REH25

Orchestrating a New Path for Multiple Sclerosis Rehabilitation: Empowering Patients Through Both Physical and Music Therapies

Megan Weigel[1], Wendy Su[2], Renée Fleming[3], Brian Hutchinson[4], Wendy L Magee[5]

[1]First Coast Integrative Medicine, Jacksonville Beach, FL, USA; [2]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [3]John F. Kennedy Center for the Performing Arts, Washington, DC, USA; [4]MS Achievement Center, Sacramento, CA, USA; [5]Temple University, Philadelphia, PA, USA

Introduction

- As multiple sclerosis (MS) progresses, a multidisciplinary approach is required to provide supportive therapy for the full range of symptoms[1-3]
- Although disease-modifying therapies (DMTs) delay disability progression (**Figure 1**), symptoms (eg, ambulation, cognition, fatigue, depression) may continue to worsen[1-3]
- Overall, 33-88% of people with MS seek complementary and alternative therapies[4,5]
- Physical therapy (PT) and music therapy (MT) may improve well-being and health-related quality of life (HRQoL) in MS
 - Access to PT and MT is limited; nonpharmacological healthcare services are often underused owing to lack of awareness and financial support[6]

Objective

- Review the role of nonpharmacological therapies in MS, focusing on PT and MT

Methods

- Narrative review of published literature relating to use and effectiveness of PT and MT in people with MS
- MEDLINE searched without date restriction to identify studies; important themes and research studies identified by a panel of PT and MT experts

Results

Physical therapy

- PT includes use of physical methods, such as exercise and neurofacilitation, to improve/regain or maintain movement, strength, endurance and flexibility
 - Early intervention may help to slow the loss of motor skills[7]
- PT can lead to improvements in mobility and balance
 - A systematic review of 16 randomized controlled trials showed that treadmill training 3 times per week for 8 weeks improved walking endurance by 26.5 m from Baseline[8]
 - Exercise intervention to improve core strength and balance led to an increase of 16.4 m in the 2 minute walk test at 30 weeks versus controls, who received standard of care on walking therapy[9]
- PT can also provide clinically meaningful improvements in fatigue, mood, cognition and HRQoL
 - In a Cochrane review of 26 clinical studies, exercise therapy led to a clinically important reduction in fatigue versus nonexercise interventions ($p<0.01$)[10]
 - 20 people with MS, who performed high-intensity resistance training twice a week for 12 weeks, had significant reductions in anxiety ($p=0.002$), depression ($p=0.019$) and fatigue ($p=0.001$); and improvements in cognitive processing speed ($p=0.04$)[11]
- There are few, if any, negative side effects of PT[12]

Music therapy

- MT addresses active patient-centered health goals through playing music, singing, songwriting, or listening and responding to music[13]
- MT may help to improve a range of functional and psychosocial domains
 - Listening to or performing music integrates functioning across multiple neurological domains (eg, cognitive, motor, language, auditory, emotional)[14,15]
- Entrainment, the use of rhythm-based training to improve gait and mobility, can successfully combine MT with PT to help patients with MS[16]

Figure 1. FDA-approved DMTs for relapsing and progressive MS

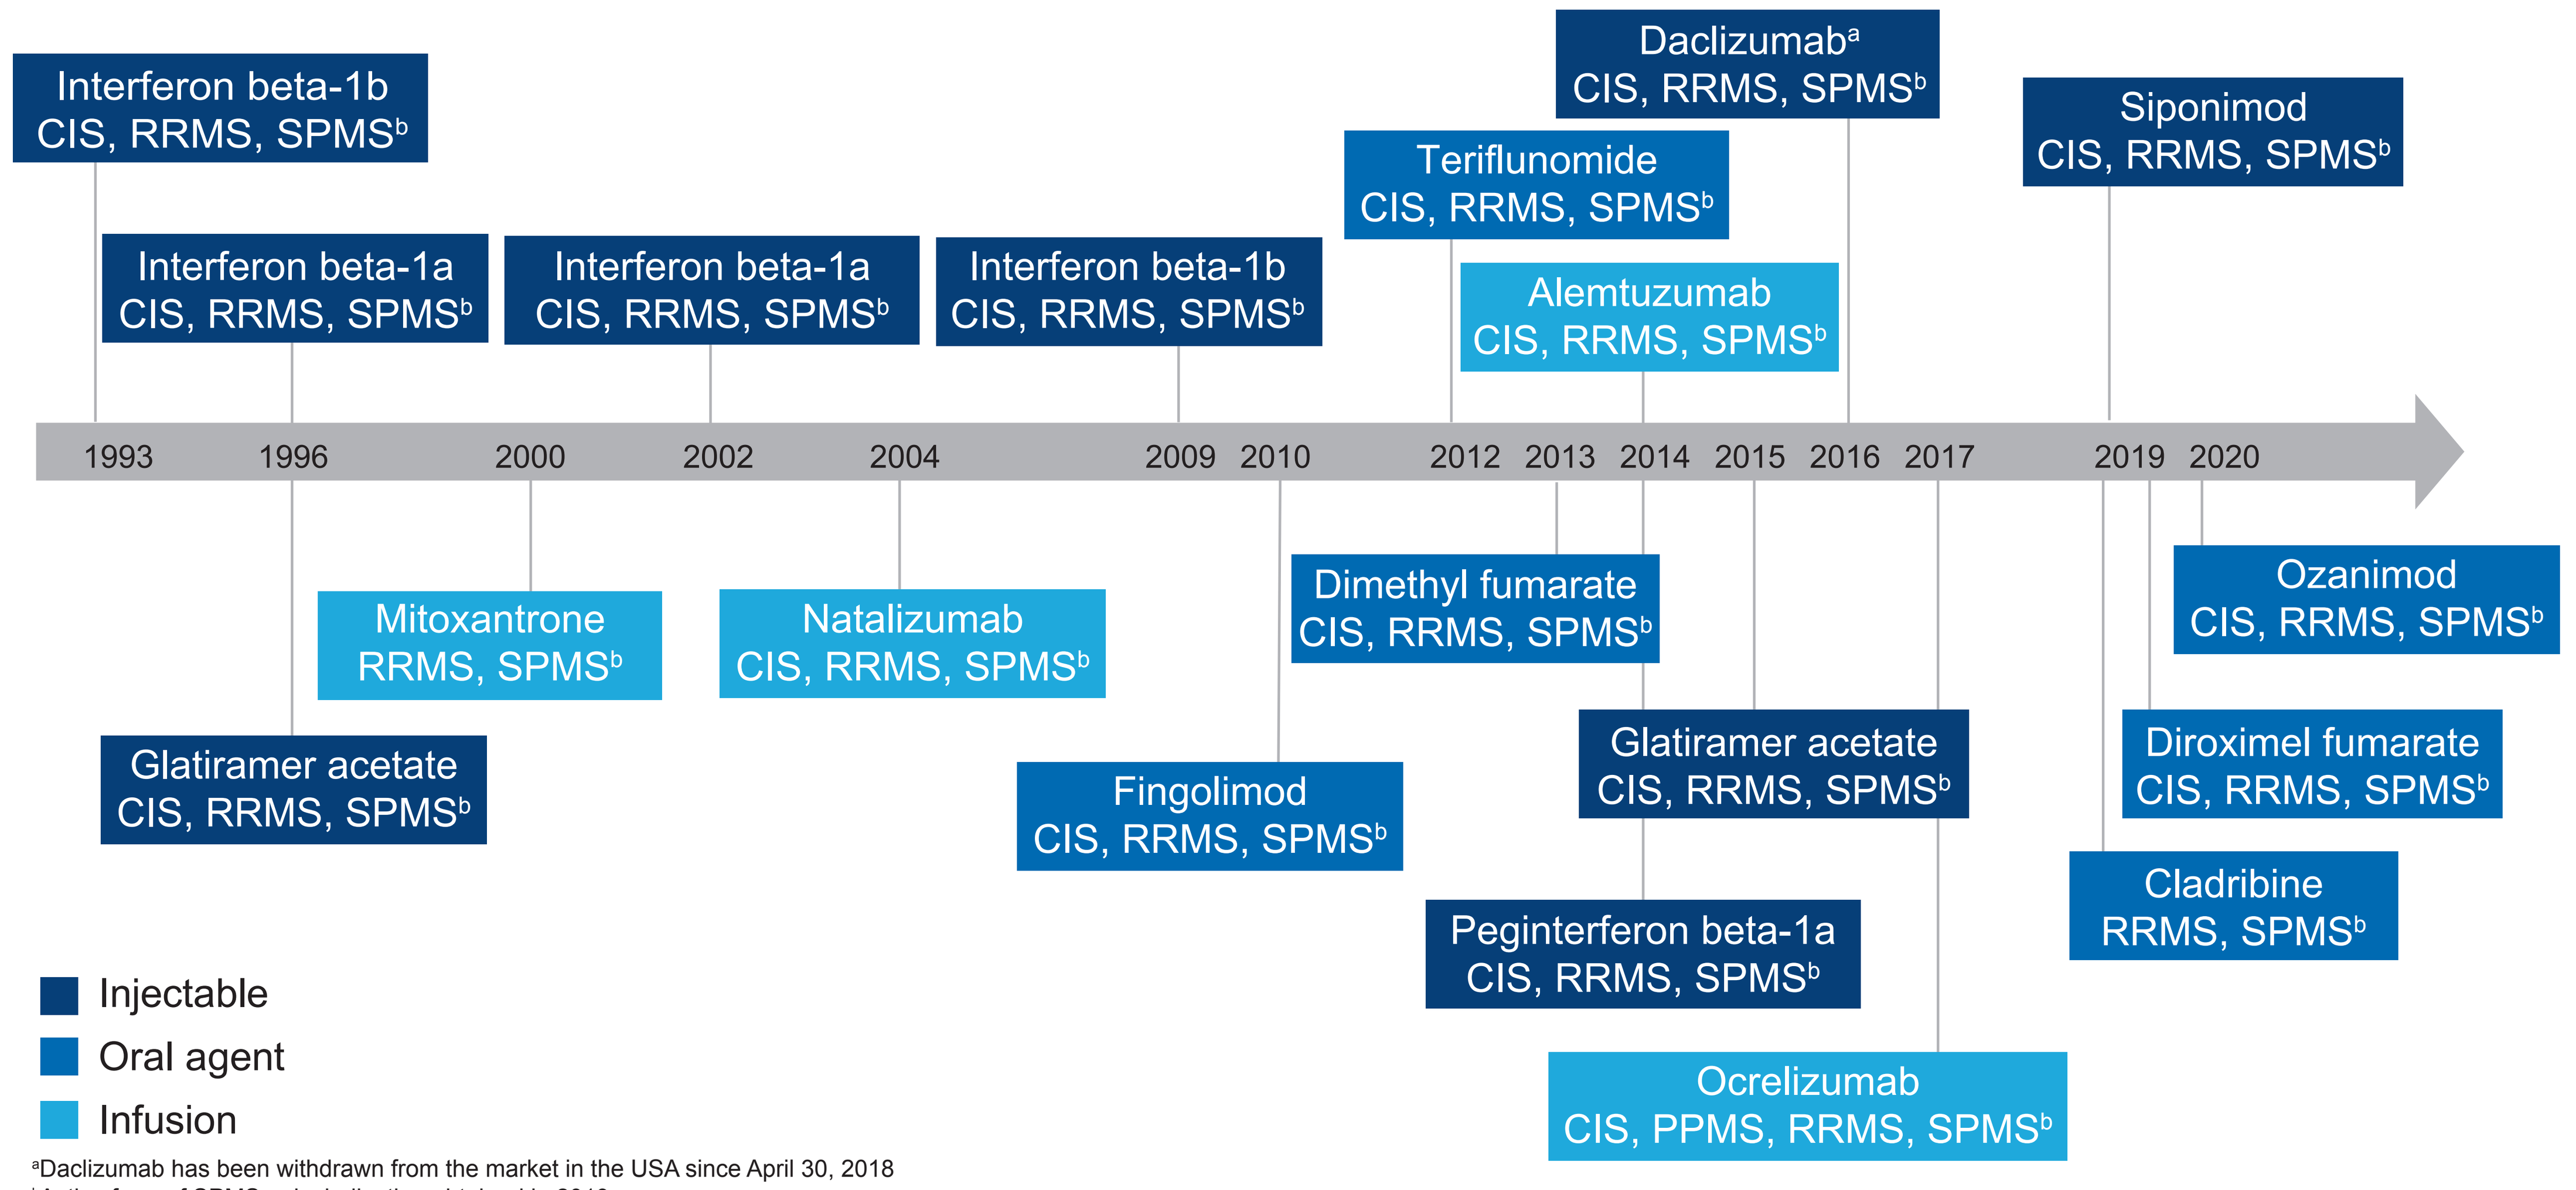
Interferon beta-1b CIS, RRMS, SPMS[b]
Interferon beta-1a CIS, RRMS, SPMS[b]
Interferon beta-1a CIS, RRMS, SPMS[b]
Interferon beta-1b CIS, RRMS, SPMS[b]
Teriflunomide CIS, RRMS, SPMS[b]
Daclizumab[a] CIS, RRMS, SPMS[b]
Alemtuzumab CIS, RRMS, SPMS[b]
Siponimod CIS, RRMS, SPMS[b]
1993 1996 2000 2002 2004 2009 2010 2012 2013 2014 2015 2016 2017 2019 2020
Mitoxantrone RRMS, SPMS[b]
Natalizumab CIS, RRMS, SPMS[b]
Glatiramer acetate CIS, RRMS, SPMS[b]
Dimethyl fumarate CIS, RRMS, SPMS[b]
Fingolimod CIS, RRMS, SPMS[b]
Glatiramer acetate CIS, RRMS, SPMS[b]
Peginterferon beta-1a CIS, RRMS, SPMS[b]
Ocrelizumab CIS, PPMS, RRMS, SPMS[b]
Ozanimod CIS, RRMS, SPMS[b]
Diroximel fumarate CIS, RRMS, SPMS[b]
Cladribine RRMS, SPMS[b]
Injectable
Oral agent
Infusion

[a]Daclizumab has been withdrawn from the market in the USA since April 30, 2018
[b]Active form of SPMS only; indication obtained in 2019
CIS, clinically isolated syndrome; DMTs, disease-modifying therapies; FDA, Food and Drug Administration (USA); MS, multiple sclerosis; PPMS, primary progressive multiple sclerosis; RRMS, relapsing-remitting multiple sclerosis; SPMS, secondary progressive multiple sclerosis
From Weigel M, *et al.* Orchestrating a new path for MS: empowering patients through physical and music therapies (in development)

Figure 2. Effect of music-cued, metronome-cued and no intervention on walking (A and B), fatigue (C), and HRQoL (D) in MS[16]

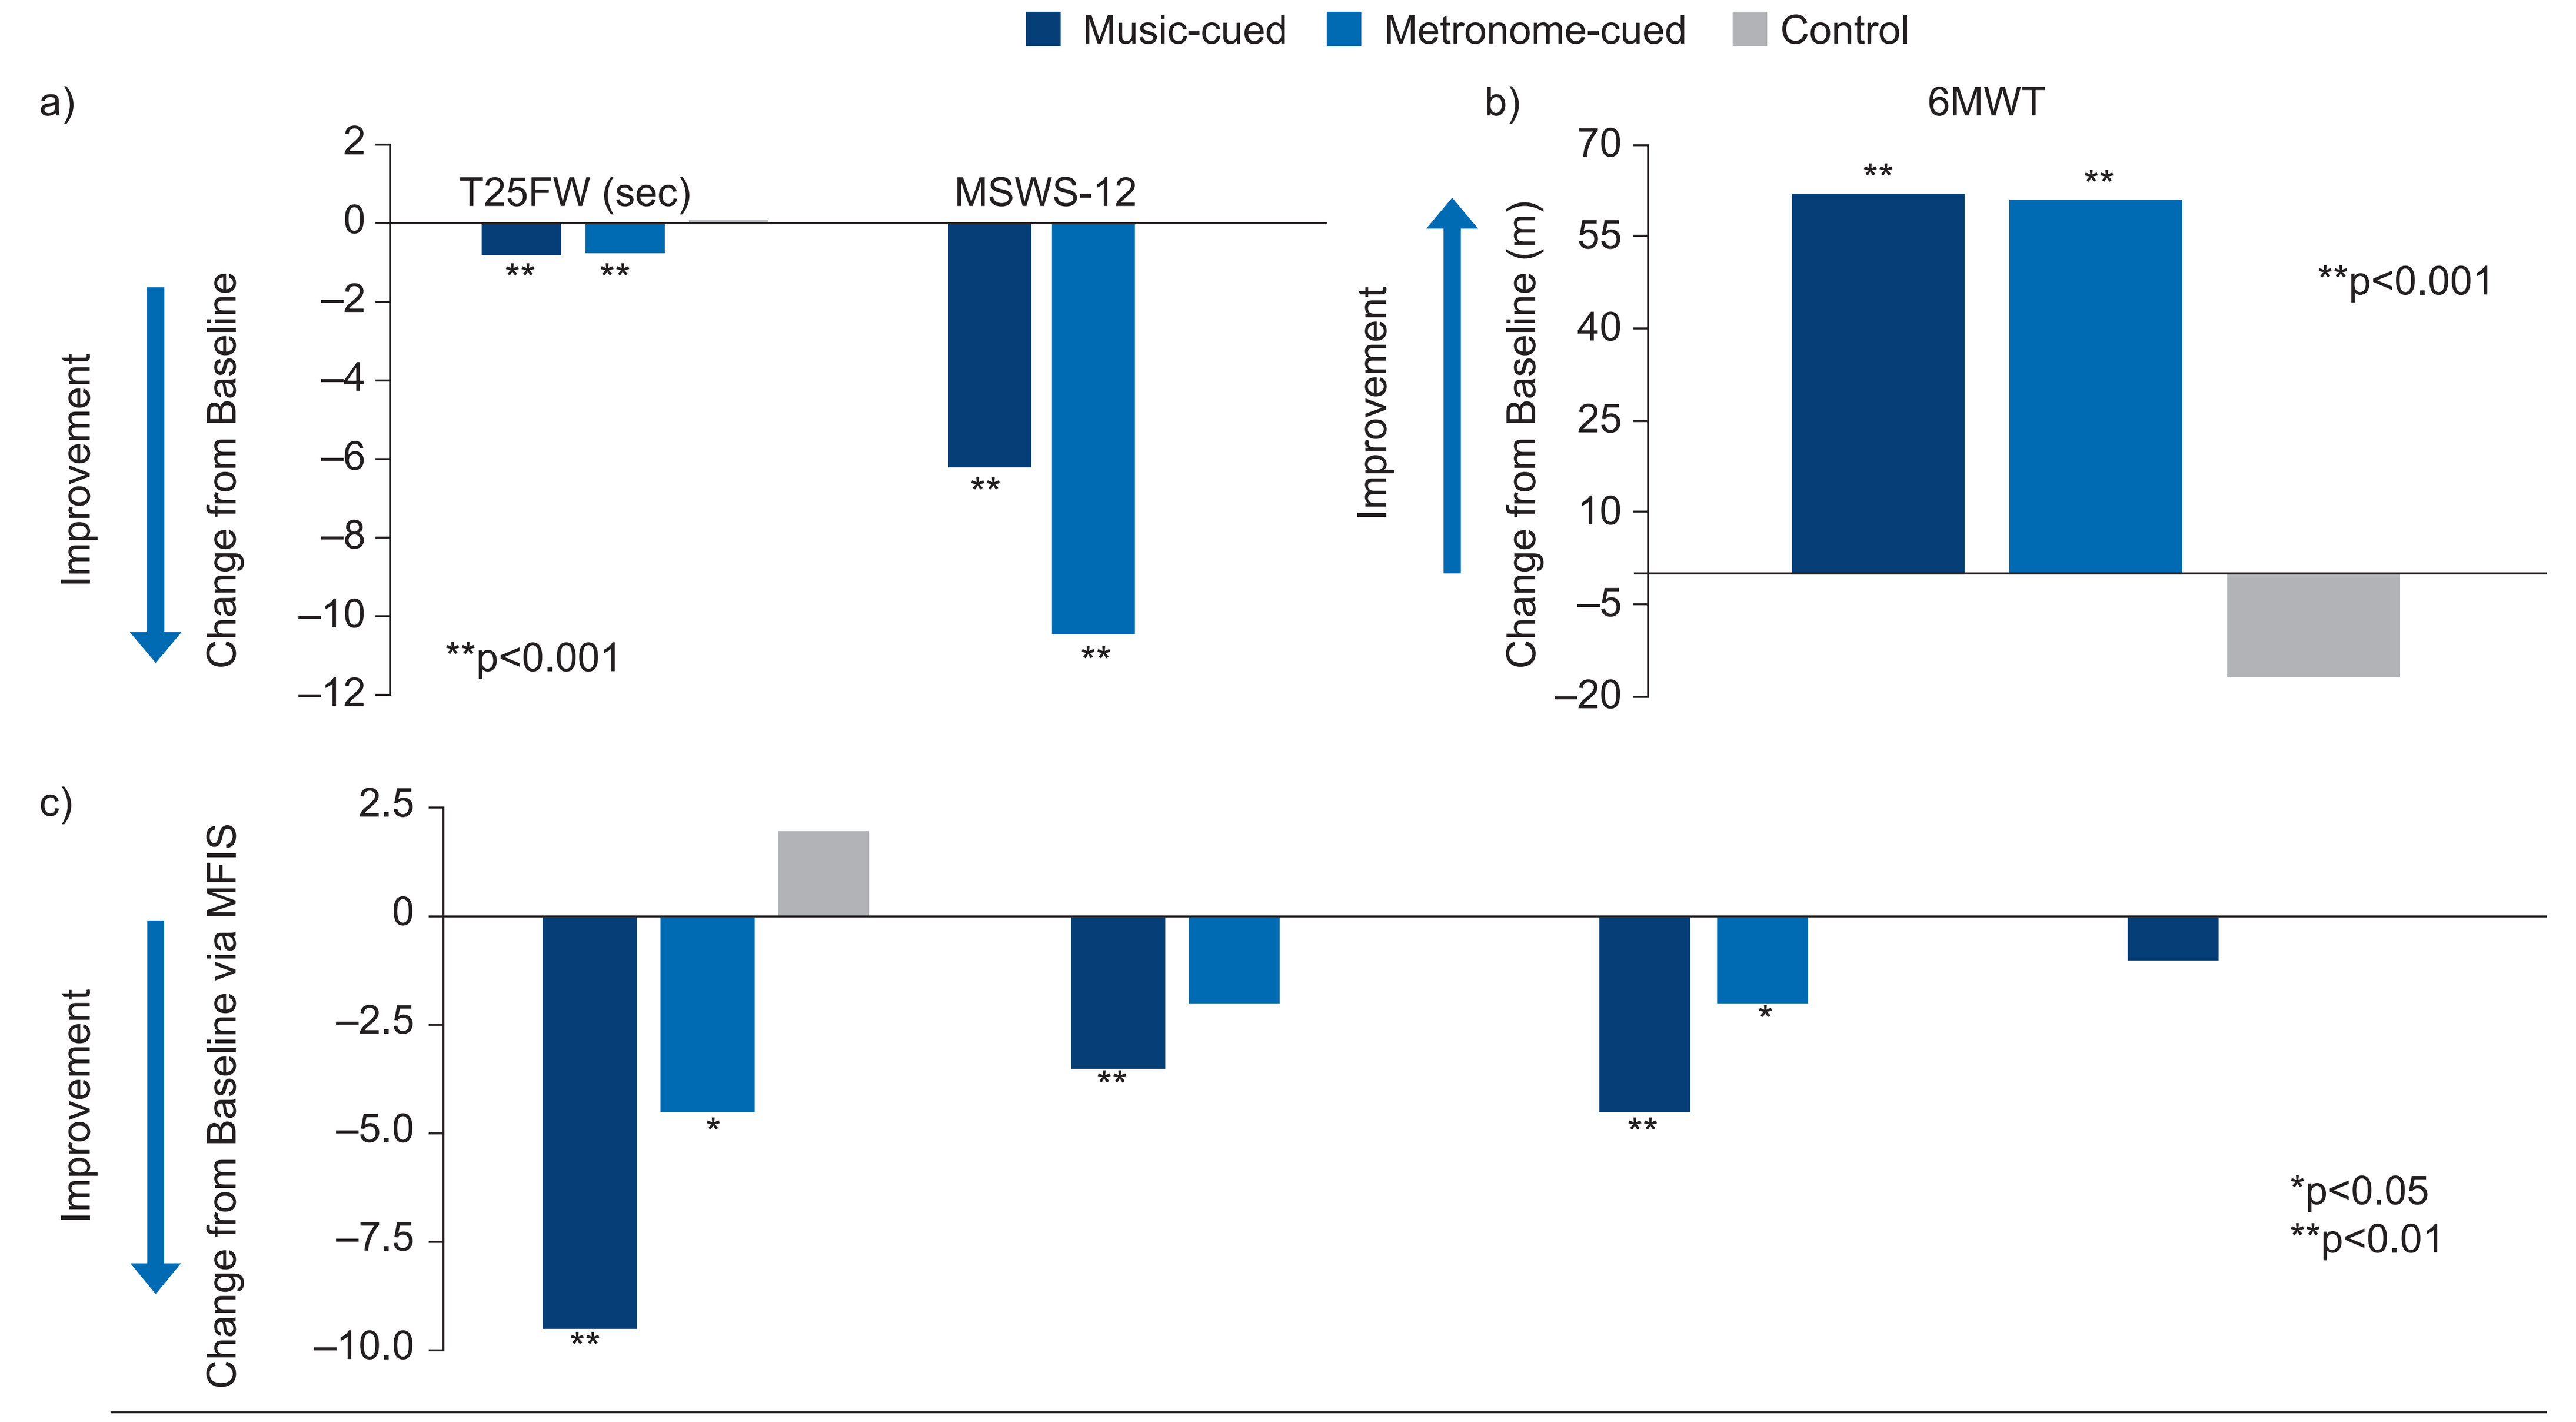
Music-cued
Metronome-cued
Control
a)
T25FW (sec)
MSWS-12
Improvement
Change from Baseline
**p<0.001
b)
6MWT
Improvement
Change from Baseline (m)
**p<0.001
c)
Improvement
Change from Baseline via MFIS
*p<0.05
**p<0.01

	Total	Physical	Cognitive	Psy-Soc
Music-cued	–9.5	–3.5	–4.5	–1.0
Metronome-cued	–4.5	–2.0	–2.0	0
Control	2.0	0	0	0

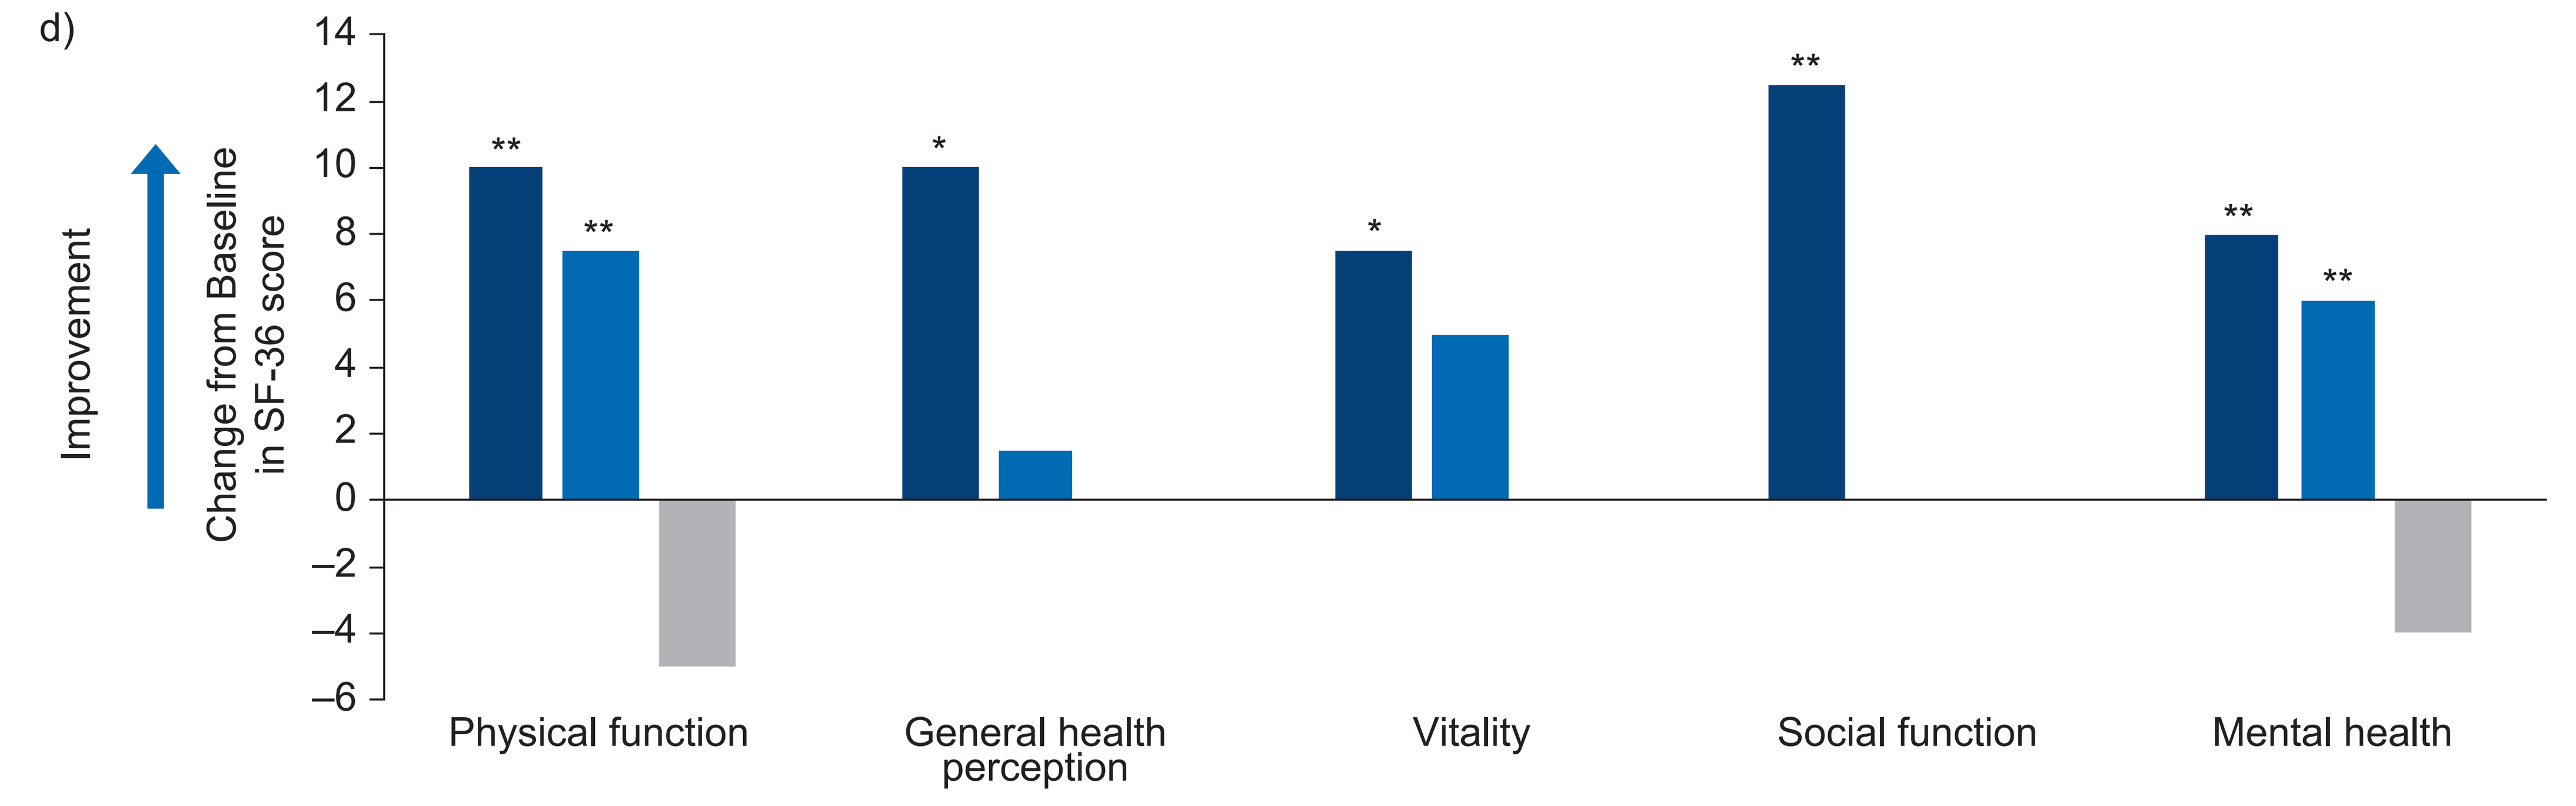
d)
Improvement
Change from Baseline in SF-36 score
Physical function
General health perception
Vitality
Social function
Mental health

6MWT, 6-minute walk test; HRQoL, health-related quality of life; MFIS, Modified Fatigue Impact Scale; MS, multiple sclerosis; MSWS-12, multiple sclerosis walking scale-12; SF-36, Short Form-36 Health Survey; T25FW, timed 25-foot walk
Adapted from Seebacher B, *et al.* 2017[16]

- MT can improve physical symptoms and HRQoL in MS (**Figure 2**)
 - A trial comparing music-cued (music and rhythm) and metronome-cued (rhythm only) motor imagery versus no intervention found that individuals in the two intervention groups could walk a mean of 62.1 m and 60.9 m further after 4 weeks, respectively, versus Baseline
 - Mean change in the no intervention group was −17.1 m[16]
 - Improvements in fatigue and HRQoL (physical function, general health perception, vitality, social function, mental health) occurred in both intervention groups versus no intervention[16]
- MT can also improve fine motor skills, learning and memory[17-19] and offers a forum for exploring emotional responses to the physical changes experienced in MS[20,21]

Conclusions

- A multidisciplinary approach is fundamental to achieve optimal results in treatment and rehabilitation of people with MS
 - Nonpharmacological rehabilitative therapies are an important adjunct for managing daily life
 - Early intervention may have the potential to lessen symptom worsening and reduce polypharmacy and long-term healthcare costs[22]
- Current studies of PT and MT indicate they are highly beneficial tools for delaying the loss of both fine and gross motor skills, improving overall well-being and psychosocial health factors and, ultimately, preserving HRQoL[23]
- Earlier intervention may potentially lessen the functional decline of patients through adaptive neuroplasticity[24]
- A multidisciplinary approach is fundamental in achieving optimal results in the treatment and rehabilitation of people with MS
- Further research on combined PT and MT interventions, and also the effect of these therapies on neuroplasticity, may further improve outcomes in MS

References

1. Macaron G, Ontaneda D. *Biomedicines.* 2019;7:56.
2. Multiple Sclerosis International Federation. Atlas of MS 2013. Available from: http://www.msif.org/wp-content/uploads/2014/09/Atlas-of-MS.pdf (Accessed March 18, 2020).
3. Lynch SG, *et al. Mult Scler.* 2001;7:411-416.
4. Bowling, A. Complementary and Alternative Medicine in MS. Available from: https://www.nationalmssociety.org/NationalMSSociety/media/MSNationalFiles/Brochures/Clinical_Bulletin_Complementary-and-Alternative-Medicine-in-MS.pdf (Accessed March 18, 2020).
5. Yadav V, *et al. Neurology.* 2014;82:1083-1092.
6. Marziniak M, *et al. JMIR Rehabil Assist Technol.* 2018;5:e5.
7. Stuifbergen AK, *et al. Arch Phys Med Rehabil.* 2006;87:935-943.
8. Robinson AG, *et al. Physiotherapy.* 2019;105:174-186.
9. Arntzen EC, *et al. Physiother Res Int.* 2019:e1798.
10. Heine M, *et al. Cochrane Database Syst Rev.* 2015:CD009956.
11. Kierkegaard M, *et al. J Neurol Sci.* 2016;362:251-257.
12. Rietberg MB, *et al. Cochrane Database Syst Rev.* 2005:CD003980.
13. Bruscia, KE. Defining Music Therapy, 3rd ed. Dallas: Barcelona Publishers; 2014.
14. Cheever T, *et al. Neuron.* 2018;97:1214-1218.
15. Collins FS, Fleming R. *JAMA.* 2017;317:2470-2471.
16. Seebacher B, *et al. Mult Scler.* 2017;23:286-296.
17. Gatti R, *et al. Physiother Res Int.* 2015;20:100-107.
18. Moumdjian L, *et al. Eur J Phys Rehabil Med.* 2017;53:466-482.
19. Thaut MH, *et al. Front Hum Neurosci.* 2014;8:395.
20. Aldridge D, *et al. Complement Ther Med.* 2005;13:25-33.
21. Schmid W, Aldridge D. *J Music Ther.* 2004;41:225-240.
22. Buchanan RJ, *et al. NeuroRehabilitation.* 2006;21:223-232.
23. Khan F, Amatya B. *Arch Phys Med Rehabil.* 2017;98:353-367.
24. Negaresh R, *et al. Eur J Neurol.* 2019;26:711-721.

Acknowledgments

Editorial support was provided by Oxford PharmaGenesis, Oxford, UK, which was sponsored by Novartis Pharmaceuticals Corporation. The final responsibility for the content lies with the authors.

Disclosures

Megan Weigel is a consultant for Biogen Inc., Celgene Corporation Inc., Novartis (independent of the submitted work) and Sanofi Genzyme; and has received speaker fees from Acorda, EMD Serono, Mallinckrodt Pharmaceuticals, Novartis (independent of the submitted work) and Sanofi Genzyme. **Renée Fleming** has received consultancy fees from Novartis (independent of the submitted work). **Brian Hutchinson** has received consultancy fees from Novartis (independent of the submitted work); and speaker fees from Biogen Inc. **Wendy L Magee** has received consultancy fees from (independent of the submitted work). **Wendy Su** is an employee of Novartis Pharmaceuticals Corporation.

© 2020 Novartis Pharmaceuticals Corporation.

Poster developed for the Consortium of Multiple Sclerosis Centers (CMSC) 2020 virtual event.

This study was sponsored by Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA.

Text: Qde8c4
To: 8NOVA (86682) US Only
+18324604729 North, Central and South Americas; Caribbean; China
+447860024038 UK, Europe and Russia
+46737494608 Sweden, Europe
Visit the web at:
http://novartis.medicalcongressposters.com/Default.aspx?doc=de8c4

Scan this QR code to download a copy of this poster

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Presenting author email address: surfernurse@me.com