P222

Tarun Singhal | tsinghal@bwh.harvard.edu

Ofatumumab Attenuates Cortical Microglial Activity in Patients With Relapsing Forms of Multiple Sclerosis Following B-Cell Depletion: Results From a 9-Month Study

Tarun Singhal[1,2], Steven Cicero[2], Ferdinand Schweser[3], John Hunter Ficke[2], Preksha Kukreja[2], Hong Pan[1,2], Eero Rissanen[1,2], Kelsey Carter[2], Steven Vaquerano[2], Caleb Hansel[2], Gauruv Bose[1,2], Kristin Galetta[1,2], Kristen Babinski[4]; Bonnie I. Glanz[1,2], Jonathan Zurawski[1,2], Maria Houtchens[1,2], Christopher Severson[1,2], Shrishti Saxena[2], Shipra Dubey[1,2], Tanuja Chitnis[1,2], Rohit Bakshi[1,2], Brandon Brown[5], Ibolya Boer[6], Howard L. Weiner[1,2]

[1]Harvard Medical School, Boston, MA, USA; [2]Department of Neurology, Brigham and Women's Hospital, Boston, MA, USA; [3]Department of Neurology, Jacobs School of Medicine & Biomedical Sciences, University of Buffalo, Buffalo Neuroimaging Analysis Center, NY, USA; [4]Tufts Medical Center, Boston, MA, USA; [5]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [6]Novartis Pharma AG, Basel, Switzerland

Scan to access full poster

To download a copy of this poster, visit the web at: https://bit.ly/actrims-forum

Copies of this poster obtained through quick response (QR) code are for personal use only and may not be reproduced without written permission of the authors

KEY FINDINGS & CONCLUSIONS

- Ofatumumab treatment was associated with decreased CoGM microglial activation, decreased brain iron, and reduced sNfL and chemokine levels at 9 months
- Depletion of peripheral CD19+ B cells observed prior to microglial activation may suggest an indirect, downstream effect of B-cell depletion on microglial activity in patients with RMS
- A decrease in CCL19 levels 1 month after treatment initiation warrants further investigation to understand if CCL19 is predictive of response to ofatumumab in the longer term
- The relationship between changes in cortical microglial activation, brain iron, and serum NfL and chemokine levels in response to ofatumumab warrants further investigation

This study was sponsored by Novartis Pharma AG, Basel, Switzerland.

Poster presented at the Americas Committee for Treatment and Research in Multiple Sclerosis (ACTRIMS) Forum, February 29–March 2, 2024.

INTRODUCTION

- Overactivation of microglia, the innate immune cells of the central nervous system, has been implicated in the pathogenesis of multiple sclerosis (MS)[1]
- Iron accumulation has been described within macrophages and/or microglia at the edges of slowly expanding and some inactive lesions. Histopathologically, chronic active lesions are also characterized by progressive tissue matrix damage, driven by a rim of iron-laden, activated microglia at the lesion edge[2,3]
- In a previous analysis, ofatumumab treatment significantly decreased microglial activation in cortical gray matter (CoGM) and brain iron levels at 9 months following treatment initiation[4]

OBJECTIVE

- To further evaluate the effect of ofatumumab on microglial activation and brain iron, in relation to peripheral B-cell depletion, brain atrophy, serum biomarker concentrations, and changes in clinical impairment, longitudinally over 9 months

METHODS

- An open-label, single-center, observational study was conducted in 10 patients with active relapsing MS (RMS; **Figure 1**)
- Statistical parametric mapping analysis was performed to assess the reduction in **positron emission tomography (PET) and quantitative susceptibility imaging (QSM) signal**
- Normal-appearing white matter (NAWM), CoGM, and thalamic regions of interest in standard atlas space were interrogated for peak cluster T values corresponding to p<0.05
- Serum biomarker assessments included measurement of serum neurofilament light chain (sNfL), glial fibrillary acidic protein (GFAP), CXCL10, and CCL19 levels
- Clinical evaluations included the change in Expanded Disability Status Scale (EDSS) score, timed 25-foot walk test (T25FWT) score, anxiety and depression scores (using Hospital Anxiety and Depression Scale), and 9-hole peg test (9-HPT) scores and cognitive assessments (included change in Symbol Digit Modalities Test, California Visuospatial Memory Test – Second Edition, and Brief Visuospatial Memory Test – Revised scores)
- All parameters were assessed at Week 1 and Months 1, 3, and 9 and were compared with baseline values

Figure 1. Study design

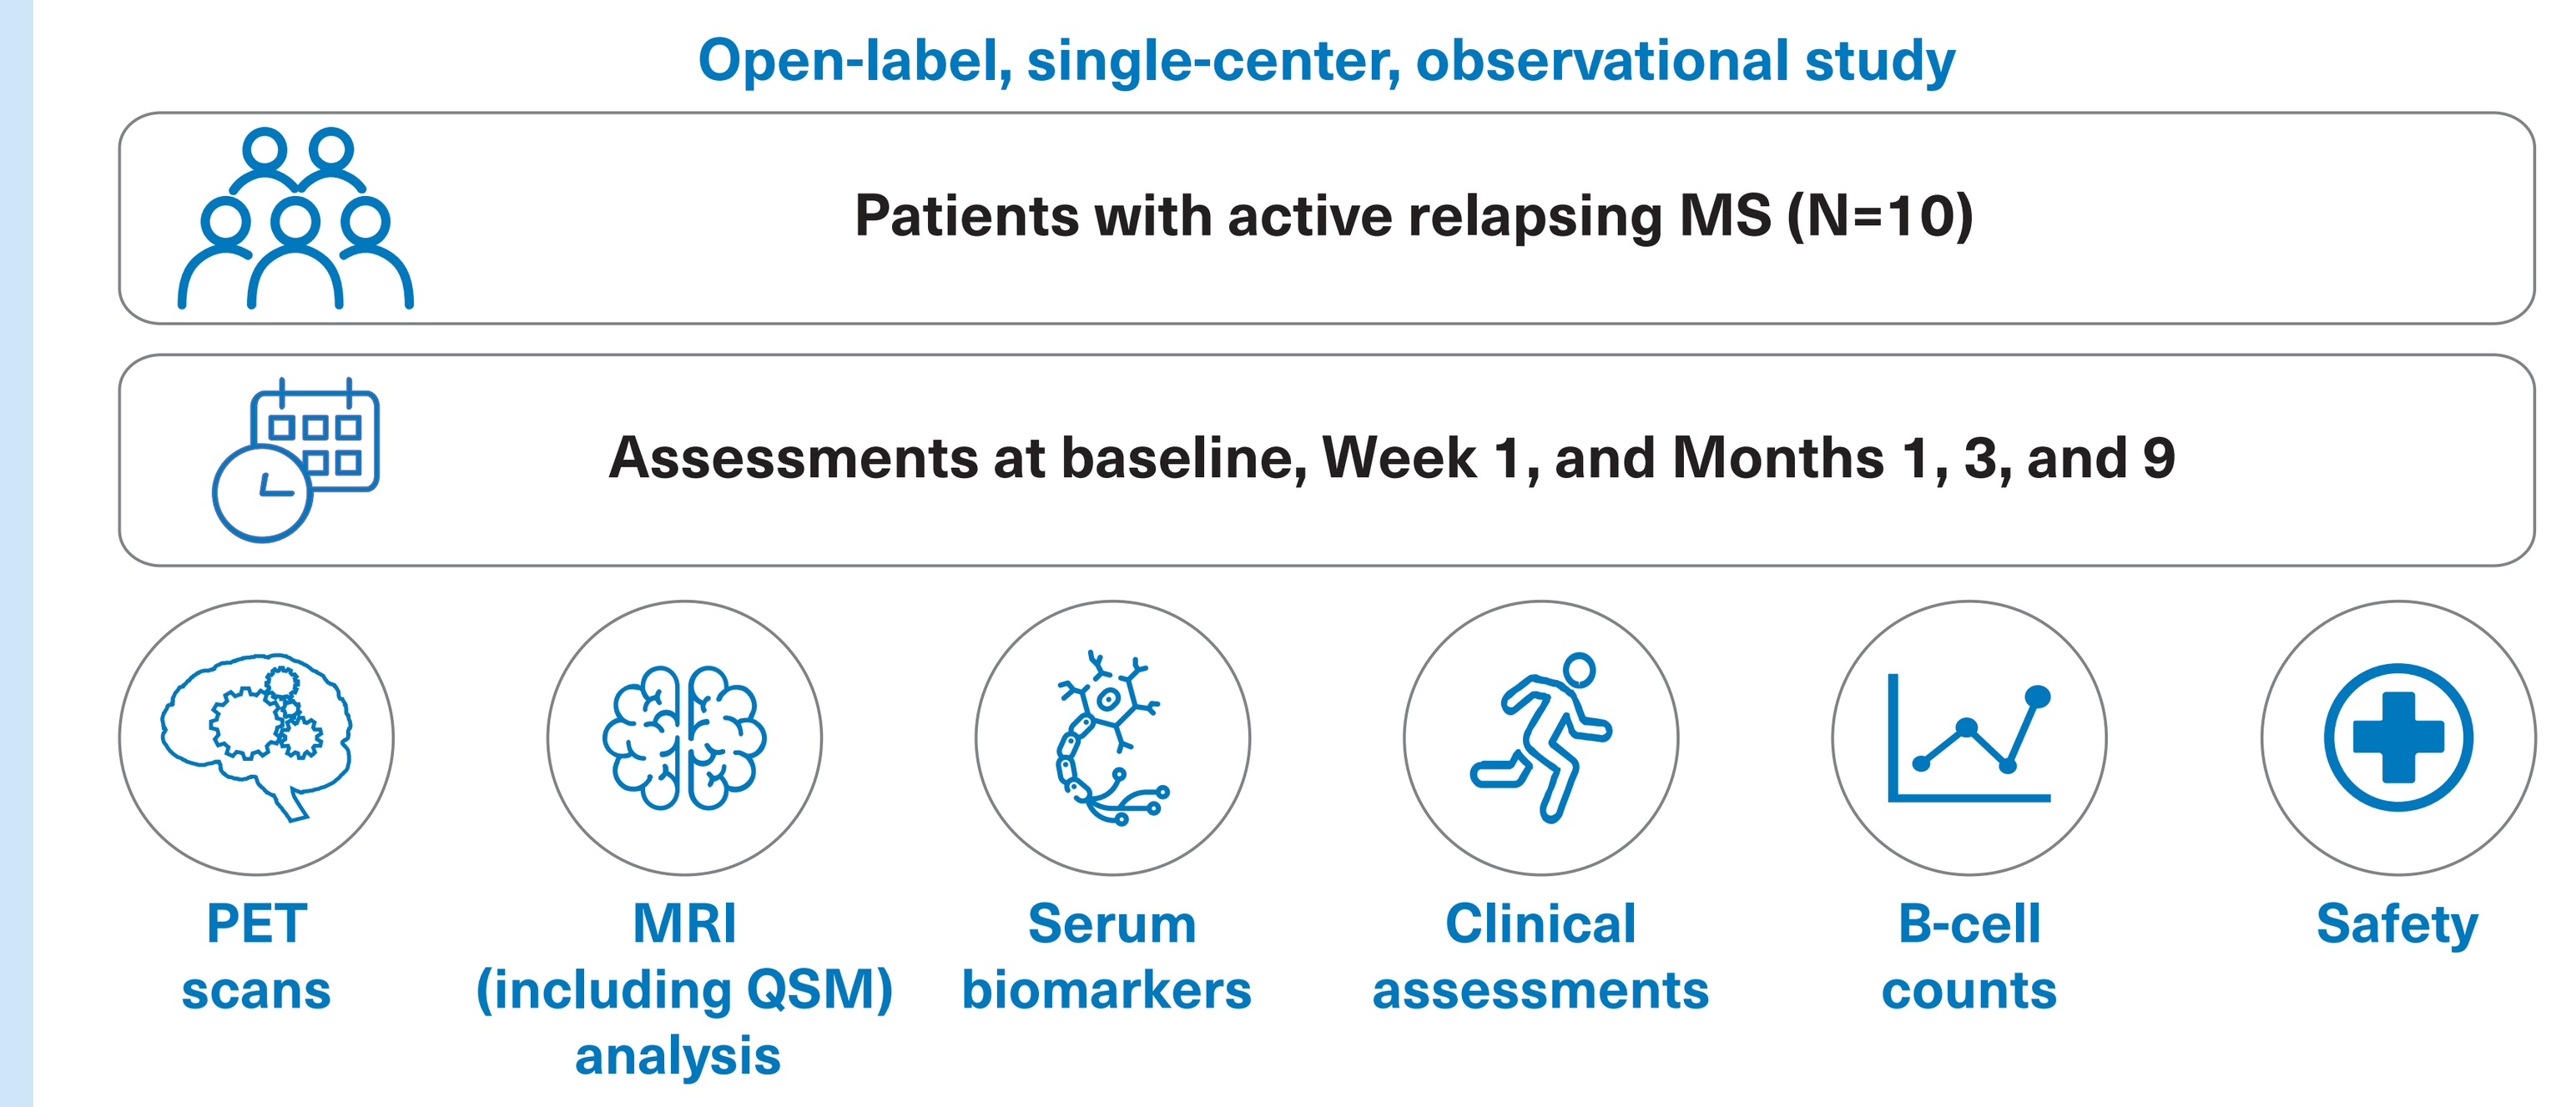
Open-label, single-center, observational study
Patients with active relapsing MS (N=10)
Assessments at baseline, Week 1, and Months 1, 3, and 9
PET scans
MRI (including QSM) analysis
Serum biomarkers
Clinical assessments
B-cell counts
Safety

MRI, magnetic-resonance imaging; MS, multiple sclerosis; PET, positron-emission tomography; QSM, quantitative susceptibility imaging.

RESULTS

Patient characteristics

- Ten patients with RMS were enrolled in the study; data for the first 5 patients (mean±SD age: 40.2±12 years; mean EDSS score: 3.0) are presented here at different time points (baseline, Week 1, and Months 1,3, and 9)

Effect of ofatumumab on microglial clusters

- At 3 and 9 months following ofatumumab initiation, voxel clusters with significantly decreased PET uptake were observed in the CoGM (p<0.01) versus baseline (**Figure 2**)
- No clusters of significantly reduced uptake were detectable in NAWM and thalamic regions of interest

Effect of ofatumumab on brain iron levels

- Clusters of decreased QSM signal, representing decreased brain iron levels, were seen in CoGM and NAWM (p<0.01) at 3 and 9 months[4]

Effect of ofatumumab on brain volume

- Whole brain and and thalamic volumes remained stable over 9 months (**Figure 3A, 3B**)

Figure 3A. Effect of ofatumumab on whole brain volume

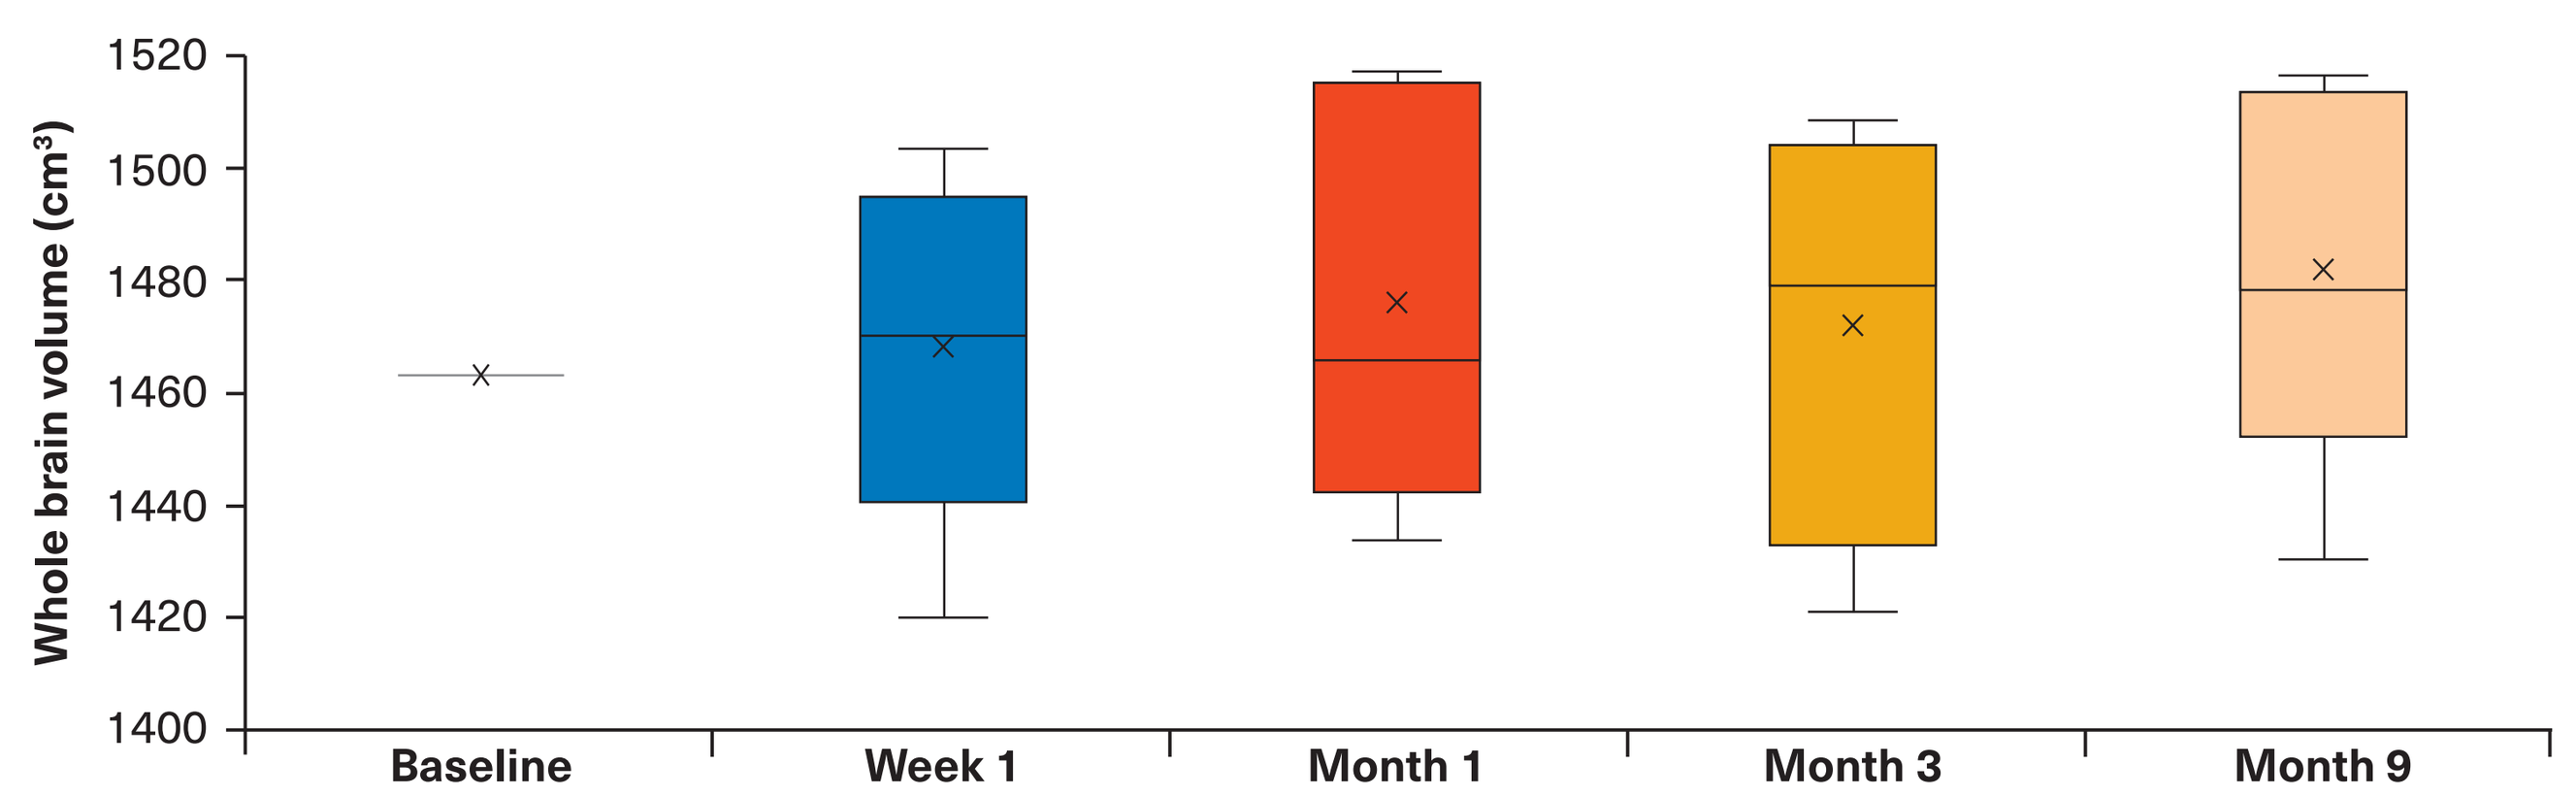
Whole brain volume (cm³)
1520
1500
1480
1460
1440
1420
1400
Baseline
Week 1
Month 1
Month 3
Month 9

Figure 3B. Effect of ofatumumab on thalamic volume

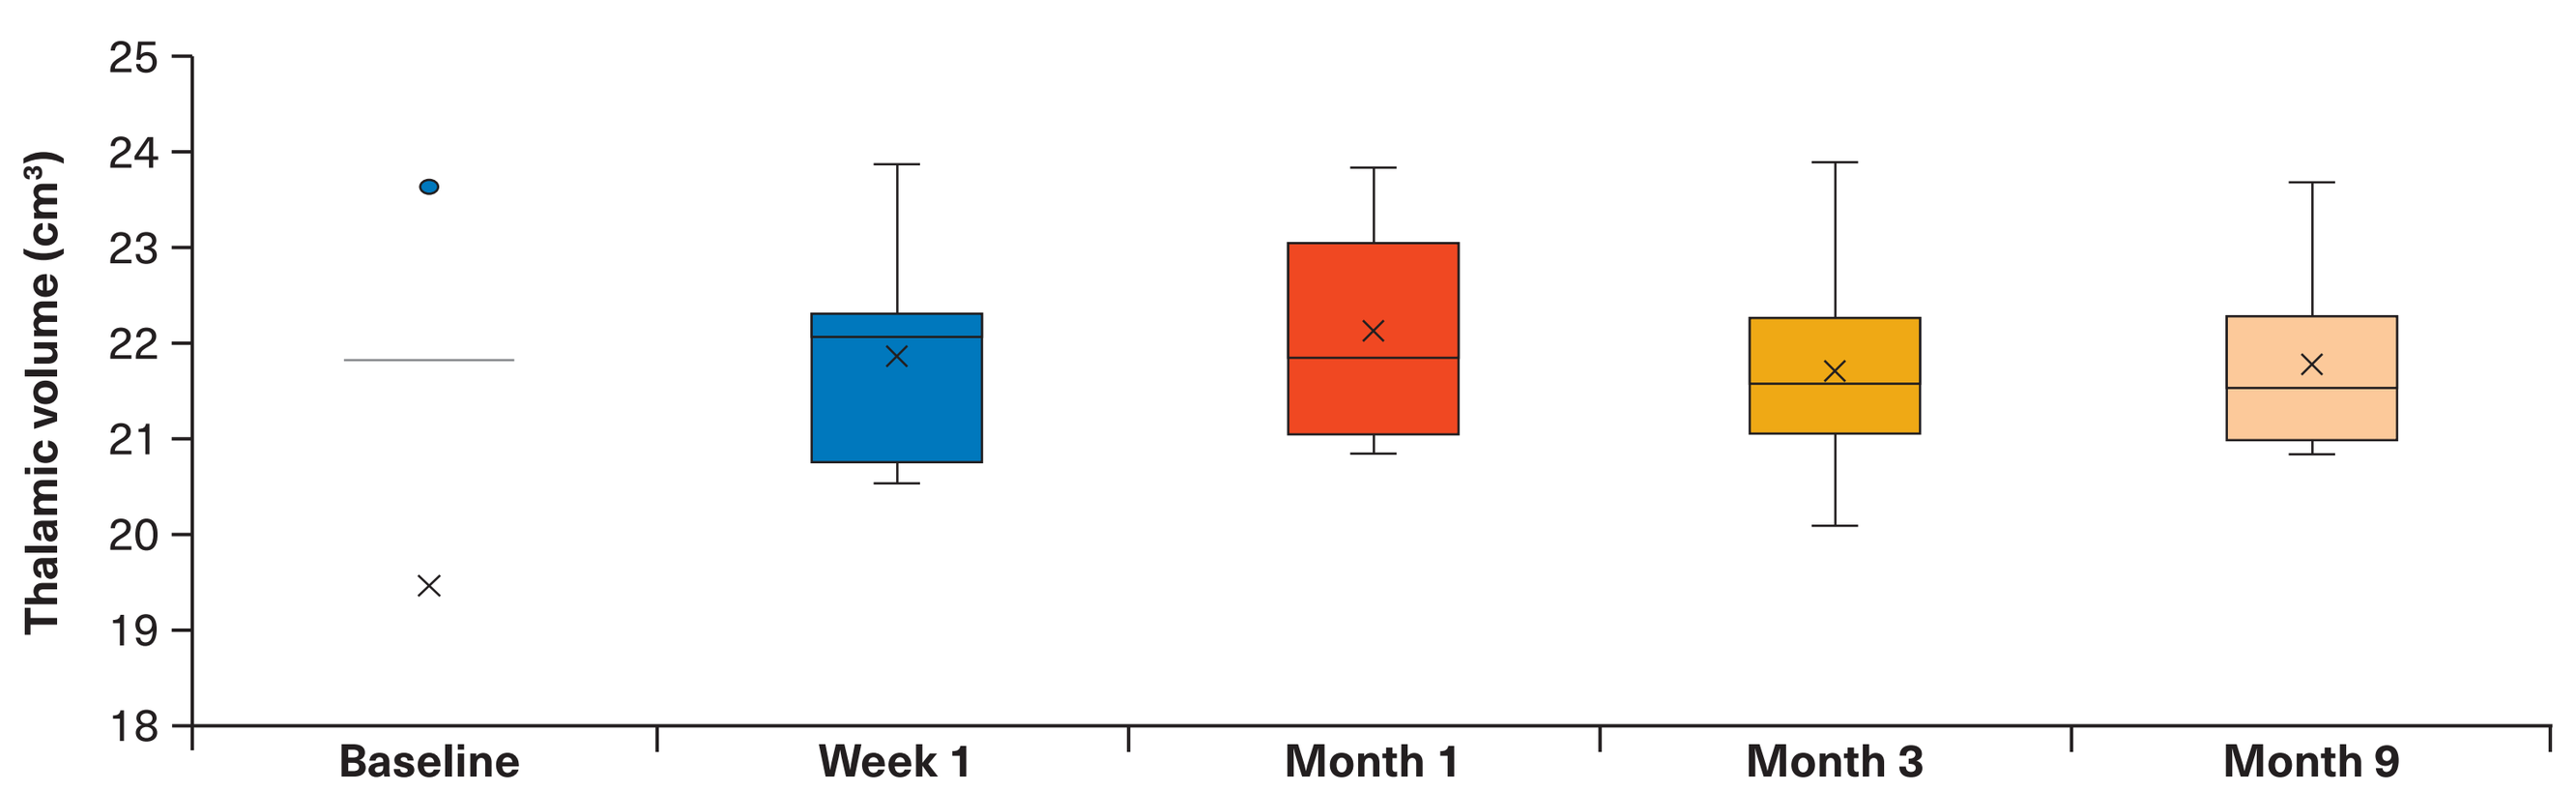
Thalamic volume (cm³)
25
24
23
22
21
20
19
18
Baseline
Week 1
Month 1
Month 3
Month 9

Note: Box limits indicate the range of the central 50% of the data, with a central line marking the median value. X represents mean value; inner points display the data points that lie between the lower whisker line and the upper whisker line. The whiskers indicate variability outside the upper and lower quartiles, and any point outside those lines or whiskers is considered an outlier.

Figure 2. [F-18]PBR06-PET showing reduction in glial activity

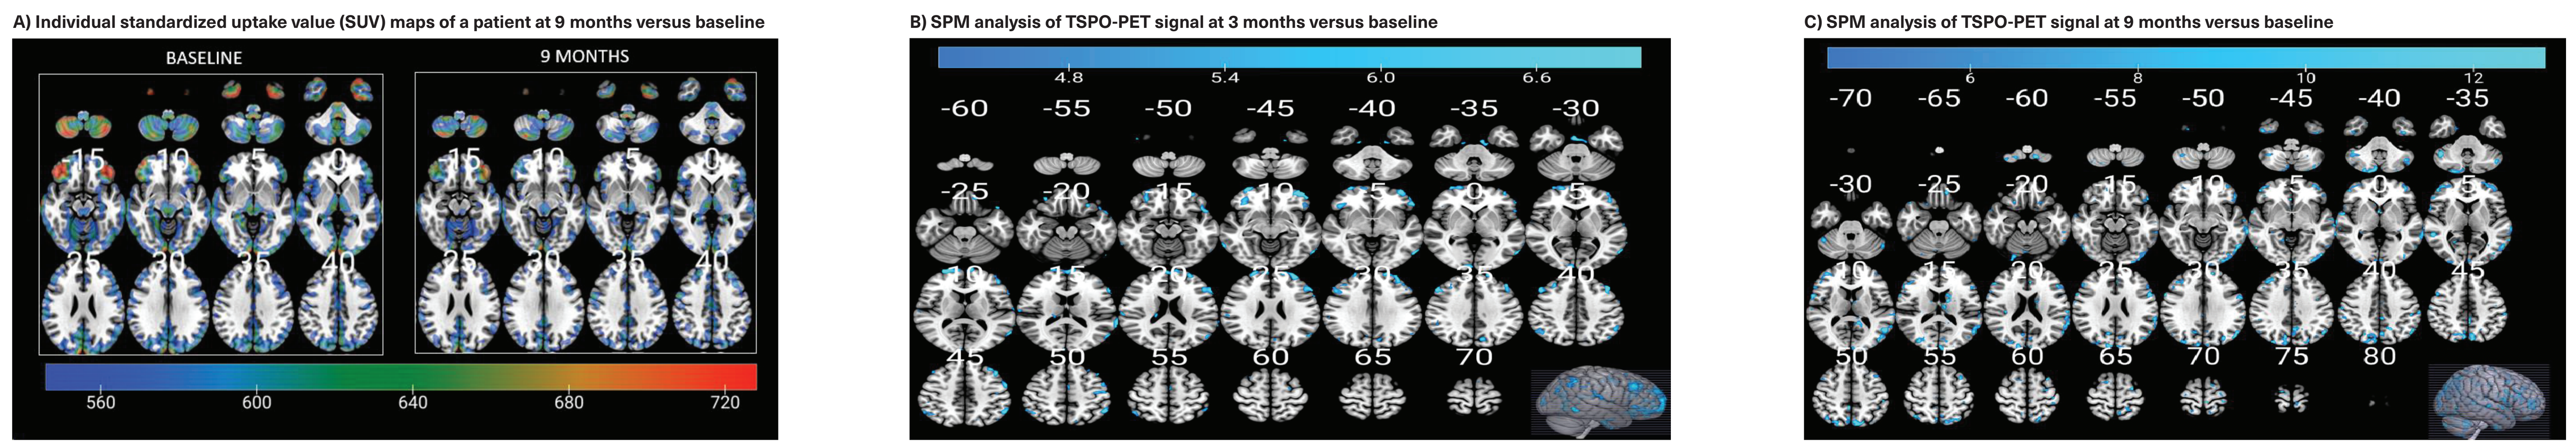
A) Individual standardized uptake value (SUV) maps of a patient at 9 months versus baseline
BASELINE
9 MONTHS
560
600
640
680
720
B) SPM analysis of TSPO-PET signal at 3 months versus baseline
4.8
5.4
6.0
6.6
C) SPM analysis of TSPO-PET signal at 9 months versus baseline
6
8
10
12

PET, positron emission tomography; SPM, statistical parametric mapping; SUV, standard uptake value; TSPO, translocator protein.

Effect of ofatumumab on serum biomarkers

- sNfL, CXCL10, and CCL19, but not serum GFAP, were significantly lower at 9 months (p<0.05) compared with baseline (**Figures 4A–4D**)
- A significant decrease in CCL19 levels was also observed at 1 month following ofatumumab initiation (**Figure 4C**)

Figure 4. Effect of ofatumumab on serum biomarkers

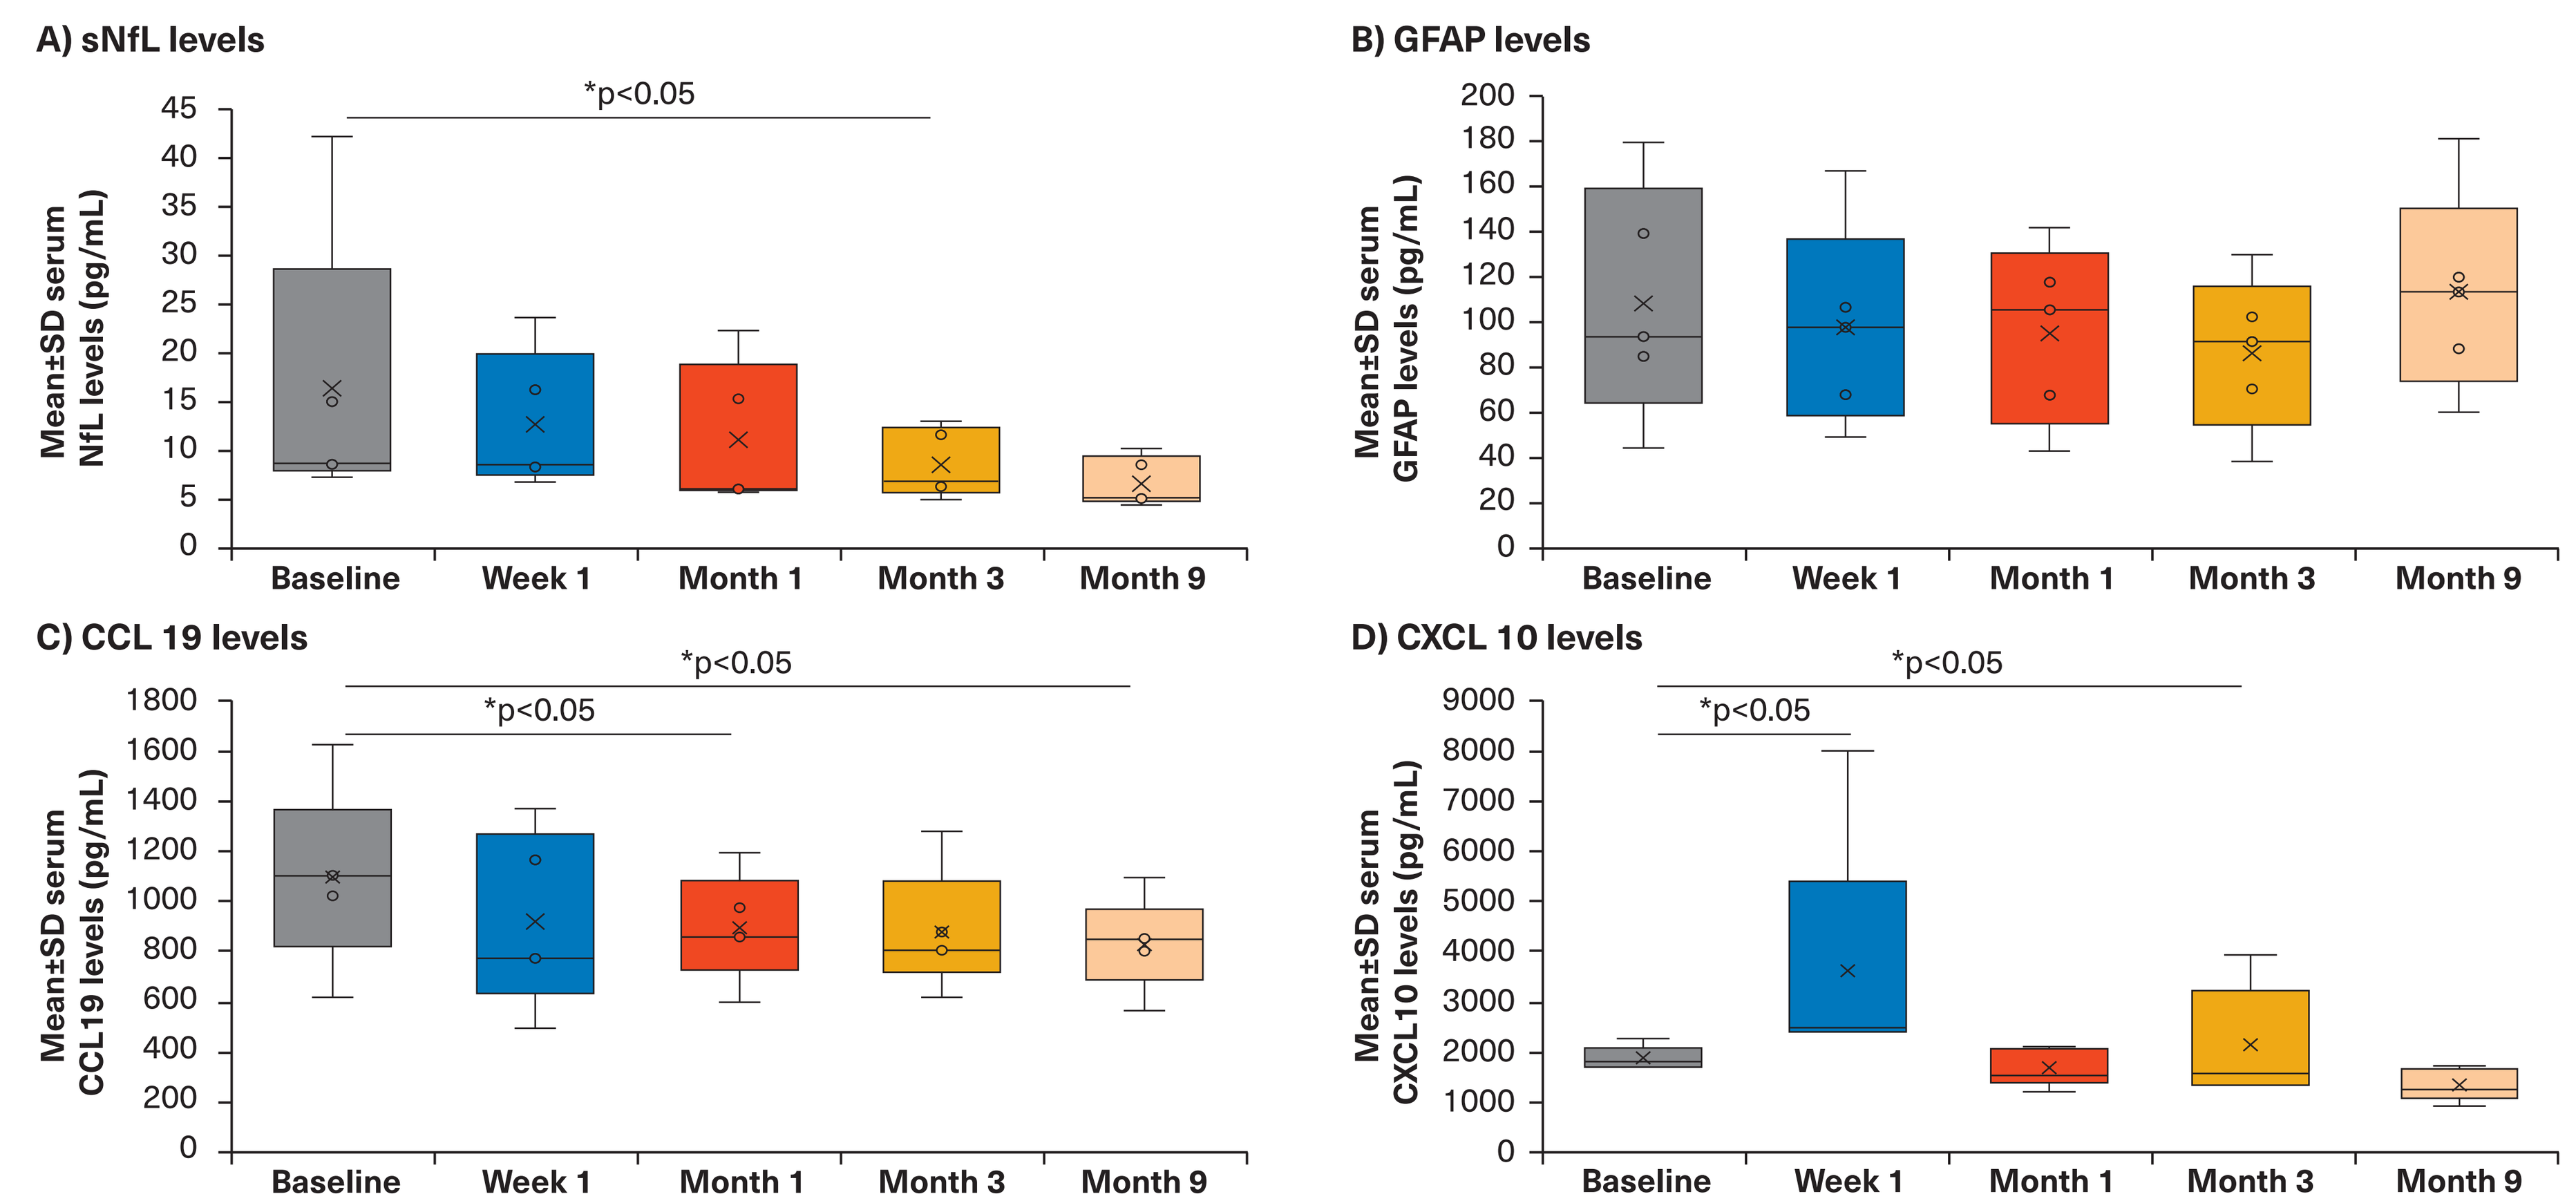
A) sNfL levels
Mean±SD serum NfL levels (pg/mL)
*p<0.05
B) GFAP levels
Mean±SD serum GFAP levels (pg/mL)
C) CCL 19 levels
Mean±SD serum CCL19 levels (pg/mL)
*p<0.05
*p<0.05
D) CXCL 10 levels
Mean±SD serum CXCL10 levels (pg/mL)
*p<0.05
*p<0.05
Baseline
Week 1
Month 1
Month 3
Month 9

GFAP, glial fibrillary acidic protein; SD, standard deviation; sNfL, serum neurofilament light chain.

Note: Box limits indicate the range of the central 50% of the data, with a central line marking the median value. X represents mean value; inner points display the data points that lie between the lower whisker line and the upper whisker line. The whiskers indicate variability outside the upper and lower quartiles, and any point outside those lines or whiskers is considered an outlier.

Effect of ofatumumab on clinical assessments

- EDSS and T25FWT scores remained stable but hospital anxiety and depression scores improved significantly at 9 months (both p<0.05)
- Cognitive function remained stable during the study
- 9-HPT scores in the dominant hand improved significantly at 9 months (p<0.05; **Figure 5**)

Figure 5. 9-HPT scores

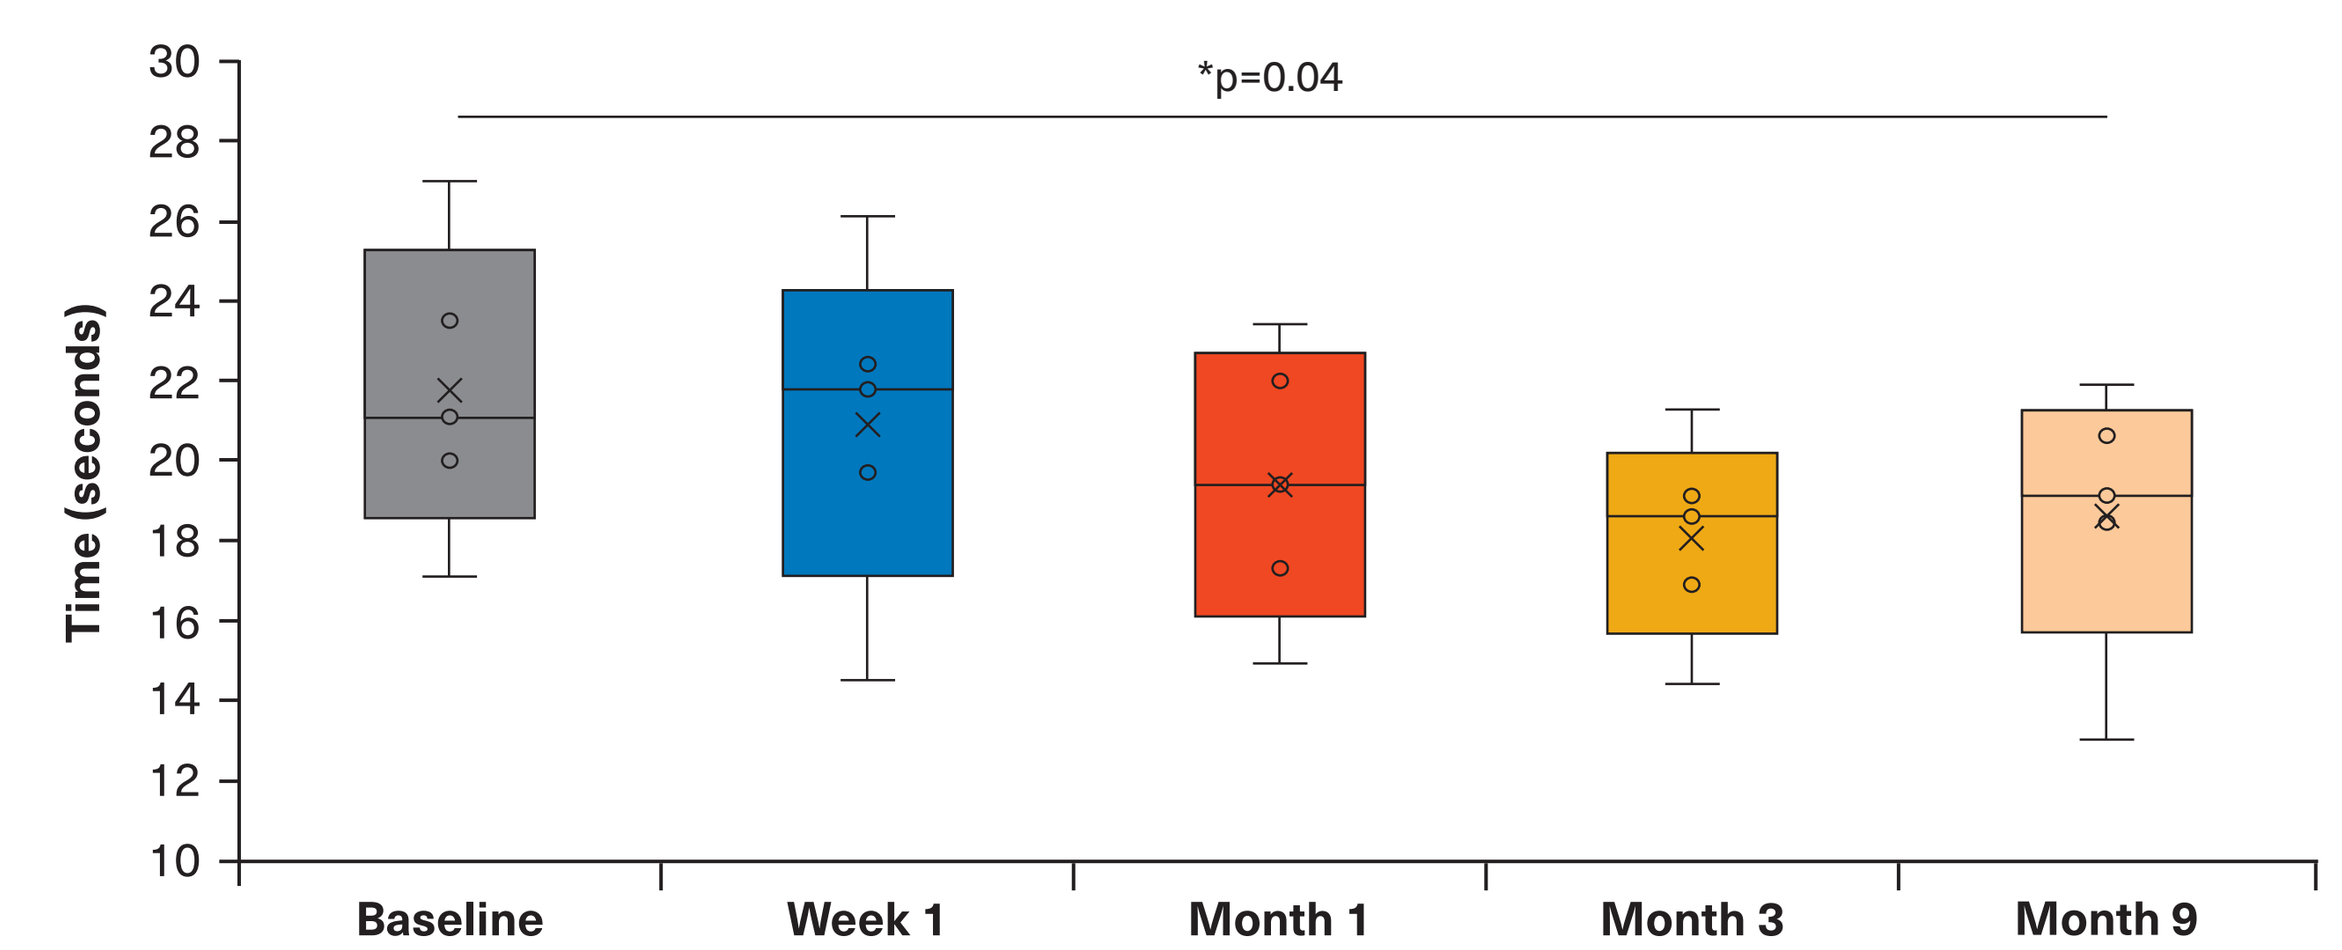
Time (seconds)
*p=0.04
30
28
26
24
22
20
18
16
14
12
10
Baseline
Week 1
Month 1
Month 3
Month 9

Note: Box limits indicate the range of the central 50% of the data, with a central line marking the median value. X represents mean value; inner points display the data points that lie between the lower whisker line and the upper whisker line. The whiskers indicate variability outside the upper and lower quartiles, and any point outside those lines or whiskers is considered an outlier.

Effect of ofatumumab on B cells

- CD19+ B cells decreased significantly after 1 week of ofatumumab treatment and remained low at 9 months

Safety

- No new safety signals were observed during the study

References

1. Luo C, et al. *Neuropsychiatr Dis Treat*. 2017;13:1661–1667
2. Dal-Bianco A, et al. *Acta Neuropathol*. 2017;133(1):25–42
3. Bagnato F, et al. *Brain*. 2011;134:3602–3615
4. Singhal T, et al. P179. Poster presented at ECTRIMS 2023.

Abbreviations

9-HPT, 9-hole peg test; **CoGM**, cortical gray matter; **EDSS**, Expanded Disability Status Scale; **GFAP**, glial fibrillary acidic protein; **MS**, multiple sclerosis; **NAWM**, normal-appearing white matter; **PET**, positron emission tomography; **QSM**, quantitative susceptibility mapping; **RMS**, relapsing multiple sclerosis; **SD**, standard deviation; **sNfL**, serum neurofilament light chain; **SPM**, statistical parametric mapping; **SUV**, standard uptake value; **T25FWT**, timed 25-foot-walk test; **TSPO**, translocator protein.

Disclosures

Tarun Singhal has received research support from Novartis Pharmaceuticals and Genzyme-Sanofi, consulting fees from Novartis Pharmaceuticals and Genentech, speaking fees from Tiziana Life Sciences, and research funding from Nancy Davis Foundation's "Race to Erase MS" program, Ann Romney Center for Neurologic Diseases, Harvard Neuro-Discovery Center, National Multiple Sclerosis Society, Department of Defense, and Water Cove Charitable Foundation. **Steven Cicero, Ferdinand Schweser, John Hunter Ficke, Preksha Kukreja, Hong Pan, Kelsey Carter, Steven Vaquerano, Gauruv Bose, Kristin Galetta, Bonnie I. Glanz, Jonathan Zurawski, Maria K. Houtchens, Caleb Hansel, Kristen Babinski, Shrishti Saxena, Christopher Severson,** and **Shipra Dubey** have nothing to disclose. **Eero Rissanen** has received a fellowship grant from the Sigrid Jusélius Foundation and research grant from the Sakari Alhopuro Foundation. **Rohit Bakshi** has received consulting fees from Bristol-Myers Squibb and EMD Serono, and research support from Bristol-Myers Squibb, EMD Serono and Novartis. **Tanuja Chitnis** has received compensation for consulting from Biogen, Novartis Pharmaceuticals, Roche Genentech, and Sanofi Genzyme. She has received research support from the National Institutes of Health, National MS Society, US Department of Defense, Sumaira Foundation, Brainstorm Cell Therapeutics, EMD Serono, I-MAB Biopharma, Mallinckrodt ARD, Novartis Pharmaceuticals, Octave Bioscience, Roche Genentech, and Tiziana Life Sciences. Disclosures do not conflict with the work being presented. **Brandon Brown** and **Ibolya Boer** are employees of Novartis. **Howard Weiner** has received research support from Cure Alzheimer's Fund, EMD Serono, Inc., Genentech, Inc., National Institutes of Health, National Multiple Sclerosis Society, Sanofi Genzyme, and Verily Life Sciences. He has received payment for consulting from Genentech, Inc, IM Therapeutics, I-MAB Biopharma, MedDay Pharmaceuticals, Tiziana Life Sciences, and vTv Therapeutics.

Acknowledgments

Medical writing support was provided by Venkateswarlu Bonala and Sreelatha Komatireddy and design support by Srinivas Kanchipati, all of Novartis Healthcare Pvt. Ltd., Hyderabad, India. The final responsibility for content lies with the authors.