Poster P272

Adnan Subei, asubei@neurologydallas.com

Immune response to influenza vaccine in patients with relapsing multiple sclerosis treated with ofatumumab: results from an open-label, multicenter, phase 4 study

Brian Steingo,[1] Adnan Subei,[2] Emily Riser,[3] Jeffrey Gitt,[4] James Stankiewicz,[5] Rebecca Piccolo,[5] Kerri Wyse,[5] Bianca Weinstock-Guttman[6]

[1]First Choice Neurology, Boca Raton, FL, USA; [2]Memorial Healthcare System, Hollywood, FL, USA; [3]University of Alabama Birmingham School of Medicine, Birmingham, AL, USA; [4]Center for Neurology & Spine, Phoenix, AZ, USA; [5]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [6]The State University of New York, University of Buffalo, Jacobs School of Medicine and Biomedical Sciences, Buffalo, NY, USA

Scan to obtain:
- Poster

https://www.medicalcongressposters.com/Default.aspx?doc=feda5
Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

KEY FINDINGS & CONCLUSIONS

- Ofatumumab (OMB)-treated patients with relapsing multiple sclerosis (RMS) are able to mount an immune response following inactivated influenza vaccination
- These results are consistent with current European and US product labeling guidelines, which recommend inactivated vaccines be administered 2 weeks prior to starting OMB
- T-cell immunologic responses were not obtained in this study

This study is sponsored by Novartis Pharmaceuticals Corporation

Poster presented at the 9th Annual Americas Committee for Treatment and Research in Multiple Sclerosis (ACTRIMS) Forum • February 29-March 2, 2024 • West Palm Beach, FL, USA

INTRODUCTION

- Ofatumumab (OMB) is a fully human anti-CD20 monoclonal antibody administered by monthly subcutaneous injection (20 mg in 0.4 mL)[1,2]
- OMB is approved for the treatment of adults with relapsing multiple sclerosis (RMS) in the European Union and the United States[1,2]
- Vaccinations comprise an important component of MS management, and there is a need for data regarding whether treatment with OMB impacts the humoral immune response to vaccines, including the influenza vaccine, in patients with RMS
- The objective of this prospective phase 4 study (NCT04667117) was to assess whether patients with RMS treated with OMB 20 mg every 4 weeks could mount a humoral immune response to the 2020-2021, 2021-2022, or 2022-2023 inactivated influenza vaccine compared with patients treated with interferon or glatiramer acetate (IFN/GA)

METHODS

Study Design

- A 3-cohort, open-label, multicenter, prospective, phase 4 study (NCT04667117; **Figure 1**)
 - Cohort 1: vaccinated ≥2 weeks before starting OMB
 - Cohort 2: vaccinated ≥4 weeks after starting OMB
 - Cohort 3: vaccinated ≥4 weeks after enrollment and currently being treated with IFN/GA
- Cohort 1 received OMB 2, 3, and 4 weeks after vaccination, followed by monthly doses from Week 6; Cohorts 2 and 3 continued OMB or other disease-modifying therapy (DMT) per their current dosing schedule
- Patients in all cohorts underwent humoral immunity titer evaluations before vaccination (baseline) and 4 weeks (Week 4) post vaccination
- Patients in Cohorts 1 and 2 could continue receiving monthly doses of OMB during an optional extension period
- Patients with recent major infections or who had been or were being treated with certain immunosuppressive or immunomodulatory therapies were excluded from the trial
- Only patients who had pre- and post-vaccination antibody titers (observed cases) were included in this analysis

Study Endpoints

- **Primary endpoint:**
 - Patients achieving seroprotection to influenza at Week 4 (defined as a post-vaccination antibody titer ≥40)
- **Secondary endpoints:**
 - Achieving seroconversion (defined as post-vaccination humoral immunity titers ≥4-fold increase or ≥40 in those with pre-vaccination titers ≥10 or <10, respectively)
 - Safety (any adverse events [AEs], serious AEs [SAEs], and AEs leading to discontinuation)

Figure 1. Study Design

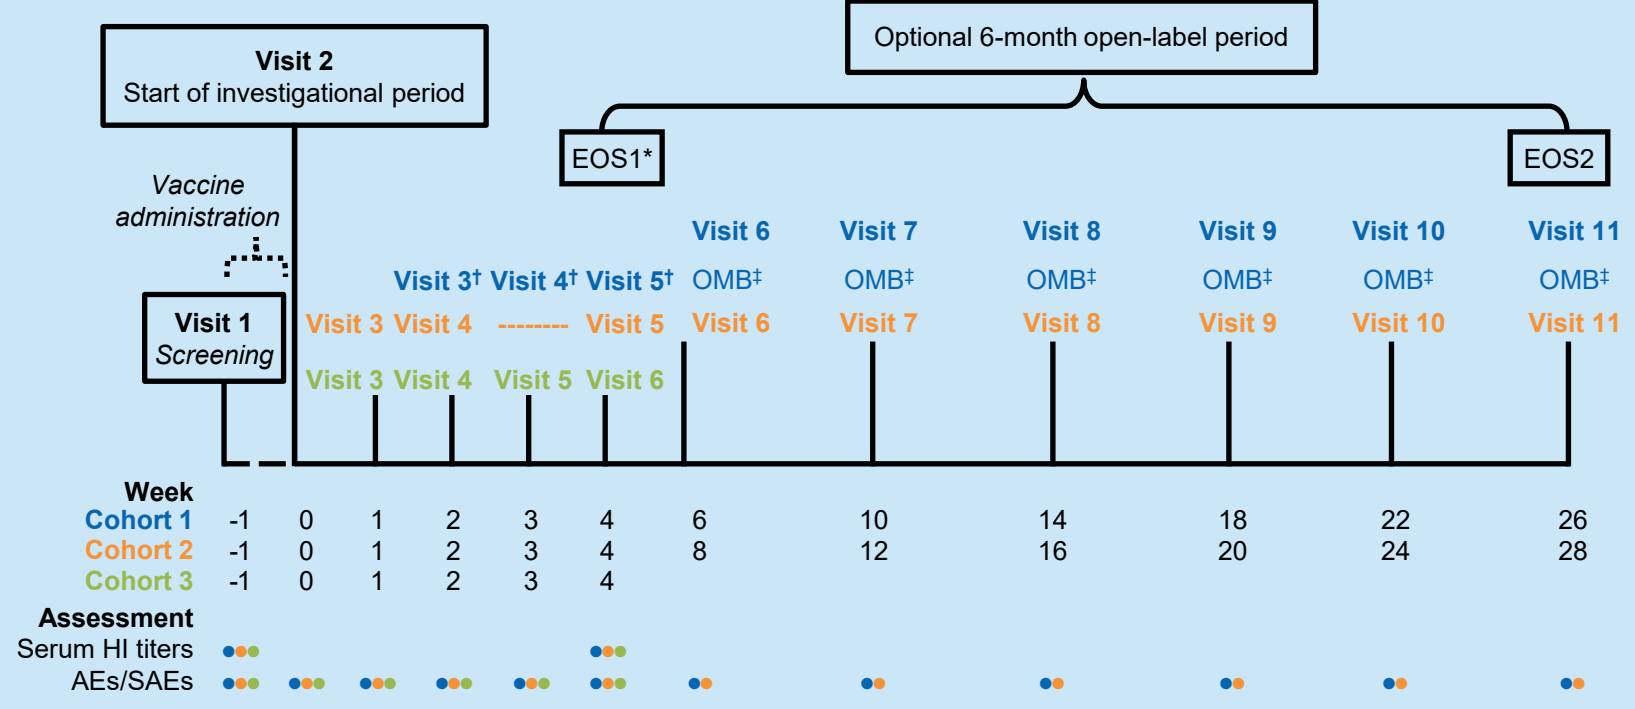
Visit 2
Start of investigational period
Optional 6-month open-label period
EOS1*
EOS2
Vaccine administration
Visit 1 Screening
Visit 3† Visit 4† Visit 5†
Visit 6 Visit 7 Visit 8 Visit 9 Visit 10 Visit 11
OMB‡ OMB‡ OMB‡ OMB‡ OMB‡ OMB‡
Visit 3 Visit 4 -------- Visit 5 Visit 6 Visit 7 Visit 8 Visit 9 Visit 10 Visit 11
Visit 3 Visit 4 Visit 5 Visit 6
Week
Cohort 1 -1 0 1 2 3 4 6 10 14 18 22 26
Cohort 2 -1 0 1 2 3 4 8 12 16 20 24 28
Cohort 3 -1 0 1 2 3 4
Assessment
Serum HI titers
AEs/SAEs

AE, adverse event; EOS, end of study; HI, hemagglutination inhibition; OMB, ofatumumab; SAE, serious adverse event
*Participants in Cohort 3 will not enter the extension; †Weekly OMB loading dose; ‡Monthly OMB dose

RESULTS

Patients

- This study included a total of 63 patients with a mean (range) age of 41 (22-54) years
- The majority of patients were female (76%) and White (86%), and 67% had used a previous DMT for MS treatment (**Table 1**)
- The mean (standard deviation) duration of exposure to the study treatment was 15 (1.5) days for Cohort 1, 320 (192.4) days for Cohort 2, and 2519 (1720.5) days for Cohort 3

Table 1. Patient Demographics and Clinical Characteristics

Characteristic	Cohort 1 (n=22)	Cohort 2 (n=22)	Cohort 3 (n=19)	Overall (N=63)
Age, years				
Mean (range)	41.4 (24-53)	38.9 (22-54)	43.3 (27-53)	41.1 (22-54)
Sex, n (%)				
Female	19 (86.4)	15 (68.2)	14 (73.7)	48 (76.2)
Male	3 (13.6)	7 (31.8)	5 (26.3)	15 (23.8)
Race, n (%)				
White	21 (95.5)	18.8 (81.8)	15 (78.9)	54 (85.7)
Black or African American	1 (4.5)	4 (18.2)	4 (21.1)	9 (14.3)
Ethnicity, n (%)				
Hispanic or Latino	6 (27.3)	4 (18.2)	0	10 (15.9)
Not Hispanic or Latino	16 (72.7)	18 (81.8)	19 (100.0)	53 (84.1)
Any previous MS DMT, n (%)	1 (4.5)	22 (100.0)	19 (100.0)	42 (66.7)
Glatiramer acetate	1 (4.5)	0	12 (63.2)	13 (20.6)
Interferon beta-1a	0	0	4 (21.1)	4 (6.3)
Interferon beta-1b	0	0	1 (5.3)	1 (1.6)
Ofatumumab	0	22 (100.0)	0	22 (34.9)
Peginterferon beta-1a	0	0	2 (10.5)	2 (3.2)

DMT, disease-modifying therapy; MS, multiple sclerosis

Seroprotection at Week 4

- At Week 4, seroprotection was generally high across strains and consistent between Cohorts (**Table 2**). Lower seroprotection rates were observed for all Influenza B strains and for Influenza A Wisconsin

Table 2. Percentages of Patients With Seroprotection at Week 4 by Influenza Strain

Strain	% of Patients With Seroprotection at Week 4		
	Cohort 1 (n=22)	Cohort 2 (n=22)	Cohort 3 (n=19)
Influenza A Brisbane	100% (5/5)	100% (2/2)	100% (1/1)
Influenza A Cambodia	100% (13/13)	80% (4/5)	85.7% (6/7)
Influenza A Kansas	100% (5/5)	100% (2/2)	100% (1/1)
Influenza A Michigan	100% (5/5)	100% (2/2)	100% (1/1)
Influenza A Singapore	100% (5/5)	100% (2/2)	100% (1/1)
Influenza A Victoria	100% (13/13)	100% (5/5)	100% (7/7)
Influenza A Wisconsin	61.5% (8/13)	40% (8/20)	68.8% (11/16)
Influenza B Colorado	60% (3/5)	50% (1/2)	100% (1/1)
Influenza B Phuket	77.8% (14/18)	68.2% (15/22)	76.5% (13/17)
Influenza B Washington	76.9% (10/13)	20% (1/5)	71.4% (5/7)

- The most commonly assessed strains (Wisconsin and Phuket) showed comparability of seroprotection between Cohorts 1 and 3 with a somewhat reduced response in Cohort 2. Of the next most tested groups (Cambodia, Victoria, and Washington), Washington also showed similarities between Cohorts 1 and 3, with a reduced response in Cohort 2, whereas Cambodia showed a diminished response in both Cohorts 2 and 3 compared with Cohort 1. Response to Victoria was uniformly robust
- The seroprotection rates presented here are similar to those previously reported for IFN/GA and ocrelizumab, another anti-CD20 DMT[3,4]

Seroconversion at Week 4

- The rates of seroconversion at Week 4 were variable across vaccine influenza strains (**Table 3**); however, trends for lower rates of seroconversion in Cohort 2 vs Cohorts 1 and 3 were broadly consistent across individual influenza A and B strains

Table 3. Percentages of Patients Achieving Seroconversion at Week 4 by Influenza Strain

Strain	% of Patients Achieving Seroconversion		
	Cohort 1 (n=22)	Cohort 2 (n=22)	Cohort 3 (n=19)
Influenza A Brisbane	80% (4/5)	0% (0/2)	0% (0/1)
Influenza A Cambodia	84.6% (11/13)	20% (1/5)	42.9% (3/7)
Influenza A Kansas	40% (2/5)	0% (0/2)	0% (0/1)
Influenza A Michigan	60% (3/5)	0% (0/2)	100% (1/1)
Influenza A Singapore	40% (2/5)	0% (0/2)	0% (0/1)
Influenza A Victoria	92.3% (12/13)	20% (1/5)	42.9% (3/7)
Influenza A Wisconsin	46.2% (6/13)	10% (2/20)	37.5% (6/16)
Influenza B Colorado	60% (3/5)	0% (0/2)	100% (1/1)
Influenza B Phuket	50% (9/18)	18.2% (4/22)	41.2% (7/17)
Influenza B Washington	38.5% (5/13)	0% (0/5)	42.9% (3/7)

- The seroconversion rates presented here are similar to those previously reported for IFN/GA and ocrelizumab, another anti-CD20 DMT[3,4]

Safety

- Overall, 38.1% (24/63) of patients experienced ≥1 AE (**Table 4**)
- AEs were most frequent in Cohort 1, where 72.7% (16/22) of patients experienced an AE vs 27.3% (6/22) in Cohort 2 and 10.5% (2/19) in Cohort 3
- The higher frequency of AEs in Cohort 1 is likely related to ofatumumab initiation (ie, IRRs)
- 1 SAE (MS pseudo relapse) was reported in Cohort 2
- No AEs resulting in discontinuation were reported

Table 4. AEs That Occurred in ≥4% of Patients and SAEs

AE, n (%)	Cohort 1 (n=22)	Cohort 2 (n=22)	Cohort 3 (n=19)	Overall (N=63)
Any AE	16 (72.7)	6 (27.3)	2 (10.5)	24 (38.1)
IRR	4 (18.2)	0	0	4 (6.3)
Headache	3 (13.6)	1 (4.5)	0	4 (6.3)
Nausea	3 (13.6)	0	0	3 (4.8)
COVID-19	2 (9.1)	0	1 (5.3)	3 (4.8)
Influenza-like illness	2 (9.1)	1 (4.5)	0	3 (4.8)
Pain	3 (13.6)	0	0	3 (4.8)
Pyrexia	2 (9.1)	0	1 (5.3)	3 (4.8)
SAE	0	1 (4.5)	0	1 (1.6)
MS pseudo relapse	0	1 (4.5)	0	1 (1.6)

AE, adverse event; COVID-19, coronavirus disease 2019; IRR, injection-related reaction; MS, multiple sclerosis; SAE, serious adverse event
IRRs were coded to the preferred term of "injection-related reaction" if specified by the investigator as related to the injection; all IRRs may not have been captured as IRRs

Limitations

- This study was conducted over multiple flu seasons, with participants receiving several different vaccines
- Previous exposure to vaccine strains may influence seroconversion rates, with greater responses seen with novel strains than in strains included in the vaccine in more than 1 consecutive year[5]
- The maximum age for this study was 55 years; therefore, the impact of immunosenescence in older patients with MS was not examined
- This study only examines the humoral immune response to the vaccine; therefore, we cannot draw conclusions regarding cell-mediated immunity. However, studies investigating response to coronavirus disease 2019 (COVID-19) mRNA vaccines in patients treated with OMB showed that, although antibody response was diminished, 100% of patients vaccinated for COVID-19 while receiving OMB treatment had severe acute respiratory syndrome coronavirus 2–specific T cells[6]

Acknowledgements

The study was supported by Novartis Pharmaceuticals Corporation. Medical writing support was provided by Frankie Sorrell, PhD, of Envision Pharma, Inc. and was funded by Novartis Pharmaceuticals Corporation. This poster was developed in accordance with Good Publication Practice (GPP 2022) guidelines. Authors had full control of the content and made the final decision on all aspects of this poster.

Disclosures

Brian Steingo has received honoraria and payments for research, speaking engagements, and ad boards from Alexion, Biogen, Bristol Myers Squibb, EMD Serono, Genentech, Janssen, Novartis Pharmaceuticals Corporation, and Sanofi. **Adnan Subei** has received consulting fees from Biogen; has received research support from Novartis; and has attended speaker bureaus for Bristol Myers Squibb and TG Therapeutics. **Emily Riser** has received research support from Alexion, Bristol Myers Squibb, EMD Serono, Mapi, Novartis, Roche, and TG Therapeutics; and has received payments for speaking engagements from Alexion, EMD Serono, and TG Therapeutics. **Jeffrey Gitt** has received honoraria for promotional programs from Biogen. **James Stankiewicz** is an employee and stockholder of Novartis Pharmaceuticals Corporation. **Rebecca Piccolo** is an employee and stockholder of Novartis Pharmaceuticals Corporation. **Kerri Wyse** is an employee and stockholder of Novartis Pharmaceuticals Corporation. **Bianca Weinstock-Guttman** has received consulting fees from Biogen, Celgene, EMD Serono, Genentech, and Janssen; and has received research support from Biogen, Celgene, EMD Serono, Genentech, and Novartis Pharmaceuticals Corporation

References

1. Novartis Pharmaceuticals Corporation. Prescribing information. Kesimpta® 2022. Accessed January 5, 2024. https://www.novartis.com/us-en/sites/novartis_us/files/kesimpta.pdf; **2.** European Medicines Agency. Summary of product characteristics. Kesimpta 2021. Accessed January 31, 2024. www.ema.europa.eu/en/documents/product-information/kesimpta-epar-product-information_en.pdf; **3.** Bar-Or A et al. *Neurology*. 2020;95(14):e1999-e2008. **4.** Metze C et al. *CNS Neurosci Ther*. 2019;25(2):245-254. **5.** Snape N et al. *NPJ Vaccines*. 2022;7(1):8. **6.** Ziemssen T et al. *Vaccines (Basel)*. 2022;10(12):2167.