P037

Anne H. Cross | crossa@wustl.edu

Prognostic Value of Serum Neurofilament Light Chain for Disease Activity in Patients With Relapsing Multiple Sclerosis: Results From Subgroup Analysis Based on Body Mass Index and Age From the Phase 3 ASCLEPIOS I/II Trials

Anne H. Cross[1], Tjalf Ziemssen[2], Douglas L. Arnold[3], Eric Thouvenot[4,5], Scott S. Zamvil[6], Alit Bhatt[7], Wenjia Wei[8], Ibolya Boer[8], Enrique Alvarez[9], Heinz Wiendl[10]

[1]Washington University School of Medicine, Saint Louis, MO, USA; [2]Center of Clinical Neuroscience, Department of Neurology, University Clinic Carl-Gustav Carus, Dresden, Germany; [3]Brain Imaging Centre, Montreal Neurological Institute and Hospital, McGill University, Montreal, QC, Canada; [4]Department of Neurology, Centre Hospitalier Universitaire (CHU) Nîmes, University of Montpellier, Nîmes, France; [5]Institut de Génomique Fonctionnelle, UMR, Institut National de la Santé et de la Recherche Médicale (INSERM), University of Montpellier, Montpellier, France; [6]UCSF Weill Institute for Neurosciences, Department of Neurology, University of California, San Francisco, CA, USA; [7]Novartis Healthcare Pvt. Ltd., Hyderabad, India; [8]Novartis Pharma AG, Basel, Switzerland; [9]Department of Neurology, Rocky Mountain MS Center at the University of Colorado, Aurora, CO, USA; [10]University of Muenster, Muenster, Germany

Scan to access full poster

To download a copy of this poster, visit the web at: https://bit.ly/actrims-forum

Copies of this poster obtained through quick response (QR) code are for personal use only and may not be reproduced without written permission of the authors

KEY FINDINGS & CONCLUSIONS

- The use of a single sNfL threshold, as defined in the ASCLEPIOS I/II study protocols, was prognostic of future lesion formation in the overall study population, including subgroups defined by BMI and age
- Overall, these data support further work on the optimization of sNfL for prognostication of future MS disease activity that would be applicable for patients with RMS

Please refer to these related posters for more details on the prognostic value of sNfL:

- Prognostic Value of On-Treatment Serum Neurofilament Light Chain for New or Enlarging T2 Lesions in People With Relapsing Multiple Sclerosis: Pooled Analysis of the ASCLEPIOS I/II Trials (**P032**)
- Baseline Serum Neurofilament Light Chain Levels Predict Future Disease Activity Irrespective of Race/Ethnicity: Results From the Phase 3 ASCLEPIOS I/II Trials (**P036**)

This study was sponsored by Novartis Pharma AG, Basel, Switzerland.

Poster presented at the Americas Committee for Treatment and Research in Multiple Sclerosis (ACTRIMS) Forum, February 29–March 2, 2024.

INTRODUCTION

- A challenge encountered in clinical practice with relapsing multiple sclerosis (RMS) is the difficulty in prognosticating the risk of future disease activity because of the variable disease course across patients[1]
- Inflammatory disease activity mostly occurs in the younger RMS population and declines with age[2]
 - A biomarker that can prognosticate disease activity may help optimize individualized patient care and limit irreversible neurological damage, even in the absence of overt clinical symptoms or radiological signs[3]
- In the phase 3 ASCLEPIOS I/II trials (ofatumumab versus teriflunomide in people with RMS [pwRMS]), a pre-planned analysis of baseline serum neurofilament light chain (sNfL) levels based on being above or below the baseline median found that sNfL levels were prognostic for on-study lesion formation and brain volume loss in the overall population and in recently diagnosed treatment-naive participants[4]
- The prognostic value of sNfL was also observed when participants were categorized by baseline sNfL concentration quartiles
 - Irrespective of treatment, participants in the lowest quartile (Q1) had the lowest risk for on-study new or enlarging [ne] T2 lesions, with the risk for neT2 lesions increasing sequentially up to the highest sNfL quartile (Q4)[5]
- Some factors such as age and/or body mass index (BMI) may influence sNfL levels; however, in younger pwRMS without comorbidities, the impact of these factors on sNfL levels is expected to be limited compared to the effect of MS neuroinflammation itself[6-10]

OBJECTIVE

- To evaluate the prognostic value of low versus high sNfL (based on a pre-planned analysis of baseline sNfL being above or below the baseline median) for future disease activity according to BMI and age in pwRMS

METHODS

Study design

- ASCLEPIOS I/II were two phase 3, double-blind, active-controlled trials in which participants with RMS were randomized to receive either ofatumumab or teriflunomide for up to 30 months
- Participants aged 18–55 years with a diagnosis of RMS, Expanded Disability Status Scale (EDSS) score 0–5.5, ≥1 relapse in the year before screening or ≥2 relapses in the last 2 years before screening, or ≥1 gadolinium-enhancing (Gd+) lesion on magnetic resonance imaging (MRI) in the year before randomization were included
- The baseline sNfL cut-off was predefined in the clinical study protocol (i.e., before measuring sNfL or any clinical or radiological outcomes), as the median sNfL value for the overall population across ASCLEPIOS I/II (9.3 pg/mL)
- Participants were stratified as having high (≥9.3 pg/mL) versus low (<9.3 pg/mL) based on the median baseline sNfL concentration

Assessments

- Quantification of sNfL levels was performed centrally (Navigate BioPharma Services, Carlsbad, CA, USA), as a single batch at the end of the trials, using a validated Quanterix Simoa® NF-light advantage kit
- MRI scans were performed at baseline, Months 12 and 24, and end of treatment/end of study
- The prognostic value of high versus low baseline sNfL for the annualized rate of neT2 lesions was assessed in baseline BMI (<24.5 vs ≥24.5 kg/m²) and age (<38.0 vs ≥38.0 years) subgroups

Statistical analysis

- Negative binomial regression model adjusting for sNfL group and sNfL group by BMI/age subgroup interaction was used to estimate the lesion rate ratio for high versus low sNfL levels in each BMI/age subgroup

RESULTS

Participant characteristics

- Of the 1882 participants randomized in ASCLEPIOS I/II trials, 1746 (92.8%) had baseline sNfL data, including 1678 (96.1%) participants with neT2 and sNfL data available for BMI and age subgroups
- Baseline characteristics were similar between sNfL groups, except the number of Gd+ lesions and T2 lesion volume, which were considerably higher in participants with high versus low baseline sNfL (**Table 1**)

Table 1. Demographic and disease characteristics by sNfL category at baseline

Characteristic	Low sNfL category (<9.3 pg/mL) N=876[a]	High sNfL category (≥9.3 pg/mL) N=870[a]
Age, years	38.6±8.5	37.8±9.7
BMI, kg/m²	27.0±6.4	24.7±5.5
Female, n (%)	588 (67.1)	602 (69.2)
MS duration since first symptom, years	8.3±7.2	7.9±6.9
Previously treated with DMT, n (%)	537 (61.3)	507 (58.3)
Number of relapses in the year before the study	1.2±0.7	1.3±0.7
Time since onset of most recent relapse, months	7.8±13.5	7.0±9.3
EDSS score	2.8±1.3	2.9±1.4
Normalized brain volume, cm³	1447.6±74.8	1437.2±81.0
Number of Gd+ T1 lesions	0.4±1.2	2.6±5.4
Participants free of Gd+ T1 lesions, n (%)	679 (77.5)	383 (44.0)
T2 lesion volume, cm³	9.4±10.6	16.7±15.0
Median sNfL, pg/mL	6.76	14.23

[a]Only participants with non-missing baseline sNfL values are included.
Data are expressed as mean±standard deviation unless specified otherwise.
BMI, body mass index; DMT, disease-modifying therapy; EDSS, Expanded Disability Status Scale; Gd+, gadolinium-enhancing; MS, multiple sclerosis; sNfL, serum neurofilament light chain.

Prognostic value of sNfL for neT2 lesions by BMI and age subgroups

- Across BMI and age subgroups, the mean annualized rate of neT2 lesions was significantly higher in participants with high sNfL (≥9.3 pg/mL) versus low sNfL (<9.3 pg/mL) levels (**Figure 1**)

Figure 1. Mean annualized rate of neT2 lesions by A) age and B) BMI subgroups[a]

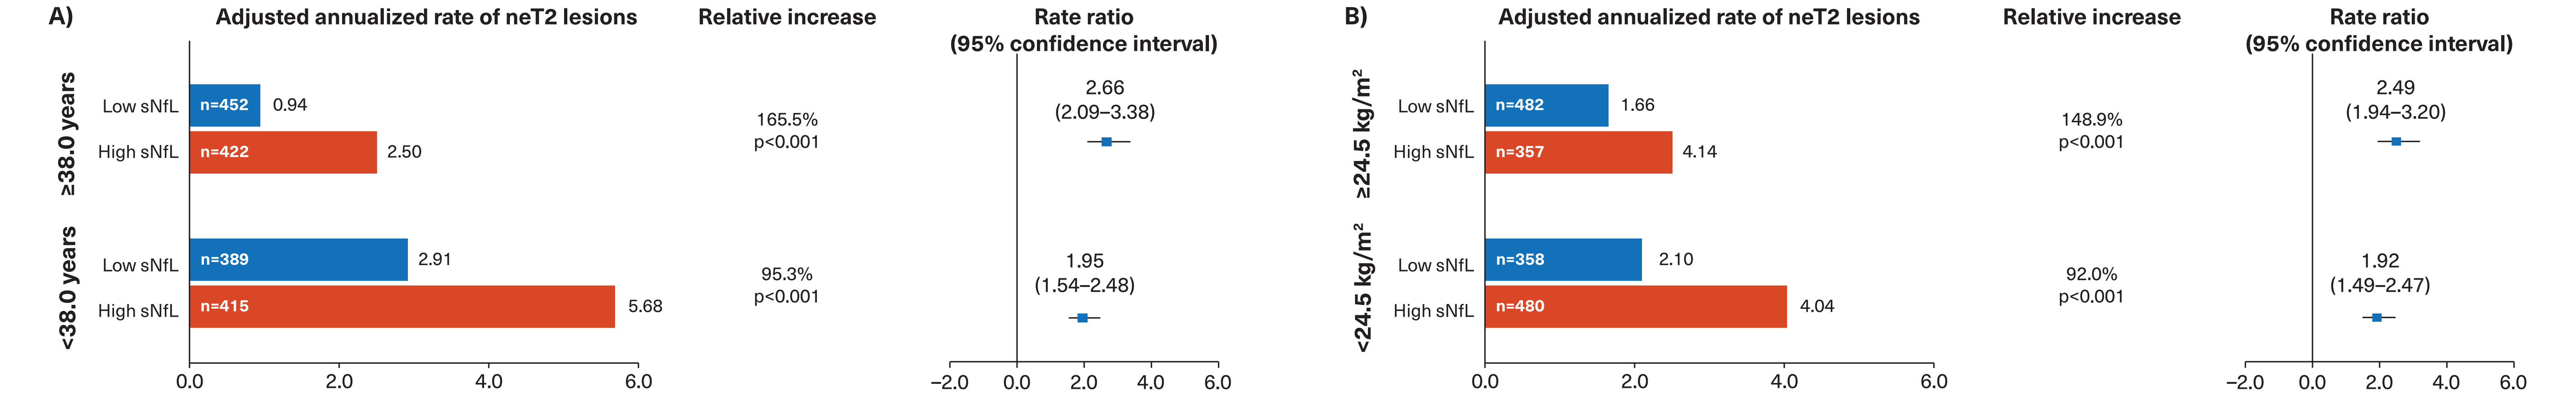
A)
Adjusted annualized rate of neT2 lesions
Relative increase
Rate ratio (95% confidence interval)
≥38.0 years
Low sNfL
n=452
0.94
High sNfL
n=422
2.50
165.5%
p<0.001
2.66
(2.09–3.38)
<38.0 years
Low sNfL
n=389
2.91
High sNfL
n=415
5.68
95.3%
p<0.001
1.95
(1.54–2.48)
0.0 2.0 4.0 6.0
−2.0 0.0 2.0 4.0 6.0
B)
Adjusted annualized rate of neT2 lesions
Relative increase
Rate ratio (95% confidence interval)
≥24.5 kg/m²
Low sNfL
n=482
1.66
High sNfL
n=357
4.14
148.9%
p<0.001
2.49
(1.94–3.20)
<24.5 kg/m²
Low sNfL
n=358
2.10
High sNfL
n=480
4.04
92.0%
p<0.001
1.92
(1.49–2.47)
0.0 2.0 4.0 6.0
−2.0 0.0 2.0 4.0 6.0

[a]Analyses were based on the population that had both baseline sNfL and neT2 data available. BMI, body mass index; neT2, new or enlarging T2 lesions; sNfL, serum neurofilament light chain.

Additional analyses of data using a single sNfL threshold by age and BMI subgroups (Figures 2A and 2B)

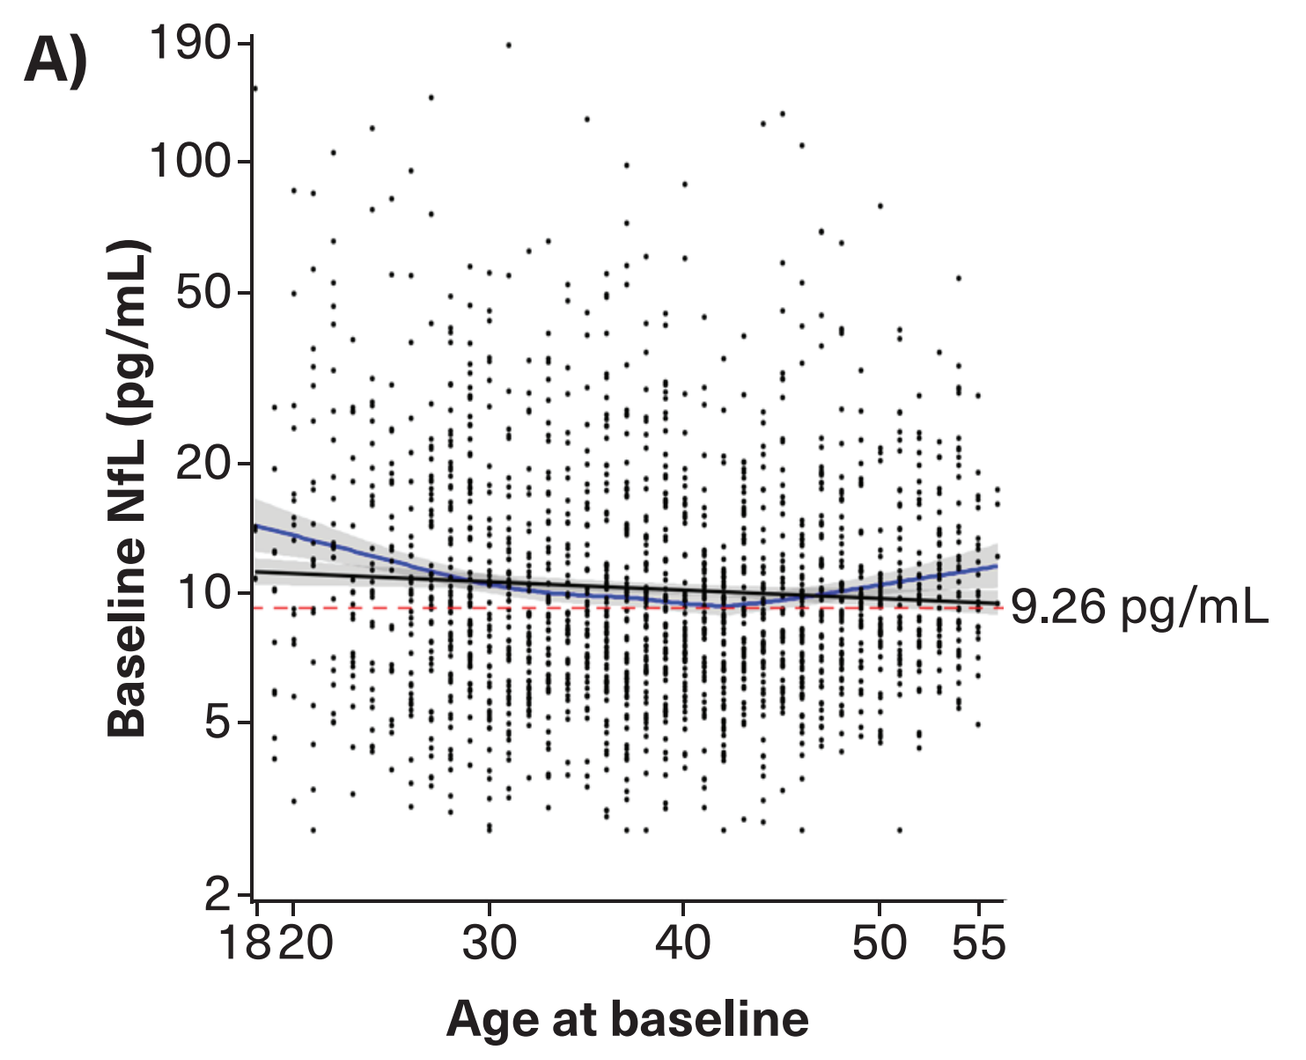
A)
Baseline NfL (pg/mL)
190
100
50
20
10
5
2
9.26 pg/mL
18 20 30 40 50 55
Age at baseline

- Based on data from participants aged 18–55 years in ASCLEPIOS I and II, the variability in sNfL levels associated with age is much lower than the observed overall variability in sNfL (likely due to MS disease activity) in patients with RMS (**Figure 2A**)
- Compared to inflammatory-associated sNfL elevations, the age dependency of sNfL may not be a relevant confounder in clinical practice in younger RMS patients without comorbidities[7]

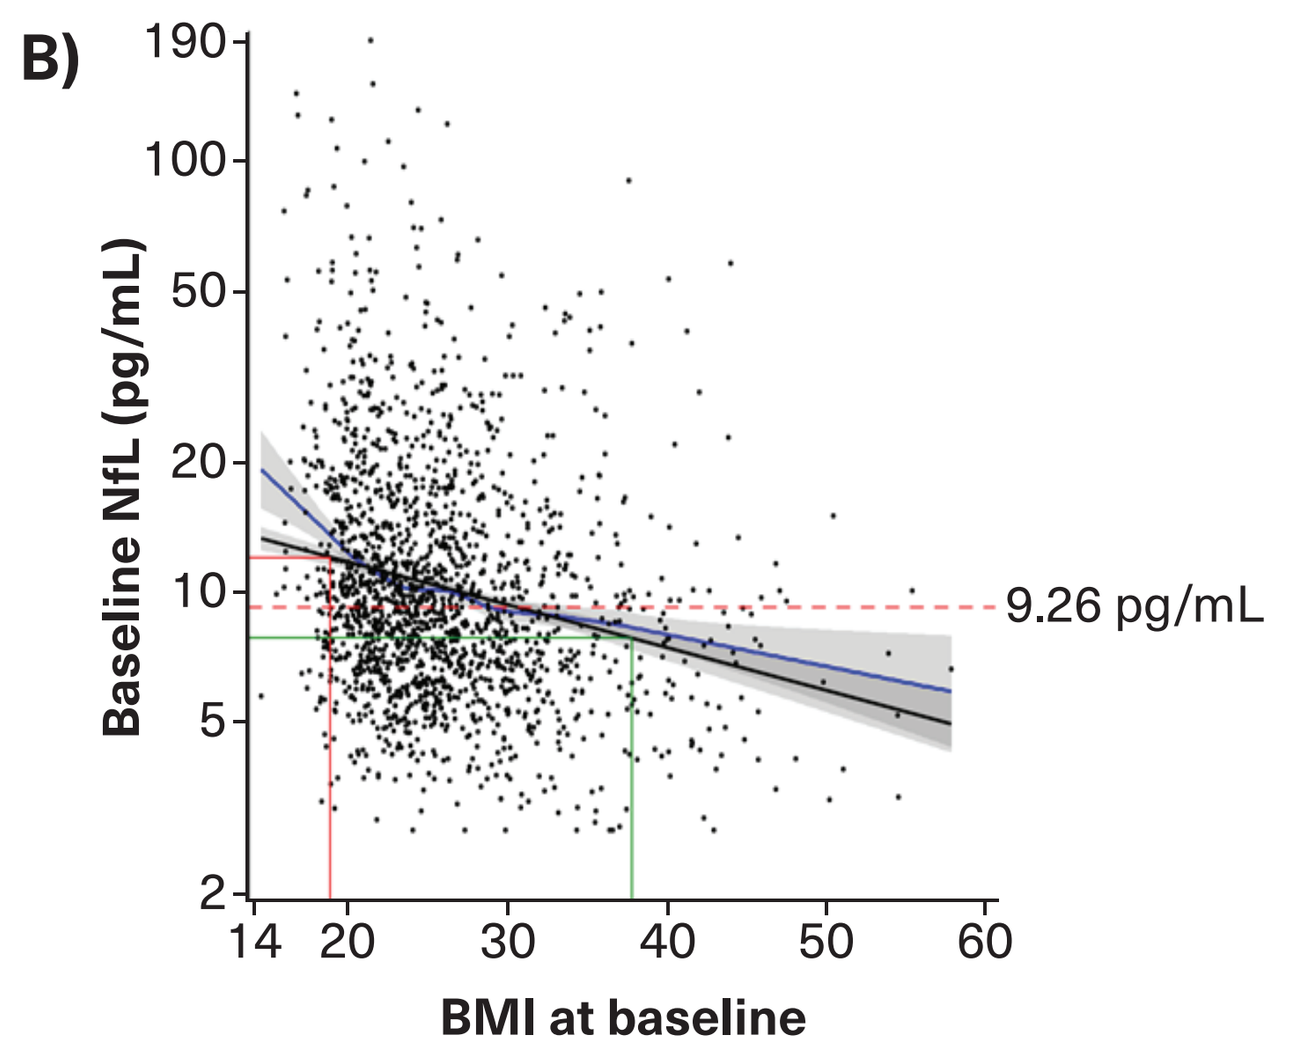
B)
Baseline NfL (pg/mL)
190
100
50
20
10
5
2
9.26 pg/mL
14 20 30 40 50 60
BMI at baseline

- In **Figure 2B**, based on data from participants aged 18–55 years in ASCLEPIOS I and II, there is an association between BMI and sNfL levels, mainly driven by extreme values of BMI
 - Underweight individuals (BMI<18.5) were associated with higher mean sNfL levels, while extremely obese individuals (BMI>40) were associated with lower mean sNfL levels
 - However, the magnitude of individual variability in sNfL driven by MS disease activity by far exceeded the magnitude of change explained by BMI (mean NfL ranges from 7.9 to 12.1 pg/mL, i.e., 4.2 pg/mL difference within the inner 90% of the BMI range)
- Thus, for most RMS patients in these studies, sNFL ≥9.3 pg/mL was prognostic of future MS disease activity irrespective of BMI (**Figure 1B**)

Figure 2A (age, left side): Black line is from linear regression of log (baseline sNfL) on age; blue line is when applying a non-linear relationship; dashed red line indicates median baseline sNfL; Figure 2B (BMI, right side): Black line is from linear regression of log (baseline sNfL) on BMI; blue line is when applying a non-linear relationship; dashed red line indicates median baseline sNfL. The vertical and horizontal red lines indicate the 5th percentile of BMI (18.78) and the corresponding mean sNfL (12.10 pg/mL). The vertical and horizontal green lines indicate the 95th percentile of BMI (37.74) and the corresponding mean sNfL (7.86 pg/mL). BMI, body mass index; MS, multiple sclerosis; RMS, relapsing multiple sclerosis; sNfL, serum neurofilament light chain.

LIMITATIONS

- Based on the pre-planned nature of the analysis, participants were stratified by baseline median sNfL value into "high" or "low" with the intention to divide a phase 3 trial RMS population into groups of equal size with higher versus lower than median sNfL
- The use of a single sNfL threshold may be mostly applicable to relatively young RMS populations (18–55 years) such as the population included in these trials; for understanding sNfL changes outside of this age range, the use of sNfL as a continuous variable may be preferable
- While results support the applicability of sNfL for most patients across the BMI range, in those with extreme BMIs (<18.5 and >40.0), prognostication using a single sNfL value should take into account the effect of BMI on sNfL
- Results reported here are based on the protocol-defined single sNfL threshold; future work should evaluate how sNfL as a prognostic factor could be optimized with a specific target and population in mind
- The data presented in this study are based on a population that was selected according to the ASCLEPIOS inclusion/exclusion criteria to represent a population suitable for phase 3 trials/regulatory purposes, and it may not reflect the broader population of individuals with RMS seen in everyday clinical practice
- Recent findings suggest that sNfL has comparable prognostic value to Gd-enhancing lesions on MRI scans for future lesion formation[11]. The relative prognostic value of sNfL and baseline MRI characteristics should be further confirmed

References

1. Oh J, et al. *Curr Opin Neurol.* 2018;31:752–759.
2. Dahlke F, et al. *Mult Scler.* 2021;27:2062–2076.
3. Thebault S, et al. *Mult Scler.* 2022;28:1491–1497.
4. Ziemssen T, et al. *Front Immunol.* 2022;13:852563.
5. Hauser SL, et al. *N Engl J Med.* 2020; 383:546–557.
6. Consortium of Multiple Sclerosis Centers. *Int J MS Care.* 2021;23:1–36.
7. Bittner S, et al. *Brain.* 2021;144:2954–2963.
8. Bar-Or A, et al. *EBioMedicine.* 2023;93:104462.
9. Kapoor R, et al. *Neurol.* 2020;95:436–444.
10. Barro C, et al. *Ann Clin Transl Neurol.* 2020;7:2508–2523.
11. Bar-Or A, et al. *Neurol Ther.* 2023 Feb;12(1):303–317.

Abbreviations

BMI, body mass index; **DMT**, disease-modifying therapy; **EDSS**, Expanded Disability Status Scale; **Gd+**, gadolinium-enhancing; **MRI**, magnetic resonance imaging; **MS**, multiple sclerosis; **neT2**, new or enlarging T2; **pwRMS**, people with relapsing multiple sclerosis; **Q**, quartile; **RMS**, relapsing multiple sclerosis; **sNfL**, serum neurofilament light chain.

Disclosures

Anne H. Cross received personal compensation from Biogen, Bristol Myers Squibb, EMD Serono, Genentech/Roche, Horizon, Novartis, Octave, and TG Therapeutics. **Tjalf Ziemssen** has received personal compensation for participating in advisory boards, trial steering committees, and data and safety monitoring committees, as well as for scientific talks and project support from Almirall, Bayer, BAT, Biogen, Celgene, Sanofi Genzyme, Merck, Novartis, Roche, Vitaccess, and Teva. **Douglas L. Arnold** reports personal fees from Biogen, Biohaven, BMS, Eli Lilly, EMD Serono, Find Therapeutics, Frequency Therapeutics, Gossamer Bio, GSK, Kiniksa, Merck, Novartis, Sanofi, Shionoggi, outside the submitted work; and ownership interest in NeuroRx. **Eric Thouvenot** has received honoraria, travel grants, or research grants from the following pharmaceutical companies: Actelion, Biogen, Bristol Myers Squibb/Celgene, Merck, Novartis, Roche, Teva. **Scott S. Zamvil** has received consulting honoraria from Alexion, Biogen-Idec, EMD-Serono, Genzyme, Novartis, Roche/Genentech, and Teva Pharmaceuticals, Inc and has served on Data Safety Monitoring Boards for Lilly, BioMS, Teva and Therapeutics. **Enrique Alvarez** received compensation for consulting from Alexion, Biogen, Celgene/BMS, EMD Serono/Merck, Genentech/Roche, Horizon/Amgen, Motric Bio, Novartis, Sanofi, Scionic, and TG Therapeutics, and for research from Atara, Biogen, Genentech/Roche, Novartis, Sanofi, TG Therapeutics, Patient-Centered Outcomes Research Initiative, National Multiple Sclerosis Society, National Institutes of Health, and Rocky Mountain MS Center. **Heinz Wiendl** declares that he has acted as a member of the Scientific Advisory Boards of Alexion, Argenx, Biocryst, Bristol Myers Squibb, Cellerys, Galapagos, Janssen, Merck, Novartis, Sandoz-Hexal, and Uniqure. He also declares that he has received speaker honoraria and travel support from Alexion, Biogen, Bristol Myers Squibb, EPG Health, Genzyme, Merck, Neurodiem, Novartis, Ology, Roche, Teva and WebMD Global and acts as a paid consultant for AbbVie, Actelion, Argenx, Biogen, Bristol Myers Squibb, EMD Serono. He is acting as a paid consultant for Actelion, Argenx, BD, Bristol Myers Squibb, Dianthus, EMD Serono, EPG Health, Fondazione Cariplo, Gossamer Bio, Idorsia, Immunic, Immunovant, Inmune Bio, Syneos Health, Janssen, LTS, Merck, NexGen, Novartis, Roche, Samsung, Sangamo, Sanofi, Swiss Multiple Sclerosis Society, Toleranzia, UCB, Viatris, VirBio, and Worldwide Clinical Trials. His research is funded by Alexion, Amicus Therapeutics, Argenx, Biogen, CSL Behring, F. Hoffmann-La Roche, Genzyme, Merck, Novartis, Roche, and UCB. **Alit Bhatt, Wenjia Wei** and **Ibolya Boer** are employees of Novartis.

Acknowledgments

Medical writing support was provided by Amitha Thakur and Paul Coyle (employees of Novartis Healthcare Pvt. Ltd., Hyderabad, India, and Novartis Ireland Ltd., Dublin, Ireland, respectively) and design support by Hareesh Cheela (employee of Novartis Healthcare Pvt. Ltd., Hyderabd, India). The final responsibility for content lies with the authors.