Presented at the American Academy of Neurology 2023 Annual Meeting • April 22-27, 2023 • Boston, MA, USA

013

Remibrutinib Inhibits Neuroinflammation Driven by B Cells and Myeloid Cells in Preclinical Models of Multiple Sclerosis

Bernd Kieseier,[1] Bruno Cenni,[2] Barbara Nuesslein-Hildesheim,[2] Enrico Ferrero,[2] Catherine Huck,[2,*] Denis Eichlisberger,[2,*] Marina Ziehn[1,*]

SUMMARY

1 **This preclinical, *in vivo* model study assessed the mechanism of action and efficacy of remibrutinib in EAE mouse models for MS**

2 **Remibrutinib exhibited dose-dependent efficacy in the HuMOG EAE model, underscoring its direct effects on pathogenic autoreactive B-cell antigen-presenting function; remibrutinib also exhibited efficacy in the B-cell–independent RatMOG EAE model, indicating significant contribution from the inhibition of pathogenic myeloid cells such as microglia**

3 **In both models, remibrutinib efficacy was correlated with sustained and high BTK occupancy in peripheral tissues and brain, supporting the use of higher doses to maximize efficacy**

[1]Novartis Pharma AG, Basel, Switzerland
[2]Novartis Institutes for Biomedical Research, Basel, Switzerland

*Employee of Novartis at the time of this study

Scan to download a copy of this poster
Copies of this poster and its content, obtained through this QR code, are for personal use only and may not be reproduced without written permission from the authors

Previously presented at the 38th Congress of the European Committee for Treatment and Research in Multiple Sclerosis, October 26-28, 2022

INTRODUCTION

- Bruton's tyrosine kinase (BTK) is a key signaling node in B-cell receptor and Fc receptor signaling[1]
- Inhibition of BTK offers an attractive mechanism to modulate immune regulatory networks and related neuroinflammation via inhibition of B cells and myeloid cells[1]
- BTK inhibitors are a novel class of oral therapies to prevent inflammation and disease progression in multiple sclerosis (MS) without depleting B cells[1]
- Remibrutinib is a potent, highly selective, covalent BTK inhibitor with a promising preclinical and clinical profile for the treatment of MS[2-4]
- Remibrutinib has been shown to exhibit improved target selectivity and potency *in vitro* (AAN 2022 [Poster 003])
- This study describes the efficacy and mechanism of action of remibrutinib in the B-cell–dependent recombinant human myelin oligodendrocyte glycoprotein (HuMOG)-induced experimental autoimmune encephalomyelitis (EAE) mouse model and in the B-cell–independent rat myelin oligodendrocyte glycoprotein (RatMOG)-induced EAE model[5]

OBJECTIVE

- To assess the mechanism of action and efficacy of remibrutinib in EAE mouse models for MS

METHODS

- In both models, C57BL/6 mice were immunized with either HuMOG or RatMOG to induce EAE. EAE clinical features were scored daily. BTK occupancy levels in spleen, lymph nodes, and brain were determined with immunoassays for free BTK and total BTK protein.[3] Additional assessments were for *ex vivo* T-cell recall proliferative response, serum anti-MOG antibody response, and serum neurofilament light chain (NfL) levels. Brain and spinal cord messenger RNA expression were analyzed in RatMOG EAE by single-cell RNA sequencing (scRNASeq)

RESULTS

REMIBRUTINIB INHIBITS B-CELL–DEPENDENT HuMOG EAE

- Oral dosing of 3 or 30 mg/kg twice-daily (BID) remibrutinib dose-dependently reduced clinical scores in HuMOG EAE mice, with a statistically significant inhibition for the 30-mg/kg dose (**Figure 1A**)
- BTK occupancy measured at early time points of dosing at the end of the study was near maximal in spleen and lymph nodes and showed the expected decay due to BTK protein resynthesis (**Figure 1B-C**)
- Brain BTK occupancy was near maximal for the 30-mg/kg dose, whereas it was marginal for 3-mg/kg dose (**Figure 1D**)

Figure 1. Efficacy and BTK Occupancy in HuMOG EAE

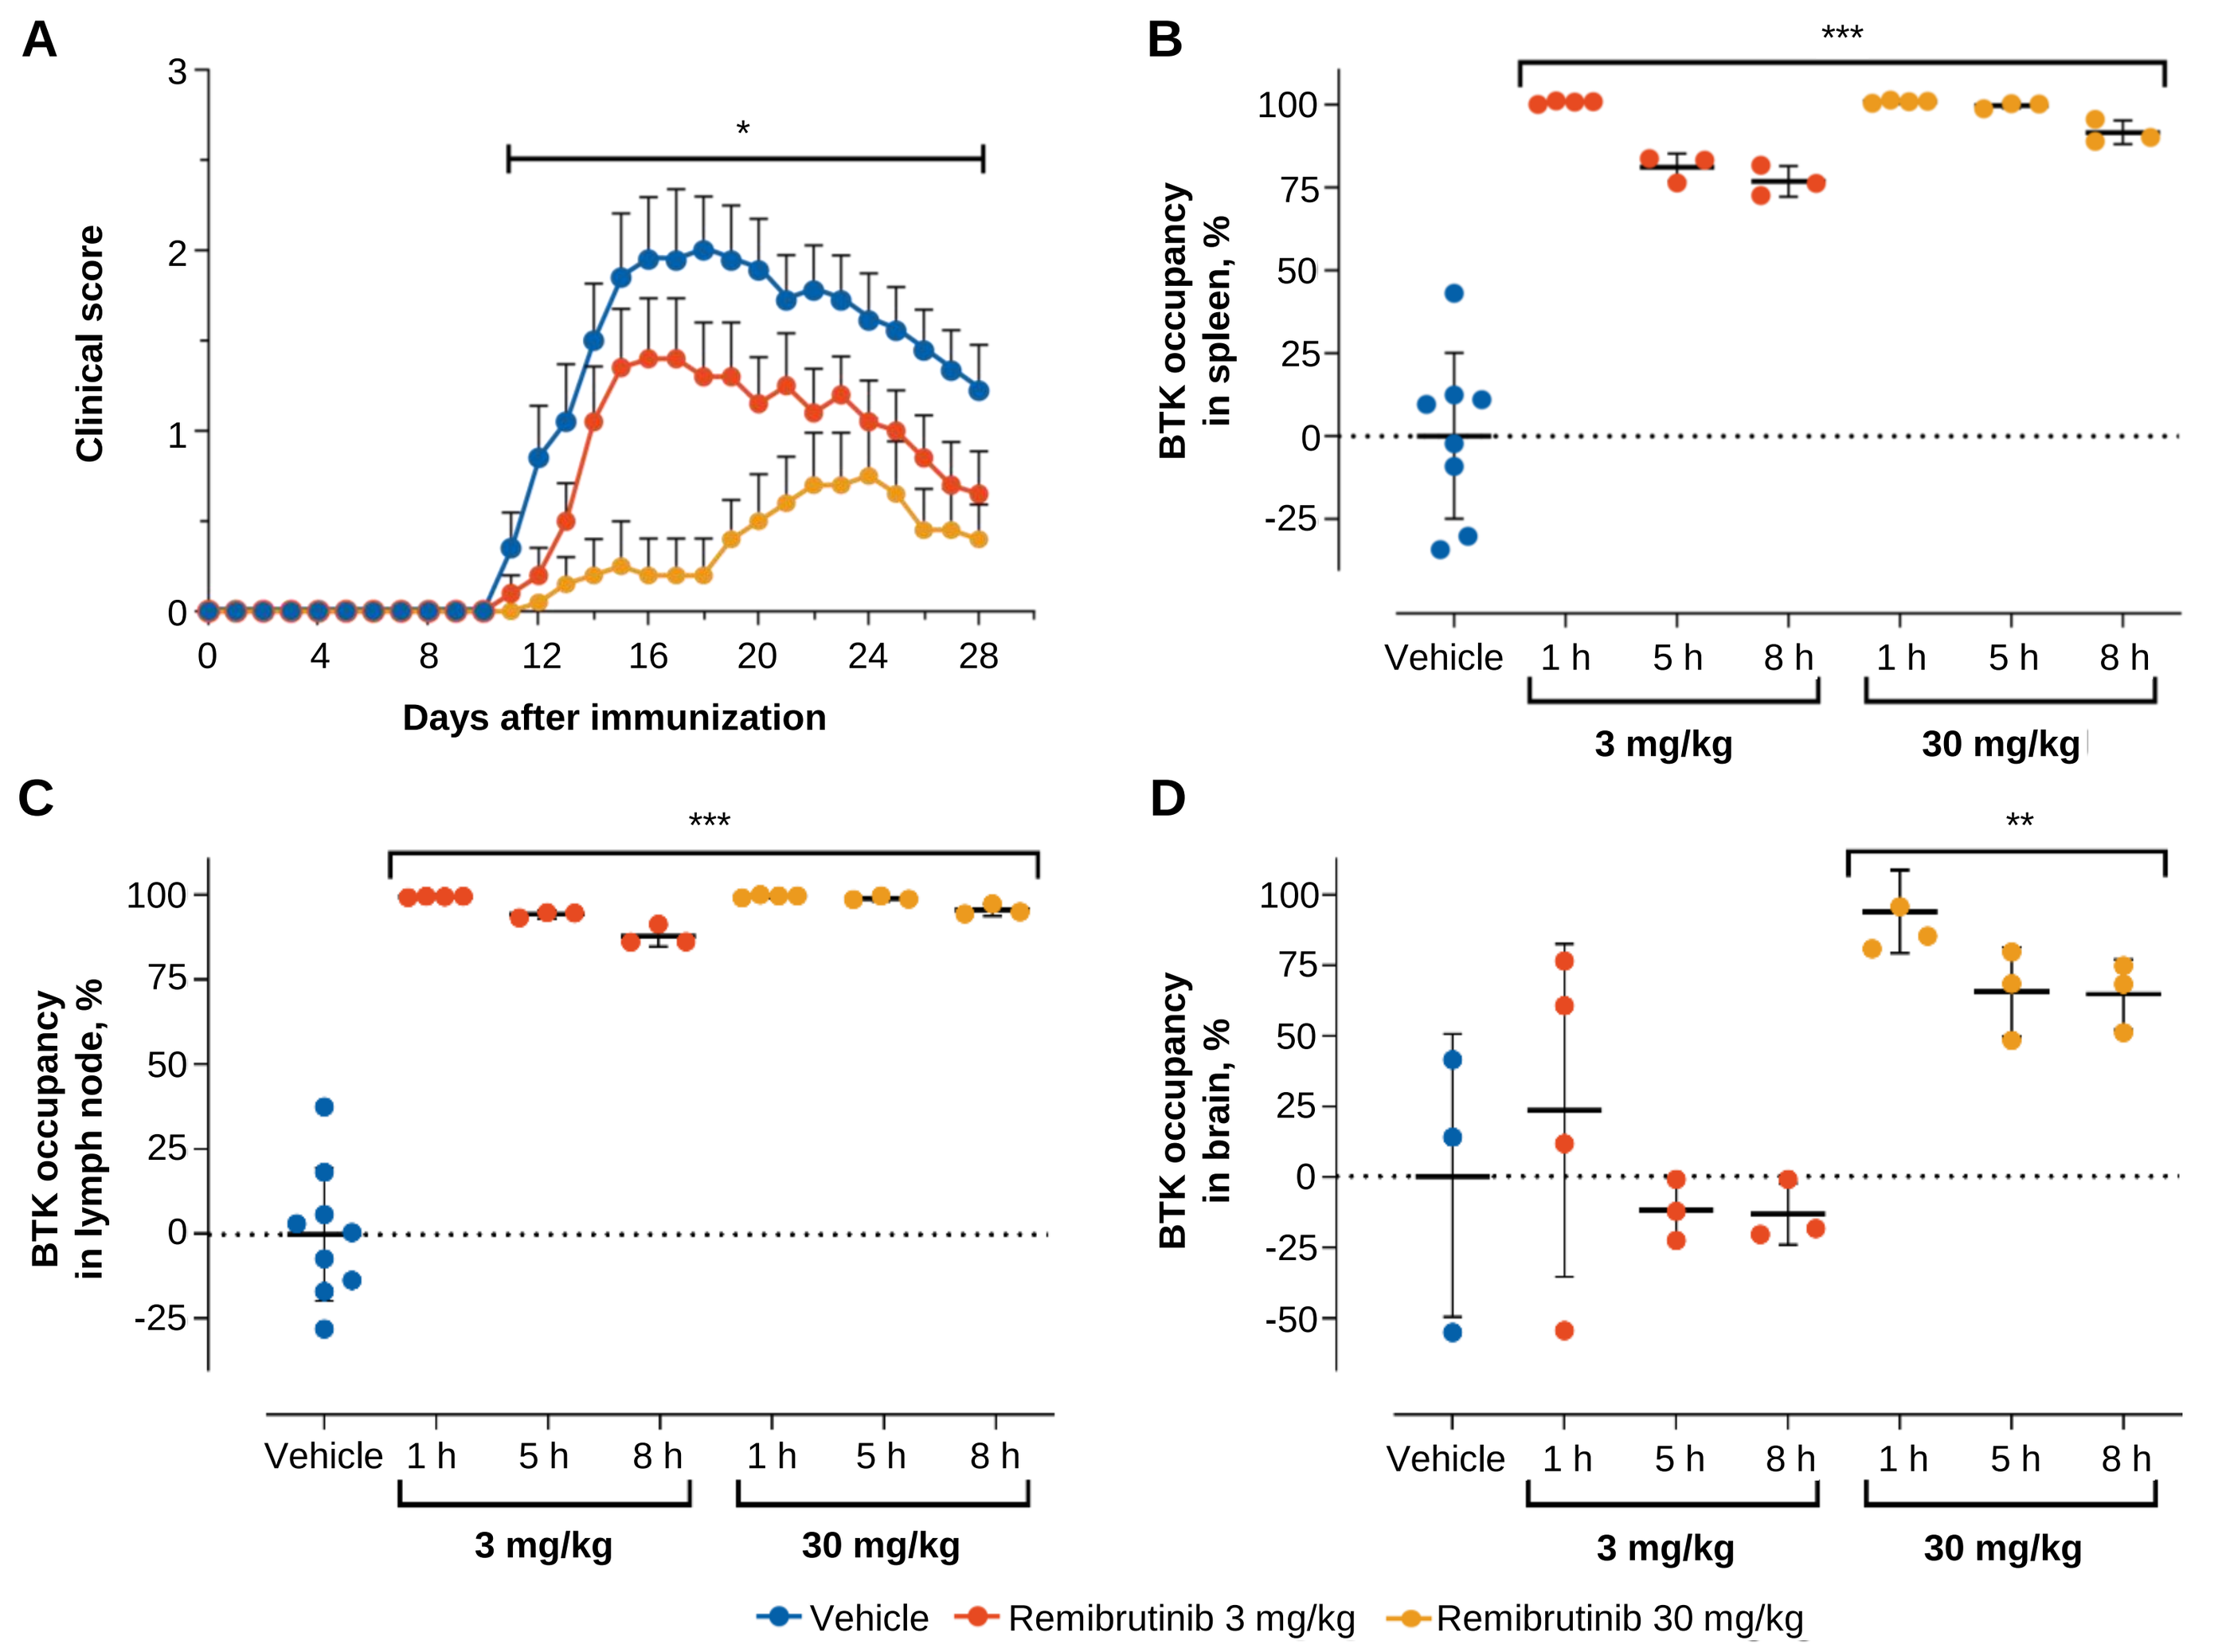
A
Clinical score
Days after immunization
B
BTK occupancy in spleen, %
C
BTK occupancy in lymph node, %
D
BTK occupancy in brain, %
Vehicle
1 h
5 h
8 h
3 mg/kg
30 mg/kg
Vehicle
Remibrutinib 3 mg/kg
Remibrutinib 30 mg/kg

ANOVA, analysis of variance; BTK, Bruton's tyrosine kinase; EAE, experimental autoimmune encephalomyelitis; huMOG, human myelin oligodendrocyte glycoprotein
Statistical significance (ANOVA with Dunnett's test) is shown as: *p<0.05; **p<0.01; ***p<0.001

- Remibrutinib inhibited *ex vivo* HuMOG-specific splenocyte T-cell recall proliferative response, but not polyclonal T-cell proliferation to anti-CD3/CD28, indicating the absence of direct T-cell immune suppression (**Figure 2**)

Figure 2. *Ex Vivo* Splenocyte HuMOG EAE Recall Proliferative Response

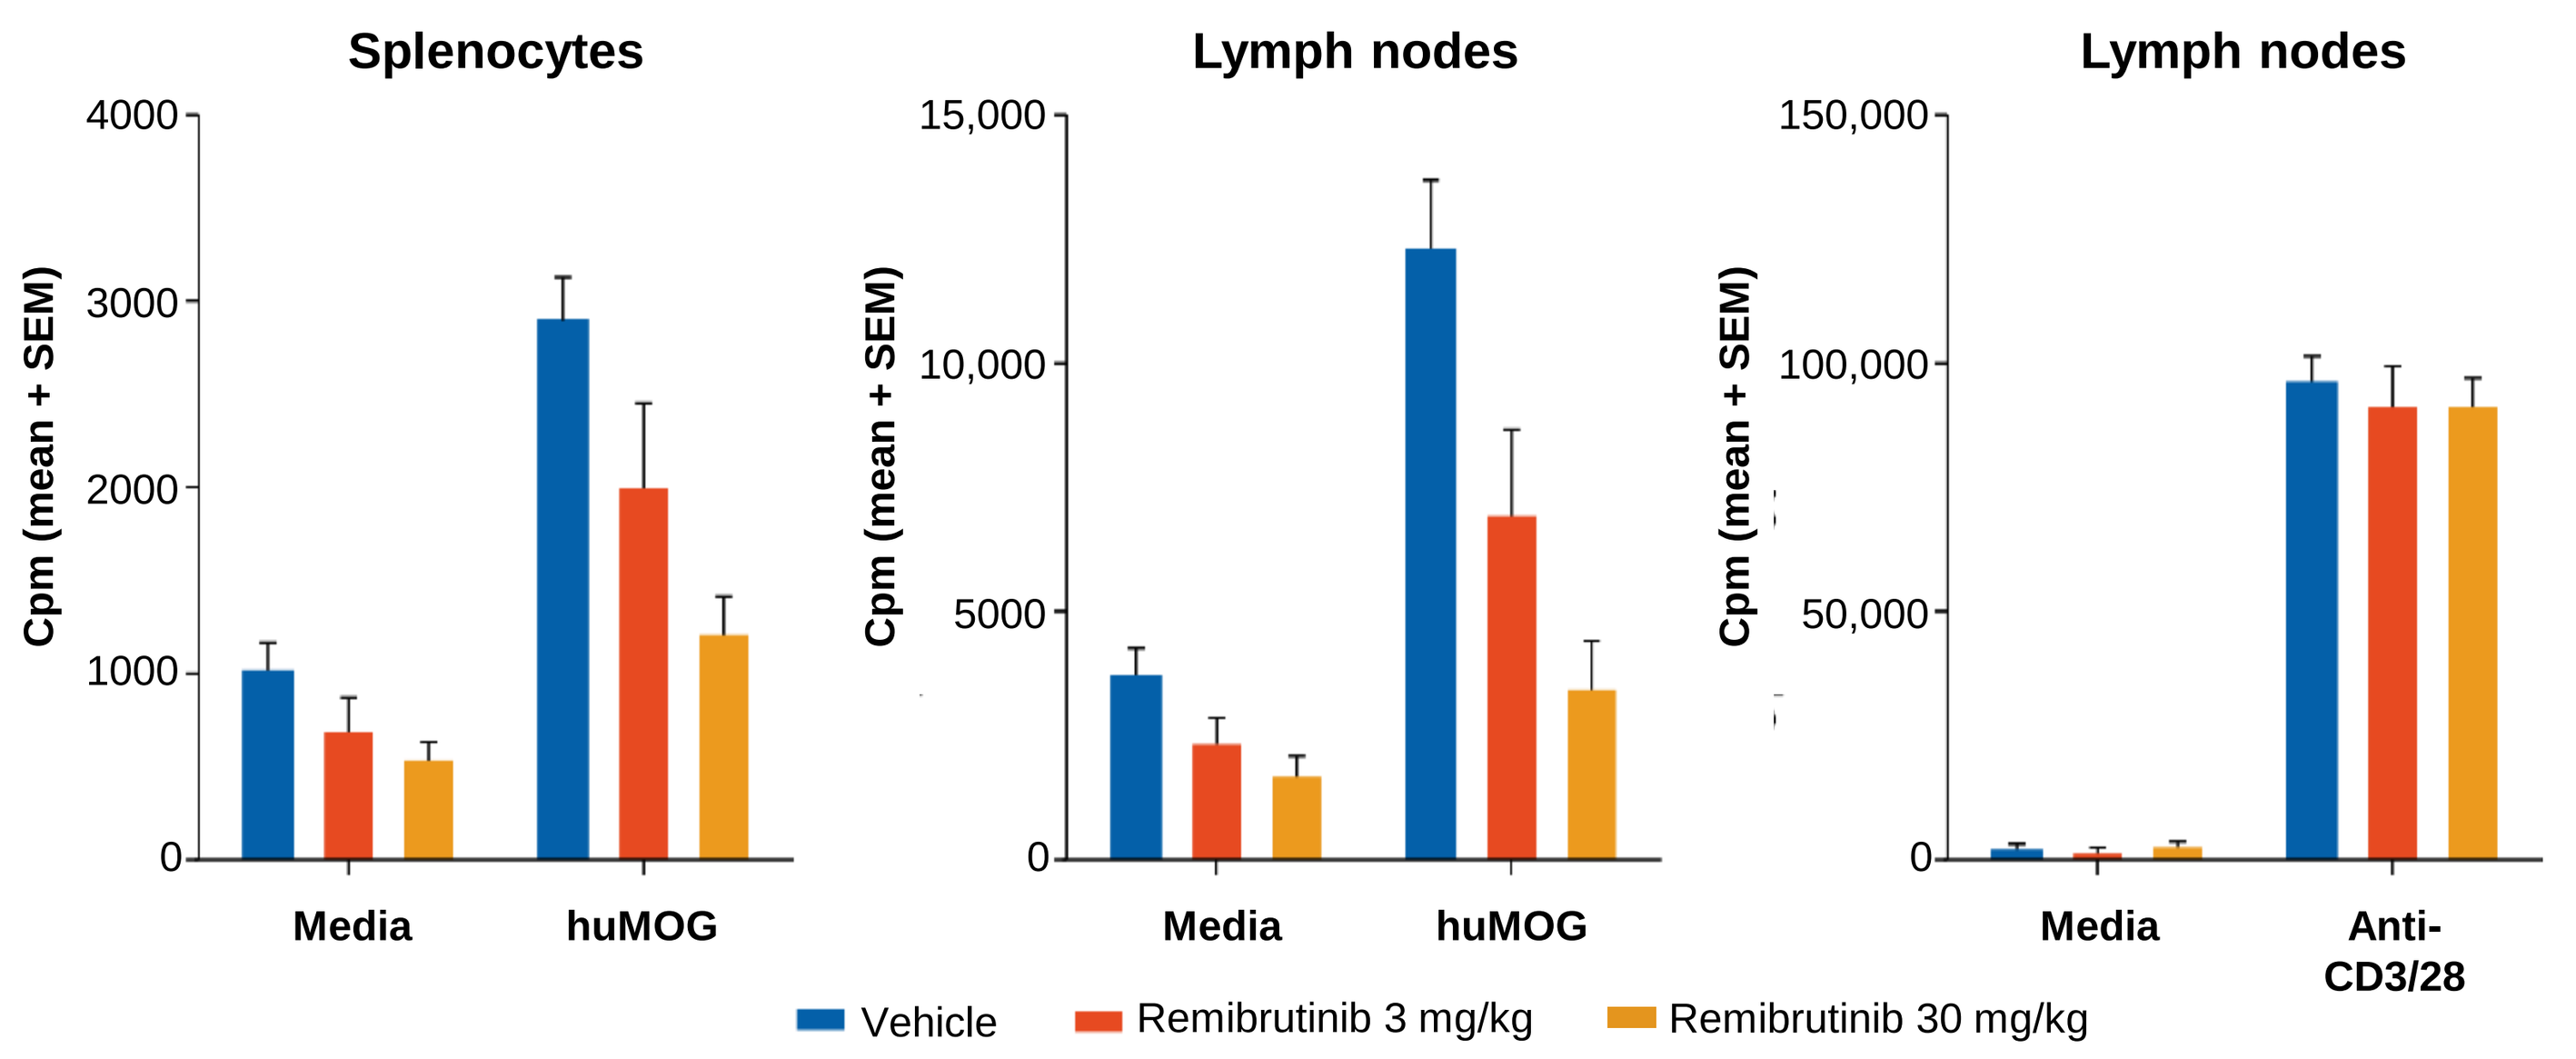
Splenocytes
Lymph nodes
Lymph nodes
Cpm (mean + SEM)
Media
huMOG
Anti-CD3/28
Vehicle
Remibrutinib 3 mg/kg
Remibrutinib 30 mg/kg

Cpm, counts per minute; EAE, experimental autoimmune encephalomyelitis; huMOG, human myelin oligodendrocyte glycoprotein; SEM, standard error of the mean

- *Ex vivo* analysis of isolated splenocytes, lymph node cells, and blood revealed no significant changes in total B-cell populations, but a clear reduction of CD4+ T-helper 17 cells (data not shown). Similarly, remibrutinib did not reduce total immunoglobulin G (IgG) antibody levels (data not shown)

REMIBRUTINIB INHIBITS RatMOG EAE

- Remibrutinib orally dosed at 30 mg/kg BID reduced EAE clinical symptoms (**Figure 3A**), suggesting that in absence of direct T-cell inhibition, the efficacy in this RatMOG EAE model is mediated by myeloid cell and microglia inhibition
- In this study, BTK occupancy was measured 16 hours after the last dose to assess minimal levels of BTK engagement, and remibrutinib showed still notable levels of BTK occupancy in the spleen (**Figure 3B**), blood (**Figure 3C**), and brain (**Figure 3D**)

Figure 3. Efficacy and BTK Occupancy in RatMOG EAE

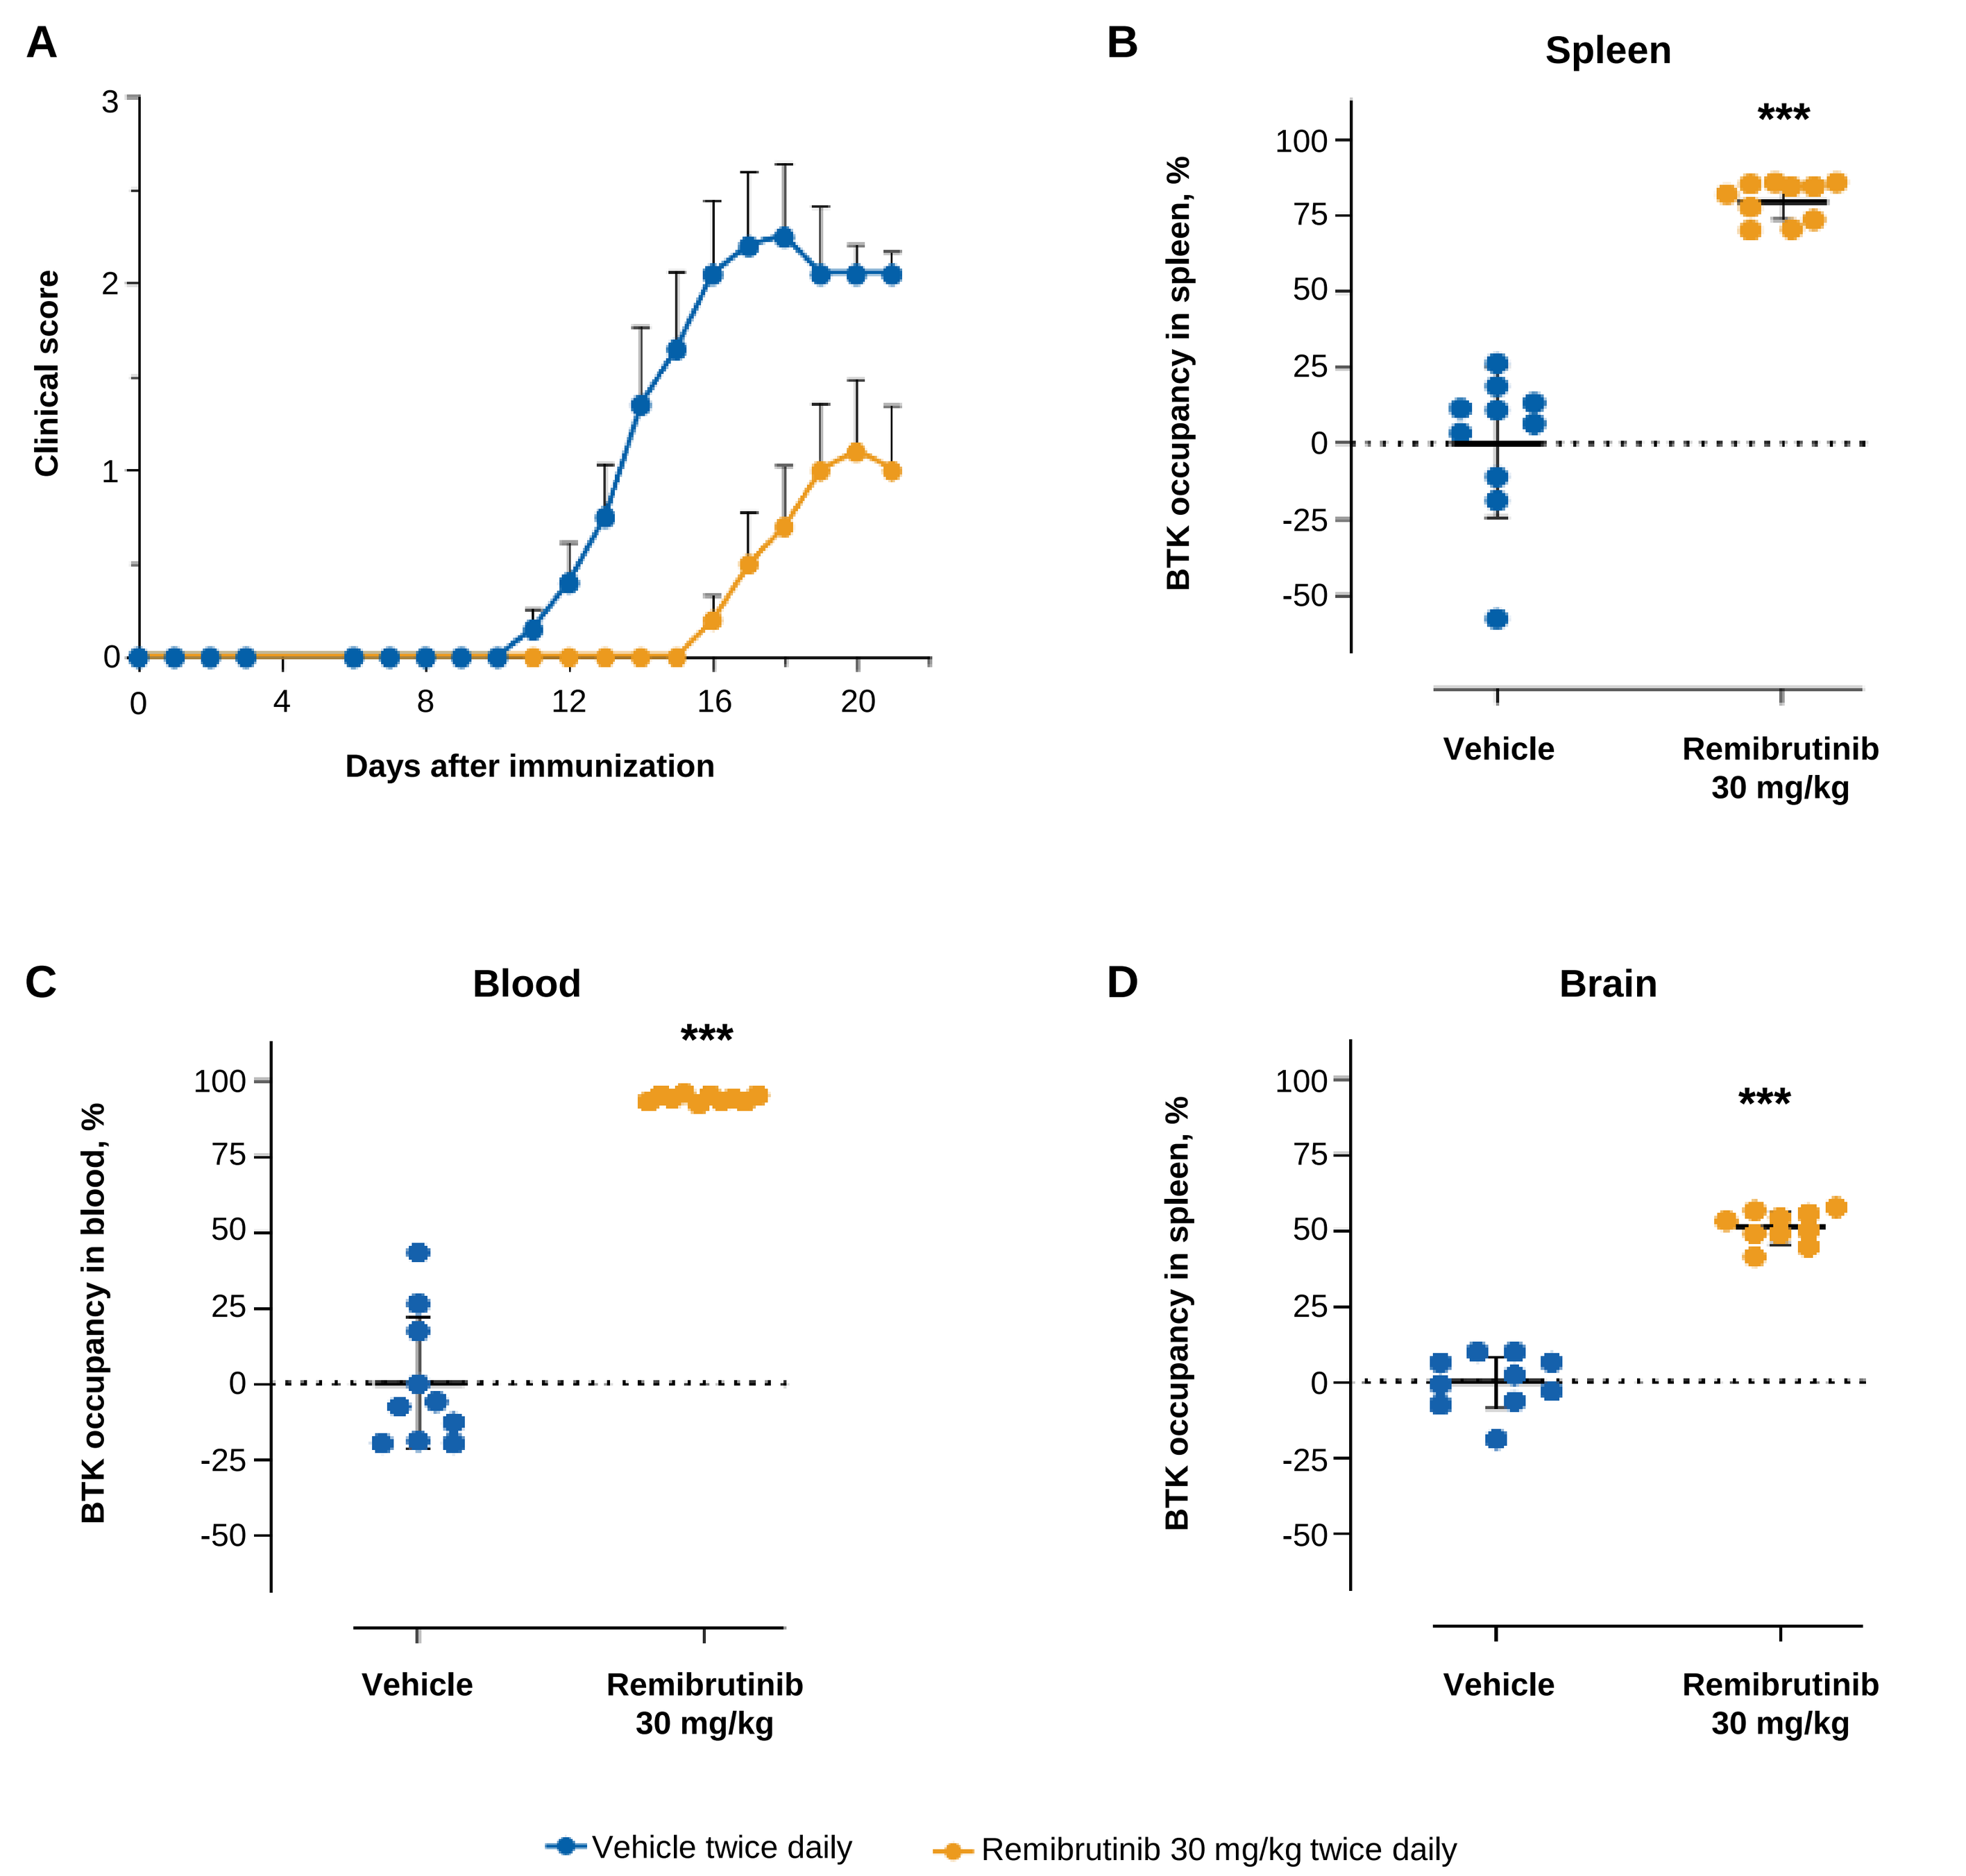
A
Clinical score
Days after immunization
B
Spleen
BTK occupancy in spleen, %
C
Blood
BTK occupancy in blood, %
D
Brain
BTK occupancy in spleen, %
Vehicle
Remibrutinib 30 mg/kg
Vehicle twice daily
Remibrutinib 30 mg/kg twice daily

ANOVA, analysis of variance; BTK, Bruton's tyrosine kinase; EAE, experimental autoimmune encephalomyelitis; RatMOG, rat myelin oligodendrocyte glycoprotein
Statistical significance (ANOVA with Dunnett's test) is shown as: *p<0.05; **p<0.01; ***p<0.001

- Remibrutinib treatment had no effect on anti-MOG serum IgM (**Figure 4A**) and IgG levels (**Figure 4B**)
- Remibrutinib reduced serum NfL levels, correlating with EAE clinical score reduction (**Figure 5**)

Figure 4. RatMOG EAE Antibody Response

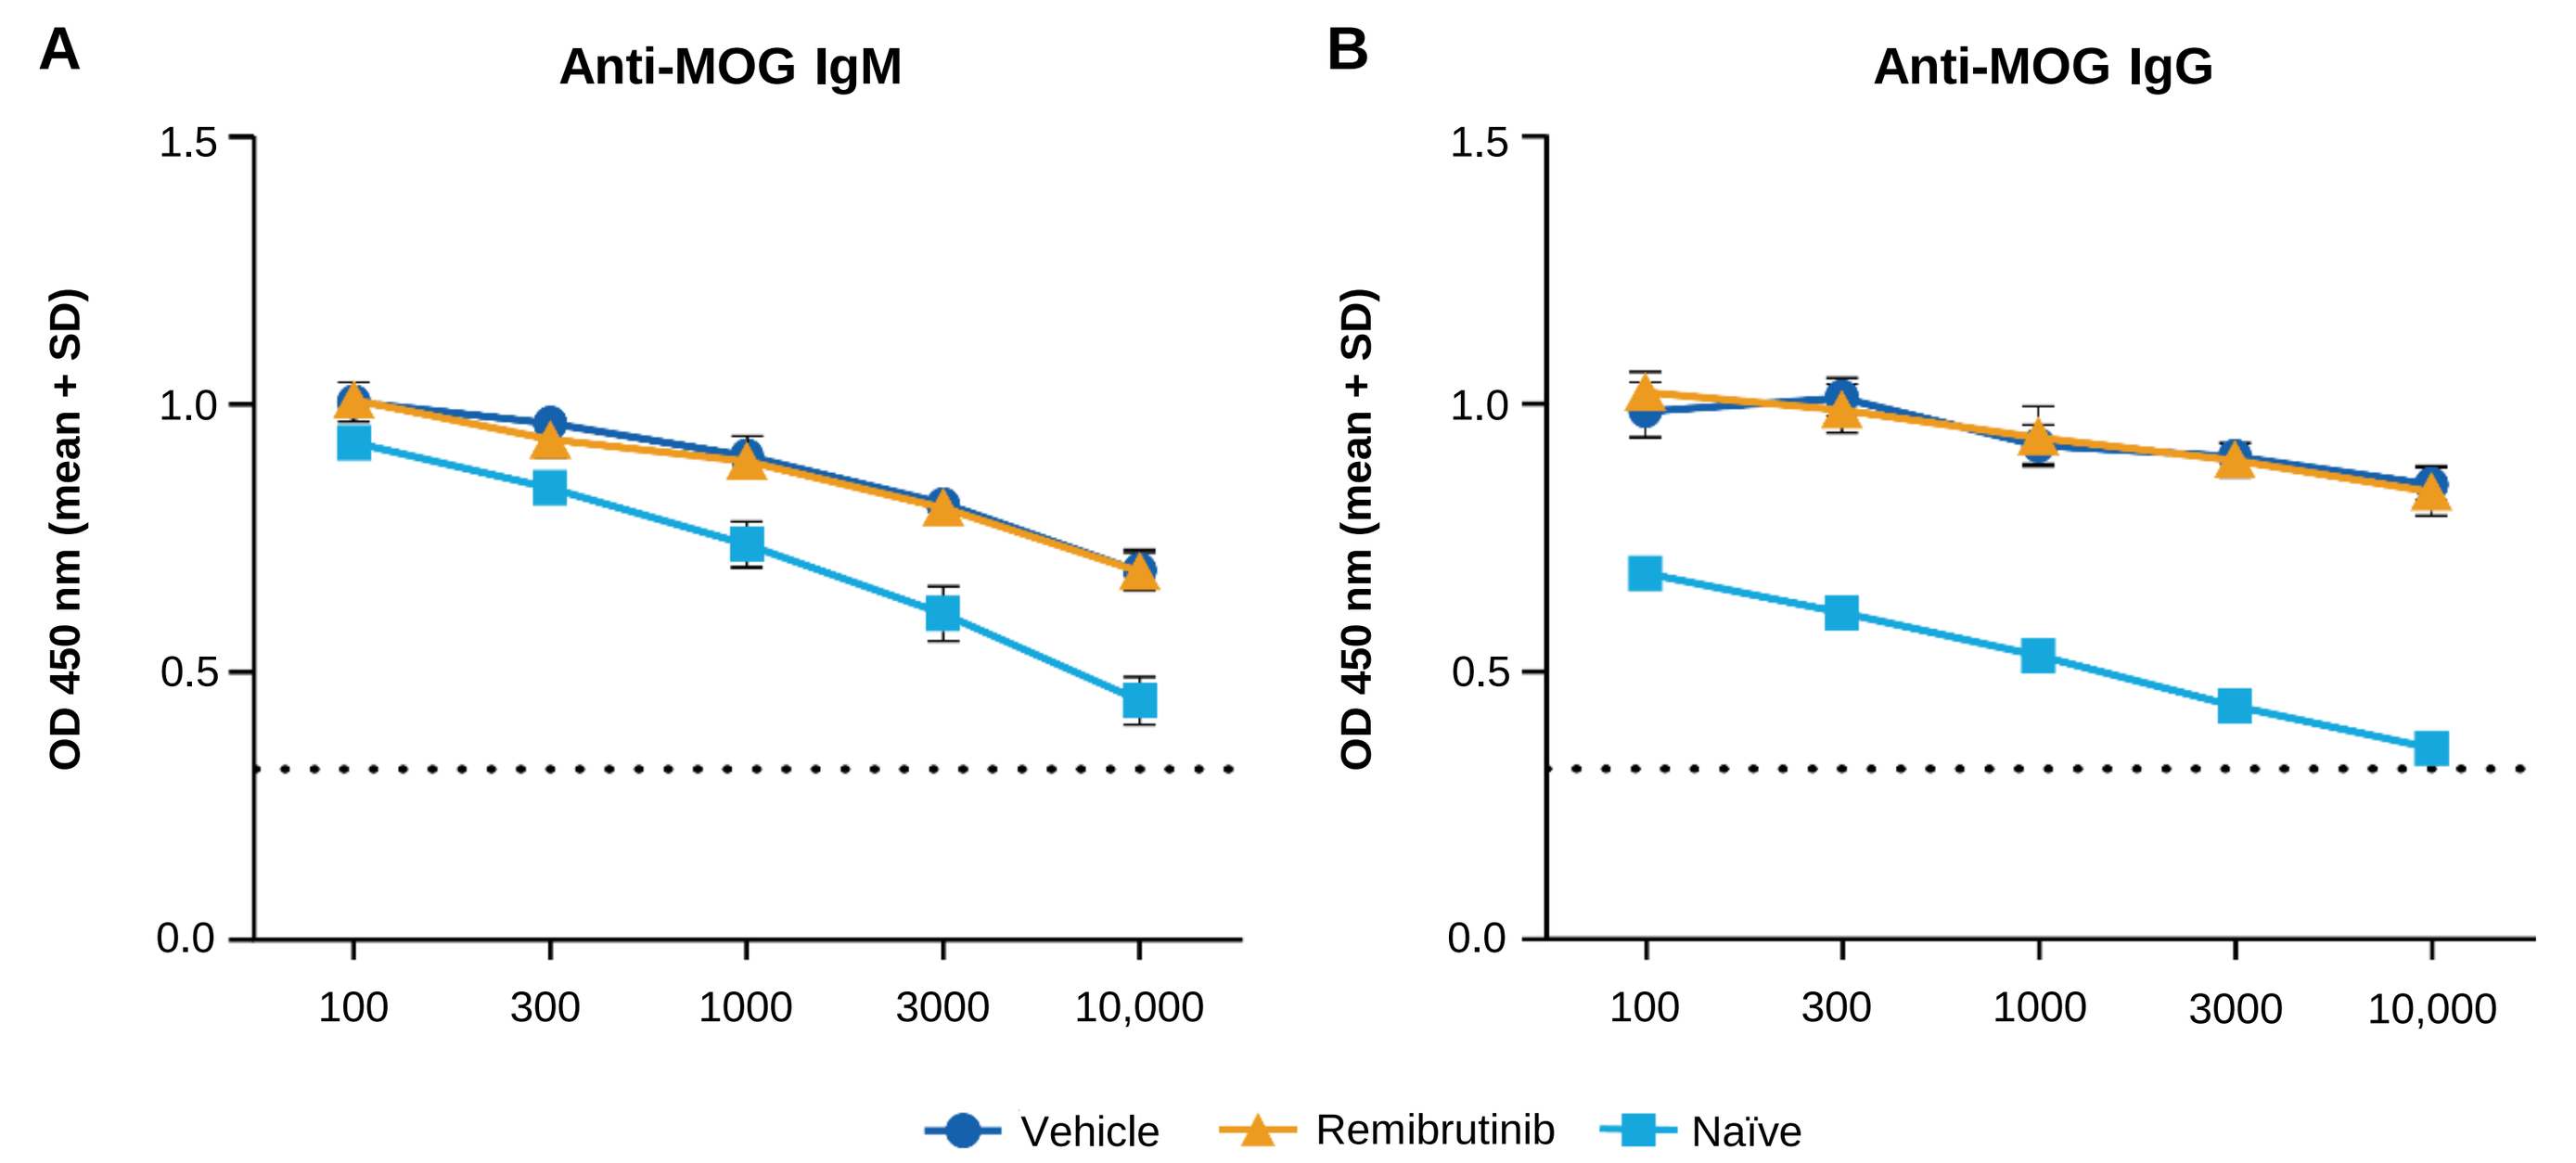
A
Anti-MOG IgM
B
Anti-MOG IgG
OD 450 nm (mean + SD)
Vehicle
Remibrutinib
Naïve

IgG, immunoglobulin G; IgM, immunoglobulin M; OD, optical density; RatMOG, rat myelin oligodendrocyte glycoprotein; SD, standard deviation

Figure 5. NfL Concentration in Serum

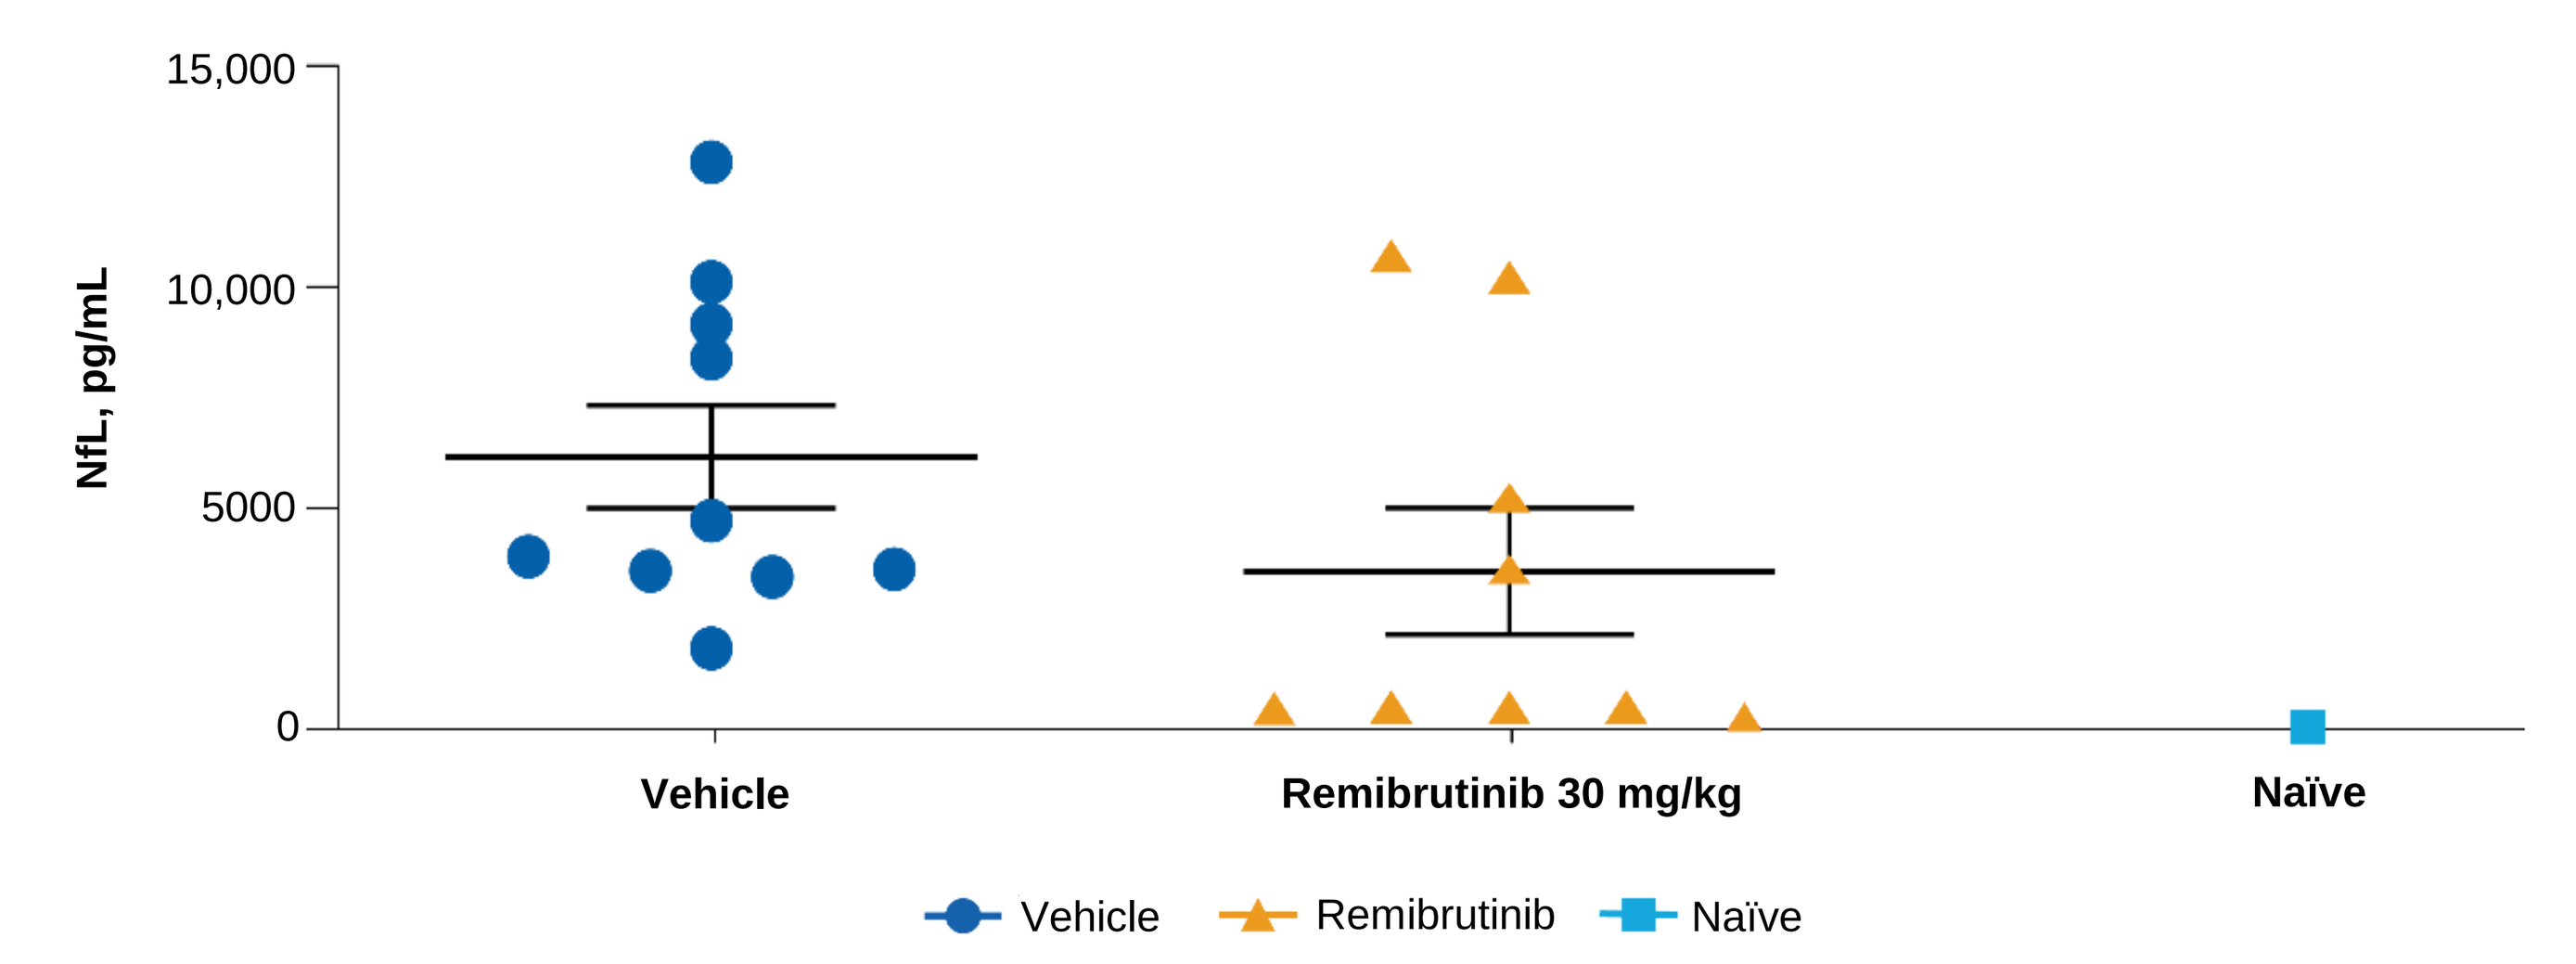
NfL, pg/mL
Vehicle
Remibrutinib 30 mg/kg
Naïve
Vehicle
Remibrutinib
Naïve

NfL, neurofilament light chain

REDUCTION OF NEUROINFLAMMATION GENE SIGNATURE IN EAE MICROGLIA

- Analysis of scRNASeq data obtained from brains and spinal cords from a separate RatMOG EAE study revealed that remibrutinib significantly downregulated multiple gene sets related to inflammation in microglia, suggesting it has an anti-inflammatory effect specifically in these cells (**Figure 6**)

Figure 6. Remibrutinib Reduces EAE Microglia Neuroinflammation Gene Expression

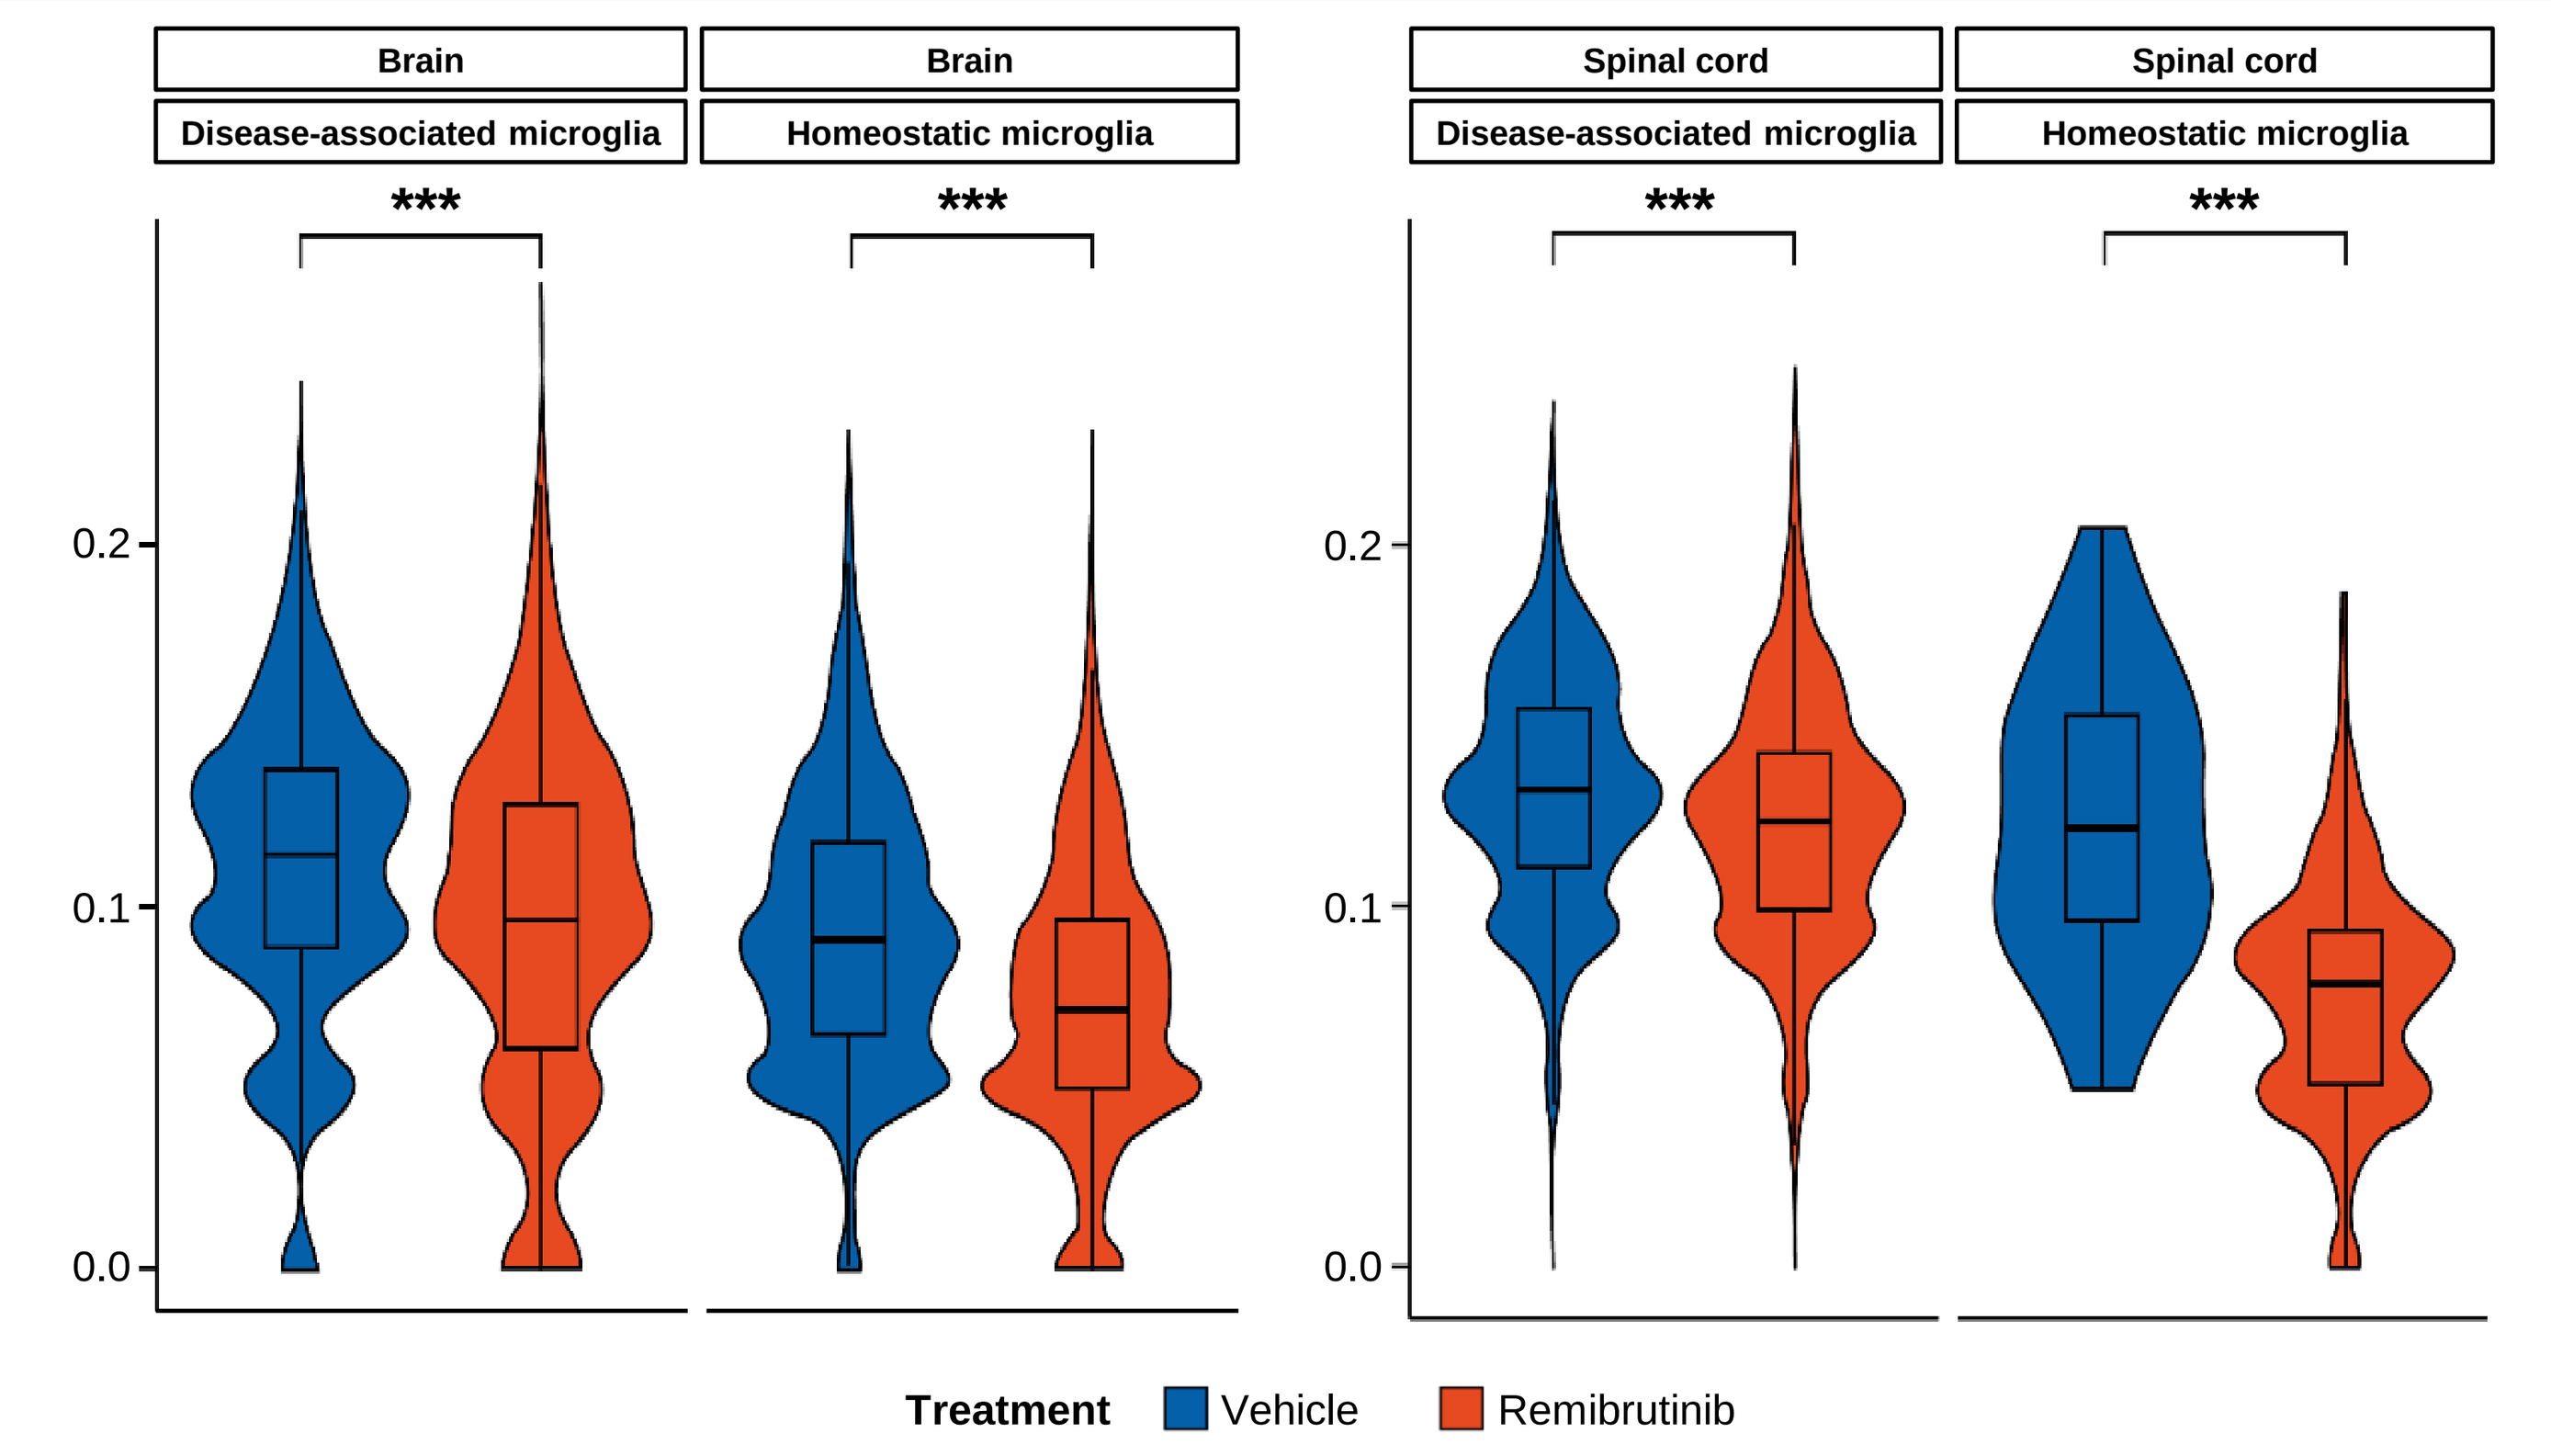
Brain
Disease-associated microglia
Brain
Homeostatic microglia
Spinal cord
Disease-associated microglia
Spinal cord
Homeostatic microglia
Treatment
Vehicle
Remibrutinib

ANOVA, analysis of variance; EAE, experimental autoimmune encephalomyelitis
Statistical significance (ANOVA with Dunnett's test) is shown as: ***p<0.001

REFERENCES: 1. Steinmaurer A et al. *Curr Pharm Des*. 2022;28(6):437-444. **2.** Kaul M et al. *Clin Transl Sci.* 2021;14(5):1756-1768. **3.** Angst D et al. *J Med Chem.* 2020;63(10):5102-5118. **4.** Maurer M et al. *J Allergy Clin Immunol.* 2022;150(6):1498-1506.e2. **5.** Smith P. *Curr Protoc*. 2021;1(6):e185.

ACKNOWLEDGMENTS: The authors acknowledge the following Novartis employees: **Vaishnavi Khandal** and **Sreelatha Komatireddy** for medical writing assistance and coordinating author reviews, and **Madhavi Kanithi** and **Mantosh Roy** for creative design assistance. Editorial assistance for this poster was provided by Envision Pharma Group and was funded by Novartis Pharmaceuticals Corporation. The final responsibility for the content lies with the authors

DISCLOSURES: The study was funded by Novartis Pharma AG, Basel, Switzerland. **Bernd Kieseier**, **Bruno Cenni**, **Barbara Nuesslein-Hildesheim**, and **Enrico Ferrero** are employees of Novartis. **Catherine Huck**, **Denis Eichlisberger**, and **Marina Ziehn** were employees of Novartis during this study