Poster P1.010

Xavier Montalban, xavier.montalban@cem-cat.org

Outcomes of COVID-19 in Ofatumumab-Treated RMS Patients: Data From the ALITHIOS Open-Label Extension Study

Xavier Montalban,[1] Ibolya Boer,[2] Bruce A. C. Cree,[3] Heinz Wiendl,[4] Anne H. Cross,[5] Sylvia R. Delgado,[6] Mario Habek,[7] Natalia Khachanova,[8] Brian J. Ward,[9] Natalia Totolyan,[10] Linda Mancione,[11] Roseanne Sullivan,[11] Ronald Zielman,[12] Xixi Hu,[11] Ayan Das Gupta,[13] Kevin Winthrop[14]

[1]Department of Neurology-Neuroimmunology, Centre d'Esclerosi Múltiple de Catalunya (Cemcat), Hospital Universitari Vall d'Hebron, Barcelona, Spain; [2]Novartis Pharma AG, Basel, Switzerland; [3]UCSF Weill Institute for Neurosciences, Department of Neurology, University of California San Francisco, San Francisco, CA, USA; [4]Department of Neurology with Institute of Translational Neurology, University of Münster, Münster, Germany; [5]Washington University School of Medicine, Saint Louis, MO, USA; [6]University of Miami Miller School of Medicine, Miami, FL, USA; [7]University Hospital Center Zagreb, School of Medicine, University of Zagreb, Zagreb, Croatia; [8]Pirogov Russian National Research Medical University, Moscow, Russia; [9]Infectious Diseases Division, Research Institute of the McGill University Health Centre, Montreal, Quebec, Canada; [10]First Saint Petersburg State Medical University, St Petersburg, Russia; [11]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [12]Novartis Pharma B.V., Amsterdam, Netherlands; [13]Novartis Healthcare Private Limited, Hyderabad, India; [14]School of Public Health at Oregon Health & Science University, Portland, OR, USA

Scan to obtain a copy of the poster

https://www.medicalcongressposters.com//Default.aspx?doc=71d23

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

KEY FINDINGS & CONCLUSIONS

- Data from ALITHIOS (as of September 25, 2022) from ~1700 patients with relapsing multiple sclerosis treated with ofatumumab showed that most COVID-19 cases were nonserious (92.3%) and mild to moderate in severity (93.9%), and most patients recovered (98.6%)
- Except for the known risk factors for serious COVID-19, such as male sex and higher body mass index, no other risk factors have been identified
- No evidence of an association between the seriousness of COVID-19 cases and ofatumumab exposure was apparent
- The COVID-19 cases observed after full vaccination (23.7%) were mostly mild to moderate in severity, and the majority of affected individuals were reported to have recovered
- The proportion of serious COVID-19 cases in the real world are consistent with the ALITHIOS trial population (9.4% vs 7.7%, respectively)

This study is sponsored by Novartis Pharma AG

Poster presented at the American Academy of Neurology (AAN) 2024 Annual Meeting, April 13-18, 2024, Denver, CO, USA

Previously presented at the 9th Congress of the European Academy of Neurology (EAN), July 1-4, 2023, Budapest, Hungary

INTRODUCTION

- The COVID-19 pandemic has created challenges in the management of patients with multiple sclerosis (MS)[1]
- B-cell–depleting therapies may compromise immune responses and lead to a higher risk of severe and prolonged COVID-19[2,3]
- There is a need for evidence from clinical studies and real-world settings to better understand the impact and effect of COVID-19 and SARS-CoV-2 vaccines in patients with MS treated with disease-modifying therapies, especially including B-cell–depleting therapies
- Data collected on COVID-19 outcomes in ofatumumab-treated patients with relapsing MS (RMS) were previously reported up to September 25, 2021 from the ongoing ALITHIOS open-label extension study and up to March 25, 2022 from progressive MS[4,5]

OBJECTIVE

- To present updated cumulative COVID-19 outcomes and vaccination statuses in patients with RMS treated with ofatumumab from the ALITHIOS study and the postmarketing population up to September 25, 2022

METHODS

- ALITHIOS (NCT03650114) is a phase 3b, open-label, single-arm extension of the core ofatumumab clinical trials[6]
- COVID-19 incidence, seriousness, severity, and outcomes, including cases after initial/booster vaccinations, were analyzed in ofatumumab-treated patients from ALITHIOS (data cutoff: September 25, 2022). Risk factors associated with serious COVID-19 were evaluated
- Postmarketing reports were collected from the Novartis Global Safety Database (data cutoff: September 25, 2022), which captures adverse events reported to Novartis by health care providers, patients, and other sources. Reporting of postmarketing cases is voluntary, with a large proportion of cases having incomplete data or incomplete follow-up

RESULTS

COVID-19 Cases Over Time in the ALITHIOS Study

- The incidence of COVID-19 cases over time in the ALITHIOS study follows the global COVID-19 incidence and incidence peaks over time (**Figure 1**)
- A clear incidence peak is observed when the SARS-CoV-2 Omicron variant was the dominant strain globally

Figure 1. COVID-19 Cases Worldwide From the WHO Dashboard[7] (A) and the ALITHIOS Study (B)

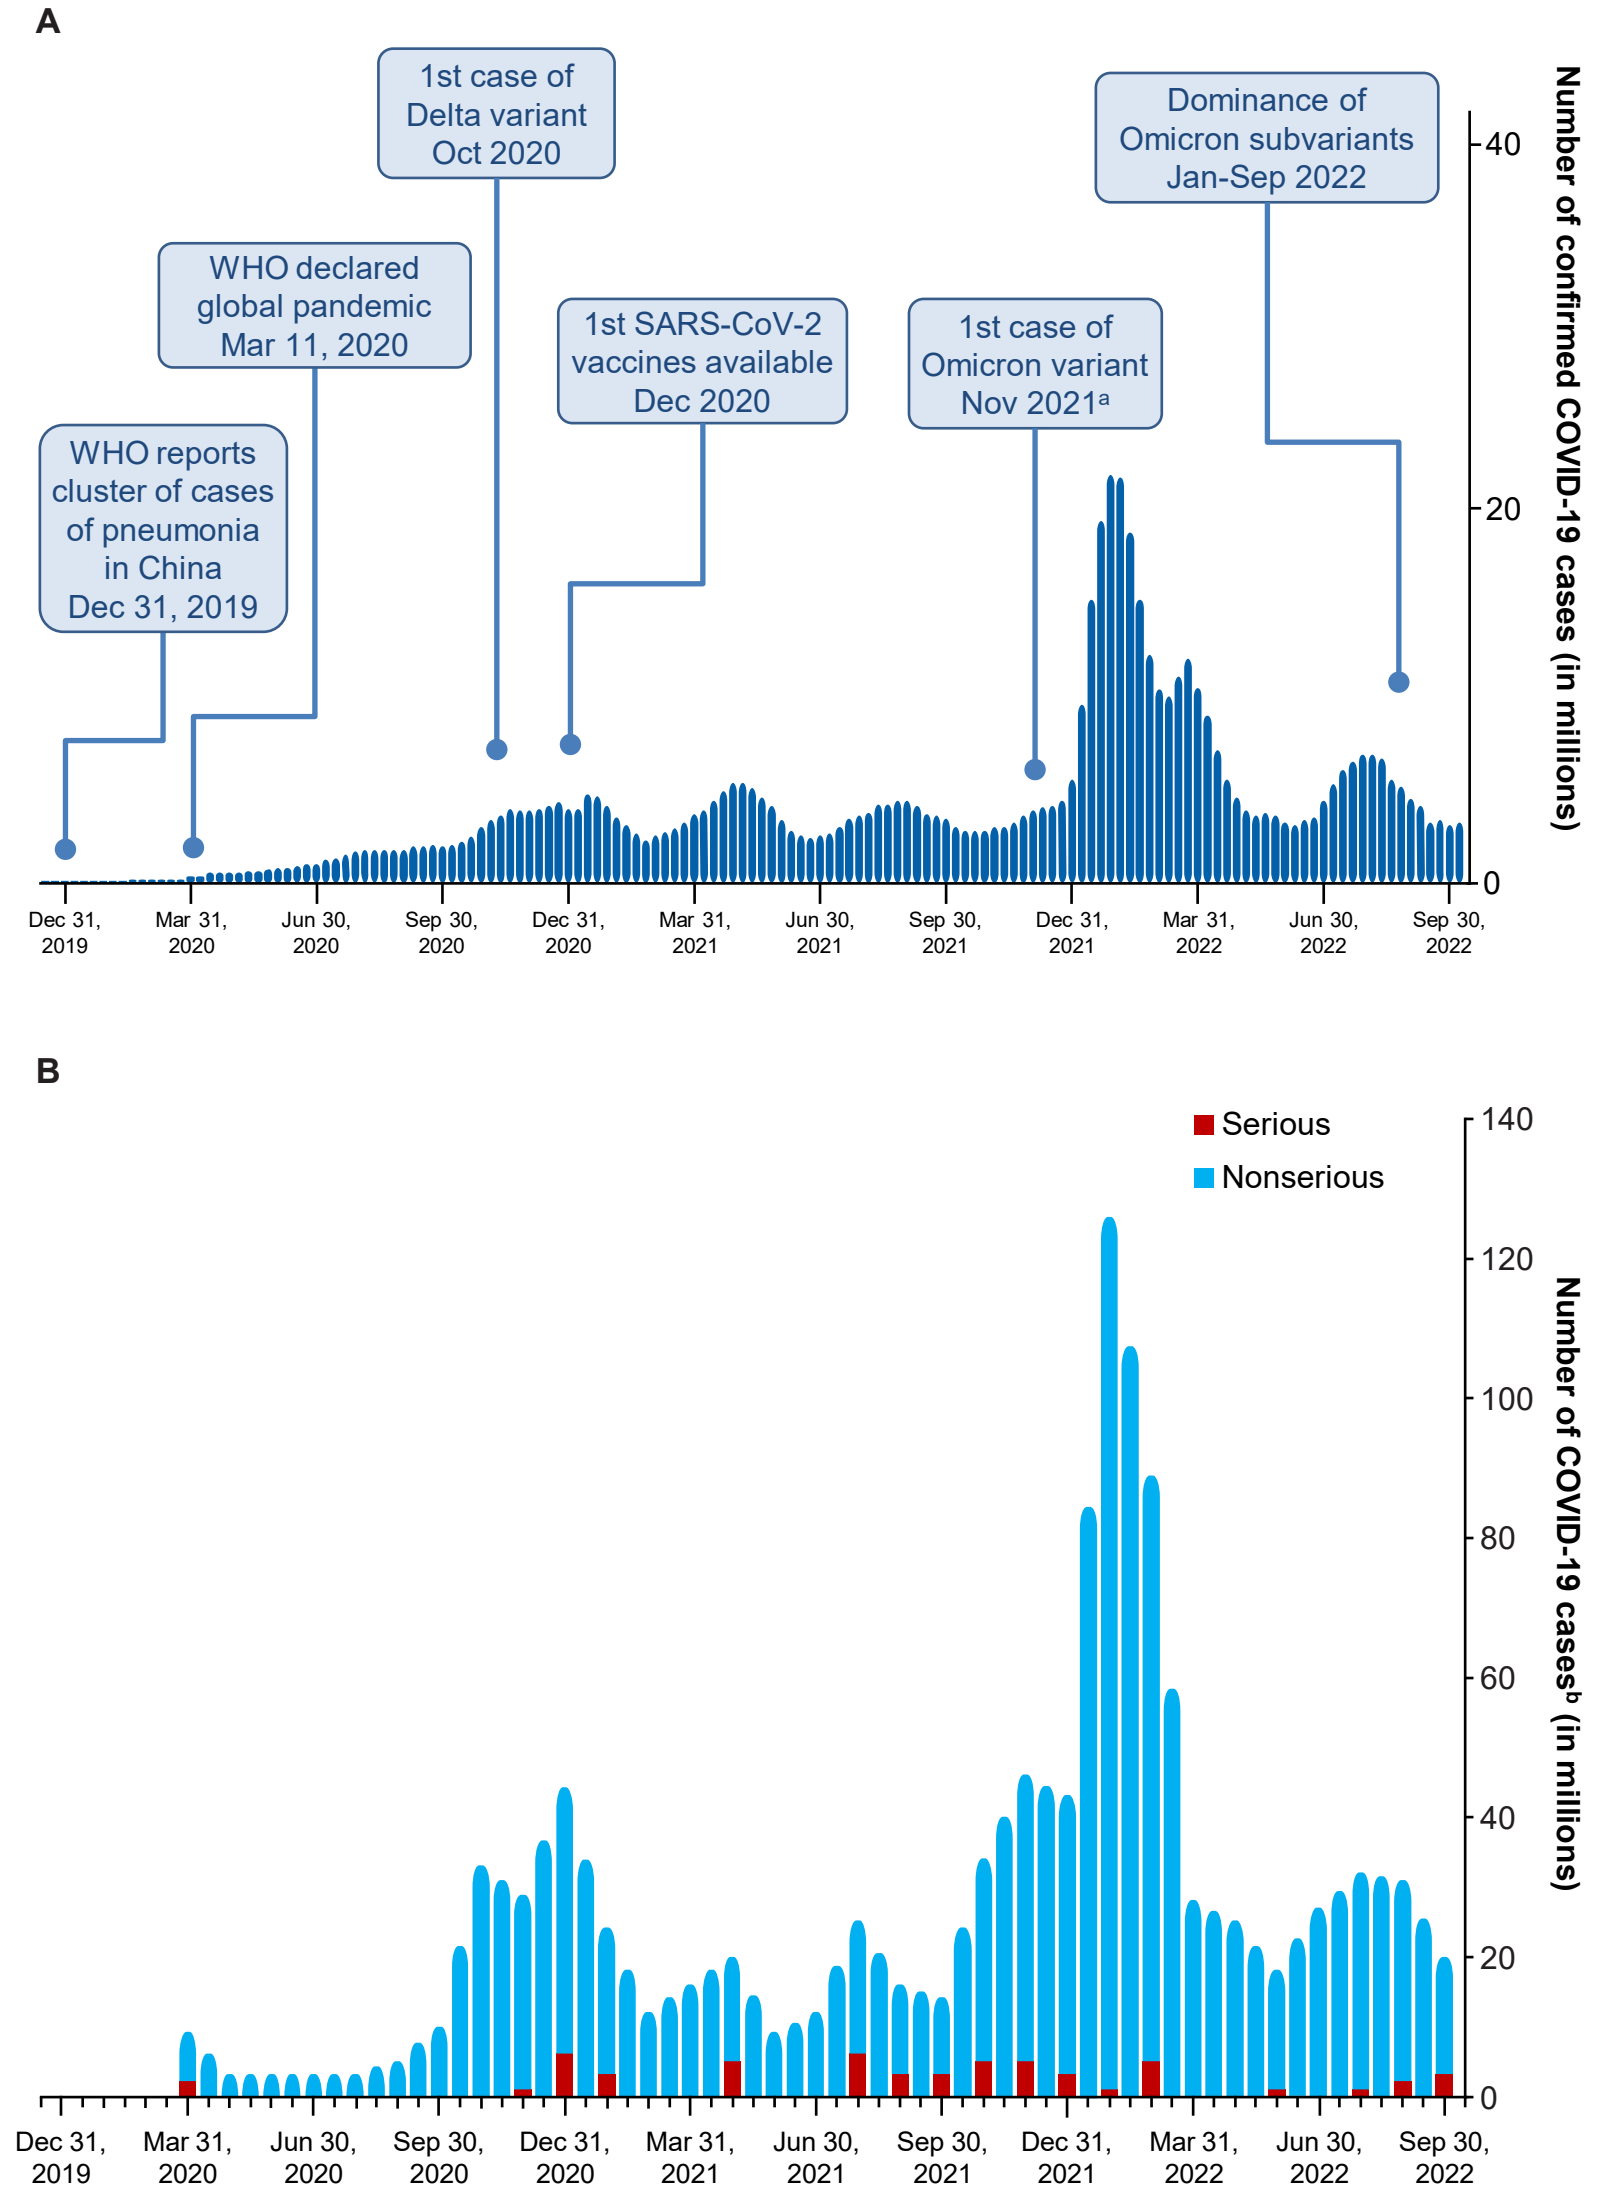
A
WHO reports cluster of cases of pneumonia in China Dec 31, 2019
WHO declared global pandemic Mar 11, 2020
1st case of Delta variant Oct 2020
1st SARS-CoV-2 vaccines available Dec 2020
1st case of Omicron variant Nov 2021a
Dominance of Omicron subvariants Jan-Sep 2022
Number of confirmed COVID-19 cases (in millions)
0
20
40
Dec 31, 2019
Mar 31, 2020
Jun 30, 2020
Sep 30, 2020
Dec 31, 2020
Mar 31, 2021
Jun 30, 2021
Sep 30, 2021
Dec 31, 2021
Mar 31, 2022
Jun 30, 2022
Sep 30, 2022
B
Serious
Nonserious
Number of COVID-19 casesb (in millions)
0
20
40
60
80
100
120
140

WHO, World Health Organization
[a]First case of Omicron variant reported after data cutoff: November 26, 2021; [b]Includes confirmed and suspected COVID-19 cases reported during the study

ALITHIOS: COVID-19 Outcomes

- 38% (648/1703) of ofatumumab-treated patients entering ALITHIOS reported COVID-19 (confirmed, n=603; suspected, n=45) (**Figure 2**)
- At baseline, patients had a mean age of 39.2 years, 69.6% were female, and 18% had a body mass index (BMI) of ≥30 kg/m^2
- There were 5 patient deaths (5/648; 0.8%)[a]; 3 patients were unvaccinated and 2 patients were fully vaccinated[b]
- Most patients (87.5%) did not interrupt ofatumumab treatment; only 5 patients discontinued the treatment due to COVID-19 or COVID-19 pneumonia
- Only 3.8% (n=64) of patients had a COVID-19 reinfection (at the onset of infection: 26 unvaccinated, 4 partially vaccinated, 22 fully vaccinated, 10 received booster doses, 2 received ≥2 booster doses)

Figure 2. COVID-19 Outcomes of Ofatumumab-Treated Patients Entering ALITHIOS

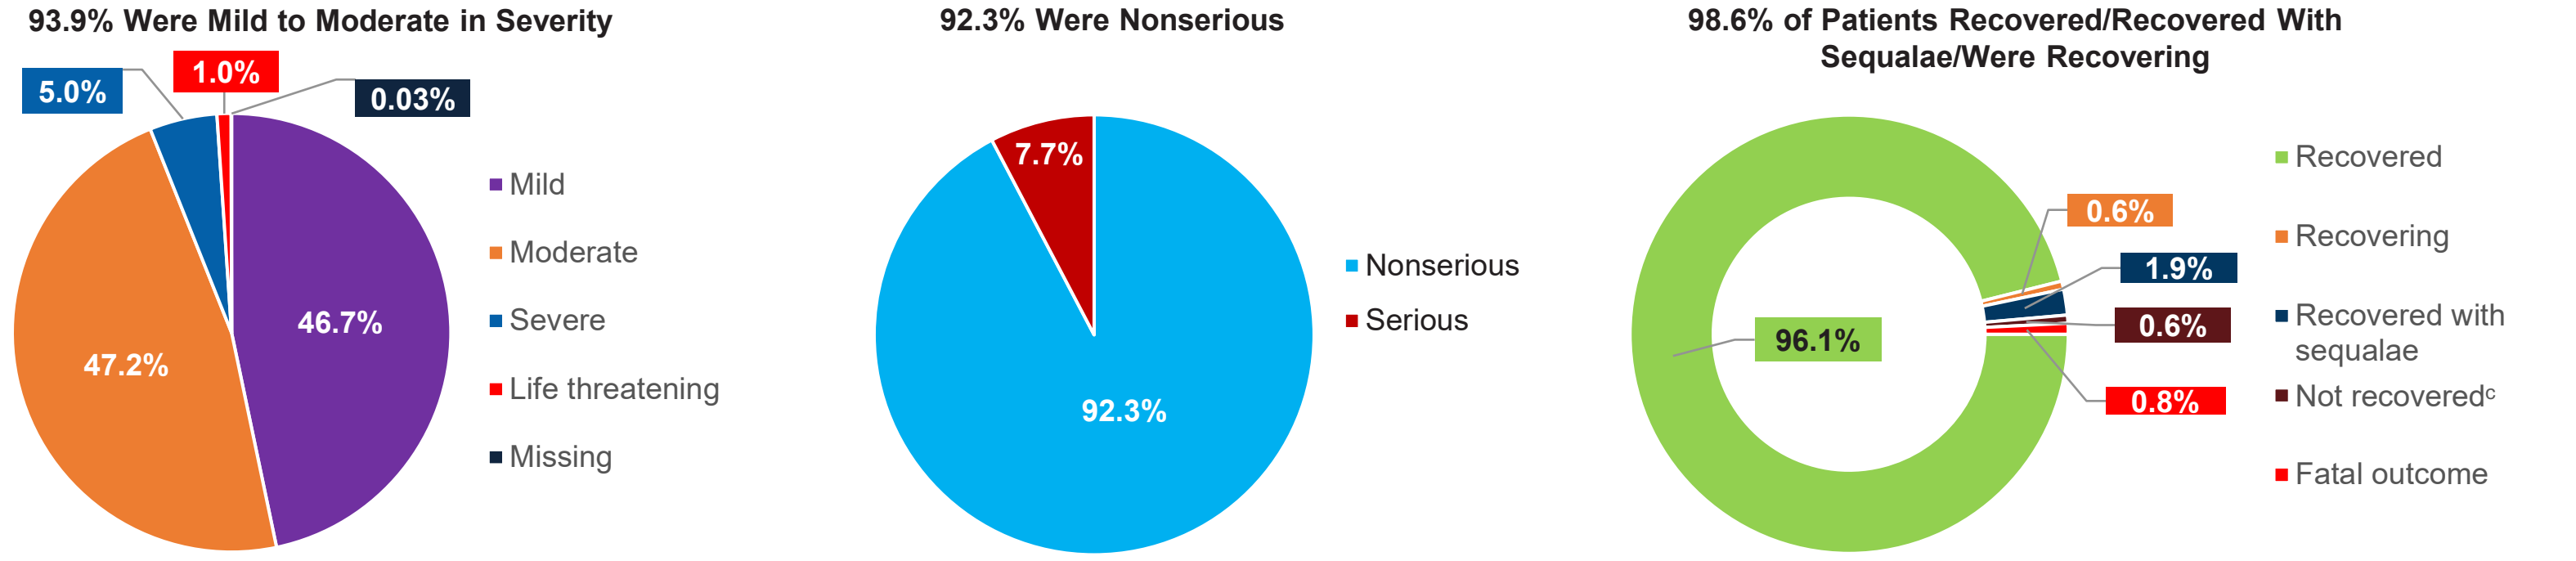
93.9% Were Mild to Moderate in Severity
5.0%
1.0%
0.03%
47.2%
46.7%
Mild
Moderate
Severe
Life threatening
Missing
92.3% Were Nonserious
7.7%
92.3%
Nonserious
Serious
98.6% of Patients Recovered/Recovered With Sequalae/Were Recovering
0.6%
1.9%
0.6%
0.8%
96.1%
Recovered
Recovering
Recovered with sequalae
Not recoveredc
Fatal outcome

[a]The 5 fatal cases consisted of: COVID-19 (n=2), COVID-19 pneumonia (n=1), COVID-19 and COVID-19 pneumonia (n=1), COVID-19 pneumonia and pneumothorax (n=1); [b]Fully vaccinated is at least 14 days after completing the primary vaccine series, which may or may not be after a booster dose. These 2 fatal cases occurred before a booster dose; 1 case had multiple risk factors for severe COVID-19, and the other case was complicated by a bilateral pneumothorax; [c]At the time of the data cutoff

ALITHIOS: COVID-19 Outcomes by Vaccination Status

- 746 patients were vaccinated (**Figure 3**). Most of the fully vaccinated patients (75.6%) received a messenger ribonucleic acid–based vaccine
- The postvaccination COVID-19 cases mostly occurred when the SARS-CoV-2 Omicron variant was the dominant strain globally
- A total of 312 COVID-19 cases were recorded, including 1 patient who had ≥3 booster doses and who recovered. The majority of cases were mild to moderate in severity (n=299/312; 95.8%) and the majority of those affected recovered[a] (n=308/312; 98.7%)
- Of the 746 patients with a COVID-19 vaccination, 55 (7.4%) had a confirmed ofatumumab dose interruption; the median duration of treatment gap was 59 days

Figure 3. COVID-19 Outcomes of Ofatumumab-Treated Patients Entering ALITHIOS by Vaccination Status

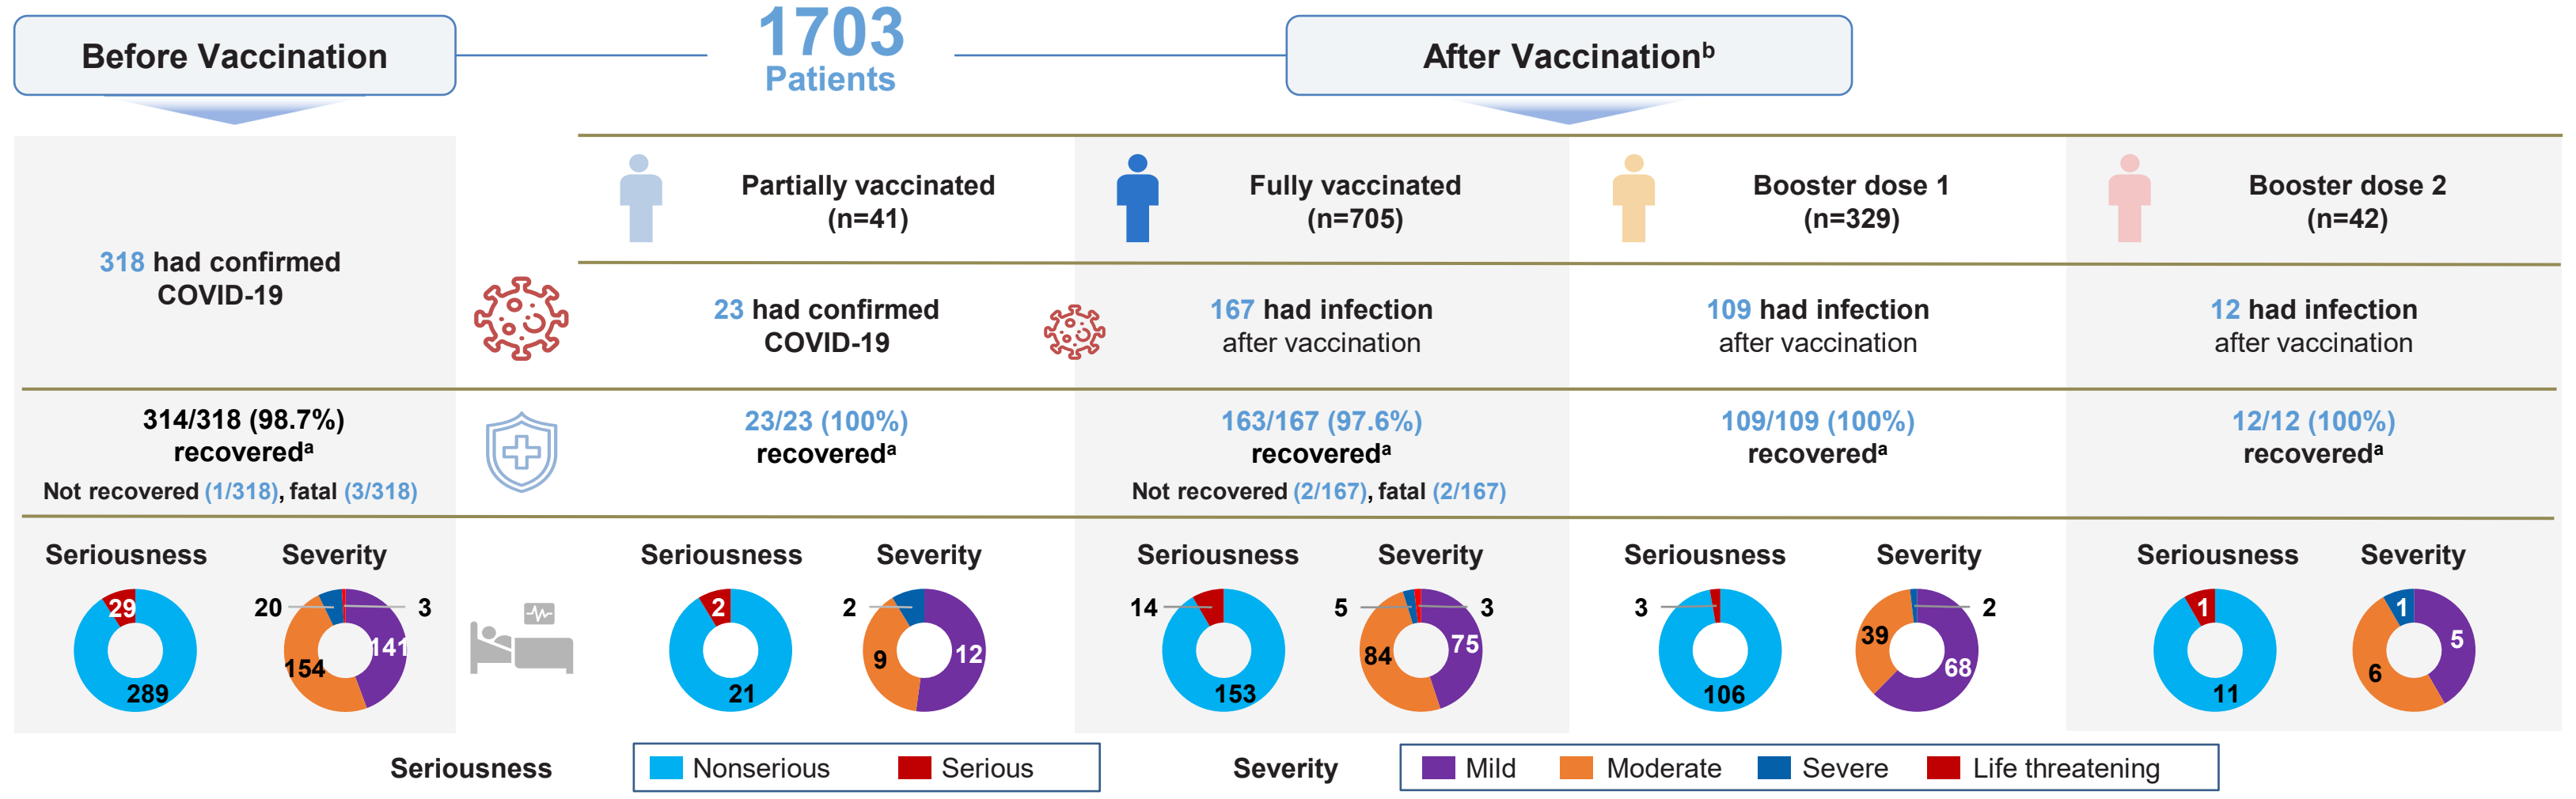
Before Vaccination
1703
Patients
After Vaccinationb
318 had confirmed COVID-19
314/318 (98.7%) recovereda
Not recovered (1/318), fatal (3/318)
Partially vaccinated (n=41)
23 had confirmed COVID-19
23/23 (100%) recovereda
Fully vaccinated (n=705)
167 had infection after vaccination
163/167 (97.6%) recovereda
Not recovered (2/167), fatal (2/167)
Booster dose 1 (n=329)
109 had infection after vaccination
109/109 (100%) recovereda
Booster dose 2 (n=42)
12 had infection after vaccination
12/12 (100%) recovereda
Seriousness
Severity
29
289
20
3
154
141
2
21
2
9
12
14
153
5
3
84
75
3
106
39
2
68
1
11
1
6
5
Seriousness
Nonserious
Serious
Severity
Mild
Moderate
Severe
Life threatening

[a]Includes patients who have recovered, were recovering, and recovered with sequalae; [b]4 patients had ≥3 booster doses, of which 1 patient had COVID-19 infection (Grade 1) and recovered

ALITHIOS: Risk Factors of Serious COVID-19

- The only identified risk factors for a serious COVID-19 infection were male sex (hazard ratio [HR] 1.89 [95% CI: 1.08-3.31]; p=0.026) and a high BMI of ≥30 kg/m^2 vs <30 kg/m^2 (HR 1.98 [95% CI: 1.06-3.67]; p=0.031) (**Figure 4**)
- Time of ofatumumab exposure was not associated with an increased risk of serious COVID-19

Figure 4. HRs From a Cox Model Analysis of Risk Factors for Serious COVID-19[a,b]

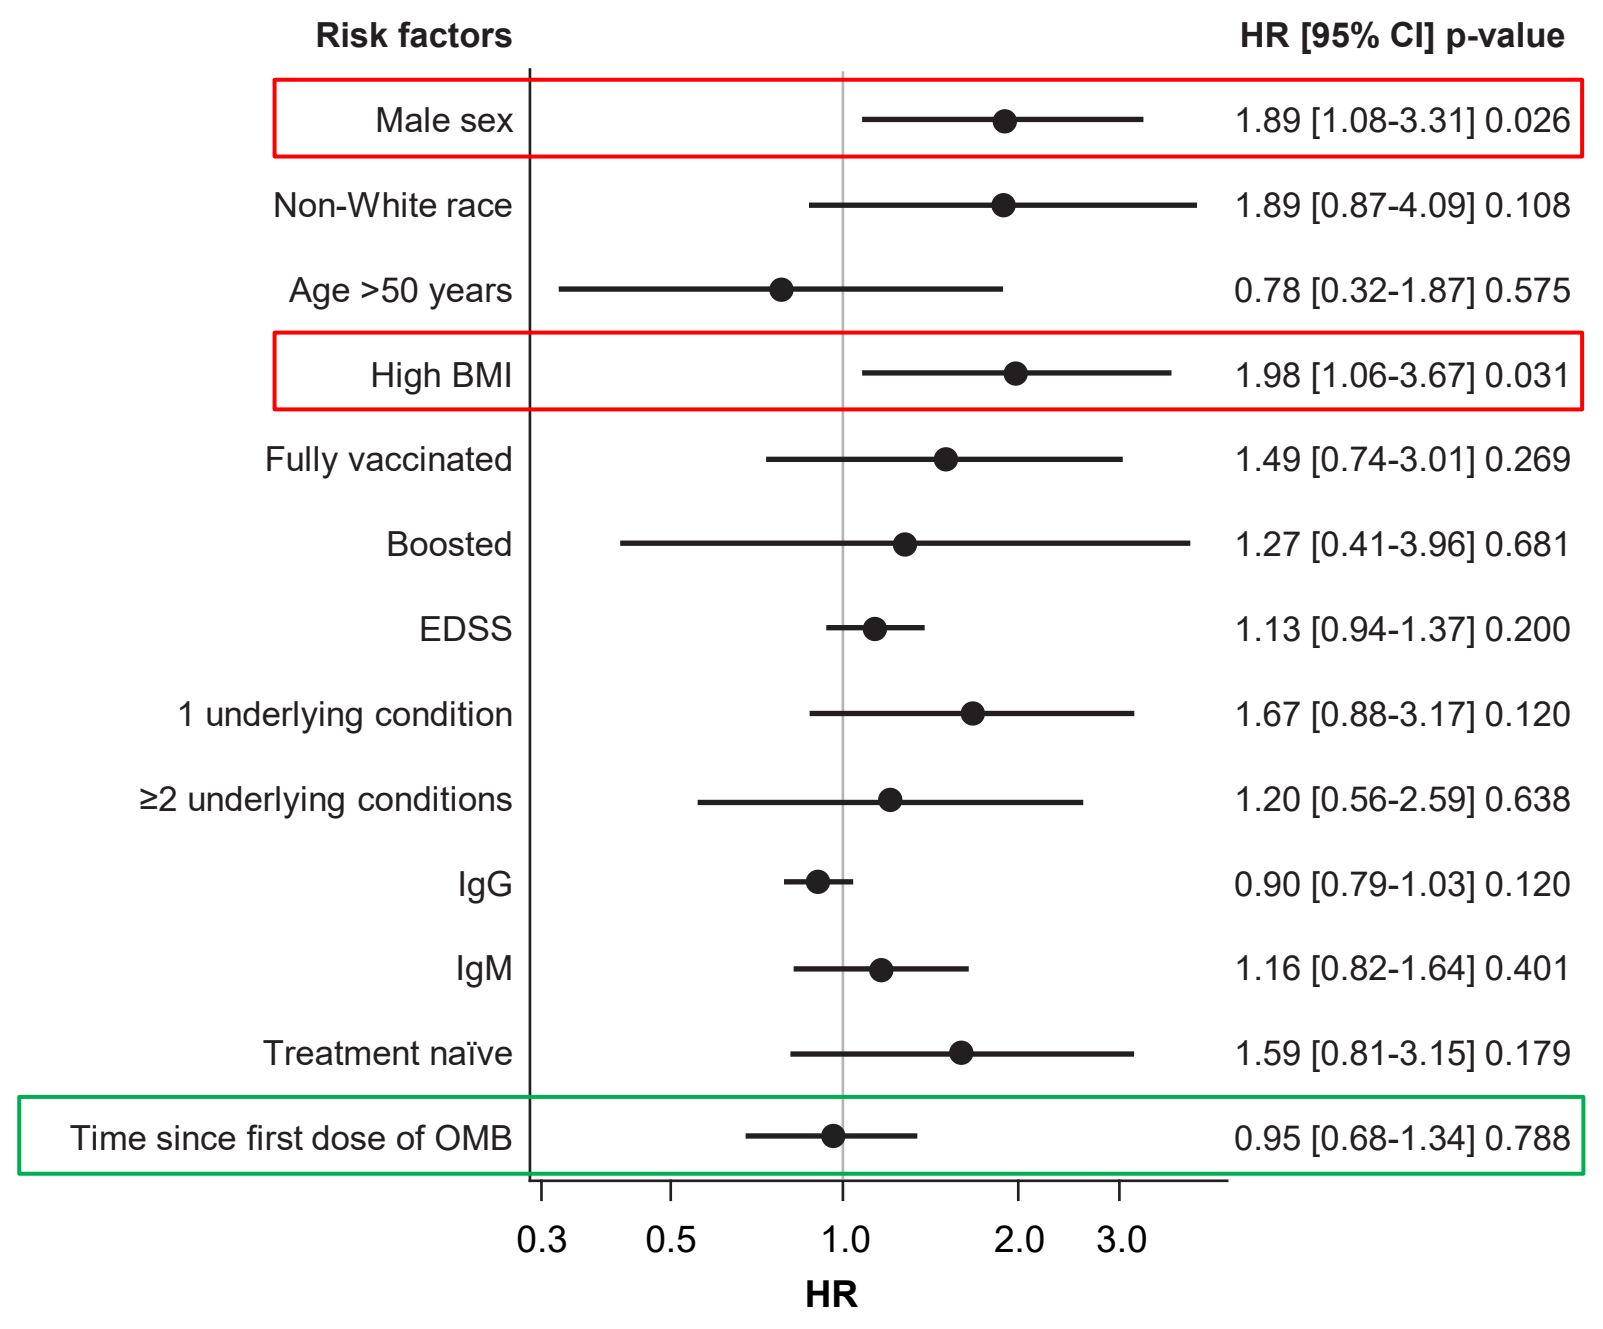
Risk factors
HR [95% CI] p-value
Male sex 1.89 [1.08-3.31] 0.026
Non-White race 1.89 [0.87-4.09] 0.108
Age >50 years 0.78 [0.32-1.87] 0.575
High BMI 1.98 [1.06-3.67] 0.031
Fully vaccinated 1.49 [0.74-3.01] 0.269
Boosted 1.27 [0.41-3.96] 0.681
EDSS 1.13 [0.94-1.37] 0.200
1 underlying condition 1.67 [0.88-3.17] 0.120
≥2 underlying conditions 1.20 [0.56-2.59] 0.638
IgG 0.90 [0.79-1.03] 0.120
IgM 1.16 [0.82-1.64] 0.401
Treatment naïve 1.59 [0.81-3.15] 0.179
Time since first dose of OMB 0.95 [0.68-1.34] 0.788
0.3
0.5
1.0
2.0
3.0
HR

BMI, body mass index; DMT, disease-modifying therapy; EDSS, Expanded Disability Status Scale; HR, hazard ratio; Ig, immunoglobulin; OMB, ofatumumab
[a]The analysis was based on ALITHIOS patients who were "on ofatumumab" (including 100 days after the last dose) as of the beginning of 2020. It confirmed the association of some factors with serious COVID-19 but did not rule out the potential causation with other factors as reported in the literature. For covariates other than vaccination status, IgG, and IgM, the last available value by January 1, 2020 was used; [b]Obtained from a Cox model with adjustment for sex, race, age (>50 vs ≤50 years), BMI (≥30 vs <30 kg/m^2), EDSS, number of underlying conditions, prior DMT, and time since first dose of OMB (years), and with vaccination status, IgG, and IgM as time-varying covariates

Postmarketing Population: COVID-19 Outcomes

- Overall, 1154 confirmed COVID-19 cases in ofatumumab-treated patients were reported in the postmarketing setting; the cumulative postauthorization patient exposure to ofatumumab since the first launch: ~37,127 patient-years
- For confirmed COVID-19 cases, the mean age (range) at baseline was 45 (17-78) years
- 108 (9.4%) were serious cases (74 hospitalizations, 38 medically significant, 2 life threatening, and 4 fatal)
- Of the 415 cases with outcomes available at the time of the data cutoff, most recovered/recovered with sequelae/were recovering (n=367, 88.4%); the remaining were condition unchanged/not recovered (n=44) and fatal (n=4)

Acknowledgements

This study is supported by Novartis Pharma AG, Basel, Switzerland. Writing support was provided by Venkateswarlu Bonala and Saimithra Thammera (employees of Novartis Healthcare Pvt. Ltd., Hyderabad, India). The final responsibility for the content lies with the authors.

Disclosures

Ibolya Boer, Linda Mancione, Ronald Zielman, Xixi Hu, and **Ayan Das Gupta** are employees of Novartis. **Bruce A. C. Cree** has received personal compensation for consulting from Alexion, Atara Biotherapeutics, Autobahn, Avotres, Biogen, EMD Serono, Novartis, Sanofi, TG Therapeutics, and Therini and has received research support from Genentech. **Heinz Wiendl** has received honoraria for acting as a member of scientific advisory boards for Biogen, Evgen, Genzyme, MedDay, Merck Serono, Novartis, Roche Pharma AG, and Sanofi-Aventis; has received speaker honoraria and travel support from Alexion, Biogen, Cognomed, F. Hoffmann-La Roche Ltd, Gemeinnützige Hertie-Stiftung, Genzyme, Merck Serono, Novartis, Roche Pharma AG, Teva, and WebMD Global; is a paid consultant for AbbVie, Actelion, Biogen, IGES, Johnson & Johnson, Novartis, Roche, Sanofi-Aventis, and the Swiss Multiple Sclerosis Society; and his research is funded by Biogen, Deutsche Forschungsgemeinschaft, the Else Kröner Fresenius Foundation, the European Union, the Fresenius Foundation, the German Ministry for Education and Research (BMBF), GlaxoSmithKline GmbH, the Hertie Foundation, the Interdisciplinary Center for Clinical Studies (IZKF) Muenster and RE Children's Foundation, the NRW Ministry of Education and Research, Roche Pharma AG, and Sanofi-Genzyme. **Anne H. Cross** has received consulting fees, research support, and honoraria from Academic CME, ACTRIMS, Biogen, Bristol Myers Squibb, Celgene, CME Outfitters, the Conrad N. Hilton Foundation, the Consortium of Multiple Sclerosis Centers, EMD Serono, Genentech, Greenwich Biosciences, Horizon, Janssen, Merck, Novartis, The Potomac Center for Medical Education, Projects in Knowledge, Roche, TG Therapeutics, and WebMD; serves on the scientific advisory board for ASCLEPIOS I/II for Novartis; has received grants from the National Institutes of Health and the US Department of Defense; has held an elected office (secretary) on the Board of Governors of the Consortium of Multiple Sclerosis Centers; has been a member of the scientific advisory board of Race to Erase MS; has been a program committee member (chair) of ACTRIMS; has been a member of the COVID-19 advisory committee of the National Multiple Sclerosis Society USA and the National Multiple Sclerosis Society representative on the Progressive MS Alliance; and has received a patent for Yablonskiy DA, Sukstansky AL, Wen J, Cross AH. Methods for simultaneous multi-angular relaxometry of tissue using magnetic resonance imaging. Patent 15060-630 (015875). **Silvia Delgado** has received research support from EMD Serono and Novartis. **Mario Habek** has participated as a clinical investigator and/or received consultation and/or speaker fees from Actelion, Alexion, Alvogen, Bayer, Biogen, Merck, Novartis, Pliva/Teva, Roche, Sanofi-Genzyme, and TG Pharmaceuticals. **Natalia Khachanova** has participated as a clinical investigator and/or received consultation and/or speaker fees from Actelion, BIOCAD, F. Hoffmann-La Roche Ltd, Generium, Janssen, Mapi, Merck, Novartis, Octapharma AG, Osmotica, Sanofi-Aventis, Teva, and TG Pharmaceuticals. **Brian J. Ward** serves on a scientific advisory board for Novartis and reports personal fees from Novartis for this activity; is a medical officer for Medicago and holds parts of patents for vaccines targeting influenza, *Clostridioides difficile*, and *Schistosoma mansoni*; and in the last 5 years has held academic industry awards with Aviex Technologies, Medicago, and MIT Canada. **Natalia Totolyan** has received fees for advisory boards or speaking from Merck and Novartis and has received institutional grants for conducting clinical trials for Alexion, BIOCAD, Janssen, Mapi, Merck, Novartis, Receptos, Roche, Sanofi, and TG Therapeutics. **Roseanne Sullivan** is an employee and stock owner of Novartis. **Xavier Montalban** has received speaking honoraria and travel expenses for participation in scientific meetings and has been a steering committee member of clinical trials or participated in advisory boards of clinical trials with AbbVie, Actelion, Alexion, Biogen, Bristol Myers Squibb/Celgene, EMD Serono, EXCEMED, F. Hoffmann-La Roche Ltd, Genzyme, Immunic, Janssen, MedDay, Merck, MS International Federation, Mylan, National MS Society, NervGen, Novartis, Sandoz, Sanofi-Genzyme, Teva, and TG Therapeutics. **Kevin Winthrop** has received consultation fees from AbbVie, AstraZeneca, Bristol Myers Squibb, Eli Lilly and Company, Galapagos, Gilead, GlaxoSmithKline, Novartis, Pfizer, Regeneron, Roche/Genentech, Sanofi, and UCB and serves on scientific advisory or data safety monitoring boards for Eli Lilly and Company, Regeneron, Roche/Genentech, and Sanofi

References

1. Hollen C, Bernard J. *Curr Neurol Neurosci Rep.* 2022;22(8):537-543. **2.** Louapre C et al. *J Neurol Neurosurg Psychiatry.* 2022;93(1):24-31. **3.** D'Abramo A et al. *Int J Infect Dis.* 2021;107:247-250. **4.** Habek M et al. ePoster presented at: *EAN* 2022; EPR169. **5.** Winthrop K et al. Poster presented at: *CMSC* 2022; DMT36. **6.** Hauser SL et al. *Mult Scler.* 2023;29(11-12):1452-1464. **7.** World Health Organization. Accessed May 26, 2023. https://covid19.who.int/