Ofatumumab vs Teriflunomide in Relapsing Multiple Sclerosis: Analysis of No Evidence of Disease Activity (NEDA-3) from ASCLEPIOS I and II Trials

Stephen L. Hauser[1], Amit Bar-Or[2], Jeffrey A. Cohen[3], Giancarlo Comi[4], Jorge Correale[5], Patricia K. Coyle[6], Anne H. Cross[7], Jérôme de Seze[8], Xavier Montalban[9,10], Krzysztof Selmaj[11], Heinz Wiendl[12], Roman Willi[13], Bingbing Li[14], Dieter A. Häring[13], Krishnan Ramanathan[13], Martin Merschhemke[13], Ludwig Kappos[15]

[1]Department of Neurology, UCSF Weill Institute for Neurosciences, University of California, San Francisco, CA, USA; [2]Center for Neuroinflammation and Experimental Therapeutics and Department of Neurology, Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA, USA; [3]Department of Neurology, Mellen MS Center, Neurological Institute, Cleveland Clinic, Cleveland, OH, USA; [4]University Vita-Salute San Raffaele, Milan, Italy; [5]Institute for Neurological Research Dr. Raul Carrea, Buenos Aires, Argentina; [6]Department of Neurology, Stony Brook University, Stony Brook, NY, USA; [7]Washington University School of Medicine, Saint Louis, MO, USA; [8]University Hospital of Strasbourg, Strasbourg, France; [9]St Michael's Hospital, University of Toronto, Toronto, Ontario, Canada; [10]Department of Neurology-Neuroimmunology, Centre d'Esclerosi Multiple de Catalunya (Cemcat), Hospital Universitario Vall d' Hebron, Barcelona, Spain; [11]Center for Neurology, Lodz, Poland; [12]University of Muenster, Germany; [13]Novartis Pharma AG, Basel, Switzerland; [14]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [15]Neurologic Clinic and Policlinic, Departments of Medicine, Clinical Research, Biomedicine and Biomedical Engineering, University Hospital and University of Basel, Basel, Switzerland

Background

- Ofatumumab, the first fully human anti-CD20 monoclonal antibody,[1] depletes CD20+ B cells and CD20+ T cells in the blood and lymphoid tissues through complement-dependent cytotoxicity and antibody-dependent cell-mediated cytotoxicity[2,3]
- In the Phase 3 ASCLEPIOS I and II trials, subcutaneous (s.c.) ofatumumab 20 mg (0.4 mL) monthly demonstrated superior efficacy versus oral teriflunomide 14 mg once daily, and a favorable safety profile in patients with relapsing multiple sclerosis (RMS)[4]
 - Relative reduction in annualized relapse rate (ARR): 50.5% (p<0.001) in ASCLEPIOS I and 58.5% (p<0.001) in ASCLEPIOS II
 - Risk reduction in 3- and 6-month confirmed disability worsening (CDW): 34.4% (p=0.002) and 32.5% (p=0.012) in the pre-specified pooled analysis
 - Risk reduction in gadolinium-enhancing (Gd+) T1 lesions and new/enlarging T2 lesions: 97.5% (p<0.001) and 82% (p<0.001) in ASCLEPIOS I; 93.8% (p<0.001) and 84.5% (p<0.001) in ASCLEPIOS II, respectively

Objective

- To investigate the effect of s.c. ofatumumab 20 mg versus oral teriflunomide 14 mg in achieving no evidence of disease activity (NEDA-3), and to separately assess the ARR in the pooled ASCLEPIOS I and II trials

Methods

Study design and treatment pattern

- Data were pooled from the randomized, double-blind, double-dummy, active comparator-controlled, parallel-group, multicenter, adaptive and flexible duration design Phase 3 ASCLEPIOS I and II trials (maximum duration of up to 30 months)[4]
- Both trials had an identical study design and were conducted in parallel, enrolling 1882 RMS patients (ASCLEPIOS I [N=927], ASCLEPIOS II [N=955]) from 385 sites across 37 countries (**Figure 1**)
- Patients received ofatumumab 20 mg (0.4 mL) s.c. injections on Days 1, 7 and 14 (*initial doses*) and every 4 weeks from Week 4 onwards (*subsequent doses*) or teriflunomide 14 mg oral once daily

Figure 1. Study design of ASCLEPIOS I and II[4]

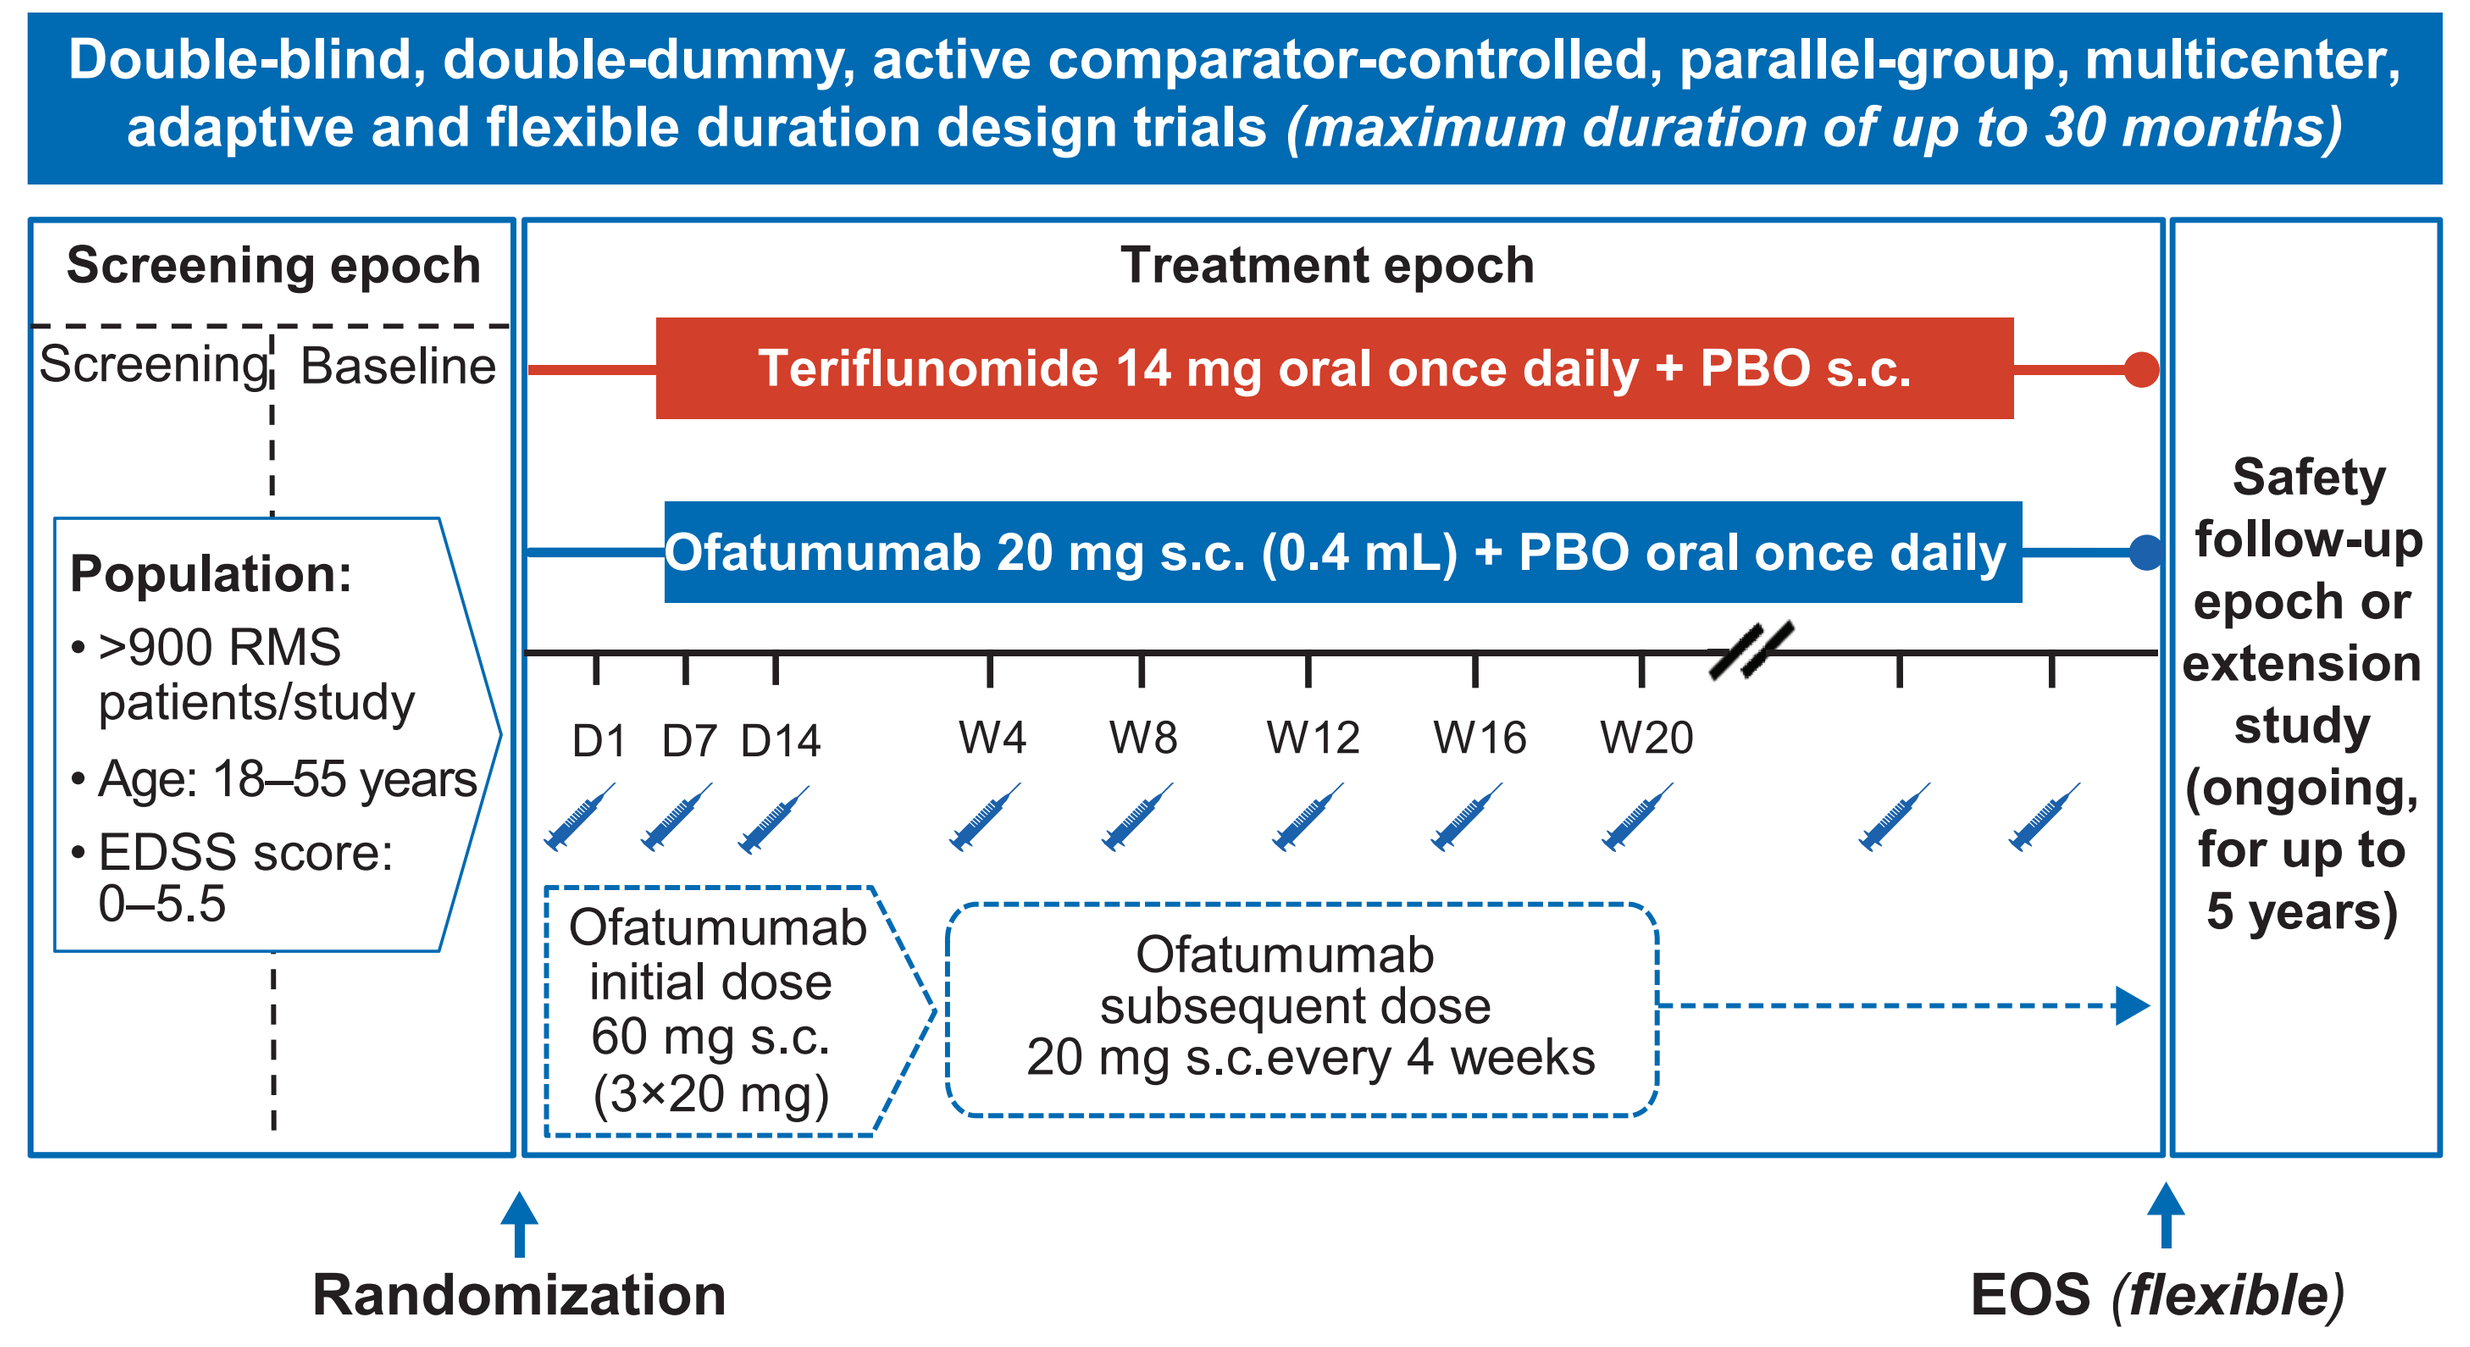

Double-blind, double-dummy, active comparator-controlled, parallel-group, multicenter, adaptive and flexible duration design trials (maximum duration of up to 30 months)
Screening epoch
Screening
Baseline
Treatment epoch
Teriflunomide 14 mg oral once daily + PBO s.c.
Ofatumumab 20 mg s.c. (0.4 mL) + PBO oral once daily
Population:
• >900 RMS patients/study
• Age: 18–55 years
• EDSS score: 0–5.5
D1 D7 D14 W4 W8 W12 W16 W20
Ofatumumab initial dose 60 mg s.c. (3×20 mg)
Ofatumumab subsequent dose 20 mg s.c.every 4 weeks
Safety follow-up epoch or extension study (ongoing, for up to 5 years)
Randomization
EOS (flexible)

D, day; EDSS, Expanded Disability Status Scale; EOS, end of study; RMS, relapsing multiple sclerosis; PBO, placebo; s.c., subcutaneous; W, week

Study assessments and statistical analysis

- NEDA-3* and its components in Year 1 and 2 were analyzed via logistic regression model[5] in modified full analysis set (mFAS)
 - The mFAS used for NEDA calculations consisted of all patients in the FAS, except those who discontinued from study drug prematurely for reasons other than 'lack of efficacy' or 'death' and had NEDA before early discontinuation. Patients who discontinued from study drug prematurely for reasons 'lack of efficacy' or 'death' were considered as having evidence of disease activity in the analysis (even if no evidence of disease activity was reported)
- ARR by time intervals were analyzed via negative binomial model in FAS

**Defined as no 6-month confirmed disability worsening [6mCDW], no confirmed multiple sclerosis [MS] relapse, no new/enlarging T2 lesions, and no gadolinium-enhancing [Gd+] T1 lesions*

Results

- Patient demographics and baseline disease characteristics of pooled ASCLEPIOS I and II trials were consistent across treatment groups and representative of a typical RMS population **(Table 1)**

Table 1. Patient demographics and baseline characteristics (pooled ASCLEPIOS I and II trials)

Characteristics Mean±SD or n (%)	Teriflunomide 14 mg (N'=936)	Ofatumumab 20 mg (N'=946)	All patients (N'=1882)
Age (years)	38.0±9.22	38.4±9.04	38.2±9.13
Female, n (%)	636 (67.9)	637 (67.3)	1273 (67.6)
Duration of MS since first symptoms (years)	8.19±7.3	8.27±7.1	8.23±7.2
Previously treated with DMTs, n (%)	573 (61.2)	560 (59.2)	1133 (60.2)
Number of relapses in last 12 months	1.3±0.71	1.2±0.69	1.3±0.70
EDSS score	2.90±1.4	2.93±1.4	2.92±1.4
T2 lesion volume (cm³)	12.5±13.8	13.7±13.8	13.1±13.8
Patients free of Gd+ T1 lesions, n (%)	584 (62.4)	561 (59.3)	1145 (60.8)
Number of Gd+ T1 lesions	1.3±3.4	1.7±4.5	1.5±4.0

DMT, disease-modifying therapy; EDSS, Expanded Disability Status Scale; FAS, full analysis set; Gd+, gadolinium-enhancing; MS, multiple sclerosis; N', number of patients in each group; RMS, relapsing multiple sclerosis; SD, standard deviation

Effect of ofatumumab on individual NEDA-3 components

- Over 2 years, more ofatumumab-treated patients were free of Gd+ T1 lesions and new/enlarging T2 lesions, compared with teriflunomide (**Figure 2A**)
- Over the entire ASCLEPIOS I and II trials, ofatumumab-treated patients (n=871) had a total of 52 Gd+ T1 lesions in 1678 magnetic resonance imaging (MRI) scans, versus 1016 Gd+ T1 lesions in 1608 MRI scans in teriflunomide-treated patients (n=856)
 - The adjusted number of Gd+ T1 lesions per scan was reduced with ofatumumab versus teriflunomide by 96%, rate ratio (95% confidence interval [CI]): 0.04 (0.03; 0.06)
- Over 2 years, more ofatumumab-treated patients were free of confirmed relapse (**Figure 2B**) and 6mCDW (**Figure 2C**), compared with teriflunomide

Effect of ofatumumab on NEDA-3 in Years 1, 2 and Years 0–2

- The odds of achieving NEDA-3 with ofatumumab versus teriflunomide was >3-fold higher in the first year and 8-fold higher in the second year of treatment (**Figure 3**)

Figure 2. Effect of ofatumumab on individual NEDA-3 components

A. MRI focal activity

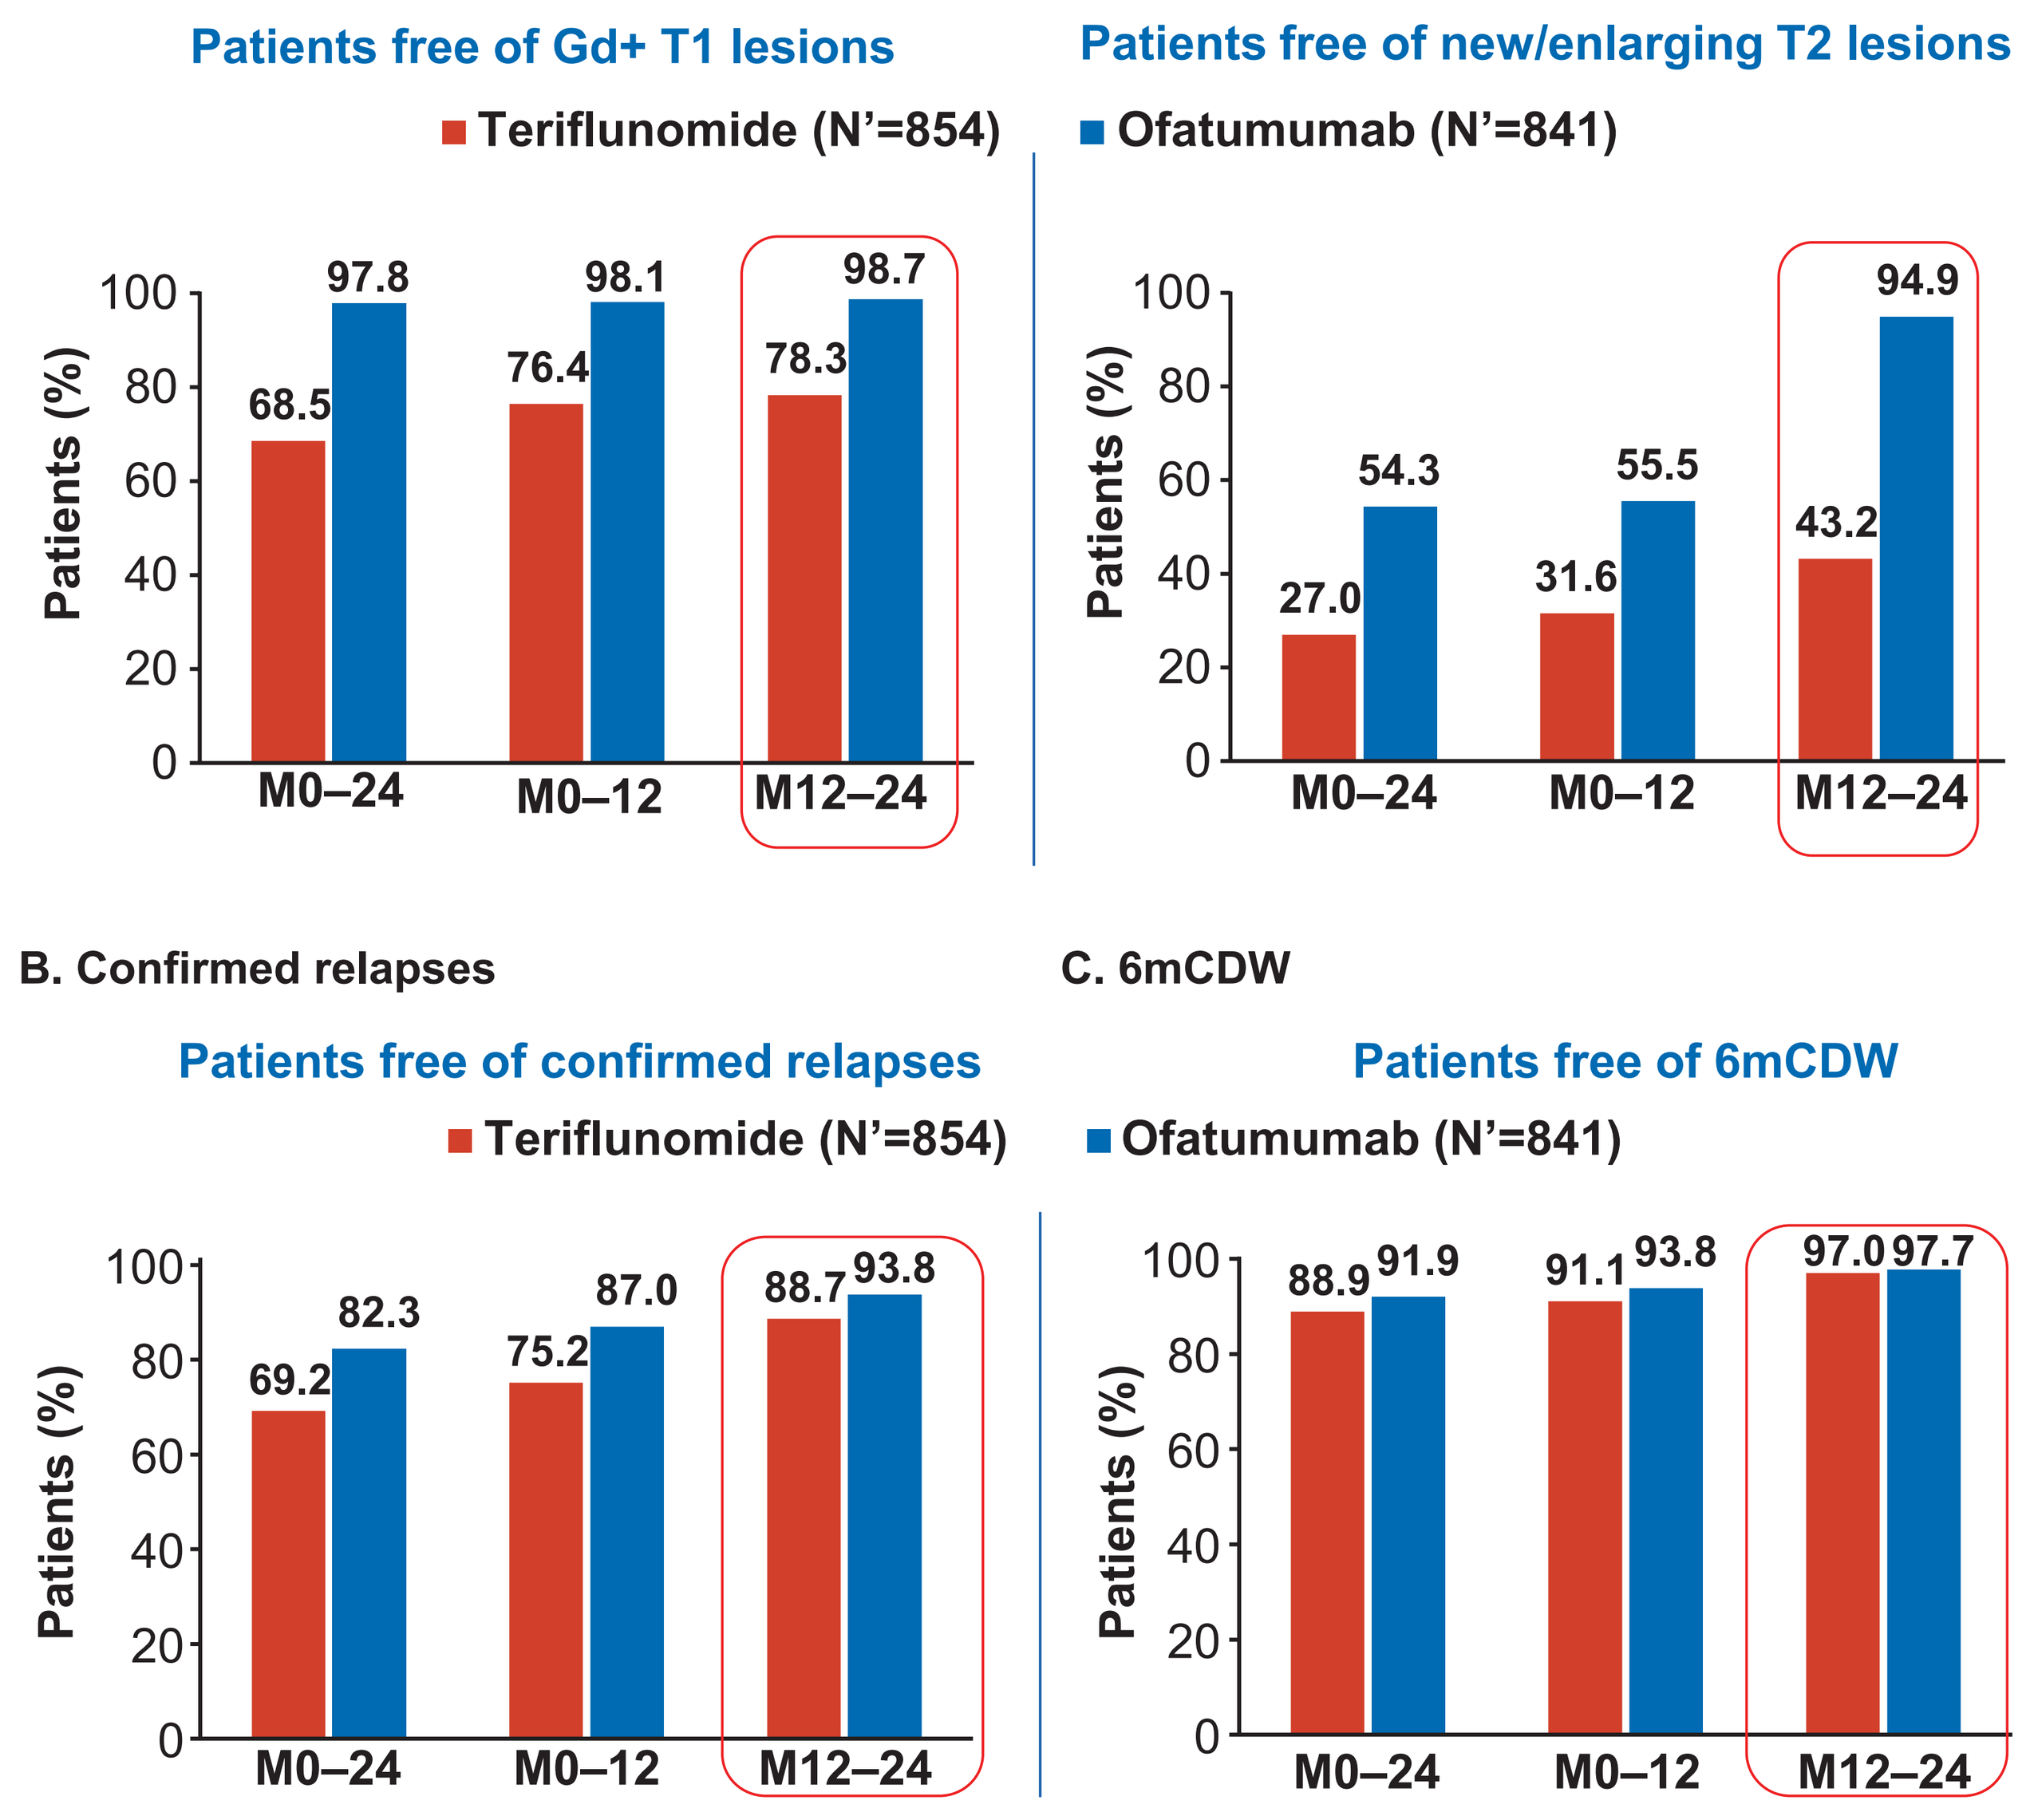

Patients free of Gd+ T1 lesions
Teriflunomide (N'=854)
Patients (%)
68.5 97.8
76.4 98.1
78.3 98.7
M0–24 M0–12 M12–24
Patients free of new/enlarging T2 lesions
Ofatumumab (N'=841)
27.0 54.3
31.6 55.5
43.2 94.9
B. Confirmed relapses
Patients free of confirmed relapses
69.2 82.3
75.2 87.0
88.7 93.8
C. 6mCDW
Patients free of 6mCDW
88.9 91.9
91.1 93.8
97.0 97.7

6mCDW, 6-month confirmed disability worsening; CI, confidence interval; Gd+, gadolinium-enhancing; M, month; MRI, magnetic resonance imaging; N', number of patients in each group; NEDA, no evidence of disease activity

Figure 3. Effect of ofatumumab on NEDA-3

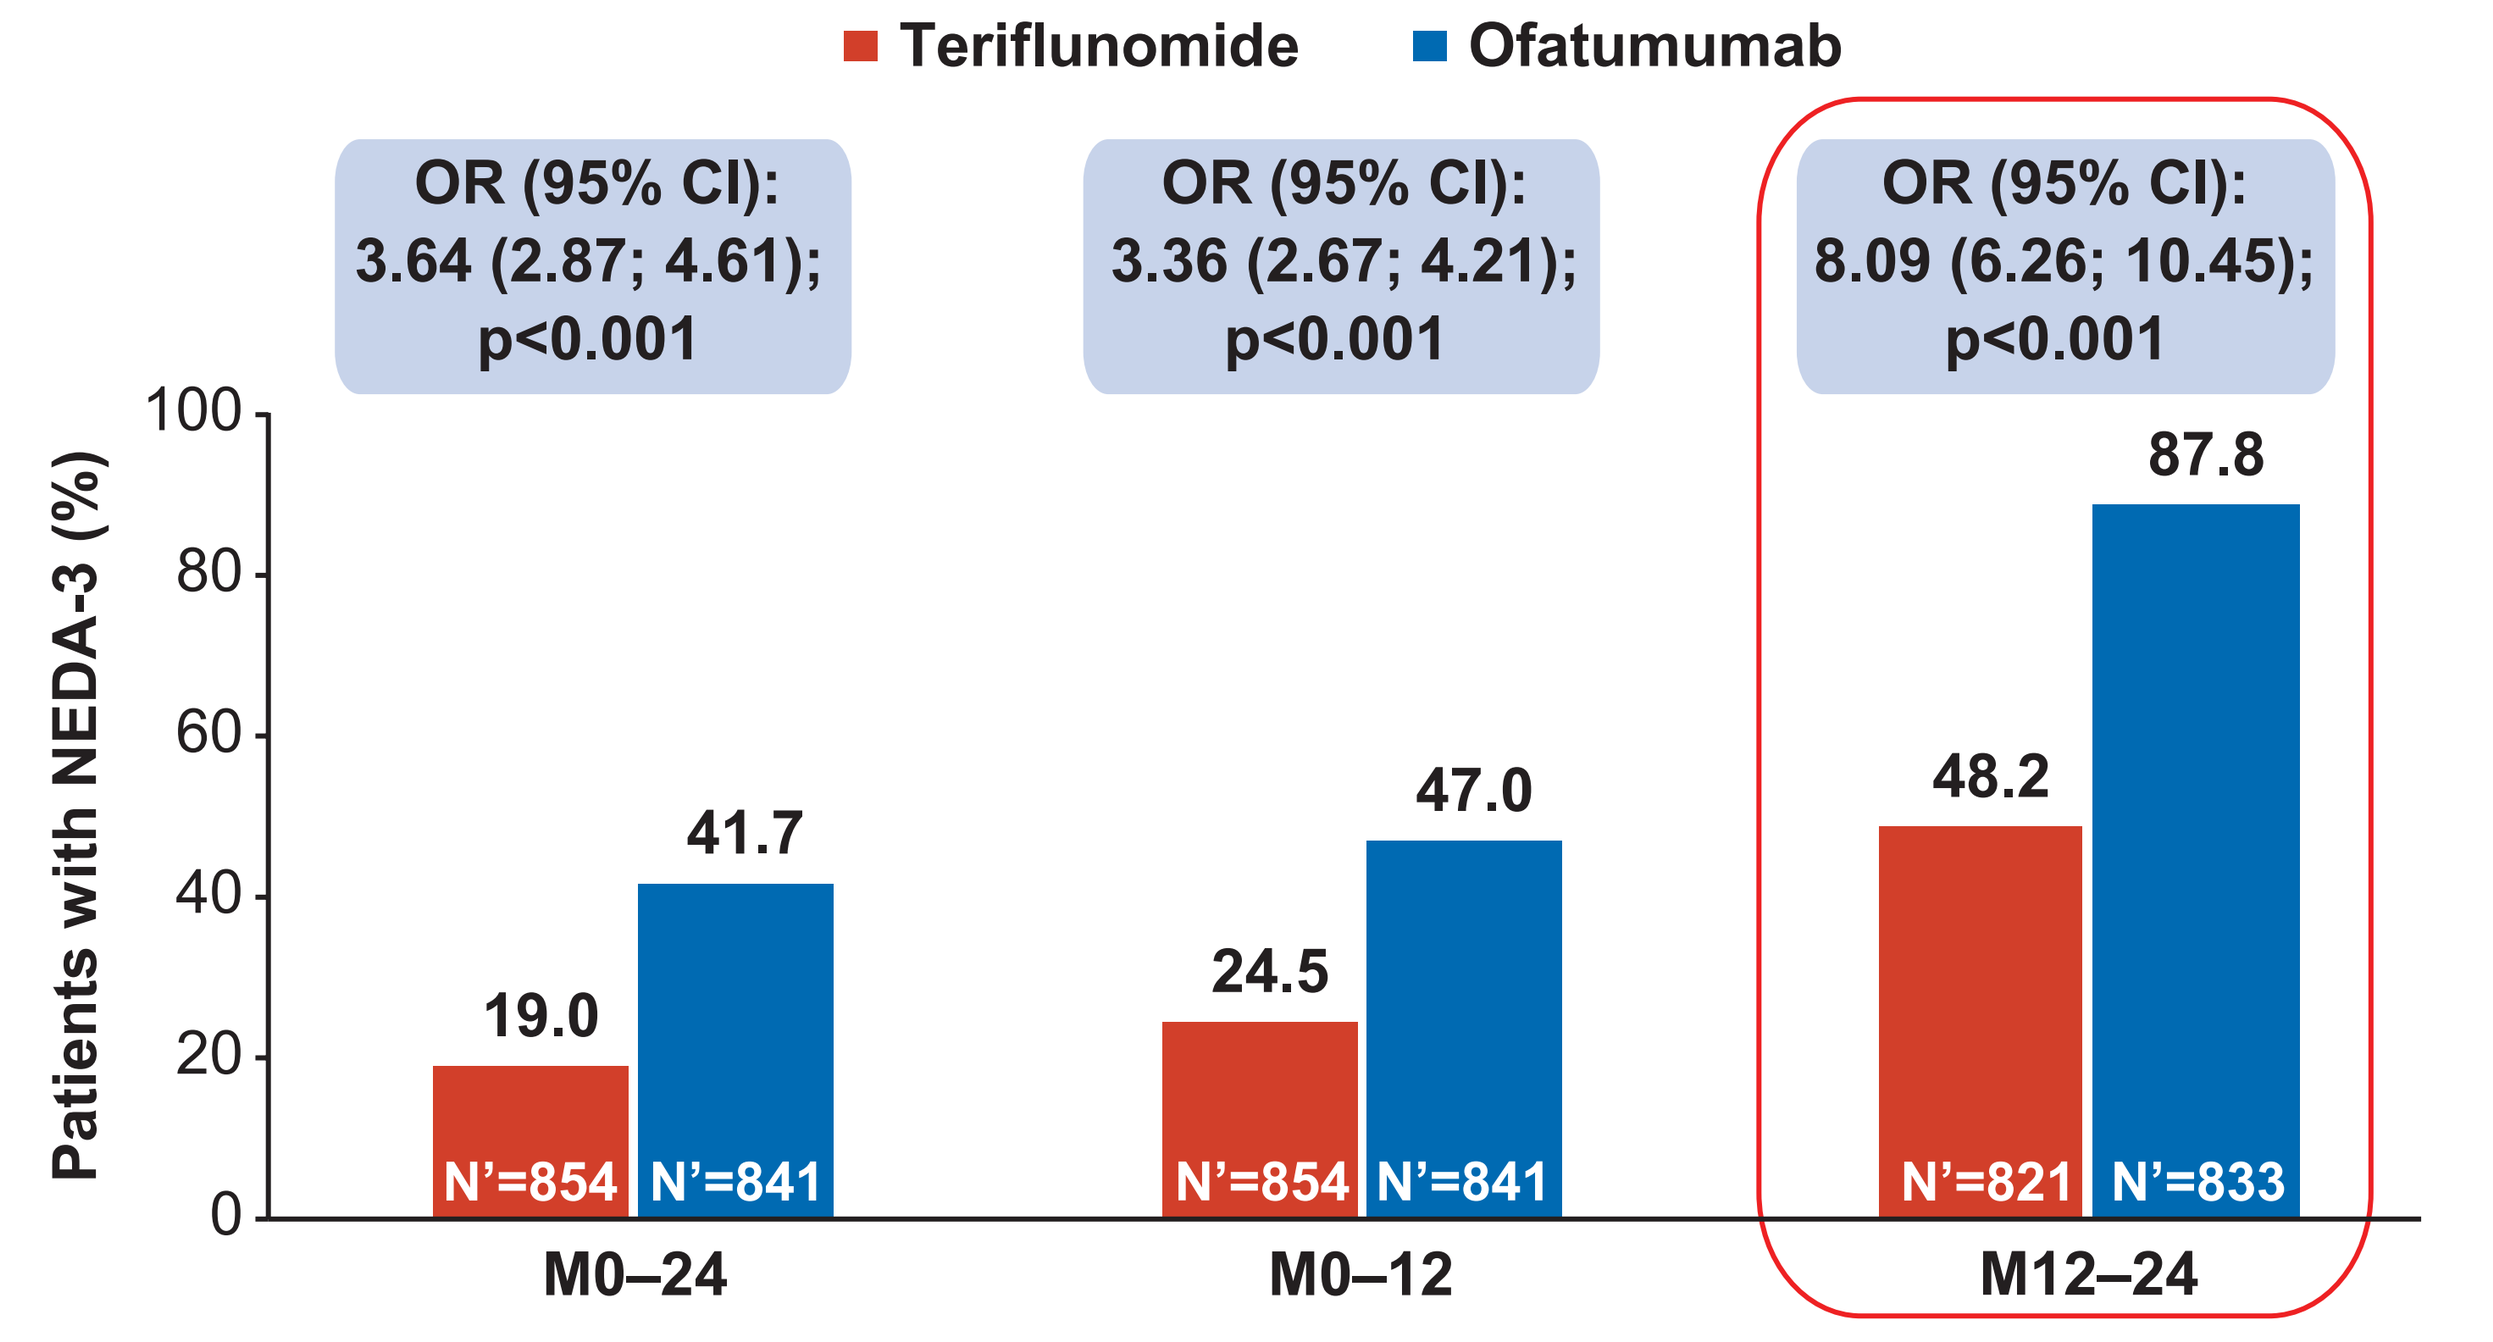

Teriflunomide
Ofatumumab
OR (95% CI): 3.64 (2.87; 4.61); p<0.001
OR (95% CI): 3.36 (2.67; 4.21); p<0.001
OR (95% CI): 8.09 (6.26; 10.45); p<0.001
Patients with NEDA-3 (%)
19.0 41.7
24.5 47.0
48.2 87.8
N'=854 N'=841
N'=854 N'=841
N'=821 N'=833
M0–24 M0–12 M12–24

CI, confidence interval; M, month; N', number of patients in each group; NEDA, no evidence of disease activity; OR, odds ratio

ARR by time interval

- Ofatumumab significantly reduced ARR versus teriflunomide within the first 3 months of treatment (M0–3: p=0.011), and in all subsequent time intervals (p<0.001) (**Figure 4**)

Figure 4. Effect of ofatumumab on ARR by time interval

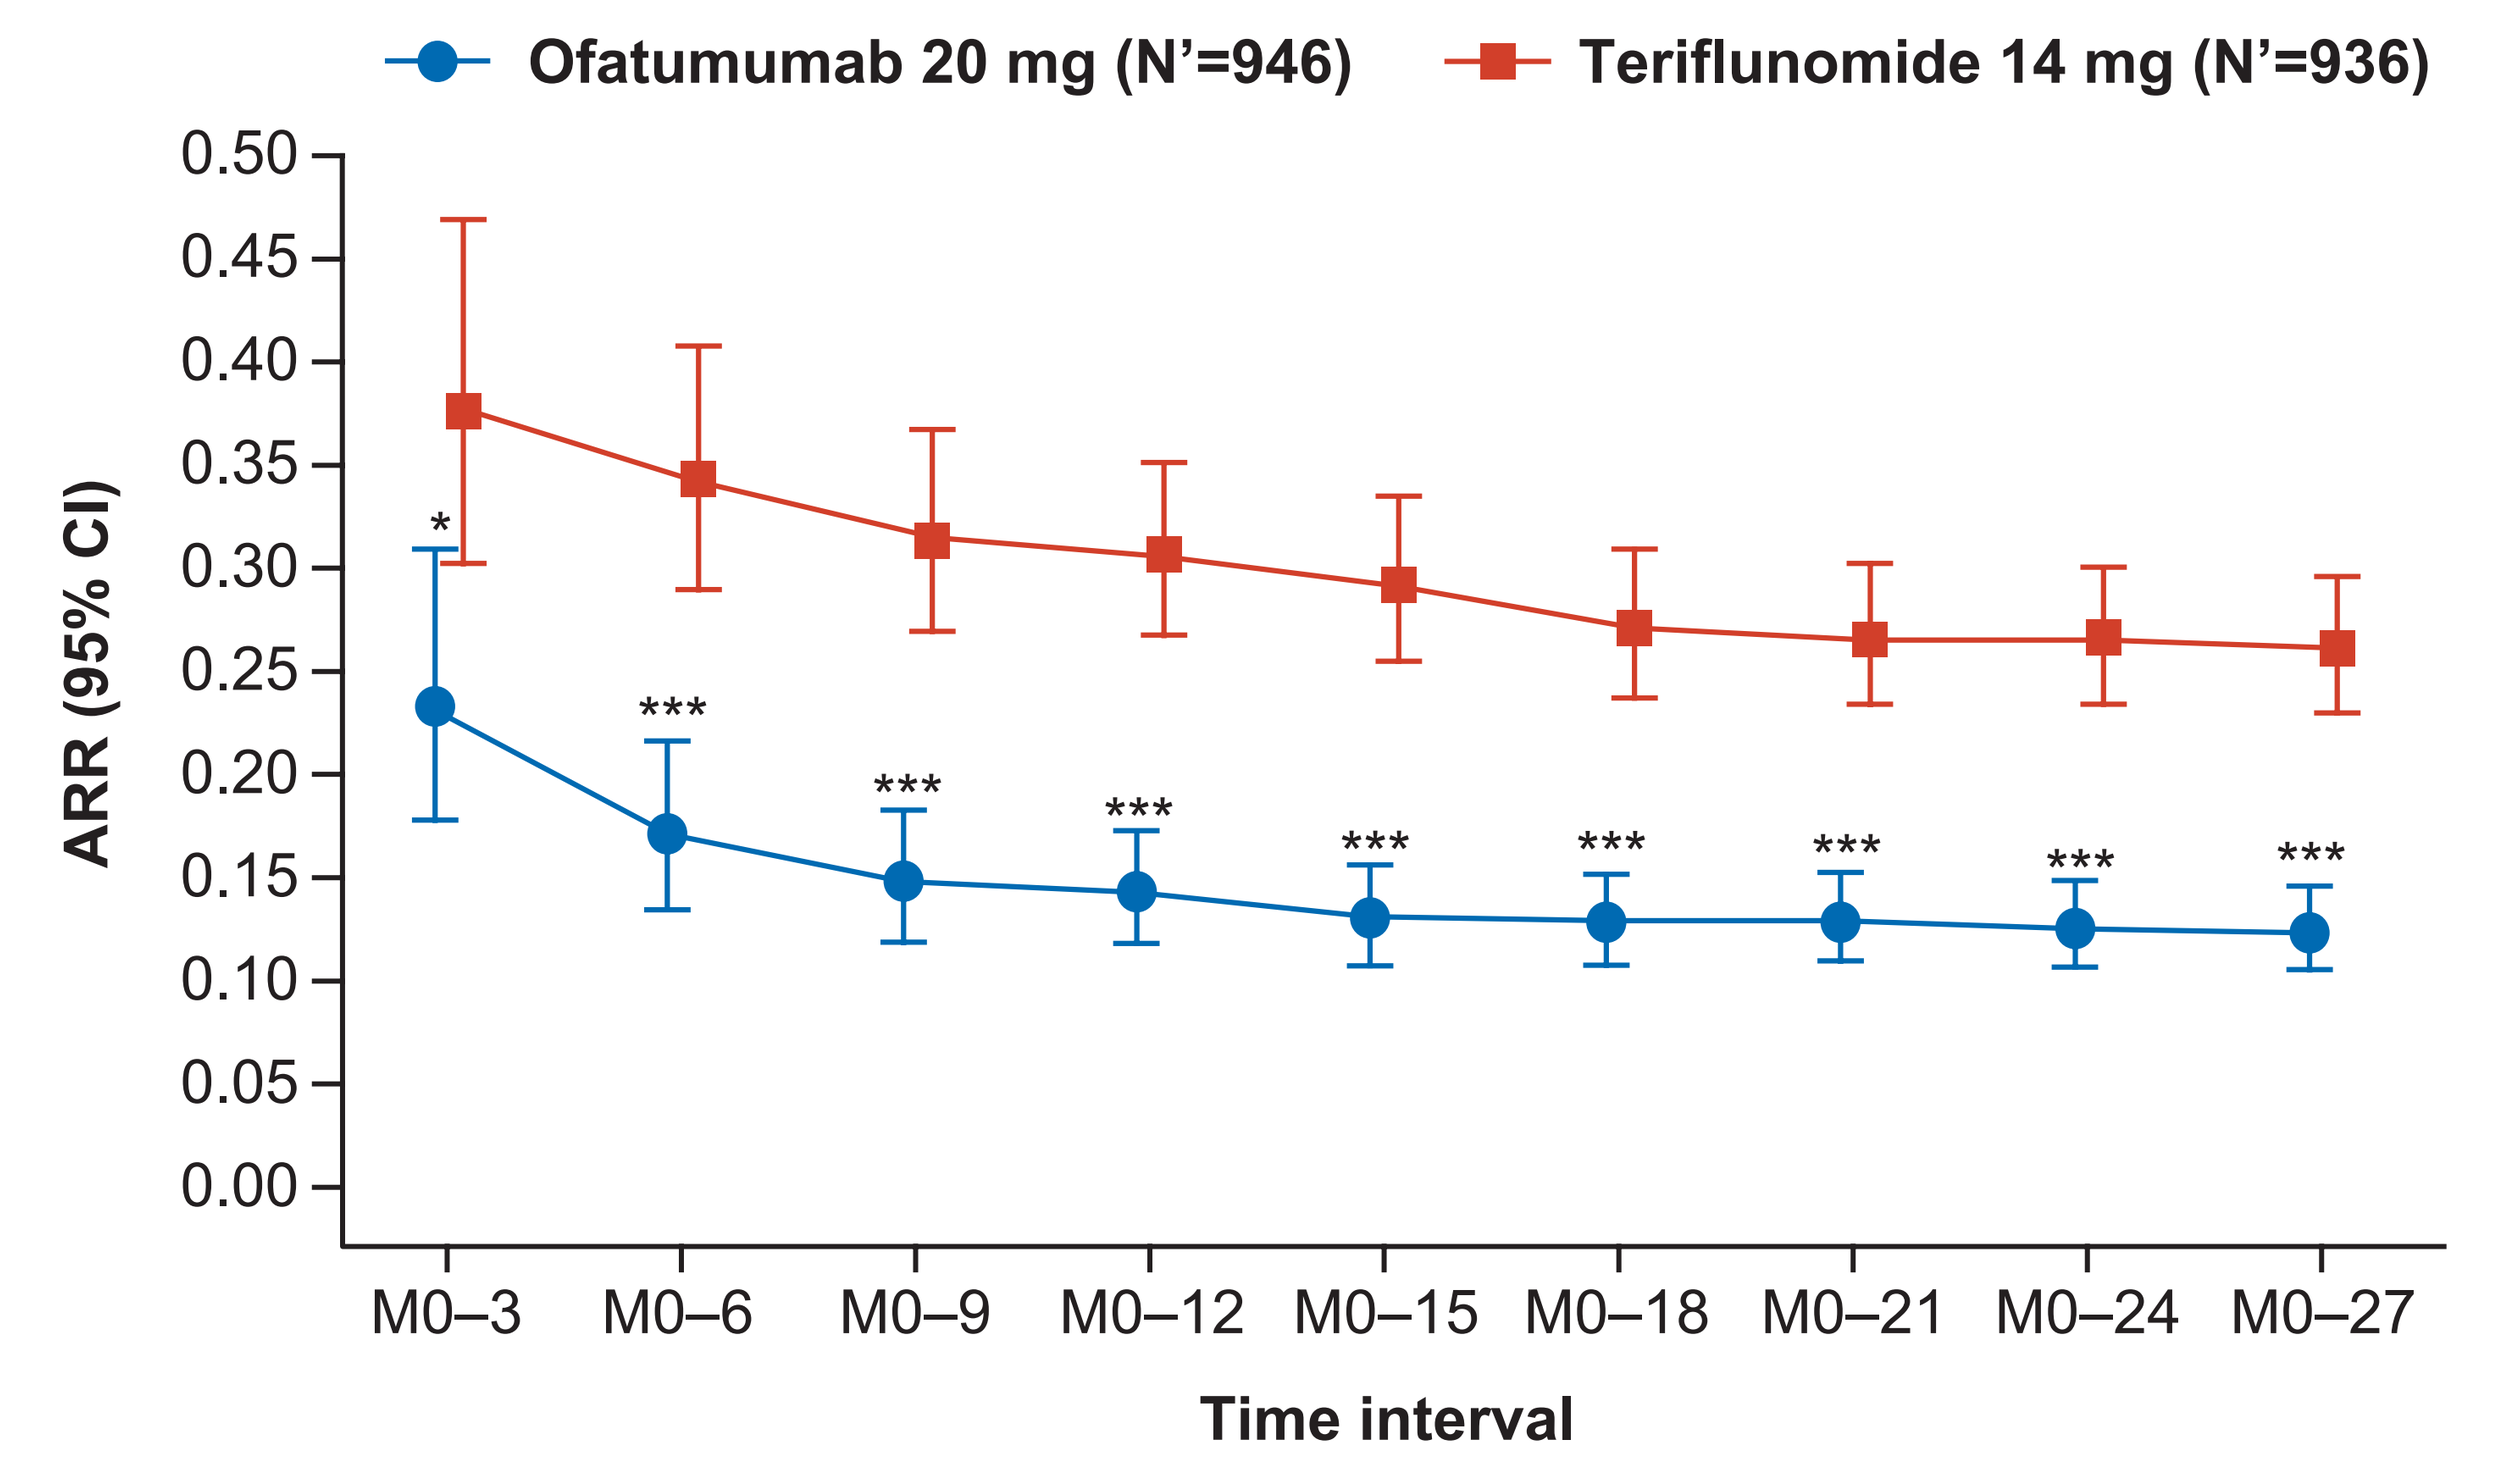

Ofatumumab 20 mg (N'=946)
Teriflunomide 14 mg (N'=936)
ARR (95% CI)
M0–3 M0–6 M0–9 M0–12 M0–15 M0–18 M0–21 M0–24 M0–27
Time interval

*p<0.05 and ***p<0.001 vs teriflunomide
ARR, annualized relapse rate; CI, confidence interval; M, month; N', total number of patients included in the analysis

Acknowledgments

The authors acknowledge the following Novartis employees: **Richa Chhabra** and **Anuja Shah** for medical writing assistance and coordinating author reviews, and **Mantosh Roy** for creative design assistance. The final responsibility for the content lies with the authors.

Disclosures

The study was funded by Novartis Pharma AG, Switzerland.

Stephen L. Hauser serves on the board of trustees for Neurona and on scientific advisory boards for Alector, Annexon, Bionure, and Molecular Stethoscope, and has received travel reimbursement and writing assistance from F. Hoffmann-La Roche Ltd and Novartis AG for CD20-related meetings and presentations.

Amit Bar-Or has participated as a speaker in meetings sponsored by and received consulting fees and/or grant support from: Janssen/Actelion; Atara Biotherapeutics, Biogen Idec, Celgene/Receptos, Roche/Genentech, Medimmune, Merck/EMD Serono, Novartis, Sanofi-Genzyme.

Jeffrey A. Cohen has received personal compensation for consulting for Adamas, Convelo, MedDay, Mylan, and Population Council; and serving as an Editor of Multiple Sclerosis Journal.

Giancarlo Comi received personal compensation for consulting and speaking activities from Novartis, Teva Pharmaceutical Industries Ltd, Teva Italia Srl, Sanofi Genzyme, Genzyme Corporation, Genzyme Europe, Merck KGgA, Merck Serono SpA, Celgene Group, Biogen Idec, Biogen Italia Srl, F. Hoffmann-La Roche, Roche SpA, Almirall SpA, Forward Pharma, MedDay and Excemed.

Jorge Correale received personal compensation from Merck Serono Argentina, Novartis Argentina, Genzyme LATAM, Genzyme Global, Biogen Idec LATAM, Merck Serono LATAM, Biogen Idec Argentina, Genzyme Argentina, Novartis LATAM, Novartis Global and Teva Argentina.

Patricia K. Coyle received personal compensation from Accordant, Alexion, Bayer, Biogen MA, Inc., Celgene Corporation, Genentech/Roche, Genzyme/Sanofi, GlaxoSmithKline, Novartis Pharmaceuticals Corporation, Serono and TG Therapeutics. She also received research support from Actelion, Alkermes, Corrona LLD, Genentech/Roche, MedDay, NINDS, Novartis Pharmaceuticals Corporation and PCORI.

Anne H. Cross received personal compensation from Biogen, Celgene, EMD Serono, Genentech/Roche, Novartis and TG Therapeutics.

Jérôme de Seze received personal compensation from Alexion, Allergan, Almirall, Bayer, Biogen, Chugai, CSL Behring, F. Hoffmann-La Roche Ltd, Genzyme, LFB, Merck, Novartis and Teva.

Xavier Montalban has received speaking honoraria and travel expenses for participation in scientific meetings, and has been a steering committee member of clinical trials or participated in advisory boards of clinical trials in the past years with Actelion, Bayer, Biogen, Celgene, Merck, Novartis, Roche, Sanofi-Genzyme, Teva Pharmaceuticals, Excemed, MSIF and NMSS.

Summary and Conclusions

- Ofatumumab significantly increased the chances of patients achieving NEDA-3 both in the first and second year of treatment, versus teriflunomide
 - In the first year of treatment, nearly 5 out of 10 patients achieved NEDA-3 with ofatumumab
 - In the second year of treatment, nearly 9 out of 10 patients achieved NEDA-3 with ofatumumab
- Treatment with ofatumumab led to nearly complete abrogation of MS disease activity and worsening in the second year of treatment:
 - 93.8% free of relapses
 - 97.7% free of 6mCDW
 - 98.7% free of Gd+ T1 lesions
 - 94.9% free of new/enlarging T2 lesions
- Significant reduction in ARR in the first three months and in all subsequent time-intervals for over 2 years with ofatumumab treatment versus teriflunomide, suggests sustained efficacy of ofatumumab in RMS patients

References

1. Bar-Or A, et al. *Neurology*. 2018;90(20):e1805–e1814.
2. Theil D, et al. Front Immunol. 2019;20(10):1340.
3. Pacheco-Fernandez T, et al. Presented at the *AAN* 2018;S52.003.
4. Hauser SL, et al. Presented at the *ECTRIMS* 2019; S17.OP336.
5. Methodology adopted from Havrdová E, et al. *Mult Scler J Exp Transl Clin.* 2018;4:2055217318760642.

Xavier Montalban has also received research support through his institution from Biogen, Merck, Novartis, Roche, Sanofi-Genzyme, and Teva Pharmaceuticals.

Krzysztof Selmaj received personal compensation from Biogen, Novartis, Roche, Merck, Genzyme and Celgene, and research support from Roche.

Heinz Wiendl has received honoraria for acting as a member of Scientific Advisory Boards Biogen, Evgen, Genzyme, MedDay Pharmaceuticals, Merck Serono, Novartis, Roche Pharma AG, and Sanofi-Aventis as well as speaker honoraria and travel support from Alexion, Biogen, Cognomed, F. Hoffmann-La Roche Ltd., Gemeinnützige Hertie-Stiftung, Merck Serono, Novartis, Roche Pharma AG, Genzyme, TEVA, and WebMD Global. Prof. Wiendl is acting as a paid consultant for Abbvie, Actelion, Biogen, IGES, Johnson & Johnson, Novartis, Roche, Sanofi-Aventis, and the Swiss Multiple Sclerosis Society. His research is funded by the German Ministry for Education and Research (BMBF), Deutsche Forschungsgemeinschaft (DFG), Else Kröner Fresenius Foundation, Fresenius Foundation, the European Union, Hertie Foundation, NRW Ministry of Education and Research, Interdisciplinary Center for Clinical Studies (IZKF) Muenster and RE Children's Foundation, Biogen, GlaxoSmithKline GmbH, Roche Pharma AG, Sanofi-Genzyme.

Ludwig Kappos' institution (University Hospital Basel) has received the following exclusively for research support: steering committee, advisory board and consultancy fees (Actelion, Addex, Bayer HealthCare, Biogen Idec, Biotica, Genzyme, Lilly, Merck, Mitsubishi, Novartis, Ono Pharma, Pfizer, Receptos, Sanofi, Santhera, Siemens, Teva, UCB and Xenoport); speaker fees (Bayer HealthCare, Biogen Idec, Merck, Novartis, Sanofi and Teva); support for educational activities (Bayer HealthCare, Biogen, CSL Behring, Genzyme, Merck, Novartis, Sanofi and Teva); license fees for Neurostatus products; and grants (Bayer HealthCare, Biogen Idec, European Union, Innoswiss, Merck, Novartis, Roche Research Foundation, Swiss MS Society and Swiss National Research Foundation).

Roman Willi, Bingbing Li, Dieter A. Häring, Krishnan Ramnathan and **Martin Merschhemke** are employees of Novartis.

Poster Presentation at the Consortium of Multiple Sclerosis Centers (CMSC) Virtual Annual Meeting, 2020.

Visit the web at:
http://novartis.medicalcongressposters.com/Default.aspx?doc=f7e89

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Presenter email address: Stephen.Hauser@ucsf.edu

Text: Qf7e89

To: 8NOVA (86682) US Only
+18324604729 North, Central and South Americas; Caribbean; China
+447860024038 UK, Europe & Russia
+46737494608 Sweden, Europe

Scan this QR code to download a copy Poster