Efficacy of Ofatumumab in Treatment-Naïve, First-Switch, and Late-Switch Patients: Insights From the ALITHIOS Open-Label Extension Study

LB8385

Jeffrey A. Cohen[1], Ralf Gold[2], Jerome de Seze[3], Derrick Robertson[4], Heinz Wiendl[5], Sibyl Wray[6], Francesco Sacca[7], Amin Azmon[8], Miriam King[8], Simone Fantaccini[8], Ronald Zielman[9], Ludwig Kappos[10]

[1]Department of Neurology, Mellen MS Center, Neurological Institute, Cleveland Clinic, Cleveland, Ohio, USA; [2]Department of Neurology, St Josef-Hospital/Ruhr-University Bochum, Bochum, Germany; [3]University Hospital of Strasbourg, Strasbourg, France; [4]Multiple Sclerosis Division, Department of Neurology, University of South Florida, Tampa, Florida, USA; [5]University of Münster, Münster, Germany; [6]Hope Neurology MS Center, Knoxville, TN, USA; [7]NSRO Department, University "Federico II" of Naples, Naples, Italy; [8]Novartis Pharma A.G., Basel, Switzerland; [9]Novartis Pharma B.V., Amsterdam, The Netherlands; [10]Research Center for Clinical Neuroimmunology and Neuroscience Basel (RC2NB) and MS Center, Departments of Head, Spine and Neuromedicine, Clinical Research, Biomedicine and Biomedical Engineering, University Hospital and University of Basel, Basel, Switzerland.

Background

- Early initiation of high-efficacy therapies for relapsing multiple sclerosis (RMS) has been shown to improve longer-term outcomes versus initiation of or escalation from low-efficacy therapies[1-3]
- Ofatumumab, a fully human anti-CD20 monoclonal antibody, reduced annualized relapse rate (ARR), magnetic resonance imaging (MRI) lesion activity, and delayed disability worsening versus teriflunomide in RMS patients who were treatment-naïve (TN) or previously treated (PT) with disease-modifying therapies (DMTs) in Phase 3 ASCLEPIOS I and II trials[4,5]
- Ofatumumab was well tolerated for up to 4 years of treatment with no new safety risks identified[5,6]
- All ASCLEPIOS patients entering the ALITHIOS extension study were switched to open-label ofatumumab, allowing further insights into ofatumumab efficacy in TN or PT patients and after first and late DMT switch

Objective

- To compare efficacy (clinical and MRI) and safety outcomes in patients initiating ofatumumab early versus switching to ofatumumab after one or multiple previous DMTs in the ASCLEPIOS I and II and ALITHIOS studies

Methods

Study design and patient population

- This analysis included cumulative data from patients randomized to ofatumumab or teriflunomide in the ASCLEPIOS I and II trials (core study) and continued on ofatumumab in the ALITHIOS open-label extension study
- Of 1882 patients randomized in the ASCLEPIOS I and II trials, 1367 (72.6%) patients enrolled into the ALITHIOS open-label extension study and received ofatumumab for up to 4 years

Figure 1. Patient disposition

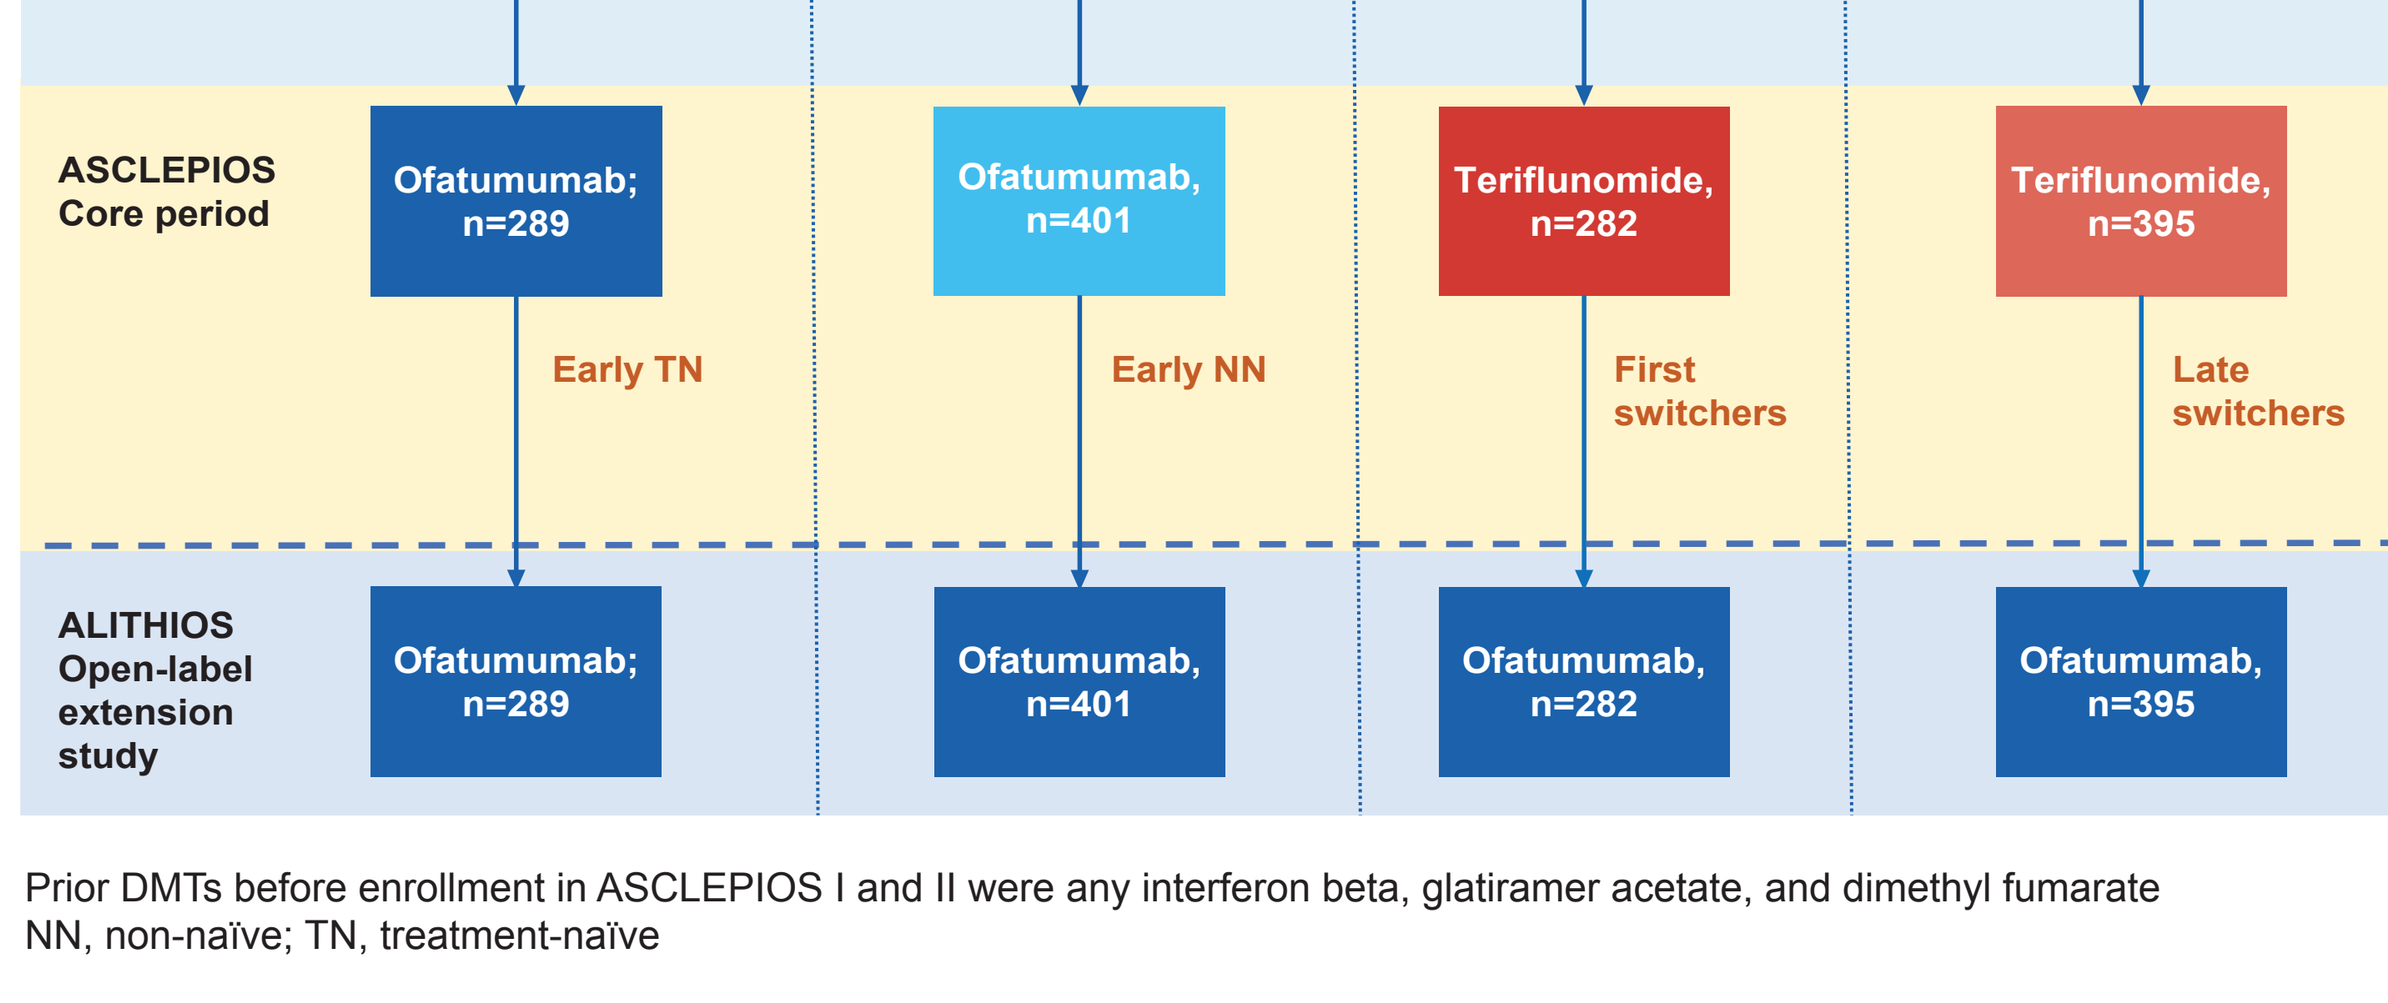

Pre-ASCLEPIOS
ASCLEPIOS Core period
ALITHIOS Open-label extension study
Naive
Previous treatment (s)
Naive
Previous treatment (s)
Ofatumumab; n=289
Ofatumumab, n=401
Teriflunomide, n=282
Teriflunomide, n=395
Early TN
Early NN
First switchers
Late switchers
Ofatumumab; n=289
Ofatumumab, n=401
Ofatumumab, n=282
Ofatumumab, n=395

Prior DMTs before enrollment in ASCLEPIOS I and II were any interferon beta, glatiramer acetate, and dimethyl fumarate
NN, non-naïve; TN, treatment-naïve

- Patients were analyzed in four treatment groups **(Figure 1)**:
 - **Early treatment-naïve (early TN)**: TN patients randomized to ofatumumab in ASCLEPIOS I and II and continued ofatumumab in ALITHIOS
 - **Early non-naïve (early NN)**: Patients who were PT, randomized to ofatumumab in ASCLEPIOS I and II, and continued ofatumumab in ALITHIOS
 - **First switchers**: TN patients randomized to teriflunomide in ASCLEPIOS I and II and switched to ofatumumab in ALITHIOS
 - **Late switchers**: Patients PT with ≥1 DMTs, randomized to teriflunomide in ASCLEPIOS I and II, and switched to ofatumumab in ALITHIOS

Key assessments

- Outcomes in ASCLEPIOS I and II (up to 30 months treatment) were compared with outcomes over 18 months in ALITHIOS (i.e., post-switch to open-label OMB)
- ARR
- 6-month confirmed disability worsening (6mCDW)
 - Confirmed disability worsening is defined as an increase from baseline in Expanded Disability Status Scale (EDSS) score sustained for at least 6 months
- Brain MRI outcomes
 - Mean number of gadolinium-enhancing (Gd+) T1 lesions per scan
 - Number of new or enlarging T2 (neT2) lesions per year
- Safety outcomes were analyzed in long-term safety set which includes all patients who received at least one dose of ofatumumab either in the core studies (ASCLEPIOS I and II, APLIOS and APOLITOS) or in the ALITHIOS, open-label extension study.

Results

Baseline characteristics

- At baseline (before entering ASCLEPIOS), mean age of patients was approximately 39 years across all the treatment groups; majority of patients were women (>65%) **(Table 1)**
- Mean EDSS score at baseline was approximately 2.8 across all the treatment groups

Table 1. Baseline demographics and disease characteristics

Demographics and Clinical Characteristics[a]	Early TN N=289	Early NN N=401	First Switchers		Late switchers	
			Core phase; N=282	Extension phase; N=282	Core phase; N=395	Extension phase; N=395
Age, years	37.8±9.16	38.3±8.35	36.9±9.15	38.9±9.16	39.0±9.18	41.0±9.16
Female, n (%)	207 (71.6)	276 (68.8)	185 (65.6)	185 (65.6)	271 (68.6)	271 (68.6)
BMI, kg/m²	25.82±6.16	25.28 ±5.83	25.64±5.48	25.68±5.33	25.58±6.11	25.70 ±6.17
EDSS score at baseline	2.43 ±1.25	3.19±1.33	2.34±1.21	2.36±1.32	3.08±1.31	3.13±1.47
Time since diagnosis of MS, years	2.26±4.18	7.84±6.01	2.13±4.20	4.01±4.20	7.83±5.97	9.7±5.99
Number of relapses in the last 12 months prior to screening	1.3±0.75	1.2±0.61	1.3±0.65	0.1±0.40	1.2±0.70	0.2±0.54
Number of Gd+ T1 lesions	1.8 ±4.53	1.7±4.78	1.3±2.74	0.6±1.85	1.2±3.92	1.0±2.67
Total volume of T2 lesions, cm³	11.44±13.03	15.27±13.75	9.68±10.27	-	14.47±14.78	-

[a]Values are represented as mean±SD unless specified otherwise.
BMI, body mass index; EDSS, Expanded Disability Status Scale; Gd+, gadolinium-enhancing; MS, multiple sclerosis; SD, standard deviation.

Annualized relapse rate

- ARR in patients who received early ofatumumab treatment (early NN and early TN) was lower compared with patients who were switched from teriflunomide (first switchers and late switchers)
- Continuous use of ofatumumab was associated with a reduction in ARR by 40.6% in the early TN group and by 43.3% in the early NN group, while switch from teriflunomide to ofatumumab resulted in a pronounced reduction in ARR (68.2% in first switchers and 65.4% in late switchers) **(Figure 2)**
- Regardless of the timing of ofatumumab treatment initiation, a significant reduction in ARR was observed across all four treatment groups

6-month confirmed disability worsening

- Continuous use of ofatumumab was associated with less number of CDW events when initiated early. Pronounced reduction of events was observed when patients switched from teriflunomide with the least benefit observed in late switchers **(Table 2)**

Figure 2. Annualized relapse rates by treatment groups

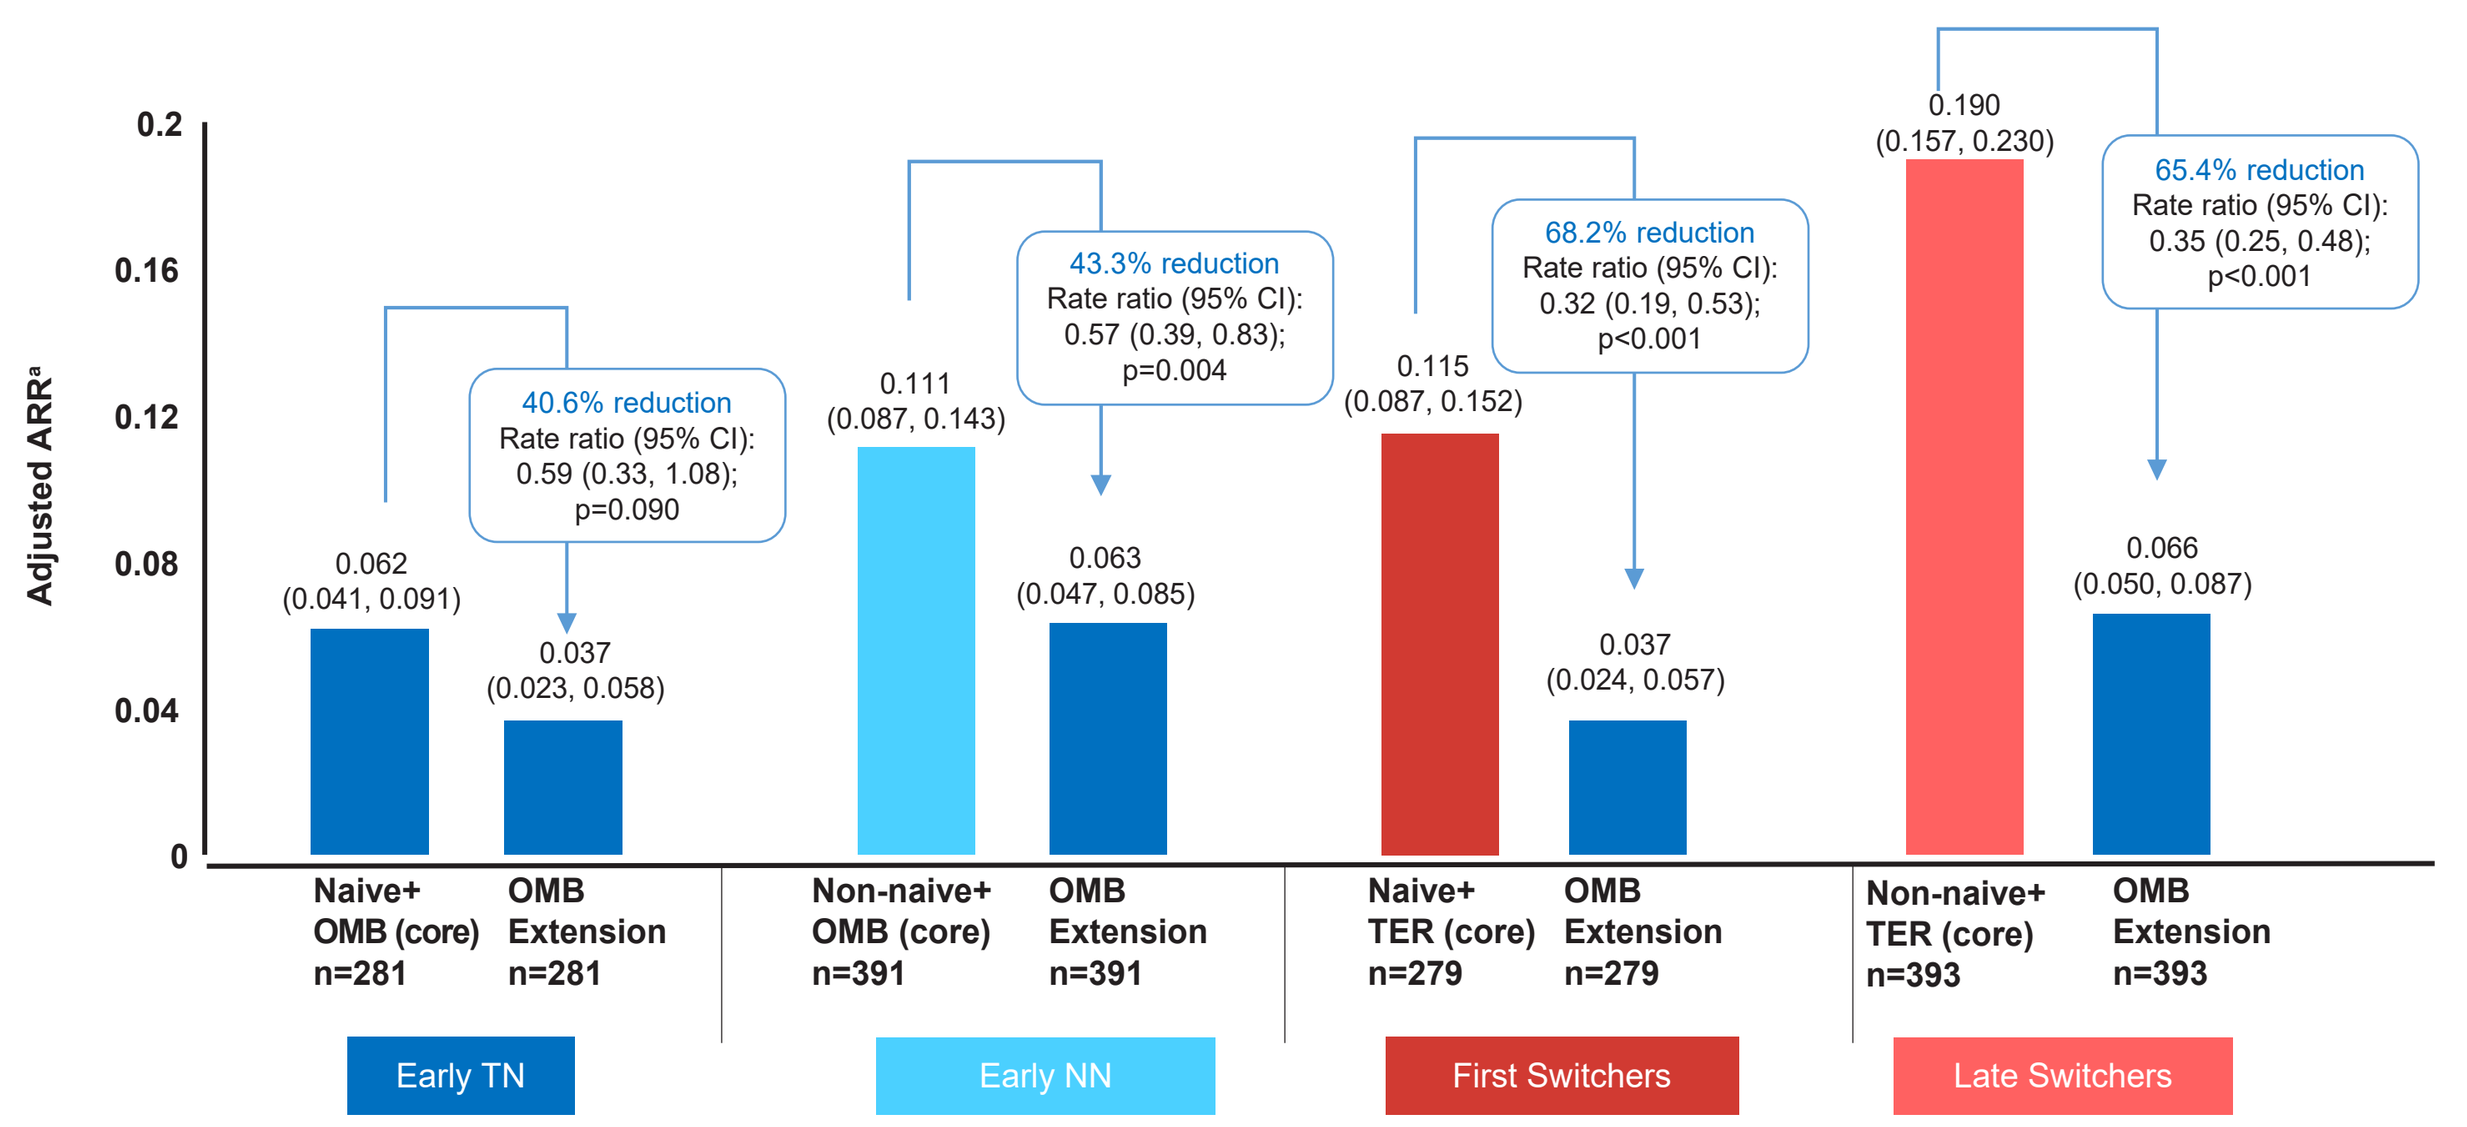

Adjusted ARR[a]
0.2
0.16
0.12
0.08
0.04
0
0.062 (0.041, 0.091)
0.037 (0.023, 0.058)
40.6% reduction Rate ratio (95% CI): 0.59 (0.33, 1.08); p=0.090
0.111 (0.087, 0.143)
0.063 (0.047, 0.085)
43.3% reduction Rate ratio (95% CI): 0.57 (0.39, 0.83); p=0.004
0.115 (0.087, 0.152)
0.037 (0.024, 0.057)
68.2% reduction Rate ratio (95% CI): 0.32 (0.19, 0.53); p<0.001
0.190 (0.157, 0.230)
0.066 (0.050, 0.087)
65.4% reduction Rate ratio (95% CI): 0.35 (0.25, 0.48); p<0.001
Naive+ OMB (core) n=281
OMB Extension n=281
Non-naive+ OMB (core) n=391
OMB Extension n=391
Naive+ TER (core) n=279
OMB Extension n=279
Non-naive+ TER (core) n=393
OMB Extension n=393
Early TN
Early NN
First Switchers
Late Switchers

[a]Obtained from fitting a piecewise negative binomial model for the time period core phase and extension phase with log-link, adjusted for treatment and region as factors, number of relapses in previous year, baseline EDSS, baseline number of Gd-enhancing lesions and the patient's age at baseline as covariates. The natural log of the time-in-study (in years) by period is used as offset to annualize the relapse rate in each period; ARR, annualized relapse rate; CI, confidence interval; early NN, early non-naïve; early TN, early treatment naïve; OMB, ofatumumab; TER, teriflunomide.

Table 2. KM estimates of the proportions with 6mCDW at various timepoints

		KM estimate with event, %	
		Month 18 %, (95% CI)	n/ N (%)
Early TN	Core phase[a]	5.6 (3.5, 9.0)	17/289 (5.9)
	Extension phase[b]	3.4 (1.8, 6.5)	9/287 (3.1)
Early NN	Core phase[a]	7.8 (5.5, 10.9)	31/401 (7.7)
	Extension phase[b]	7.7 (5.4, 10.8)	29/397 (7.3)
First switchers	Core phase[a]	7.1 (4.7, 10.8)	20/282 (7.1)
	Extension phase[b]	4.1 (2.3, 7.4)	11/281 (3.9)
Late switchers	Core phase[a]	9.7 (7.2, 13.1)	42/395 (10.6)
	Extension phase[b]	7.1 (5.0, 10.2)	30/392 (7.7)

[a]ASCLEPIOS up to Month 18; [b]ALITHIOS (post switch to ofatumumab) up to Month 18; early NN, early non-naïve; early TN, early treatment naïve; KM, Kaplan Meier; n=number of patients with events; N=Total number of patients in the group.

MRI lesion activity

Mean number of Gd+ T1 lesions per scan

- Mean number of Gd+ T1 lesions in patients who received early ofatumumab treatment (early NN and early TN) was lower compared with patients who were switched from teriflunomide (first switchers and late switchers)
- Despite a reduced lesion load in early TN and early NN patients, a reduction of 65.1% and 76.2%, respectively, was observed
- An almost complete suppression of Gd+ T1 lesion activity was observed in first and late switchers (99.5% and 95.1%, respectively) **(Figure 3)**

Number of neT2 lesions per year

- Mean number of neT2 lesions in patients who received early ofatumumab treatment (early NN and early TN) was lower compared with patients who were switched from teriflunomide (first switchers and late switchers)
- Despite a reduced lesion load in early TN and early NN patients, a significant reduction of 88.2% and 91%, respectively was observed
- A similar significant reduction in neT2 lesion activity was observed in first and late switchers (86.9% and 80.9%, respectively) **(Figure 4)**

Figure 3. Gd+ T1 lesions by treatment groups

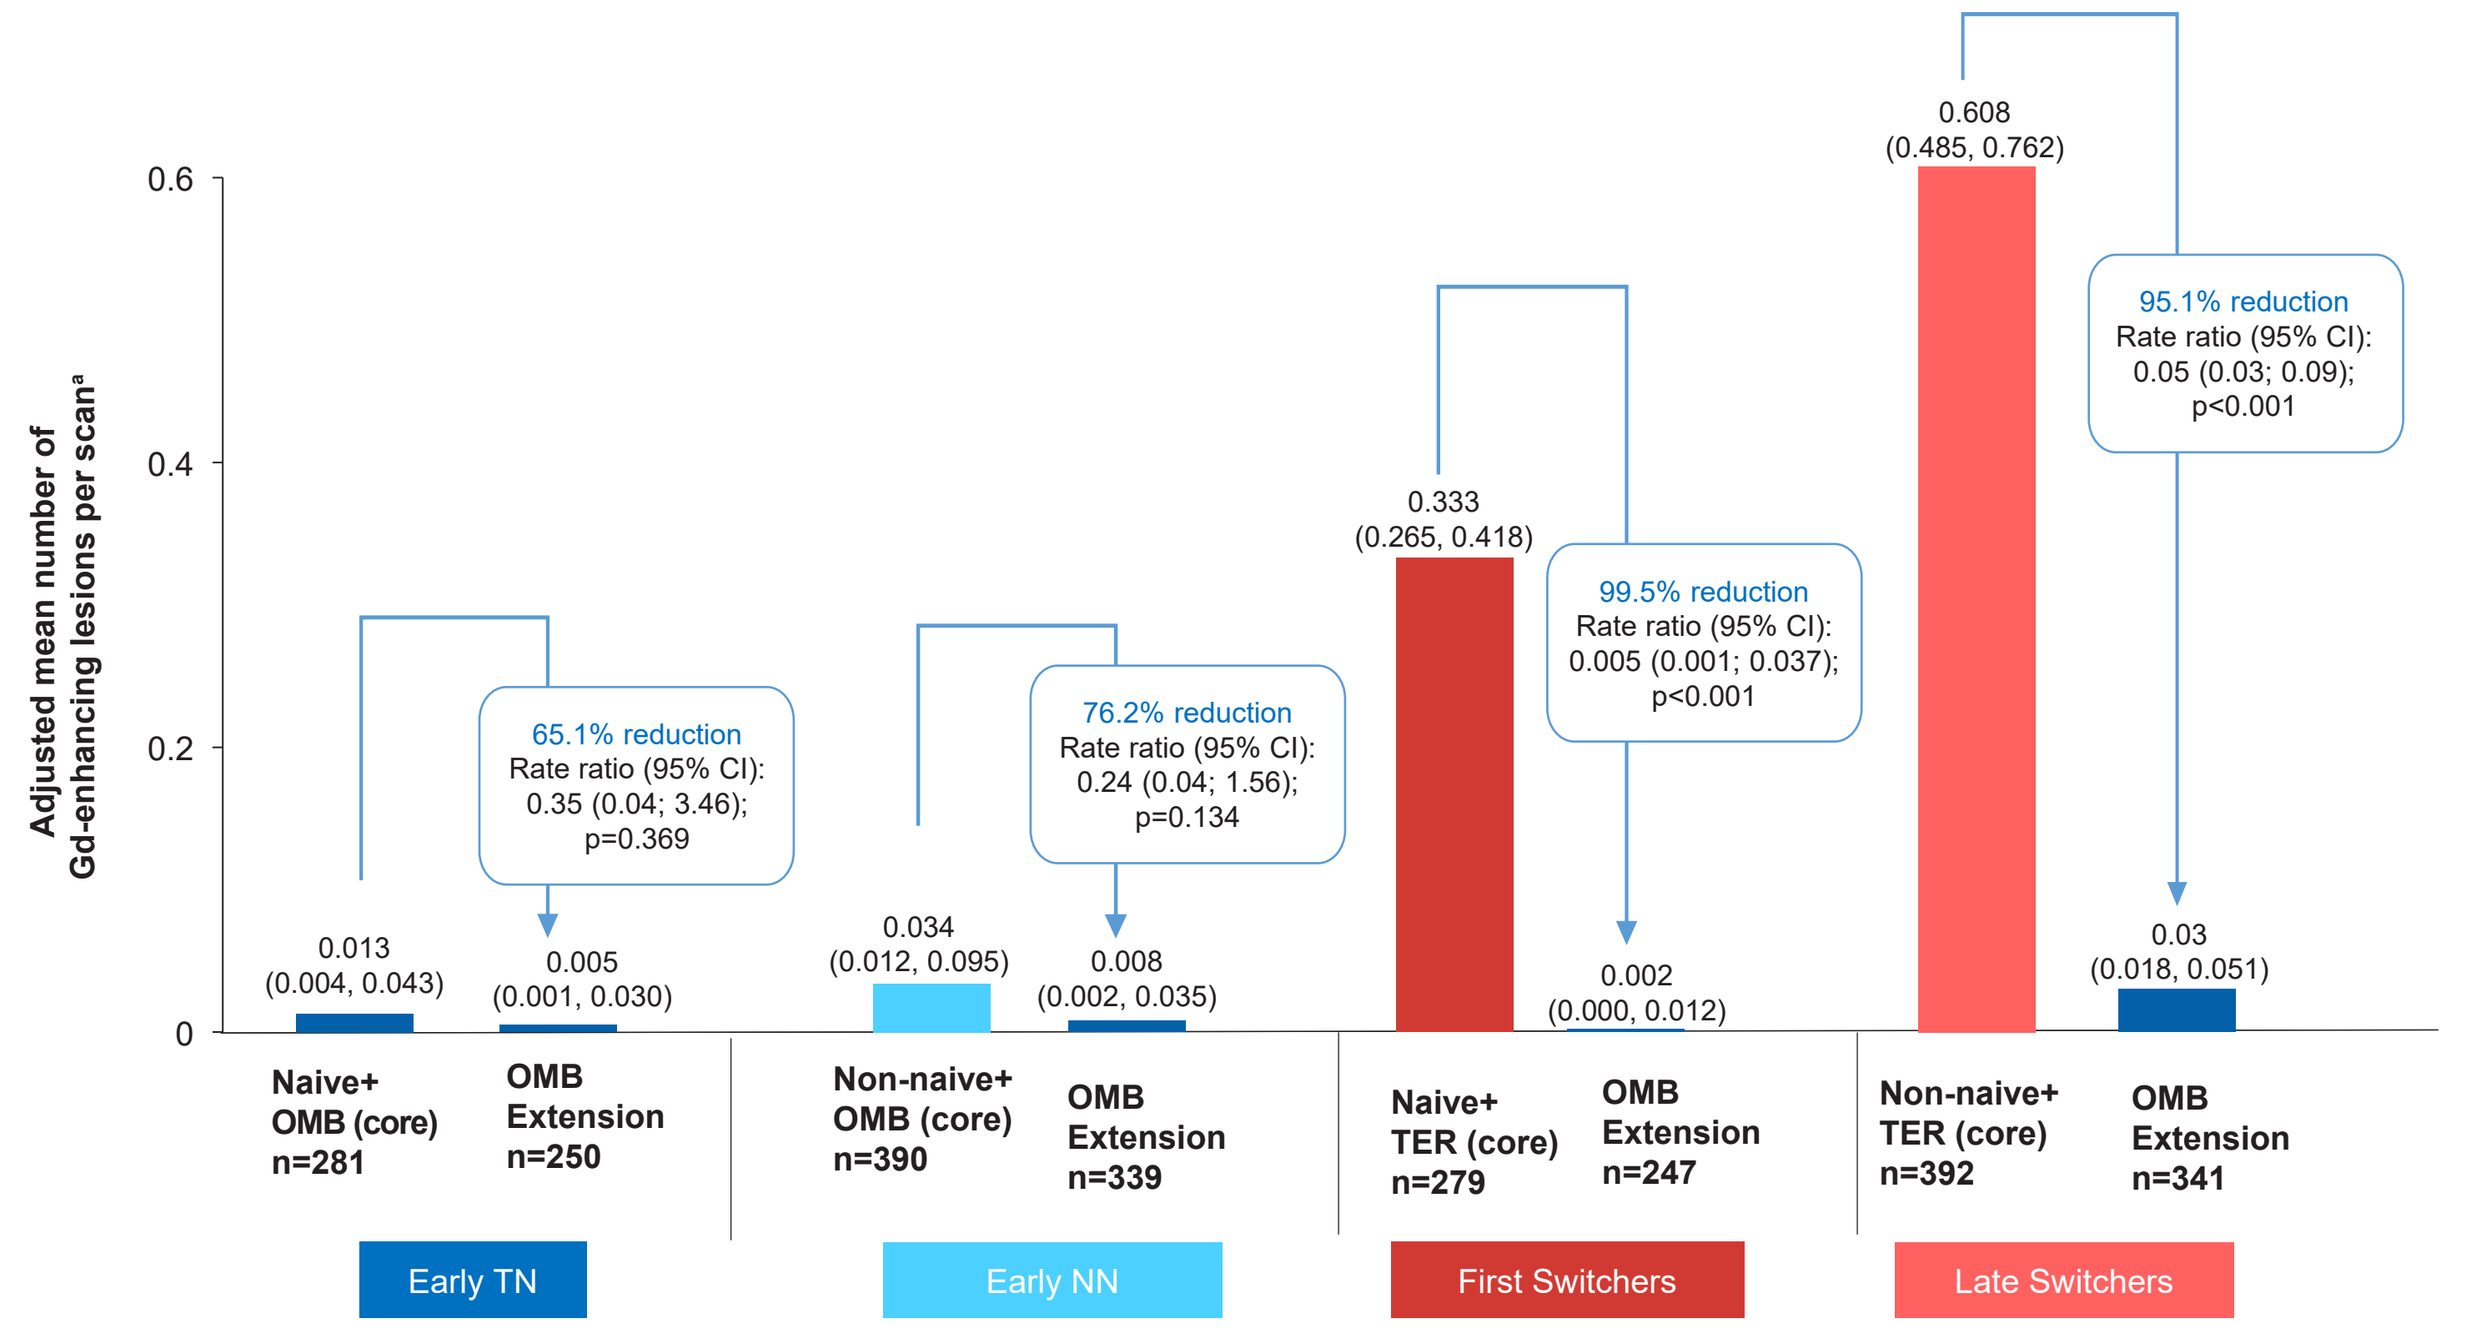

Adjusted mean number of Gd-enhancing lesions per scan[a]
0.6
0.4
0.2
0
0.013 (0.004, 0.043)
0.005 (0.001, 0.030)
65.1% reduction Rate ratio (95% CI): 0.35 (0.04; 3.46); p=0.369
0.034 (0.012, 0.095)
0.008 (0.002, 0.035)
76.2% reduction Rate ratio (95% CI): 0.24 (0.04; 1.56); p=0.134
0.333 (0.265, 0.418)
0.002 (0.000, 0.012)
99.5% reduction Rate ratio (95% CI): 0.005 (0.001; 0.037); p<0.001
0.608 (0.485, 0.762)
0.03 (0.018, 0.051)
95.1% reduction Rate ratio (95% CI): 0.05 (0.03; 0.09); p<0.001
Naive+ OMB (core) n=281
OMB Extension n=250
Non-naive+ OMB (core) n=390
OMB Extension n=339
Naive+ TER (core) n=279
OMB Extension n=247
Non-naive+ TER (core) n=392
OMB Extension n=341
Early TN
Early NN
First Switchers
Late Switchers

[a]Estimated from fitting a piecewise negative binomial model for the time period core phase and extension phase with log-link, adjusted for treatment and region as factors, baseline number of T1 Gd+ lesions and patient's age at baseline as covariates. The natural log of the number of scans with evaluable Gd-enhancing lesion counts by period is used as offset to obtain the lesion rate per scan in each period. Baseline variables are from the core study baseline; CI, confidence interval; early NN, early non-naïve; early TN, early treatment naïve; Gd, gadolinium; OMB, ofatumumab; TER, teriflunomide.

Figure 4. neT2 lesions by treatment groups

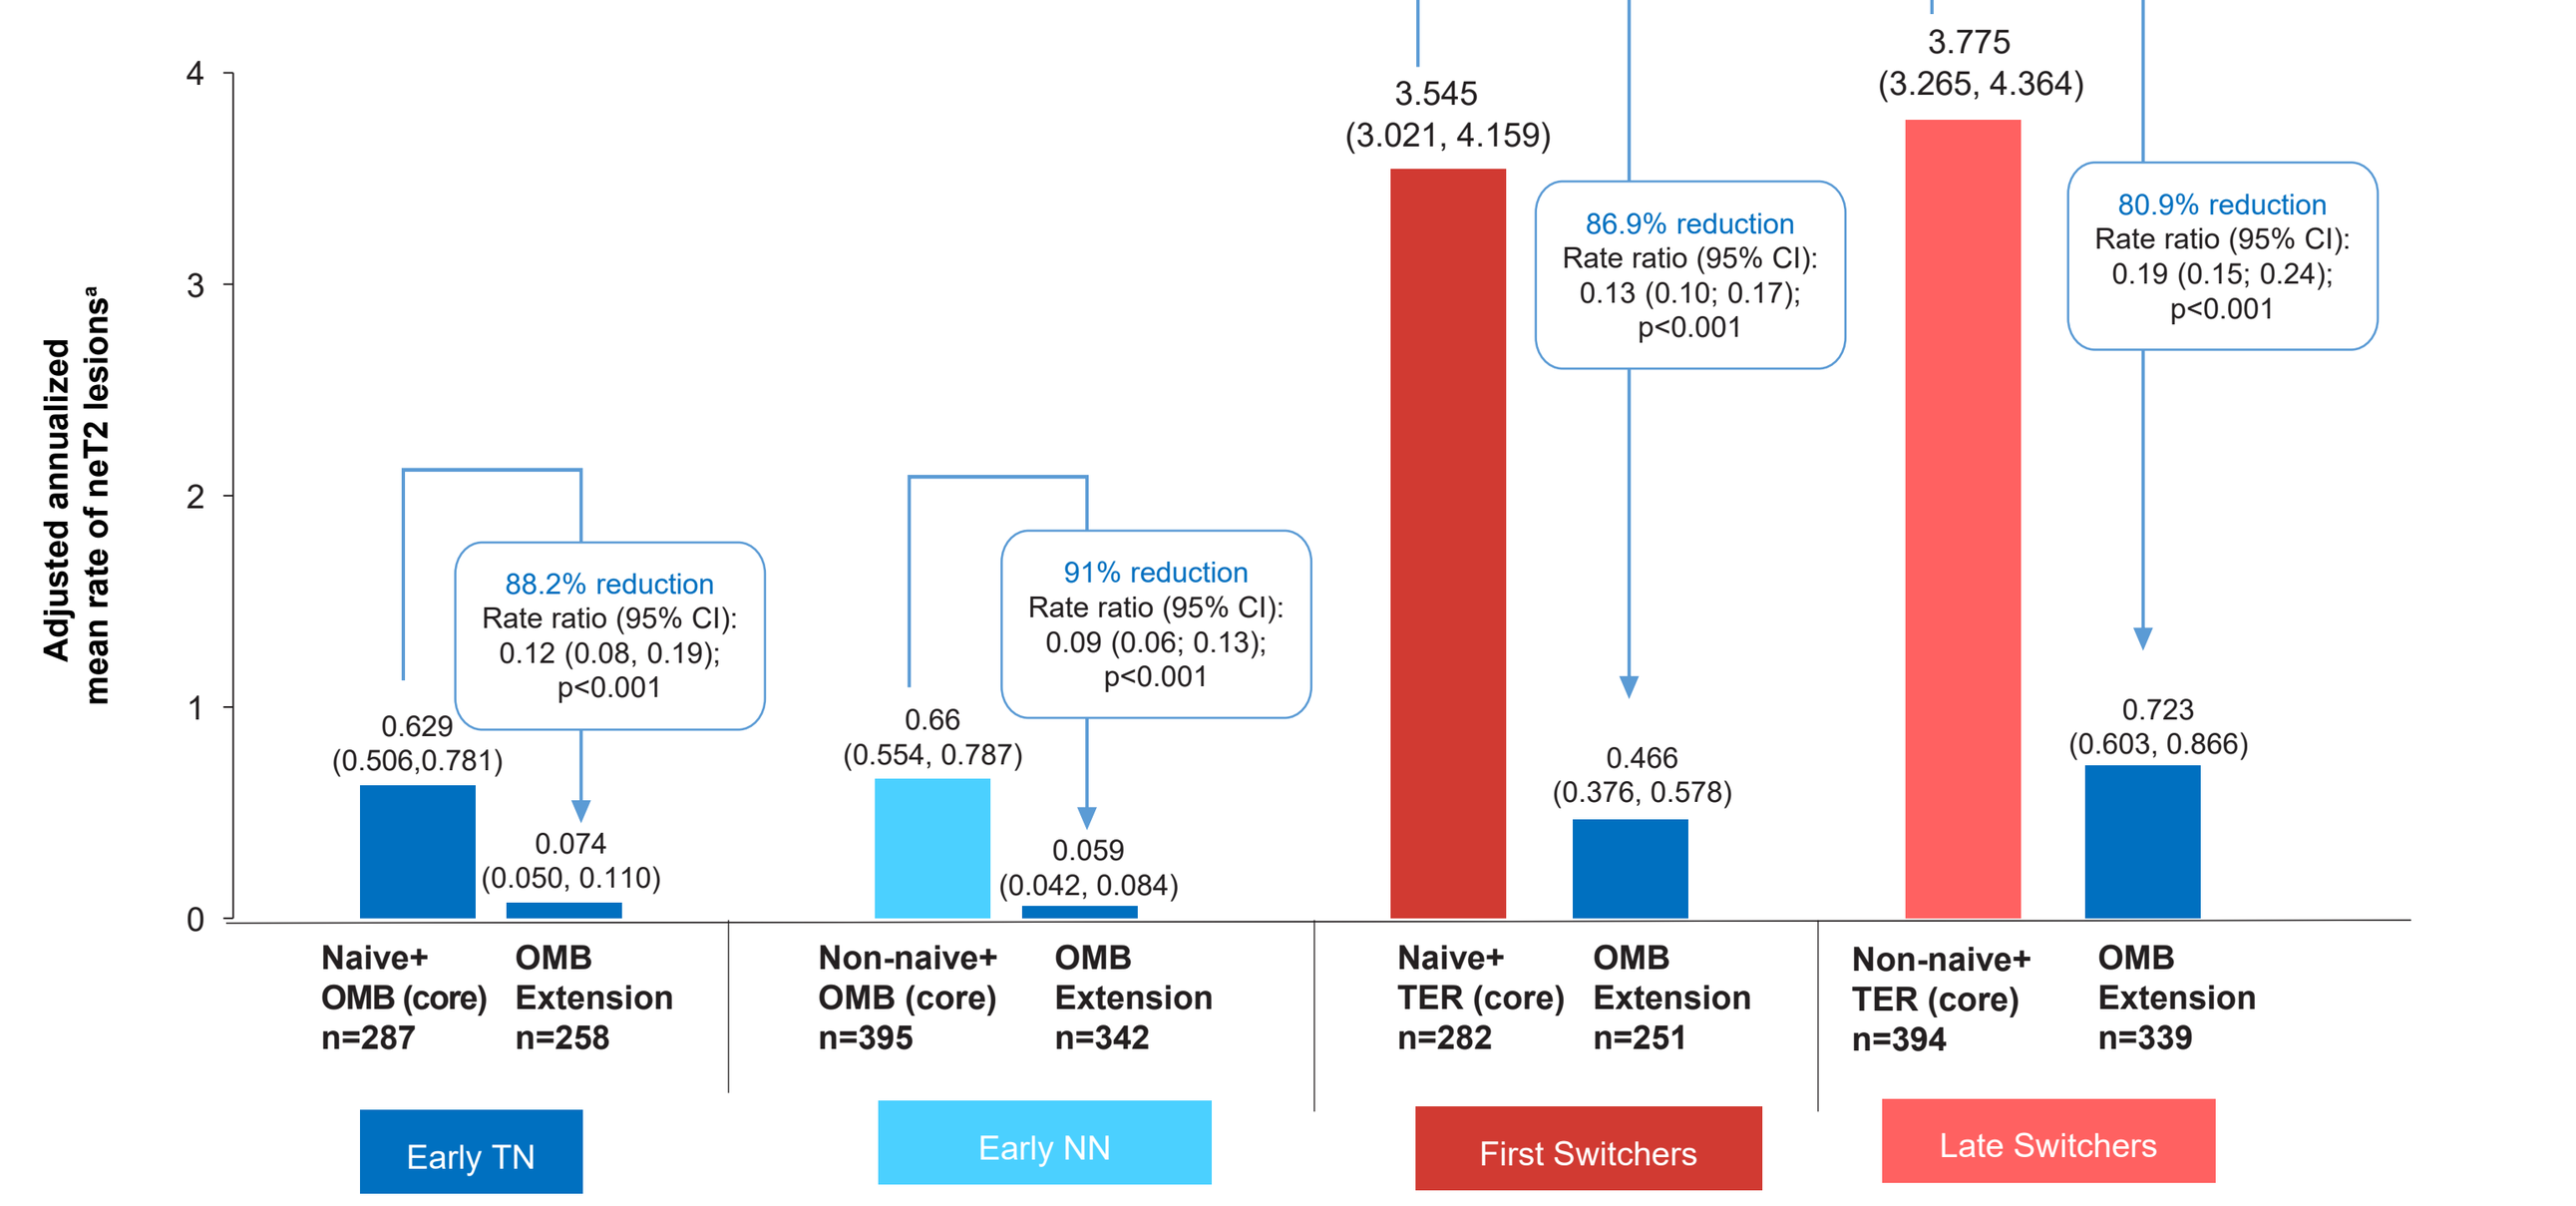

Adjusted annualized mean rate of neT2 lesions[a]
4
3
2
1
0
0.629 (0.506,0.781)
0.074 (0.050, 0.110)
88.2% reduction Rate ratio (95% CI): 0.12 (0.08, 0.19); p<0.001
0.66 (0.554, 0.787)
0.059 (0.042, 0.084)
91% reduction Rate ratio (95% CI): 0.09 (0.06; 0.13); p<0.001
3.545 (3.021, 4.159)
0.466 (0.376, 0.578)
86.9% reduction Rate ratio (95% CI): 0.13 (0.10; 0.17); p<0.001
3.775 (3.265, 4.364)
0.723 (0.603, 0.866)
80.9% reduction Rate ratio (95% CI): 0.19 (0.15; 0.24); p<0.001
Naive+ OMB (core) n=287
OMB Extension n=258
Non-naive+ OMB (core) n=395
OMB Extension n=342
Naive+ TER (core) n=282
OMB Extension n=251
Non-naive+ TER (core) n=394
OMB Extension n=339
Early TN
Early NN
First Switchers
Late Switchers

[a]Estimated from fitting a piecewise negative binomial model for the time period core phase and extension phase with log-link, adjusted for treatment as factor, baseline volume of T2 lesions and patient's age at baseline as covariates. The natural log of the time-in-study (in years) by period is used as offset to annualize the lesion rate in each period. Baseline variables are from the core study baseline. All P values are nominal P values; CI, confidence interval; early NN, early non-naïve; early TN, early treatment naïve; neT2, new or enlarging T2; OMB, ofatumumab; TER, teriflunomide.

Safety outcomes

- The proportion of patients with ≥1 AE with ofatumumab in early TN, early NN, first switchers, and late switchers was 94.1%, 92.5%, 82.6%, and 80.3%, respectively
- Infection rates observed in early TN, early NN, first switchers, and late switchers were 73.7%, 68.1%, 50.7%, and 53.2%, respectively
- Most common AEs were injection-related reaction, COVID-19, nasopharyngitis **(Table 3)**
- Across all the treatment groups, both mean IgG and IgM were above the lower limit of normal (LLN) for majority of patients
 - IgG levels: early TN: 99.3%, early NN: 98.3%, first switchers: 99.6%, late switchers: 98.2%
 - IgM levels: early TN: 70.6%, early NN: 70.3%, first switchers: 79%, late switchers: 72.9%

Table 3. Most common TEAEs[a] (>10% in any group)

Event, n (%)	Early TN, N=289	Early NN, N=401	First Switchers, N=282	Late Switchers, N=395
Any AE	272 (94.1)	371 (92.5)	233 (82.6)	317 (80.3)
Injection-related reaction	79 (27.3)	105 (26.2)	67 (23.8)	88 (22.3)
COVID-19	37 (12.8)	48 (12.0)	39 (13.8)	47 (11.9)
Nasopharyngitis	89 (30.8)	86 (21.4)	41 (14.5)	40 (10.1)
Urinary tract infection	34 (11.8)	77 (19.2)	13 (4.6)	36 (9.1)
Upper respiratory tract infection	47 (16.3)	64 (16.0)	23 (8.2)	38 (9.6)
IgM decreased	44 (15.2)	59 (14.7)	24 (8.5)	51 (12.9)
Headache	63 (21.8)	55 (13.7)	23 (8.2)	26 (6.6)
Injection-site reaction	49 (17.0)	50 (12.5)	28 (9.9)	30 (7.6)
Backpain	37 (12.8)	44 (11.0)	18 (6.4)	18 (4.6)
Arthralgia	32 (11.1)	34 (8.5)	13 (4.6)	19 (4.8)
Diarrhea	20 (6.9)	35 (8.7)	34 (12.1)	50 (12.7)
Alopecia	16 (5.5)	30 (7.5)	5 (1.8)	11 (2.8)
Fatigue	40 (13.8)	31 (7.7)	11 (3.9)	16 (4.1)
Hypertension	14 (14.8)	27 (6.7)	2 (0.7)	11 (2.8)

Data from the time of the first dose of the ofatumumab is included; [a]TEAEs is defined as any AE which started on or after the day of the first dose of the study medication; AEs occurring at any time during the course of the 4 year period; AE, adverse event; COVID-19, corona virus disease; Ig, immunoglobulin; TEAE, treatment emergent AE.

Conclusions

- These results show consistent efficacy in reducing relapses, MRI lesion activity, and the risk of disability worsening observed in early and treatment naïve vs switching from other DMTs, highlighting the value of earlier initiation of ofatumumab in patients with RMS
- Early use of ofatumumab either as first or second line of therapy is beneficial in patients compared to later switchers
- Ofatumumab treatment was well-tolerated across all subgroups of patients and the safety findings were consistent with the overall ASCLEPIOS I and II study populations

References

1. He A, et al. *Lancet Neurol*. 2020;19:307–316;
2. Harding K, et al. *AMA Neurol*. 2019;76:536–541;
3. Iaffaldano P, et al. *Ther Adv Neurol Disord*. 2021;14:17562864211019574;
4. KESIMPTA® (ofatumumab) Prescribing Information. https://www.novartis.us/sites/www.novartis.us/files/kesimpta.pdf (accessed March 29, 2022);
5. Hauser SL, et al. *N Engl J Med*. 2020;383:546–57
6. Hauser SL. oral presentation presented at AAN 2022; S14.004

Acknowledgments

The study was funded by Novartis Pharma AG, Basel, Switzerland. Medical writing support was provided by **Amitha thakur** and **Saimithra Thammera** and design support by **Srinivas Kanchipati** and **Bal Reddy Telekala**, both of Novartis Healthcare Pvt. Ltd., Hyderabad, India. The final responsibility for the content lies with the authors.

Disclosures

Jeffrey A. Cohen received personal compensation for consulting for Biogen, Bristol-Myers Squibb, Convelo, Genentech, Janssen, NervGen, Novartis, and PSI; speaking for H3 Communications; and serving as an Editor of Multiple Sclerosis Journal. **Ralf Gold** has received compensation for serving as a consultant or speaker from Bayer HealthCare, Biogen Idec, Merck Serono, Novartis and Teva Neuroscience. He, or the institution he works for, has received research support from Bayer HealthCare, Biogen Idec, Merck Serono, Novartis and Teva Neuroscience. He has also received honoraria as a Journal Editor from SAGE and Thieme Verlag. **Jérôme de Seze** received personal compensation from Alexion, Allergan, Almirall, Bayer, Biogen, Chugai, CSL Behring, F. Hoffmann-La Roche Ltd., Genzyme, LFB, Merck, Novartis and Teva. **Derrick Robertson** has received fees for consulting, contracted research and speaker's bureau from Biogen, Celgene, EMD Serono, Genentech, Sanofi Genzyme, Janssen, TG therapeutics; consulting fees and speakers bureau for Bristol Myers Squibb, Horizon, and Alexion; consulting fees and contracted research for Novartis; consulting fees for Greenwich biosciences; contracted research for GW Pharmaceuticals, PCORI, Atara Biotherapeutics and CorEvitas. **Heinz Wiendl** has received honoraria for acting as a member of scientific advisory boards for Biogen, Evgen, Genzyme, MedDay Pharmaceuticals, Merck Serono, Novartis, Roche Pharma AG, and Sanofi-Aventis, as well as speaker honoraria and travel support from Alexion, Biogen, Cognomed, F. Hoffmann-La Roche Ltd., Gemeinnützige Hertie-Stiftung, Merck Serono, Novartis, Roche Pharma AG, Genzyme, Teva, and WebMD Global. Heinz Wiendl is acting as a paid consultant for AbbVie, Actelion, Biogen, IGES, Johnson & Johnson, Novartis, Roche, Sanofi-Aventis, and the Swiss Multiple Sclerosis Society. His research is funded by the German Ministry for Education and Research (BMBF), Deutsche Forschungsgemeinschaft (DFG), Else Kröner Fresenius Foundation, Fresenius Foundation, the European Union, Hertie Foundation, NRW Ministry of Education and Research, Interdisciplinary Center for Clinical Studies (IZKF) Muenster and RE Children's Foundation, Biogen, GlaxoSmithKline GmbH, Roche Pharma AG, and Sanofi-Genzyme. **Sibyl Wray** received consulting fees from and advisory boards for Biogen, Celgene, and EMO Serano; speaker bureaus for Biogen, Celgene, EMO Serono, Genentech-Roche, and Sanofi-Genzyme; research support from Biogen, Celgene, EMO Sereno, Genentech-Roche, Novartis, Receptos, Sanofi-Genzyme, and TG Therapeutics. **Francesco Saccà** served on advisory boards for Almirall, Argenx, Avexis, Biogen, Forward Pharma, Merck, Novartis, Pomona, Roche, Sanofi, Alexion, and Takeda. He received public speaking or travel honoraria from Biogen, Mylan, Novartis, Roche, Sanofi, and Teva. He received honoraria from Almirall, Novartis, and Sanofi for educational editorial work. He received consultancy fees from Argenx, Forward Pharma, Novartis, and Novatek. **Amin Azmon, Miriam King, Simone Fantaccini, Ronald Zeilman** are employees of Novartis. **Ludwig Kappos** has received consultancy fees from Actelion, Bayer HealthCare, Biogen, BMS, Genzyme, Janssen, Japan Tobacco, Merck, Novartis, Roche, Sanofi, Santhera, and TG therapeutics; contracted research from Bayer HealthCare, Biogen, European Union, InnoSwiss, Merck, Novartis, Roche, Swiss MS Society, and Swiss National Research Foundation; speaker fees from Bayer HealthCare, Biogen, Merck, Novartis, Roche, and Sanofi; serves on the steering committee for Actelion, Bayer HealthCare, Biogen, BMS, Genzyme, Janssen, Japan Tobacco, Merck, Novartis, Roche, Sanofi, Santhera, TG Therapeutics; Support of educational activities from Allergan, Bayer HealthCare, Biogen, CSL Behring, Desitin, Genzyme, Merck, Novartis, Roche, Pfizer, Sanofi, Shire, and Teva; License fees for Neurostatus products

Poster presented at the Consortium of Multiple Sclerosis Centers in National Harbor, Maryland, June 01-04, 2022

Visit

https://bit.ly/MSKCCMSC

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Presenter email address: cohenj@ccf.org

Scan this QR code to download a copy Poster