MULTIPLE SCLEROSIS TREATMENT AND HOLISTIC PATIENT CARE CONSENSUS BY THE SPANISH SOCIETY OF NEUROLOGY

6-013

José E. Meca Lallana[1], Sergio Martínez Yélamos[2], Sara Eichau[3], Miguel Ángel Llaneza[4], Jesús Martín Martínez[5], Joaquín Peña Martínez[6], Virginia Meca Lallana[7], Ana María Alonso Torres[8], Ester Moral Torres[9], Jordi Río[10], Carmen Calles[11], Adrián Ares Luque[12], Lluis Ramio Torrenta[13], María Eugenia Marzo Sola[14], José María Prieto[15], Maria Luisa Martínez Ginés[16], Rafael Arroyo[17], María Angeles Otano Martinez[18], Luis Brieva Ruiz[19], Montserrat Gómez Gutiérrez[20], Alfredo Rodriguez-Antigüedad Zarranz[21], Victoria Galán Sánchez Seco[22], Lucienne Costa-Frossard[23], Miguel Ángel Hernández Pérez[24], Lamberto Landete Pascual[25], Montserrat González Platas[26], Celia Oreja-Guevara[27]

[1] Clinic Neuroimmunology Unit and Multiple Sclerosis CSUR, Department of Neurology. Hospital Clínico Universitario Virgen de la Arrixaca (IMIB-Arrixaca)/Cátedra de Neuroinmunología Clínica y Esclerosis Múltiple, Universidad Católica San Antonio (UCAM), Murcia, Spain; [2] Multiple Sclerosis Unit "EMxarxa". Department of Neurology. H.U. de Bellvitge. IDIBELL. Department of Clinical Science. Universitat de Barcelona, Spain; [3] Department of Neurology, Hospital Universitario Virgen Macarena, Sevilla, Spain; [4] Department of Neurology, Complejo Hospitalario Universitario de Ferrol, Spain; [5] Department of Neurology, Hospital Universitario Miguel Servet, Zaragoza, Spain;[6] Department of Neurology, Hospital Universitario San Agustín, Avilés, Asturias, Spain; [7] Department of Neurology, Hospital Universitario La Princesa, Madrid, Spain; [8] Multiple Sclerosis Unit, Department of Neurology, Hospital Regional Universitario de Málaga, Spain; [9] Department of Neurology, Hospital de Sant Joan Despí Moisès Broggi, Sant Joan Despí, Barcelona, Spain; [10] Department of Neurology, CemCat-Vall d'Hebrón, Barcelona, Spain; [11] Department of Neurology, Hospital Universitari Son Espases, Palma de Mallorca, Spain; [12] Department of Neurology, Complejo Asistencial Universitario de León, Spain; [13] Girona Neuroimmunology and Multiple Sclerosis Unit, Neurology Department, Hospital Universitario Dr. Josep Trueta and Hospital Santa Caterina. Neurodegeneration and Neuroinflammation research group, IDIBGI. Department of Medical Sciences, University of Girona; Girona, Spain; [14] Department of Neurology, Hospital San Pedro, Logroño, Spain; [15] Fundación Instituto de Investigación Sanitaria de Santiago (IDIS), Department of Neurology, Complejo Hospitalario Universitario de Santiago, Santiago de Compostela, Spain; [16] Department of Neurology, Hospital Universitario Gregorio Marañón / Red Española de Esclerosis Múltiple (REEM), Madrid, Spain; [17] Department of Neurology, Hospital Universitario Quiron Salud Madrid, Madrid, Spain; [18] Department of Neurology, Complejo Hospitalario de Navarra, Navarra, Spain; [19] Universitat de Lleida, Hospital Universitari Arnau de Vilanova, Lleida, Spain; [20] Department of Neurology, Hospital Universitario de Cáceres, Cáceres Spain; [21] Department of Neurology, Hospital Universitario Cruces, Barakaldo, Spain; [22] Department of Neurology, Hospital Universitario de Toledo, Toledo, Spain; [23] Multiple Sclerosis Unit, Ramón y Cajal University Hospital, Madrid, Spain; [24] Department of Neurology, Hospital Nuestra Señora de Candelaria, Santa Cruz de Tenerife, Spain; [25] Department of Neurology, Hospital Universitario Dr Peset, Valencia, Spain; [25] Department of Neurology, Hospital Universitario de Canarias. La Laguna, Spain; [27] Department of Neurology, Hospital Clínico San Carlos, IdISSC,Madrid, Spain / Department of Medicine , Facultad de Medicina, Universidad Complutense de Madrid (UCM), Madrid, Spain.

INTRODUCTION

- Patients diagnosed with multiple sclerosis (MS) encounter many obstacles at diagnosis, monitoring and treatment [1-4].
- The availability of a wide range of disease modifying therapies (DMTs) present a challenge given the absence of reliable biomarkers to predict their effectiveness and safety.
- Effective management of MS requires the development of clear recommendations, guidelines and consensus. Previous guidelines did not emphasize the importance of initiating treatments with high efficacy (HE)-DMTs [5-9].
- Additionally, the increasing number of remote follow-ups, along with special situations as pregnancies, complicate the process of therapeutic decision-making.

OBJECTIVE

- Our objective of this collaborative project was to develop a comprehensive set of recommendations for the management of patients with multiple sclerosis (MS) within the Spanish clinical practice [10].

METHODS

- The development of the recommendations employed the Delphi methodology: an iterative process ensuring participant anonymity and that incorporates the participants' feedback.
- The scientific committee was formed by five nationally recognized MS experts. This committee undertook a literature review, identified relevant dimensions and items and extended invitations to panelists. The items were grouped into nine dimensions:
 1. Early diagnosis
 2. Early start of DMTs
 3. Escalation vs early start of HE-DMTs
 4. Face-to-face and remote follow-up
 5. Detection of suboptimal response and treatment optimization
 6. Patient perspective
 7. Biomarkers
 8. Pregnancy
 9. Vaccination
- The panelists were 21 experts in MS from different regions of Spain. They rated each item using a 9-point Likert scale and those items were classified as either rejected (1-3), indeterminate (4-6) or accepted (7-9). Consensus was deemed to have been reached when the agreement among the panelists was at least 66.6% **(Figure 1)**.
- Feedback from the initial round was used to revise and refine items that did not reach consensus, which were re-evaluated in the second round **(Figure 1)**.

Figure 1: The Delphi method process

First round (Dec 2021-Jan 2022)

- 128 items defined by the scientific committee (5 members) were evaluated by 21 panelists
 92 consensus items
- 36 items did not reach consensus and were reviewed and reformulated or divided

Second round (May 2022)

- 46 items evaluated by 21 panelists
- 12 consensus items
- 34 items did not reach consensus and were reviewed

Final evaluation (June 2022)

- 34 items evaluated by the scientific committee
- 8 consensus items
- 26 items did not reach consensus

110 consensus items

Some consensus items from round 1 also move to round 2; for this reason, the sum of the number of consensus items in the first round, second round and final evaluation (112) is higher than the number of items agreed upon at the end (110).

RESULTS

- Here we present a selection of items that have significant implications for clinical practice.

Diagnosis and treatment with DMTs

- The panel recommended the incorporation of assessments of optic nerve lesions and spinal MRI into the diagnostic process to enhance the information available for therapeutic decision-making.
- Early identification of progression can be very challenging. The panel considered that progression can be confirmed upon detection of worsening measures of disability, functional examination of the lower limbs and upper limbs and cognitive assessment, together with the presence or absence of new MRI lesions **(Figure 2)**.

Figure 2: Assessments for confirming disease progression

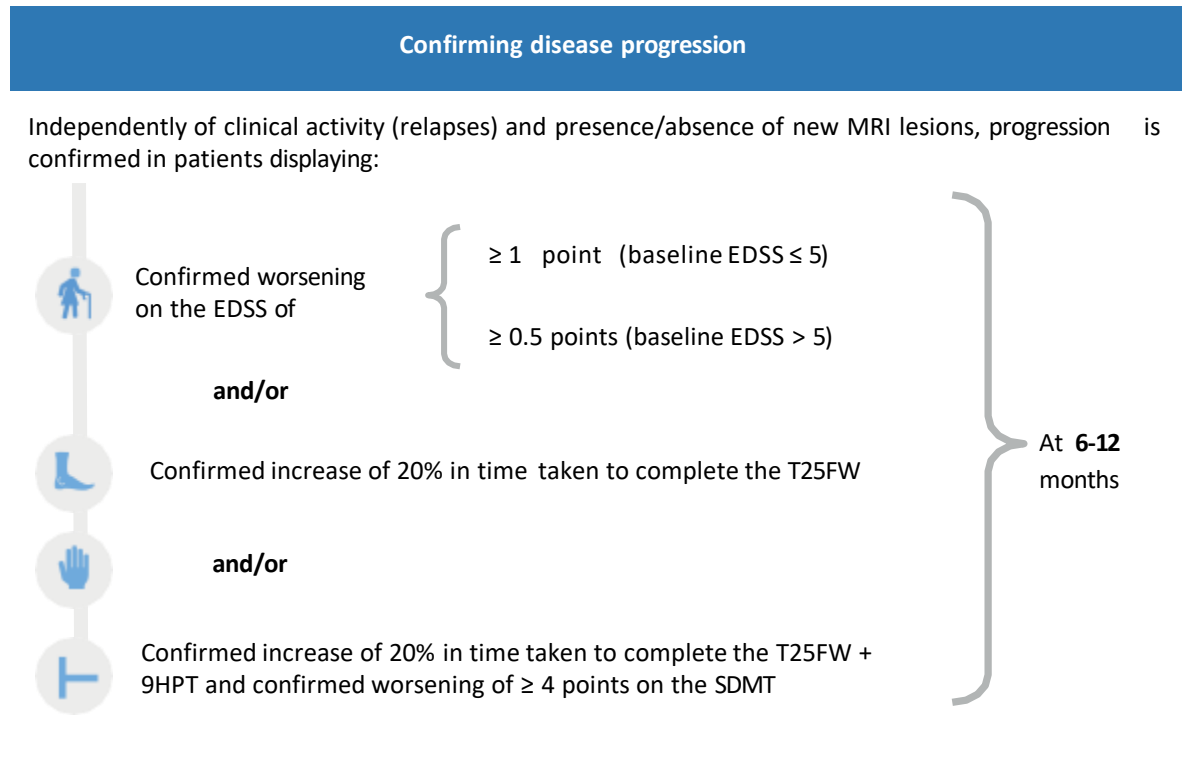
Confirming disease progression
Independently of clinical activity (relapses) and presence/absence of new MRI lesions, progression is confirmed in patients displaying:
Confirmed worsening on the EDSS of
≥ 1 point (baseline EDSS ≤ 5)
≥ 0.5 points (baseline EDSS > 5)
and/or
Confirmed increase of 20% in time taken to complete the T25FW
and/or
Confirmed increase of 20% in time taken to complete the T25FW + 9HPT and confirmed worsening of ≥ 4 points on the SDMT
At 6-12 months

9HPT: Nine-Hole Peg Test; EDSS: Expanded Disability Status Scale; SDMT: Symbol Digit Modalities Test; T25FW: Timed 25-Foo Walk test.

- The initial treatment should not be determined by a sequence of "treatment lines", but rather should be based on the presence or absence of poor prognostic factors that indicate a higher risk of relapse or progression of disability **(Figure 3)**.

Figure 3: Early onset of disease-modifying treatment

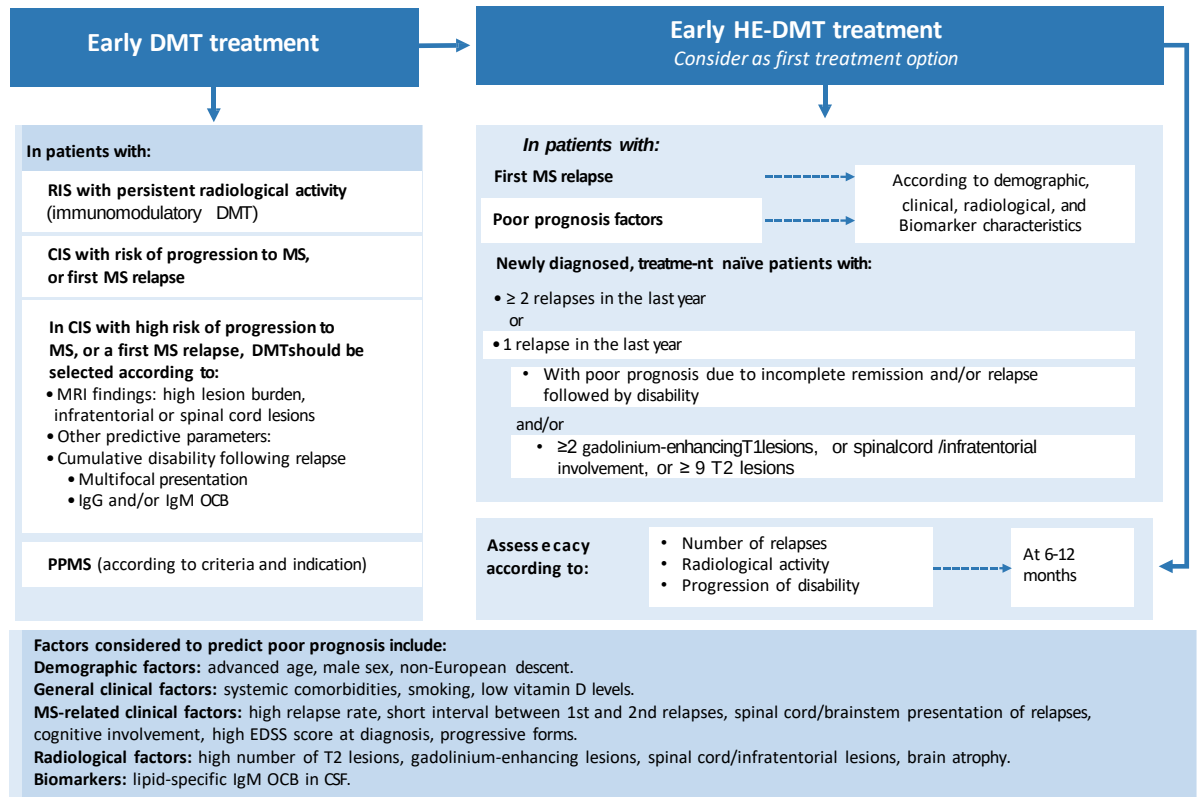
Early DMT treatment
In patients with:
RIS with persistent radiological activity (immunomodulatory DMT)
CIS with risk of progression to MS, or first MS relapse
In CIS with high risk of progression to MS, or a first MS relapse, DMTshould be selected according to:
• MRI findings: high lesion burden, infratentorial or spinal cord lesions
• Other predictive parameters:
• Cumulative disability following relapse
• Multifocal presentation
• IgG and/or IgM OCB
PPMS (according to criteria and indication)
Early HE-DMT treatment
Consider as first treatment option
In patients with:
First MS relapse
Poor prognosis factors
According to demographic, clinical, radiological, and Biomarker characteristics
Newly diagnosed, treatme-nt naïve patients with:
• ≥ 2 relapses in the last year
or
• 1 relapse in the last year
• With poor prognosis due to incomplete remission and/or relapse followed by disability
and/or
• ≥2 gadolinium-enhancingT1lesions, or spinalcord /infratentorial involvement, or ≥ 9 T2 lesions
Assess ecacy according to:
• Number of relapses
• Radiological activity
• Progression of disability
At 6-12 months
Factors considered to predict poor prognosis include:
Demographic factors: advanced age, male sex, non-European descent.
General clinical factors: systemic comorbidities, smoking, low vitamin D levels.
MS-related clinical factors: high relapse rate, short interval between 1st and 2nd relapses, spinal cord/brainstem presentation of relapses, cognitive involvement, high EDSS score at diagnosis, progressive forms.
Radiological factors: high number of T2 lesions, gadolinium-enhancing lesions, spinal cord/infratentorial lesions, brain atrophy.
Biomarkers: lipid-specific IgM OCB in CSF.

CIS: clinically isolated syndrome; CSF: cerebrospinal fluid; DMT: disease-modifying therapy; MRI: magnetic resonance imaging; MS: multiple sclerosis; OCB: oligoclonal bands; PPMS: primary progressive MS; RIS: radiologically isolated syndrome.

Follow-up, detection of suboptimal response and treatment optimization

- While telemedicine may be a supplementary or alternative approach for certain follow-up consultations, an in-person follow-up is recommended in some situations, as after the initiation of the first DMT (Figure 4) or for those patients exhibiting instability.
- Treatment should be switched if a suboptimal response is detected. The panel agreed on criteria that should be used to identify suboptimal response in patients receiving DMTs **(Figure 4)**.

Figure 4: Follow-up during treatment and after detection of suboptimal treatment response

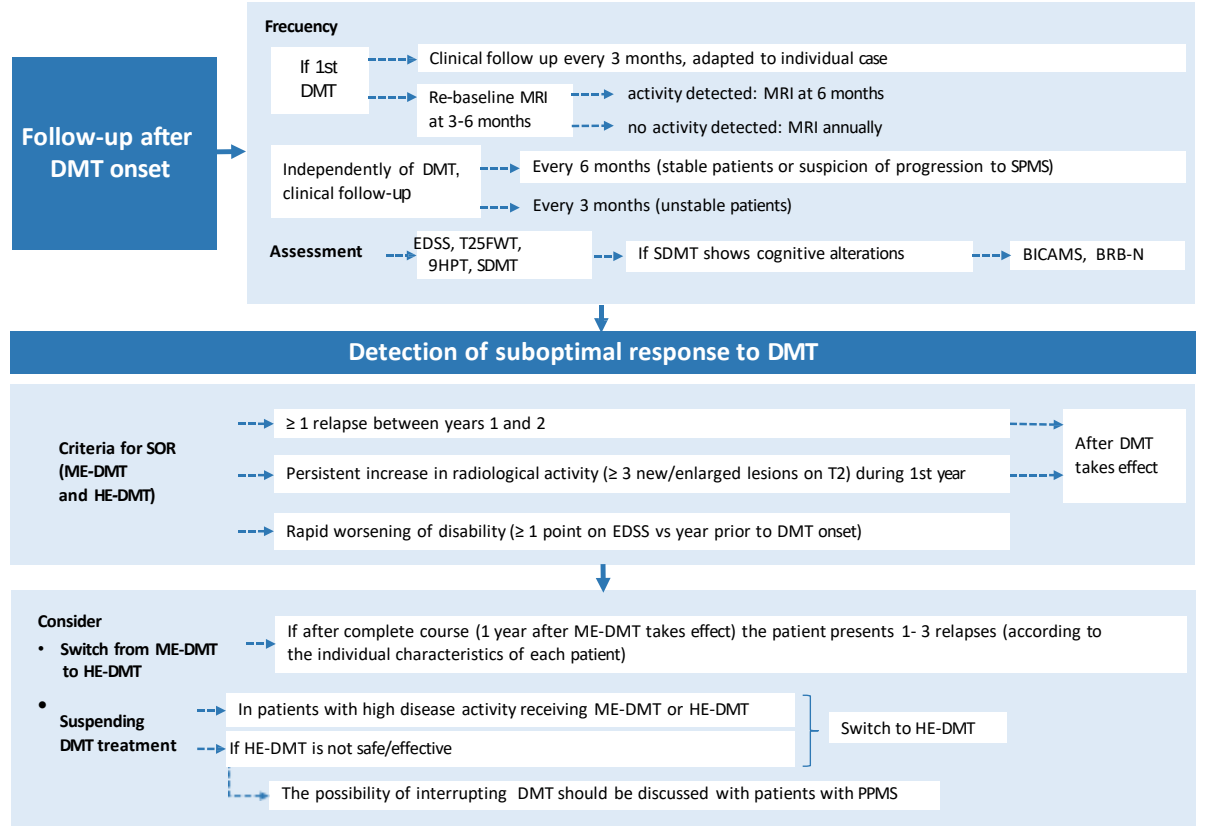
Follow-up after DMT onset
Frecuency
If 1st DMT
Clinical follow up every 3 months, adapted to individual case
Re-baseline MRI at 3-6 months
activity detected: MRI at 6 months
no activity detected: MRI annually
Independently of DMT, clinical follow-up
Every 6 months (stable patients or suspicion of progression to SPMS)
Every 3 months (unstable patients)
Assessment
EDSS, T25FWT, 9HPT, SDMT
If SDMT shows cognitive alterations
BICAMS, BRB-N
Detection of suboptimal response to DMT
Criteria for SOR (ME-DMT and HE-DMT)
≥ 1 relapse between years 1 and 2
Persistent increase in radiological activity (≥ 3 new/enlarged lesions on T2) during 1st year
Rapid worsening of disability (≥ 1 point on EDSS vs year prior to DMT onset)
After DMT takes effect
Consider
• Switch from ME-DMT to HE-DMT
If after complete course (1 year after ME-DMT takes effect) the patient presents 1- 3 relapses (according to the individual characteristics of each patient)
• Suspending DMT treatment
In patients with high disease activity receiving ME-DMT or HE-DMT
If HE-DMT is not safe/effective
Switch to HE-DMT
The possibility of interrupting DMT should be discussed with patients with PPMS

9HPT: Nine-Hole Peg Test; BICAMS: Brief International Cognitive Assessment for Multiple Sclerosis; BRB-N: Brief Repeatable Battery of Neuropsychological Tests; DMT: disease-modifying treatment; EDSS: Expanded Disability Status Scale; HE-DMT: high-efficacy DMT; ME-DMT: moderate-efficacy DMT; MRI: magnetic resonance imaging; MS: multiple sclerosis; PPMS: primary progressive MS; SDMT: Symbol Digit Modalities Test; SOR: suboptimal response; SPMS: secondary-progressive MS; T25FW: Timed 25-Foot Walk test.

Biomarkers

- The majority of biomarkers lack reliability and utility in routine clinical practice.
- Serum levels of neurofilament light chain (NfL) could be a feasible biomarker to predict disease progression, response to treatment and for follow-up.
- Immunoglobulin M (IgM) oligoclonal bands (OCBs) are a useful and feasible biomarker to predict prognosis of the disease.

Pregnancies

- In women considering becoming pregnant, it is recommended to optimize treatment and delay pregnancy to stabilize the patient.
- If an unplanned pregnancy occurs, the risk/benefit of each DMT should be evaluated.

CONCLUSIONS

- This consensus offers guidelines for the holistic care of MS patients within Spanish clinical setting [10].
- It underscores the importance of early diagnosis and the initiation of DMTs.
- Suboptimal response is characterized by relapses, new lesions on MRI, or a rise in confirmed disability. Upon identifying a suboptimal response, the approach to treatment should be adjusted, ensuring that HE-DMTs are continued in cases where the patient's condition remains stable.
- Currently, few biomarkers are considered feasible in routine clinical practice. NfL and IgM OCBs garnered substantial agreement for their utility in tracking the evolution of the disease.
- We expect that these guidelines will enhance patient care and subsequently improve the quality of life for those living with MS.

References

1. Oreja-Guevara C, et al. (2017) Multiple Sclerosis Journal 23(2_suppl):166-178; 2. Thompson AJ, et al. (2018) Lancet Neurol 17(2):162-173; 3. Spelman T, et al. (2021) JAMA Neurology; 4. D'Amico E, et al. (2019) Mult Scler Relat Disord 33:61-66; 5. Montalban X, et al. (2018) Eur J Neurol 25(2):215-237; 6. Wiendl H, et al. (2021) Ther Adv Neurol Disord 14:17562864211039648; 7. García Merino A, et al. (2016) Neurología (English Edition) 32(2):113-119; 8. García Merino A, et al. (2010) Neurología 25(6):378-390; 9.García-Merino A, et al. (2013) Neurología 28(6):375-378; 10. Meca-Lallana JE, et al. (2024) Neurologia (Engl Ed)

Acknowledgements

The authors thank Carmela García Doval (Evidenze Health España S.L.U.) for her support in medical writing, in accordance with the Good Publication Practices GPP 2022 (http://www.ismpp.org/gpp-2022) of medical writing.

Conflicts of interest

The authors declare fees for lectures, consultations, assistance to congresses, advisory meetings, personal compensation, teaching or research from: Actelion, Alexion, Almirall, Aventis, Bayer, Bial, Biogen Idec, BMS, Bristol Myers Squibb, Celgene, Daiichi Sankyo, Genzyme, GW Pharma, Janssen, Merck, Novartis, Roche, Sandoz, Sanofi, Teva, UCB Pharma and Viatris.

The study was sponsored by Novartis Pharma AG, Basel, Switzerland.

Poster presented at: AAN 2024 Annual Meeting – 13-18 April, Denver, US.