Poster P043

Paul S. Giacomini
paul.giacomini@innodemneurosciences.com

Eye Movement Biomarkers for Classification of Multiple Sclerosis Progression vs Non-Progression in a Cohort of Multiple Sclerosis Patients

Paul S. Giacomini[1,3], Natacha Bastien[2], Patrice Voss[3], François Blanchette[2], Francis Arseneau[3], Shamiza Hussein[2], Rosemberg Ramos[3], Etienne de Villers-Sidani[3]

[1]Neurology and Neurosurgery, Montreal Neurological Institute, Montréal, QC, CANADA; [2]Novartis Pharmaceuticals Canada, Montréal, QC, CANADA; [3]Innodem Neurosciences, Montréal, QC, CANADA

Scan to access full poster

https://bit.ly/actrims-forum

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

KEY FINDINGS & CONCLUSIONS

- The preliminary findings presented here suggest that eye movement biomarkers (EMBs) could potentially serve as novel biomarkers in Multiple Sclerosis (MS) to detect early progression before irreversible brain injury occurs, thereby facilitating treatment optimization and improving patient outcomes.
- In this early analysis, we were able to establish that select eye movement metrics can discriminate between MS patients with progression from those without progression with a relatively high sensitivity and specificity, as early as six months from baseline.
- When completed, this trial will hopefully demonstrate the reliability of mobile oculomotor assessments for the monitoring of MS progression as a non-invasive, accessible, scalable, and sensitive novel digital biomarker of disease progression - both for cognitive and physical disability.

Poster presented at the ACTRIMS Forum 2024, held on February 29 – March 2, 2024, at West Palm Beach, Florida

INTRODUCTION

- Multiple sclerosis (MS) is characterized by subtle progression of disease that is difficult to detect in clinical practice. Existing tools designed to help clinicians monitor progression may not be sensitive enough to detect early changes in disease[1].
- There is increasing evidence linking abnormal eye movements in patients with MS (PwMS) to disease severity and cognition. Indeed, not only are there a growing number of studies documenting the existence of oculomotor anomalies in PwMS[2], but several recent studies have linked these anomalies to brain health via correlations between several eye movement parameters and disease or cognitive status, as measured by tools such as the Expanded Disability Status Scale (EDSS) and the Symbol Digit Modalities Test (SDMT)[3-5]. Translating these discoveries into clinical practice is difficult due the cost-prohibitive nature and limited scalability of the specialized equipment required to capture eye-movements.
- A patented gaze-tracking technology was developed that can reliably and accurately track eye movements with precision without the need for infrared cameras and using only the embedded camera of a tablet computer. Using this novel technology, we have recently replicated sets of well-known oculomotor findings in both individuals with MS[6] and Parkinson's disease[7]. However, whether the information captured via this device can be used to detect disease progression in PwMS remains to be determined.

OBJECTIVE

The objective is to determine if a mobile device tool that automatically extracts and combines multiple eye-movement metrics can detect, with high accuracy, whether PwMS are experiencing disease progression.

METHODS

Study design and subject population:

- PwMS in an ongoing 48-month longitudinal proof-of-concept study are undergoing eye-tracking testing (weekly for the first 12 months; every 6 months thereafter) and clinical assessments every 6 months (including assessment at baseline) (**Figure 1**). The data presented herein were collected from 56 PwMS who completed at least 6 months of follow-up and 27 of whom completed follow-up at 12-months. The study was approved by both the Veritas and the McGill University Health Center (MUHC) research ethics boards (ClinicalTrials.gov Identifier: NCT05061953).
- The main inclusion criteria were adults with a confirmed diagnosis of MS with no signs of progressive increase in physical disability within the past 6 months and sufficient corrected visual acuity to allow for the accurate reading of the on-screen visual task instructions. The main exclusion criteria were the presence of comorbid neurological or psychiatric conditions, the recent start of medications known to influence ocular motor visual function (e.g., benzodiazepines) to avoid eye movement anomaly confounders, and patients who experienced an MS relapse at the time of assessment.

Clinical and cognitive assessments:

- To assess disease and cognitive status, three of the most commonly employed MS assessment tools were used: EDSS, Brief International Cognitive Assessment for MS (BICAMS), and two subtests of the Multiple Sclerosis Functional Composite (MSFC).
- The BICAMS consists of a test of information processing speed (SDMT), as well as tests for verbal (California Verbal Learning Test [CVLT-II]) and visual memory (Brief Visuospatial Memory Test-Revised [BVMT-R]), representing the most frequent cognitive deficits observed in MS. However, we substituted the CVLT-II with the Rey Auditory Verbal Learning Test (RAVLT) due to it having a validated and normed version for French Canadians.
- The MSFC comprises quantitative functional measures of three key clinical dimensions of MS: leg function/ambulation (timed 25-foot walk test [T25-FW]), arm/hand function (9-hole pegboard test (9-HPT)), and cognitive function (the Paced Auditory Serial Addition Task test [PASAT]). However, we only performed the two subtests with a motor component (T25-FW and 9-HPT) and replaced the PASAT score with the SDMT score to reduce testing time.

Oculomotor assessment:

- All tests were performed using a 12.9-inch tablet computer with the patented software for gaze-tracking installed. The software enables simultaneous video recordings of the eyes at 60 frames per second using the embedded front-facing camera and the presentation of visual stimuli on the screen. The software's gaze-tracking algorithms, as estimated from a normative sample of 196 healthy control individuals, have an accuracy of 0.47 degrees and precision of 0.33 degrees.
- All participants performed five oculomotor tasks (a fixation task, a pro-saccade task, an anti-saccade task, a smooth pursuit task, and an optokinetic nystagmus (OKN) task), which were preceded by a calibration step to ensure proper gaze-tracking. All five tasks, as well as the calibration step, were completed in under 15 minutes. All tasks were performed with the tablet screen placed vertically, camera side up, and secured at eye level using a tablet pole mount. Participants were positioned approximately 45 cm from the tablet screen and were allowed to use their best-corrected vision, with glasses or lenses if necessary. Safeguards within the gaze-tracking software ensured the participant's head was properly positioned and visible via the embedded camera, at an acceptable angle and distance from the screen.

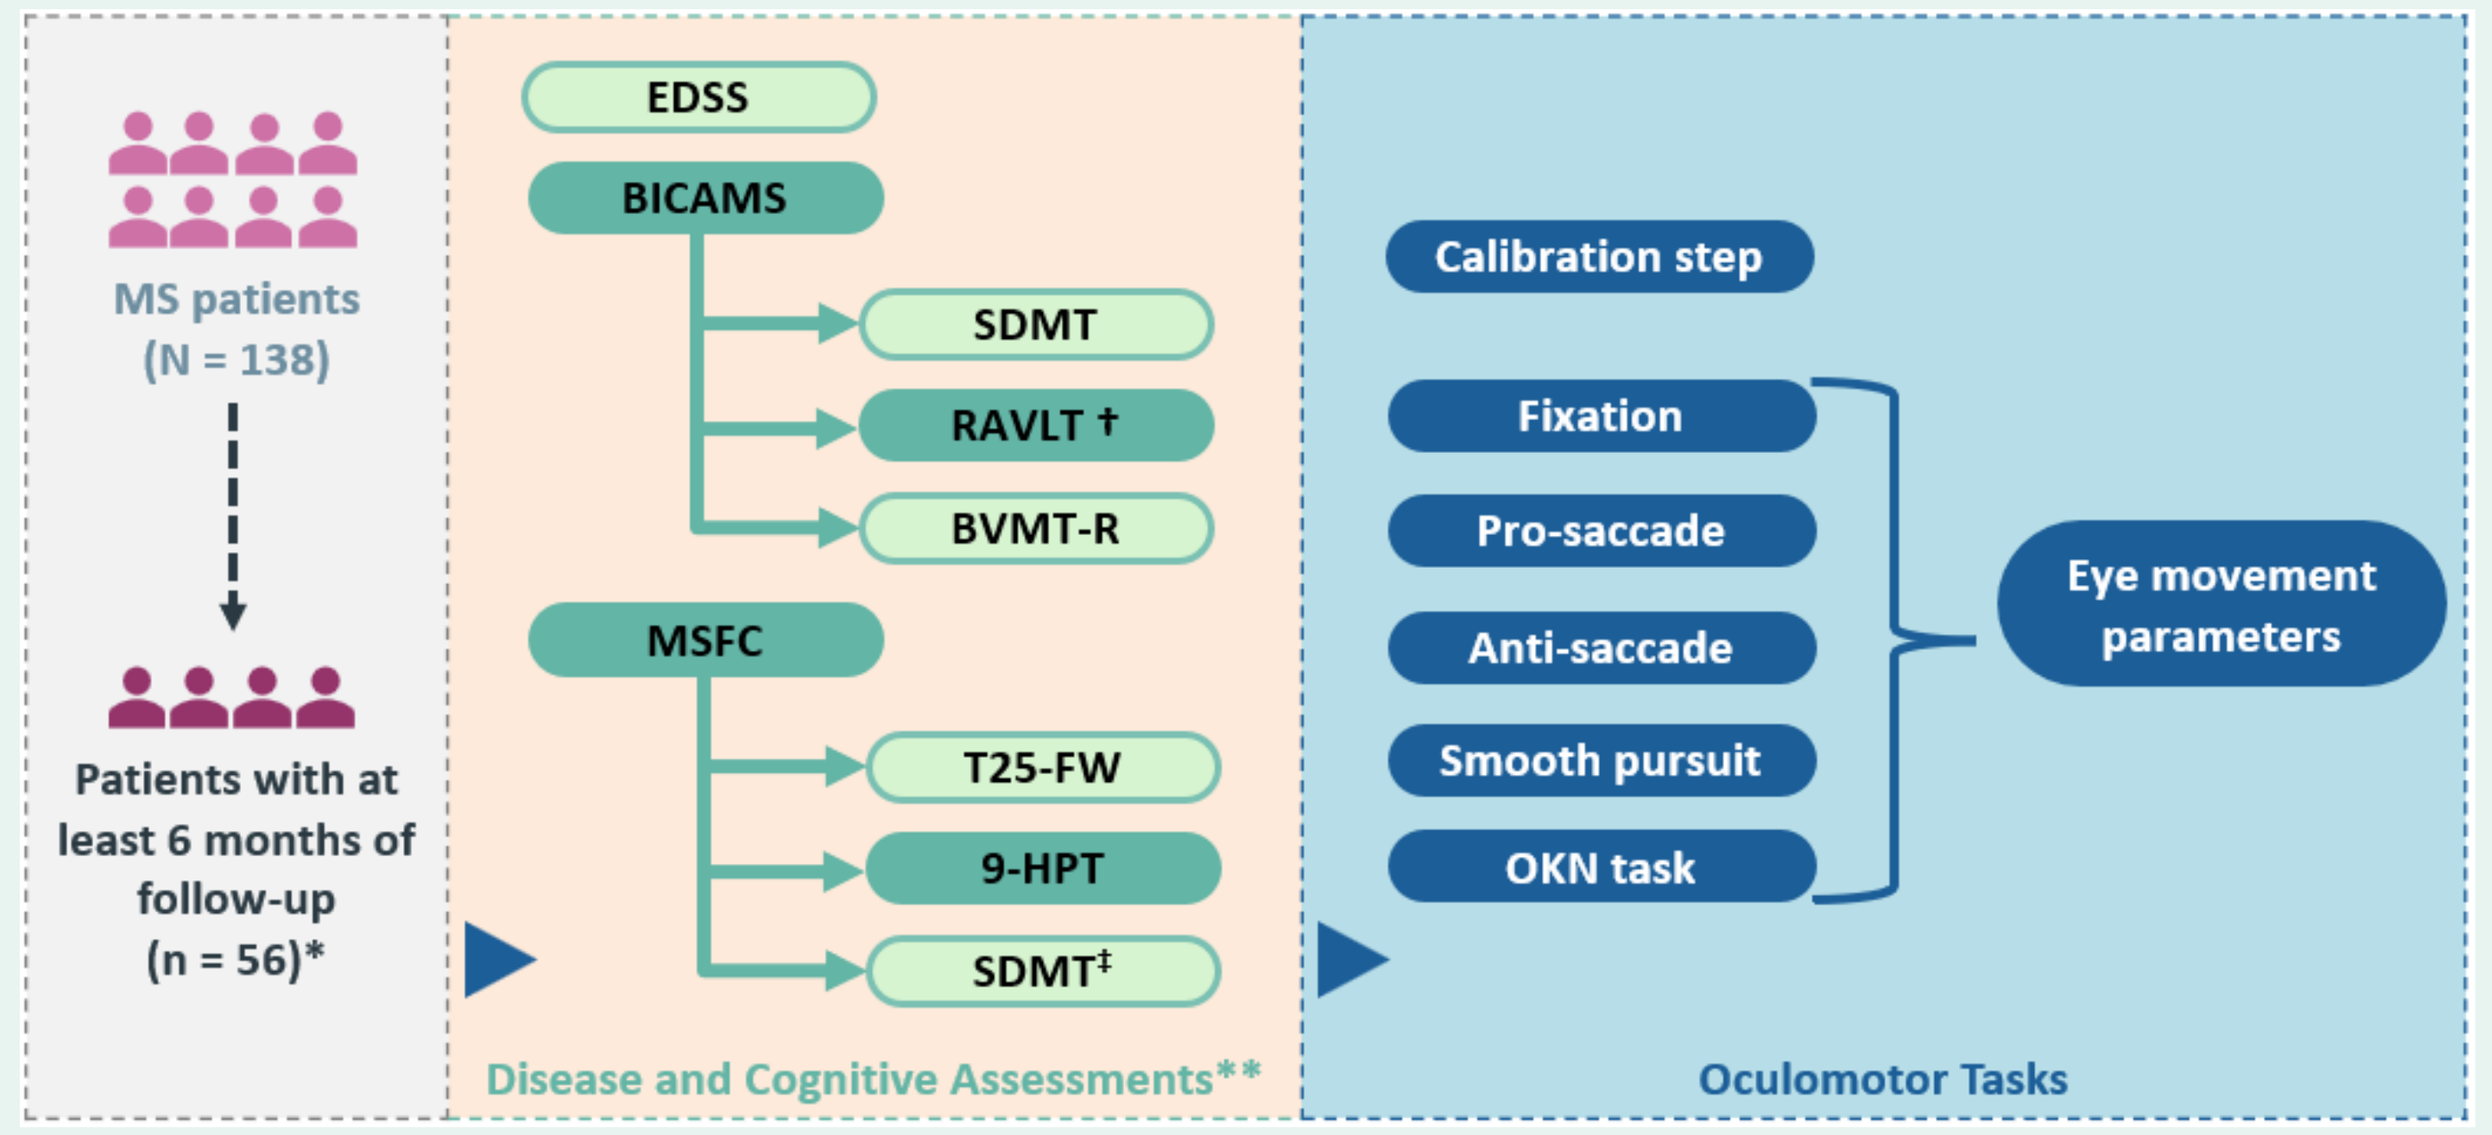

MS patients
(N = 138)
Patients with at least 6 months of follow-up
(n = 56)*
EDSS
BICAMS
SDMT
RAVLT †
BVMT-R
MSFC
T25-FW
9-HPT
SDMT‡
Disease and Cognitive Assessments**
Calibration step
Fixation
Pro-saccade
Anti-saccade
Smooth pursuit
OKN task
Eye movement parameters
Oculomotor Tasks

Figure 1: Study design

EDSS, Expanded Disability Status Scale; BICAMS, Brief International Cognitive Assessment for Multiple Sclerosis; MS, Multiple Sclerosis; MSFC, Multiple Sclerosis Functional Composite; SDMT, Symbol Digit Modalities Test; RAVLT, Rey Auditory Verbal Learning Test; BVMT-R, Brief Visuospatial Memory Test-Revised; T25-FW, timed 25-foot walk; 9-HPT, 9-hole pegboard test.

**This interim analysis included 56 out of 138 Multiple Sclerosis patients with EDSS scores ranging from 0 to 7.5.*

†RAVLT was selected as a surrogate for the California Verbal Learning Test – Second Edition (CVLT-II) due to its availability in French.

‡SDMT was selected as a surrogate for the Paced Auditory Serial Addition Test (PASAT) to reduce testing time.

***Analysis of EDSS, SDMT, T25-FW and BVMT-R (highlighted in light green) were used to identify patients with clinically meaningful change.*

Data Analysis:

- To determine if patients progressed during the course of the study, analysis was performed to identify those that exhibited a clinically meaningful change (CMC+) in at least one of the scores measured: an EDSS increase of 1.0 for scores equal to or below 5.5, or 0.5 for patients with a higher score, a decrease in SDMT score of 4, a 20% increase in the T25-FW time, and a 20% decrease in BVMT-R.
- Subsequently, oculomotor features were examined to determine if any had progression slopes that differed between those that were determined to progress on the basis of clinical data (CMC+) and those that did not (CMC-).
- Finally, in a third step, a support vector classifier (SVC) was built using only the oculomotor features identified in the previous step to see if progressors (CMC+) could be accurately separated from non-progressors (CMC-).

RESULTS

Baseline characteristics:

- Of the 56 patients included, 70% were female and the mean age was 51.9 years (SD ± 9.8) at baseline.
- The mean EDSS at baseline was 3.4 (SD ± 1.9) while the mean baseline SDMT was 50.0 (SD ± 12.8).

Table 1: Baseline Characteristics (n = 56)

Characteristics and Results		Baseline Visit, (n = 56)
Age	Mean (SD)	51.9 (9.8)
	Min-Max	26 – 70
Sex	Female, n (%)	39 (70)
Expanded Disability Status Scale (EDSS)	Mean (SD)	3.4 (1.9)
	Min-Max	1 – 7.5
Symbol Digit Modalities Test (SDMT)	Mean (SD)	50.0 (12.8)
	Min-Max	22 – 80
Brief Visuospatial Memory Test-Revised (BVMT-R)	Mean (SD)	24.2 (6.5)
	Min-Max	8 – 36
Timed 25-Foot Walk (T25-FW)	Mean (SD)	1.4 (0.6)
	Min-Max	0.0 – 2.7

Descriptive statistics are reported as mean, standard deviation (SD); minimum to maximum range for continuous variables; and percentage (%) for categorical variables.

Outcome data:

- We found that 44.6% of the cohort of PwMS (n = 56) had a clinically meaningful change (CMC+) in at least one of the scores measured at 12 months (**Figure 2**).
- In the analysis dataset, 17 distinct eye metrics were identified that had an average slope over time that was significantly different between CMC+ and CMC- patients (see **Figure 3** for two representative oculomotor features).
- An SVC was employed with parameters determined using a 5-fold cross-validated grid search and utilizing as inputs the five eye movement metrics that demonstrated the best model performance. The SVC separated progressors from non-progressors with a sensitivity to detect progression of 78%, a specificity of 68%, and a balanced accuracy of 76% (**Figure 4**). Balanced accuracy was used as a discriminatory measure.

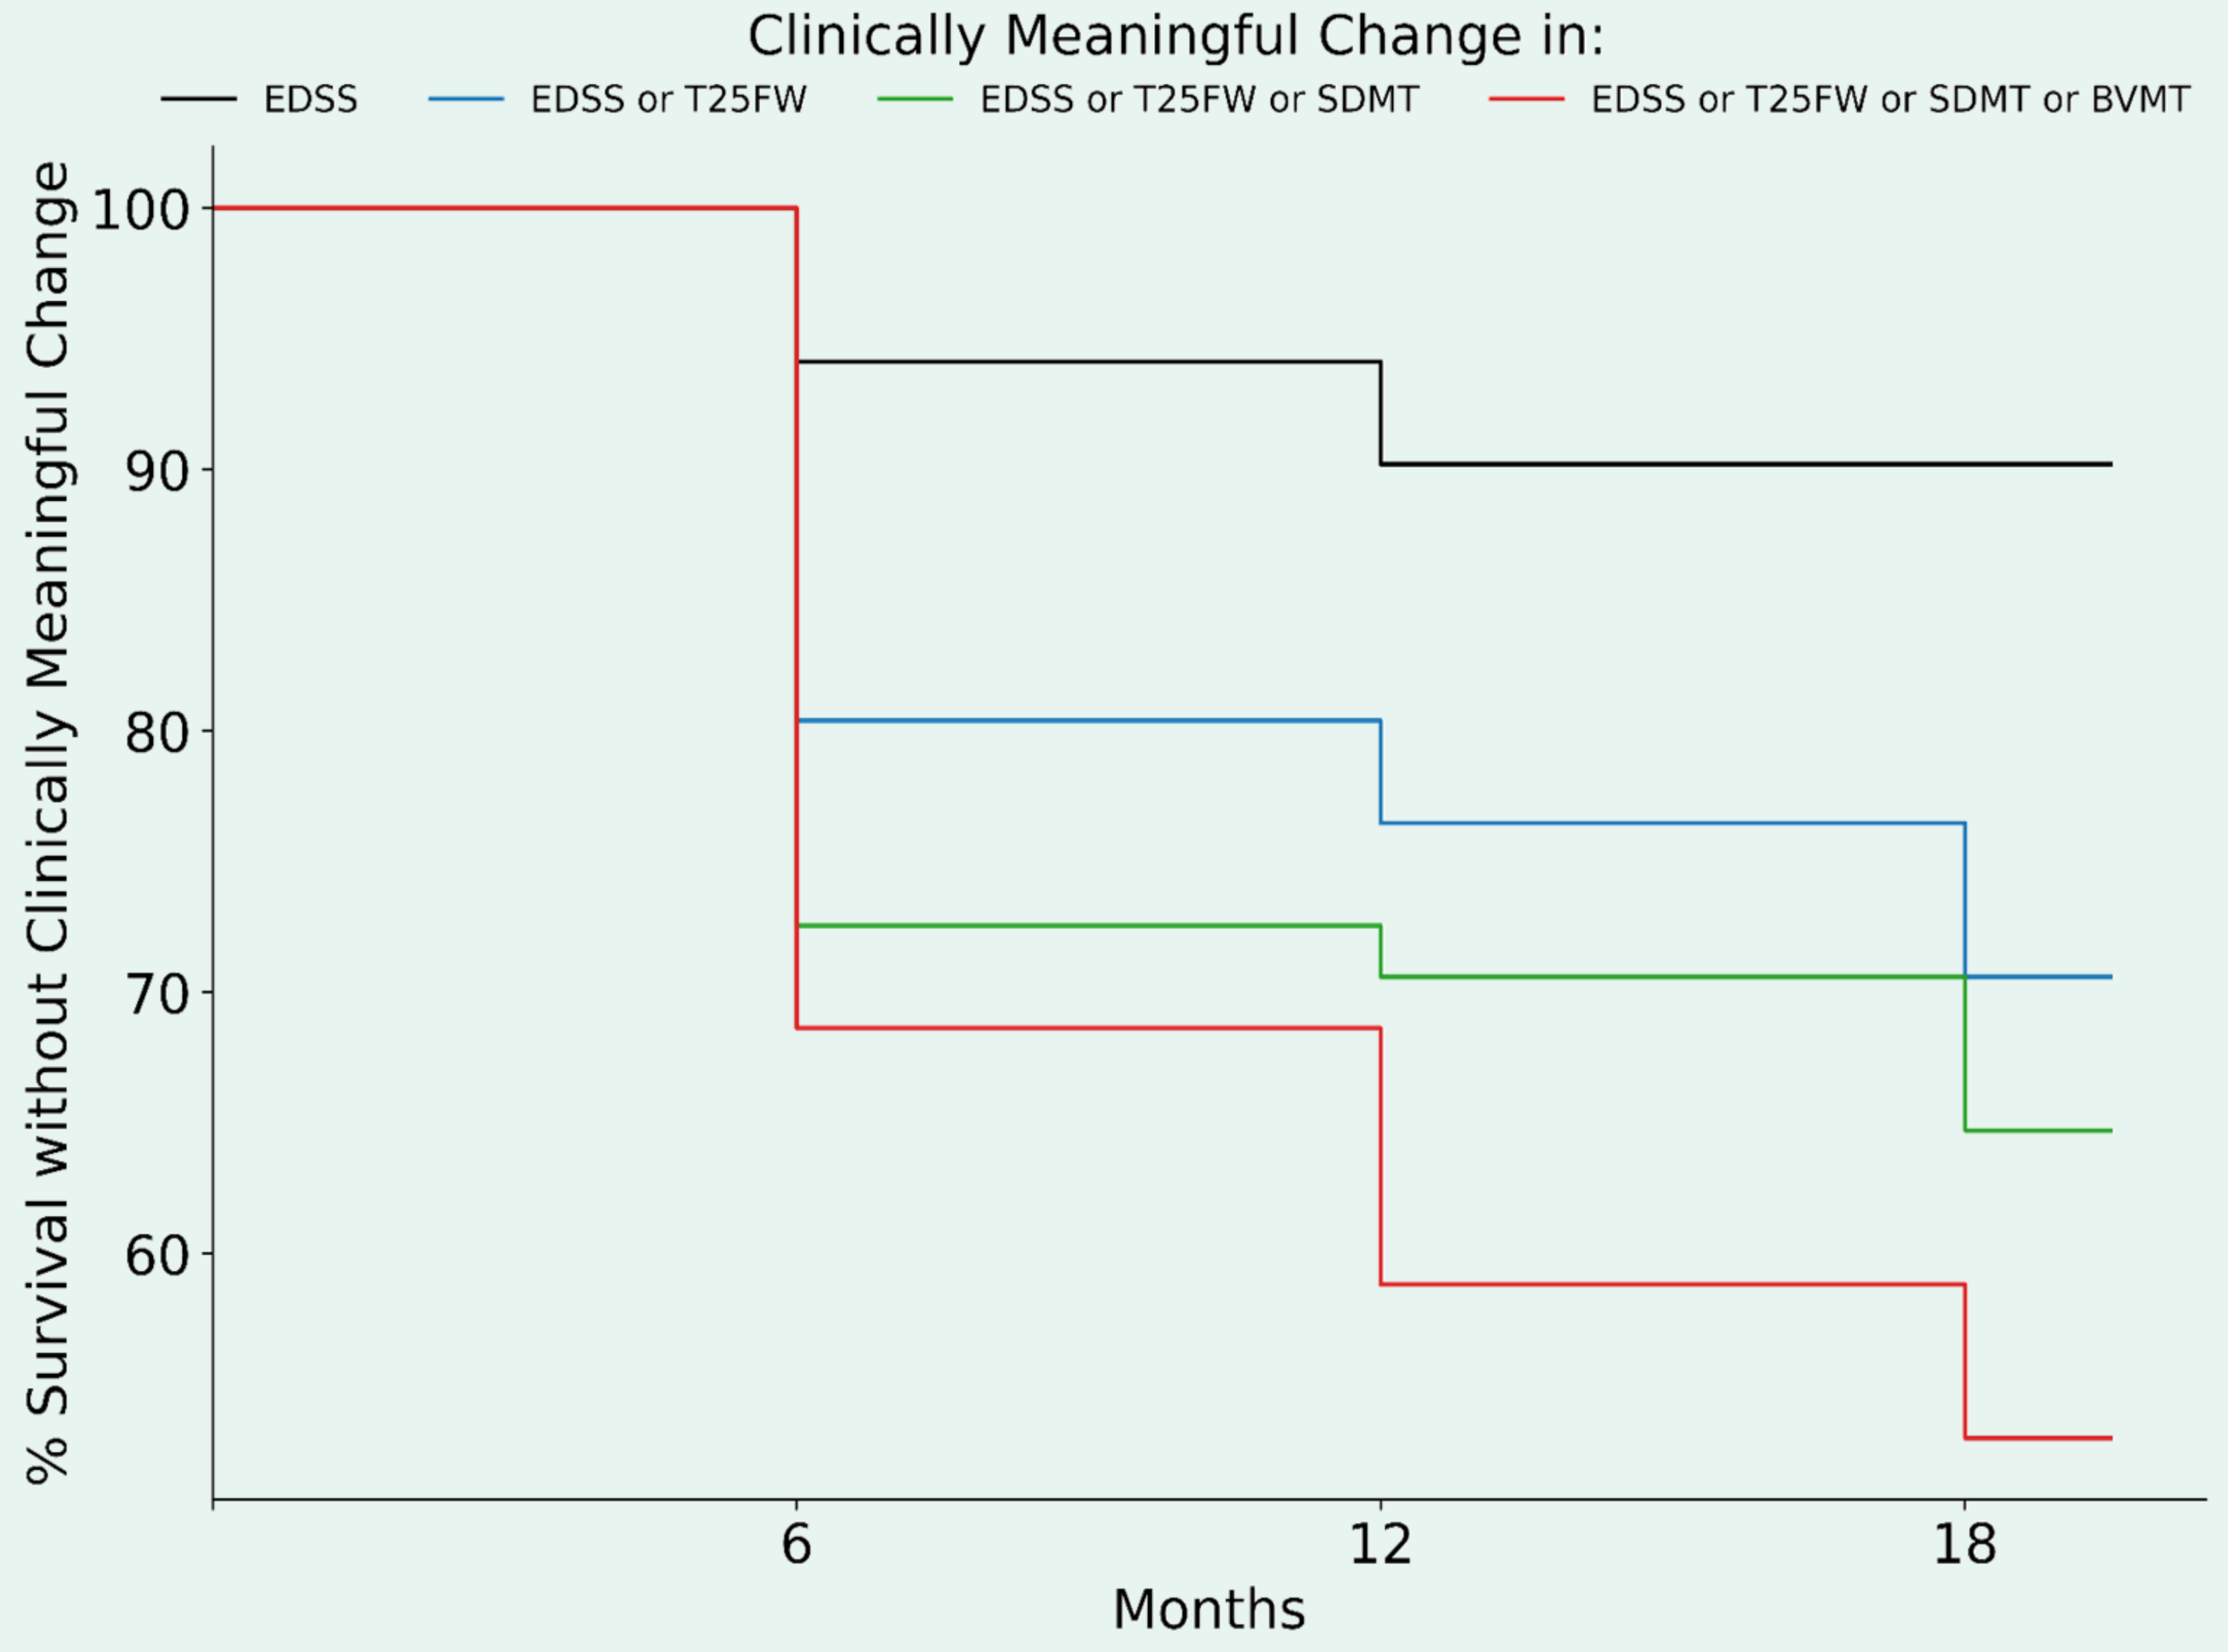

Clinically Meaningful Change in:
EDSS
EDSS or T25FW
EDSS or T25FW or SDMT
EDSS or T25FW or SDMT or BVMT
% Survival without Clinically Meaningful Change
100
90
80
70
60
6
12
18
Months

Figure 2: Kaplan-Meier graphs of time to 6-month confirmed disability worsening on clinical measures
EDSS, Expanded Disability Status Scale; SDMT, Symbol Digit Modalities Test; BVMT-R, Brief Visuospatial Memory Test-Revised; T25FW, timed 25-foot walk.

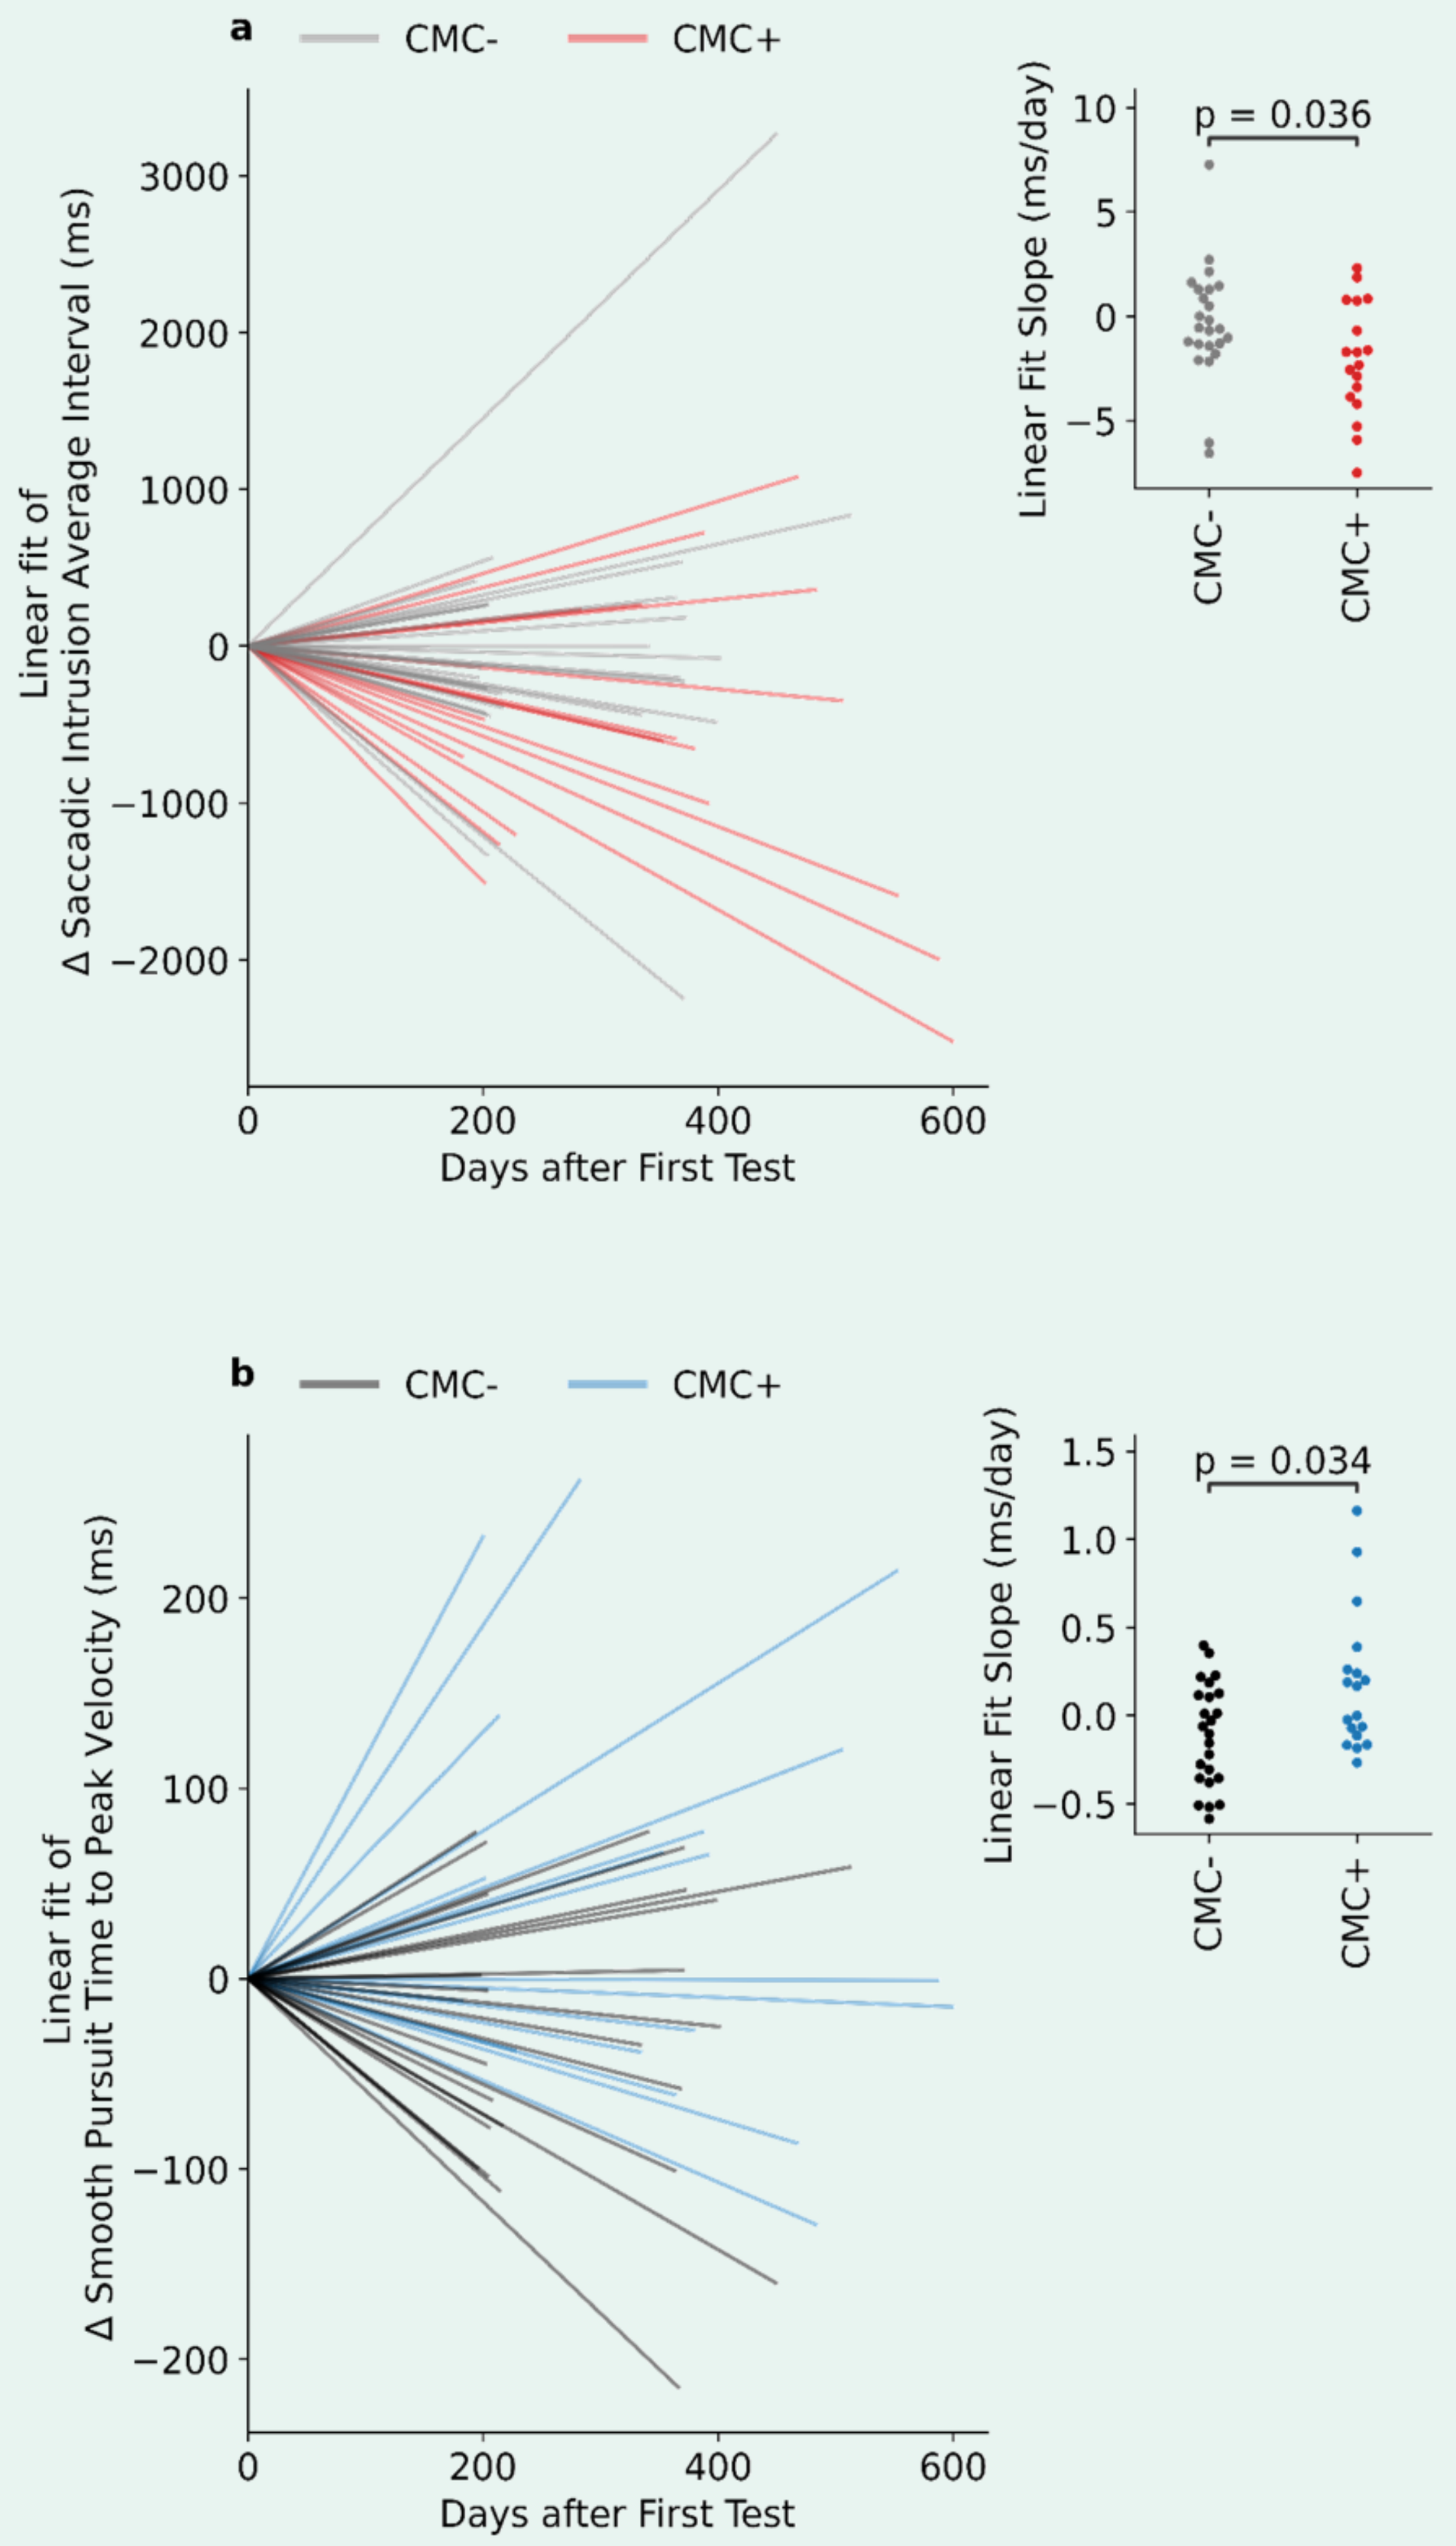

a
CMC-
CMC+
Linear fit of Δ Saccadic Intrusion Average Interval (ms)
Days after First Test
Linear Fit Slope (ms/day)
p = 0.036
b
CMC-
CMC+
Linear fit of Δ Smooth Pursuit Time to Peak Velocity (ms)
Days after First Test
Linear Fit Slope (ms/day)
p = 0.034

Figure 3: Plots illustrating and comparing progressor (clinically meaningful change, CMC+) and non-progressor (CMC-) patient slopes measured for two individual oculomotor features: (a) average time interval between saccadic intrusions during fixation and (b) average time elapsed between onset of smooth pursuit movement and peak velocity attainment.

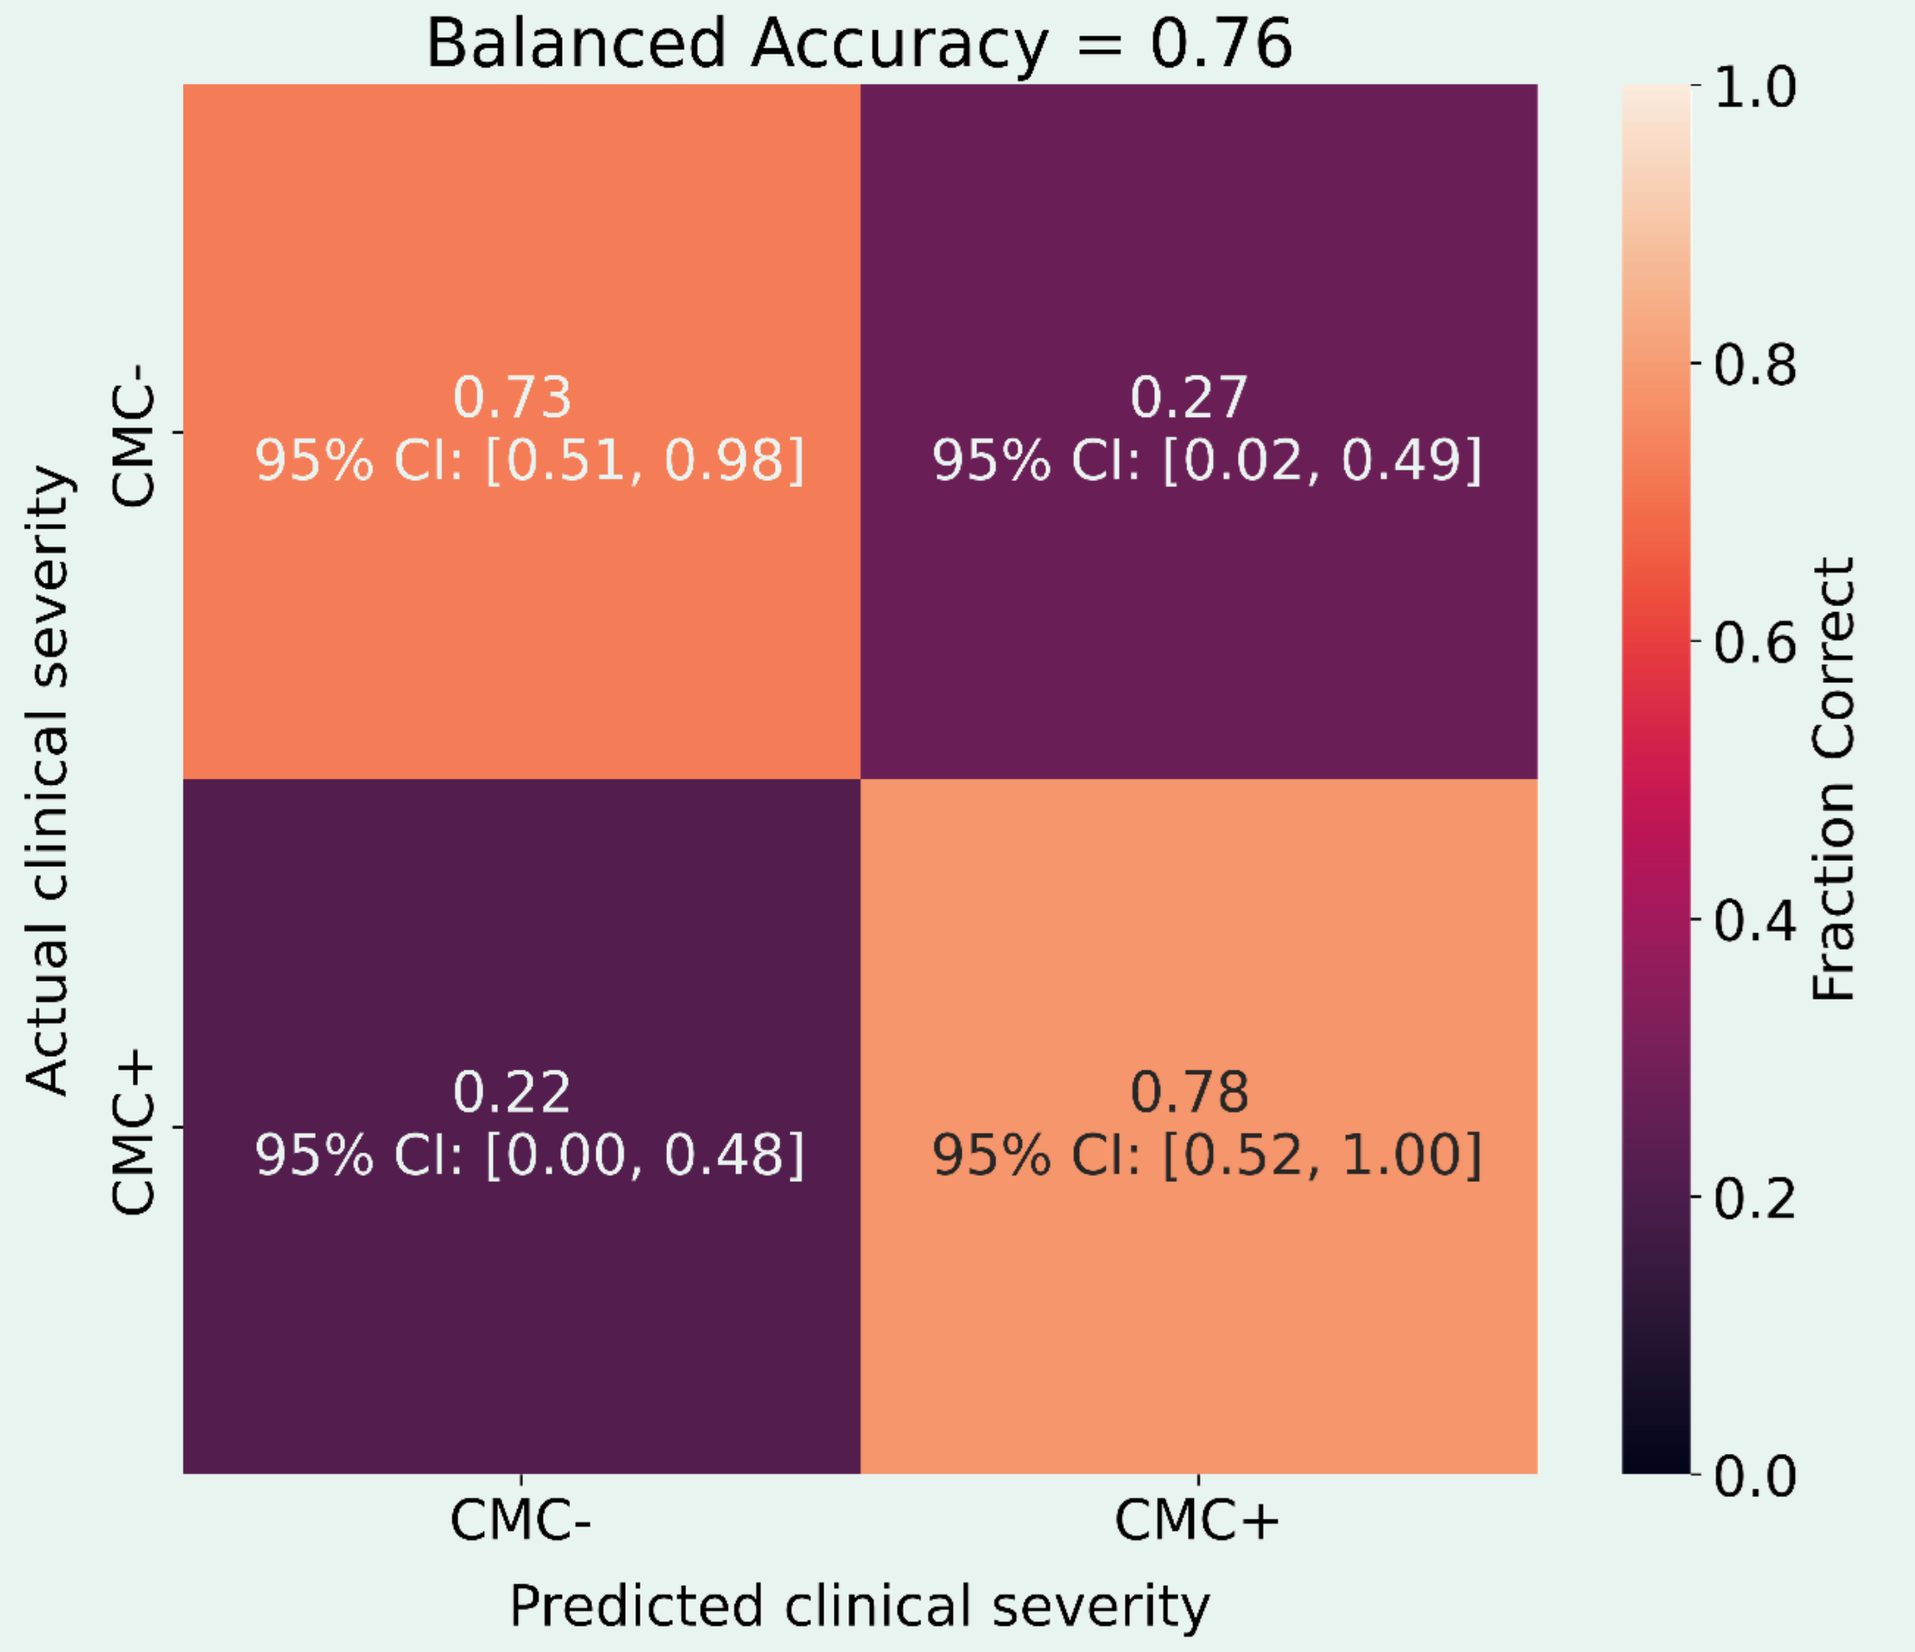

Balanced Accuracy = 0.76
0.73
95% CI: [0.51, 0.98]
0.27
95% CI: [0.02, 0.49]
0.22
95% CI: [0.00, 0.48]
0.78
95% CI: [0.52, 1.00]
Actual clinical severity
Predicted clinical severity
CMC-
CMC+
Fraction Correct

Figure 4: Performance of the support vector classifier: confusion matrix for classification of patients with MS (PwMS) as either progressors (clinically meaningful change, CMC+) or non-progressors (CMC-) using eye tracking parameters.

Disclosures

Paul Giacomini - McGill University Health Centre, Montréal, QC, Canada

PG has received research grants from: EMD Serono and F. Hoffmann-La Roche. PG has received honoraria for speaking, advisory board participation or consultation fees from: Alexion, Biogen, Bristol Myers Squibb-Celgene, EMD Serono, Genzyme-Sanofi, Innodem Neurosciences, McKesson, Novartis, Pendopharm, F. Hoffmann-La Roche, and Teva Neuroscience. PG is the Chief Medical Officer at Innodem Neurosciences and has stock options from Innodem Neurosciences.

Acknowledgements

This study was funded by Novartis Pharmaceuticals Canada, Montréal, QC, Canada. Medical writing and design support was provided by Sabitha Rajaruban and Tanya Patrawala from IQVIA Solutions Canada Inc., Mississauga, ON, Canada. The final responsibility for the content lies with the authors.

References

1. Kuhlmann, T., M. Moccia, T. Coetzee, et al., Multiple sclerosis progression: time for a new mechanism-driven framework. Lancet Neurol, 2023. 22(1): p. 78-88.
2. Serra, A., C.G. Chisari, and M. Matta, Eye Movement Abnormalities in Multiple Sclerosis: Pathogenesis, Modeling, and Treatment. Front Neurol, 2018. 9: p. 31.
3. Gajamange, S., A. Shelton, M. Clough, et al., Functional correlates of cognitive dysfunction in clinically isolated syndromes. PLoS One, 2019. 14(7): p. e0219590.
4. Sheehy, C.K., E.S. Bensinger, A. Romeo, et al., Fixational microsaccades: A quantitative and objective measure of disability in multiple sclerosis. Mult Scler, 2020. 26(3): p. 343-353.
5. Polet, K., S. Hesse, M. Cohen, et al., Video-oculography in multiple sclerosis: Links between oculomotor disorders and brain magnetic resonance imaging (MRI). Multiple sclerosis and related disorders, 2020. 40: p. 101969.
6. de Villers-Sidani, É., P. Voss, N. Bastien, et al., Oculomotor analysis to assess brain health: preliminary findings from a longitudinal study of multiple sclerosis using novel tablet-based eye-tracking software. Front Neurol, 2023. 14: p. 1243594.
7. de Villers-Sidani, É., P. Voss, D. Guitton, et al., A novel tablet-based software for the acquisition and analysis of gaze and eye movement parameters: a preliminary validation study in Parkinson's disease. Front Neurol, 2023. 14: p. 1204733.