Poster presented at CMSC Annual Meeting 2023, May 31-June 3, 2023, Aurora, Colorado, USA

LB09

Real-world Satisfaction and Experience with Injection and Autoinjector Device for Ofatumumab indicated for Multiple Sclerosis

Amy Perrin Ross[1], Jacqueline Nicholas[2], Ming-Hui Tai[3], Stephen Yeung[3], Nazneen Fatima Shaikh[4], Helen Chen[4], Mariana Fernandes[4], Kevin Hawkins[4]

[1]Neuroscience Nurse Consultant, Oak Brook, IL 60660, USA; [2]OhioHealth Multiple Sclerosis Center, 3535 Olentangy River Rd, Suite 1501, Columbus, OH 43214, USA; [3]Novartis Pharmaceuticals Corporation, One Health Plaza, East Hanover, NJ 07936, USA; [4]Real World Solutions, IQVIA, 2400 Ellis Road Durham, NC 27703, USA

CONCLUSIONS

1. MS patients reported high satisfaction levels and positive experiences with the use of the OMB Sensoready pen in real-world practice, mostly driven by reasonable administration time and ease-of-use
2. These findings provide early insights for clinicians and MS patients to build confidence for the use of Sensoready pen for OMB administration in the US

Copyright © 2023 Novartis Pharma AG. All rights reserved.

Scan to download a copy of this poster

Copies of this poster and its content, obtained through this QR code, are for personal use only and may not be reproduced without written permission from the authors

INTRODUCTION

- Ofatumumab (OMB; Kesimpta®) is a high-efficacy disease-modifying therapy (DMT) approved in August 2020 for the treatment of relapsing multiple sclerosis (MS) in the US[1]
- OMB is self-administered subcutaneously once a month via a single-use Sensoready® autoinjector pen.[1] Real-world evidence on patients' satisfaction and experiences with the Sensoready pen used to administer OMB is limited

OBJECTIVE

- To evaluate the overall satisfaction, device usability, and injection experience of MS patients self-administering OMB using the Sensoready pen

METHODS

STUDY DESIGN

- This was a US-based, observational cross-sectional study with primary data collection via a questionnaire directly administered to patients with MS who received OMB using the Sensoready pen within the previous 12 months

STUDY INCLUSION/EXCLUSION CRITERIA

- Patients aged ≥18 years at the time of the survey, having a diagnosis of MS based on 2017 McDonald criteria,[2] newly prescribed OMB within the prior 12 months, and currently self-administering treatment using the Sensoready pen, were eligible for inclusion
- Patients who had received OMB injection as part of the randomized clinical trial, having active hepatitis B virus, and those having cognitive impairment at the time of the survey were excluded

VARIABLES

- Socio-demographic and clinical characteristics, and prior DMT experience were assessed at the time of survey participation
- Eligible patients were administered a 30-item de novo questionnaire that focused on overall device satisfaction, device usability, convenience/flexibility for travel with the device, user confidence, and injection experience
- Ratings were measured on Likert and numeric rating scales, with higher scores indicating positive responses

STATISTICAL ANALYSES

- Descriptive statistics were used to summarize all study variables. Data were summarized using frequency counts and percentages for categorical variables and mean and standard deviation (SD) for continuous variables
- All analyses were performed by IQVIA using SAS® statistical software

RESULTS

STUDY POPULATION

- Among 130 patients enrolled by physician/neurologist, 105 patients met the study eligibility criteria and were included in the analysis (**Figure 1**)

PATIENT SOCIO-DEMOGRAPHIC CHARACTERISTICS

- The mean (SD) age of patients was 42.5 (SD: 12.2) years and 63.8% were female
- Most patients were Caucasian (51.4%), resided in the Northeast US geographic region (72.4%), and had commercial insurance (66.7%)
- Most patients were married (61.0%), attended school/college/vocational training or had an associate degree (58.1%), and were employed/self-employed (62.9%) (**Figure 2**)

Figure 1. Patient selection

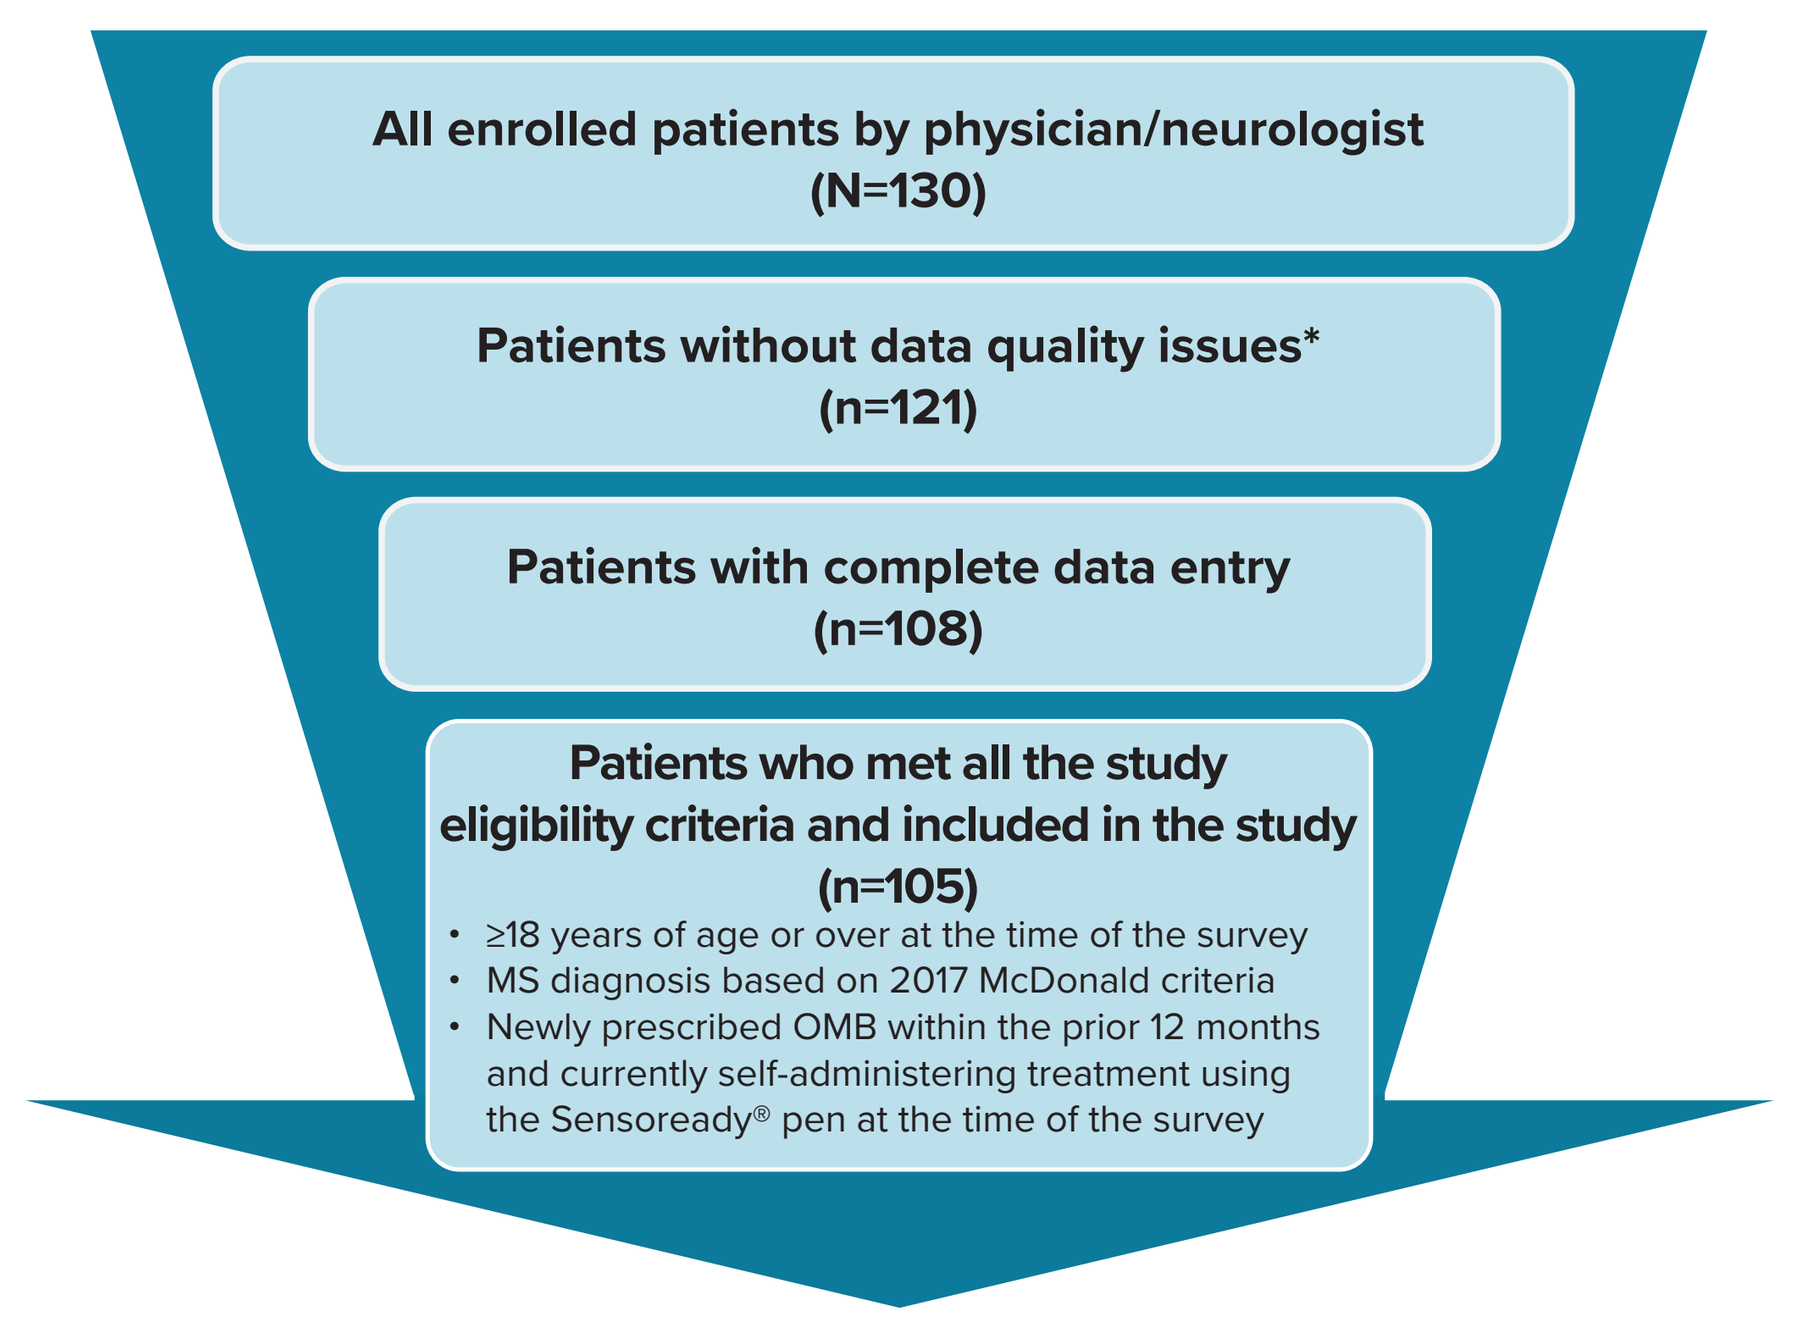
All enrolled patients by physician/neurologist (N=130)
Patients without data quality issues* (n=121)
Patients with complete data entry (n=108)
Patients who met all the study eligibility criteria and included in the study (n=105)
≥18 years of age or over at the time of the survey
MS diagnosis based on 2017 McDonald criteria
Newly prescribed OMB within the prior 12 months and currently self-administering treatment using the Sensoready® pen at the time of the survey

OMB, ofatumumab
*Amount of time participants spend on the survey was too short

Figure 2. Socio-demographic characteristics of patients who received OMB using the Sensoready pen (N=105)

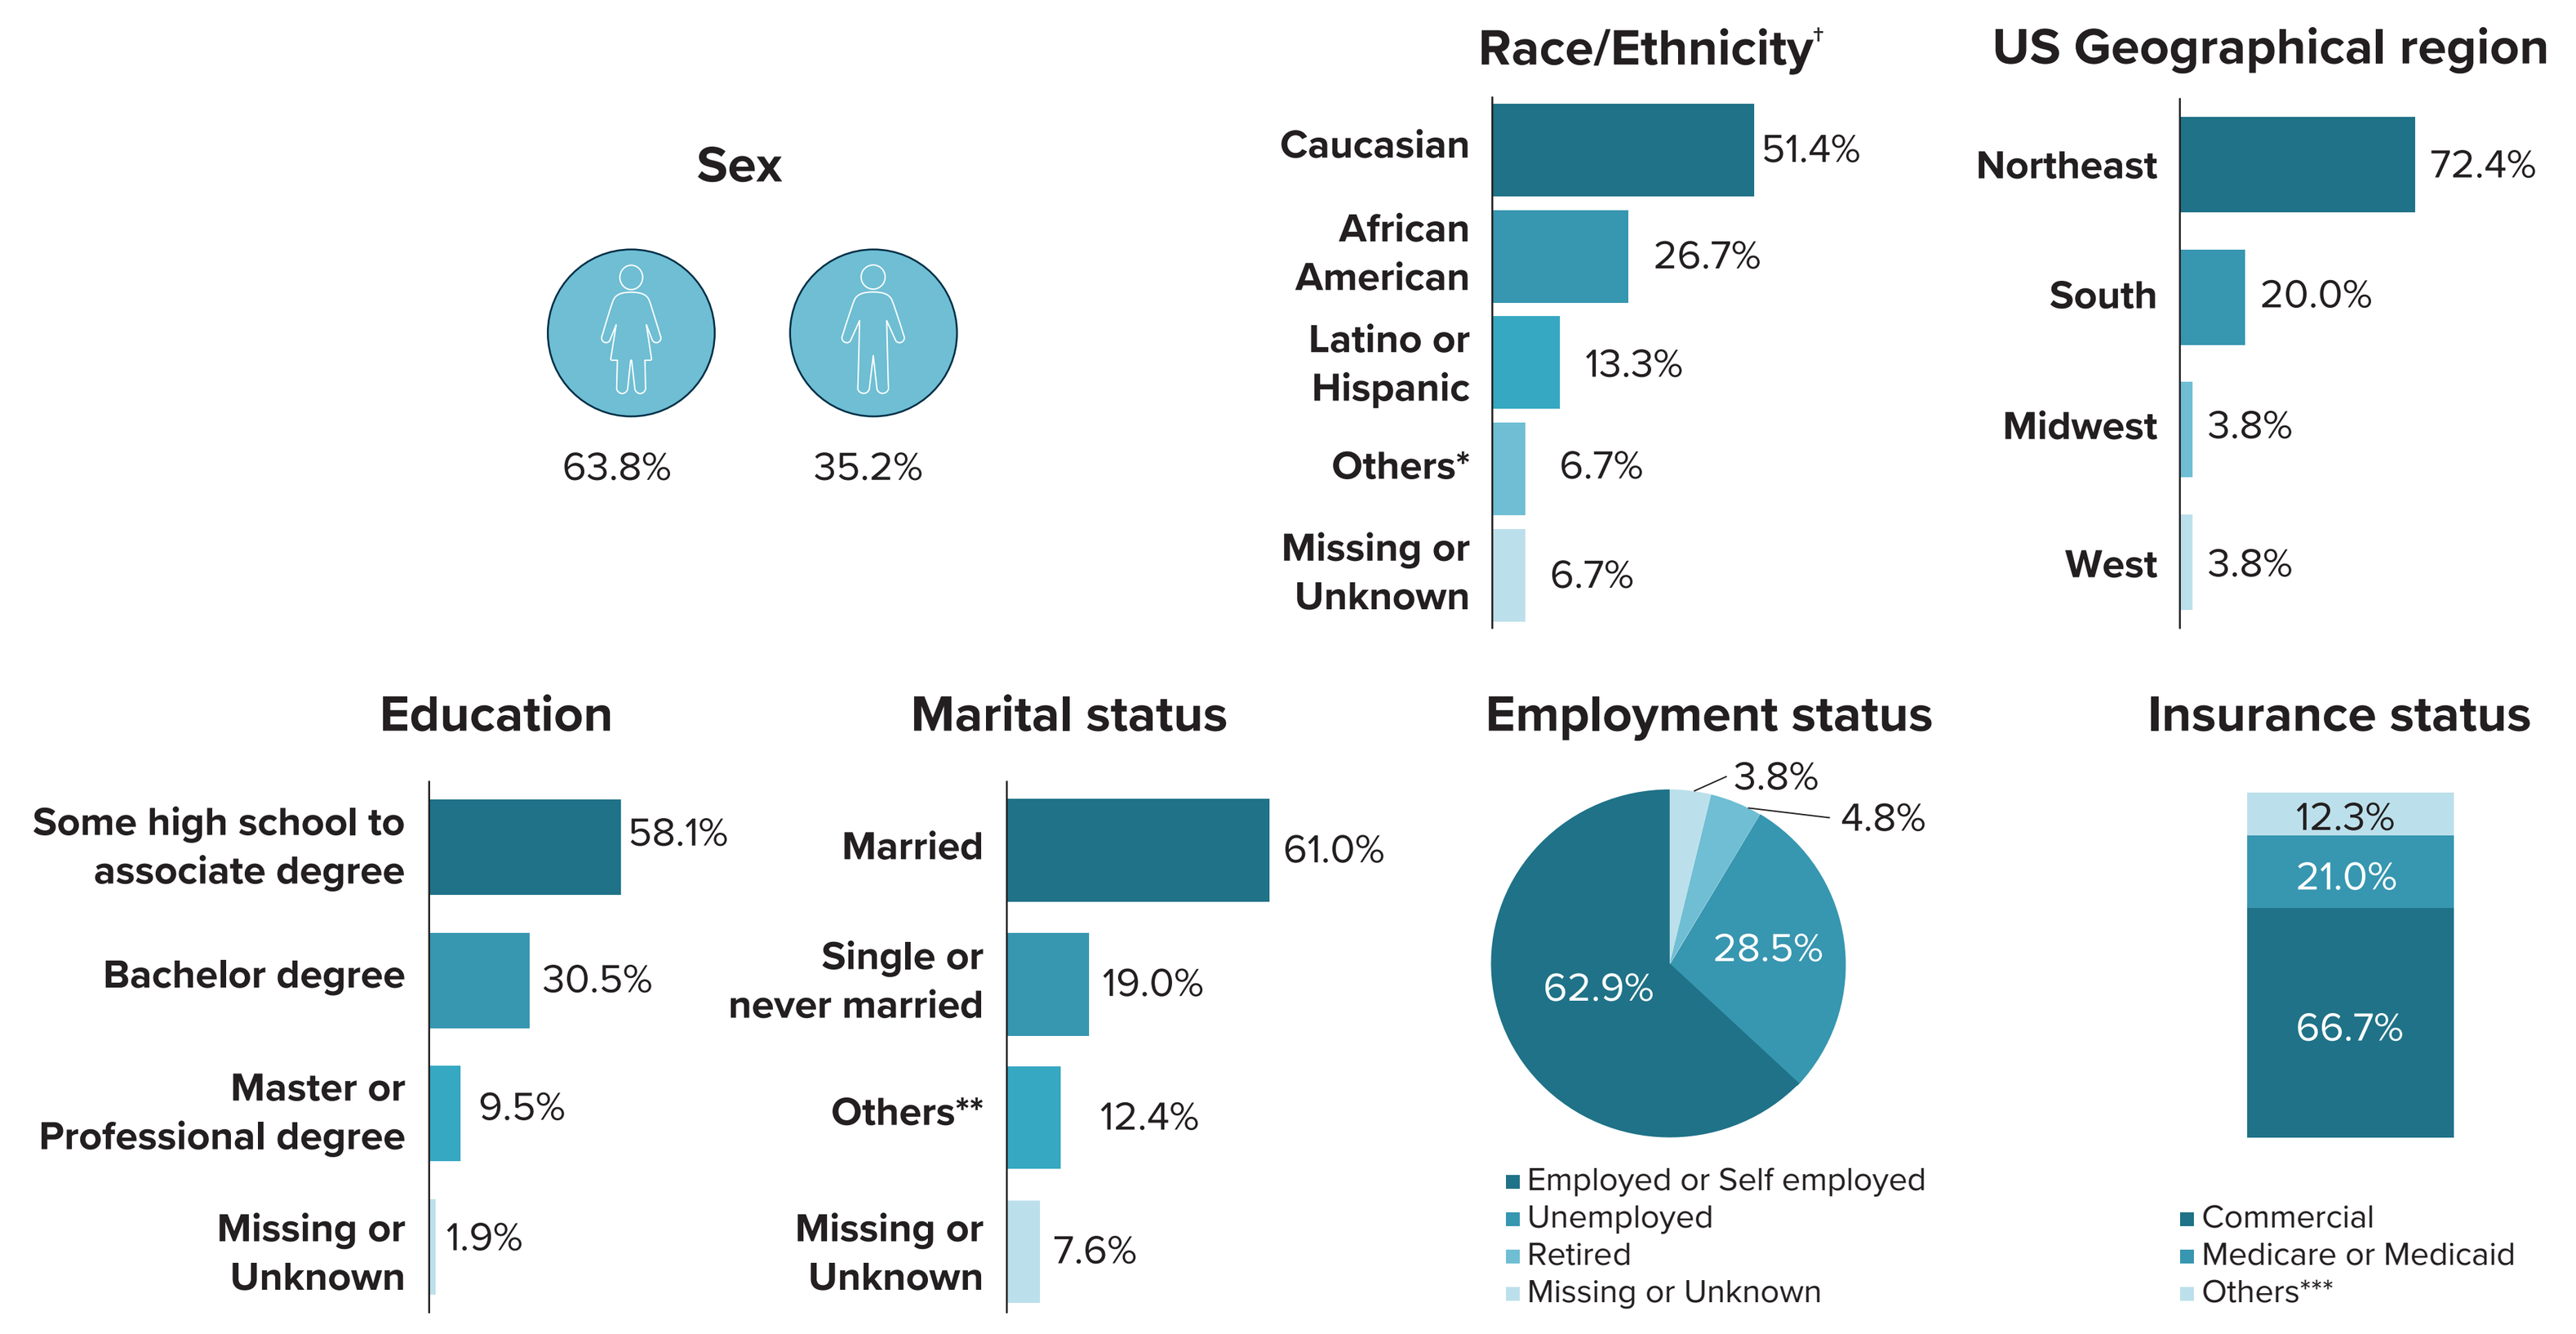
Sex
63.8%
35.2%
Race/Ethnicity†
Caucasian 51.4%
African American 26.7%
Latino or Hispanic 13.3%
Others* 6.7%
Missing or Unknown 6.7%
US Geographical region
Northeast 72.4%
South 20.0%
Midwest 3.8%
West 3.8%
Education
Some high school to associate degree 58.1%
Bachelor degree 30.5%
Master or Professional degree 9.5%
Missing or Unknown 1.9%
Marital status
Married 61.0%
Single or never married 19.0%
Others** 12.4%
Missing or Unknown 7.6%
Employment status
62.9%
28.5%
4.8%
3.8%
Employed or Self employed
Unemployed
Retired
Missing or Unknown
Insurance status
12.3%
21.0%
66.7%
Commercial
Medicare or Medicaid
Others***

OMF, ofatumumab
*Asians and Native Americans; **Domestic partnership, widowed, divorced, separated; ***Veterans Health Administration plan or Department of Defense (DoD)/ TRICARE, No insurance coverage/pay by cash, other categories; †Can be more than one answer per patient.

PATIENT CLINICAL CHARACTERISTICS

- The mean Patient Determined Disease Steps (PDDS) score of the study cohort was 2.3 (SD: 1.5), with the majority of patients reporting mild to severe disability/walking ability (84.8%)
- The majority of patients rated their general health status as good to excellent (78.1%)
- The mean time since MS diagnosis was 33.6 (SD: 36.8) months, with most patients having relapsing-remitting MS (41.9%) followed by primary progressive MS (33.3%). Most of the patients were DMT experienced (61.9%) at the time of the survey
- The average number of comorbid conditions was 2.0 (SD: 1.3) per patient. The most common comorbid conditions were anxiety (36.2%), high cholesterol (26.7%), and hypertension (22.9%) (**Table 1**)

Table 1. Clinical characteristics of patients who received OMB using the Sensoready pen (N=105)

Characteristics	Overall (N=105)
PDDS	
Mean (SD)	2.3 (1.5)
Category, n (%)	
Normal	16 (15.1%)
Mild to severe disability	89 (84.9%)
General health	
Mean (SD)	3.2 (0.9)
Category, n (%)	
Poor or fair	23 (21.9%)
Good to excellent	82 (78.1%)
MS phenotype, n (%)	
RRMS	44 (41.9%)
PPMS	35 (33.3%)
SPMS	15 (14.3%)
Missing or Unknown	11 (10.5%)
Mean (SD) time since MS diagnosis, months*	33.6 (36.8)
Prior DMT experience, n (%)	
Yes	65 (61.9%)
No	40 (38.1%)
Mean (SD) number of comorbid conditions per patient	2.0 (1.3)
Key comorbid conditions, n (%)**	
Anxiety	38 (36.2%)
High cholesterol	28 (26.7%)
Hypertension	24 (22.9%)
Chronic pain	21 (20.0%)
Depression	19 (18.1%)

DMT, disease-modifying therapy; MS, multiple sclerosis; OMB, ofatumumab; PPMS, primary progressive MS; RRMS, relapsing-remitting MS; SPMS, secondary progressive MS; PDDS, Patients Determined Disease Steps; SD, standard deviation
*Time since MS diagnosis (months) = ([survey completion date] – [diagnosis date] + 1)/30.44.**Can be more than one answer per patient.

PATIENT SATISFACTION

- The overall mean device satisfaction score was 4.4 (SD: 0.7)
- The majority of the patients (86.7%) expressed high satisfaction (i.e., rated either 4 [satisfied] or 5 [extremely satisfied] on a 5-point Likert scale) with the autoinjector device used for OMB administration (**Figure 3**)
- None of the patients were dissatisfied or extremely dissatisfied with the autoinjector device used for OMB administration

Figure 3. Overall satisfaction* with the Sensoready pen (N=105)

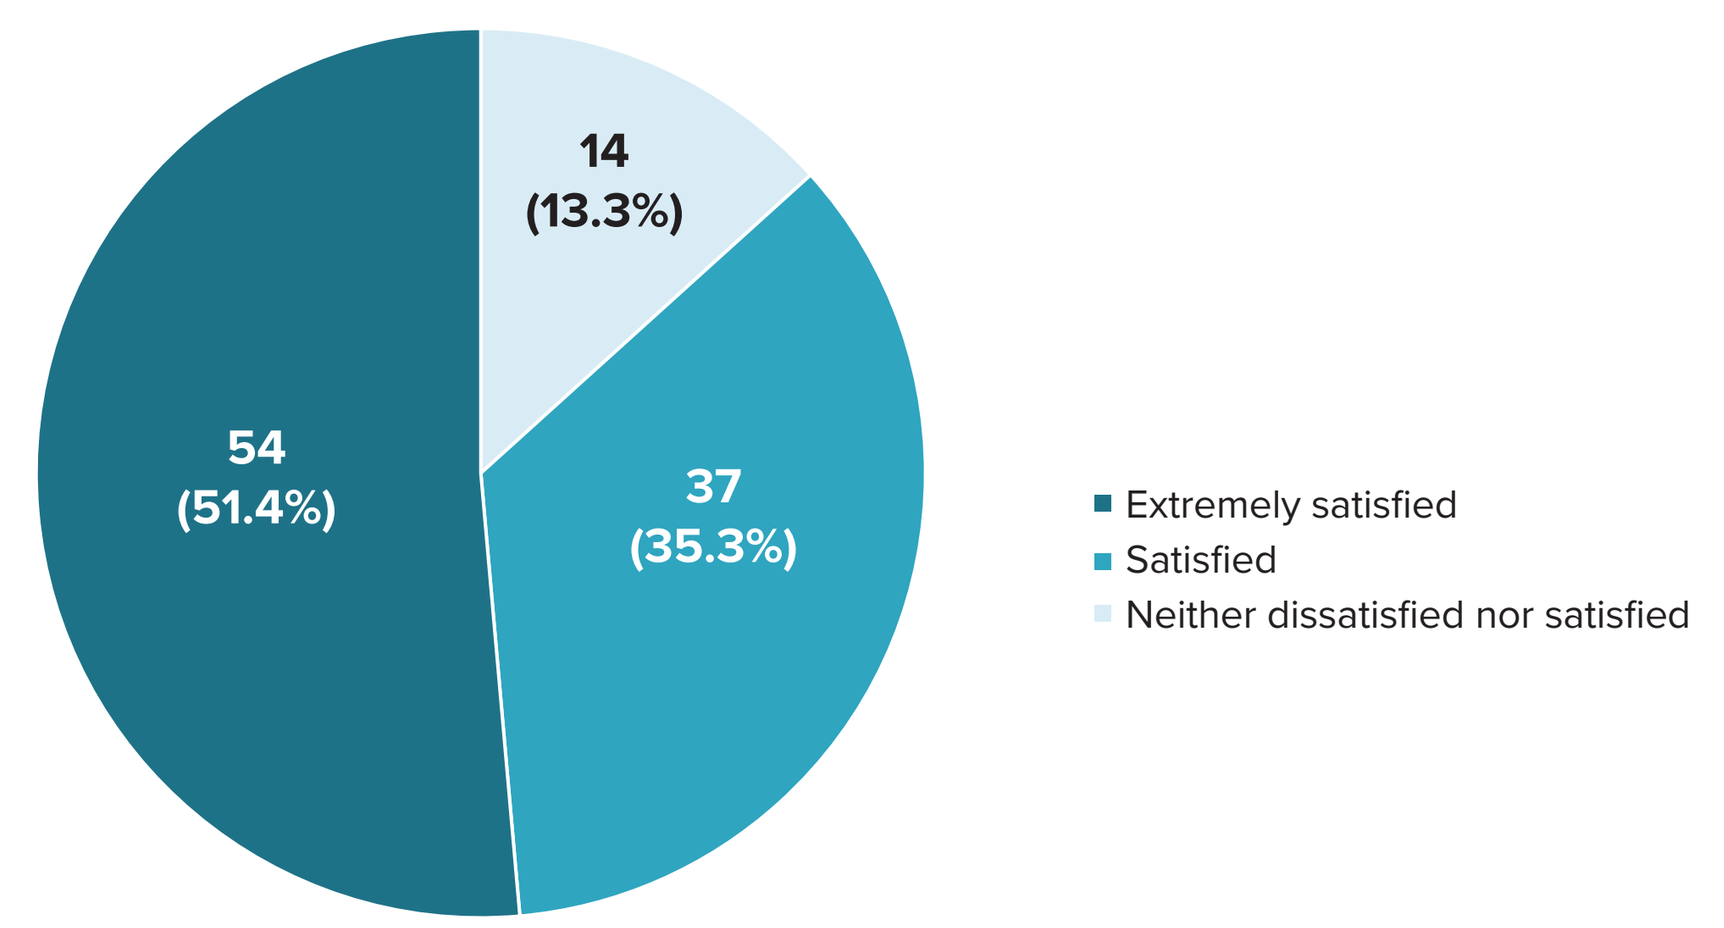
54 (51.4%)
37 (35.3%)
14 (13.3%)
Extremely satisfied
Satisfied
Neither dissatisfied nor satisfied

*Assessed using a 5-point Likert scale (1=extremely dissatisfied to 5=extremely satisfied)

- The most common reasons for high satisfaction included reasonable administration time (90.5%), overall ease of use (89.5%), a monthly dosing schedule of OMB (89.5%), the time required for device preparation (86.7%), ease of device preparation (81.9%), device ergonomics (76.2%), and portability (73.3%) (**Figure 4**)
- The majority of patients felt confident to self-administer OMB using the device (85.7%); the majority expressed their intention to continue with the OMB Sensoready pen (89.4%) and would recommend OMB to others (87.6%). Moreover, 77.1% of patients indicated that the use of an OMB Sensoready pen did not interfere with their daily activities

Figure 4. Device usability characteristics* (N=105)

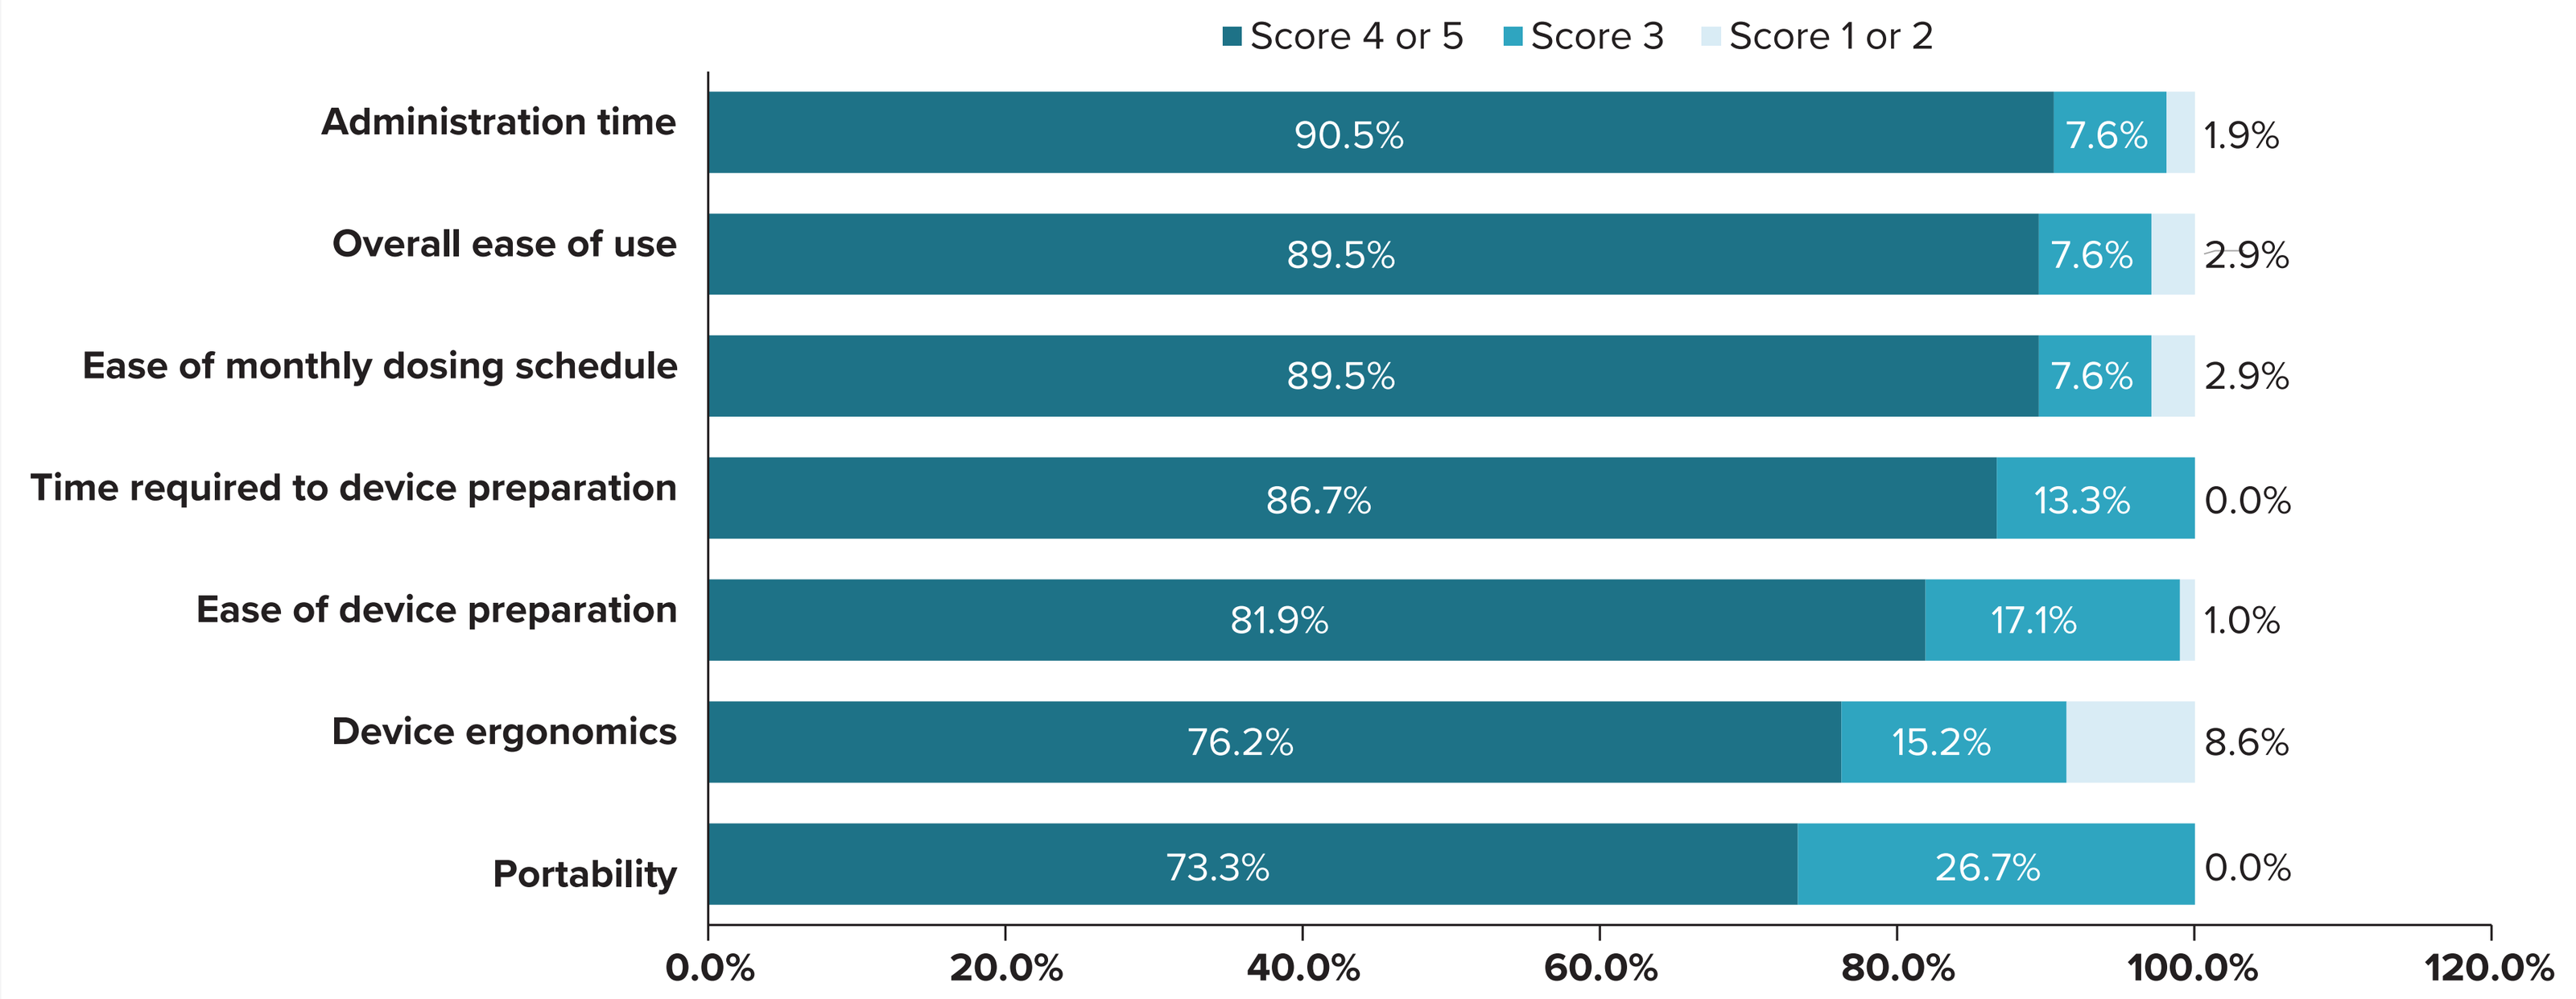
Score 4 or 5
Score 3
Score 1 or 2
Administration time 90.5% 7.6% 1.9%
Overall ease of use 89.5% 7.6% 2.9%
Ease of monthly dosing schedule 89.5% 7.6% 2.9%
Time required to device preparation 86.7% 13.3% 0.0%
Ease of device preparation 81.9% 17.1% 1.0%
Device ergonomics 76.2% 15.2% 8.6%
Portability 73.3% 26.7% 0.0%
0.0% 20.0% 40.0% 60.0% 80.0% 100.0% 120.0%

*Assessed using a 5-point Likert scale (1=strongly disagree to 5=strongly agree)

DISCUSSION & LIMITATIONS

- Results from this study provide evidence of the real-world satisfaction and experience with the use of Sensoready pen for OMB administration in patients with MS
- The limitations of this analysis include:
 - The cross-sectional nature of the study
 - Initiation of OMB in patients may be influenced by insurance and availability (among others) and may not fully represent intended target OMB patients. Hence, data should be cautiously interpreted to understand initial landscape of DMT use within the evaluated time period
 - All questionnaires used are subject to recall bias

REFERENCES: 1. United States Food and Drug Administration. Ofatumumab (Kesimpta®). (Available at: https://www.accessdata.fda.gov/drugsatfda_docs/label/2020/125326s070lbl.pdf. Accessed May 7, 2023). **2.** Thompson A J, et al. *Lancet Neurol.* 2018;17:162–173.

DISCLOSURES: Amy Perrin Ross has received consulting fee from EMD Serono, BMS, Horizon, Alexion, TG Therapeutics, Novartis, Roche, and Sanofi. Jacqueline Nicholas has received research grants from Biogen, Novartis, PCORI, Genentech, University of Buffalo. She has also received consulting fee from EMD Serono, Genentech, Greenwich Biosciences, Novartis, TG Therapeutics, Sanofi, and speaking honoraria from BMS, EMD Serono, Horizon, TG Therapeutics. Ming-Hui Tai and Stephen Yeung are employees of Novartis Pharmaceuticals Corporation. Nazneen Fatima Shaikh, Helen Chen, Mariana Fernandes, and Kevin Hawkins are employees of Real World Solutions, IQVIA, 2400 Ellis Road Durham, NC 27703, USA.

ACKNOWLEDGEMENTS: This study was funded by Novartis Pharmaceuticals Corporation. The authors would like to thank **Dr. Santosh Tiwari** (Novartis Healthcare Private Limited) for writing support and **Hareesh Cheela** (Novartis Healthcare Private Limited) for designing the poster layout, which was funded by Novartis Pharmaceuticals Corporation.