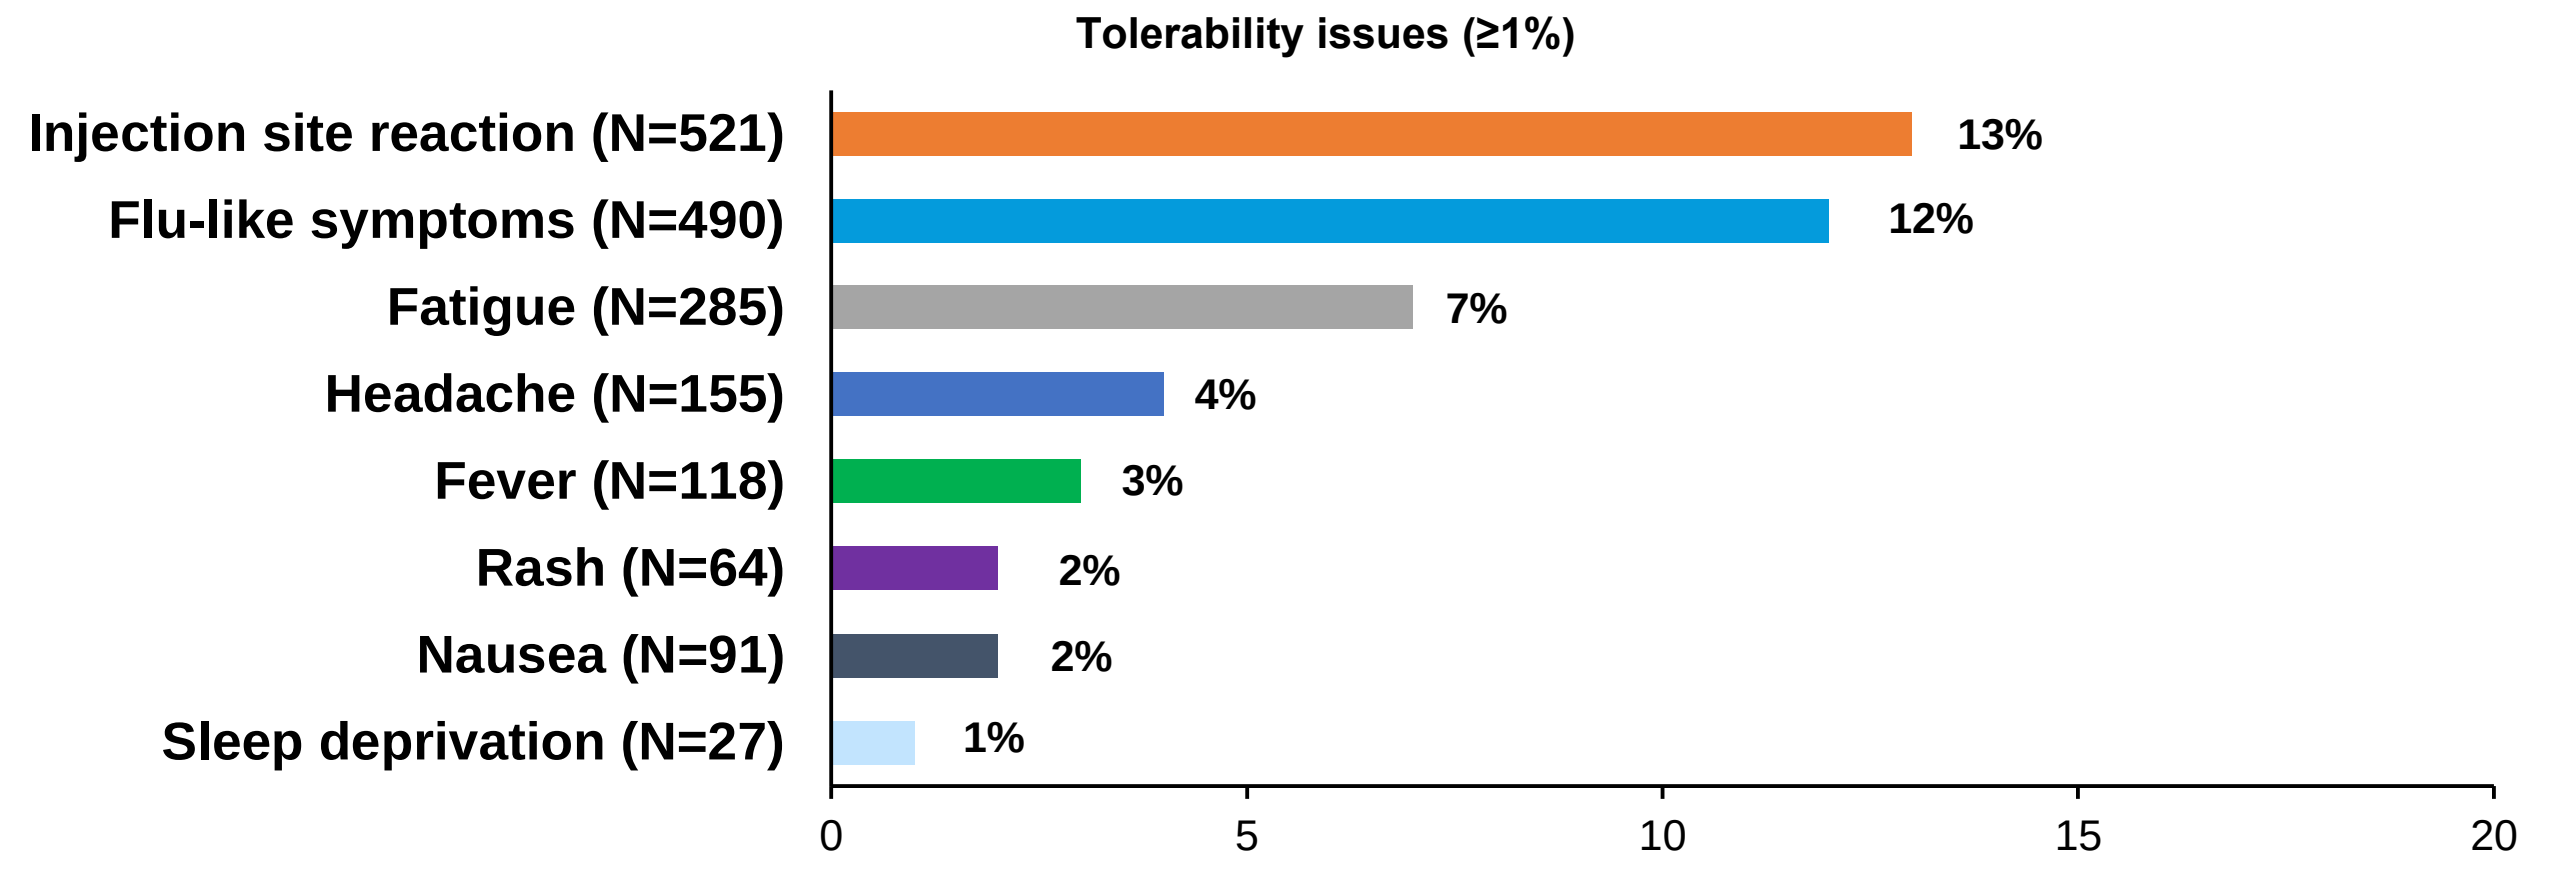
Tolerability issues (≥1%)
Injection site reaction (N=521)
Flu-like symptoms (N=490)
Fatigue (N=285)
Headache (N=155)
Fever (N=118)
Rash (N=64)
Nausea (N=91)
Sleep deprivation (N=27)
13%
12%
7%
4%
3%
2%
2%
1%
0
5
10
15
20

*Lymphocytopenia, thrombocytopenia, risk of progressive multifocal leukoencephalopathy, risk of malignancies, risk of infection, cardiac events, macular oedema, urticaria, urinary tract infections, slow/irregular heart beat, and GI risk accounted independently for <1% of tolerability issues

Occurrence of ISR by drug class and country

- GA accounted for the majority of the reported ISRs (51%; **Figure 3A**)
- When segregated by country, Italy (18%), the UK (16%), and the US (15%) had the highest share of ISR prevalence **(Figure 3B)**
- Physicians reported that 27.9% of patients who were on injectable DMTs wanted to have less frequent injections and/or reduced ISR/pain

Figure 3. Prevalence of ISRs per drug class (A) and by country* (B) in those patients currently using injectable IFNβ and GA DMTs and experiencing ISRs, N=521 (%)

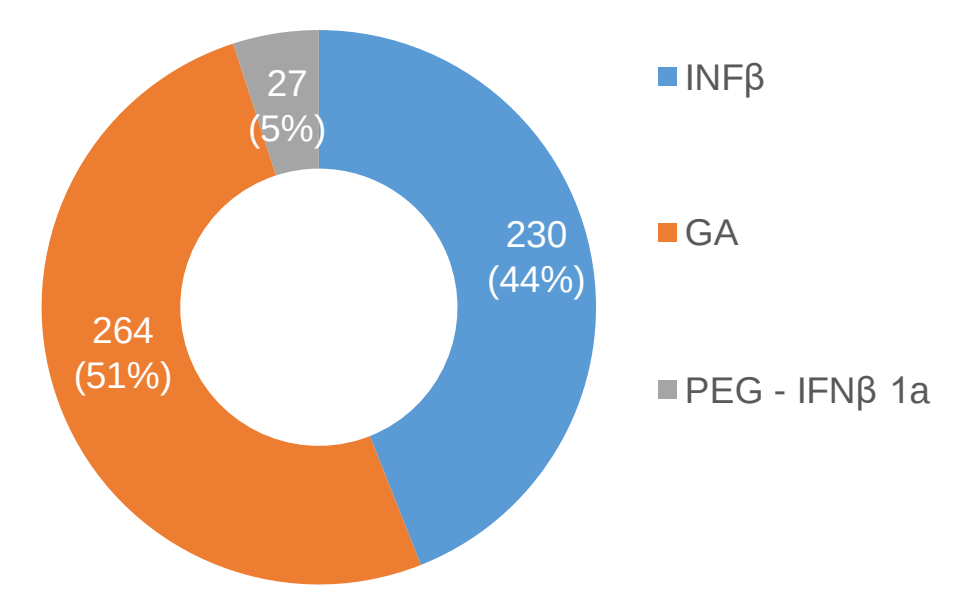
INFβ
GA
PEG - IFNβ 1a
27 (5%)
230 (44%)
264 (51%)

Figure 3A. Patients, n (%)

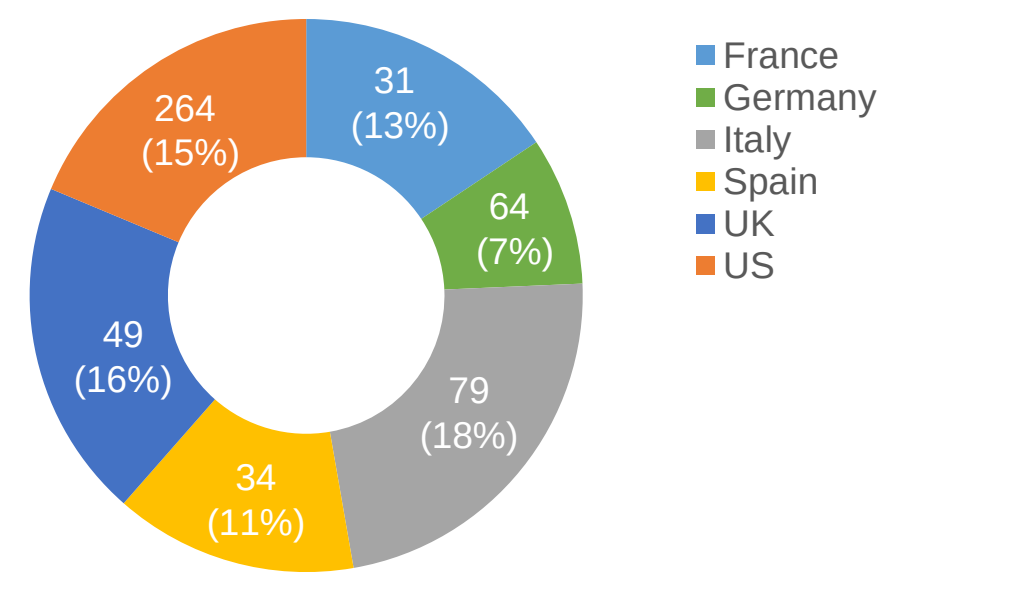
France
Germany
Italy
Spain
UK
US
31 (13%)
64 (7%)
79 (18%)
34 (11%)
49 (16%)
264 (15%)

Figure 3B. Patients, n (%)*

*This % is relating to ISRs experienced by patients in the individual countries.

Proportion of MS patients previously using injectable IFNβ and GA DMTs who switched treatment

- Of the 3002 patients who switched from an injectable DMT to their current treatment, 21% switched due to ISRs within the same time period **(Table 2)**
- Despite the high prevalence of ISRs, the majority of patients currently using injectable DMTs, also had an injectable DMT as their most recent previous therapy **(Figure 4)**

Table 2. Percentage of patients previously using injectable IFNβ and GA who switched due to ISR per country

	All patients (N=3002)	France (N=239)	Germany (N=707)	Italy (N=237)	Spain (N=196)	UK (N=237)	US (N=1386)
Injection Site Reaction	639	31	108	39	54	71	336
Proportion of switched injectable patients who switched off due to injection site reaction (%)	21	13	15	16	28	30	24

N= Patients who switched off an injectable from previous regimen to current treatment

Figure 4. Treatment Pathway showing the most recent previous treatment of those patients currently prescribed injectable DMTs

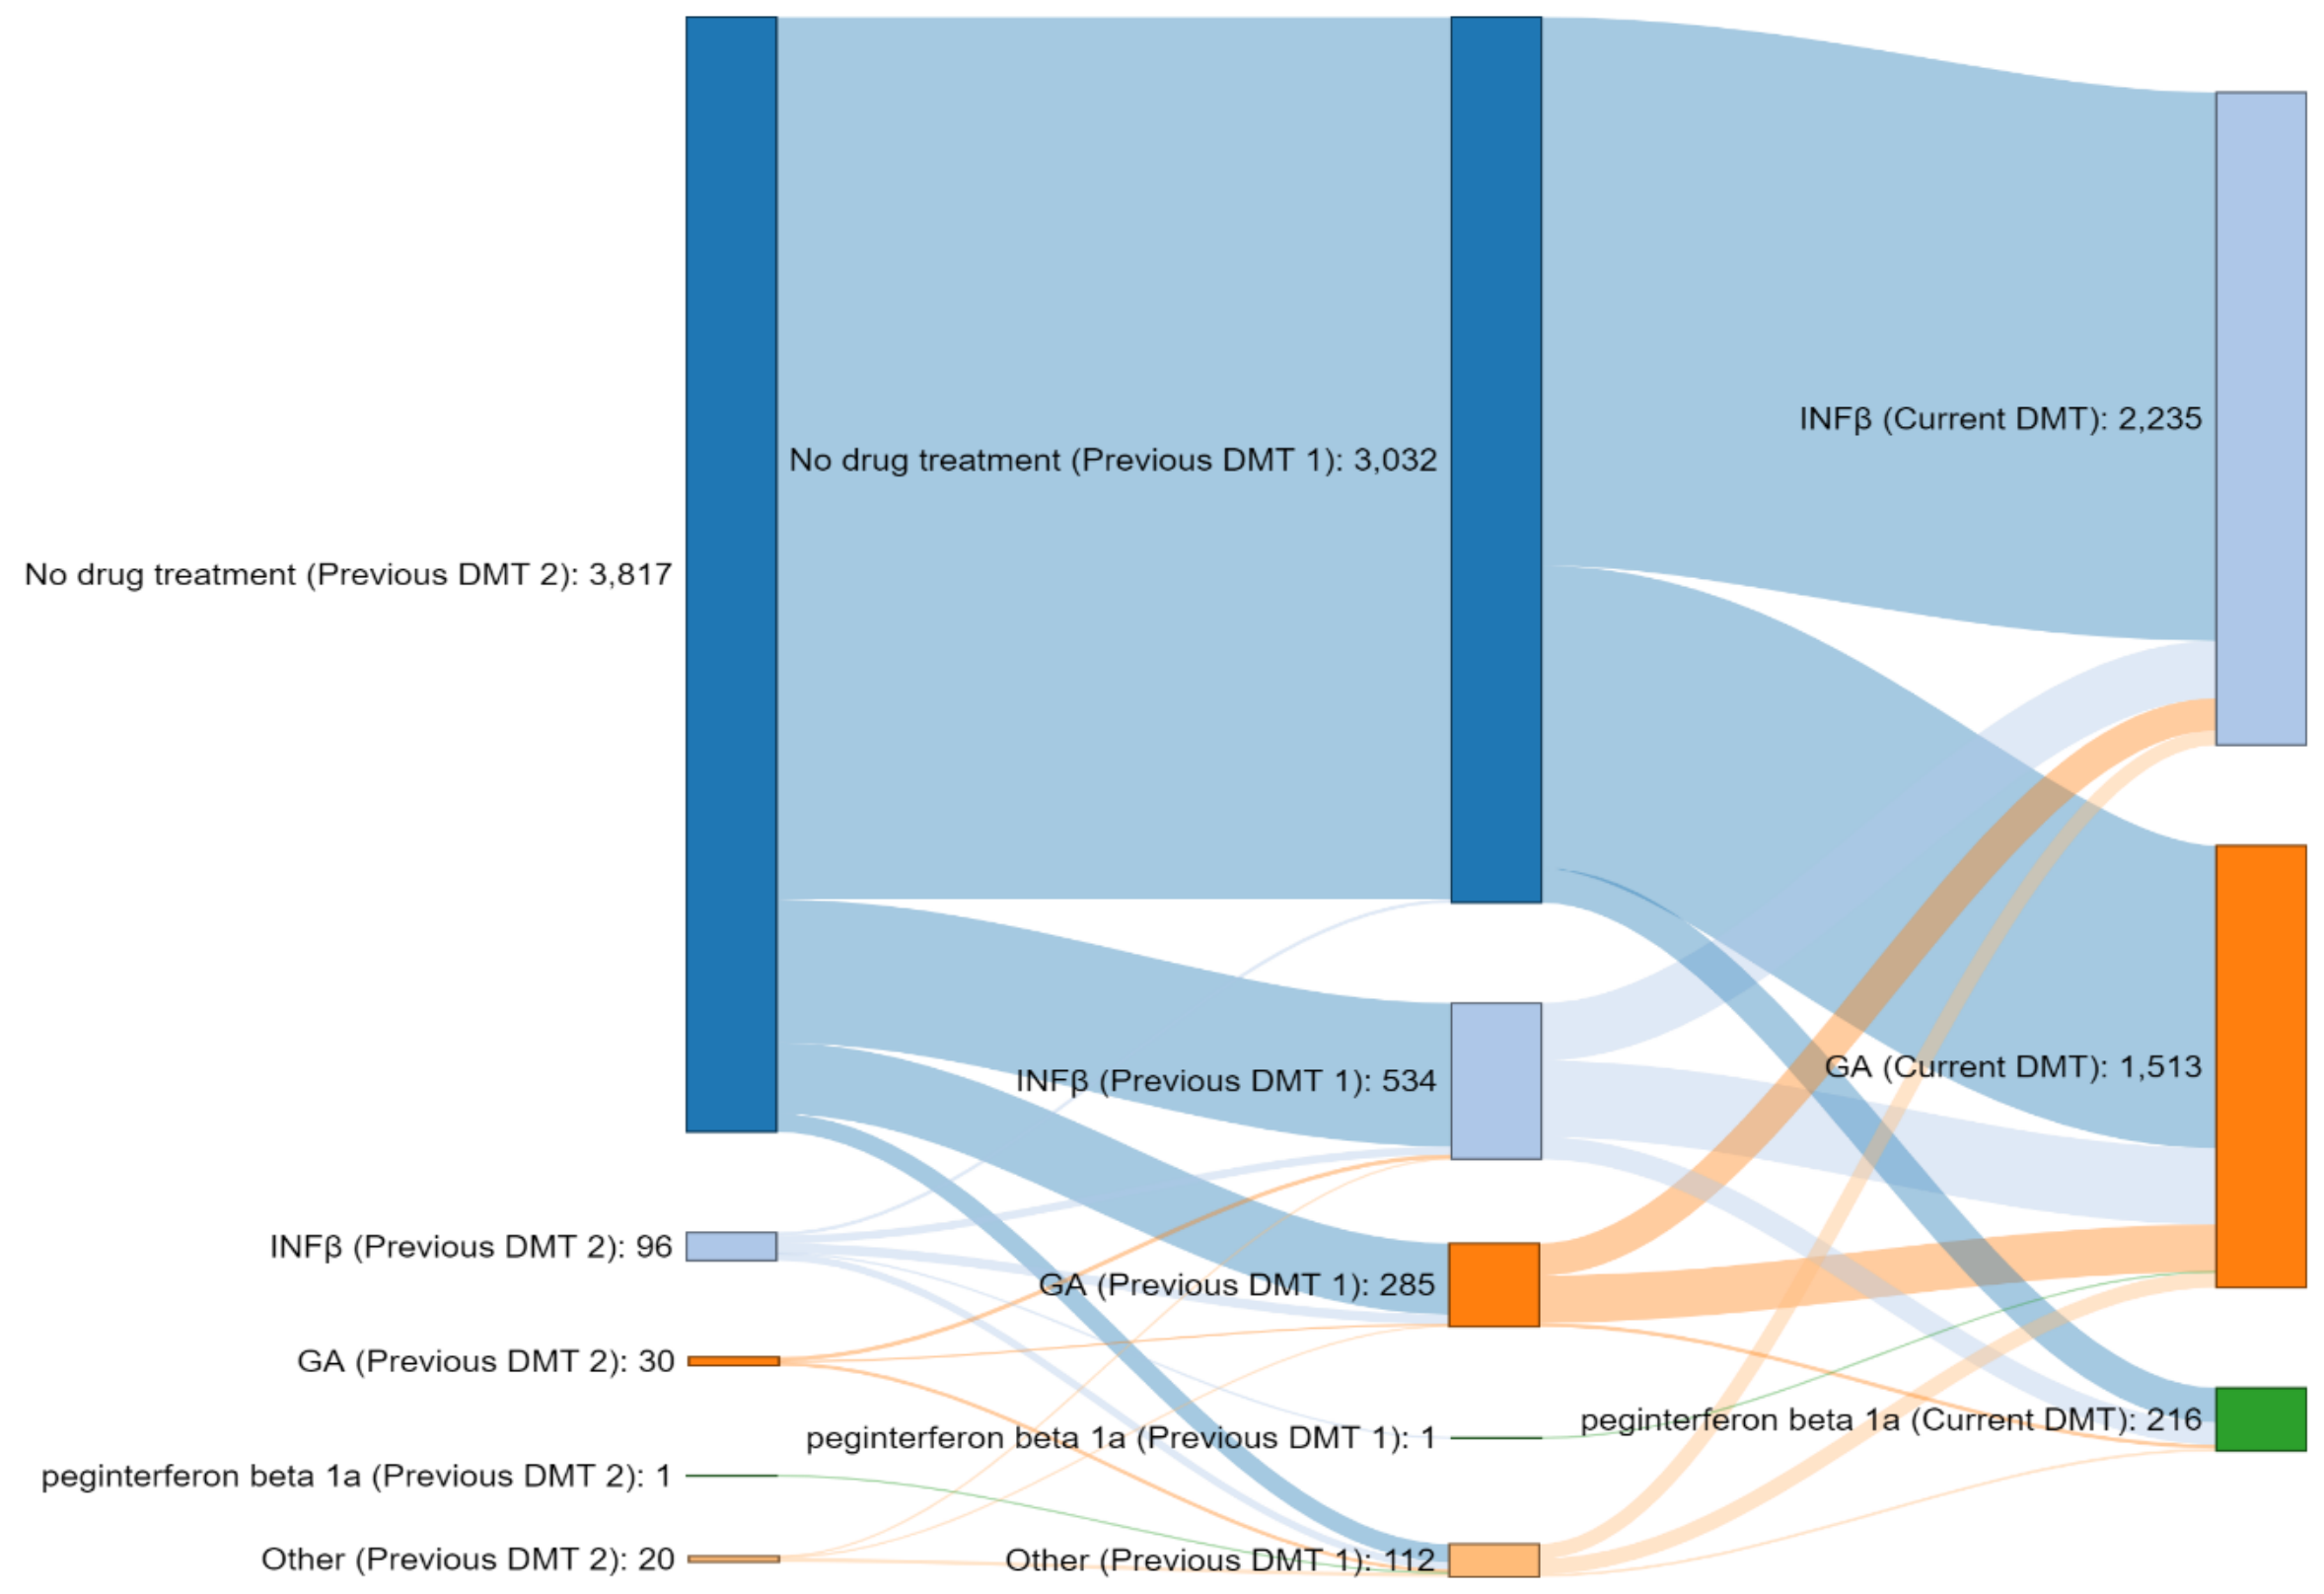
No drug treatment (Previous DMT 2): 3,817
INFβ (Previous DMT 2): 96
GA (Previous DMT 2): 30
peginterferon beta 1a (Previous DMT 2): 1
Other (Previous DMT 2): 20
No drug treatment (Previous DMT 1): 3,032
INFβ (Previous DMT 1): 534
GA (Previous DMT 1): 285
peginterferon beta 1a (Previous DMT 1): 1
Other (Previous DMT 1): 112
INFβ (Current DMT): 2,235
GA (Current DMT): 1,513
peginterferon beta 1a (Current DMT): 216

Summary

- Our results suggest that a large percentage of patients with MS on IFNβ and GA experience tolerability issues that lead to treatment discontinuation, with ISRs being the most common tolerability issue reported
- Despite the high prevalence of ISRs in those currently using injectable IFNβ and GA, many patients had been previously using injectable IFNβ and GA
- Future studies could evaluate whether the duration and frequency of ISRs are similar or different amongst available injectable DMTs
- Further assessment of the impact of ISRs on DMT adherence and persistence and associated patient outcomes is recommended

References

1. Melendez-Torres GJ, et al. *BMC Neurology* 2018; 18:162.
2. Menzin J, et al. *J Manag Care Pharm*. 2013. 19 (1 Suppl A):S24-40).
3. Anderson P, et al., *Curr Med Res Opin*. 2008. Nov;24(11):3063-72.

Disclosures

Jong-Mi Lee has served on advisory boards and speakers bureau for Biogen, Novartis, Sanofi Genzyme, Bristol Myers Squibb, and EMD Serono. **Jacqueline Nicholas** has received research grant support from Biogen, Novartis, PCORI, ADAMAS, Genzyme. She has received honoraria from consulting with Biogen, Bristol Myers Squib, EMD Serono, Genetech, Greenwich Biosciences and Novartis. She has received speaker fees from Alexion, Biogen, Bristol Myers Squibb, EMD Serono, Genetech, Novartis and Viela Bio. **Carrie M. Hersh** has received speaking, consulting, and advisory board fees from Genentech, Genzyme, Biogen, Novartis, EMD-Serono, Bristol Myers Squibb, and TG Therapeutics. She has received research support paid to her institution by Biogen, Novartis, Genentech, Patient-Centered Outcomes Research Institute (PCORI) and NIH - NINDS 1U01NS111678-01A1 sub-award. **Eddie Jones** and **James Pike** are full-time employees of Adelphi Real World Ltd, United Kingdom, a health care research consultancy. **Patricia Dominguez-Castro, Vladimir Bezlyak, Carol Lines and Marina Ziehn** are employees of Novartis. **Gavin Giovannoni** received consulting fee from AbbVie, Actelion, Atara Bio, Biogen, Celgene, Sanofi-Genzyme, Genentech, GlaxoSmithKline, Merck-Serono, Novartis, Roche and Teva, for research from Biogen, Roche, Merck, Merck-Serono, Novartis, Sanofi-Genzyme and Takeda.

Acknowledgements
The authors would like to thank **Emma Houchen, Sourav Biswas** and **Lukas Sobisek** from the Novartis Global Service Center, Dublin and Novartis Healthcare Private Limited, Hyderabad for their contribution to the development of the study documentation. The authors also acknowledge the following Novartis employees: **Bhavesh Kshirsagar** and **Saimithra Thammera** for medical writing assistance and coordinating author reviews. The final responsibility for the content lies with the authors

Poster presented at 37th Congress of the European Committee for Treatment and Research in Multiple Sclerosis (ECTRIMS), 13-15 October 2021
This study was sponsored by Novartis Pharma AG, Switzerland

Scan this QR code to download a copy of Poster