Poster presented at the CMSC Annual Meeting, Aurora, Colorado, USA, May 31 – June 3, 2023.

DMT27

Effect of Longer-term Ofatumumab Treatment on Disability Worsening and Brain Volume Change

Derrick Robertson[1], Jeffrey A. Cohen[2], Stephen L. Hauser[3], Ronald Zielman[4], Ayan Das Gupta[5], Amin Azmon[6], Jing Xi[7], Gina Mavrikis Cox[8], Heinz Wiendl[9], Ludwig Kappos[10]

[1]Multiple Sclerosis Division, Department of Neurology, University of South Florida, Tampa, FL, USA; [2]Department of Neurology, Mellen MS Center, Neurological Institute, Cleveland Clinic, Cleveland, OH, USA; [3]UCSF Weill Institute for Neurosciences, Department of Neurology, University of California - San Francisco, San Francisco, CA, USA; [4]Novartis Pharma B.V., Amsterdam, The Netherlands; [5]Novartis Healthcare Pvt. Ltd., Hyderabad, India; [6]Novartis Pharma A.G., Basel, Switzerland; [7]China Novartis Institutes for BioMedical Research Co., Ltd, Shanghai, China; [8]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [9]University of Muenster, Muenster, Germany; [10]Research Center for Clinical Neuroimmunology and Neuroscience Basel (RC2NB) and MS Center, Departments of Head, Spine and Neuromedicine, Clinical Research, Biomedicine and Biomedical Engineering, University Hospital and University of Basel, Basel, Switzerland.

CONCLUSIONS

1. **With up to 5 years treatment with ofatumumab, >80% of patients remained free of 6m-CDW**
2. **Fewer disability events including PIRA and RAW, were experienced by patients who received ofatumumab continuously versus those initially randomized to teriflunomide**
3. **PBVC remained low (<1.5% loss) over 5 years in patients continuously on ofatumumab**
 - **Early ofatumumab initiation resulted in statistically significantly lower levels of PBVC versus later switch from teriflunomide to ofatumumab at year 5**
4. **Overall, these data support the early use of ofatumumab in patients with RMS**

Copyright © 2023 Novartis Pharma AG. All rights reserved.

Scan to download a copy of this poster

Copies of this poster and its content, obtained through this QR code, are for personal use only and may not be reproduced without written permission from the authors

INTRODUCTION

- Ofatumumab is the first fully human anti-CD20 monoclonal antibody with a 20 mg s.c. monthly dosing regimen in 0.4ml, approved for the treatment of RMS in adults in the US[1] and other countries*
- Data from the ASCLEPIOS I/II trials and the open-label ALITHIOS extension with up to 4 years treatment with ofatumumab showed:
 - Significantly delayed disability accrual compared with teriflunomide[2]
 - Sustained efficacy by reducing relapses and magnetic resonance imaging (MRI) lesions[3,4]
 - 93% of patients receiving continuous ofatumumab achieved no evidence of disease activity (NEDA-3)[†5]
- Relapse-associated disability worsening (RAW) and progression independent of relapse activity (PIRA)[6,7] both contribute to confirmed disability worsening (CDW) in patients with relapsing multiple sclerosis (RMS)[8]

OBJECTIVE

- To assess longer-term effects (up to 5 years of treatment) of continuous ofatumumab treatment on disability outcomes (CDW, PIRA, RAW) and brain volume change compared with delayed ofatumumab treatment (after switching from teriflunomide)

KEY ASSESSMENTS

- 6-month confirmed disability worsening (6m-CDW).
 - 6-month confirmed relapse associated worsening (6m-RAW)
 - 6-month confirmed progression independent of relapse activity (6m-PIRA)
 - 6-month confirmed and sustained progression independent of relapse activity (6m-sPIRA)
- Brain Volume Change (BVC) MRI outcomes
 - Annual rate of brain volume change (ABVC) over the study duration (Core & Extension)
 - Percentage brain volume change (PBVC) at each year
- Refer to Table 1 for Patient demographics and clincal characteristics

RESULTS

- Of 1882 patients randomized in the ASCLEPIOS I/II trials, 1367 (72.6%) patients enrolled into the ALITHIOS open-label extension study and received ofatumumab for up to 5 years cumulatively (**Figure 1**)
- Of these, 1145/1367 (83.8%) patients were still receiving ofatumumab treatment at the time of data cut-off (25-Sep-2022)
- Overall, fewer CDW, PIRA and RAW events were observed in patients receiving continuous ofatumumab versus those initially randomized to teriflunomide and switched to ofatumumab (**Figures 2 & 3**); similar patterns were identified for the 3m-CDW
- RMS patients treated with disease modifying therapies accumulate disability primarily via PIRA (**Figure 3**)
- Ofatumumab's high anti-inflammatory efficacy is evident with flattening of the 6m-RAW curve in the TER-OMB switch group in the extension period (**Figure 3**)

Figure 1. Participant disposition

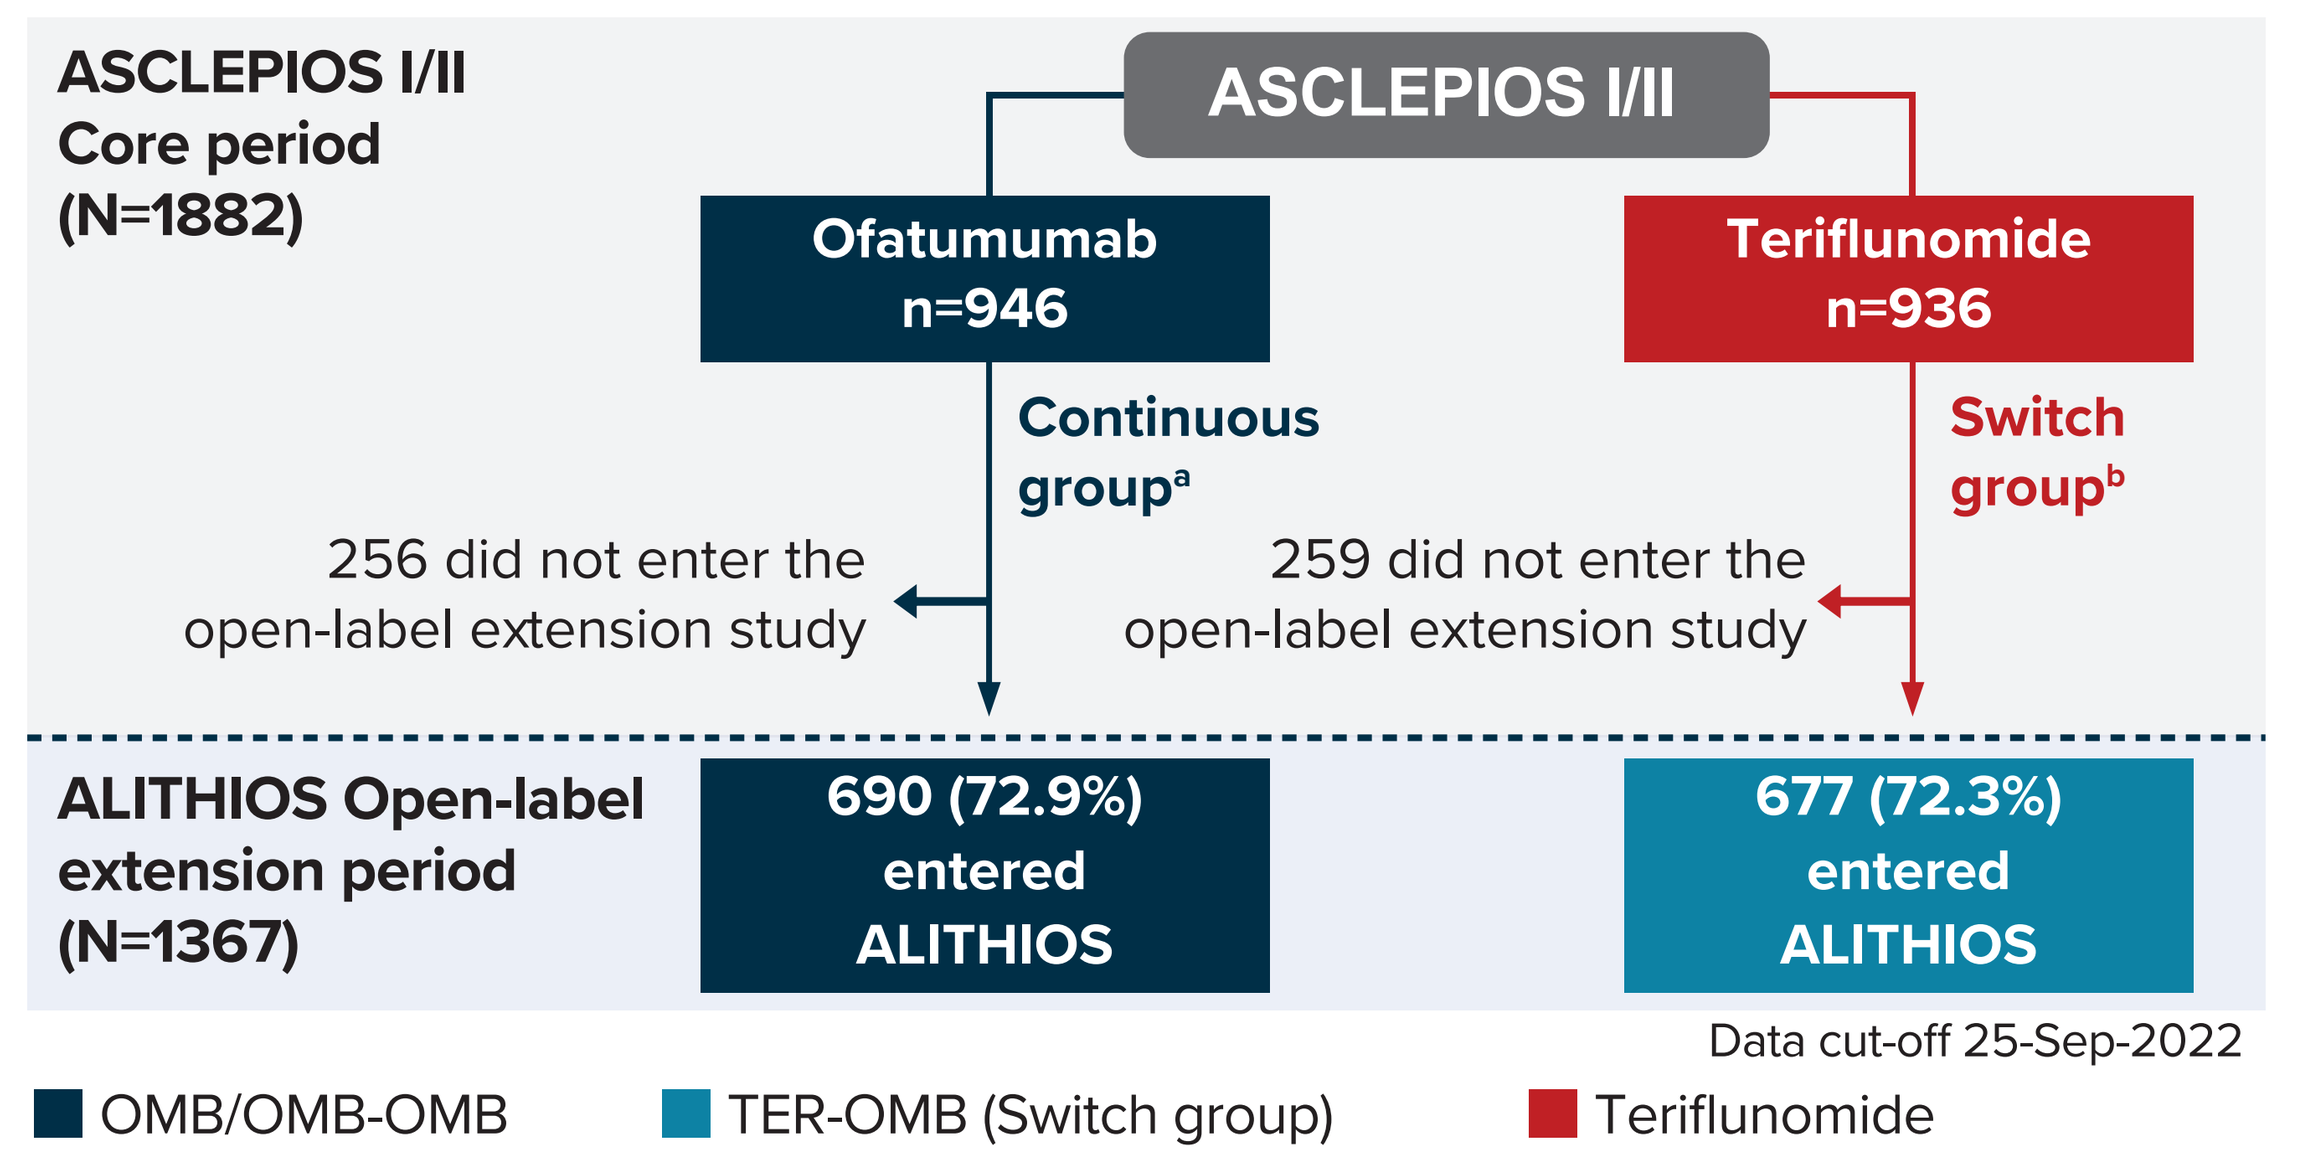

ASCLEPIOS I/II Core period (N=1882)
ASCLEPIOS I/II
Ofatumumab n=946
Teriflunomide n=936
Continuous group[a]
Switch group[b]
256 did not enter the open-label extension study
259 did not enter the open-label extension study
ALITHIOS Open-label extension period (N=1367)
690 (72.9%) entered ALITHIOS
677 (72.3%) entered ALITHIOS
Data cut-off 25-Sep-2022
OMB/OMB-OMB
TER-OMB (Switch group)
Teriflunomide

All percentages are calculated based on the number of patients in full analysis set in the corresponding column. Dotted line represents the first dose of ofatumumab in extension period.
Core period is period before the dotted line.

Only patients from the ASCLEPIOS I/II studies are included in the analyses presented here; [a]randomized to ofatumumab in the core; [b]switch group refers to the patients who were randomized to teriflunomide in the core and switched to ofatumumab during the extension period.

The 259 patients who took teriflunomide in the core period but did not enter the extension study are included in the switch group.

Note: Patients in ASCLEPIOS I/II had a flexible trial duration with individual patient's exposure of up to 2.3 years; Hauser et al. N Engl J Med. 2020; 383:546-557, supplementary figure S2

Table 1. Baseline demographics and clinical characteristics

Demographics and clinical characteristics[a]	Ofatumumab continuous (N=946)		Switch from teriflunomide to ofatumumab (N=936)	
	Baseline of core study (N=946)	Baseline of extension study (N=690)	Baseline of core study (N=936)	Baseline of extension study[d] (N=677)
Age, years	38.4±9.04	38.1±8.69	38.0±9.22	40.1±9.21
Female, n (%)	637 (67.3)	483 (70.0)	636 (67.9)	456 (67.4)
BMI, kg/m²	25.86±6.22	25.73±6.00	25.93±6.02	25.61±5.85
Treatment-naive patients[b], n (%)	386 (40.8)	Not applicable[c]	363 (38.8)	Not applicable[c]
EDSS score at baseline	2.93±1.35	2.80±1.49	2.90±1.36	2.81±1.46
Number of relapses in the last 12 months prior to screening, n (%)	1.2±0.69	0.1±0.35	1.3±0.71	0.2±0.49
Number of Gd+ T1 lesions	1.7±4.51	0.0±0.21	1.3±3.43	0.8±2.37
Total volume of T2 lesions, cm³	13.72±13.79	Not available	12.55±13.81	Not available

[a]Values are represented as mean±SD unless specified otherwise; [b]Treatment-naive patients are those who have not received a prior multiple sclerosis disease modifying therapy; [c]not applicable since all patients have been pre-treated with either ofatumumab or teriflunomide in the core period; [d]the baseline from the extension study in the ofatumumab continuous and the ofatumumab switch from teriflunomide groups reflect the relative treatment effects during the double-blind treatment phase in the ASCLEPIOS I/II studies.

BMI, body mass index; EDSS, Expanded Disability Status Scale; Gd+, gadolinium enhancing.

Figure 2. 6m-CDW

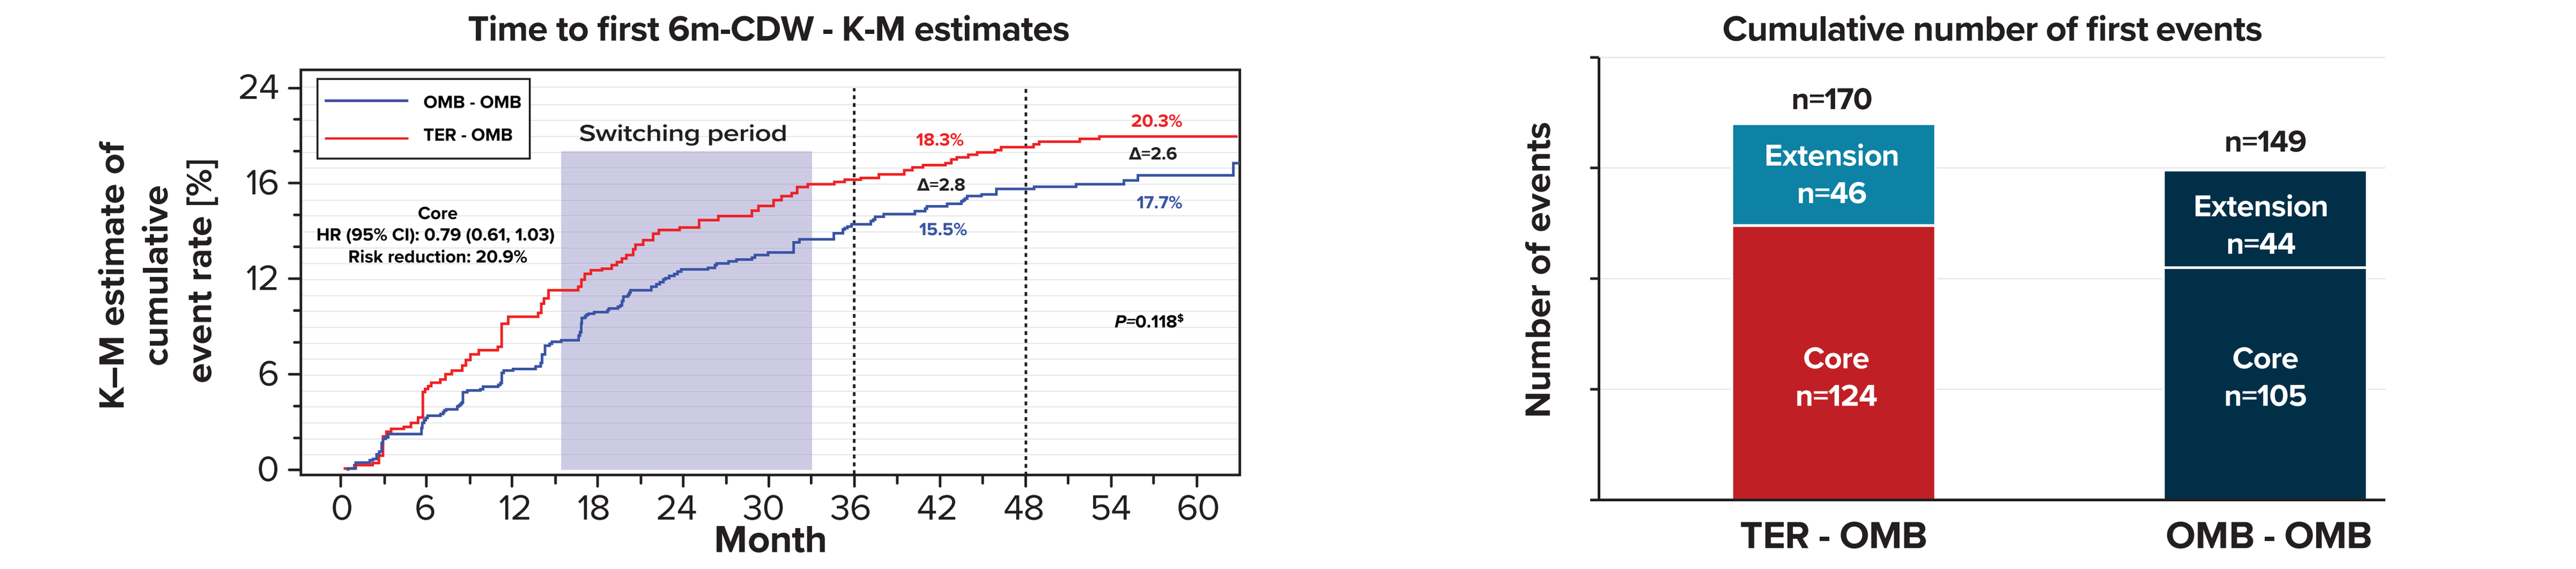

Time to first 6m-CDW - K-M estimates
OMB - OMB
TER - OMB
Switching period
Core
HR (95% CI): 0.79 (0.61, 1.03)
Risk reduction: 20.9%
18.3%
Δ=2.8
15.5%
20.3%
Δ=2.6
17.7%
P=0.118$
K–M estimate of cumulative event rate [%]
Month
Cumulative number of first events
Number of events
n=170
Extension n=46
Core n=124
n=149
Extension n=44
Core n=105
TER - OMB
OMB - OMB

$p-value from log-rank test on difference between the cumulative K-M curves of the two treatment groups

6m-CDW, 6-month CDW; CDW, confirmed disability worsening; K-M, Kaplan-Meier; OMB-OMB, patients randomized to ofatumumab in the Core period and continuing ofatumumab in ALITHIOS; TER-OMB, patients randomized to teriflunomide during the Core and switched to ofatumumab during ALITHIOS.

- Patients receiving continuous ofatumumab had a lower number of cumulative events versus the switch group

Figure 3. Kaplan-Meier estimates of 6m-CDW, 6m-PIRA, 6m-sPIRA, and 6m-RAW up to 5 years

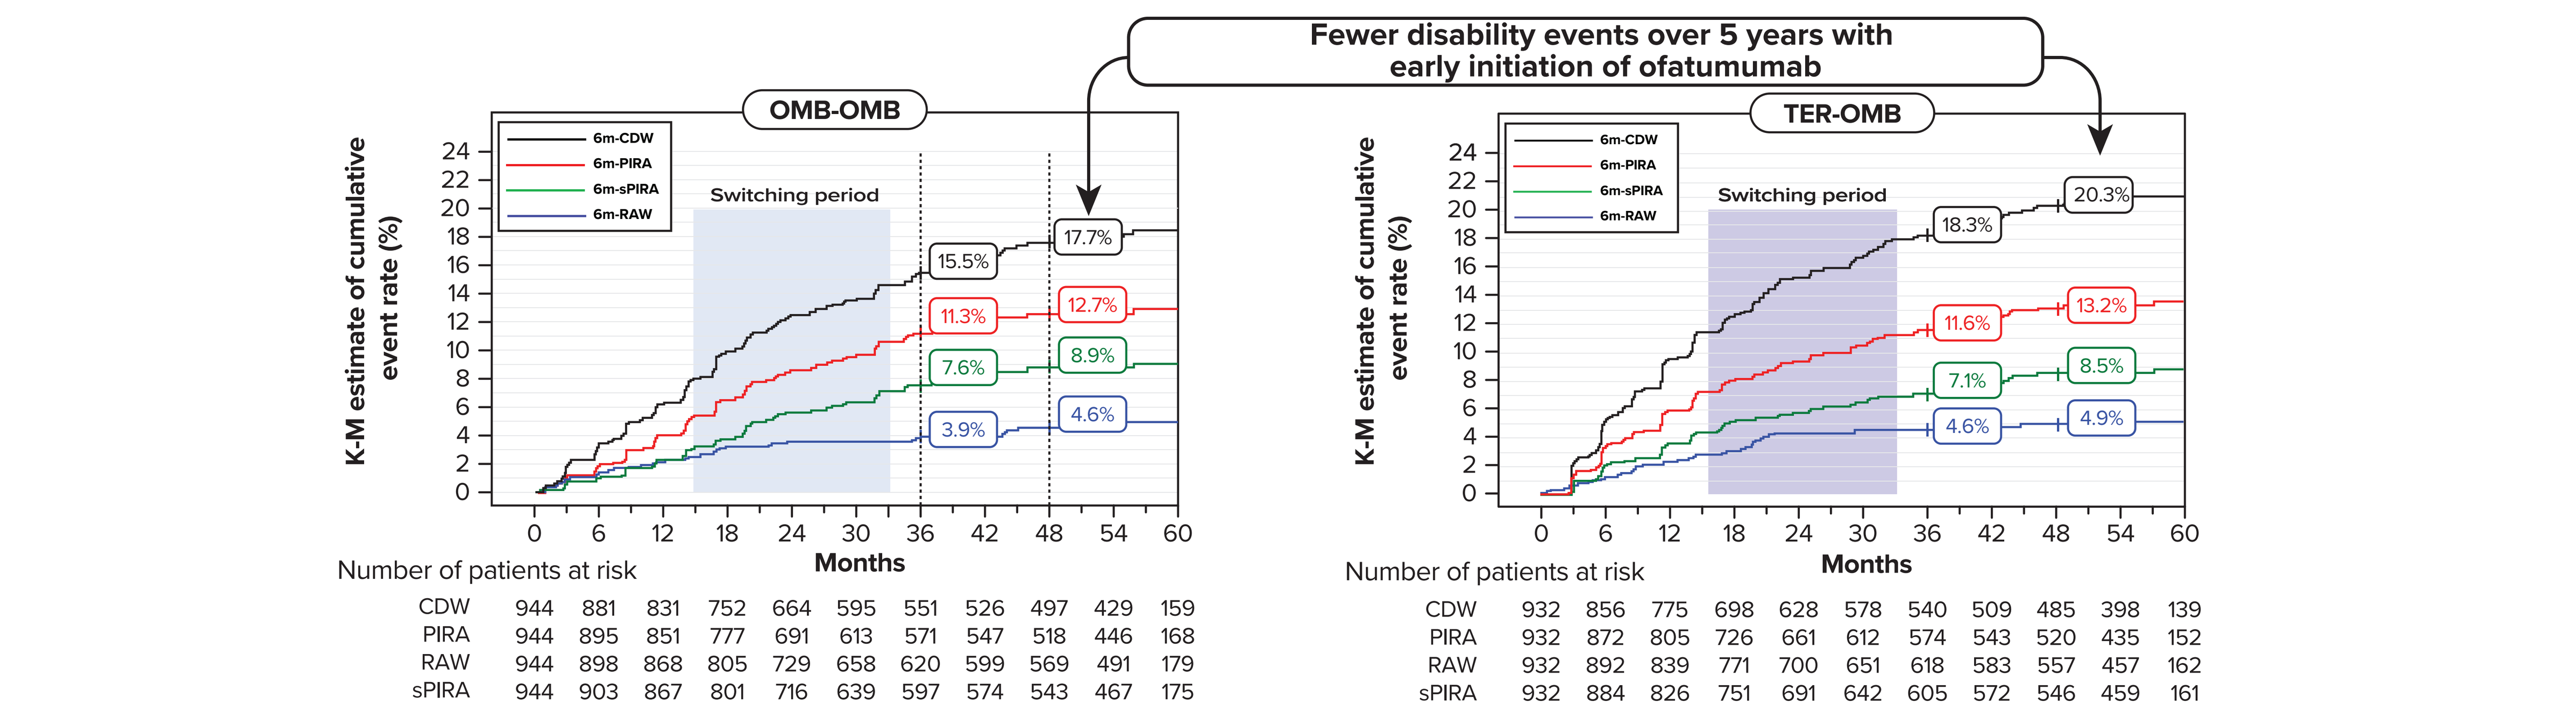

Fewer disability events over 5 years with early initiation of ofatumumab
OMB-OMB
6m-CDW
6m-PIRA
6m-sPIRA
6m-RAW
Switching period
15.5%
17.7%
11.3%
12.7%
7.6%
8.9%
3.9%
4.6%
K-M estimate of cumulative event rate (%)
Months
Number of patients at risk
CDW 944 881 831 752 664 595 551 526 497 429 159
PIRA 944 895 851 777 691 613 571 547 518 446 168
RAW 944 898 868 805 729 658 620 599 569 491 179
sPIRA 944 903 867 801 716 639 597 574 543 467 175
TER-OMB
18.3%
20.3%
11.6%
13.2%
7.1%
8.5%
4.6%
4.9%
CDW 932 856 775 698 628 578 540 509 485 398 139
PIRA 932 872 805 726 661 612 574 543 520 435 152
RAW 932 892 839 771 700 651 618 583 557 457 162
sPIRA 932 884 826 751 691 642 605 572 546 459 161

6m-CDW, 6-month CDW; 6m-PIRA, 6-month confirmed PIRA; 6m-RAW, 6-month confirmed RAW; 6m-sPIRA, 6-month confirmed sustained PIRA; CDW, confirmed disability worsening; K-M, Kaplan-Meier; OMB-OMB, patients randomized to ofatumumab in the Core period and continuing ofatumumab in ALITHIOS; PIRA, progression independent of relapse activity; RAW, relapse associated worsening; RMS, relapsing multiple sclerosis; sPIRA, sustained progression independent of relapse activity; TER-OMB, patients randomized to teriflunomide during the Core and switched to ofatumumab during ALITHIOS.

Figure 4. PBVC up to 5 years

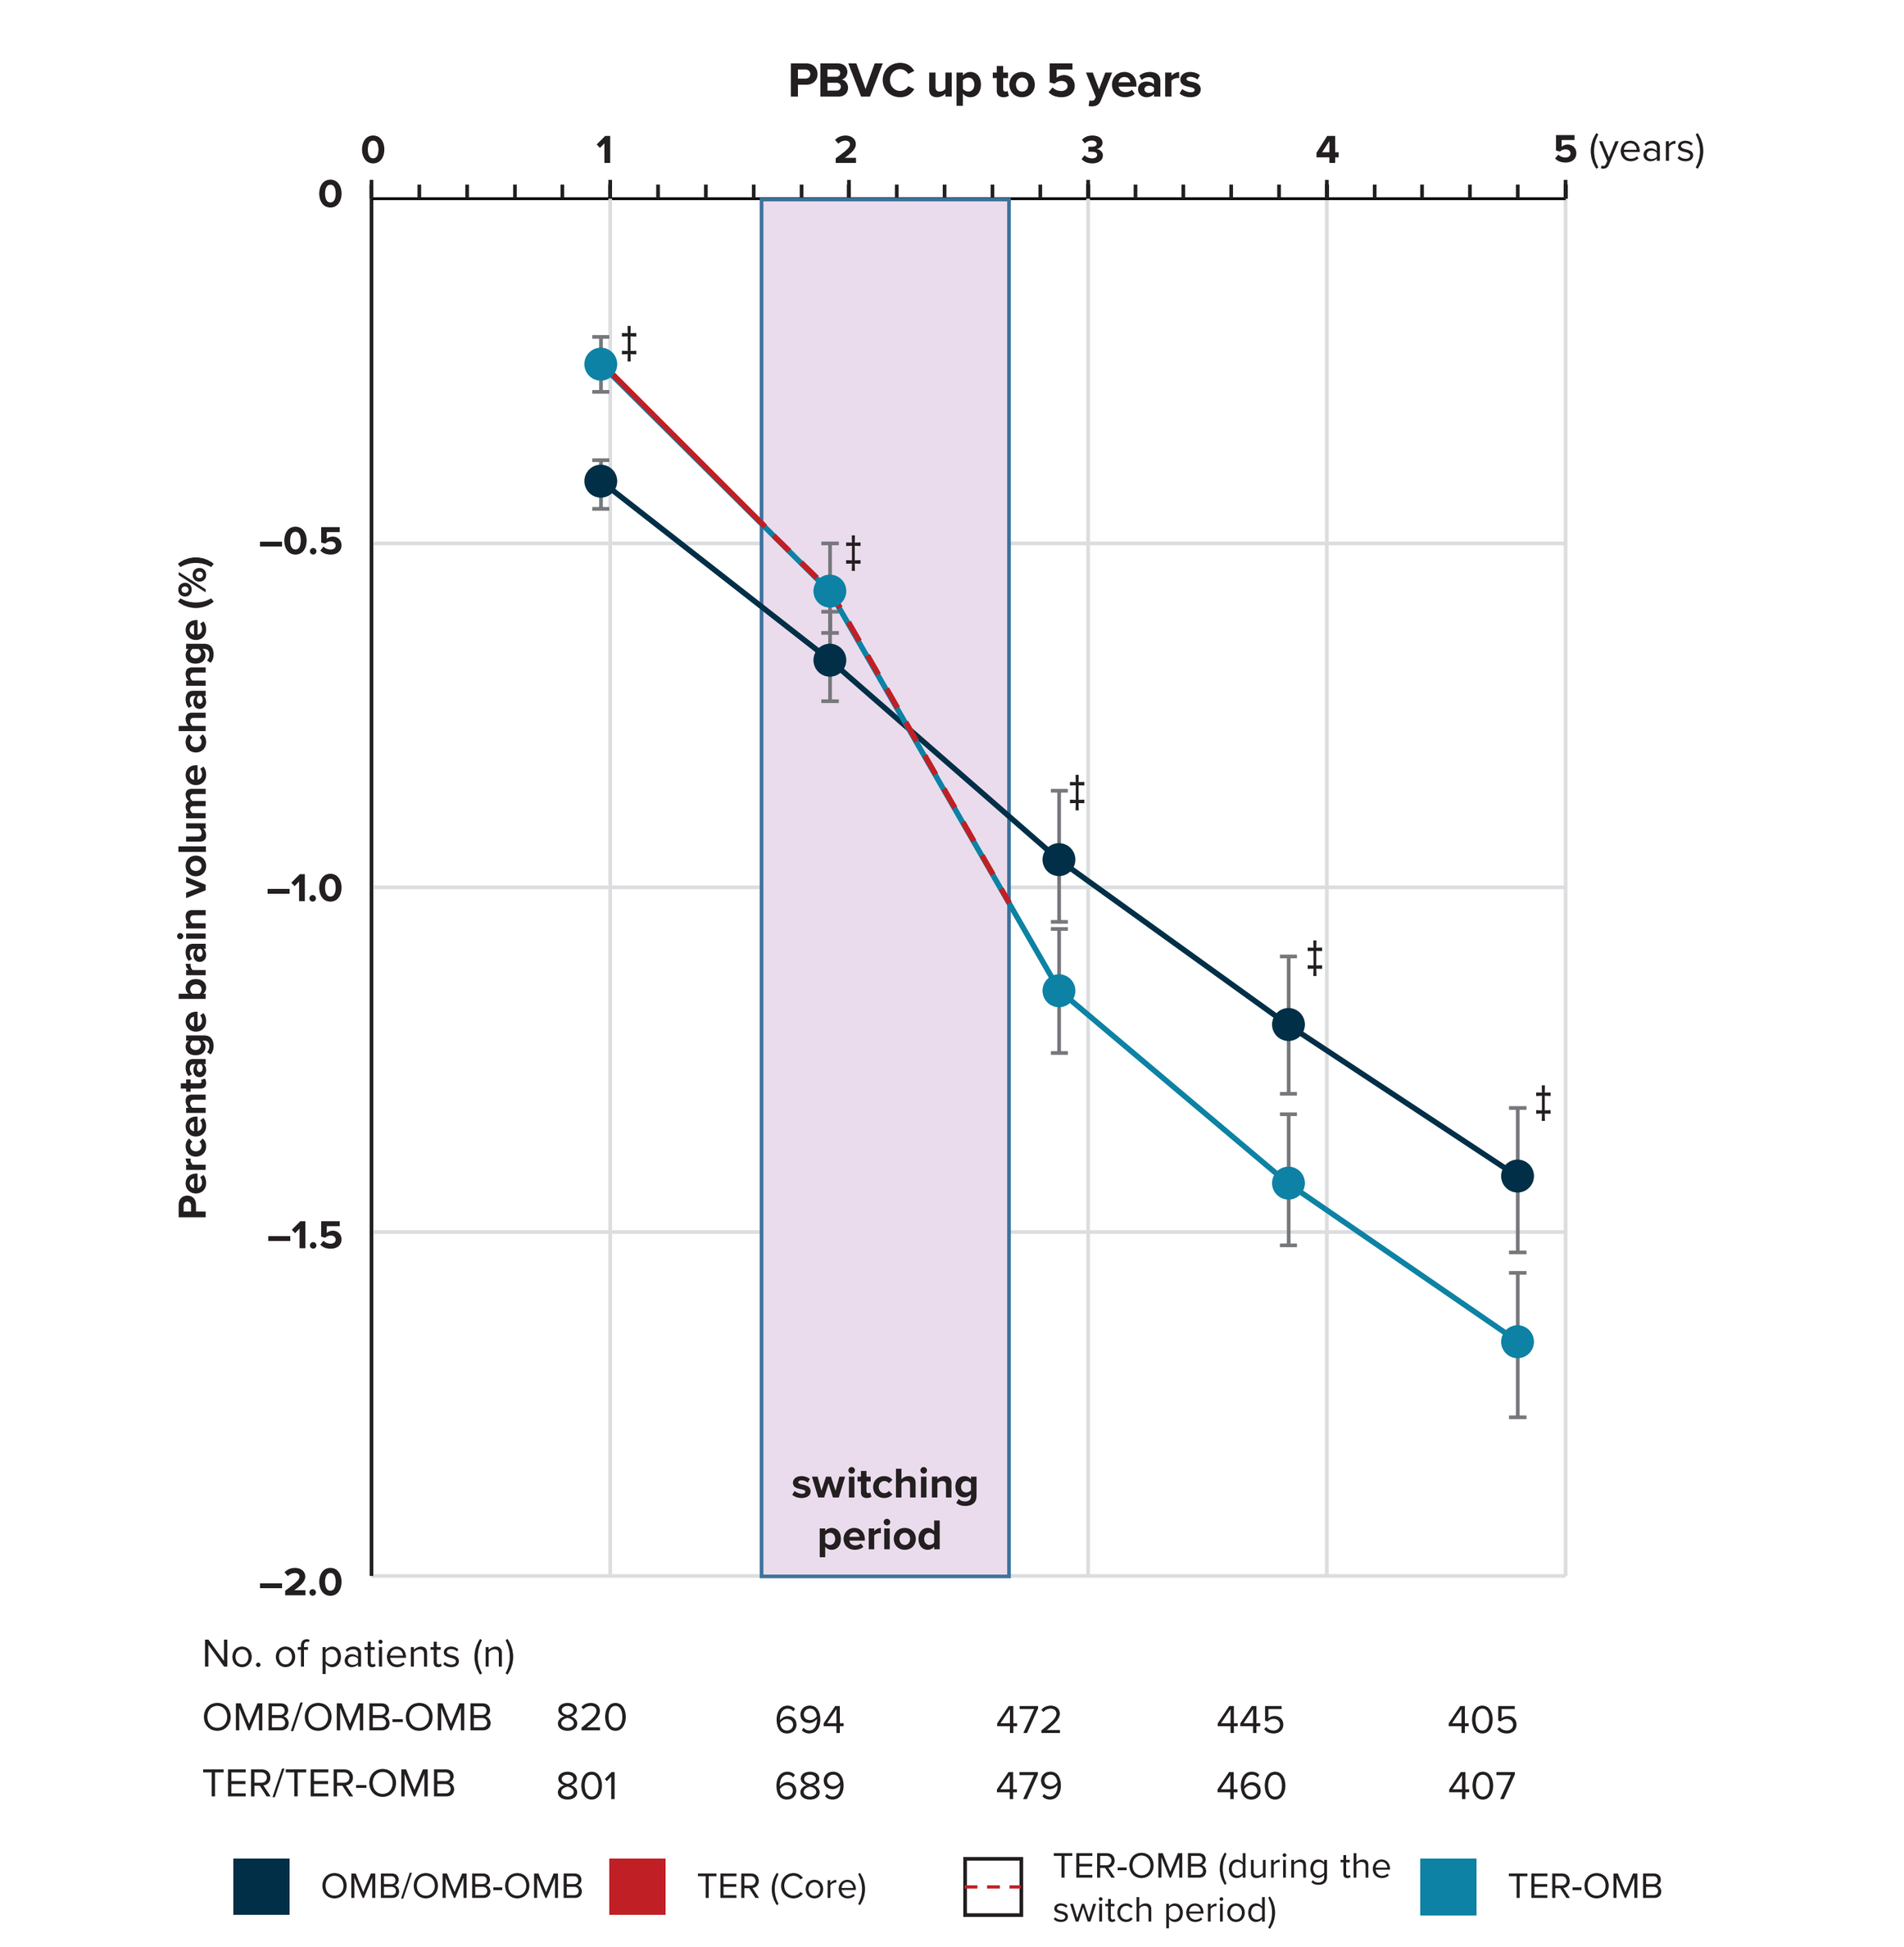

PBVC up to 5 years
(years)
Percentage brain volume change (%)
switching period
No. of patients (n)
OMB/OMB-OMB 820 694 472 445 405
TER/TER-OMB 801 689 479 460 407
OMB/OMB-OMB
TER (Core)
TER-OMB (during the switch period)
TER-OMB

[‡]Statistically significant between-group comparison at each year for PBVC

OMB/OMB-OMB, patients treated with ofatumumab during the Core period (OMB) and continued with ofatumumab (OMB-OMB) during the Extension period; PBVC, percentage brain volume change; TER-OMB, patients randomised to teriflunomide during the Core period and switched to ofatumumab during the Extension period (ALITHIOS); n, total number of patients included in the analysis.

Note, all estimates are obtained from a mixed model for repeated measured with treatment and visit window as interacting factors, region as a factor and with age, baseline number of Gd-enhanced lesions, baseline T2 volume, and baseline normalized brain volume as continuous covariates.

- PBVC remained low (<1.5% loss) up to 5 years with continuous ofatumumab treatment[#] (**Figure 4**)
- Overall, ABVC remained low in patients receiving continuous ofatumumab with –0.27% BVC per year during Extension

Table 2. Annual Brain Volume Change

Group	Core (%)	P-value[ππ]	Extension (%)	P-value[ππ]
OMB/OMB-OMB	–0.34	0.115	–0.27	0.666
TER-OMB	–0.42		–0.28	

[ππ]Between-group comparison for ABVC

- A steeper decline was observed with TER-OMB vs OMB-OMB during Core while the annual rate became similar to OMB-OMB during Extension indicating a slowing of BVC following switch to ofatumumab

[#]The initial difference between groups in brain volume in the first year and during the switch period may be due to the strong effect of ofatumumab on lesions and lesion-related edema (pseudo-atrophy).

Note, all ABVC estimates are obtained from a random coefficient model with treatment, time period (core vs. extension) and region as factors, time, baseline number of Gd-enhanced lesions, baseline T2 volume, and baseline normalized brain volume as continuous covariates, the three-way interaction (treatment group x time period x time) and all its associated two-way interactions.

REFERENCES: 1. KESIMPTA® (ofatumumab) Prescribing information. https://www.novartis.us/sites/www.novartis.us/files/kesimpta.pdf (accessed March 22, 2023); **2.** Hauser et al. *N Engl J Med.* 2020; 383:546-557; **3.** Kappos et al. ERP161 Poster Presented at EAN 2022; **4.** Hauser SL, et al. ePoster #004 presented at AAN 2022; **5.** Kuhle et al. P1198 poster presented at ECTRIMS 2022; **6.** Kappos et al. *Mult Scler* 2018; 24: 963–739; **7.** Kappos et al. Clinical Trials. *JAMA Neurol* 2020; 77: 1132–40; **8.** Lublin F, et al. *Brain* 2022;145,9:3147-3161.

*Kesimpta® (ofatumumab) has now been approved in many countries including US, Canada, Switzerland, Singapore, Australia, Japan and the EU

†NEDA-3 is defined as no 6-month confirmed disability worsening, no confirmed MS relapse, no new or enlarging T2 lesions compared to baseline and no T1 gadolinium-enhancing lesions.

DISCLOSURES: The study was funded by Novartis Pharma AG, Basel, Switzerland. **Ronald Zielman, Ayan Das Gupta, Amin Azmon, Jing Xi, Gina Mavrikis Cox** are employees of Novartis.
Derrick Robertson has received fees for consulting, contracted research and speaker's bureau from Biogen, Celgene, EMD Serono, Genentech, Sanofi Genzyme, Janssen, TG therapeutics; consulting fees and speakers bureau for Bristol Myers Squibb, Horizon, and Alexion; consulting fees and contracted research for Novartis; consulting fees for Greenwich biosciences; contracted research for GW Pharmaceuticals, PCORI, Atara Biotherapeutics and CorEvitas. **Jeffrey A. Cohen** received personal compensation for consulting for Biogen, Bristol-Myers Squibb, Convelo, Genentech, Janssen, NervGen, Novartis, and PSI; speaking for H3 Communications; and serving as an Editor of Multiple Sclerosis Journal. **Stephen Hauser** serves on the board of trustees for Neurona and serves on scientific advisory boards for Accure, Alector and Annexon, received travel reimbursement and writing assistance for CD-20 related meeting and presentations from Roche and Novartis. **Heinz Wiendl** is acting as a paid consultant for AbbVie, Actelion, Biogen, IGES, Johnson & Johnson, Novartis, Roche, Sanofi-Aventis, and the Swiss Multiple Sclerosis Society. His research is funded by the German Ministry for Education and Research (BMBF), Deutsche Forschungsgemeinschaft (DFG), Else Kröner Fresenius Foundation, Fresenius Foundation, the European Union, Hertie Foundation, NRW Ministry of Education and Research, Interdisciplinary Center for Clinical Studies (IZKF) Muenster and RE Children's Foundation, Biogen, GlaxoSmithKline GmbH, Roche Pharma AG, and Sanofi-Genzyme. **Ludwig Kappos** has received consultancy fees from Actelion, Bayer HealthCare, Biogen, BMS, Genzyme, Janssen, Japan Tobacco, Merck, Novartis, Roche, Sanofi, Santhera, and TG therapeutics; contracted research from Bayer HealthCare, Biogen, European Union, InnoSwiss, Merck, Novartis, Roche, Swiss MS Society, and Swiss National Research Foundation; speaker fees from Bayer HealthCare, Biogen, Merck, Novartis, Roche, and Sanofi; serves on the steering committee for Actelion, Bayer HealthCare, Biogen, BMS, Genzyme, Janssen, Japan Tobacco, Merck, Novartis, Roche, Sanofi, Santhera, TG Therapeutics; Support of educational activities from Allergan, Bayer HealthCare, Biogen, CSL Behring, Desitin, Genzyme, Merck, Novartis, Roche, Pfizer, Sanofi, Shire, and Teva; License fees for Neurostatus products.

ACKNOWLEDGEMENTS: The authors acknowledge the following Novartis employees: Writing support was provided by **Erik O'Hanlon** a and **Paul Coyle** (employees of Novartis Ireland Ltd., Dublin Ireland), and **Saketh Vellanki** for creative design assistance. The final responsibility for the content lies with the authors.