P098

Anne H. Cross | crossa@wustl.edu

B-Cell Depletion and Efficacy Outcomes of Ofatumumab Are Consistent Across Different Body Mass Index Categories: Insights From ASCLEPIOS I and II Trials

Anne H. Cross[1], Stephen L. Hauser[2], Heinz Wiendl[3], Amit Bar-Or[4], Patricia K. Coyle[5], Xavier Montalban[6], Jérôme de Seze[7], Haoyi Fu[8], Alit Bhatt[9], Ibolya Boer[10], Ludwig Kappos[11]

[1]Washington University School of Medicine, Saint Louis, MO, USA; [2]UCSF Weill Institute for Neurosciences, University of California, San Francisco, CA, USA; [3]University of Muenster, Muenster, Germany; [4]Center for Neuroinflammation and Experimental Therapeutics and Department of Neurology, Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA, USA; [5]Department of Neurology, Stony Brook University, Stony Brook, NY, USA; [6]Department of Neurology-Neuroimmunology, Centre d'Esclerosi Múltiple de Catalunya (Cemcat), Hospital Universitario Vall d'Hebron, Barcelona, Spain; [7]University Hospital of Strasbourg, Strasbourg, France; [8]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [9]Novartis Healthcare Pvt. Ltd., Hyderabad, India; [10]Novartis Pharma AG, Basel, Switzerland; [11]Research Center for Clinical Neuroimmunology and Neuroscience Basel (RC2NB) and MS Center, Departments of Headorgans, Spine and Neuromedicine, Clinical Research, Biomedicine and Biomedical Engineering, University Hospital and University of Basel, Basel, Switzerland

Scan to access full poster

To download a copy of this poster, visit the web at: https://bit.ly/actrims-forum

Copies of this poster obtained through quick response (QR) code are for personal use only and may not be reproduced without written permission of the authors

KEY FINDINGS & CONCLUSIONS

- Monthly 20 mg subcutaneous administration of ofatumumab showed rapid B-cell depletion in pwRMS, independent of BMI
 - Ofatumumab achieves rapid and sustained B-cell depletion independent of BMI
 - Ofatumumab demonstrated consistent treatment benefits on clinical outcomes (ARR and 3m/6mCDW), as well as MRI across all BMI subgroups and consistent with those observed in the overall pooled phase 3 ASCLEPIOS I and II patient population[1]
- The subcutaneous administration of ofatumumab allows for patients to have a home-based, high-efficacy therapy with demonstrated ease of use and without the need for dose adjustment based on BMI

This study was sponsored by Novartis Pharma AG, Basel, Switzerland.

Poster Presented at the Americas Committee for Treatment and Research in Multiple Sclerosis (ACTRIMS) Forum, February 29–March 2, 2024.

INTRODUCTION

- In the ASCLEPIOS I and II trials, ofatumumab demonstrated superior efficacy over teriflunomide while maintaining a favorable safety profile in people with relapsing multiple sclerosis (pwRMS)[1]
- Previous analyses from pooled ASCLEPIOS I/II trials evaluated the effect of ofatumumab on B-cell depletion and efficacy outcomes in subgroups of patients defined by baseline demographic and disease characteristics, and revealed consistent treatment benefits and rapid B-cell depletion across diverse subgroups, suggesting that the approved dose of ofatumumab achieves consistent efficacy across a wide patient spectrum[2,3]
- As body mass index (BMI) can be a possible confounding factor affecting multiple sclerosis disease activity, it is important to understand the effect of BMI on B-cell depletion and efficacy outcomes across subgroups

OBJECTIVE

- To evaluate the effect of ofatumumab on B-cell depletion and efficacy outcomes in patients from the ASCLEPIOS I/II trials defined by their baseline BMI

METHODS

- In the ASCLEPIOS I/II trials, patients were randomized to receive either ofatumumab 20 mg subcutaneous or teriflunomide 14 mg oral for up to 30 months

Outcomes

B-cell levels (over 96 weeks)
- Median B-cell counts[a]
- Proportion of patients with B-cell counts ≤10 cells/µL

Efficacy outcomes (up to end of study [EOS])
- Annualized relapse rate (ARR)
- 3-month/6-month confirmed disability worsening (3m/6mCDW)
- Gadolinium-enhancing (Gd+) T1 lesions
- New/enlarging (ne) T2 lesions

Assessments

By typical BMI cutoffs (kg/m²)
- Underweight: BMI <18.5
- Normal weight: BMI ≥18.5 to <25
- Overweight: BMI ≥25 to <30
- Obesity: BMI ≥30

By BMI baseline quartiles (kg/m²)
- Q1: BMI <21.5
- Q2: BMI ≥21.5 to <24.6
- Q3: BMI ≥24.6 to <28.7
- Q4: BMI ≥28.7

Statistical analyses

- Descriptive statistics for categorical data (B-cell counts)
- Negative binomial regression model (ARR, Gd+ T1, and neT2 lesions)
- Cox regression model (3mCDW and 6mCDW)

[a]B-cell counts were measured categorically in the categories of 0–4, 5–14, 15–24, and up to 250 cells/µL.

3mCDW, 3-month confirmed disability worsening; 6mCDW, 6-month confirmed disability worsening; ARR, annualized relapse rate; BMI, body mass index; EOS, end of study; Gd+, gadolinium-enhancing; ne, new or enlarging; Q, quartile.

RESULTS

Baseline demographics and disease characteristics

- Baseline demographics and disease characteristics of patient subgroups categorized by typical BMI cutoffs included a mean Expanded Disability Status Scale score of ~2.9, ~70% of female patients, and a mean age of approximately 39 years
- Similar baseline demographics and disease characteristics were observed for patients across BMI quartiles

Effect of ofatumumab on B-cell counts over 96 weeks

- Across all BMI categories by typical BMI cutoffs, the median B-cell counts reduced rapidly with ofatumumab by Week 2 (≤10 cells/µL) and sustained at 0 cells/µL up to Week 96 (**Figure 1A**)
- When analyzed by BMI quartiles, the results were consistent with those of BMI cutoffs (median B-cell counts were ≤10 cells/µL at Week 2 and 0 cells/µL until Week 96)
- In the subgroups receiving teriflunomide, B-cell counts ranged between 120 and 230 cells/µL (by BMI cutoffs) and 115–230 cells/µL (by BMI quartiles) throughout the observation period

Proportion of patients with B-cell counts ≤10 cells/µL

- Irrespective of typical BMI cutoff, >75% of ofatumumab-treated patients achieved B-cell counts ≤10 cells/µL by Week 2 and ≥90% by Week 4, which was maintained up to Week 96 (**Figure 1B**)
- When analyzed by BMI quartiles, the results were consistent with those of BMI cutoffs (proportion of patients with B-cell counts ≤10 cells/µL were >75% at Week 2 and >93% at Week 96)
- In the subgroups receiving teriflunomide, B-cell counts ≤10 cells/µL were found in 0% to 7.1% (by BMI cutoffs) and 0% to 4.1% (by BMI quartiles) of patients at any given time point

Figure 1. (A) Median B-cell counts over 96 weeks by typical BMI cutoffs; (B) proportion of patients with B-cell counts ≤10 cells/µL over 96 weeks by typical BMI cutoffs

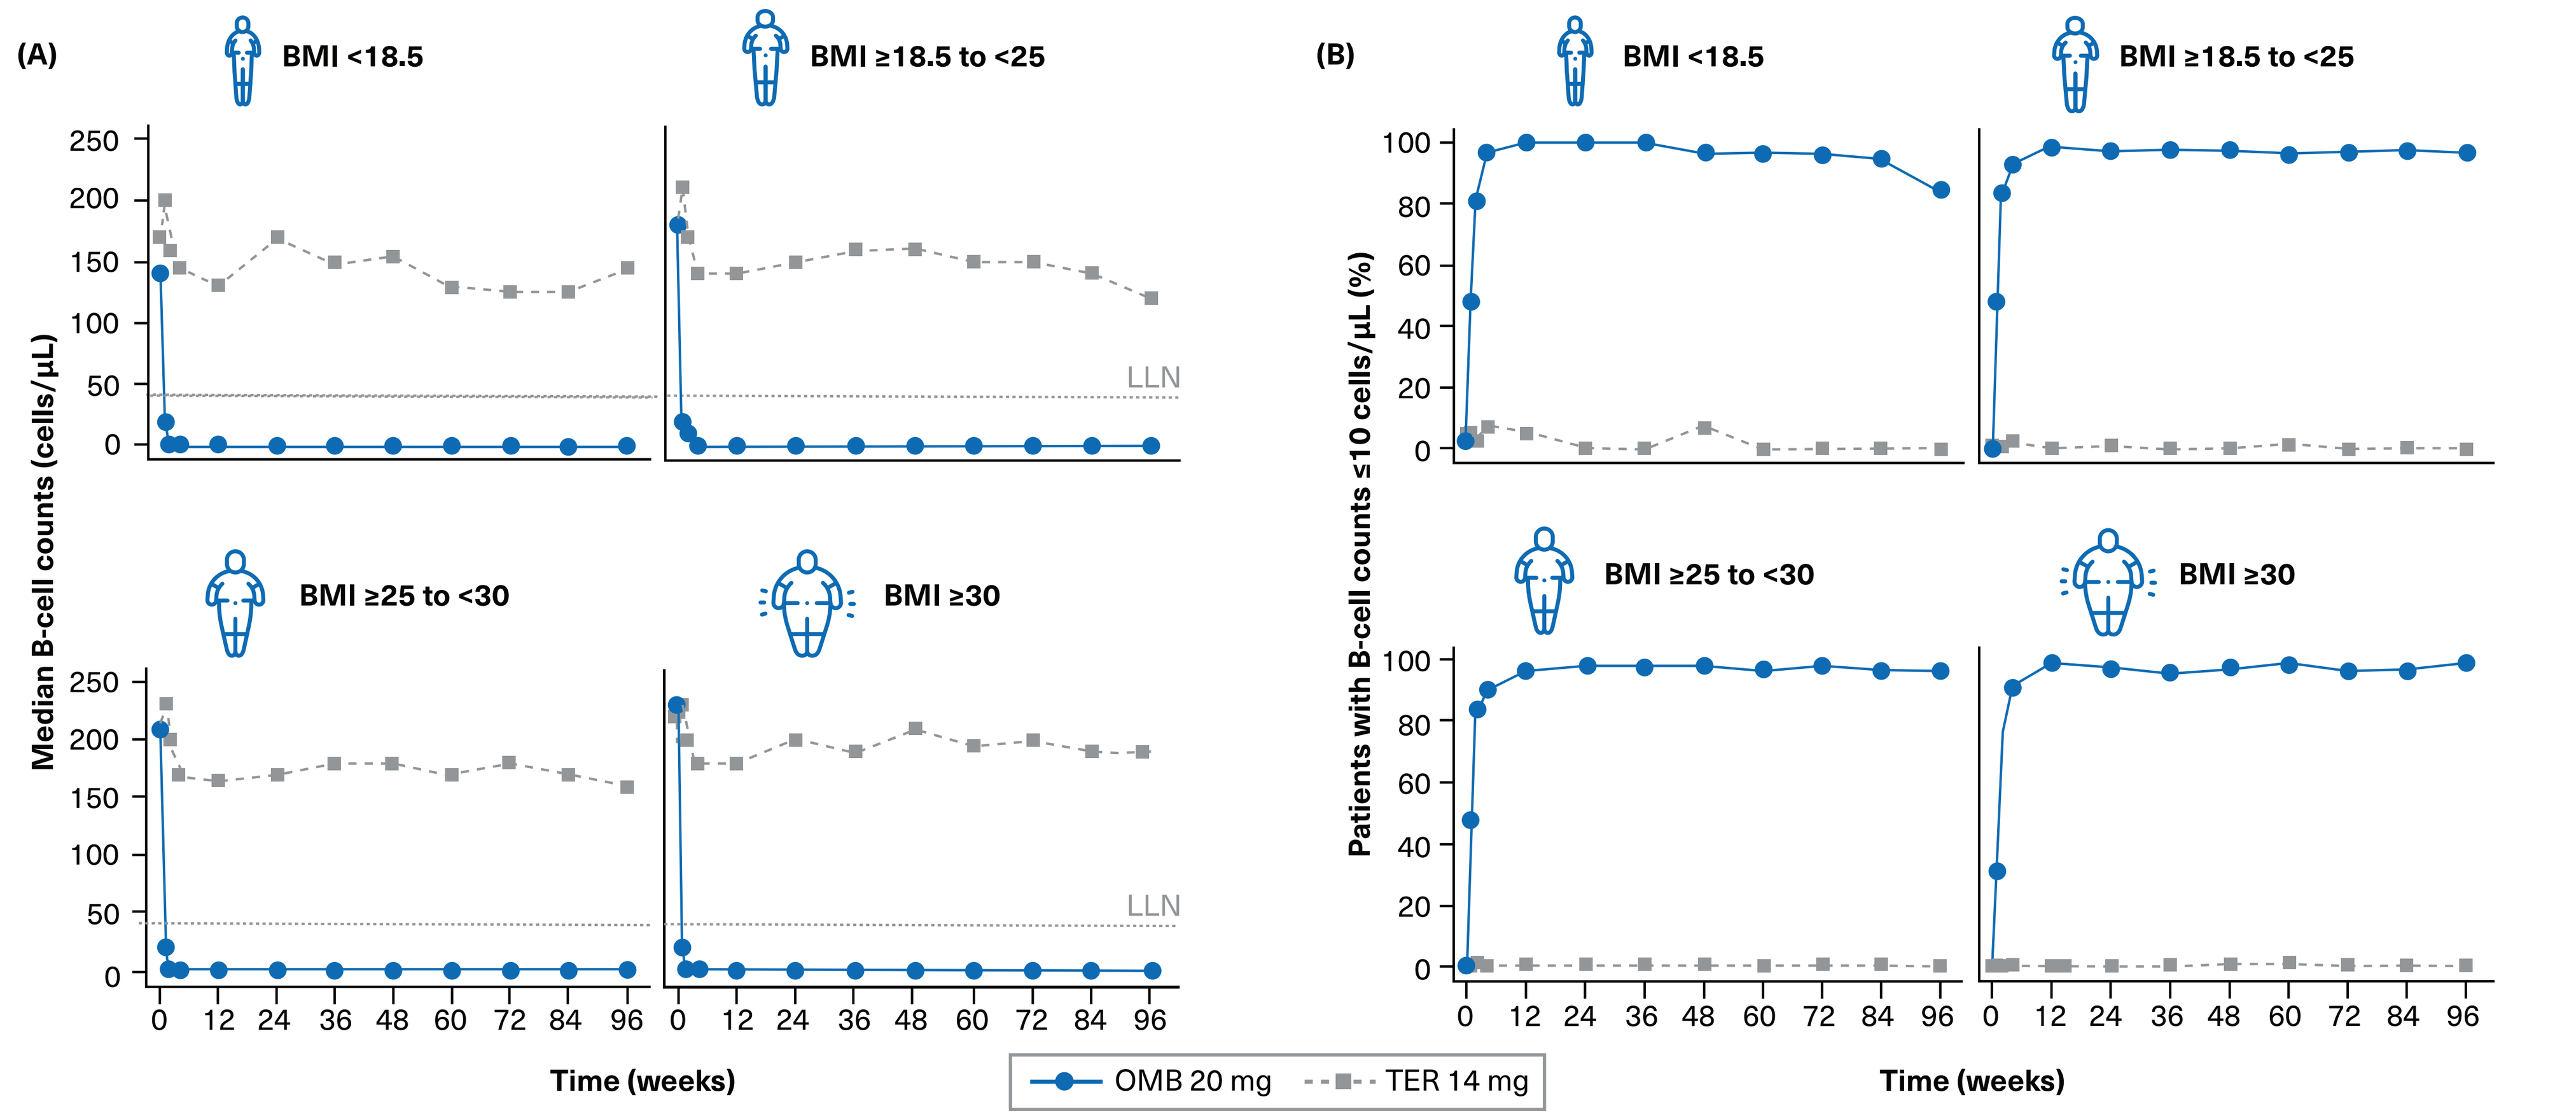
(A)
BMI <18.5
BMI ≥18.5 to <25
BMI ≥25 to <30
BMI ≥30
Median B-cell counts (cells/µL)
LLN
(B)
Patients with B-cell counts ≤10 cells/µL (%)
Time (weeks)
OMB 20 mg
TER 14 mg

BMI, body mass index; LLN, lower limit of normal; OMB, ofatumumab; TER, teriflunomide.

Effect of ofatumumab on ARR across subgroups

- Ofatumumab demonstrated higher efficacy versus teriflunomide for ARR across BMI categories by typical cutoffs (**Figure 2**)
- Similar results were observed across different BMI quartiles
- The magnitude of ofatumumab treatment effect was consistent among all subgroups

Figure 2. ARR by typical BMI cutoffs

	N/Adj Rate (95% CI) OMB 20 mg	N/Adj Rate (95% CI) TER 14 mg	Interact. p value[a]
			0.953
BMI <18.5	34/0.10 (0.04–0.25)	42/0.22 (0.12–0.40)	
BMI ≥18.5 to <25	487/0.13 (0.11–0.17)	434/0.27 (0.23–0.32)	
BMI ≥25 to <30	235/0.11 (0.08–0.16)	276/0.26 (0.21–0.33)	
BMI ≥30	190/0.10 (0.07–0.15)	182/0.22 (0.16–0.29)	

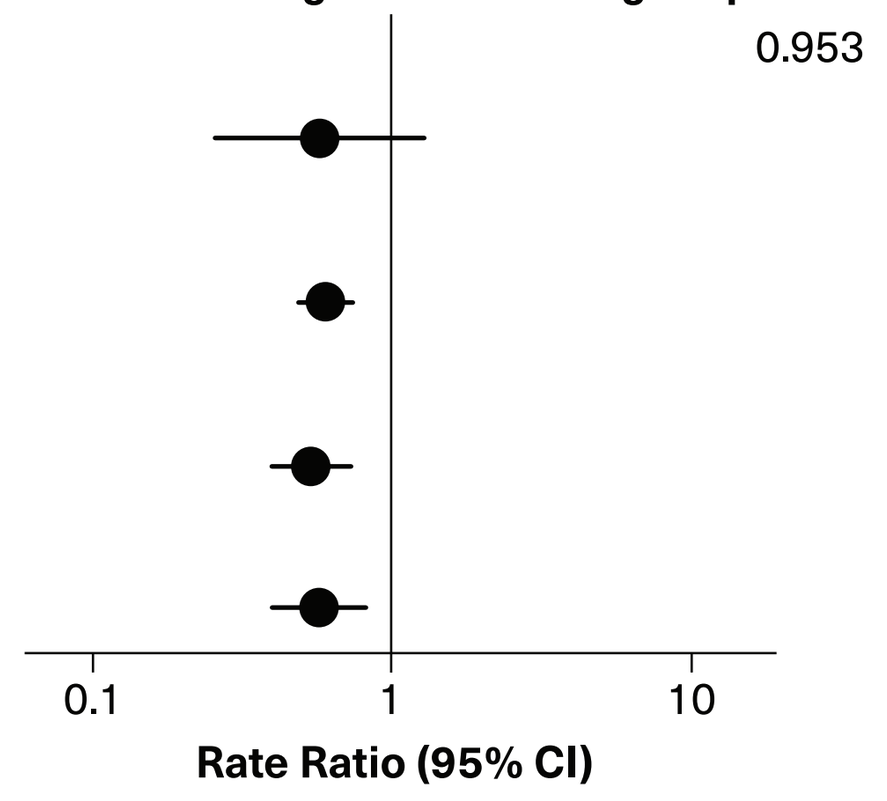
Favors OMB 20 mg
Favors TER 14 mg
0.1
1
10
Rate Ratio (95% CI)

[a]P value for the type-3 test of the treatment by subgroup interaction is a heterogeneity test (the treatment effect is similar between subgroups if the test is non-significant). Results obtained from the statistical model were adjusted with additional cofactors of subgroup and treatment by subgroup interaction for subgroup analysis. Natural log of the time-in-study was used as offset to annualize the relapse rate; N, total number of patients included in the analysis.

Adj, adjusted; ARR, annualized relapse rate; BMI, body mass index; CI, confidence interval; Interact., interaction; OMB, ofatumumab; TER, teriflunomide.

Effect of ofatumumab on 3m/6mCDW across subgroups

- Reductions in 3m/6mCDW favored ofatumumab versus teriflunomide across all BMI subgroups (**Figure 3**)
- Similar results were observed across different BMI quartiles

Figure 3. 3m/6mCDW by typical BMI cutoffs

	Event Rate, n/N (%) OMB 20 mg	Event Rate, n/N (%) TER 14 mg	Interact. p value[a]
3mCDW			0.578
BMI <18.5	2/34 (5.9)	4/42 (9.5)	
BMI ≥18.5 to <25	41/487 (8.4)	56/434 (12.9)	
BMI ≥25 to <30	21/235 (8.9)	41/276 (14.9)	
BMI ≥30	24/190 (12.6)	24/182 (13.2)	
6mCDW			0.515
BMI <18.5	2/34 (5.9)	4/42 (9.5)	
BMI ≥18.5 to <25	34/487 (7.0)	40/434 (9.2)	
BMI ≥25 to <30	16/235 (6.8)	36/276 (13.0)	
BMI ≥30	19/190 (10.0)	19/182 (10.4)	

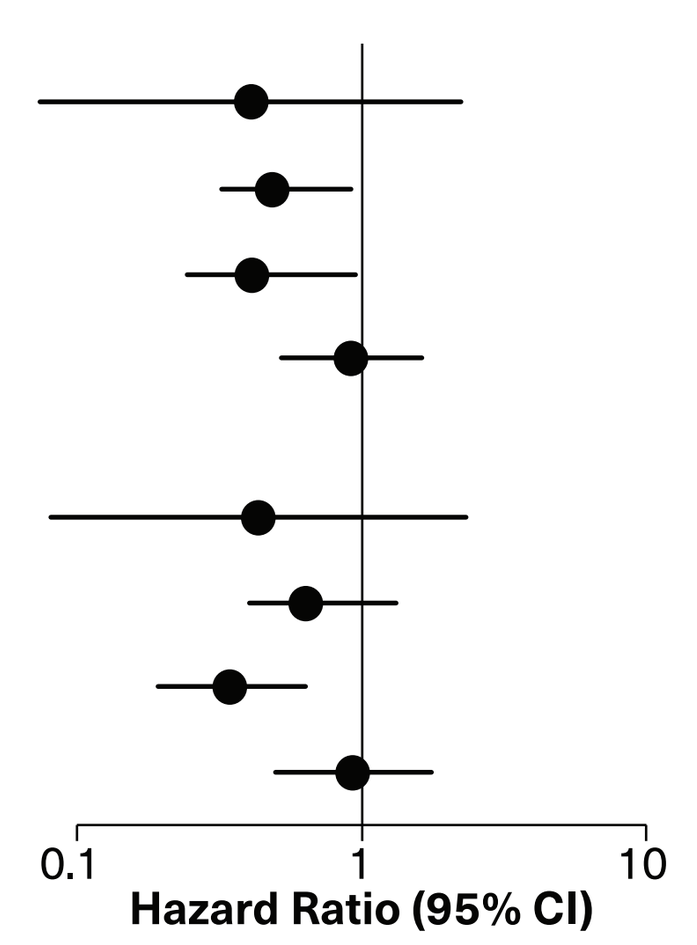
Favors OMB 20 mg
Favors TER 14 mg
0.1
1
10
Hazard Ratio (95% CI)

[a]P value for the type-3 test of the treatment by subgroup interaction is a heterogeneity test (the treatment effect is similar between subgroups if the test is non-significant). Results obtained from the statistical model were adjusted with additional cofactors of subgroup and treatment by subgroup interaction for subgroup analysis; n, total number of events included in the analysis; N, total number of patients included in the analysis.

3m/6mCDW, 3-month/6-month confirmed disability worsening; BMI, body mass index; CI, confidence interval; Interact., interaction; OMB, ofatumumab; TER, teriflunomide.

Effect of ofatumumab on MRI lesions across subgroups

- Ofatumumab demonstrated higher efficacy versus teriflunomide for Gd+ T1 and neT2 lesions across BMI categories by typical cutoffs (**Figure 4**)
- The magnitude of ofatumumab treatment effect was consistent among all BMI subgroups
- Similar results were observed across different BMI quartiles

Figure 4. MRI lesions by typical BMI cutoffs

	Adj mean number of Gd+ lesions per scan (95% CI) OMB 20 mg	Adj mean number of Gd+ lesions per scan (95% CI) TER 14 mg	Rate Ratio (95% CI)	Rate Reduction (%)/p value
				0.242[a]
BMI <18.5	0.02 (0.002–0.137)	1.27 (0.558–2.887)	0.01 (0.001–0.126)	98.7/<0.001*
BMI ≥18.5 to <25	0.03 (0.018–0.044)	0.64 (0.494–0.824)	0.04 (0.026–0.074)	95.6/<0.001*
BMI ≥25 to <30	0.01 (0.006–0.034)	0.62 (0.448–0.861)	0.02 (0.009–0.058)	97.7/<0.001*
BMI ≥30	0.05 (0.027–0.087)	0.79 (0.528–1.173)	0.06 (0.030–0.125)	93.9/<0.001*

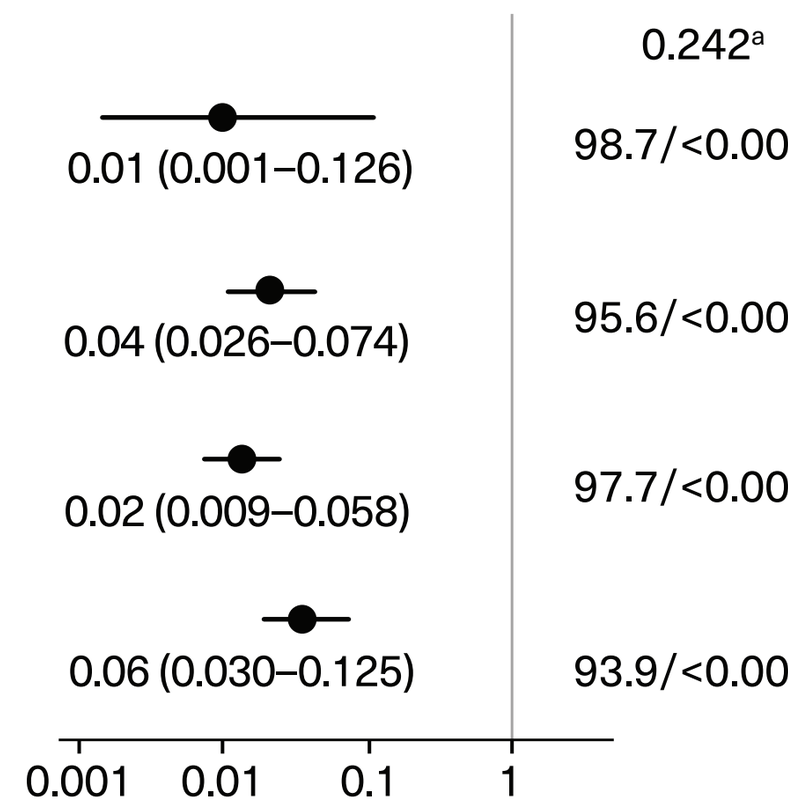
0.001
0.01
0.1
1

	Adj annualized mean rate of neT2 lesions (95% CI) OMB 20 mg	Adj annualized mean rate of neT2 lesions (95% CI) TER 14 mg	Rate Ratio (95% CI)	Rate Reduction (%)/p value
				0.078[a]
BMI <18.5	1.34 (0.750–2.394)	7.54 (4.569–12.453)	0.18 (0.083–0.382)	82.2/<0.001*
BMI ≥18.5 to <25	0.85 (0.723–1.004)	5.54 (4.748–6.459)	0.15 (0.123–0.193)	84.6/<0.001*
BMI ≥25 to <30	1.07 (0.848–1.341)	4.50 (3.699–5.466)	0.24 (0.175–0.321)	76.3/<0.001*
BMI ≥30	0.62 (0.471–0.817)	4.54 (3.559–5.792)	0.14 (0.095–0.197)	86.3/<0.001*

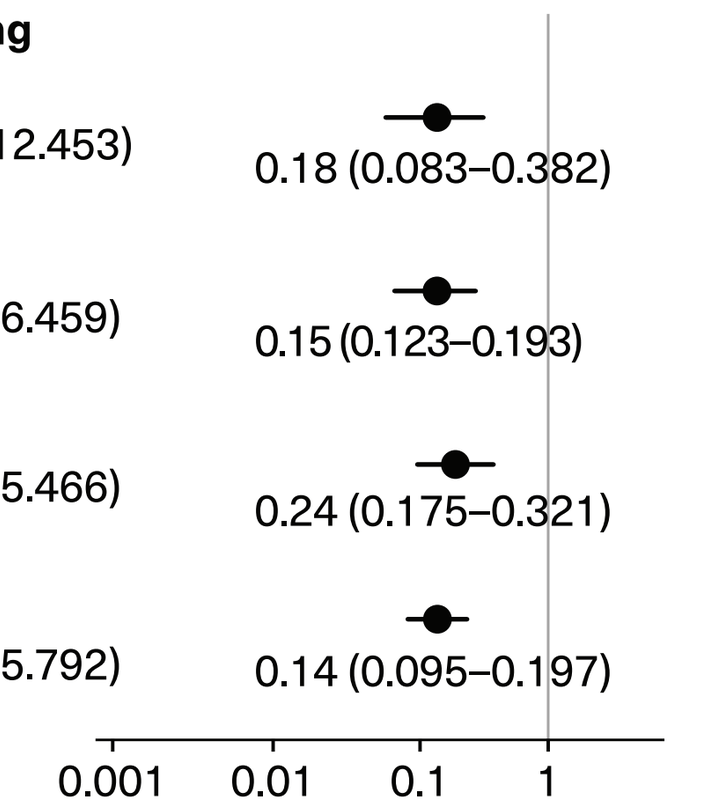
0.001
0.01
0.1
1

[a]P value for the type-3 test of the treatment by subgroup interaction is a heterogeneity test (the treatment effect is similar between subgroups if the test is non-significant). Results obtained from the statistical model were adjusted with additional cofactors of subgroup and treatment by subgroup interaction for subgroup analysis. For Gd+ T1 lesions, the natural log of the number of MRI scans with evaluable Gd+ lesion counts is used as the offset to obtain the lesion rate per scan. For neT2 lesions, the natural log of the time from the baseline scan (in years) is used as the offset. *Indicates statistical significance (two sided) at the 0.05 level.

Adj, adjusted; BMI, body mass index; CI, confidence interval; Gd+, gadolinium-enhancing; MRI, magnetic resonance imaging; neT2, new or enlarging T2 lesions; OMB, ofatumumab; TER, teriflunomide.

References

1. Hauser SL, et al. *N Engl J Med.* 2020;383:546–557.
2. Wiendl H, et al. E-presentation at EAN 2020. EPR3101.
3. Hauser SL, et al. Poster Presentation at AAN 2020. P7.1–013.

Abbreviations

3mCDW, 3-month confirmed disability worsening; **6mCDW**, 6-month confirmed disability worsening; **Adj**, adjusted; **ARR**, annualized relapse rate; **BMI**, body mass index; **CI**, confidence interval; **EOS**, end of study; **Gd+**, gadolinium-enhancing; **Interact.**, interaction; **LLN**, lower limit of normal; **MRI**, magnetic resonance imaging; **neT2**, new or enlarging T2; **OMB**, ofatumumab; **pwRMS**, people with relapsing multiple sclerosis; **Q**, quartile; **TER**, teriflunomide.

Disclosures

Anne H. Cross received personal compensation from Biogen, Bristol Myers Squibb, EMD Serono, Genentech/Roche, Horizon, Novartis, Octave, and TG Therapeutics. **Stephen L. Hauser** currently serves on the scientific advisory board of Accure, Alector, Annexon. He has previously consulted for BD, Moderna, NGM Bio, Pheno Therapeutics and previously served on the Board of Directors of Neurona. Dr. Hauser also has received travel reimbursement and writing support from F. Hoffmann-La Roche and Novartis AG for anti-CD20-therapy-related meetings and presentations. Grants: NIH/NINDS (R35NS111644), NMSS (SI-2001-35701) and Valhalla Foundation. **Heinz Wiendl** declares that he has acted as a member of the Scientific Advisory Boards of Alexion, Argenx, Biocryst, Bristol Myers Squibb, Cellerys, Galapagos, Janssen, Merck, Novartis, Sandoz-Hexal, and Unique. He also declares that he has received speaker honoraria and travel support from Alexion, Biogen, Bristol Myers Squibb, EPG Health, Genzyme, Merck, Neurodiem, Novartis, Ology, Roche, Teva and WebMD Global and acts as a paid consultant for AbbVie, Actelion, Argenx, Biogen, Bristol Myers Squibb, EMD Serono. He is acting as a paid consultant for Actelion, Argenx, BD, Bristol Myers Squibb, Dianthus, EMD Serono, EPG Health, Fondazione Cariplo, Gossamer Bio, Idorsia, Immunic, Immunovant, Inmune Bio, Syneos Health, Janssen, LTS, Merck, NexGen, Novartis, Roche, Samsung, Sangamo, Sanofi, Swiss Multiple Sclerosis Society, Toleranzia, UCB, Viatris, VirBio, and Worldwide Clinical Trials. His research is funded by Alexion, Amicus Therapeutics, Argenx, Biogen, CSL Behring, F. Hoffmann-La Roche, Genzyme, Merck, Novartis, Roche, and UCB. **Amit Bar-Or** has participated as a speaker in meetings sponsored by and received consulting fees and/or grant support from Janssen/Actelion, Atara Biotherapeutics, Biogen Idec, Celgene/Receptos, Roche/Genentech, Medimmune, Merck/EMD Serono, Novartis, and Sanofi-Genzyme. **Patricia K. Coyle** received consulting and nonbranded speaker fees from Accordant, Biogen, Bristol Myers Squibb, Eli Lilly and Company, EMD Serono, GlaxoSmithKline, Genentech, Horizon Therapeutics, LabCorp, Mylan, Novartis, Sanofi Genzyme, and Viatris. She also received research support from Celgene, CorEvitas LLC, Genentech/Roche, NINDS, Sanofi Genzyme. **Xavier Montalban** has received speaking honoraria and travel expenses for participation in scientific meetings, has been a steering committee member of clinical trials or participated in advisory boards of clinical trials in the past years with AbbVie, Actelion, Alexion, Biogen, Bristol Myers Squibb/Celgene, EMD Serono, Genzyme, Hoffmann-La Roche, Immunic, Janssen Pharmaceuticals, MedDay, Merck, Mylan, NervGen, Novartis, Sandoz, Sanofi-Genzyme, Teva, TG Therapeutics, EXCEMED, MSIF, and NMSS. **Jérôme de Seze** received personal compensation from Alexion, Biogen, F. Hoffmann-La Roche Ltd, Sanofi, LFB, Merck, Novartis, Horizon-Amgen, Argenx, and UCB. **Ludwig Kappos'** institution (University Hospital Basel) has received the following exclusively for research support: Steering committee, advisory board and/or consultancy fees (Biogen, EMD Serono Research and development, Genentech, Janssen, Novartis, Clene Nanomedicine Inc., Bayer, Bristol Myers Squibb, Celltrion Inc, Eli Lilly [Suisse] SA, EMD Serono Research and development, Galapagos NV, Kiniksa Pharmaceuticals, Merck Healthcare AG, Minoryx and Santhera, Neurostatus UHB AG, Roche, Sanofi, Santhera Pharmaceuticals, Shionogi BV, Wellmera AG, Zai Lab); speaker fees (Bristol Myers Squibb, Janssen, Novartis, Roche, Sanofi); grants (European Union, Innosuisse, Merck Healthcare AG, Novartis, Roche); and testimony. **Haoyi Fu, Alit Bhatt, and Ibolya Boer** are employees of Novartis.

Acknowledgments

Medical writing support was provided by Amitha Thakur and Saimithra Thammera and design support by Ravi Kishor, all of Novartis Healthcare Pvt. Ltd., Hyderabad, India. The final responsibility for content lies with the authors.