P533

Effect of Ofatumumab on Brain Volume Loss Versus Historical Placebo in Relapsing Multiple Sclerosis

Habib Ganjgahi[1], Bernd Kieseier[2], Dieter A. Häring[2], Wendy Su[2], Thomas E. Nichols[1]

[1]Oxford Big Data Institute, Li Ka Shing Centre for Health Information and Discovery, Nuffield Department of Population Health, University of Oxford, Oxford, UK; [2]Novartis Pharma AG, Basel, Switzerland

Introduction

- Ofatumumab, a fully human anti-CD20 monoclonal antibody, is approved for the treatment of relapsing multiple sclerosis (RMS) in adults in the US[1], EU[2], and other countries*
- In the phase 3 ASCLEPIOS I/II trials in patients with RMS, ofatumumab 20 mg administered subcutaneously every 4 weeks (q4w) was superior to oral teriflunomide 14 mg once daily, with significant reductions in relapse rates and conventional brain lesion numbers on magnetic resonance imaging (MRI), lower neurofilament light levels and greater delays in disability worsening[3]
- However, in the preplanned analysis (ASCELPIOS I/II trials), ofatumumab was not superior on whole brain volume loss (BVL) compared with the active control arm (teriflunomide); teriflunomide demonstrated superior efficacy on brain volume change in a prior placebo controlled trial[4]

*Australia, Canada, Singapore, Switzerland, UAE, Albania, Argentina, Japan and India.

Objective

- To assess the effect of ofatumumab on the percentage whole brain volume change (PBVC) over 2 years compared with historical placebo in patients with RMS

Methods

Study design

- MRI scans for RMS patients treated with ofatumumab were collected at baseline and at year 1 and 2 in the ASCLEPIOS I/II trials
- MRI scans from RMS patients treated with placebo were collected at baseline, month 6, and at year 1 and 2 in the FREEDOMS and FREEDOMS II trials, two fingolimod Phase 3 trials
 - Harmonization of MRI methodology was used to analyze PBVC and re-analyze lesion volumes and numbers across all trials at the Oxford BDI using SIENA as the MRI scans from the fingolimod and ofatumumab trials were assessed by different MRI reading centers[5]
- Statistical methods to allow causal inference in a non-randomized population include propensity methods or Bayesian adaptive regression trees (BART)
- In the past, BART has outperformed methods based on propensity scores and allowed for causal inference between non-randomized cohorts[6]

Endpoints

- The effect of ofatumumab versus historical placebo on PBVC over 1 and 2 years after initiating treatment while adjusting for baseline differences with regard to age, sex, duration of MS, disability level (Expanded Disability Status Scale [EDSS]), relapses in the last 2 years, the T2 volume and the number of gadolinium-enhancing (Gd+) T1 lesions using counterfactual inference based on BART
- A deep learning model has been trained to obtain the number of Gd-lesions and the T2 lesion volume across trials
- Correlation between absolute BVL and number of Gd-enhancing lesions and T2 lesion volume at baseline

Results

Patient demographics and baseline characteristics

- The mean age (years) was 37.9 in the ofatumumab group (N=681) versus 38.2 in the historical placebo group (N=530); in both groups, the majority of the patients were female (65.1% and 76.8%, respectively; **Table 1**)
- The baseline number (mean) of Gd-enhancing lesions was significantly lower in the ofatumumab group (2.45) than in the historical placebo group (16.62) (**Table 1, Figure 1**)
- The T2 lesion volume (mL) at baseline was higher in the placebo than in the ofatumumab group (5.24 versus 10.73, respectively; **Table 1, Figure 2**)

Table 1. Patient demographics and baseline characteristics

Characteristics	Ofatumumab (N=681)	Placebo (N=530)
Age, years, mean (SD)	37.92 (9.23)	38.20 (8.65)
Sex, male, N (%)	238 (34.9)	123 (23.2)
Duration of MS, mean (SD)	9.16 (7.94)	10.72 (8.47)
Baseline EDSS score, mean (SD)	2.85 (1.34)	2.39 (1.28)
Relapses in the last 2 years, mean (SD)	2.01 (1.26)	2.17 (1.31)
Number of Gd-enhancing lesions, n, mean, (SD)	2.45 (4.57)	16.62 (16.45)
Baseline normalized brain volume, cm³, mean (SD)	1543.51 (87.81)	1532.18 (87.08)
Baseline T2-weighted lesion volume, mL, mean (SD)	10.73 (10.88)	5.24 (6.21)

Figure 1. Number of Gd-enhancing lesions at baseline

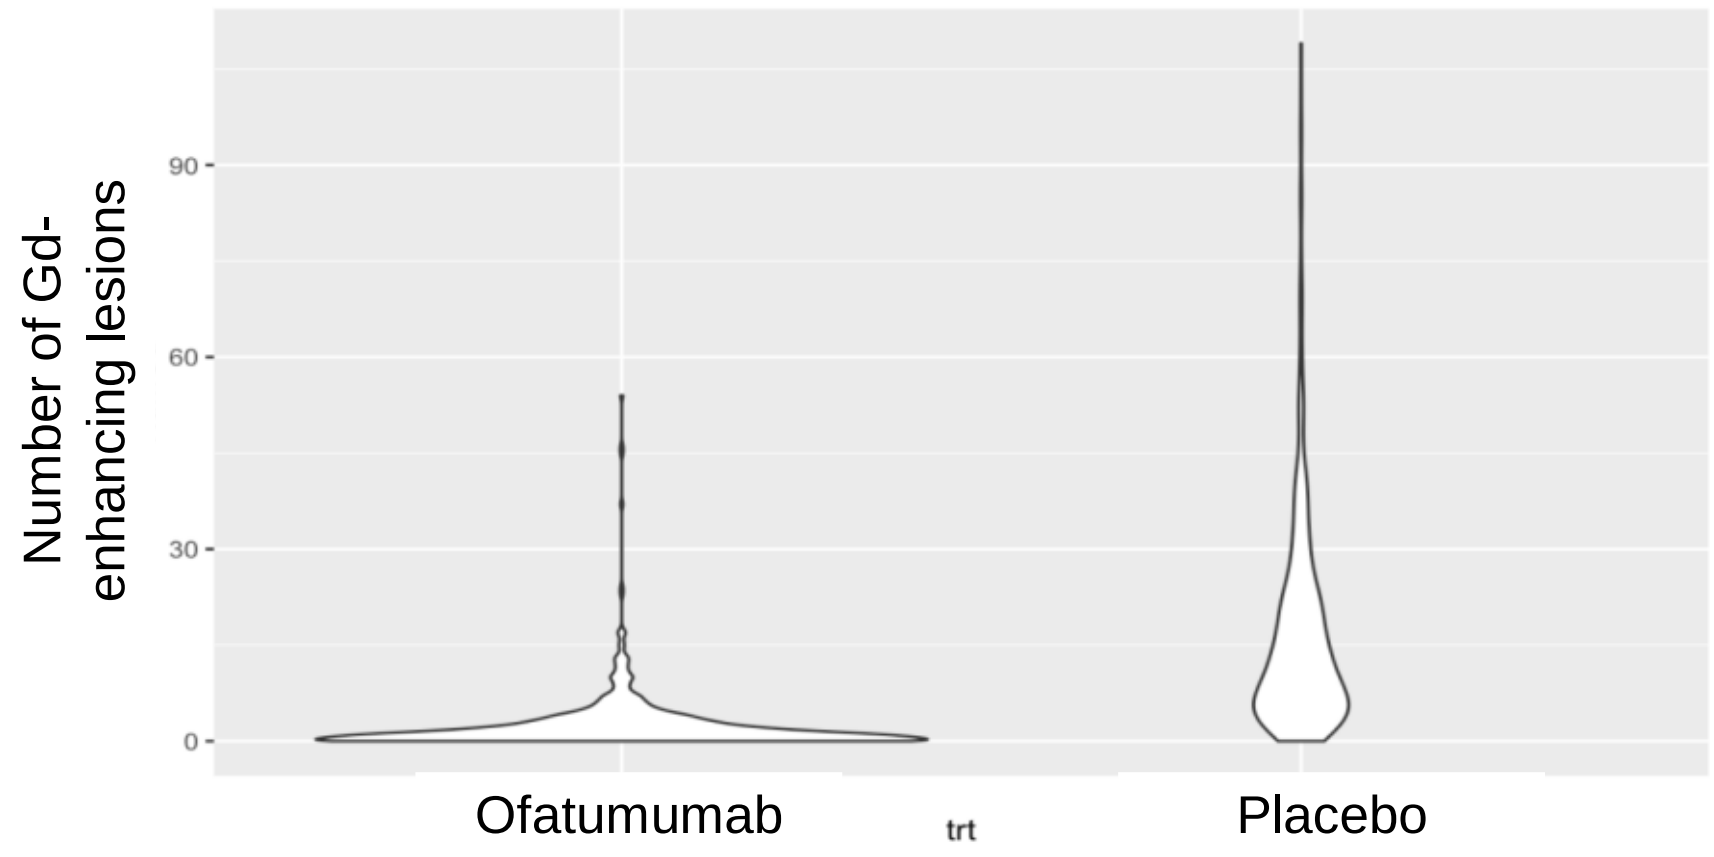

Number of Gd-enhancing lesions
90
60
30
0
Ofatumumab
trt
Placebo

Figure 2. T2 lesion volume at baseline

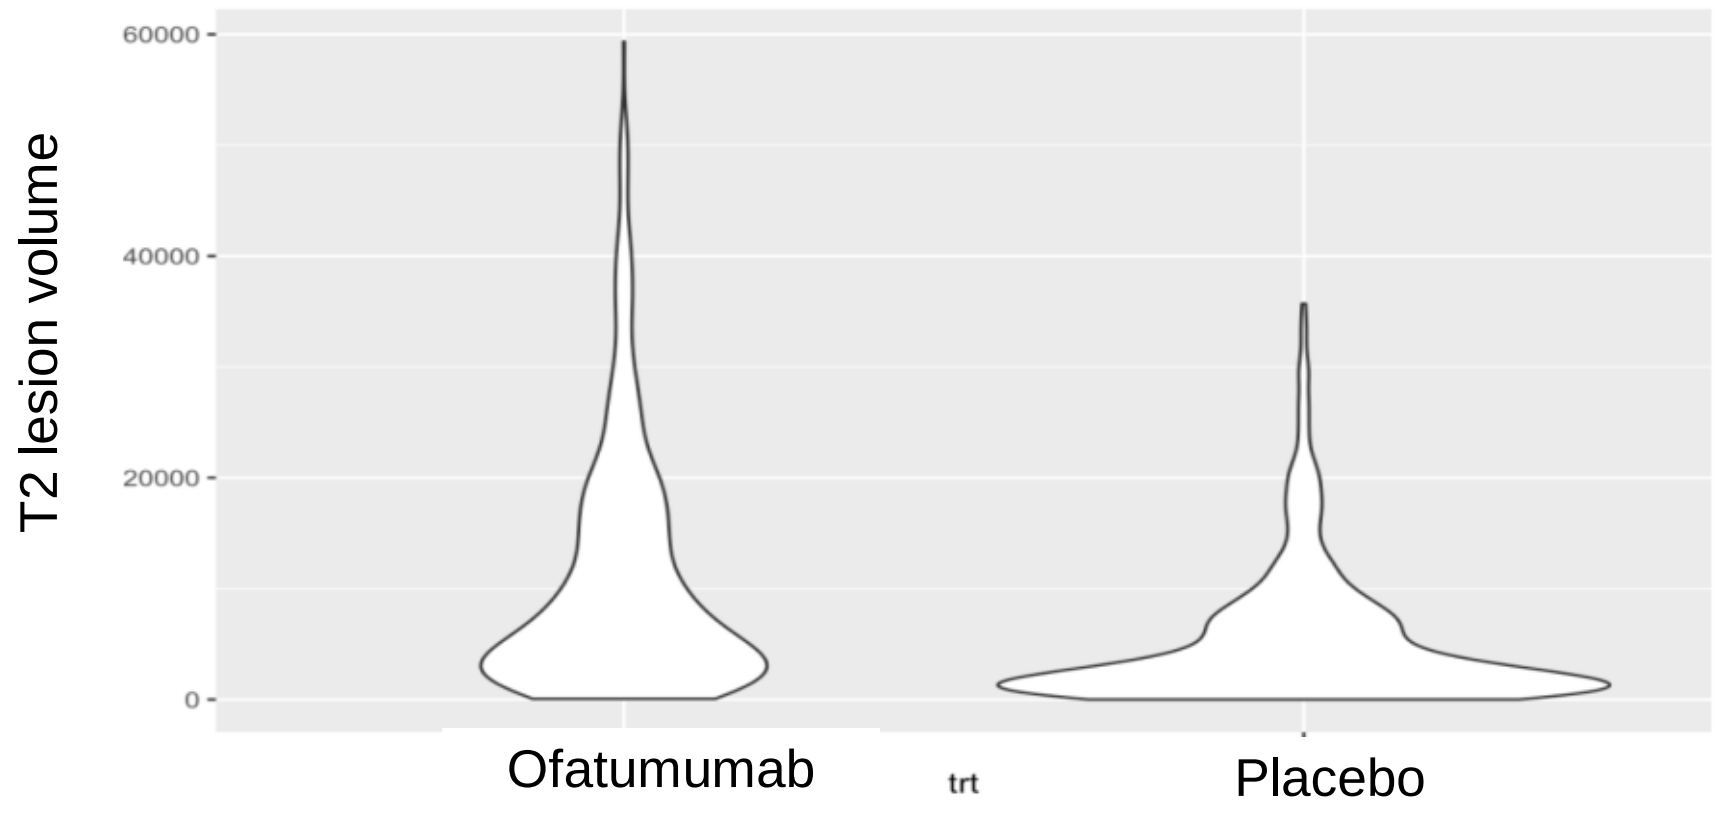

T2 lesion volume
60000
40000
20000
0
Ofatumumab
trt
Placebo

EDSS, Expanded Disability Status Scale; SD, Standard deviation

Effect of ofatumumab on the PBVC over 2 years compared to historical placebo

- The PBVC was lower with ofatumumab compared with placebo over 1 year (−0.57% versus −0.58%; p=0.431), and over 2 years (−1.00% versus −1.25%; p=0.015*) (**Figure 3**)

Figure 3. Brain volume change

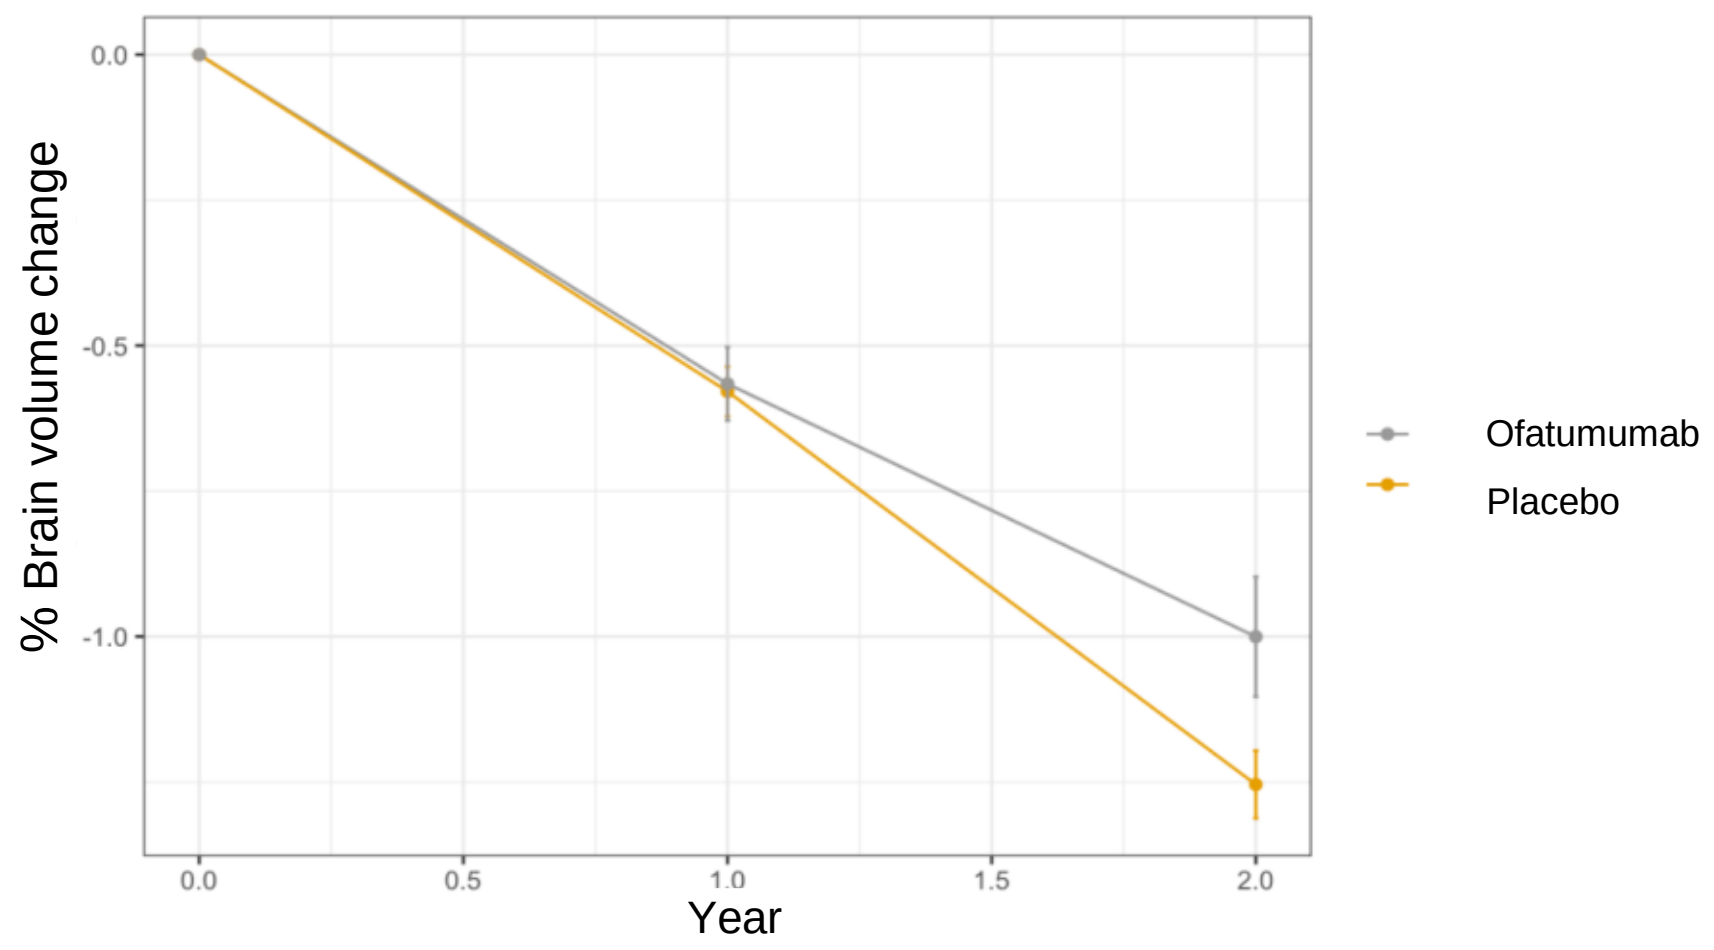

% Brain volume change
0.0
-0.5
-1.0
0.0
0.5
1.0
1.5
2.0
Year
Ofatumumab
Placebo

*Statistically significant PVBC difference at year two (-0.254)

Correlation between BVL and baseline lesion burden

- As shown in **Figure 4**, PBVC in the historical placebo group showed a moderate dependency on baseline inflammatory activity (number of Gd+ lesions at baseline)
 - In the ofatumumab group, a distinct change in slope is notable in patients who had many (>11) Gd-enhancing lesions at baseline, consistent with the hypothesis of 'pseudo-atrophy' and resolution of brain edema caused by the anti-inflammatory treatment
- **Figure 5** shows the significant dependency of PBVC on baseline T2 lesion volume (consistent with Radue EW et al., 2015[7])
- The treatment benefit of ofatumumab on PBVC was most apparent in those with a substantial T2 lesion volume at baseline **(Figure 5)**

Figure 4. Interdependence of PBVC and number of Gd+ lesions

PBVC by quartile of Gd+ lesion number at baseline

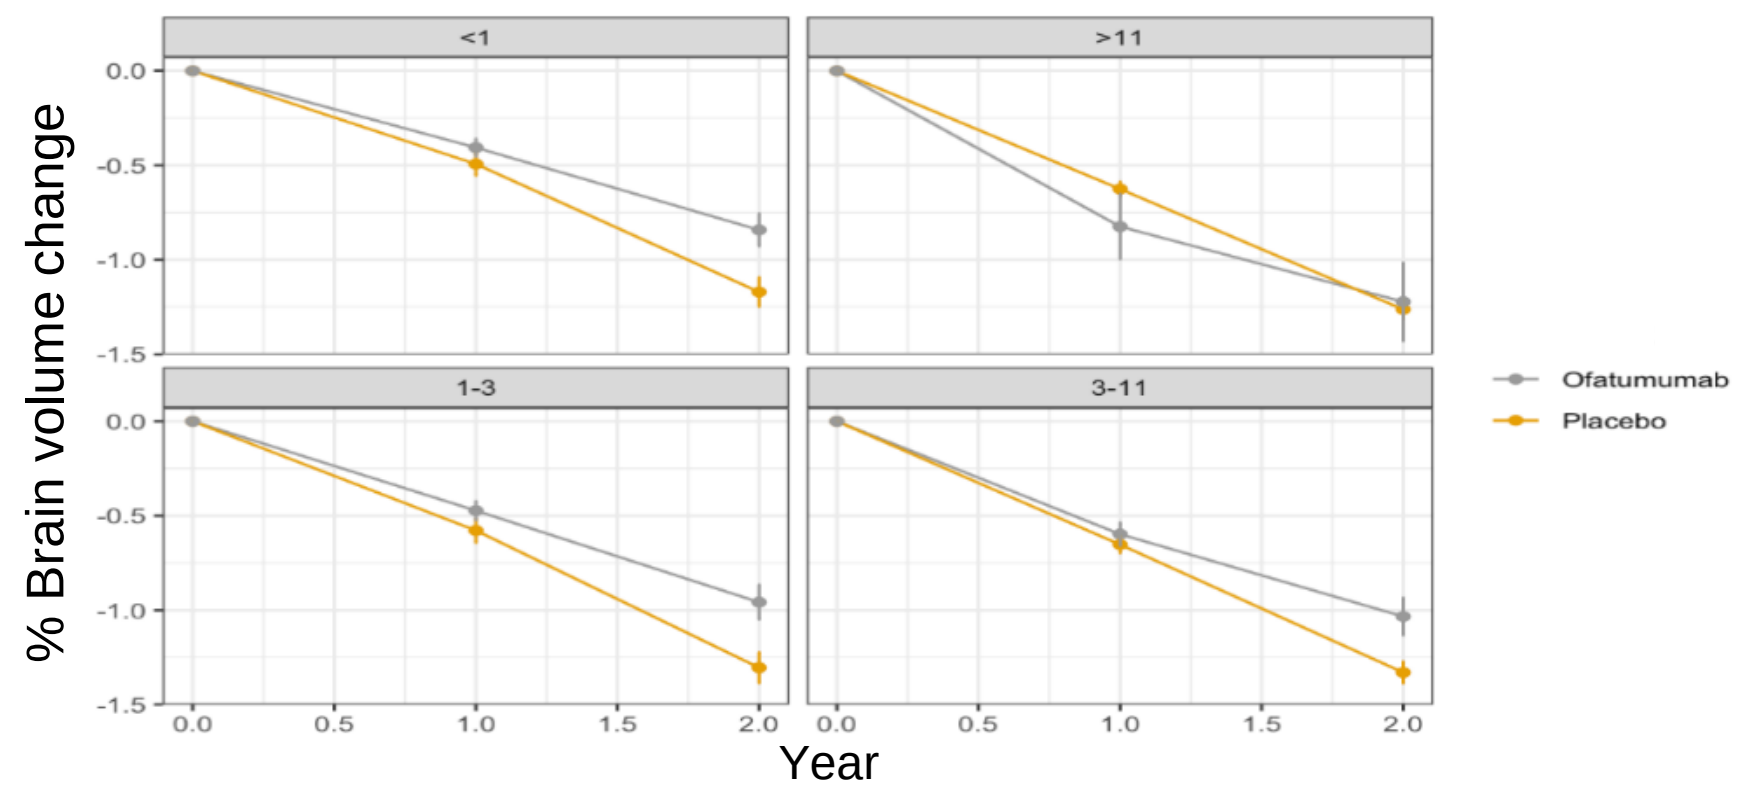

% Brain volume change
<1
>11
1-3
3-11
Year
Ofatumumab
Placebo

Figure 5. Interdependence of PBVC and T2 lesion volume

PBVC by quartile of T2 lesion volume at baseline

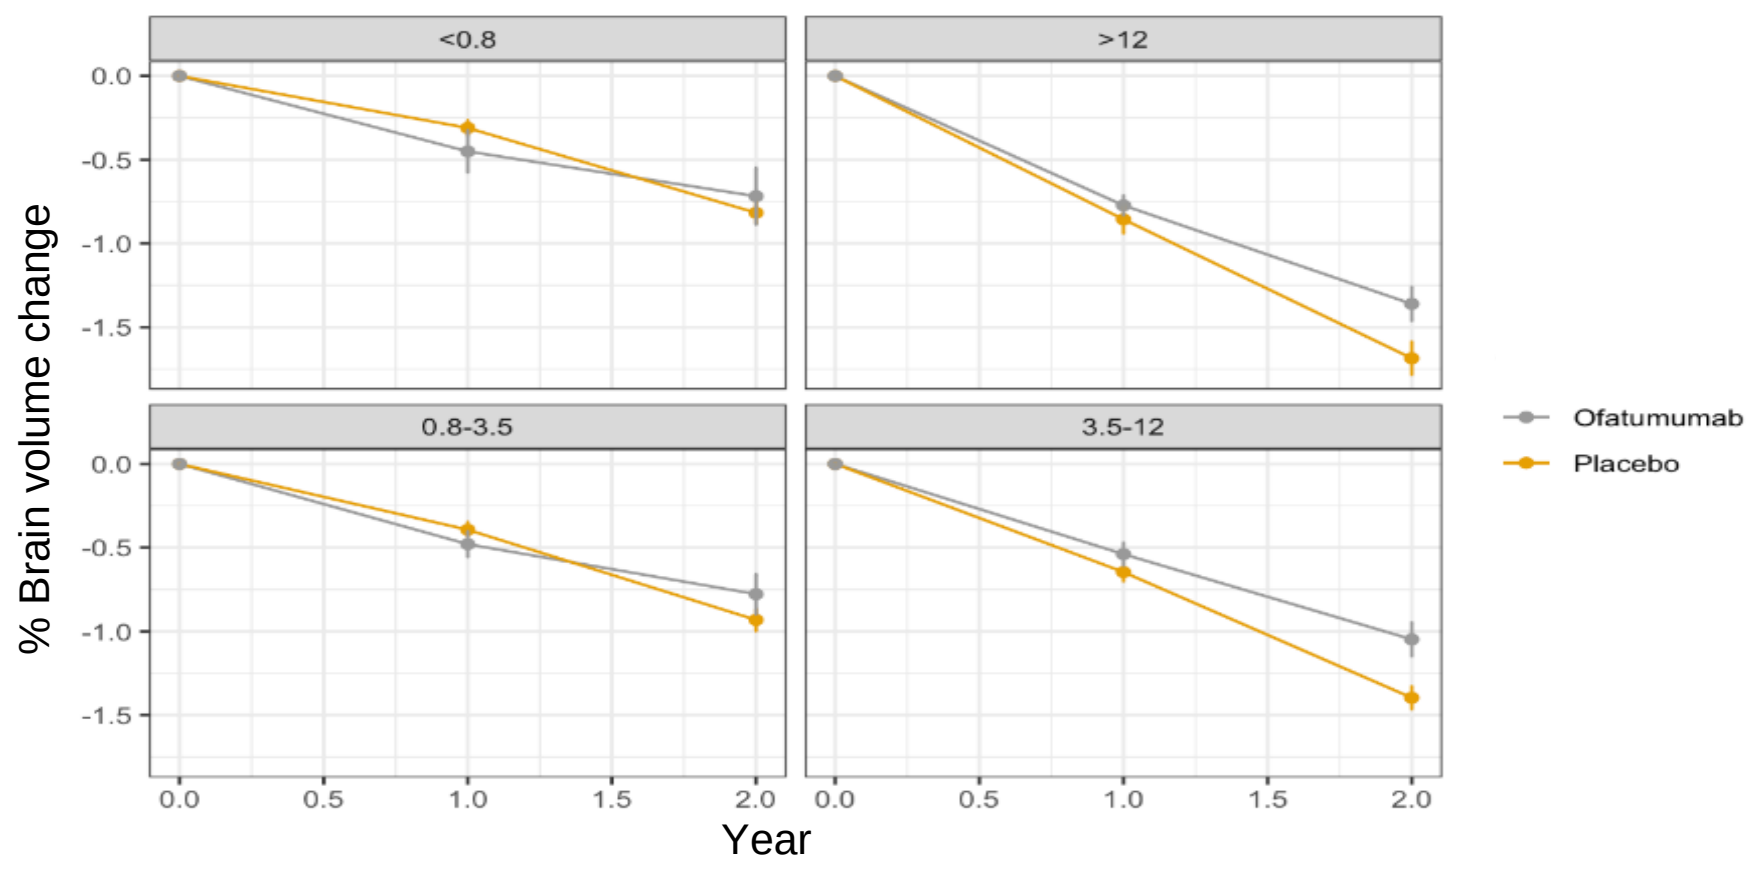

% Brain volume change
<0.8
>12
0.8-3.5
3.5-12
Year
Ofatumumab
Placebo

Gd+, gadolinium-enhancing; PBVC, percentage whole brain volume change

Summary

- This is the first comparison of an anti-CD20 mAb versus historical placebo in MS
- BART and a harmonized MRI methodology were used to overcome the ethical limitations of testing ofatumumab versus placebo
- The harmonized MRI methodology was used to assess the number of Gd-lesions, the T2 lesion volume and the brain volume change to minimize methodological differences between trials
- In the placebo group, PBVC depended more on the T2 lesion volume than the number of Gd-enhancing lesions (in line with Radue EW et al. 2015[7])
- The dependence of the PBVC on the T2 lesion volume is supportive of the topographical hypothesis of MS,[8] which hypothesizes that the progression recapitulates prior damage to the CNS, even if this was initially clinically asymptomatic
- Ofatumumab significantly reduced brain volume change versus historical placebo over 2 years; the change seen over 1 year might reflect pseud-atrophy

References

1. Kesimpta [prescribing information]. East Hanover, NJ: Novartis Pharmaceuticals Corporation; 2020.
2. Kesimpta. EMA 2021: Kesimpta, INN-ofatumumab (europa.eu)
3. Hauser SL et al. *N Engl J Med* 2017;376:221-234
4. Sprenger T et al. *MSJ* 2020,26(10) 1207-1216
5. Schach S et al. *Mult Scler.* 2007;13(4):509-16.
6. Hill JL et al. *JCGS 2011,* 20(1):217-240
7. Radue EW et al. *Neurology.* 2015;84:784–793
8. Krieger SC et al. *Neuroinflamm* 2016;3:e279

Disclosures

Dieter Häring, Wendy Su, and Bernd Kieseier are employees of Novartis. **Habib Gangjahi and Thomas E. Nichols** are supported by Novartis-funded Novartis-Oxford BDI collaboration.

Acknowledgments
The authors acknowledge the following Novartis employees: **Bhavesh Kshirsagar** and **Paul Coyle** for medical writing assistance and coordinating author reviews. The final responsibility for the content lies with the authors.
Poster presented at 37th Congress of the European Committee for Treatment and Research in Multiple Sclerosis (ECTRIMS), 13-15 October 2021
This study was sponsored by Novartis Pharma AG, Switzerland

Scan this QR code to download a copyPoster