Injection-Related Reactions With Subcutaneous Administration of Ofatumumab in Relapsing Multiple Sclerosis: Pooled Analysis of the Phase 3 ASCLEPIOS I and II Trials

DXT61

Ratnakar Pingili, MBBS[1], Amit Bar-Or, MD, FRCPC[2], Jeffrey A. Cohen, MD[3], Patricia K. Coyle, MD[4], Anne H. Cross, MD[5], Stephen L. Hauser, MD[6], Ludwig Kappos, MD[7], Cecile Kerloeguen, MSc[8], Ayan Das Gupta, MS[9], Valentine Jehl, MSc[8], Dieter A. Haering, PhD[8], Krishnan Ramanathan, PhD[8], Martin Merschhemke, MD[8]

[1]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [2]Center for Neuroinflammation and Experimental Therapeutics and Department of Neurology, Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA, USA; [3]Department of Neurology, Mellen MS Center, Neurological Institute, Cleveland Clinic, Cleveland, OH, USA; [4]Department of Neurology, Stony Brook University, Stony Brook, NY, USA; [5]Department of Neurology, Division of Neuroimmunology, Washington University School of Medicine, St Louis, MO, USA; [6]Department of Neurology, UCSF Weill Institute for Neurosciences, University of California San Francisco, San Francisco, CA, USA; [7]Neurologic Clinic and Policlinic, Departments of Medicine, Clinical Research, Biomedicine and Biomedical Engineering, University Hospital and University of Basel, Basel, Switzerland; [8]Novartis Pharma AG, Basel, Switzerland; [9]Novartis Healthcare Pvt. Ltd. Hyderabad, India

Background

- Ofatumumab is the first fully human anti-CD20 monoclonal antibody,[1] administered with a monthly 20 mg subcutaneous (s.c.) dosing regimen in multiple sclerosis[2]
- In the Phase 3 ASCLEPIOS I and II trials, ofatumumab 20 mg s.c. demonstrated superior efficacy versus teriflunomide and a favorable safety profile in patients with relapsing multiple sclerosis (RMS)[3]
 - Relative reduction in ARR: 50.5% (p<0.001) in ASCLEPIOS I, and 58.5% (p<0.001) in ASCLEPIOS II and risk reduction in 3- and 6-month CDW: 34.4% (p=0.002) and 32.5% (p=0.012) in the pre-specified pooled analysis[3]
 - The total incidence of adverse events (AEs) was similar between the ofatumumab (83.6%) and teriflunomide (84.2%) treatment groups
 - The majority of AEs (>90%) with both ofatumumab and teriflunomide were of Grade 1/2 in severity. The incidence of Grade 3/4 AEs was low (8.0% vs 8.4%)
- Injection-related reactions (IRRs) were the most common AEs by preferred term (PT) observed in these trials (cumulative: ofatumumab group, 20.6%; matching placebo injection in the teriflunomide group, 15.3%).

Objective

- To characterize the risk of IRRs (systemic and local site reactions) observed with ofatumumab in the ASCLEPIOS I/II trials in RMS patients

Methods

Study design and treatment pattern

- ASCLEPIOS I and II were double-blind, double-dummy, active comparator-controlled, parallel-group, multicenter, adaptive and flexible duration design trials (maximum duration of up to 30 months) (**Figure 1**)

Figure 1. Study design – treatment period

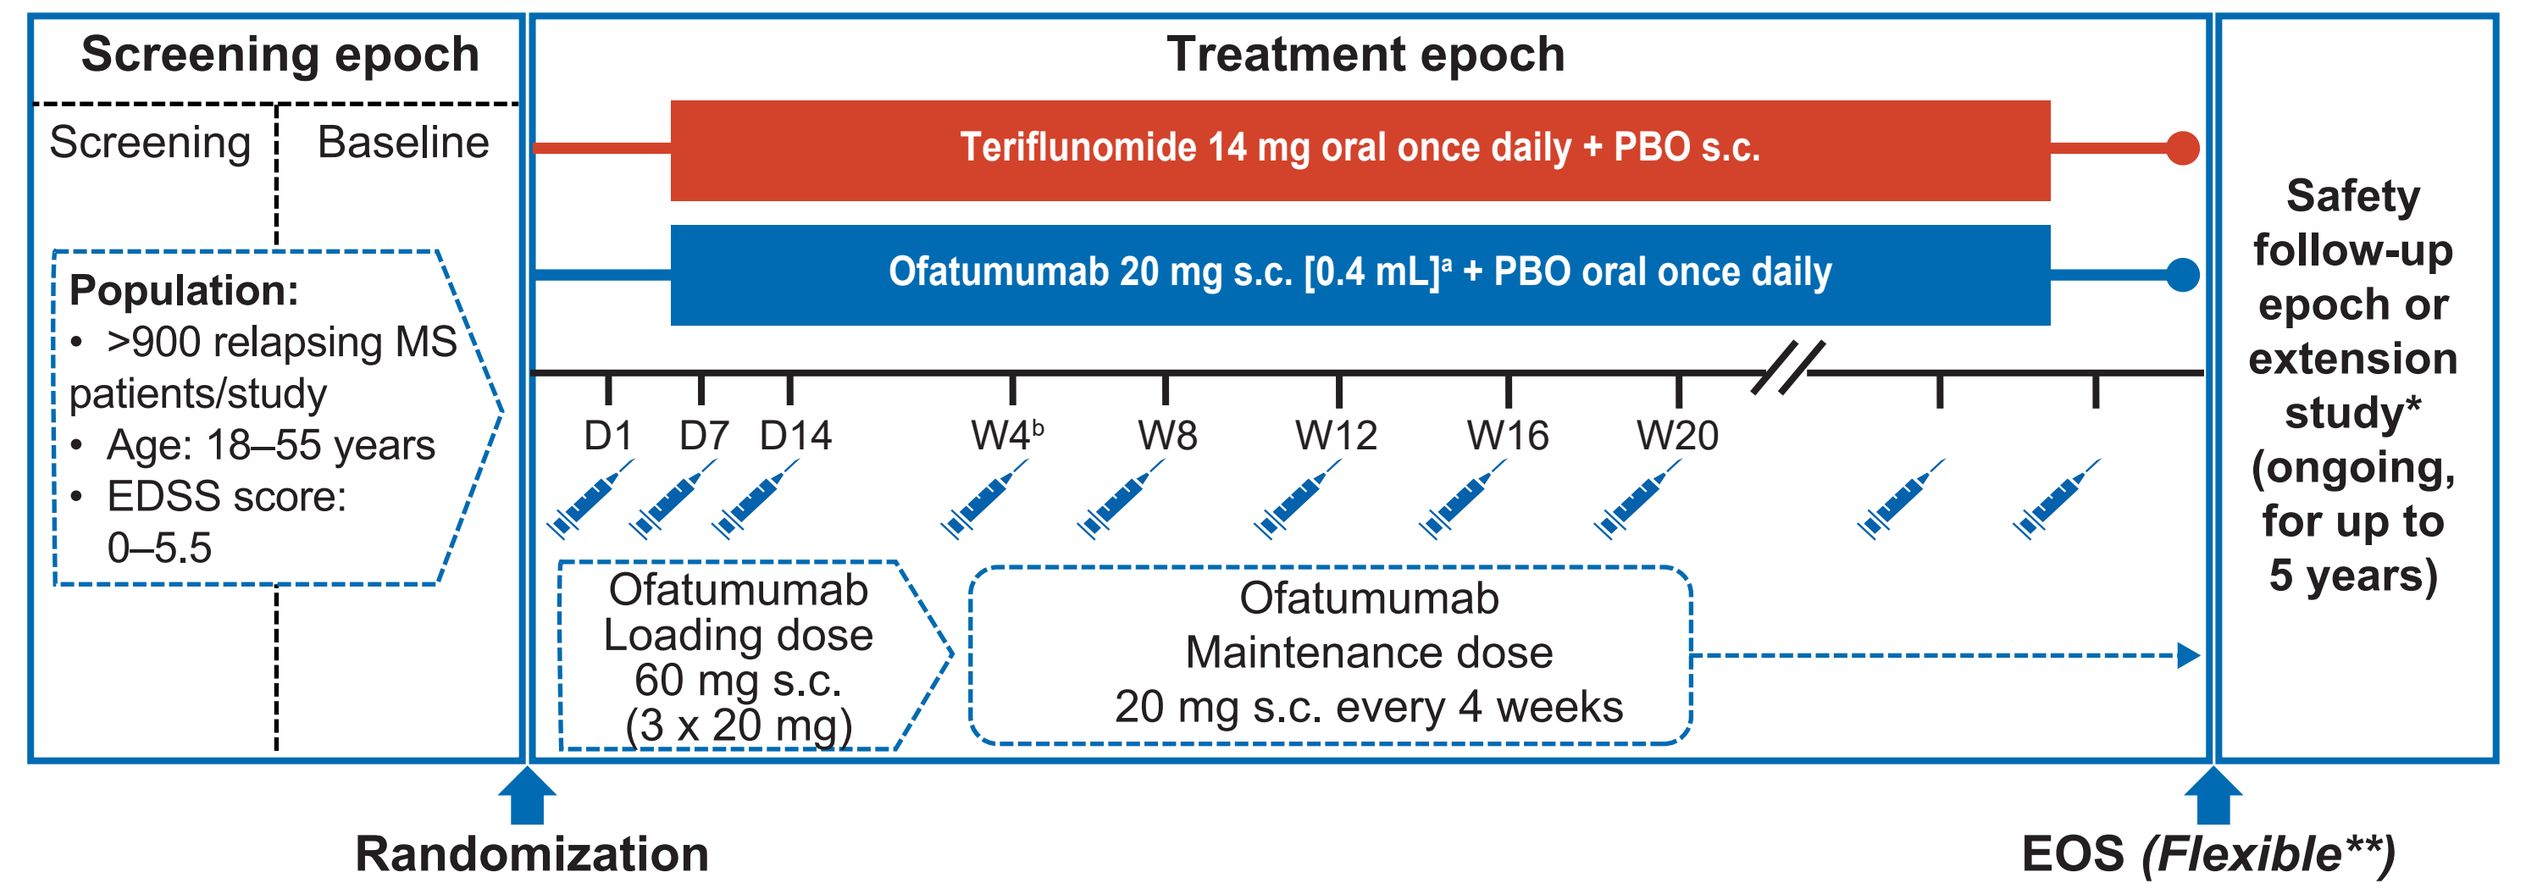
Screening epoch
Treatment epoch
Screening
Baseline
Teriflunomide 14 mg oral once daily + PBO s.c.
Ofatumumab 20 mg s.c. [0.4 mL]ᵃ + PBO oral once daily
Population:
• >900 relapsing MS patients/study
• Age: 18–55 years
• EDSS score: 0–5.5
D1 D7 D14 W4ᵇ W8 W12 W16 W20
Ofatumumab Loading dose 60 mg s.c. (3 x 20 mg)
Ofatumumab Maintenance dose 20 mg s.c. every 4 weeks
Safety follow-up epoch or extension study* (ongoing, for up to 5 years)
Randomization
EOS (Flexible**)

*Open Label Extension study (up to 5 years) via separate protocol. Patients who complete the Treatment Epoch while on study drug, may be eligible to participate. Safety FU epoch is included to ensure all patients not entering Extension can have at least 9 months follow up after last dose of study drug

**The end of study was projected based on a prospectively planned analysis of blinded data to provide 90% power for the primary endpoint, and 90% and 80% power for 3- and 6-month confirmed disability worsening. EOS was defined by the amount of statistical information collected in the trial (relapses and disability events), instead of relying on a fixed time after the last patient has been randomized

[a]20 mg of ofatumumab was administered in an injection volume of 0.4 mL; [b]Week 4 (Month 1) and every 4 weeks thereafter

D, day; EDSS, Expanded Disability Status Scale; EOS, end of study; MS, multiple sclerosis; PBO, placebo; s.c., subcutaneous; W, week

- Patients were randomized (1:1) to receive ofatumumab 20 mg s.c. (n=946) (loading dose: Days 1, 7 and 14; maintenance dose: every 4 weeks from Week 4) or oral teriflunomide 14 mg once daily (n=936) for up to 30 months
- Patients in the teriflunomide group received matching placebo injections
- As per protocol, the patients received or self-administered the first four injections at the clinic/under supervision; after the fourth injection, the majority of patients (74.4%) self-administered ofatumumab at home
- Premedication with acetaminophen and/or antihistamines (or equivalent) was recommended and administered at the discretion of the investigator, 30 to 60 minutes prior to the injection of the study drug

Study assessments and analysis

- The investigator reported IRRs on dedicated CRF pages as systemic IRRs or local site IRRs
- For analysis, systemic reactions that occurred within 24 hours after the injection (i.e., time to onset of reaction ≤24 hours) were assumed to be injection-related and accordingly included in the analysis as PT 'injection-related reaction'
- Local site IRRs could be reported without any time limit from the time the injection was administered and were included in the analysis as PT 'injection-site reactions'
- The proportions of patients with systemic and local site IRRs were analyzed by treatment group against injection sequence numbers (0 to 10) and cumulatively for all injections
- The severity of these reactions was reported using the Common Terminology Criteria for Adverse Events (CTCAE) grading, and categorized as mild (Grade 1), moderate (Grade 2), severe (Grade 3), and life threatening (Grade 4)
- Symptoms were summarized by the number and percentage of patients by each injection and cumulatively for all injections
- Safety analyses were conducted using the safety set (all patients who received at least one dose of trial medication)

Results

- The median (range) duration (patient-years) of exposure was similar between the treatment groups
- Ofatumumab: 599.5 (31–873) days (1486.7 patient-years); 87.9% of patients had ≥1 study year of exposure to the study drug
- Teriflunomide: 576.0 (13–848) days (1397.8 patient-years); 87.1% of patients had ≥1 study year of exposure to the study drug

Systemic IRRs (that occurred within 24 hours after the injection)

- Over the duration of the studies, a higher proportion of patients in the ofatumumab group experienced ≥1 systemic IRR versus teriflunomide/placebo injections (20.2% vs 15.0%)
- The incidence of systemic IRRs was highest with the first injection (ofatumumab, 14.4% versus matching placebo, 7.5%); the incidence decreased substantially for all subsequent injections and was similar to the matching placebo injections in the teriflunomide group (**Figure 2**)
- Majority of the IRRs (99.7%) reported with ofatumumab were non-serious in nature and of Grade 1/2 in severity
- Grade 3 IRRs were observed in two patients (0.2%) with ofatumumab at the first injection
 - In one patient, the event was also reported as a serious AE. The symptoms (fever, nausea, tachycardia and vomiting) reported 4 hours after first injection were resolved with symptomatic medication and the patient completed the study with no recurrences
 - In the other patient, the event was not assessed as a serious AE (as per investigator), but the patient discontinued the study treatment due to IRR. The symptoms (abdominal pain, asthenia, pruritis general, and urticaria) resolved with antihistamine treatment
- One additional IRR (Grade 1 with symptoms of chills, asthenia, arthralgia, and muscle spasm) was reported as a serious AE with ofatumumab. This patient recovered and no recurrences of an IRR on subsequent injections were observed
- All IRRs were manageable with very low withdrawal rate (0.1%); 9.2% required symptomatic treatment in the ofatumumab group vs 4.2% in the teriflunomide group
- In the teriflunomide group, there were no serious or Grade 3 IRRs
- In both the treatment groups, no Grade 4 systemic IRRs were reported during the study
- In the teriflunomide group, none of IRRs led to discontinuation of the study drug

Symptoms associated with systemic IRRs

- The most common (≥2%) systemic IRR symptoms observed during all injections (**Table 1**) were as follows:
 - Ofatumumab: Fever, other systemic reactions, headache, myalgia, chills, and fatigue
 - Teriflunomide: Other systemic reactions, headache, and flushing

Table 1. Incidence of symptoms (≥2% in any group) related to systemic IRRs

Symptoms	Ofatumumab (N=946) n (%)	Teriflunomide (N=936) n (%)
Any symptoms	191 (20.2)	140 (15.0)
Fever	66 (7.0)	17 (1.8)
Other, systemic	58 (6.1)	55 (5.9)
Headache	50 (5.3)	29 (3.1)
Myalgia	37 (3.9)	14 (1.5)
Chills	35 (3.7)	16 (1.7)
Fatigue	25 (2.6)	18 (1.9)
Flushing	13 (1.4)	20 (2.1)

IRRs, injection-related reactions

Local site IRRs

- A higher proportion of patients in the ofatumumab group experienced ≥1 local site IRR versus teriflunomide/placebo injections (10.8% vs 5.6%)
- The incidence of injection site reactions with the first injection was 2.7% with ofatumumab versus 1.6% with matching placebo; the incidence decreased with subsequent injections and was similar to the matching placebo injections in the teriflunomide group (**Figure 3**)
- The majority of injection site reactions (99.9%) were of Grade 1/2 in severity.
- Only one patient reported Grade 3 injection site reaction with injection 1 (**Figure 3**) with symptoms (pain and warmth) resolved in 2 days with symptomatic medication; the same patient also reported Grade 3 injection systemic reactions
- No serious or Grade 4 injection site reactions were reported during the study
- No injection site reactions leading to discontinuation of the study treatment were observed in either treatment group

Symptoms associated with local site IRRs

- The most common injection-site reactions symptoms (≥1%) observed during all injections (**Table 2**) were as follows:
 - Ofatumumab: Erythema/redness, other site reactions, pain, itching, and induration/swelling
 - Teriflunomide: Other site reactions, pain, erythema/redness, and itching

Table 2. Incidence of symptoms (≥2% in any group) related to local site IRRs

Symptoms	Ofatumumab (N=946) n (%)	Teriflunomide (N=936) n (%)
Any symptoms	102 (10.8)	52 (5.6)
Erythema/redness	55 (5.8)	15 (1.6)
Other, site reactions	35 (3.7)	21 (2.2)
Pain	29 (3.1)	19 (2.0)
Itching	25 (2.6)	4 (0.4)
Induration/swelling	22 (2.3)	3 (0.3)

IRRs, injection-related reactions

Premedication

- At the first injection, more than 30% of patients in both treatment groups did not receive any premedication (ofatumumab, 30.8%; teriflunomide, 31.1%); the majority of patients (ofatumumab, 63.6%; teriflunomide, 61.8%) received a steroid premedication and 5.6% of patients in the ofatumumab group and 7.2% in the teriflunomide group received a non-steroid premedication

Figure 2. Incidence of systemic IRRs (safety set)

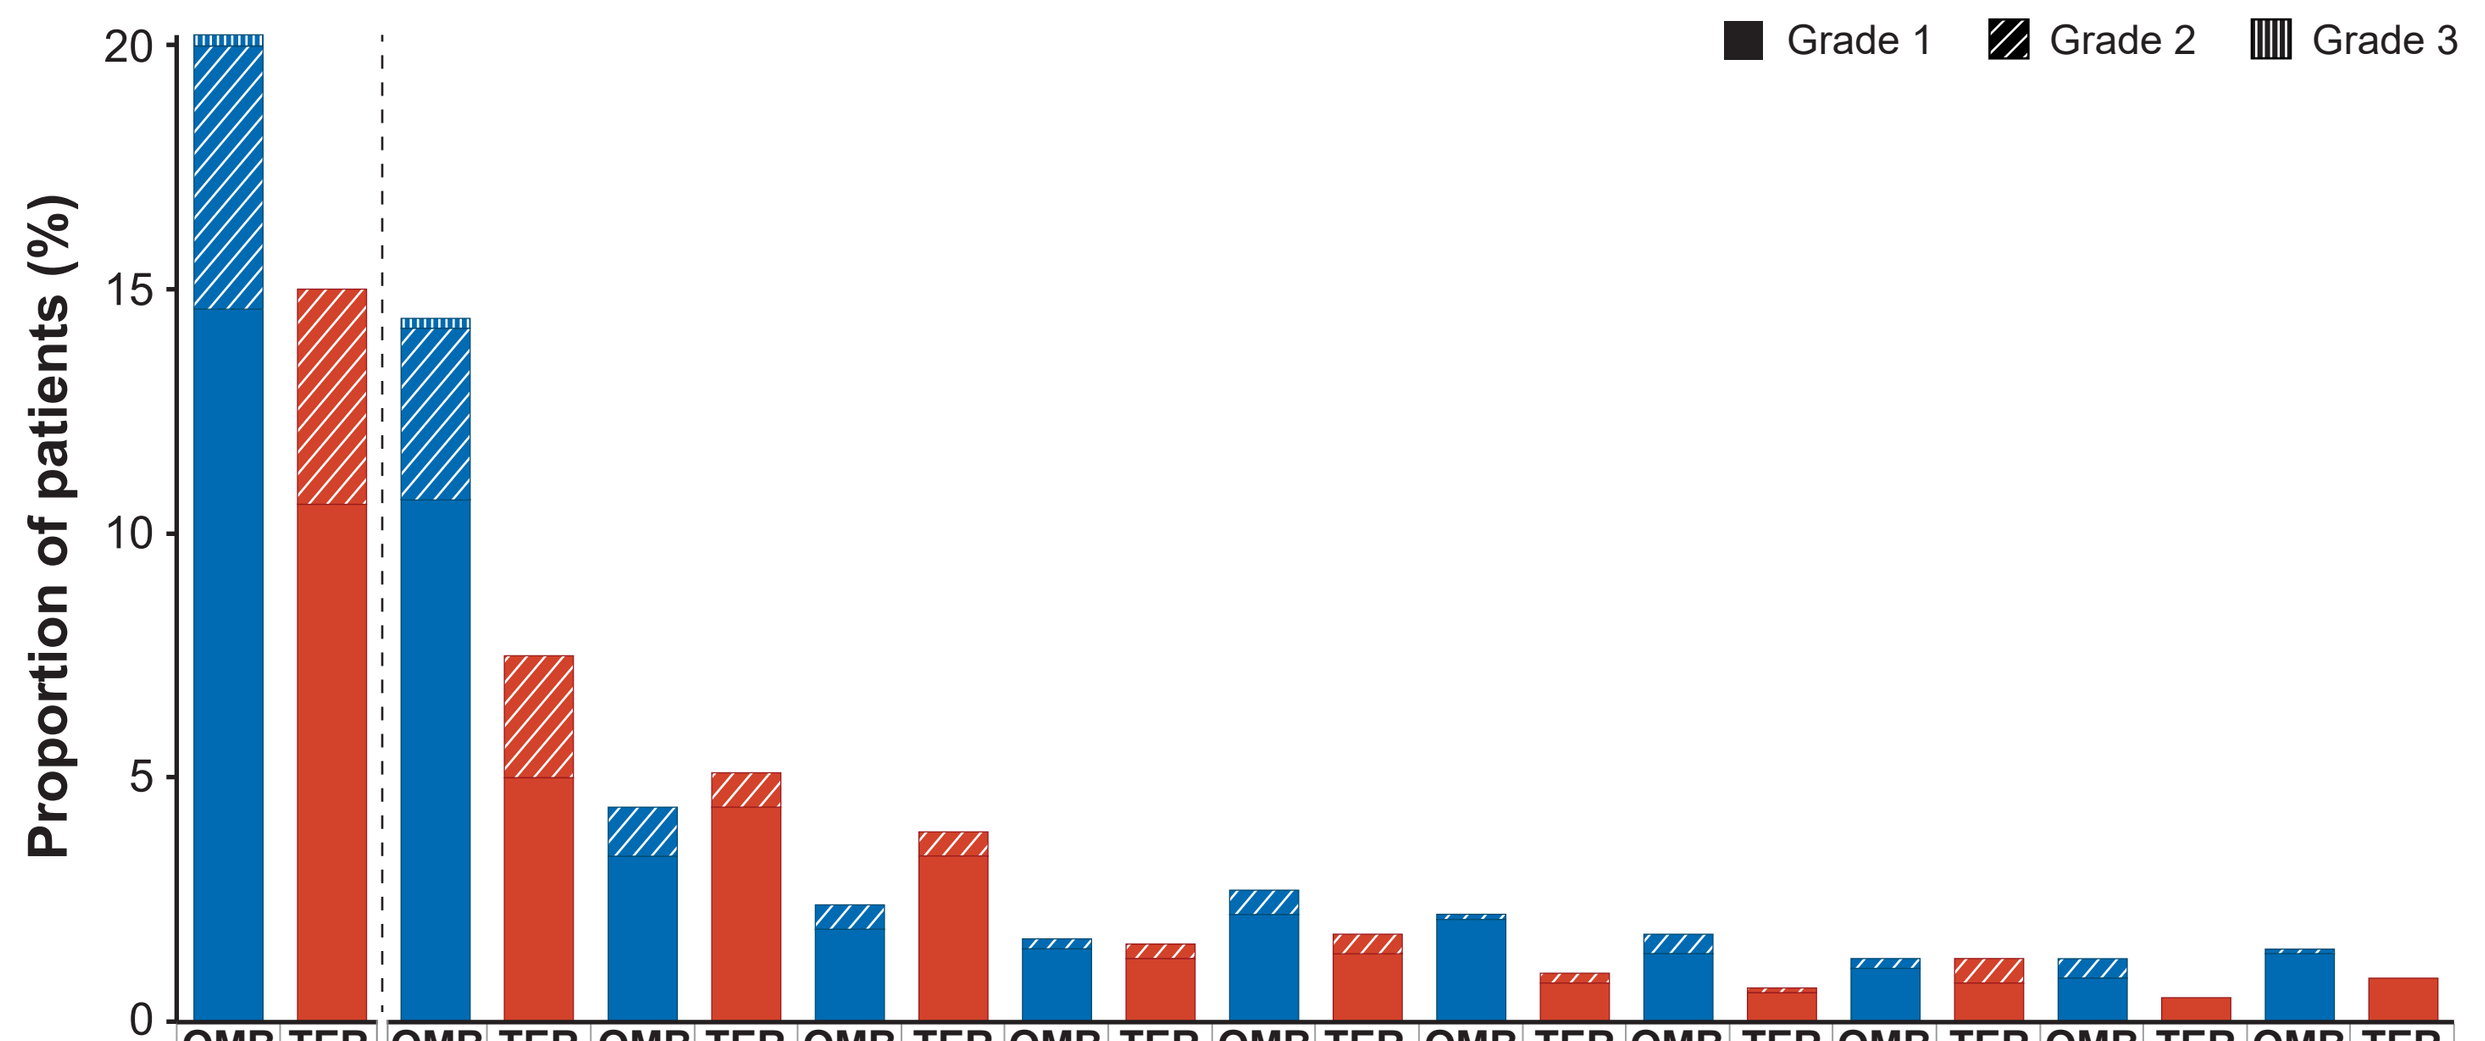
Grade 1
Grade 2
Grade 3
Proportion of patients (%)
20
15
10
5
0

Injections	All OMB	All TER	1 OMB	1 TER	2 OMB	2 TER	3 OMB	3 TER	4 OMB	4 TER	5 OMB	5 TER
Grade 3	0.2	0	0.2	0	0	0	0	0	0	0	0	0
Grade 2	5.4	4.4	3.5	2.5	1	0.7	0.5	0.5	0.2	0.3	0.5	0.4
Grade 1	14.6	10.6	10.7	5	3.4	4.4	1.9	3.4	1.5	1.3	2.2	1.4

Injections	6 OMB	6 TER	7 OMB	7 TER	8 OMB	8 TER	9 OMB	9 TER	10 OMB	10 TER
Grade 3	0	0	0	0	0	0	0	0	0	0
Grade 2	0.1	0.2	0.4	0.1	0.2	0.5	0.4	0	0.1	0
Grade 1	2.1	0.8	1.4	0.6	1.1	0.8	0.9	0.5	1.4	0.9

IRRs, injection-related reactions; OMB, ofatumumab 20 mg; TER, teriflunomide 14 mg

Figure 3. Incidence of local site IRRs (safety set)

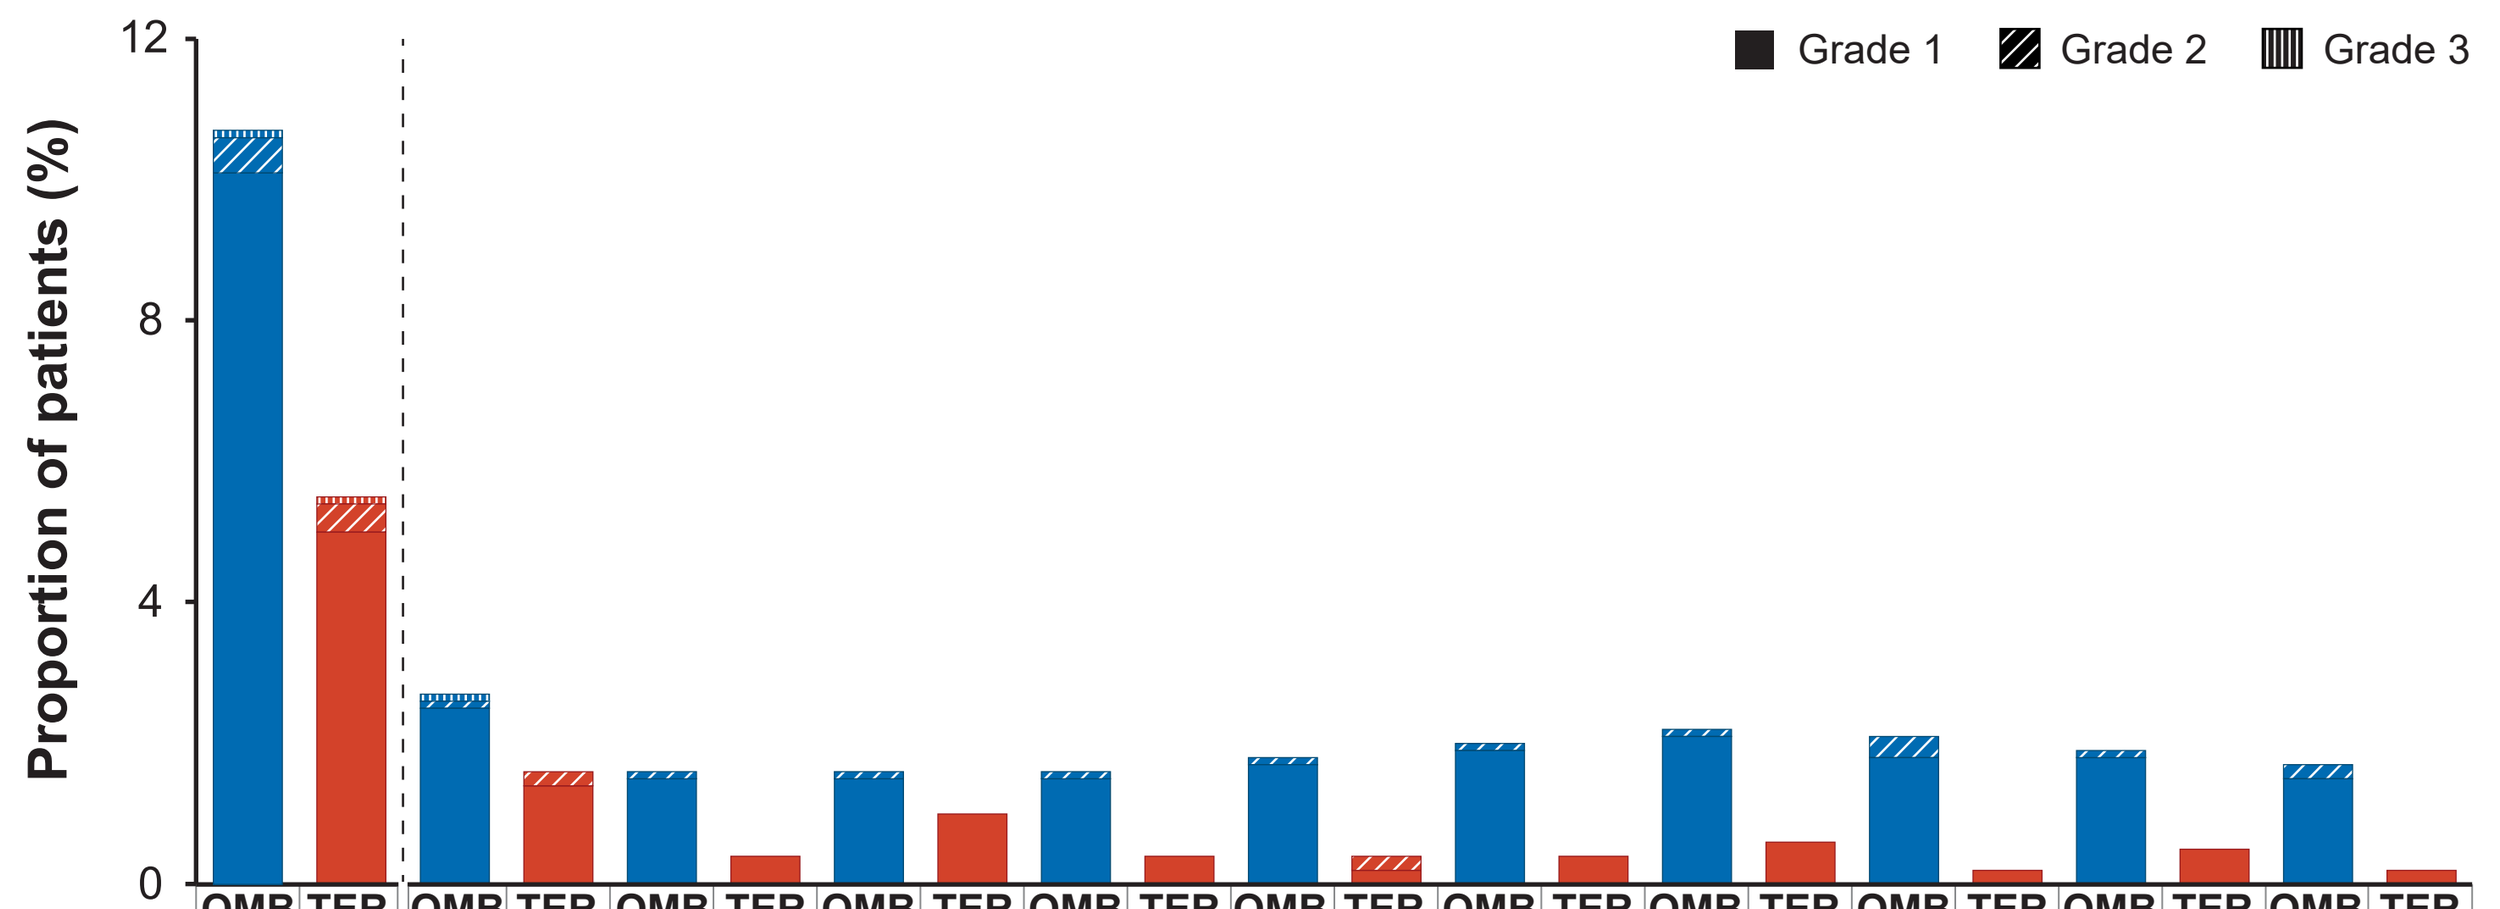
Grade 1
Grade 2
Grade 3
Proportion of patients (%)
12
8
4
0

Injections	All OMB	All TER	1 OMB	1 TER	2 OMB	2 TER	3 OMB	3 TER	4 OMB	4 TER	5 OMB	5 TER
Grade 3	0.1	0.1	0.1	0	0	0	0	0	0	0	0	0
Grade 2	0.5	0.4	0.1	0.2	0.1	0	0.1	0	0.1	0	0.1	0.2
Grade 1	10.1	5	2.5	1.4	1.5	0.4	1.5	1	1.5	0.4	1.7	0.2

Injections	6 OMB	6 TER	7 OMB	7 TER	8 OMB	8 TER	9 OMB	9 TER	10 OMB	10 TER
Grade 3	0	0	0	0	0	0	0	0	0	0
Grade 2	0.1	0	0.1	0	0.3	0	0.1	0	0.2	0
Grade 1	1.9	0.4	2.1	0.6	1.8	0.2	1.8	0.5	1.5	0.2

IRRs, injection-related reactions; OMB, ofatumumab 20 mg; TER, teriflunomide 14 mg

Figure 4. Incidence of systemic IRRs at all injections by premedication category (safety set)

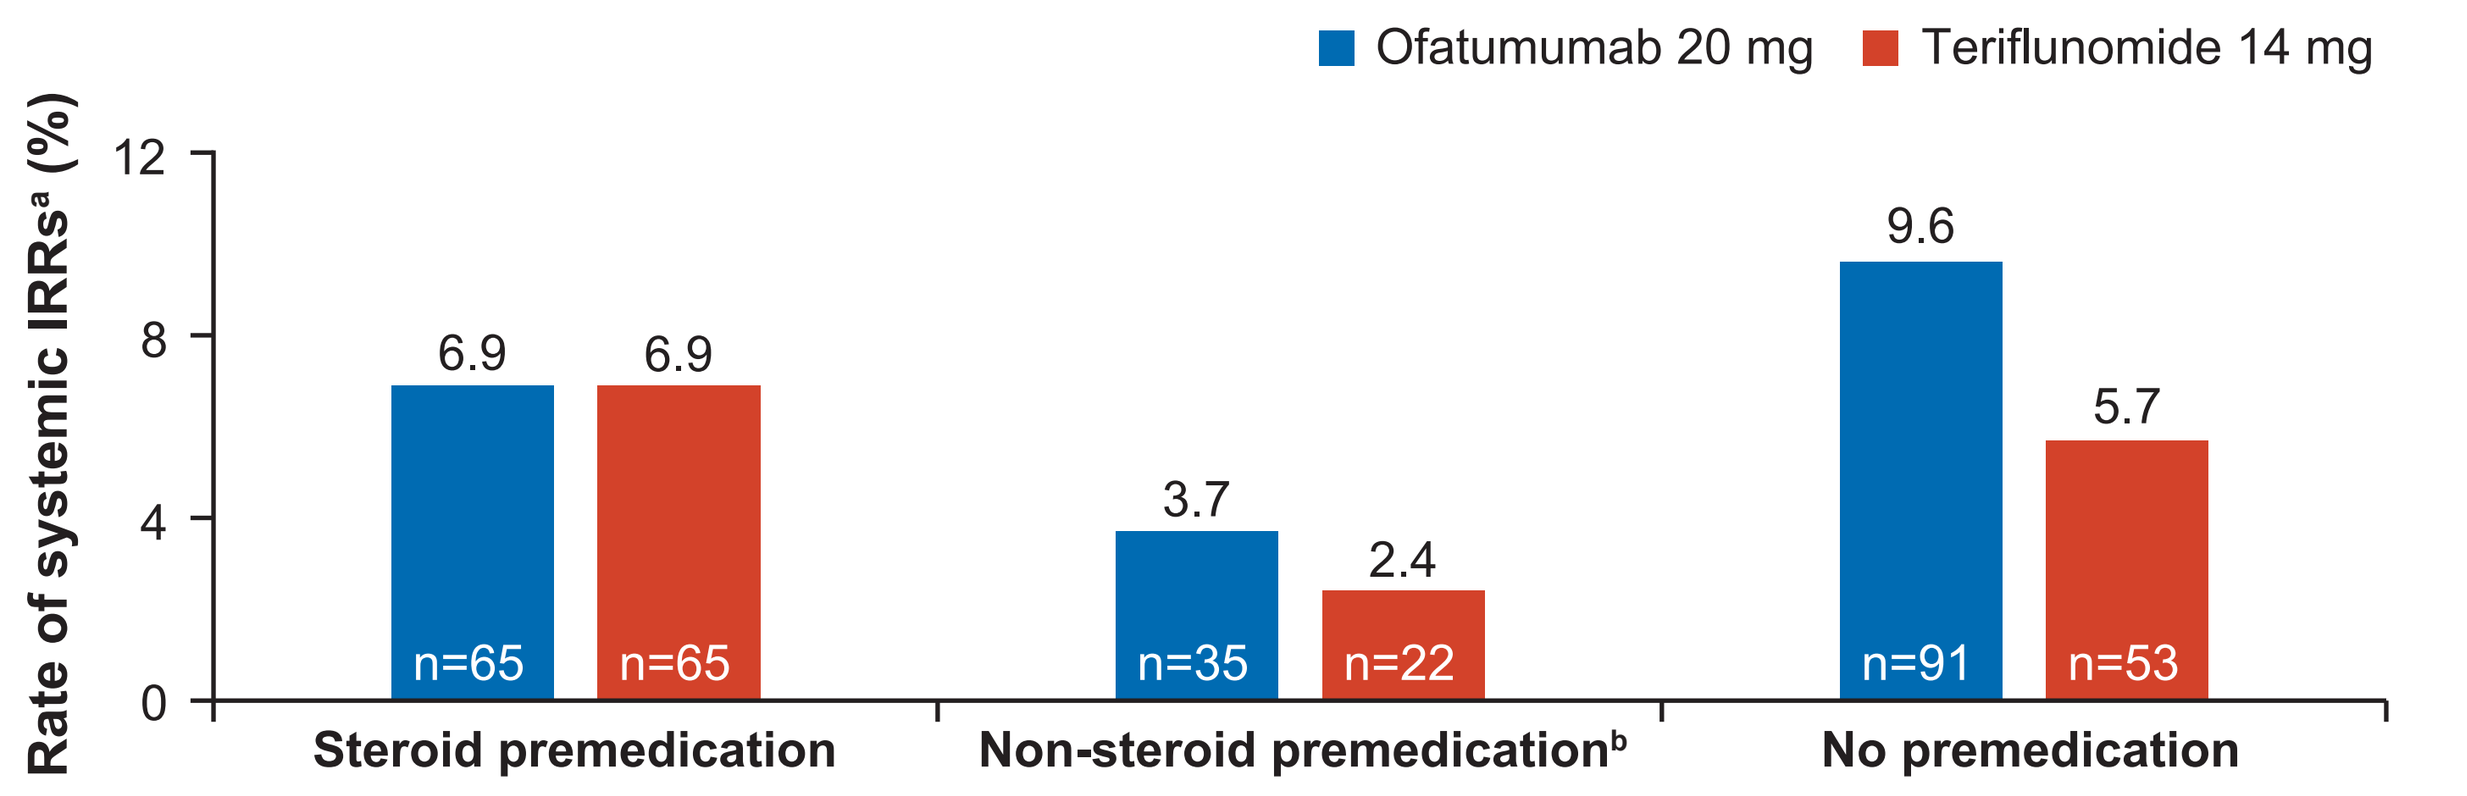
Ofatumumab 20 mg
Teriflunomide 14 mg
Rate of systemic IRRsᵃ (%)
12
8
4
0
6.9
6.9
n=65
n=65
3.7
2.4
n=35
n=22
9.6
5.7
n=91
n=53
Steroid premedication
Non-steroid premedicationᵇ
No premedication

IRRs, injection-related reactions

[a]Percentages are based on number of patients in the safety set; [b]Patients who never received any steroid premedication

- When no premedication was used at all injections, although the number of reactions overall was low, a higher proportion of patients in the ofatumumab group reported systemic IRRs compared with matching placebo injections in the teriflunomide group (9.6% vs 5.7%)
- With any premedication, a limited benefit in reduction in IRRs was observed in the ofatumumab group versus matching placebo injections in the teriflunomide group (steroids [6.9% vs 6.9%]; non-steroids [3.7% vs 2.4%]); **Figure 4**)

Conclusions

- Both systemic and local IRRs observed with ofatumumab 20 mg s.c. were mostly (99.8%) mild to moderate in severity and non-serious in nature
- The IRRs were predominantly reported with the first injection with ofatumumab compared with the matching placebo in teriflunomide group and the incidence decreased and was similar with subsequent injections in both treatment groups
- All IRRs were manageable with very low withdrawal rate (0.1%) and recovered with symptomatic treatment even in the absence of premedication
- Only limited benefit of premedication with steroids and non-steroids was observed in RMS clinical trials; premedication may not be required with ofatumumab

References

1. Bar-Or A, et al, *Neurology*. 2018;90(20):e1805-e1814.
2. Smith P, et al. Presented at *ECTRIMS* 2016;P1143.
3. Hauser SL, et al. Presented at *ECTRIMS* 2019; S17.OP336.

Acknowledgments

The authors acknowledge the following Novartis employees: **Saimithra Thammera** for medical writing assistance and coordinating author reviews, and **Mantosh Roy** for creative design assistance. The final responsibility for the content lies with the authors.

Disclosures

The study was supported by Novartis Pharma AG, Switzerland.

Amit Bar-Or participated as a speaker in meetings sponsored by and received consulting fees and/or grant support from: Janssen/Actelion; Atara Biotherapeutics, Biogen Idec, Celgene/Receptos, Roche/Genentech, Medimmune, Merck/EMD Serono, Novartis, Sanofi-Genzyme.

Jeffrey A. Cohen received personal compensation for consulting for Adamas, Convelo, MedDay, Mylan, and Population Council; and serving as an Editor of Multiple Sclerosis Journal

Patricia K. Coyle has received consulting fees from Accordant, Alexion, Bayer, Biogen Idec, Celgene, Genentech/Roche, Genzyme/Sanofi, Novartis, Serono and TG Therapeutics; and research support from Actelion, Alkermes, Genentech/Roche, MedDay, Novartis and NINDS.

Anne H. Cross has consulted for AbbVie, Bayer, Biogen, EMD Serono, Genentech/Roche, Genzyme/Sanofi, Mallinckrodt, Novartis and Teva

Stephen L Hauser serves on the board of trustees for Neurona and on scientific advisory boards for Alector, Annexon, Bionure, and Molecular Stethoscope, and has received travel reimbursement and writing assistance from F. Hoffmann-La Roche Ltd and Novartis AG for CD20-related meetings and presentations.

Ludwig Kappos' institution (University Hospital Basel) has received the following exclusively for research support: steering committee, advisory board and consultancy fees (Actelion, Addex, Bayer HealthCare, Biogen Idec, Biotica, Genzyme, Lilly, Merck, Mitsubishi, Novartis, Ono Pharma, Pfizer, Receptos, Sanofi, Santhera, Siemens, Teva, UCB and Xenoport); speaker fees (Bayer HealthCare, Biogen Idec, Merck, Novartis, Sanofi and Teva); support for educational activities (Bayer HealthCare, Biogen, CSL Behring, Genzyme, Merck, Novartis, Sanofi and Teva); license fees for Neurostatus products; and grants (Bayer HealthCare, Biogen Idec, European Union, Innoswiss, Merck, Novartis, Roche Research Foundation, Swiss MS Society and Swiss National Research Foundation).

Ratnakar Pingili, Cecile Kerloeguen, Ayan Das Gupta, Valentine Jehl, Dieter A. Haering, Krishnan Ramanathan, Martin Merschhemke, are employees of Novartis.

Poster presentation at the Consortium of Multiple Sclerosis Centers (CMSC) Virtual Annual Meeting, 2020.

Visit the web at:
http://novartis.medicalcongressposters.com/Default.aspx?doc=3418a

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Presenter email address: ratnakar.pingili@novartis.com

Text: Q3418a

To: 8NOVA (86682) US Only
+18324604729 North, Central and South Americas; Caribbean; China
+447860024038 UK, Europe & Russia
+46737494608 Sweden, Europe

Scan this QR code to download a copy Poster